519

Efficacy and Safety of Doravirine/Islatravir (100/0.25 mg) in Adults with HIV and Renal Impairment

Frank Post, MD[1]; Princy Kumar, MD[2]; Chloe Orkin, MD[3]; Amy Colson, MD, MPH[4]; Moti N. Ramgopal, MD[5]; Alexandra Calmy, MD, PhD[6]; Hiroyuki Gatanaga, MD[7]; Jonathan Underwood, MBBS, PhD[8]; Stephanie O. Klopfer, PhD[9]; Anjana Grandhi, PhD[9]; Ciaran J. McMullan, MB, BCh, BAO (Hon), MMSc[9]; Matthew G. Johnson, MD[9]; Jason Y. Kim, MD, MSCE[9]; Rima Lahoulou, MD[9]; Michelle C. Fox, MD[9]

[1]King's College Hospital NHS Foundation Trust, London, UK; [2]Division of Infectious Diseases, Georgetown University Medical Center, Washington DC, USA; [3]SHARE Collaborative, Faculty of Medicine and Dentistry, Queen Mary University of London, London, UK; [4]Community Resource Initiative, Boston, MA and Cambridge Health Alliance, Cambridge, MA, USA; [5]Midway Immunology and Research Center, Fort Pierce, FL, USA; [6]HIV/AIDS Unit, Division of Infectious Diseases, Geneva University Hospitals, Geneva, Switzerland; [7]AIDS Clinical Center, NCGM, Tokyo, Japan; [8]Infection and Immunity, Cardiff University, Cardiff, UK and Infectious Diseases, Cardiff and Vale University Health Board, Cardiff, UK; [9]Merck & Co., Inc., Rahway, NJ, USA

Background

- Renal impairment (RI) is a common comorbidity in people living with HIV[1]. The availability of safe and effective antiretroviral therapy (ART) in people with RI is important, especially as people with HIV age
- Some antiretroviral drugs can adversely impact renal function
 - Tenofovir disoproxil fumarate (TDF) has been linked to proximal tubular dysfunction, acute kidney injury, progressive eGFR decline, and chronic kidney disease (CKD)[2]
 - Indinavir, atazanavir and lopinavir have been associated with crystal nephropathy, interstitial nephritis, and/or CKD[3]
- Other antiretrovirals (dolutegravir, bictegravir, rilpivirine, and cobicistat) alter tubular secretion of creatinine[3]
 - Using creatinine-based eGFR to assess renal function in people taking these agents may cause misclassification of kidney disease outcomes or stage[4]
- Doravirine (DOR) 100 mg, an approved NNRTI, and islatravir (ISL) 0.25 mg, an investigational nucleoside reverse transcriptase translocation inhibitor (NRTTI), is a once-daily single-tablet regimen in development for the treatment of HIV-1
 - Renal excretion of DOR is minimal (~10%), and renal function does not have a clinically relevant effect on the pharmacokinetics of DOR[5]
 - ~30% to 60% of ISL is excreted renally, and ISL is not a substrate of renal transporters; a single 60-mg dose of ISL was generally well tolerated in individuals with severe renal insufficiency[6]
- In two Phase 3 studies of virologically suppressed adults with HIV-1[7,8]
 - Switching to DOR/ISL (100/0.25 mg) was non-inferior at Week 48 to continuing the previous ART regimen (any oral ART[7] or bictegravir/emtricitabine/tenofovir alafenamide [BIC/FTC/TAF][8]) and maintained high rates of virologic suppression through Week 96
 - DOR/ISL (100/0.25 mg) was generally well tolerated through Week 96 with a safety profile similar to BIC/FTC/TAF
- In previous studies evaluating a higher dose of ISL, DOR/ISL had no adverse effects on renal function[9,10]
- This post hoc analysis explored the efficacy and safety of DOR/ISL (100/0.25 mg) by baseline renal function through Week 48 of the Phase 3, non-inferiority switch studies (**Figure 1**)
 - MK-8591A-051 (P051): open-label switch from any oral 2- or 3-drug ART (baseline ART)[7]
 - MK-8591A-052 (P052): double-blind switch from BIC/FTC/TAF[8]

Figure 1. DOR/ISL (100/0.25 mg) vs Baseline ART (P051) or BIC/FTC/TAF (P052)

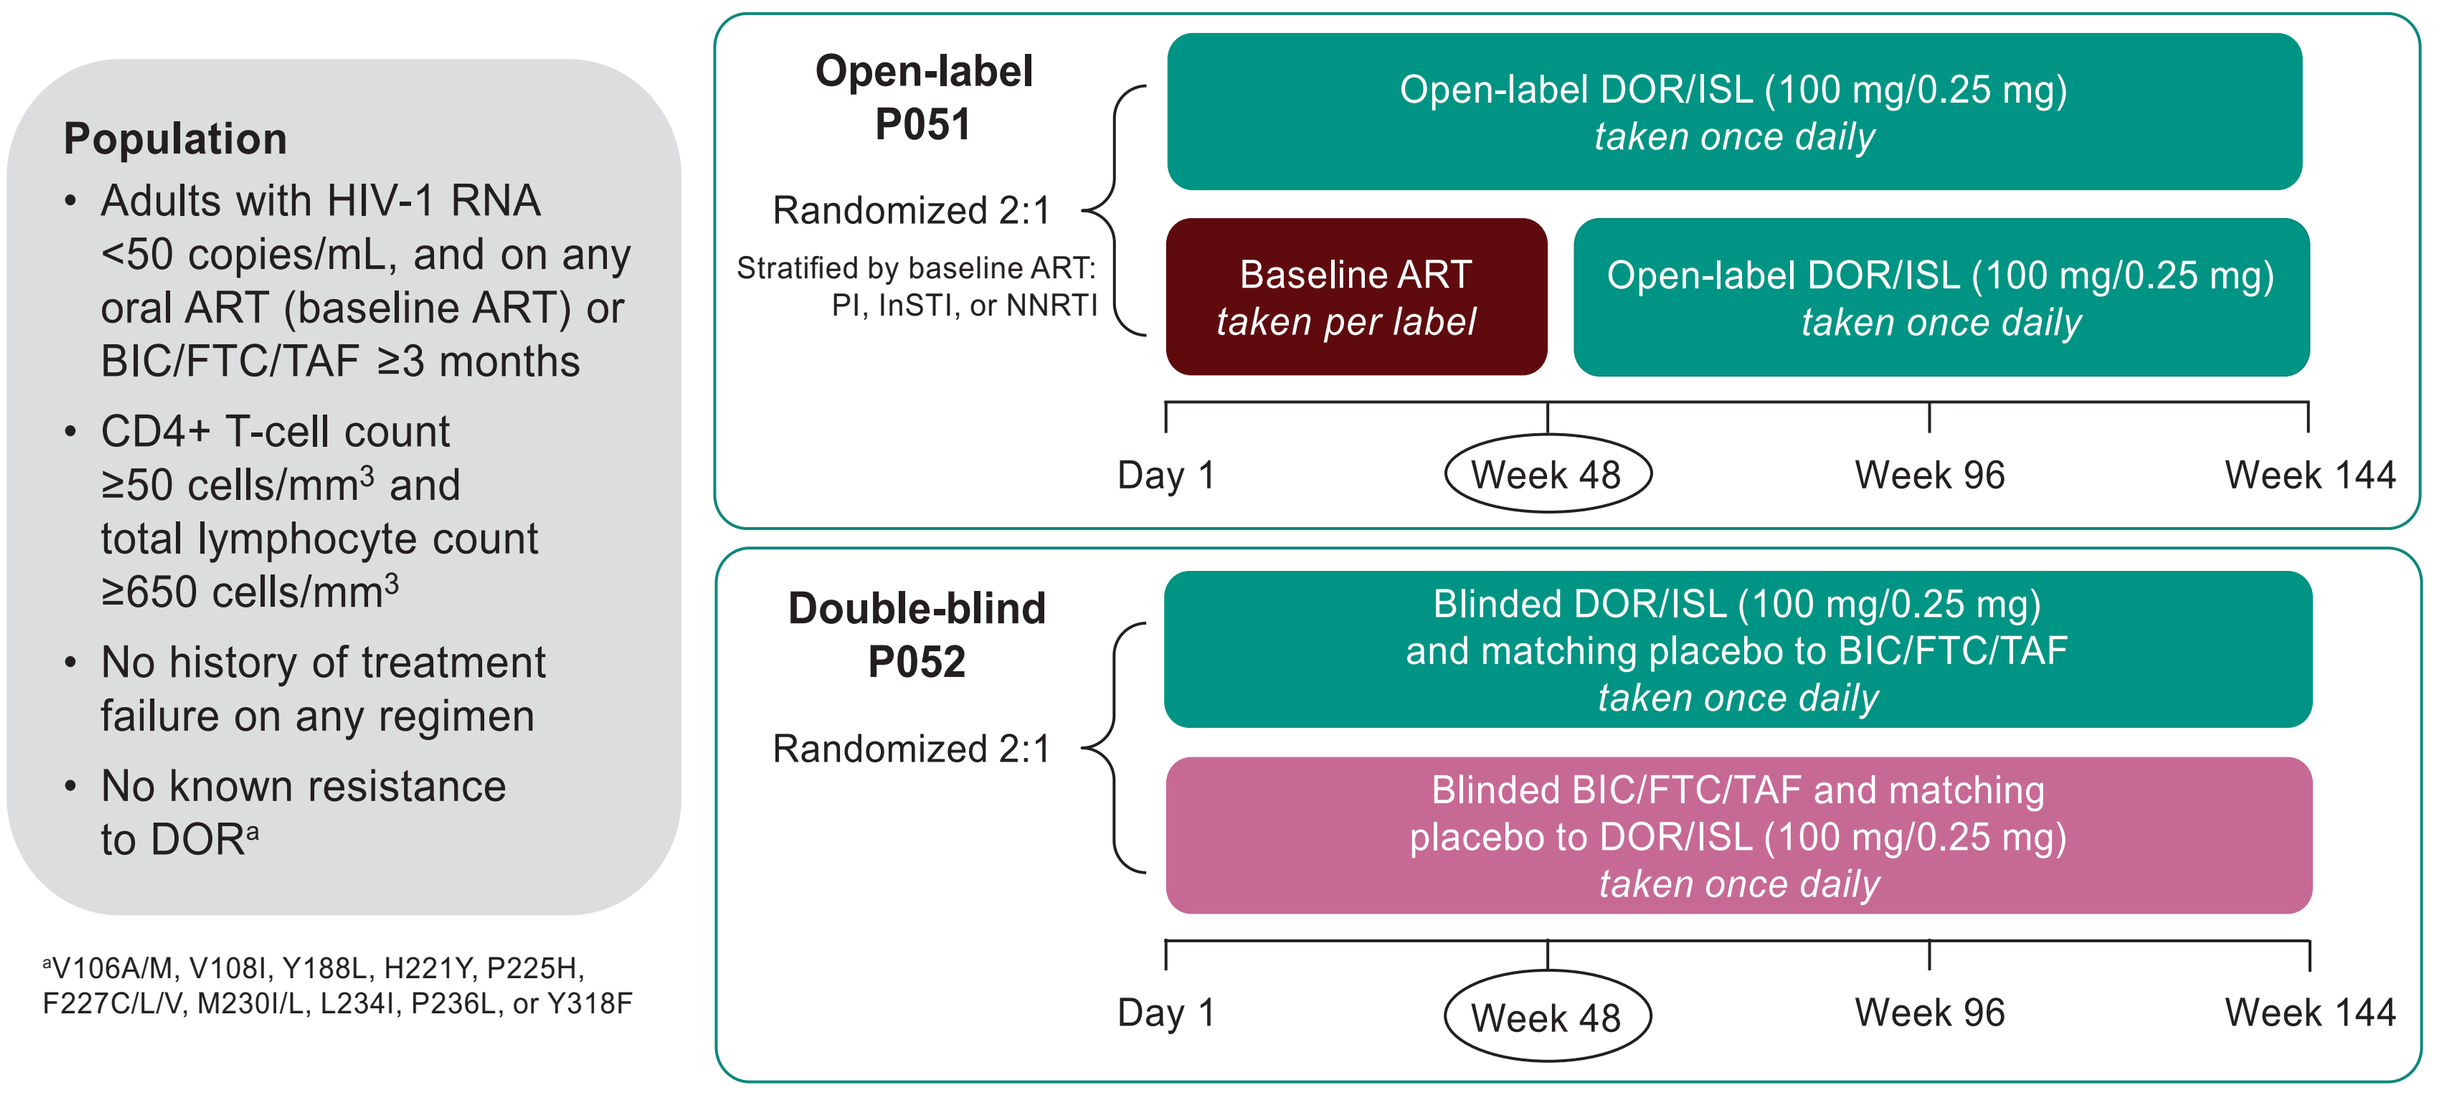
Population
Adults with HIV-1 RNA <50 copies/mL, and on any oral ART (baseline ART) or BIC/FTC/TAF ≥3 months
CD4+ T-cell count ≥50 cells/mm³ and total lymphocyte count ≥650 cells/mm³
No history of treatment failure on any regimen
No known resistance to DOR[a]
[a]V106A/M, V108I, Y188L, H221Y, P225H, F227C/L/V, M230I/L, L234I, P236L, or Y318F
Open-label P051
Randomized 2:1
Stratified by baseline ART: PI, InSTI, or NNRTI
Open-label DOR/ISL (100 mg/0.25 mg) taken once daily
Baseline ART taken per label
Open-label DOR/ISL (100 mg/0.25 mg) taken once daily
Day 1
Week 48
Week 96
Week 144
Double-blind P052
Randomized 2:1
Blinded DOR/ISL (100 mg/0.25 mg) and matching placebo to BIC/FTC/TAF taken once daily
Blinded BIC/FTC/TAF and matching placebo to DOR/ISL (100 mg/0.25 mg) taken once daily
Day 1
Week 48
Week 96
Week 144

Participants with a screening CrCl ≤30 mL/min (Cockcroft-Gault) were excluded.

Methods

- Baseline renal function was determined by creatinine-based eGFR (MDRD equation):
 - Normal renal function = eGFR ≥90 mL/min/1.73 m²
 - Mild renal impairment = eGFR 60 to <90 mL/min/1.73 m²
 - Moderate renal impairment = eGFR 30 to <60 mL/min/1.73 m²
- Efficacy and Safety endpoints at Week 48 were evaluated by baseline renal function:
 - Proportion of participants with HIV-1 RNA <50 copies/mL
 - Mean change from baseline in CD4+ T-cell count
 - Incidence of adverse events (AEs), drug-related AEs, and serious AEs
 - Mean change from baseline in eGFR based on (1) creatinine (by MDRD) and (2) cystatin C (by CKD-EPI)
- The DOR/ISL groups were pooled for the efficacy endpoints and mean change in eGFR analysis
- Additional safety endpoints included: the incidence of decreased eGFR (based on DAIDS criteria) and mean change from baseline in serum creatinine, cystatin C, urine albumin/creatinine ratio, and retinol-binding protein/creatinine ratio

Results

Participants

- Among the 708 DOR/ISL participants, 36.5% had normal renal function at baseline, 54.0% had mild renal impairment, and 9.5% had moderate renal impairment (**Table 1**)
- The majority of DOR/ISL participants (81.2%) were receiving an InSTI-based regimen before study entry
- In P051, 27.9% of the DOR/ISL group switched from a previous regimen containing TDF

Table 1. Demographics and baseline characteristics

	Pooled DOR/ISL N=708		Baseline ART N=185		BIC/FTC/TAF N=171	
Female sex at birth, n (%)	226	(31.9)	67	(36.2)	36	(21.1)
Age (years), median (IQR)	49.5	(39 to 59)	51.0	(42 to 59)	48.0	(37 to 60)
Race, n (%)						
White	347	(49.0)	74	(40.0)	106	(62.0)
Black or African American	277	(39.1)	84	(45.4)	47	(27.5)
Asian	38	(5.4)	10	(5.4)	10	(5.8)
Other	43	(6.1)	17	(9.2)	7	(4.1)
Hispanic or Latino Ethnicity, n (%)	127	(17.9)	26	(14.1)	44	(25.7)
Years since HIV-1 diagnosis, median (IQR)	12.3	(6 to 20)	13.3	(7 to 19)	10.6	(5 to 19)
Years on previous ART before enrollment, median (IQR)	3.5	(2.0 to 5.3)	3.8	(2.1 to 6.3)	3.6	(2.0 to 4.7)
Previous ART class, n (%)						
PI (including PI + InSTI)	22	(3.1)	8	(4.3)	0	(0.0)
InSTI (non-PI)	575	(81.2)	121	(65.4)	171	(100)
NNRTI (non-PI, non-InSTI)	111	(15.7)	56	(30.3)	0	(0.0)
Tenofovir in previous ART regimen, n (%)						
Tenofovir alafenamide	497	(70.2)	80	(43.2)	171	(100)
Tenofovir disoproxil fumarate	102	(14.4)	66	(35.7)	0	(0.0)
Other*	104	(14.7)	37	(20.0)	0	(0.0)
CD4+ T-cell count (cells/mm3), median (IQR)	691.5	(514 to 899)	725.0	(566 to 918)	705.0	(512 to 920)
eGFR from creatinine (mL/min/1.73 m²)						
Mean (SD)	85.3	(20.9)	88.6	(23.5)	83.6	(18.0)
Normal renal function (eGFR ≥90), n (%)	258	(36.4)	76	(41.1)	56	(32.7)
Mild renal impairment (eGFR 60 to <90), n (%)	382	(54.0)	96	(51.9)	99	(57.9)
Moderate renal impairment (eGFR 30 to <60), n (%)	67	(9.5)	12	(6.5)	16	(9.4)
eGFR from cystatin C (mL/min/1.73 m²)						
Mean (SD)	91.9	(20.4)	92.9	(20.0)	92.8	(19.5)
Normal renal function (eGFR ≥90), n (%)	397	(56.1)	105	(56.8)	97	(56.7)
Mild renal impairment (eGFR 60 to <90), n (%)	256	(36.2)	67	(36.2)	64	(37.4)
Moderate renal impairment (eGFR 30 to <60), n (%)	51	(7.2)	12	(6.5)	9	(5.3)

*Other NRTIs include: Zidovudine, Didanosine, Zalcitabine, Stavudine, Lamivudine, Abacavir, Emtricitabine, and Tenofovir not specified as alafenamide or disoproxil fumarate.

PI, protease inhibitor; InSTI, integrase inhibitor; NNRTI, non-nucleoside reverse transcriptase inhibitor.

Efficacy

- In the pooled DOR/ISL group, the proportion of participants with HIV RNA <50 copies/mL at Week 48 was generally similar for participants with normal renal function and those with mild RI or moderate RI (**Figure 2**)
- Similar efficacy by baseline renal function was also observed in the pooled comparator group

Figure 2. HIV-RNA <50 copies/mL by baseline renal function[a]

	DOR/ISL n/N (%)	Comparators n/N (%)	Treatment Difference (95% CI)[b]
All participants	663/708 (93.6)	331/356 (93.0)	0.7 (-2.4, 4.2)
Normal renal function	241/258 (93.4)	120/132 (90.9)	2.5 (-2.8, 9.1)
Mild impairment	356/382 (93.2)	183/195 (93.8)	-0.7 (-4.7, 4.1)
Moderate impairment	65/67 (97.0)	27/28 (96.4)	0.6 (-7.4, 15.1)

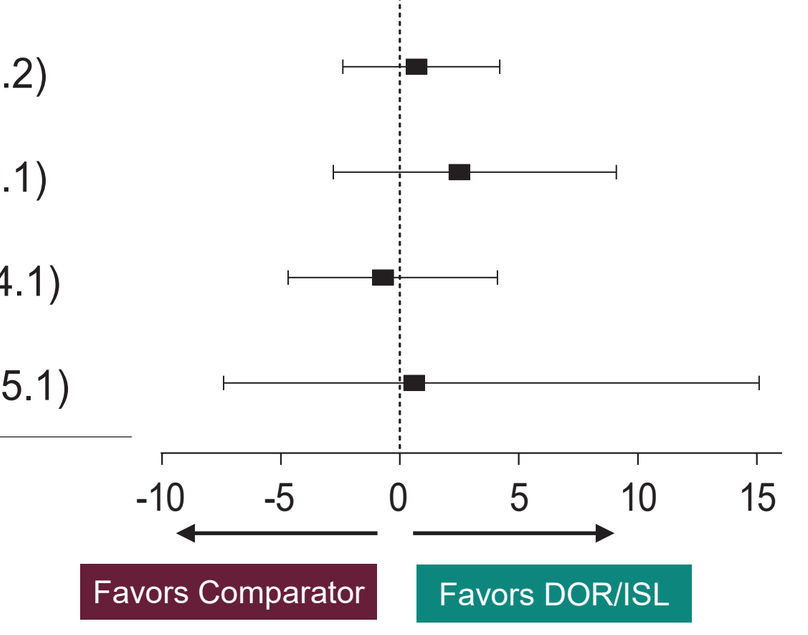
-10
-5
0
5
10
15
Favors Comparator
Favors DOR/ISL

[a]Renal function determined by creatinine-based eGFR.
[b]95% confidence intervals calculated by unstratified Miettinen and Nurminen method.

- In the pooled DOR/ISL group, mean change in CD4+ T-cell count at Week 48 was generally similar for participants with normal renal function and those with mild RI or moderate RI (**Figure 3**)
- Mean change in CD4+ T-cell count was also similar across baseline renal function categories in the pooled comparator group

Figure 3. Mean change in CD4+ T-cell count by baseline renal function*

	DOR/ISL N	DOR/ISL mean (95% CI)	Comparators N	Comparators mean (95% CI)	Treatment Difference (95% CI)[b]
All participants	667	17.2 (4.2, 30.2)	338	23.0 (0.9, 45.0)	-4.4 (-27.7, 18.8)
Normal renal function	243	20.3 (-0.3, 40.9)	127	35.5 (-2.1, 73.1)	-17.7 (-53.9, 18.5)
Mild impairment	359	17.9 (-0.7, 36.5)	185	6.3 (-23.1, 35.6)	11.2 (-20.6, 43.0)
Moderate impairment	64	4.6 (-35.2, 44.4)	26	76.8 (0.4, 153.1)	-70.1 (-156.4, 16.2)

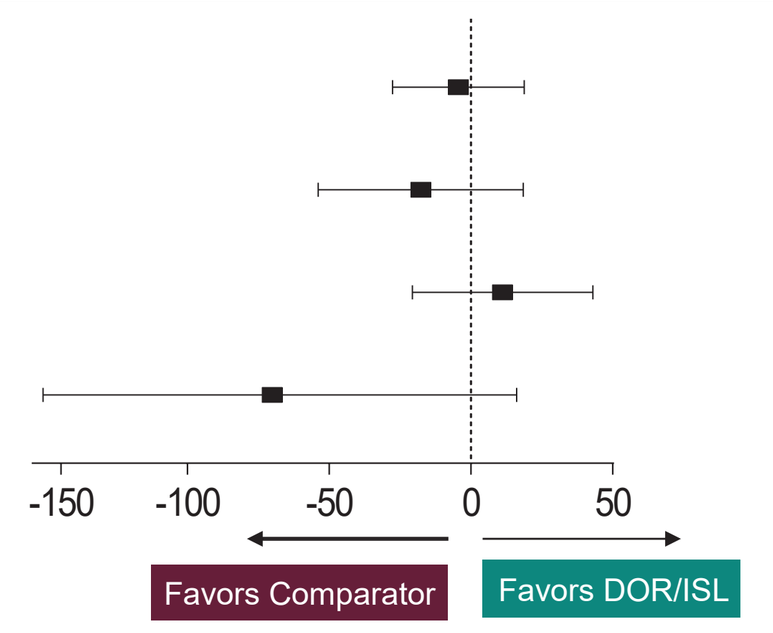
-150
-100
-50
0
50
Favors Comparator
Favors DOR/ISL

[a]Renal function determined by creatinine-based eGFR.
[b]95% CI for treatment difference based on cLDA models adjusted by treatment group, time, and interaction of time-by-treatment group.

Safety

- In the DOR/ISL groups from both studies, incidence rates for any AE, drug-related AEs, and serious AEs were generally similar for participants with normal renal function and those with mild RI or moderate RI (**Table 2**)
 - In P051, rates of drug-related AEs were higher with DOR/ISL vs bART, which has been attributed to the open-label study design

Table 2. Summary of adverse events by baseline renal function

	P051 DOR/ISL N	P051 DOR/ISL n (%)	P051 Baseline ART N	P051 Baseline ART n (%)	P052 DOR/ISL N	P052 DOR/ISL n (%)	052 BIC/FTC/TAF N	052 BIC/FTC/TAF n (%)
Any AE								
Overall	366	291 (79.5)	185	155 (83.8)	342	255 (74.6)	171	122 (71.3)
Normal renal function	140	117 (83.6)	76	69 (90.8)	118	82 (69.5)	56	38 (67.9)
Mild RI	192	151 (78.6)	96	75 (78.1)	190	145 (76.3)	99	71 (71.7)
Moderate RI	33	22 (66.7)	12	10 (83.3)	34	28 (82.4)	16	13 (81.3)
Drug-related AEs								
Overall	366	44 (12.0)	185	9 (4.9)	342	35 (10.2)	171	16 (9.4)
Normal renal function	140	17 (12.1)	76	5 (6.6)	118	10 (8.5)	56	7 (12.5)
Mild RI	192	25 (13.0)	96	3 (3.1)	190	22 (11.6)	99	6 (6.1)
Moderate RI	33	2 (6.1)	12	1 (8.3)	34	3 (8.8)	16	3 (18.8)
Serious AEs								
Overall	366	23 (6.3)	185	9 (4.9)	342	15 (4.4)	171	11 (6.4)
Normal renal function	140	6 (4.3)	76	6 (7.9)	118	6 (5.1)	56	2 (3.6)
Mild RI	192	14 (7.3)	96	3 (3.1)	190	9 (4.7)	99	9 (9.1)
Moderate RI	33	3 (9.1)	12	0 (0.0)	34	0 (0.0)	16	0 (0.0)

Glomerular Filtration Rate (eGFR)

- In all renal function categories, eGFR-creatinine increased in participants who switched to DOR/ISL vs those who remained on bART or BIC/FTC/TAF, consistent with the inhibitory effects of some antiretrovirals on creatinine secretion, while eGFR-cystatin C was comparable across all groups (**Figure 4**)

Figure 4. Mean change from baseline to Week 48 (95% CI) in eGFR-creatinine vs eGFR-cystatin C by baseline renal function

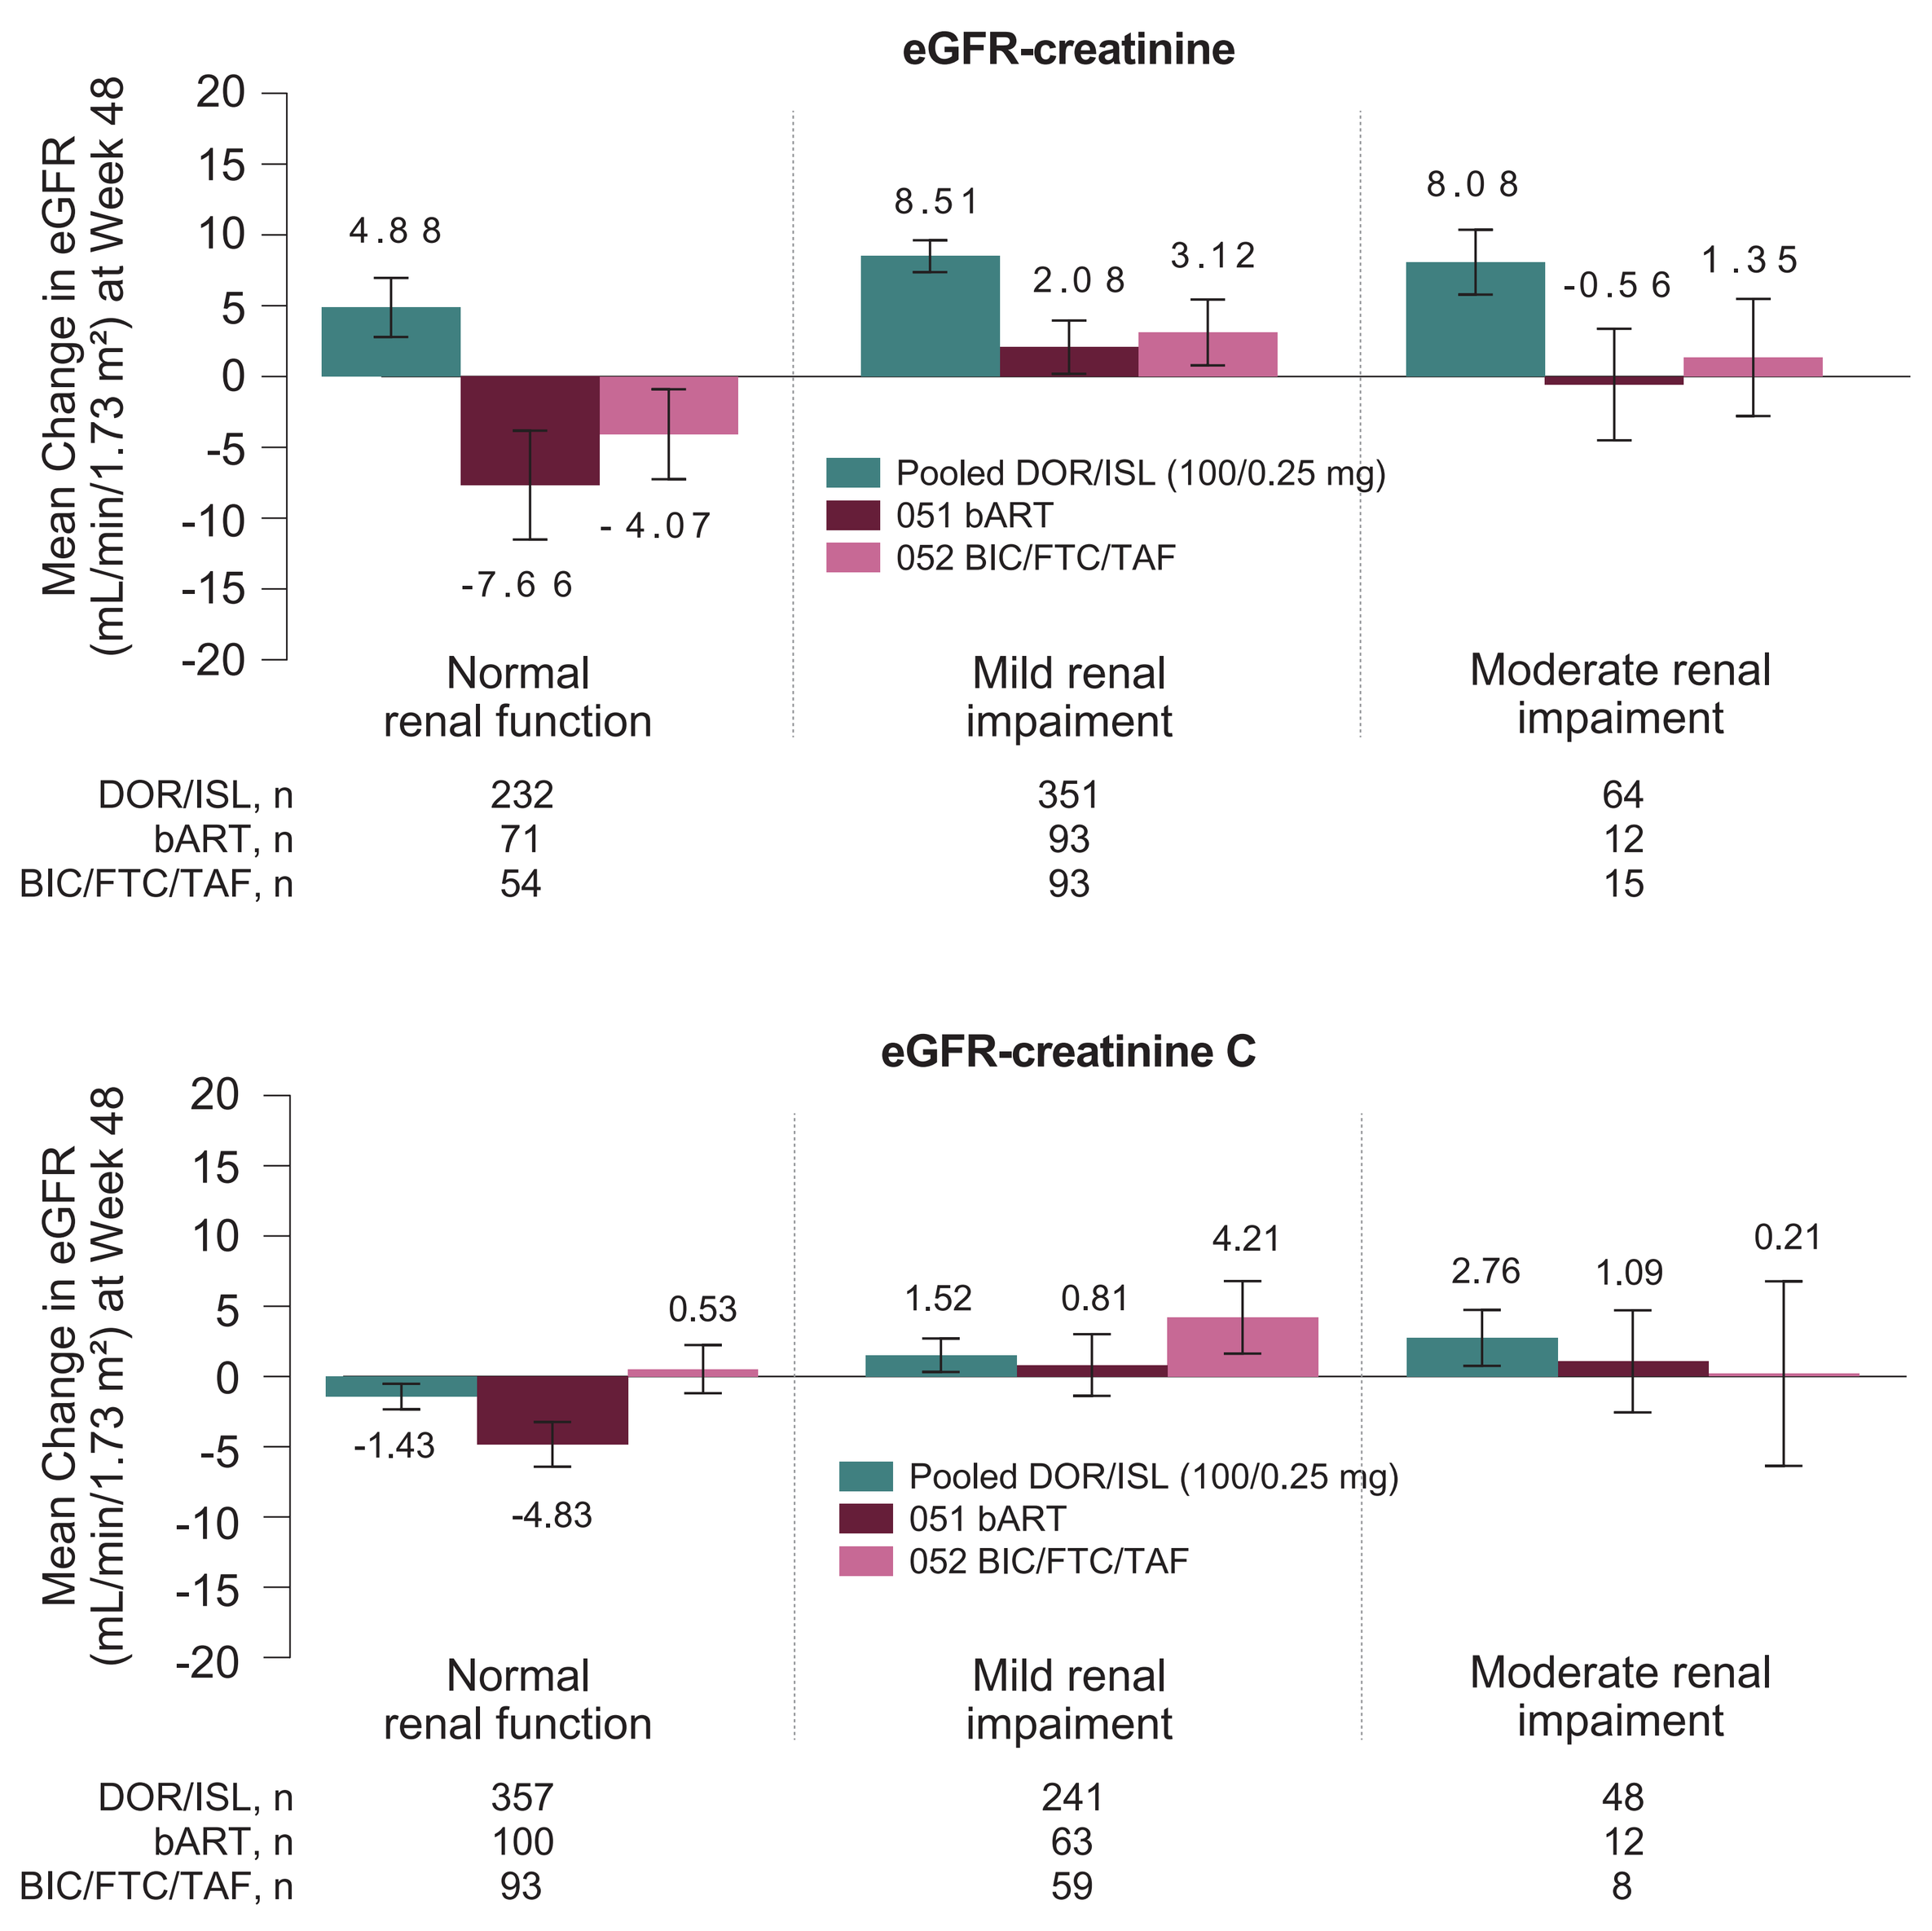
eGFR-creatinine
Mean Change in eGFR (mL/min/1.73 m²) at Week 48
4.8 8
-7.6 6
- 4.07
8.51
2.0 8
3.12
8.0 8
-0.5 6
1.3 5
Pooled DOR/ISL (100/0.25 mg)
051 bART
052 BIC/FTC/TAF
Normal renal function
Mild renal impairment
Moderate renal impairment
DOR/ISL, n 232 351 64
bART, n 71 93 12
BIC/FTC/TAF, n 54 93 15
eGFR-creatinine C
Mean Change in eGFR (mL/min/1.73 m²) at Week 48
-1.43
-4.83
0.53
1.52
0.81
4.21
2.76
1.09
0.21
Pooled DOR/ISL (100/0.25 mg)
051 bART
052 BIC/FTC/TAF
Normal renal function
Mild renal impairment
Moderate renal impairment
DOR/ISL, n 357 241 48
bART, n 100 63 12
BIC/FTC/TAF, n 93 59 8

- Decreases in eGFR-creatinine were less common with DOR/ISL than with bART or BIC/FTC/TAF, while decreases in eGFR-cystatin C were similar across the treatment groups (**Table 3**)
- Few participants had DAIDS grade 3 or 4 decreases in eGFR through Week 48

Table 3. Incidence of decreased eGFR, by DAIDS grading system

Criterion	P051 DOR/ISL	P051 bART	P052 DOR/ISL	P052 BIC/FTC/TAF
eGFR-creatinine, n/N (%)				
Grade 2: <90 to 60, or 10 to <30%↓from baseline	58/365 (15.9)	54/185 (29.2)	43/340 (12.6)	39/171 (22.8)
Grade 3: <60 to 30, or 30 to <50%↓from baseline	14/365 (3.8)	14/185 (7.6)	9/340 (2.6)	17/171 (9.9)
Grade 4: <30 or ≥50%↓from baseline	2/365 (0.5)	0/185	0/340	0/171
eGFR-cystatin C, n/N (%)				
Grade 2: <90 to 60, or 10 to <30%↓from baseline	50/358 (14.0)	46/180 (25.6)	50/319 (15.7)	24/165 (14.5)
Grade 3: <60 to 30, or 30 to <50%↓from baseline	12/358 (3.4)	2/180 (1.1)	8/319 (2.5)	3/165 (1.8)
Grade 4: <30 or ≥50%↓from baseline	1/358 (0.3)	0/180	0/319	0/165

Grades are based on the NIH DAIDS Table for Grading the Severity of Adult and Pediatric Adverse Events, version 2.1.

Renal Laboratory Markers

- For serum creatinine, serum cystatin C, and urine albumin/creatinine ratio, the mean change from baseline to Week 48 was minimal and generally similar between DOR/ISL and the comparators (**Table 4**)
- The mean change from baseline in retinol binding protein/creatinine ratio was minimal for both treatment groups in P052 (**Table 4**) and was influenced by large outliers in P051, where the median change (95% CI) was similar for DOR/ISL (−9.9 [-14.3, -3.3]) and bART (-8.6 [-18.9, 0.7])

Table 4. Mean change from baseline (95% CI) in renal laboratory markers

	n	P051 DOR/ISL	n	P051 bART	n	P052 DOR/ISL	n	P052 BIC/FTC/TAF
Serum Creatinine (mg/dL)								
Baseline mean	320	0.95	177	0.95	314	1.01	162	1.00
Mean change (95% CI)	320	-0.07 (-0.08, -0.05)	177	0.01 (-0.01, 0.02)	314	-0.07 (-0.08, -0.06)	162	-0.01 (-0.02, 0.01)
Serum Cystatin C (mg/L)								
Baseline mean	336	0.92	175	0.90	310	0.92	160	0.91
Mean change (95% CI)	336	0.00 (-0.01, 0.01)	175	0.02 (0.01, 0.03)	310	-0.00 (-0.01, 0.01)	160	-0.01 (-0.03, -0.00)
Urine Albumin/Creatinine Ratio (mg/g)								
Baseline mean	344	0.03	174	0.02	311	0.02	160	0.01
Mean change (95% CI)	344	-0.01 (-0.02, 0.00)	174	0.00 (-0.01, 0.01)	311	-0.01 (-0.01, 0.00)	160	-0.00 (-0.00, 0.00)
Retinol Binding Protein/Creatinine Ratio (µg/g)								
Baseline mean	334	672	164	478	298	499	154	153
Mean change (95% CI)	334	-475 (-1195, 246)	164	612 (-891, 2115)	298	25.8 (-66.6, 118)	154	-3.99 (-30.8, 22.8)

Within-group 95% CIs are based on the t-distribution.

Conclusions

- DOR/ISL (100/0.25 mg) demonstrated similar efficacy and safety in participants with mild or moderate RI compared to those with normal renal function
- For participants in all renal function categories, switching to DOR/ISL did not adversely impact renal function through Week 48

References

1. Naicker S. *Clin Nephrol.* 2020 Supplement-Jan;93(1):87-93.
2. Hughes C. *Current Opinion in HIV and AIDS.* 2021;16:303-308.
3. Heron JE, et al. *AIDS Res Ther.* 2020;17:11.
4. Brunet L, et al. *Antiviral Ther.* 2020;25:287–292.
5. Khalilieh S, et al. *Clinical Drug Investigation* 2020; 40:927–946.
6. Matthews RP, et al. *Antimicrob Agents Chemother.* 2022; 66(12):e0093122.
7. Orkin C, et al. *Lancet.* 2026;407:599–610.
8. Colson A, et al. *Lancet.* 2026;407:611-621.
9. Post F, et al. Renal safety parameters after switch to doravirine/islatravir (DOR/ISL 100/0.75mg) once-daily: week 48 results from 2 randomized, active-controlled phase 3 trials, MK8591A-017 (P017) and MK8591A-018 (P018). Presented at the 12th International AIDS Society (IAS) Conference on HIV Science 2023; Brisbane, Australia; July 23-26, 2023.
10. Post F, et al. Renal safety parameters of doravirine/islatravir (100/0.75 mg) vs bictegravir/emtricitabine/tenofovir alafenamide once-daily as initial HIV-1 treatment: week 48 results from a randomized, double-blind phase 3 trial. Presented at 19th European AIDS Conference; Warsaw, Poland; October 2023.

Acknowledgments

We thank the study participants, their families, and all investigators and site personnel for their contributions to the DOR/ISL program.

Funding for this research was provided by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA (MSD). Medical writing and editorial support were provided by Kim Strohmaier, MPH; Dorothy McCoy, PharmD, BCPS, BCIDP; and Carol Zecca, BS, all employees of MSD.

Presented at Conference on Retroviruses and Opportunistic Infections (CROI); February 22-25, 2026; Denver, CO, USA.

Copyright © 2026 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.