Once-Weekly Islatravir Plus Lenacapavir Maintains HIV-1 Suppression Through 96 Weeks: Phase 2 Study

Poster #516

Amy E. Colson[1]*, Gordon E. Crofoot[2], Peter J. Ruane[3], Moti N. Ramgopal[4], Alexandra W. Dretler[5], Ronald G. Nahass[6], Gary I. Sinclair[7], Mezgebe Berhe[8], Shan-Yu Liu[9], Sharline Madera[9], Hadas Dvory-Sobol[9], Cyril Llamoso[10], Elizabeth G. Rhee[10], Devi SenGupta[9], Joseph Eron[11]

[1]Community Resource Initiative, Boston, Massachusetts, USA; [2]The Crofoot Research Center, Houston, Texas, USA; [3]Ruane Clinical Research, Los Angeles, California, USA; [4]Midway Immunology & Research Center, Fort Pierce, Florida, USA; [5]Metro Infectious Disease Consultants, Decatur, Georgia, USA; [6]IDCare, Hillsborough, New Jersey, USA; [7]Prism Health North Texas, Dallas, Texas, USA; [8]North Texas Infectious Diseases Consultants, Dallas, Texas, USA; [9]Gilead Sciences, Inc., Foster City, California, USA; [10]Merck & Co., Inc., Rahway, New Jersey, USA; [11]University of North Carolina, Chapel Hill, North Carolina, USA.

*Presenting author

Conclusions

- All participants receiving once-weekly oral islatravir (ISL) + lenacapavir (LEN) maintained virologic suppression at study Week 96
 - — No participant in either group had HIV-1 RNA ≥50 copies/mL at study discontinuation
 - — No emergent resistance to ISL or LEN was detected
- Once-weekly oral ISL+LEN was well tolerated; no Grade ≥3 adverse events (AEs), serious AEs, or AEs leading to study drug discontinuation were related to ISL+LEN
- CD4+ T-cell counts and lymphocyte counts remained stable in both groups
- These results support ISL+LEN as a potential first, once-weekly complete oral HIV-1 treatment regimen
- Two ongoing Phase 3 studies (ISLEND-1, NCT06630286; ISLEND-2, NCT06630299) are investigating an ISL/LEN fixed-dose combination in virologically suppressed people with HIV-1 (PWH)

Plain Language Summary

- Lenacapavir is a medicine which is approved to treat HIV in people who have already tried many other HIV medicines, and is usually given by injection under the skin after oral (by mouth) doses; islatravir is a new medicine taken orally that is being tested to treat HIV
- Islatravir and lenacapavir can be given orally together once a week, which is different from most HIV treatments that are usually taken once a day. These two medicines are being tested together in a study with people on HIV treatment who already have very low levels of the HIV virus in their blood
 - — At study Week 96, all participants who remained in the study still had very low levels of the HIV virus in their blood (virologically suppressed), and there were no serious or severe side effects related to islatravir and lenacapavir

Background

- Once-weekly oral antiretroviral (ARV) therapy may help to address adherence challenges associated with daily oral treatment, including pill fatigue and privacy concerns[1-3]
- ISL, a nucleoside reverse transcriptase translocation inhibitor,[4] and LEN, a capsid inhibitor,[5] have distinct mechanisms of action, potent ARV activity at low doses, long half-lives that allow for weekly dosing, and a longer forgiveness window than daily oral therapy[6-8]
- A Phase 2 study (NCT05052996) is investigating the combination of ISL and LEN as the first once-weekly, complete oral regimen for HIV-1 treatment
 - — ISL+LEN maintained a high rate of virologic suppression (94.2%) at Week 48 in virologically suppressed PWH[9]
- Week 96 outcomes for participants who received continuous ISL+LEN (Early Switch Group) were reported previously[10]; here, we also present outcomes for the group who switched to ISL+LEN at Week 48 (Late Switch Group)

References: **1.** Claborn KR, et al. *Psychol Health Med*. 2015; 20(3): 255–65. **2.** Mate KKV, et al. *Int J STD AIDS.* 2023; 34(10):677–686. **3.** Scarsi KK, et al. *J Int Assoc Provid AIDS Care*. 2021;20:1–10. **4.** Schürmann D et al. *Lancet HIV*. 2020;7:e164–72. **5.** Squires K, et al. CROI. 2023; Abstract 192. **6.** Zhang H, et al. CROI. 2022; Abstract 433. **7.** Shaik N, et al. AIDS. 2022; Poster PESUB23. **8.** Matthews R, et al. *Clin Trans Sci*. 2021;14:1935–44. **9.** Colson A, et al. IDWeek. 2024; Abstract 577. **10.** Colson A, et al. EACS. 2025; Presentation PS15.5.LB. **11.** Colson A, et al. CROI. 2024; Abstract 208.

Acknowledgments: We extend our thanks to the participants, their families, and all participating investigators. This study was funded by Gilead Sciences, Inc. All authors contributed to and approved the presentation; medical writing and editorial support were provided by Sophie Roberts and Sherriden Beard of Ashfield MedComms (Macclesfield, UK), an Inizio company, and was funded by Gilead Sciences, Inc. Correspondence: Amy Colson, acolson@crihealth.org

Author Disclosures: **Amy E. Colson:** Gilead Sciences, Inc. (consulting fees); ViiV (honoraria); Merck(advisory board participation). **Gordon E. Crofoot:** Gilead Sciences, Inc.(grant/research support); ViiV (grant/research support); Janssen Pharmaceuticals, Inc.(grant/research support); Merck (grant/research support); AbbVie (grant/research support). **Peter J. Ruane:** Gilead Sciences, Inc. (advisor/consultant, honoraria); ViiV(advisor/consultant, honoraria). **Moti N. Ramgopal:** Gilead Sciences, Inc.(advisor/consultant, honoraria, research support); ViiV (advisor/consultant, honoraria); AbbVie (honoraria). **Alexandra W. Dretler:** Gilead Sciences, Inc. (grant/research support); ViiV (grant/research support, advisor/consultant); AbbVie (grant/research support). **Ronald G. Nahass:** Gilead Sciences, Inc. (grants); Merck (grants); ViiV (grants); GSK (grants); Insmed (grants); IDSA (leadership or fiduciary role). **Gary I. Sinclair:** Gilead Sciences, Inc. (advisor/consultant; grant/research support); Janssen (advisor/consultant; grant/research support; honoraria); ViiV (advisor/consultant; grant/research support; honoraria); Theratechnologies (advisor/consultant; grant/research support; honoraria); Merck (advisor/consultant; grant/research support; honoraria); AbbVie (grant/research support). **Mezgebe Berhe:** none. **Shan-Yu Liu, Sharline Madera, Hadas Dvory-Sobol, and Devi SenGupta** are all employees and shareholders of Gilead Sciences, Inc. **Cyril Llamoso,** and **Elizabeth G. Rhee** are employees of Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, New Jersey, USA; and shareholders of Merck & Co., Inc., Rahway, New Jersey, USA. **Joseph J. Eron:** Gilead Sciences, Inc. (grant/contract payments made to his institution; consulting fees); ViiV (grant/contract payments made to his institution; consulting fees); Merck (consulting fees); AbbVie (consulting fees); Invivyd (fees, DSMB/advisory board); Taimed (fees, DSMB/advisory board).

Objective

- Here, we report efficacy, safety, and adherence outcomes through study Week 96 in all participants receiving ISL+LEN

Methods

- In this Phase 2, randomized, open-label, active-controlled study (NCT05052996), virologically suppressed adults with HIV-1 on daily bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF) were randomized 1:1 to switch to weekly oral ISL 2 mg + LEN 300 mg (Early Switch Group) or to continue daily oral B/F/TAF for 48 weeks (**Figure 1**)
 - — At Week 48, participants on B/F/TAF were given the opportunity to switch to weekly ISL+LEN (Late Switch Group) in the extension phase
- Baseline values were defined as last measurement prior to the first dose of ISL+LEN
- Outcomes assessed at Week 96 included HIV-1 RNA ≥50 copies/mL, adherence by pill count, AEs, and changes in CD4+ T-cell and lymphocyte count, weight and BMI
- This Week 96 analysis includes participants in both the Early Switch Group and Late Switch Group

Figure 1. Study Design

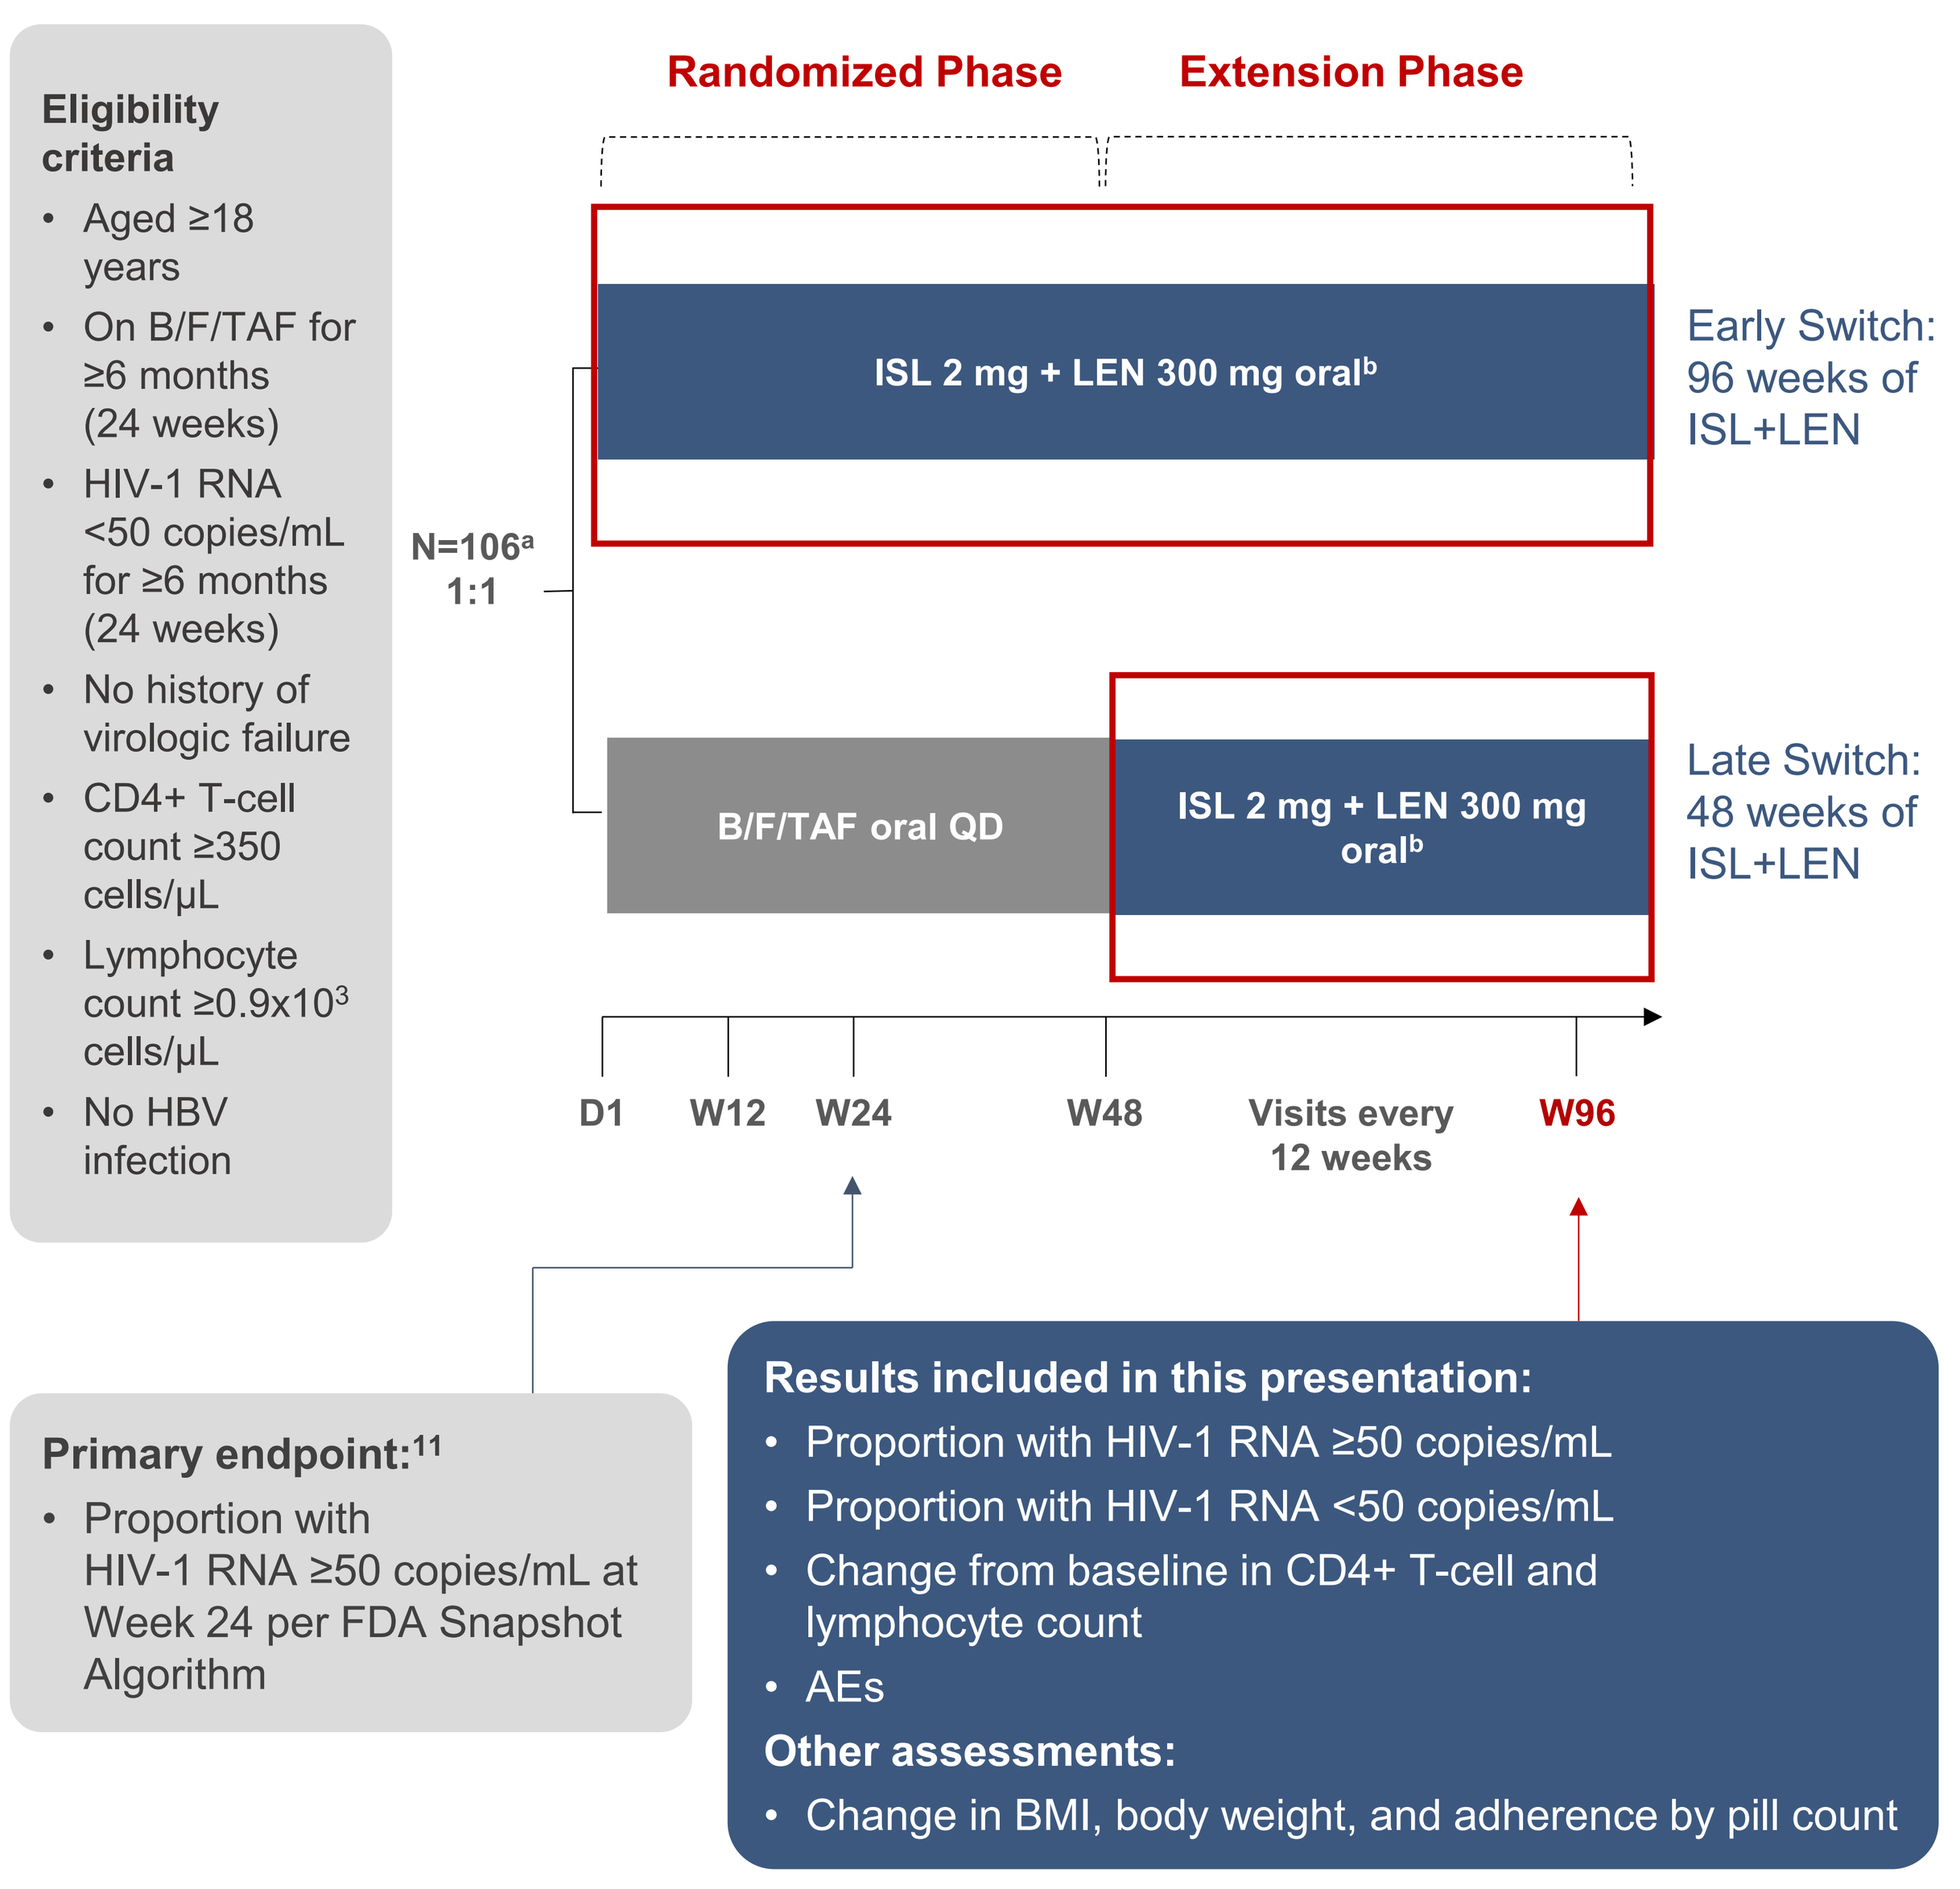
Eligibility criteria
Aged ≥18 years
On B/F/TAF for ≥6 months (24 weeks)
HIV-1 RNA <50 copies/mL for ≥6 months (24 weeks)
No history of virologic failure
CD4+ T-cell count ≥350 cells/μL
Lymphocyte count ≥0.9x10³ cells/μL
No HBV infection
Randomized Phase
Extension Phase
N=106ᵃ
1:1
ISL 2 mg + LEN 300 mg oralᵇ
Early Switch: 96 weeks of ISL+LEN
B/F/TAF oral QD
ISL 2 mg + LEN 300 mg oralᵇ
Late Switch: 48 weeks of ISL+LEN
D1
W12
W24
W48
Visits every 12 weeks
W96
Primary endpoint:¹¹
Proportion with HIV-1 RNA ≥50 copies/mL at Week 24 per FDA Snapshot Algorithm
Results included in this presentation:
Proportion with HIV-1 RNA ≥50 copies/mL
Proportion with HIV-1 RNA <50 copies/mL
Change from baseline in CD4+ T-cell and lymphocyte count
AEs
Other assessments:
Change in BMI, body weight, and adherence by pill count

[a]Randomized, N=106; dosed, n=104. [b]600 mg of LEN was given on Day 1 and Day 2 for pharmacologic loading.
AE, adverse event; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BMI, body mass index; D, Day; FDA, Food and Drug Administration; HBV, hepatitis B virus; ISL, islatravir; LEN, lenacapavir; QD, daily; QW, weekly; W, Week.

Results

Participants

- Ninety-seven participants received ISL+LEN
 - — Early Switch Group: 52 participants were randomized to ISL+LEN at baseline
 - — Late Switch Group: of the 52 participants who received B/F/TAF in the randomized phase, 45 switched to ISL+LEN at Week 48 of the study
- Baseline characteristics were similar between the two groups: for the combined population, median age was 40 years, 18.6% were assigned female at birth, and 50.5% were White (**Table 1**)

Table 1. Baseline Characteristics

	Early Switch Group 96 Weeks of ISL+LEN (n=52)	Late Switch Group 48 Weeks of ISL+LEN (n=45)	Total (N=97)
Median (range) age, years	40 (28–66)	40 (26–75)	40 (26–75)
Assigned female at birth, n (%)	10 (19.2)	8 (17.8)	18 (18.6)
Gender identity, n (%)			
Transgender female	1 (1.9)	0	1 (1.0)
Non-binary/third gender	0	1 (2.2)	1 (1.0)
Race, n (%)			
White	25 (48.1)	24 (53.3)	49 (50.5)
Black	21 (40.4)	14 (31.1)	35 (36.1)
Asian	2 (3.8)	1 (2.2)	3 (3.1)
American Indian or Alaska Native	1 (1.9)	1 (2.2)	2 (2.1)
Other	3 (5.8)	5 (11.1)	8 (8.2)
Hispanic or Latine ethnicity, n (%)	13 (25.0)	14 (31.1)	27 (27.8)
Mean (95% CI) CD4+ T-cell count, cells/μL	755 (692; 817)	793 (728; 857)	772 (728; 816)
Mean (95% CI) lymphocyte count, x10³ cells/uL	1.94 (1.8; 2.1)	2.01 (1.8; 2.2)	1.97 (1.9; 2.1)
Median (IQR) body weight, kg	79.3 (70.4; 87.4)	83.2 (77.0; 91.6)	80.5 (73.7; 89.0)
Median (IQR) BMI, kg/m²	26.9 (23.8; 30.0)	27.4 (24.8; 29.8)	27.1 (24.4; 29.9)

BMI, body mass index; CI, confidence interval; IQR, interquartile range; ISL, islatravir; LEN, lenacapavir

Virologic Outcomes After 96 Weeks on ISL+LEN for the Early Switch Group

- At study Week 96, virologic data were available for 46/52 participants in the Early Switch Group
 - — Three participants discontinued prior to Week 48[a]
 - — Two participants completed 48 weeks of the randomized phase and did not enter the extension phase[b]
 - — One participant discontinued during the extension phase[c]
- No participants with virologic data available had HIV-1 RNA ≥50 copies/mL at Week 96 or at discontinuation
- At Week 96, all participants continuing the study maintained virologic suppression (HIV-1 RNA <50 copies/mL) (**Figure 2**)
- No emergent resistance to ISL or LEN was detected in the Early Switch Group through study Week 96
- Mean adherence to ISL+LEN was 99.3% in the Early Switch Group[d]

Virologic Outcomes After 48 Weeks on ISL+LEN for the Late Switch Group

- At study Week 48, 45/52 participants in the Late Switch Group switched from B/F/TAF to ISL+LEN
 - — Three participants discontinued prior to Week 48[e]
 - — Four participants did not enter the extension phase at Week 48[f]
- After 48 weeks of treatment with ISL+LEN in the extension phase (study Week 96), virologic data were available for 44/45 participants in the Late Switch Group; one participant discontinued at study Week 60 due to personal reasons
- No participants in the Late Switch Group with virologic data available had HIV-1 RNA ≥50 copies/mL after 48 weeks on ISL+LEN or at study discontinuation; all participants who remained in the study after 48 weeks on ISL+LEN maintained viral suppression (HIV-1 RNA <50 copies/mL) (**Figure 2**)
- No emergent resistance to ISL or LEN was detected in the Late Switch Group through study Week 96 (48 weeks of ISL+LEN)
- Mean adherence to ISL+LEN was 98.6% in the Late Switch Group[d]

[a]Two participants discontinued due to AEs unrelated to study drug (large intestine perforation and renal colic in same participant [n=1]; acute hepatitis B [n=1]) and one discontinued due to personal reasons. [b]Personal reasons (n=2). [c]Personal reasons (n=1). [d]Adherence calculated by pill count. [e]Personal reasons (n=1); lost to follow-up (n=2). [f]Withdrew consent (n=2); lost to follow-up (n=2).

Figure 2. Study Week 96 Virologic Outcomes for Early Switch Group and Late Switch Group

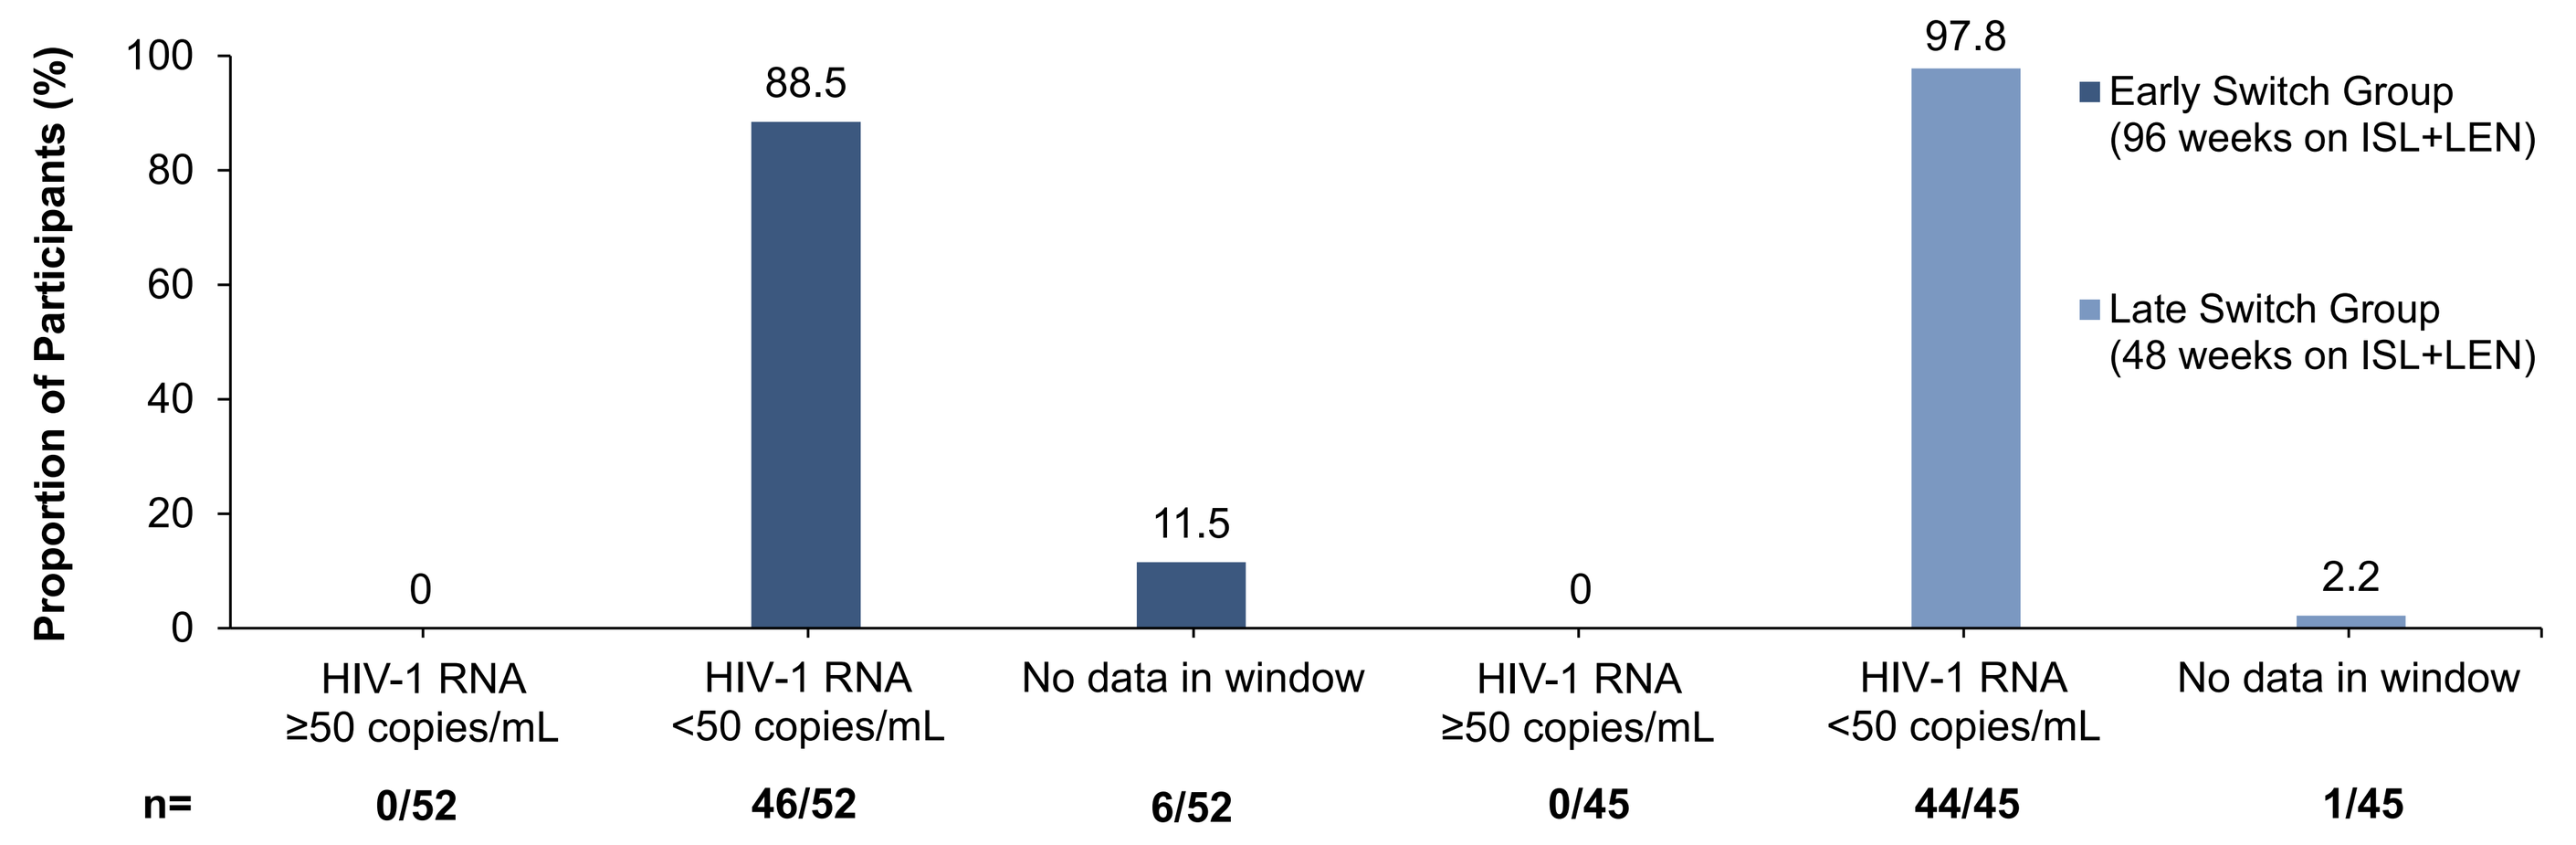
Proportion of Participants (%)
100
80
60
40
20
0
Early Switch Group (96 weeks on ISL+LEN)
Late Switch Group (48 weeks on ISL+LEN)
0
88.5
11.5
0
97.8
2.2
HIV-1 RNA ≥50 copies/mL
HIV-1 RNA <50 copies/mL
No data in window
HIV-1 RNA ≥50 copies/mL
HIV-1 RNA <50 copies/mL
No data in window
n=
0/52
46/52
6/52
0/45
44/45
1/45

ISL, islatravir; LEN, lenacapavir

Table 2. Adverse Events

Participants, n (%)	Early Switch Group 96 Weeks of ISL+LEN (n=52)	Late Switch Group 48 Weeks of ISL+LEN (n=45)	Total (N=97)
Any AE	46 (88.5)[a]	35 (77.8)[b]	81 (83.5)
Treatment-related AEs	10 (19.2)[c]	2 (4.4)[d]	12 (12.4)
Grade 1 or 2	10 (19.2)[c]	2 (4.4)[d]	12 (12.4)
≥2 participants in any group			
Dry mouth	2 (3.8)	0	2 (2.1)
Nausea	2 (3.8)	0	2 (2.1)
Fatigue	1 (1.9)	1 (2.2)	2 (2.1)
Grade 3 or higher	0	0	0
Serious AE	3 (5.8)[e]	0	3 (3.1)
Treatment related	0	0	0
AE leading to study drug discontinuation	2 (3.8)[f]	0	2 (2.1)
Treatment related	0	0	0

[a]AEs occurring in >10%: diarrhea (n=9), upper respiratory tract infection (n=8), arthralgia (n=6), COVID 19 (n=6); [b]AEs occurring in >10%: arthralgia (n=6), upper respiratory tract infection (n=5); [c]Treatment-related AEs: dry mouth (n=2), nausea (n=2), fatigue (n=1), abdominal distension (n=1), upper abdominal pain (n=1), brain fog (n=1), diarrhoea (n=1), headache (n=1), nightmare (n=1), taste disorder (n=1). [d]Treatment-related AEs: fatigue (n=1) and sleep disorder (n=1). [e]Serious AEs: neurologic anaesthesia complication (n=1), renal colic/colon perforation in same participant (n=1); pneumonia (n=1); [f]AEs leading to study discontinuation: acute hepatitis B virus infection (n=1), renal colic/colon perforation in same participant (n=1).
AE, adverse event; ISL, islatravir; LEN, lenacapavir.

Safety

- Median (interquartile range) duration of exposure to ISL+LEN was 103.9 (98.1–112.0) weeks in the Early Switch Group and 57.9 (53.7–63.9) weeks in the Late Switch Group
- No Grade ≥3 AEs, serious AEs, or AEs leading to study drug discontinuation were related to study drug (**Table 2**)

CD4 and Lymphocytes

- Mean changes in CD4+ T-cell (**Figure 3A**) and lymphocyte counts (**Figure 3B**) were minimal in both groups
- No participants discontinued due to changes in CD4+ T-cell counts or lymphocyte count

Figure 3. (A) CD4+ T-cell and (B) Lymphocyte Changes

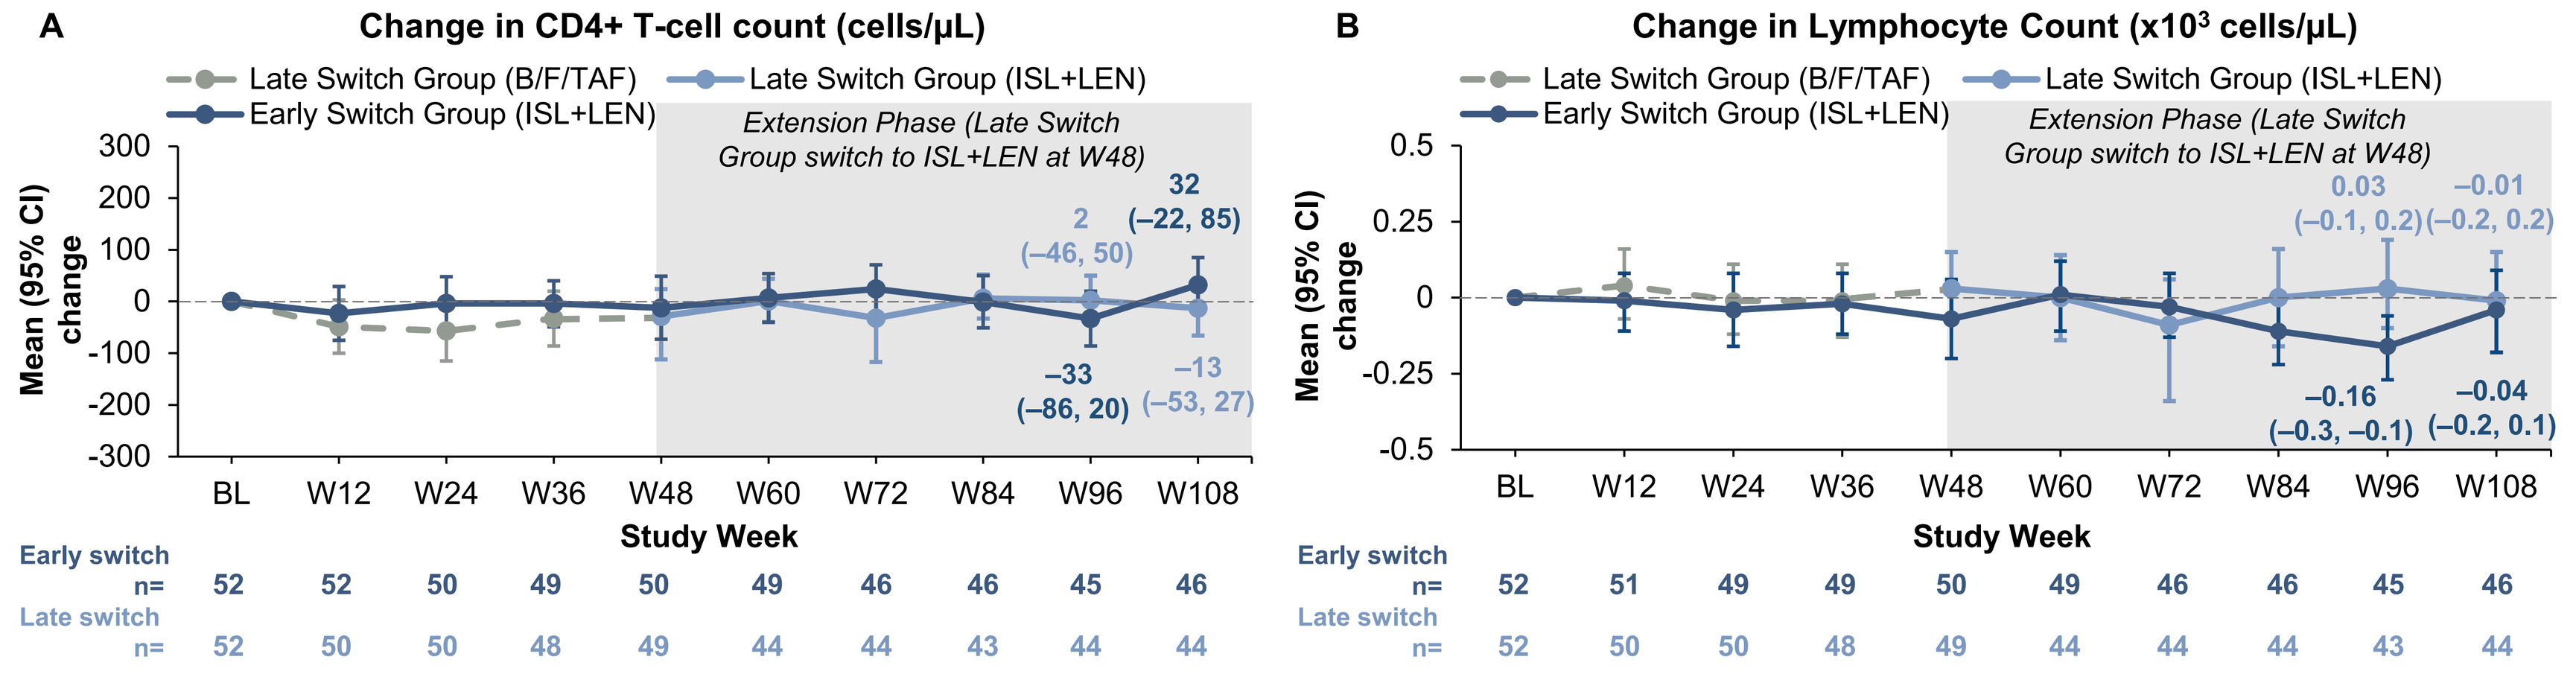
A
Change in CD4+ T-cell count (cells/μL)
Late Switch Group (B/F/TAF)
Late Switch Group (ISL+LEN)
Early Switch Group (ISL+LEN)
Extension Phase (Late Switch Group switch to ISL+LEN at W48)
Mean (95% CI) change
32 (−22, 85)
2 (−46, 50)
−33 (−86, 20)
−13 (−53, 27)
BL W12 W24 W36 W48 W60 W72 W84 W96 W108
Study Week
Early switch n= 52 52 50 49 50 49 46 46 45 46
Late switch n= 52 50 50 48 49 44 44 43 44 44
B
Change in Lymphocyte Count (x10³ cells/μL)
Late Switch Group (B/F/TAF)
Late Switch Group (ISL+LEN)
Early Switch Group (ISL+LEN)
Extension Phase (Late Switch Group switch to ISL+LEN at W48)
Mean (95% CI) change
0.03 (−0.1, 0.2)
−0.01 (−0.2, 0.2)
−0.16 (−0.3, −0.1)
−0.04 (−0.2, 0.1)
BL W12 W24 W36 W48 W60 W72 W84 W96 W108
Study Week
Early switch n= 52 51 49 49 50 49 46 46 45 46
Late switch n= 52 50 50 48 49 44 44 44 43 44

Figure also presents post-hoc analysis beyond the W96 analysis at W108.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BL, baseline; CI, confidence interval; ISL, islatravir; LEN, lenacapavir; W, week.

Weight and BMI

- Mean changes in body weight (**Figure 4A**) and BMI (**Figure 4B**) were minimal in both groups
- No participants discontinued due to changes in body weight or BMI

Figure 4. (A) Weight and (B) BMI Changes

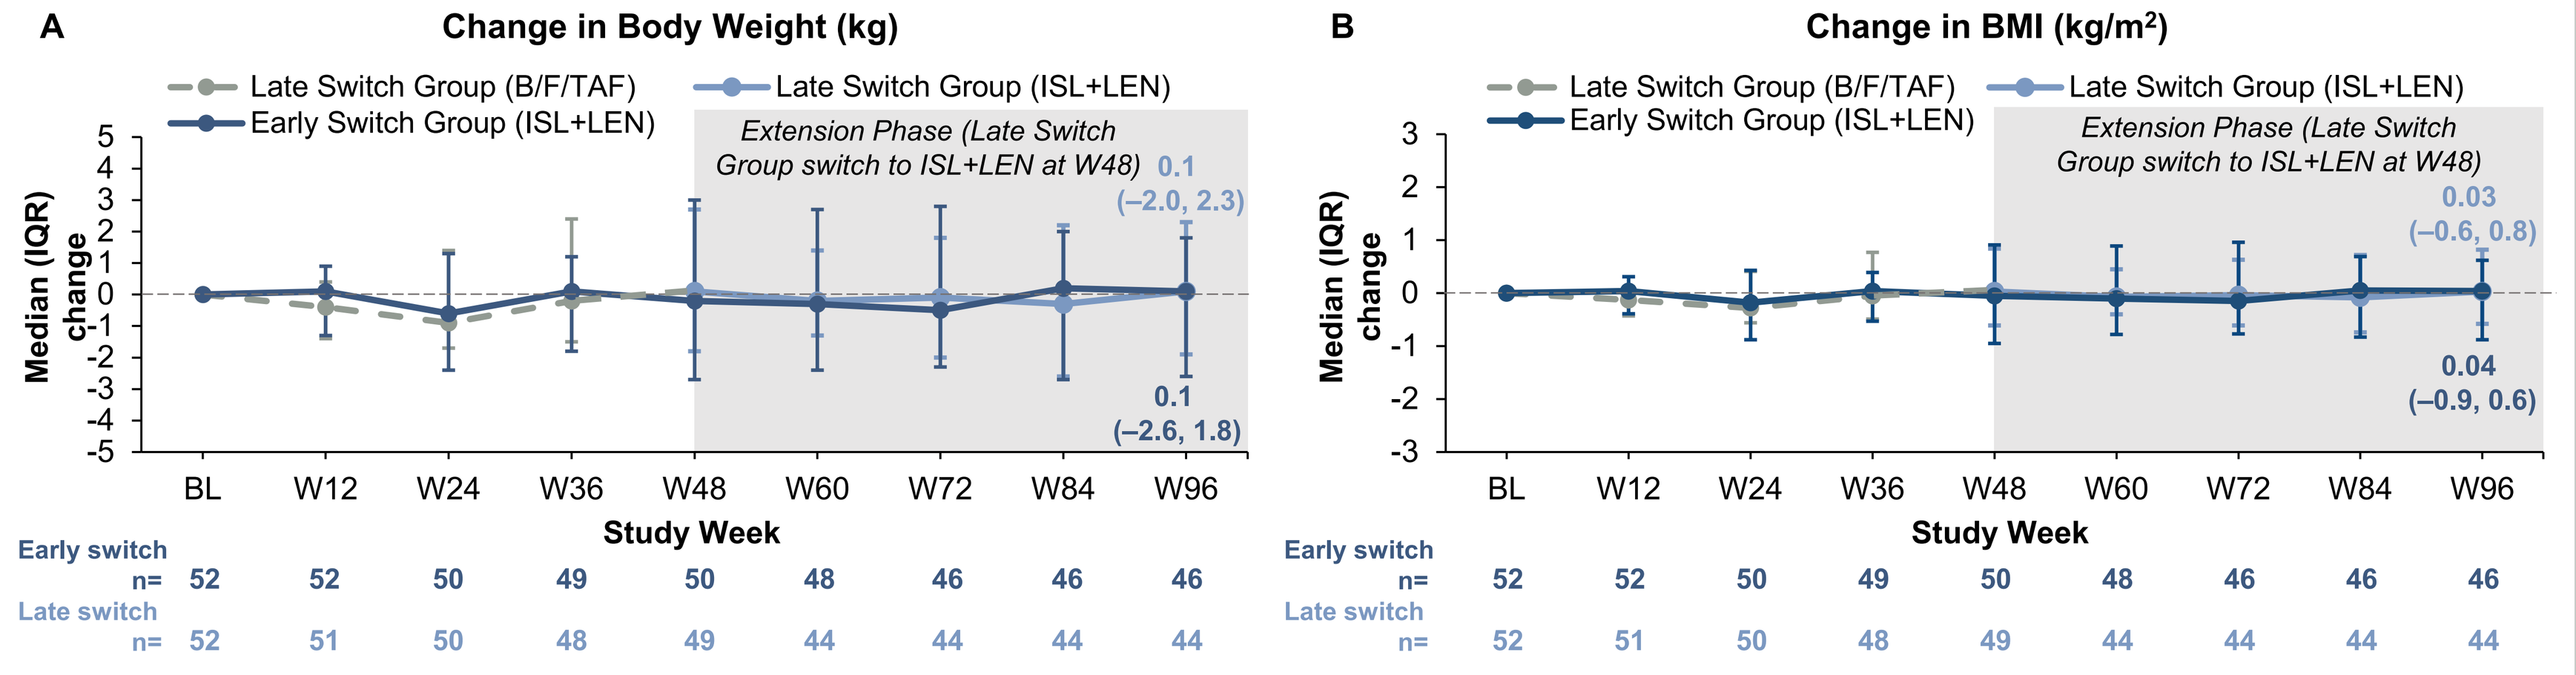
A
Change in Body Weight (kg)
Late Switch Group (B/F/TAF)
Late Switch Group (ISL+LEN)
Early Switch Group (ISL+LEN)
Extension Phase (Late Switch Group switch to ISL+LEN at W48)
Median (IQR) change
0.1 (−2.0, 2.3)
0.1 (−2.6, 1.8)
BL W12 W24 W36 W48 W60 W72 W84 W96
Study Week
Early switch n= 52 52 50 49 50 48 46 46 46
Late switch n= 52 51 50 48 49 44 44 44 44
B
Change in BMI (kg/m²)
Late Switch Group (B/F/TAF)
Late Switch Group (ISL+LEN)
Early Switch Group (ISL+LEN)
Extension Phase (Late Switch Group switch to ISL+LEN at W48)
Median (IQR) change
0.03 (−0.6, 0.8)
0.04 (−0.9, 0.6)
BL W12 W24 W36 W48 W60 W72 W84 W96
Study Week
Early switch n= 52 52 50 49 50 48 46 46 46
Late switch n= 52 51 50 48 49 44 44 44 44

B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BL, baseline; BMI, body mass index; IQR, interquartile range; ISL, islatravir; LEN, lenacapavir; W, week

CROI 2026, February 22–25 2026, Denver, Colorado, USA