515

Switch to DOR/ISL (100/0.25 mg) QD From Oral ART: Week 96 Update From an Open-Label Phase 3 Study

Chloe Orkin, MD[1]; Rosie Mngqibisa, MD[2]; Juan Diego Velez, MD[3]; Princy Kumar, MD[4]; Dominique Braun, MD[5]; Andrew Carr, DSc[6]; Mark Bloch, MD[7]; Sharon Walmsley, MD[8]; Pablo Tebas, MD[9]; Alexandra Calmy, MD[10]; Monica Fuszard, MS[11]; Tracy Diamond, PhD[11]; Beth Jackson, BS[11]; Michelle C. Fox, MD[11]; Jason Kim, MD, MSCE[11]

[1]Queen Mary University of London, London, UK; [2]Enhancing Care Foundation, Wentworth Hospital, Durban, South Africa; [3]Fundación Valle del Lili, Cali, Colombia; [4]Division of Infectious Diseases, Georgetown University Medical Center, Washington DC, USA; [5]University Hospital, Zurich, Switzerland; [6]University of New South Wales, Sydney, Australia; [7]Holdsworth House, Sydney, Australia; [8]University of Toronto Department of Medicine, Ontario, Canada; [9]Penn Center for AIDS Research, University of Pennsylvania, Philadelphia, PA, USA; [10]HIV/AIDS Unit, Division of Infectious Diseases, Geneva University Hospitals, Geneva, Switzerland; [11]Merck & Co., Inc., Rahway, NJ, USA

Background

- Given the longer life expectancy and high prevalence of age-related comorbidities in people living with HIV-1, there is an unmet need for effective and tolerable two-drug regimens that minimize the risk of drug-disease and drug-drug interactions and limit lifetime exposure to antiretroviral medication[1]
- The combination of doravirine (DOR) and islatravir (ISL) has the potential to be a simplified once-daily (QD) treatment option for HIV-1
 - DOR is a non-nucleoside reverse transcriptase inhibitor (NNRTI) with activity against common NNRTI-resistance-associated variants and a favorable safety profile[2]
 - ISL is an investigational nucleoside reverse transcriptase translocation inhibitor (NRTTI) with multiple mechanisms of action and potent in vitro antiviral activity (including against common resistance-associated variants) and a high barrier to the emergence of resistance[3,4]
 - DOR and ISL have complementary resistance profiles; variants encoding the most common DOR resistance-associated mutations observed in DOR/3TC/TDF Phase 3 studies are hypersusceptible to ISL[3]
- MK-8591A-051 is an open-label, randomized clinical study of switching to DOR/ISL (100/0.25 mg) QD from any 2- or 3-drug oral antiretroviral therapy (ART) in virologically suppressed adults with HIV-1[5]
 - In the primary analysis at Week 48, the efficacy of DOR/ISL was noninferior to continued baseline ART (bART): HIV-1 RNA ≥50 copies/mL in 1.4% vs 4.9% of participants (difference -3.6, 95% CI -7.8 to -0.8)
 - DOR/ISL was generally well tolerated through Week 48 with a safety profile similar to continued bART
- Here we present the Week 96 efficacy and safety results from this ongoing study

Methods

Study design

- MK-8591A-051 (NCT05631093) is a randomized, active-controlled, open-label, Phase 3 non-inferiority study in virologically suppressed adults with HIV-1 (**Figure 1**)
 - Participants were assigned (2:1) to switch to DOR/ISL (100/0.25 mg) QD (Group 1) or to continue their bART regimen (Group 2) for up to 48 weeks; randomization was stratified by bART regimen (PI-, InSTI-, or NNRTI-based)
 - After Week 48, both groups receive DOR/ISL (100/0.25 mg) QD through Week 144
- At Week 96, efficacy was assessed by the proportion of participants with HIV-1 RNA ≥50 copies/mL, <50 copies/mL, and <200 copies/mL (FDA snapshot approach) in the modified Full Analysis Set (all participants who received ≥1 dose of DOR/ISL); no formal hypothesis testing was performed
- Resistance analysis was performed for participants with confirmed HIV-1 RNA ≥200 copies/mL and for those with HIV-1 RNA ≥200 copies/mL at discontinuation

Figure 1. MK-8591A-051: a Phase 3, randomized, open-label switch study

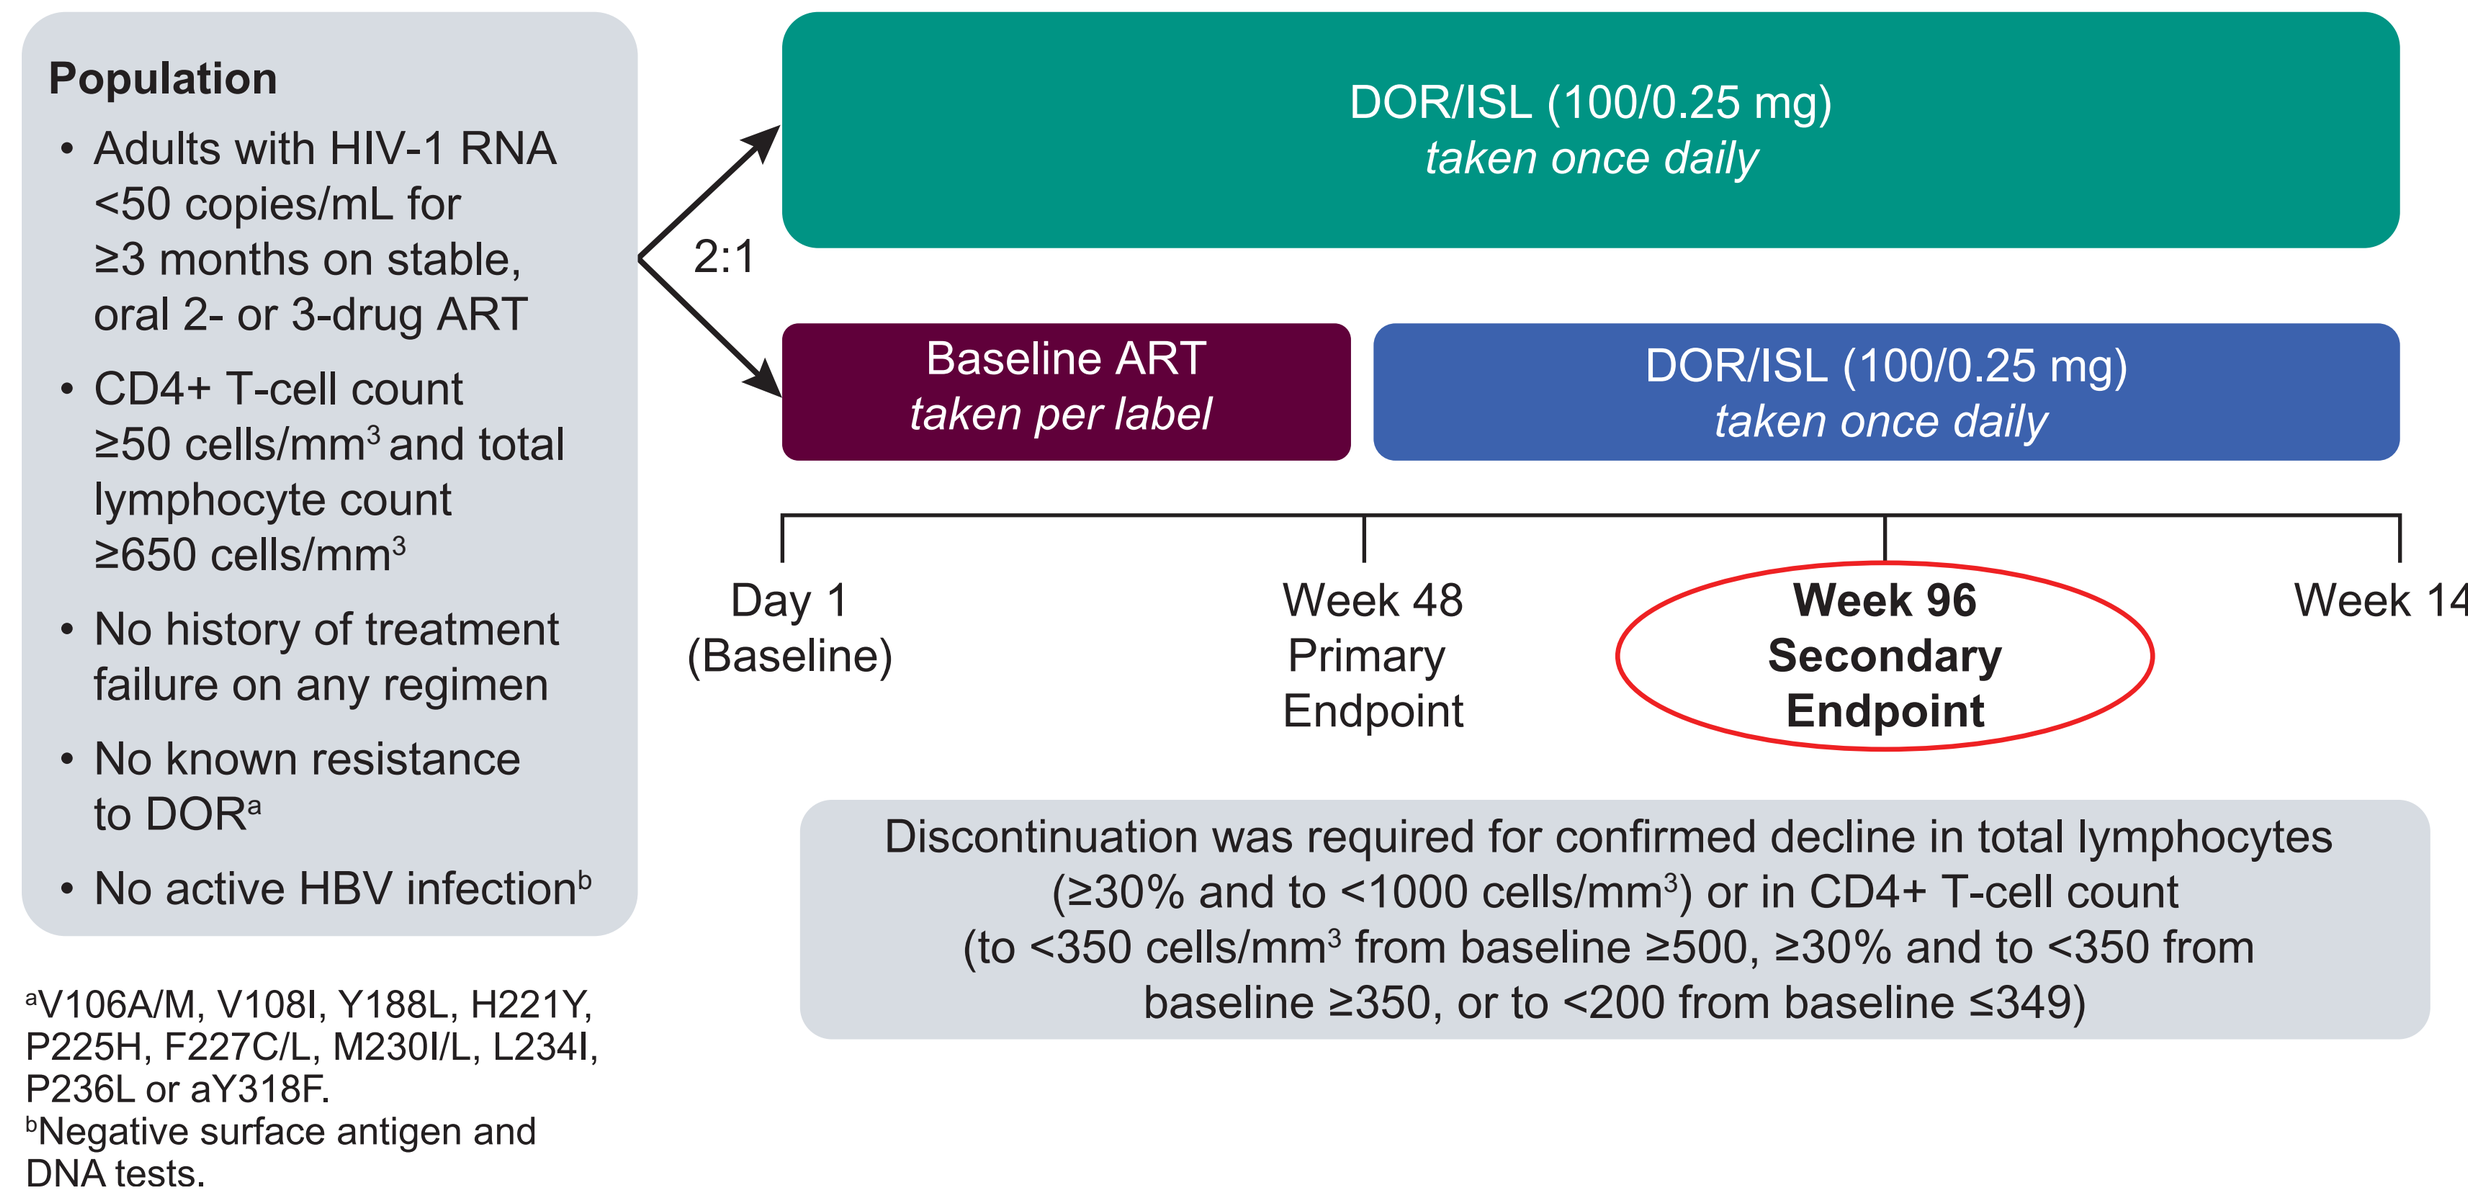
Population
• Adults with HIV-1 RNA <50 copies/mL for ≥3 months on stable, oral 2- or 3-drug ART
• CD4+ T-cell count ≥50 cells/mm³ and total lymphocyte count ≥650 cells/mm³
• No history of treatment failure on any regimen
• No known resistance to DOR[a]
• No active HBV infection[b]
2:1
DOR/ISL (100/0.25 mg) taken once daily
Baseline ART taken per label
DOR/ISL (100/0.25 mg) taken once daily
Day 1 (Baseline)
Week 48 Primary Endpoint
Week 96 Secondary Endpoint
Week 144
Discontinuation was required for confirmed decline in total lymphocytes (≥30% and to <1000 cells/mm³) or in CD4+ T-cell count (to <350 cells/mm³ from baseline ≥500, ≥30% and to <350 from baseline ≥350, or to <200 from baseline ≤349)

[a]V106A/M, V108I, Y188L, H221Y, P225H, F227C/L, M230I/L, L234I, P236L or aY318F.
[b]Negative surface antigen and DNA tests.

References

1. Gibas KM, et al. *Lancet HIV.* 2022;9(12):e868-e83.
2. Stockdale AJ, Khoo S. *Curr Opin HIV AIDS.* 2022;7(1):4-14.
3. Diamond TL, et al. *Antimicrob Agents Chemother.* 2022;66(6):e0013322.
4. Lai MT, et al. *Antimicrob Agents Chemother.* 2022;66(5):e0222321.
5. Orkin C, et al. *Lancet.* 2026;407:599-610.

Acknowledgments

We thank all the participants and their families, along with the investigators and their staff, for their many contributions to this study. We are also grateful for the critical review of the data by scientific advisors.

Funding for this study was provided by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA. Medical writing and editorial support were provided by Kim Strohmaier, MPH, and Carol Zecca, BS, employees of MSD.

Presented at Conference on Retroviruses and Opportunistic Infections (CROI); February 22-25, 2026; Denver, CO, USA.

Results

- On Day 1, 366 participants switched to DOR/ISL (Group 1) and 185 continued their bART regimen (Group 2)
- The study population was diverse (**Table 1**): 39.7% female at birth, 45.4% Black, and 14.5% Hispanic or Latino/a
- bART was InSTI-based in 64.2% of participants, NNRTI-based in 30.3%, and PI-based in 5.4%
- At Week 48, 177 participants (95.7%) in Group 2 switched from bART to DOR/ISL, for a total of 543 receiving DOR/ISL

Table 1. Baseline characteristics were balanced across treatment groups

Baseline Characteristic	Total Population N=551	DOR/ISL (100/0.25 mg) N=366	Baseline ART N=185
Age (years), median (IQR)	51.0 (41.0 – 59.0)	51.0 (41.0 – 59.0)	51.0 (42.0 – 59.0)
Female sex at birth, n (%)	219 (39.7)	152 (41.5)	67 (36.2)
Race, n (%): Black or African American	250 (45.4)	166 (45.4)	84 (45.4)
White	215 (39.0)	141 (38.5)	74 (40.0)
Asian	28 (5.1)	18 (4.9)	10 (5.4)
Ethnicity, n (%): Hispanic or Latine	80 (14.5)	54 (14.8)	26 (14.1)
Years since HIV-1 diagnosis, median (IQR)	13.3 (7.2 – 20.4)	13.3 (7.7 – 21.2)	13.3 (6.8 – 19.1)
Years on current ART, median (IQR)	3.8 (2.0 – 6.3)	3.7 (2.0 – 6.3)	3.8 (2.1 – 6.3)
Baseline ART, n (%): PI-based	30 (5.4)	22 (6.0)	8 (4.3)
InSTI-based	354 (64.2)	233 (63.7)	121 (65.4)
NNRTI-based	167 (30.3)	111 (30.3)	56 (30.3)
CD4+ T-cell count (cells/mm³), median (IQR)	706 (545 – 921)	697 (525 – 922)	725 (566 – 918)
CD4+ T-cell count ≥350 cells/mm³, n (%)	505 (91.7)	332 (90.7)	173 (93.5)

Efficacy (Figure 2)

- In Group 1 DOR/ISL, virologic suppression (HIV-1 RNA <50 copies/mL) was maintained in 92.6% of participants at Week 96
- 7 participants (1.9%) in Group 1 had HIV-1 RNA ≥50 copies/mL at Week 96:
 - As previously reported, 5 had HIV-1 RNA ≥50 copies/mL at Week 48 and discontinued; no treatment-emergent resistance-associated substitutions to either DOR or ISL were detected
 - 2 had HIV-1 RNA ≥50 copies/mL after Week 48 (neither met criteria for resistance testing)
 - 1 withdrew at Week 84 with last on-treatment HIV-1 RNA ≥50 copies/mL; DOR and ISL levels were in expected ranges
 - 1 had HIV-1 RNA ≥50 and <200 copies/mL at Week 96 and remains in the study; DOR and ISL levels were intermittently below expected ranges
- In Group 2 DOR/ISL, virologic suppression was maintained in 96.6% of participants at Week 96 (48 weeks on DOR/ISL), similar to Group 1 at Week 48 (95.6%)
- 2 participants (1.1%) in Group 2 DOR/ISL had HIV-1 RNA ≥50 copies/mL at Week 96:
 - 1 had HIV-1 RNA ≥200 copies/mL at Week 108 and discontinued due to lack of efficacy; DOR and ISL levels were in expected ranges at time of viremia (no resistance results available due to assay failure)
 - 1 resuppressed on DOR/ISL with HIV-1 RNA <50 copies/mL at Week 108 and remains in the study

Figure 2. Virologic outcomes at Week 48 and Week 96 (FDA snapshot approach)

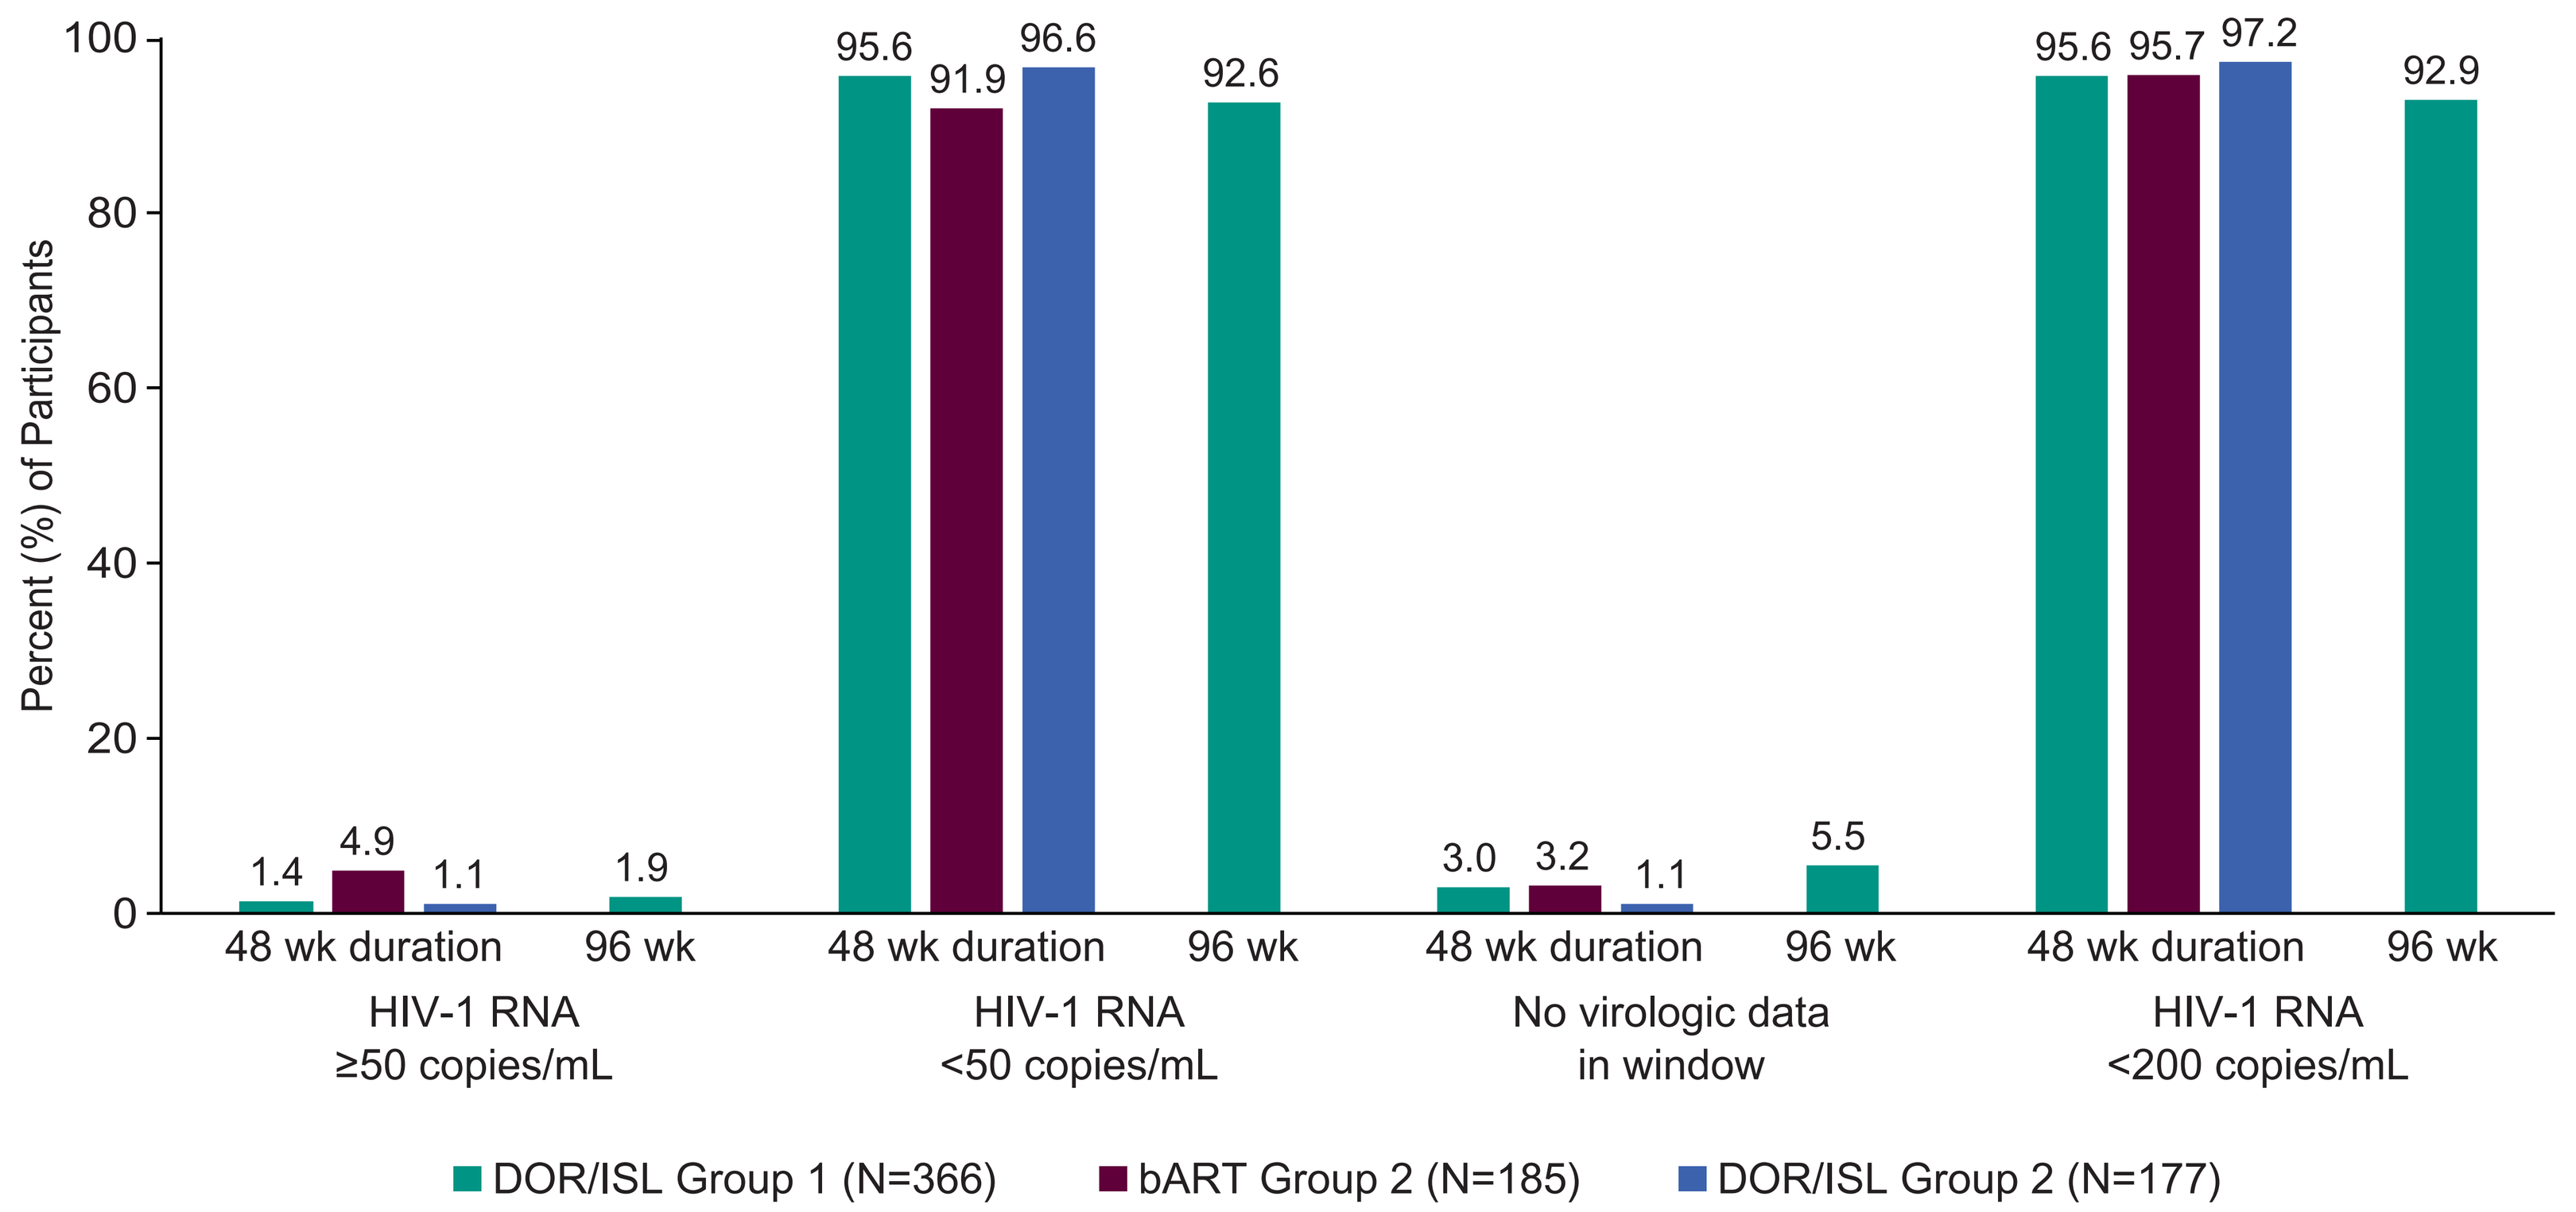
Percent (%) of Participants
100
80
60
40
20
0
1.4
4.9
1.1
1.9
95.6
91.9
96.6
92.6
3.0
3.2
1.1
5.5
95.6
95.7
97.2
92.9
48 wk duration
96 wk
HIV-1 RNA ≥50 copies/mL
HIV-1 RNA <50 copies/mL
No virologic data in window
HIV-1 RNA <200 copies/mL
DOR/ISL Group 1 (N=366)
bART Group 2 (N=185)
DOR/ISL Group 2 (N=177)

Safety

- During the first 48 weeks of DOR/ISL, mean percent changes in total lymphocyte count and CD4+ T-cell count were comparable between treatment groups (**Figure 3**)
- At Week 96, Group 1 had a mean increase of 3.6% in total lymphocyte count and 4.5% in CD4+ T-cell count
- No participants met discontinuation criteria for decrease in total lymphocyte count or CD4+ T-cell count

Figure 3. Mean percent change in total lymphocyte and CD4+ T-cell counts from start of DOR/ISL

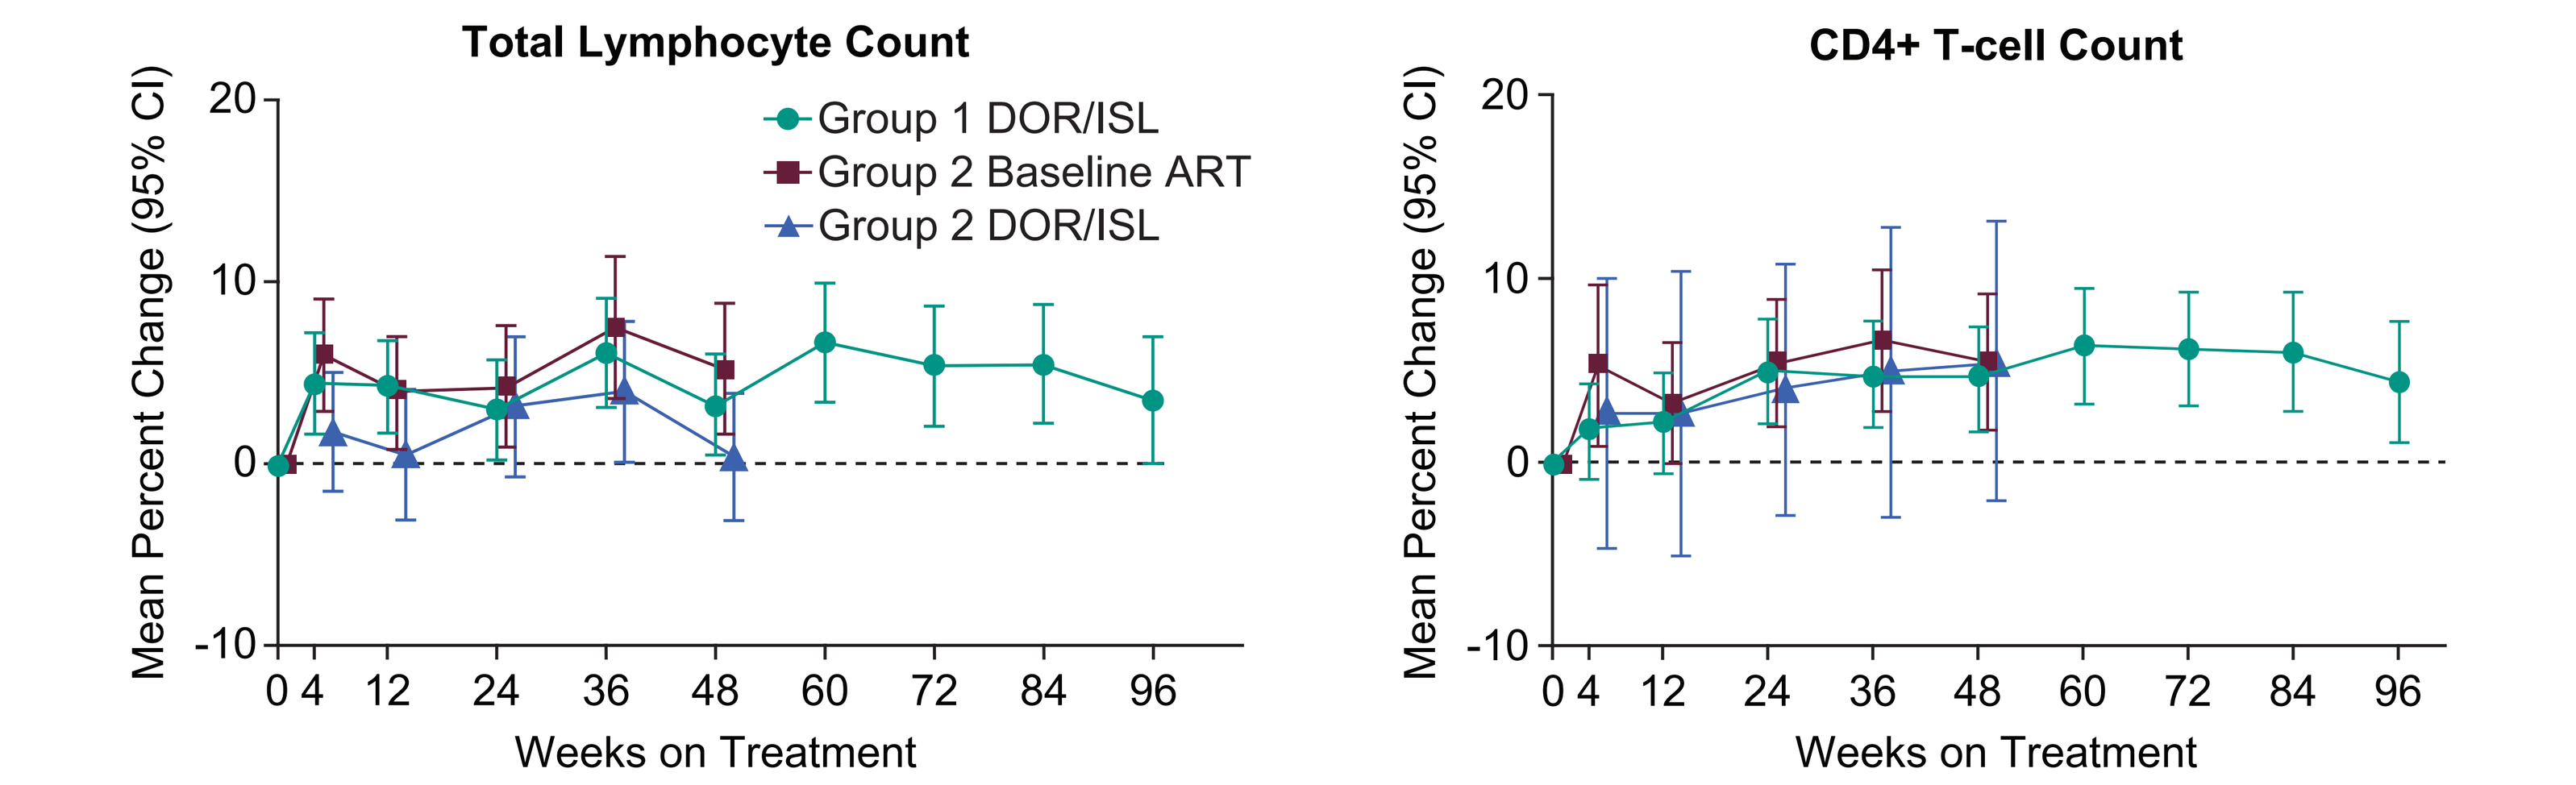
Total Lymphocyte Count
CD4+ T-cell Count
Group 1 DOR/ISL
Group 2 Baseline ART
Group 2 DOR/ISL
Mean Percent Change (95% CI)
20
10
0
-10
0 4 12 24 36 48 60 72 84 96
Weeks on Treatment

- AE rates during the first 48 weeks on DOR/ISL were similar in Group 1 (Week 0-48) and Group 2 (Week 48-96) (**Table 2**)
- In Group 1, fewer participants reported drug-related AEs during Week 48-96 (1.7%) compared to Week 0-48 (12.3%)
- The most common drug-related AEs were diarrhea (3.3%) and weight increase (2.2%) in Group 1 DOR/ISL and weight increase (1.7%) and abnormal dreams (1.7%) in Group 2 DOR/ISL
- One drug-related serious AE occurred on DOR/ISL (Group 1, Week 54): toxicity to various agents (previously reported multiple-drug overdose)
- One death occurred on DOR/ISL (Group 1, Week 62): myocardial infarction, considered not drug-related

Table 2. Summary of adverse events (AE) through Week 96

n (%) of participants	Group 1: DOR/ISL Week 0-96 N=366	Group 1: DOR/ISL Week 0-48 N=366	Group 1: DOR/ISL Week 48-96 N=353	Group 2: bART Week 0-48 N=185	Group 2: DOR/ISL Week 48-96 N=177
One or more AEs	328 (89.6)	296 (80.9)	270 (76.5)	155 (83.8)	141 (79.7)
Drug-related[a] AEs	50 (13.7)	45 (12.3)	6 (1.7)	9 (4.9)	20 (11.3)
Grade 3-4 AEs	54 (14.8)	40 (10.9)	24 (6.8)	18 (9.7)	11 (6.2)
Serious AEs	34 (9.3)	24 (6.6)	16 (4.5)	9 (4.9)	9 (5.1)
Drug-related serious AEs	1 (0.3)	0 (0.0)	1 (0.3)	1 (0.5)	0 (0.0)
Deaths	1 (0.3)	0 (0.0)	1 (0.3)	1 (0.5)	0 (0.0)
Discontinued due to AE	4 (1.1)	2 (0.5)	2 (0.6)	4 (2.2)	1 (0.6)
Due to drug-related AE	2 (0.5)	1 (0.3)	1 (0.3)	2 (1.1)	0 (0.0)
Due to serious drug-related AE	1 (0.3)	0 (0.0)	1 (0.3)	1 (0.5)	0 (0.0)

[a]Relationship to study drug was determined by the investigator.

- No cases of clinical HBV reactivation occurred; 3 participants (who were HBcAb+ at screening) had transient low-level HBV viremia (DNA <450 IU/mL) that resolved on DOR/ISL (**Table 3**); all were asymptomatic with no surface antigenemia or elevated transaminases
- One participant in Group 2 DOR/ISL had a new HBV infection (HBV DNA ~20 million IU/mL) with DAIDS Grade 1-2 elevations in ALT and AST (and normal bilirubin); DOR/ISL was stopped and BIC/FTC/TAF started

Table 3. Hepatitis B Virus (HBV) events through Week 96 by serology at screening

HBV Serology	Group 1: DOR/ISL Week 0-48 N=366		Group 1: DOR/ISL Week 48-96 N=353		Group 2: bART Week 0-48 N=185		Group 2: DOR/ISL Week 48-96 N=177	
HBcAb positive, HBsAb positive at screening, n (%)	95	(25.9)	92	(26.1)	47	(25.4)	45	(25.4)
HBV DNA detected, m/n (%)[a]	1/95	(1.1)	0	(0.0)	0	(0.0)	0	(0.0)
HBcAb positive, HBsAb negative at screening, n (%)	11	(3.0)	11	(3.1)	7	(3.8)	6	(3.4)
HBV DNA detected, m/n (%)[a]	0	(0.0)	0	(0.0)	0	(0.0)	2/6	(33.3)
HBcAb negative at screening, n (%)	260	(71.0)	260	(71.0)	131	(70.8)	131	(70.8)
New HBV infection, m/n (%)[b]	0	(0.0)	0	(0.0)	0	(0.0)	1/131	(0.8)

[a]Confirmed HBV DNA >10 IU/mL with positive baseline HBcAb. [b]Confirmed HBV DNA >10 IU/mL with negative baseline HBcAb.

- No clinically relevant changes were observed in LDL-cholesterol, non-HDL-cholesterol, or insulin resistance (**Table 4**)
- Creatinine-based eGFR showed statistically significant increase in the first 48 weeks after switch to DOR/ISL, which was not observed in cystatin-C-based eGFR

Table 4. Mean change in fasting lipids, insulin resistance, and renal function through Week 96

Laboratory parameter	Group 1: DOR/ISL Week 0-48 N=366 n	Mean change*	Group 1: DOR/ISL Week 48-96 N=353 n	Mean change*	Group 2: bART Week 0-48 N=185 n	Mean change*	Group 2: DOR/ISL Week 48-96 N=177 n	Mean change*
Fasting LDL-C, mg/dL	247	2.8 (0.2, 5.5)	226	-1.0 (-3.5, 1.5)	129	-0.2 (-3.8, 3.3)	121	2.1 (-1.6, 5.7)
Fasting non-HDL-C, mg/dL	247	3.5 (0.5, 6.4)	226	0.4 (-2.2, 3.1)	129	2.2 (-4.1, 8.5)	121	-0.4 (-6.4, 5.6)
Insulin resistance (HOMA-IR)	261	-0.3 (-1.5, 1.0)	253	0.3 (0.0, 0.7)	123	0.1 (-0.3, 0.4)	130	0.1 (-0.1, 0.3)
eGFR from creatinine, mL/min/1.73m²	333	6.8 (5.3, 8.3)	340	-1.7 (-3.1, -0.3)	177	-2.0 (-3.9, -0.0)	172	7.1 (5.1, 9.1)
eGFR from cystatin C, mL/min/1.73m²	336	-0.1 (-1.1, 0.9)	339	-2.4 (-3.4, -1.5)	175	-2.4 (-3.7, -1.1)	169	-1.9 (-3.4, -0.4)

*With 95% confidence internal (CI) based on the t-distribution.
Participants taking lipid-lowering agents were excluded from the LDL-C and non-HDL-C analyses.
HOMA-IR=homeostatic model assessment-insulin resistance; participants taking diabetes medication were excluded from this analysis.

- In Group 1, weight gain was minimal from Week 48 to Week 96 (mean change -0.01 kg; **Figure 4**)
- In Group 2, mean weight change during the first 48 weeks after switching to DOR/ISL (Week 48-96; 0.89 kg) was similar to Group 1 (Week 0-48; 0.94 kg)
- Weight change was greater in participants who switched from weight-suppressive bART containing efavirenz (EFV) and/or tenofovir disoproxil fumarate (TDF)
 - Weight gain after removal of EFV or TDF in Group 2 (mean 2.45 kg) was commensurate with Group 1 Week 0-48 (mean 2.48 kg)
 - Participants who switched from a regimen that did not contain EFV or TDF had minimal weight change at Week 48 and Week 96

Figure 4. Mean change in weight at Week 48 and Week 96

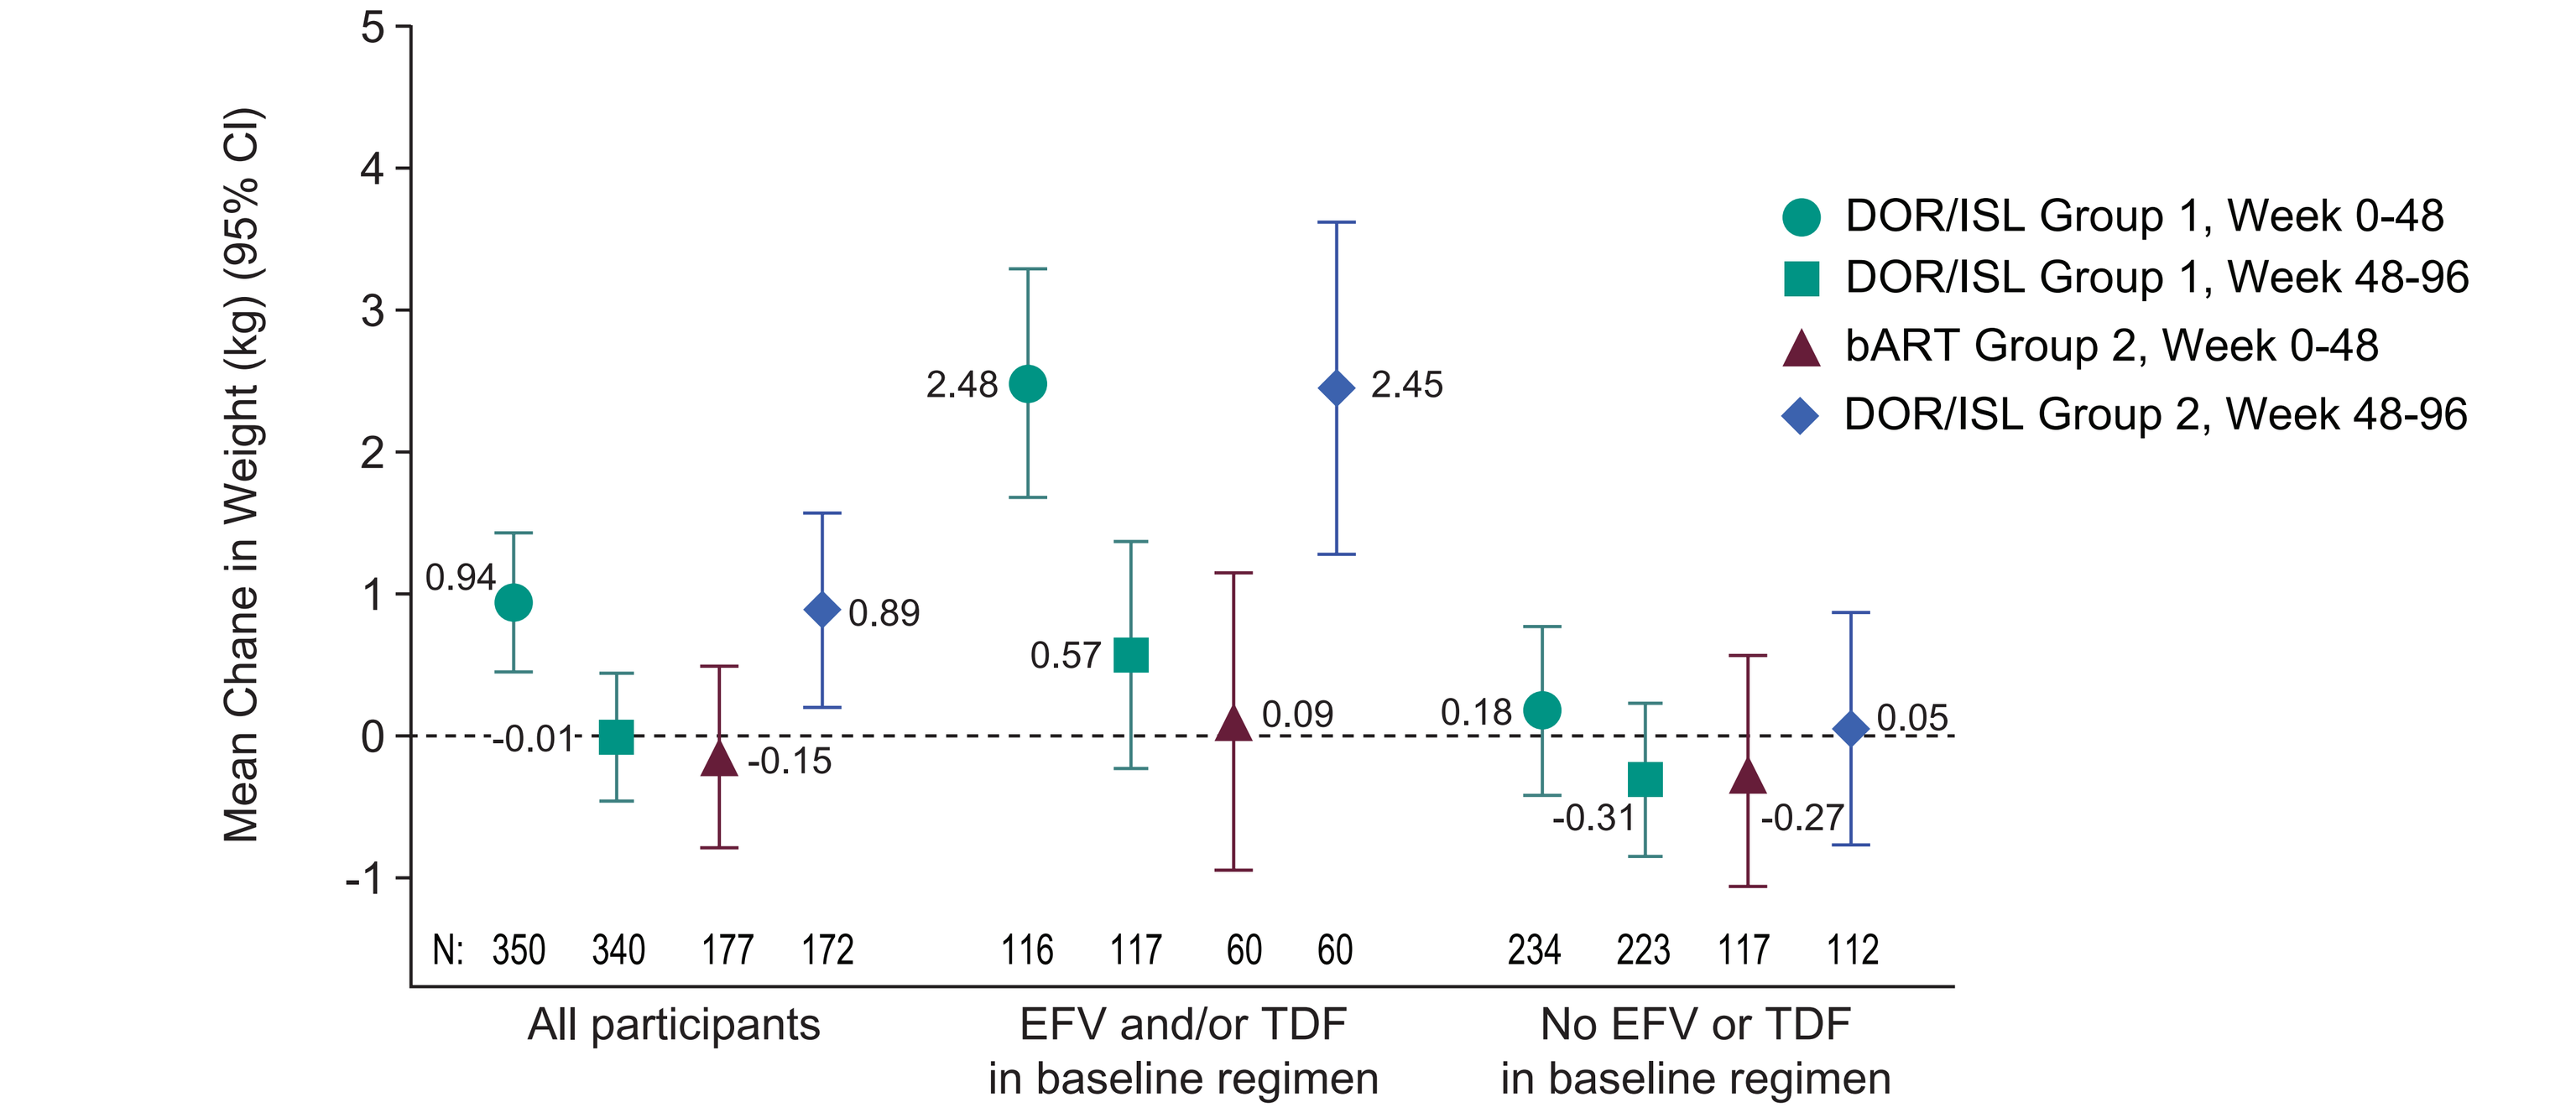
Mean Chane in Weight (kg) (95% CI)
5
4
3
2
1
0
-1
0.94
-0.01
-0.15
0.89
2.48
0.57
0.09
2.45
0.18
-0.31
-0.27
0.05
N: 350 340 177 172 116 117 60 60 234 223 117 112
All participants
EFV and/or TDF in baseline regimen
No EFV or TDF in baseline regimen
DOR/ISL Group 1, Week 0-48
DOR/ISL Group 1, Week 48-96
bART Group 2, Week 0-48
DOR/ISL Group 2, Week 48-96

Conclusions

- Switching to DOR/ISL (100/0.25 mg) maintained high rates of virologic suppression (HIV-1 RNA <50 copies/mL) through 96 weeks of treatment
 - 48 weeks of DOR/ISL: 95.6% in Group 1 (Week 0-48) and 96.6% in Group 2 (Week 48-96)
 - 96 weeks of DOR/ISL: 92.6% in Group 1 (Week 0-96)
- No treatment-emergent resistance to DOR or ISL was observed through Week 96
- Switching to DOR/ISL had no adverse effects on total lymphocyte or CD4+ T-cell counts through Week 96
- DOR/ISL maintained a favorable safety and tolerability profile over 96 weeks

Plain language summary

In this open-label clinical study, switching from any oral HIV treatment regimen to once-daily DOR/ISL (100/0.25 mg) maintained viral suppression for 96 weeks in more than 92% of the study participants, with no evidence of viral resistance to either DOR or ISL in any participant. DOR/ISL showed a favorable safety and tolerability profile with very few treatment-related side effects reported after week 48, and no negative effects on total lymphocyte or CD4+ T-cell counts observed through week 96.

Copyright © 2026 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.