Type I IFN Signaling and Regulation in Vesatolimod-Treated Virally Suppressed Adults With HIV-1

Susie SY Huang, Liao Zhang, Donovan Verrill, Christiaan R de Vries, Elena Vendrame, Devi SenGupta, Jeffrey J Wallin, Yanhui Cai

Gilead Sciences, Foster City, CA, USA

Poster 00542

GS-US-382-1450

Conclusions

- Whole blood transcriptome analysis identified differences in interferon-stimulated gene (ISG) expression based on vesatolimod (VES) concentration and the order of VES dose administration
- Signals of positive and negative regulation of type I interferon (IFN) signaling were induced post-dose at all VES concentrations evaluated
- Induction of type I IFN responses decreased over multiple doses at 8 mg, but were more sustained at 6 mg
- The results suggest that a cumulative biological effect from multiple VES doses may modulate the VES pharmacodynamic (PD) response

Plain Language Summary

Selective and potent toll-like receptor (TLR) agonists are under clinical investigation for the treatment of viral infections. VES is an oral, selective TLR-7 agonist that has been shown to increase immune cell activation with a peak PD response at 8 mg. In this study, we identified differences in VES PD response based on VES concentration and the cumulative effect of multiple VES doses. These findings inform the use of VES in future HIV cure combination studies.

Introduction

- VES (formerly known as GS-9620) is a well-tolerated, orally administered toll-like receptor-7 (TLR-7) agonist under development as a component of an HIV cure regimen[1]

Figure 1. VES Mechanism of Action

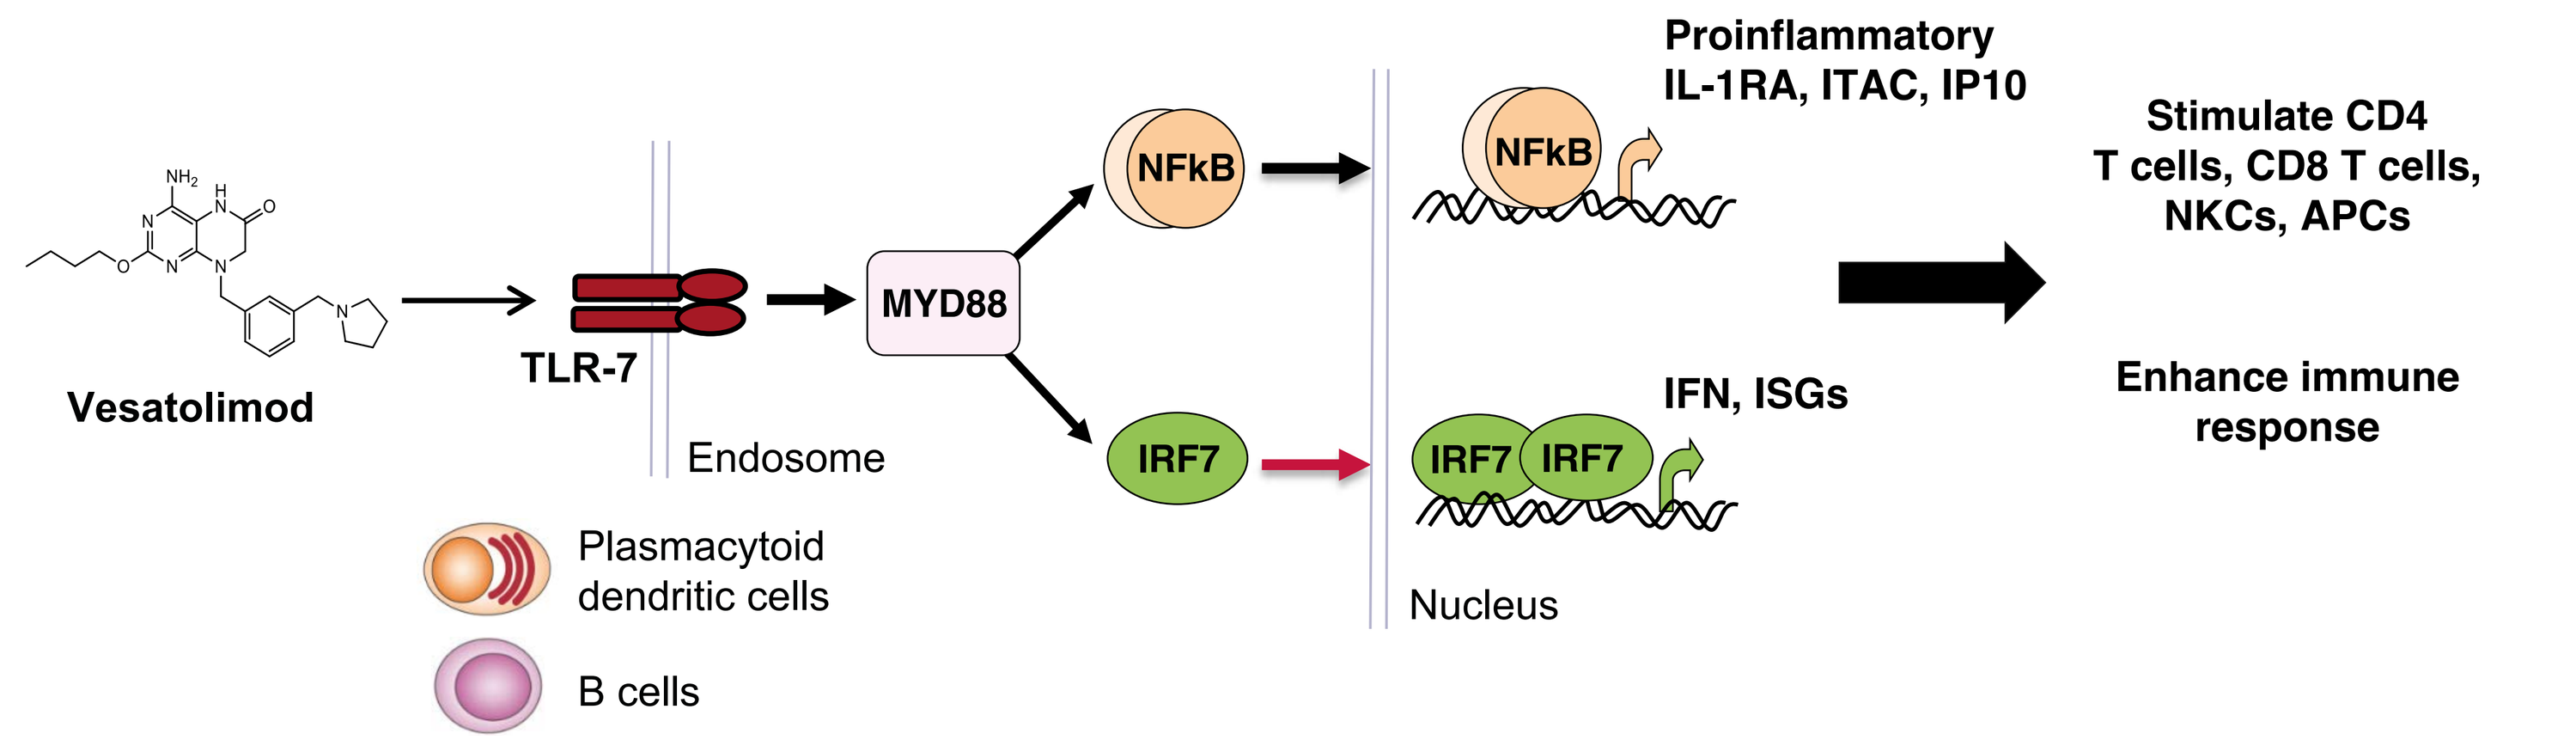
Vesatolimod
TLR-7
Endosome
MYD88
NFkB
IRF7
Proinflammatory IL-1RA, ITAC, IP10
IFN, ISGs
Nucleus
Stimulate CD4 T cells, CD8 T cells, NKCs, APCs
Enhance immune response
Plasmacytoid dendritic cells
B cells

APC, antigen-presenting cell; IFN, interferon; IL, interleukin; IRF, IFN regulatory factor; IP, IFN-gamma-inducible protein; ISG, IFN-stimulated gene; ITAC, interferon-inducible T-cell alpha chemoattractant; MYD, myeloid differentiation; NKC, natural killer cells; RA, receptor antagonist; TLR-7, toll-like receptor 7.

- VES increases expression of ISGs and immune cell activation (**Figure 1**) in people living with HIV (PLWH), and VES monotherapy promotes a modest delay in HIV rebound in HIV controllers[1,2]
- In a phase 1b study, VES PD response was higher with 8 mg compared with 10-12 mg doses in PLWH treated during chronic infection[1]
- This study investigated the immune mechanism and regulation of IFN signaling pathways in response to VES (6, 8, and 10/12 mg) from whole blood transcriptome

References: **1.** Riddler SA, et al. *Clin Infect Dis*. 2021;72:e815-e824. **2.** SenGupta D, et al. *Sci Transl Med*. 2021;13:eabg3071.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators. This study was funded by Gilead Sciences, Inc. Editing and production assistance were provided by Parexel, and funded by Gilead Sciences, Inc.

Disclosures: All authors are employees of Gilead Sciences, Inc., and may own stock in Gilead Sciences, Inc.

Correspondence: Susie SY Huang, Susie.Huang9@gilead.com.

Methods

Figure 2. Study Design

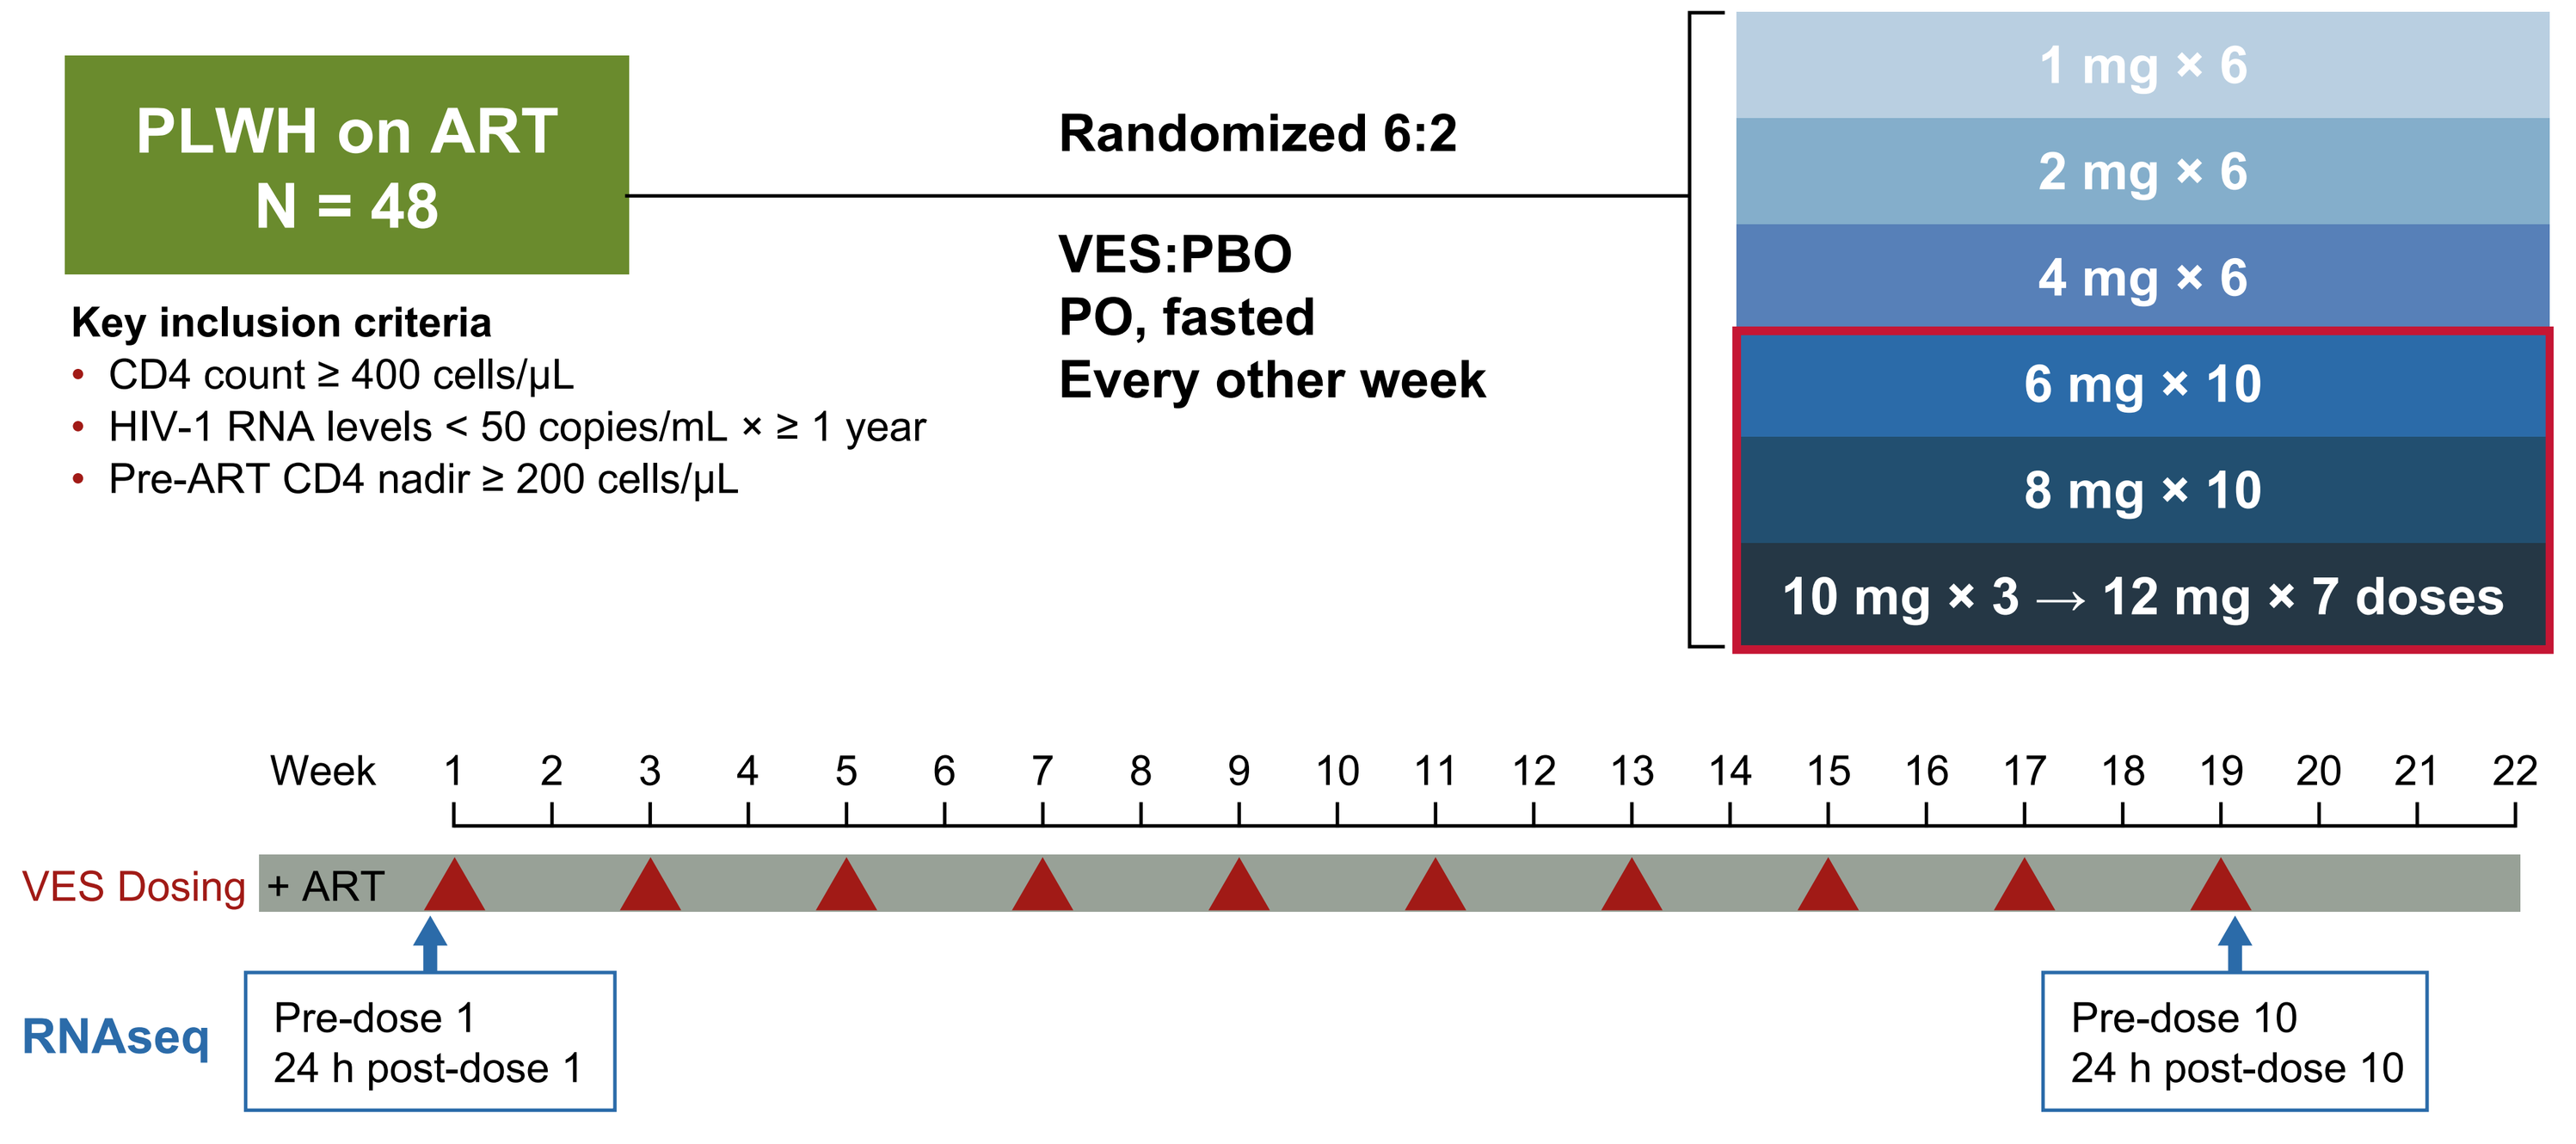
PLWH on ART
N = 48
Key inclusion criteria
CD4 count ≥ 400 cells/μL
HIV-1 RNA levels < 50 copies/mL × ≥ 1 year
Pre-ART CD4 nadir ≥ 200 cells/μL
Randomized 6:2
VES:PBO
PO, fasted
Every other week
1 mg × 6
2 mg × 6
4 mg × 6
6 mg × 10
8 mg × 10
10 mg × 3 → 12 mg × 7 doses
Week
VES Dosing
+ ART
RNAseq
Pre-dose 1
24 h post-dose 1
Pre-dose 10
24 h post-dose 10

ART, antiretroviral therapy; PBO, placebo; PLWH, people living with HIV; PO, orally; VES, vesatolimod.

- GS-US-382-1450 (NCT02858401) is a phase 1b, double-blind, placebo-controlled trial of VES PD and immune cell activation in PLWH on antiretroviral therapy (ART) (**Figure 2**)
- The study enrolled 48 virally suppressed PLWH (age ≥ 18 years, plasma HIV-1 RNA < 50 copies/mL) and randomized 6:2 to receive VES or placebo (PBO) for 10 doses
- Whole blood collected predose and 24 hours after doses 1 and 10 from participants who received VES 6, 8, or 10-12 mg (n = 24) was used for mRNA-Seq (Illumina Stranded mRNA Prep; NovaSeq 6000) and aligned and mapped to Gencode v38
- A linear mixed-effects model tested for differentially expressed genes (DEGs) associated with type I IFN regulation and regulators of pattern recognition receptors (false discovery rate [FDR] < 0.1), including
 - — Nanostring type I IFN gene signature, negative regulators for type I IFN response, IFN regulatory factors activation, and VES PD: ISG15, MIX1, OAS1

Results

Figure 3. After the First VES Dose, the Highest Number of DEGs Associated With Type I IFN Responses Was at 8 mg

	VES 6 mg	VES 8 mg	VES 10/12 mg
FDR < 0.1	21	38	15
FDR < 0.05	17	34	11

N = 6; padj-values only.

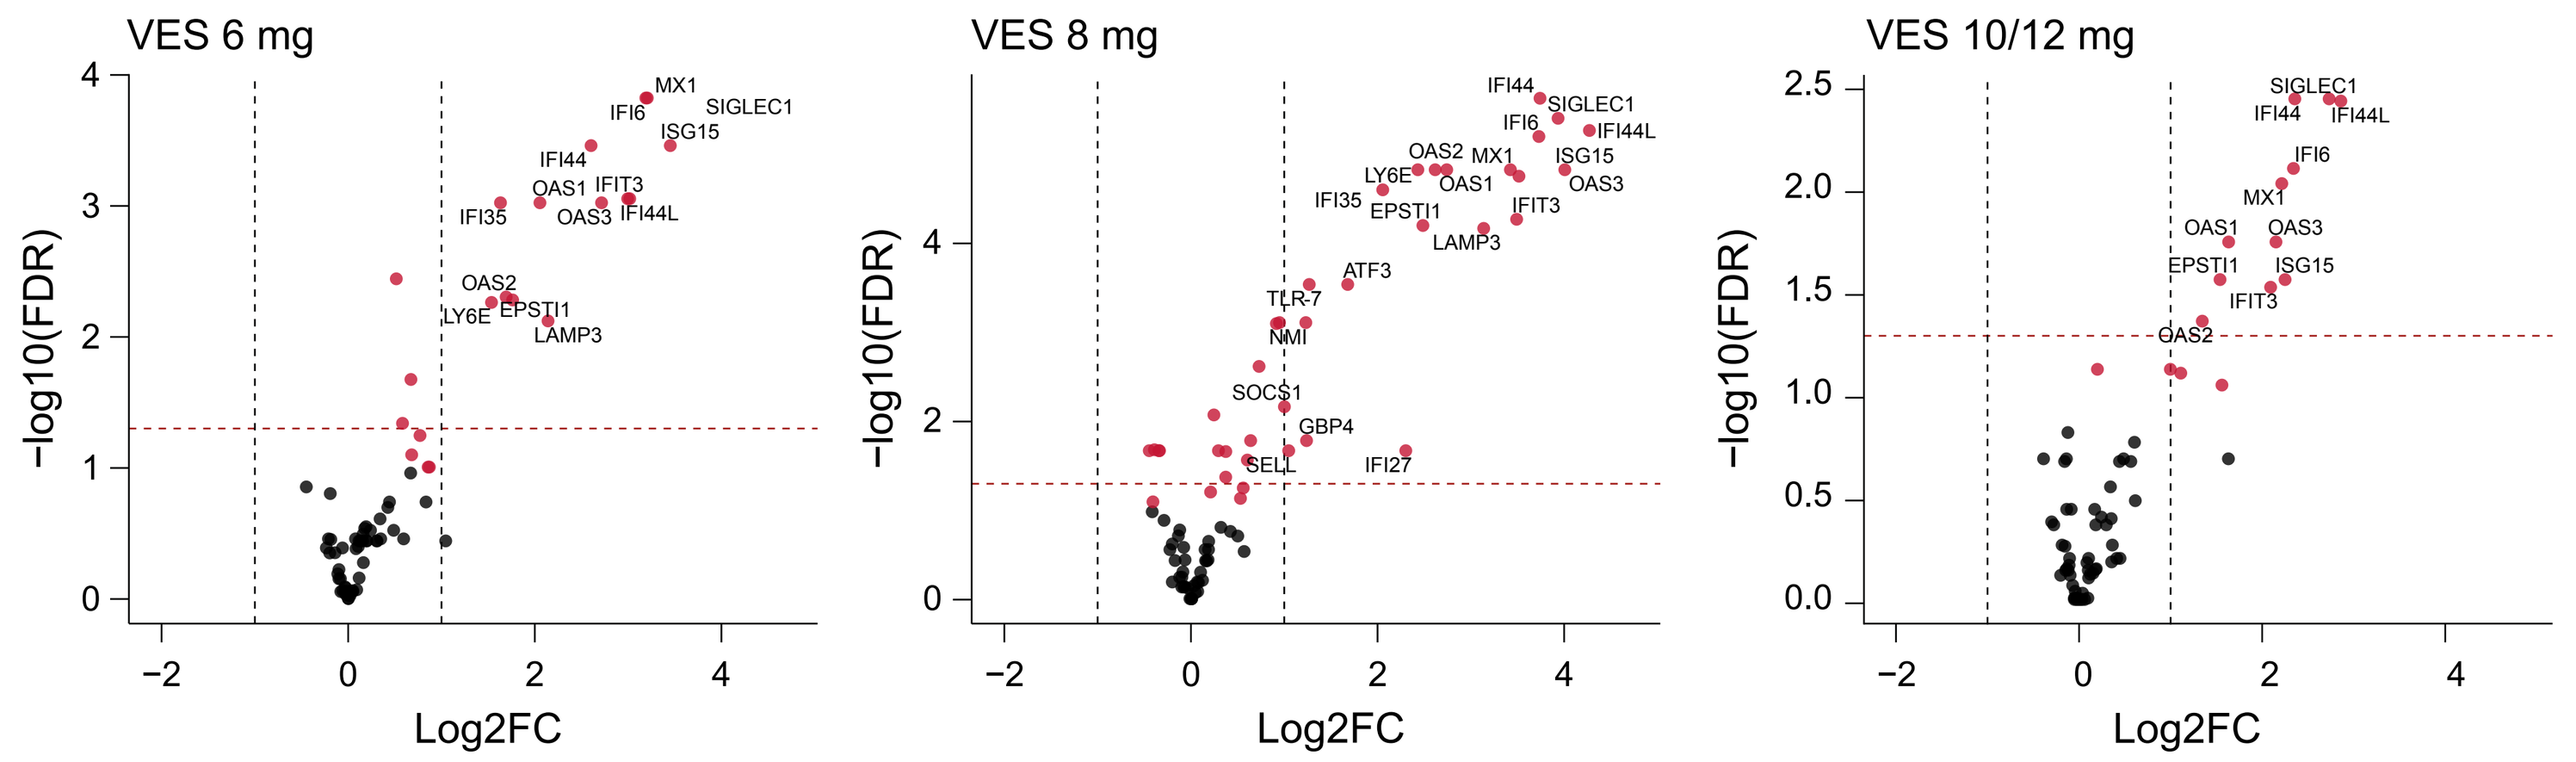
VES 6 mg
VES 8 mg
VES 10/12 mg
−log10(FDR)
Log2FC

Significance denoted at FDR < 0.1; black dashed lines denote abs(log2FC)=1; black circles denote non-significant genes; red dashed lines denote FDR < 0.05. DEG, differentially expressed gene; FC, fold change; FDR, false discovery rate; IFN, interferon; VES, vesatolimod.

- The highest number of DEGs (FDR < 0.1), including those associated with activation and inhibition, was observed with VES 8 mg (38 genes), followed by VES 6 mg (21 genes) (**Figure 3**)

Figure 4. Fourteen DEGs Were Common Among VES Concentrations Post-Dose 1 (FDR < 0.1)

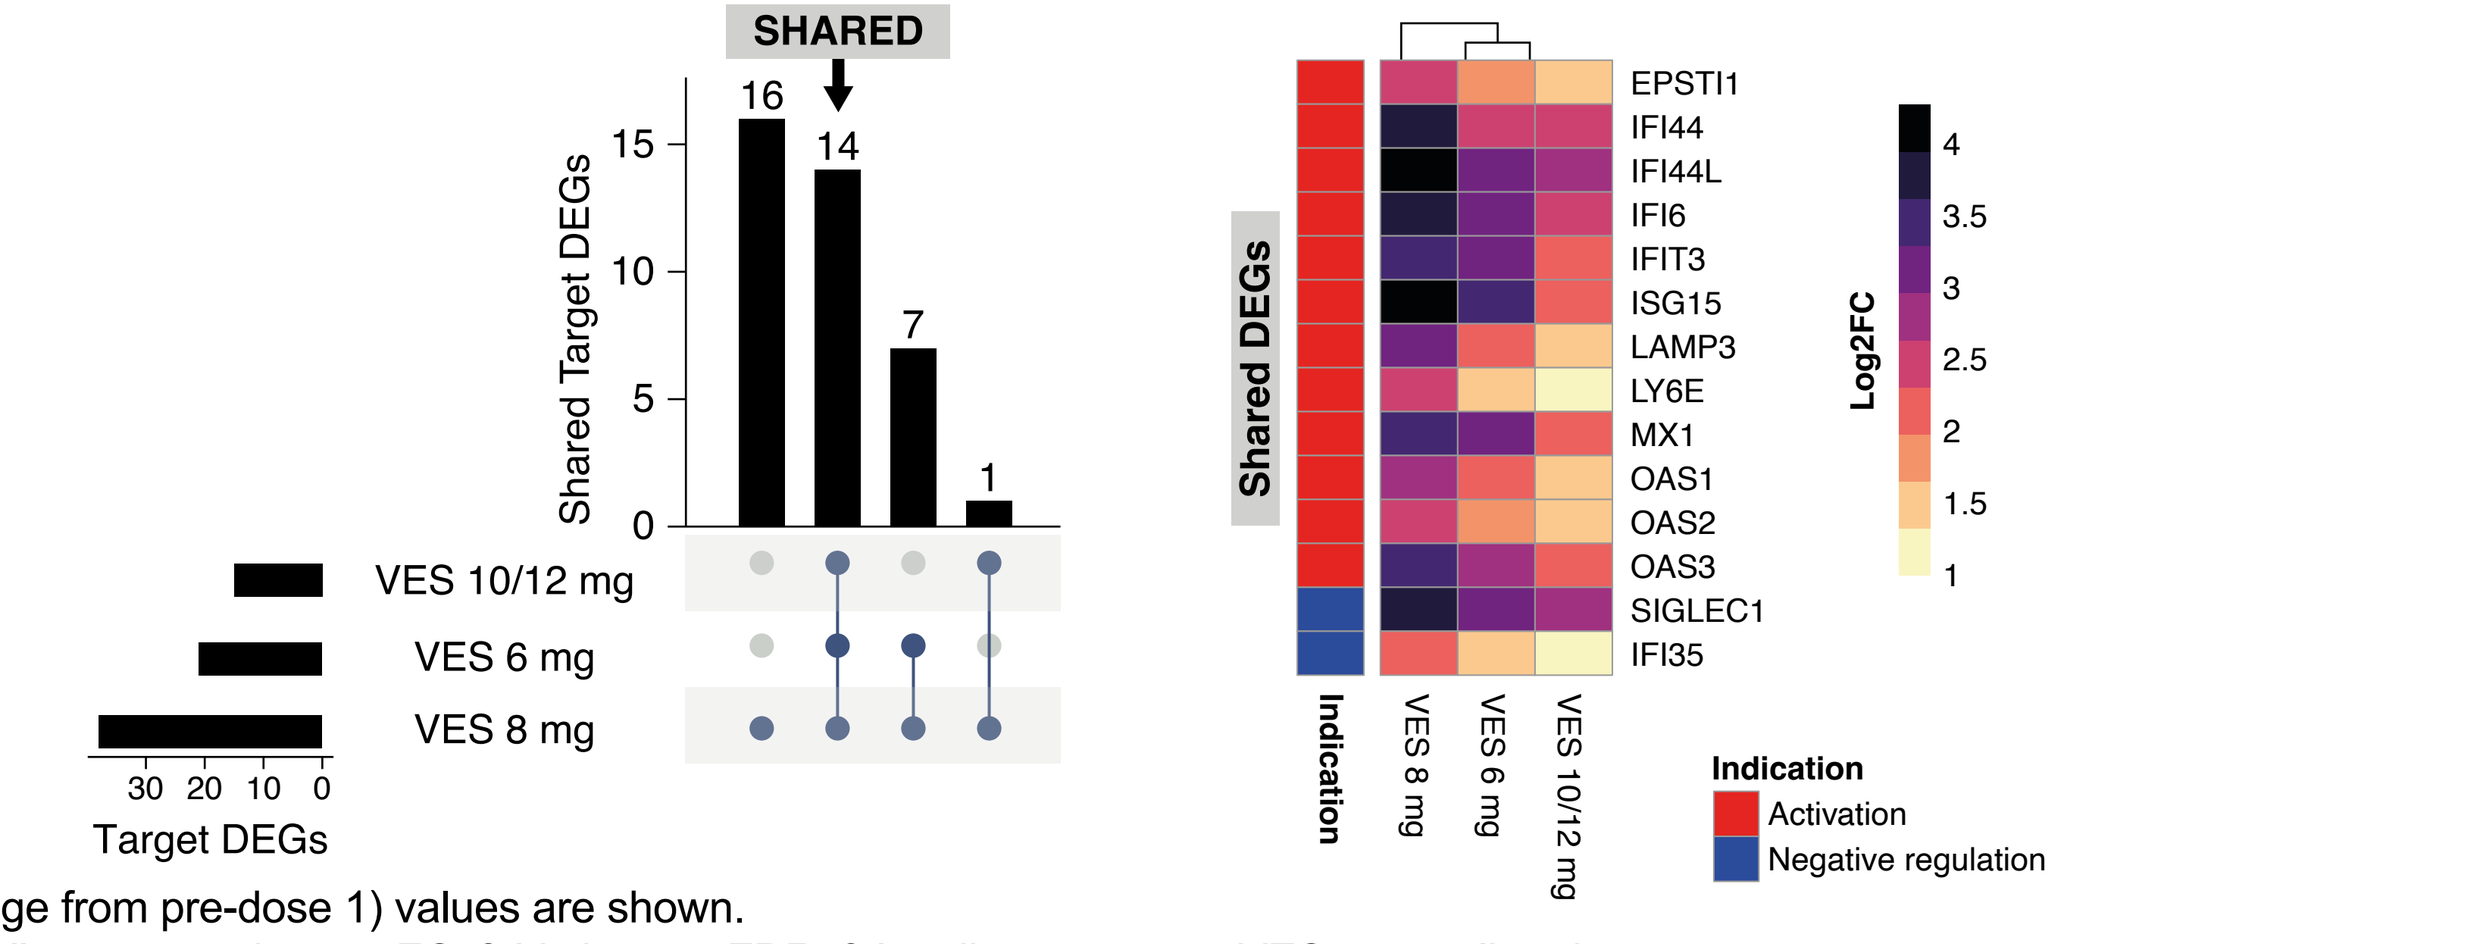
SHARED
Shared Target DEGs
Target DEGs
VES 10/12 mg
VES 6 mg
VES 8 mg
Shared DEGs
EPSTI1
IFI44
IFI44L
IFI6
IFIT3
ISG15
LAMP3
LY6E
MX1
OAS1
OAS2
OAS3
SIGLEC1
IFI35
Log2FC
Indication
Activation
Negative regulation

Log2 (fold change from pre-dose 1) values are shown.
DEG, differentially expressed gene; FC, fold change; FDR, false discovery rate; VES, vesatolimod.

- Seven genes were upregulated and shared between VES 6 mg and 8 mg (**Figure 4**)
 - — *ISG15*, *MX1*, *IFI44*, *IFI6*, *SIGLEC1*, *OAS3*, and *IFIT3*

Figure 5. Greatest Upregulation of Type I IFN Responses After the First VES Dose Was at 8 mg

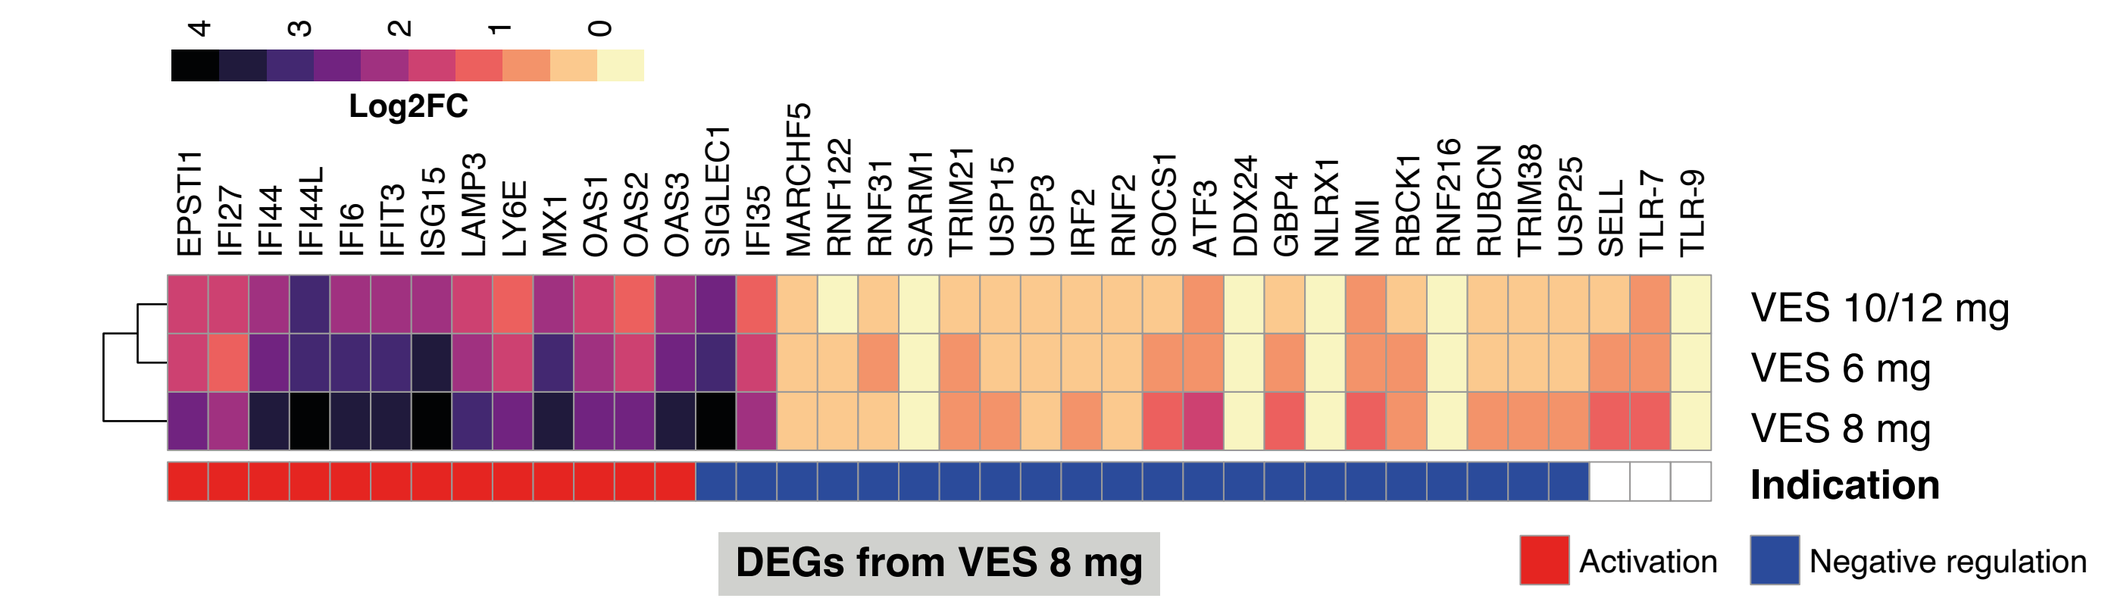
Log2FC
VES 10/12 mg
VES 6 mg
VES 8 mg
Indication
DEGs from VES 8 mg
Activation
Negative regulation

Residual Response[a] (38 Genes)

VES Concentration	Residual
8 mg	2.84
6 mg	2.22
10/12 mg	1.90

[a]Residual = median of activation[log2FC] – median of negative regulation[log2FC].
DEG, differentially expressed gene; FC, fold change; VES, vesatolimod.

- Activation and negative regulation of type I IFN signaling were induced post-dose at all VES concentrations evaluated (6-12 mg), but the residual value (indicating summed effect of type I IFN response to VES) was highest with 8 mg and lowest with 10/12 mg (**Figure 5**)

Figure 6. VES Induced the Most DEGs Associated With Type I IFN Response Post-Dose 10 at 6 mg

	VES 6 mg	VES 8 mg	VES 10/12 mg
FDR < 0.1	49	31	27
FDR < 0.05	45	26	0

N = 6; VES 10/12 mg N=5; padj-values only.

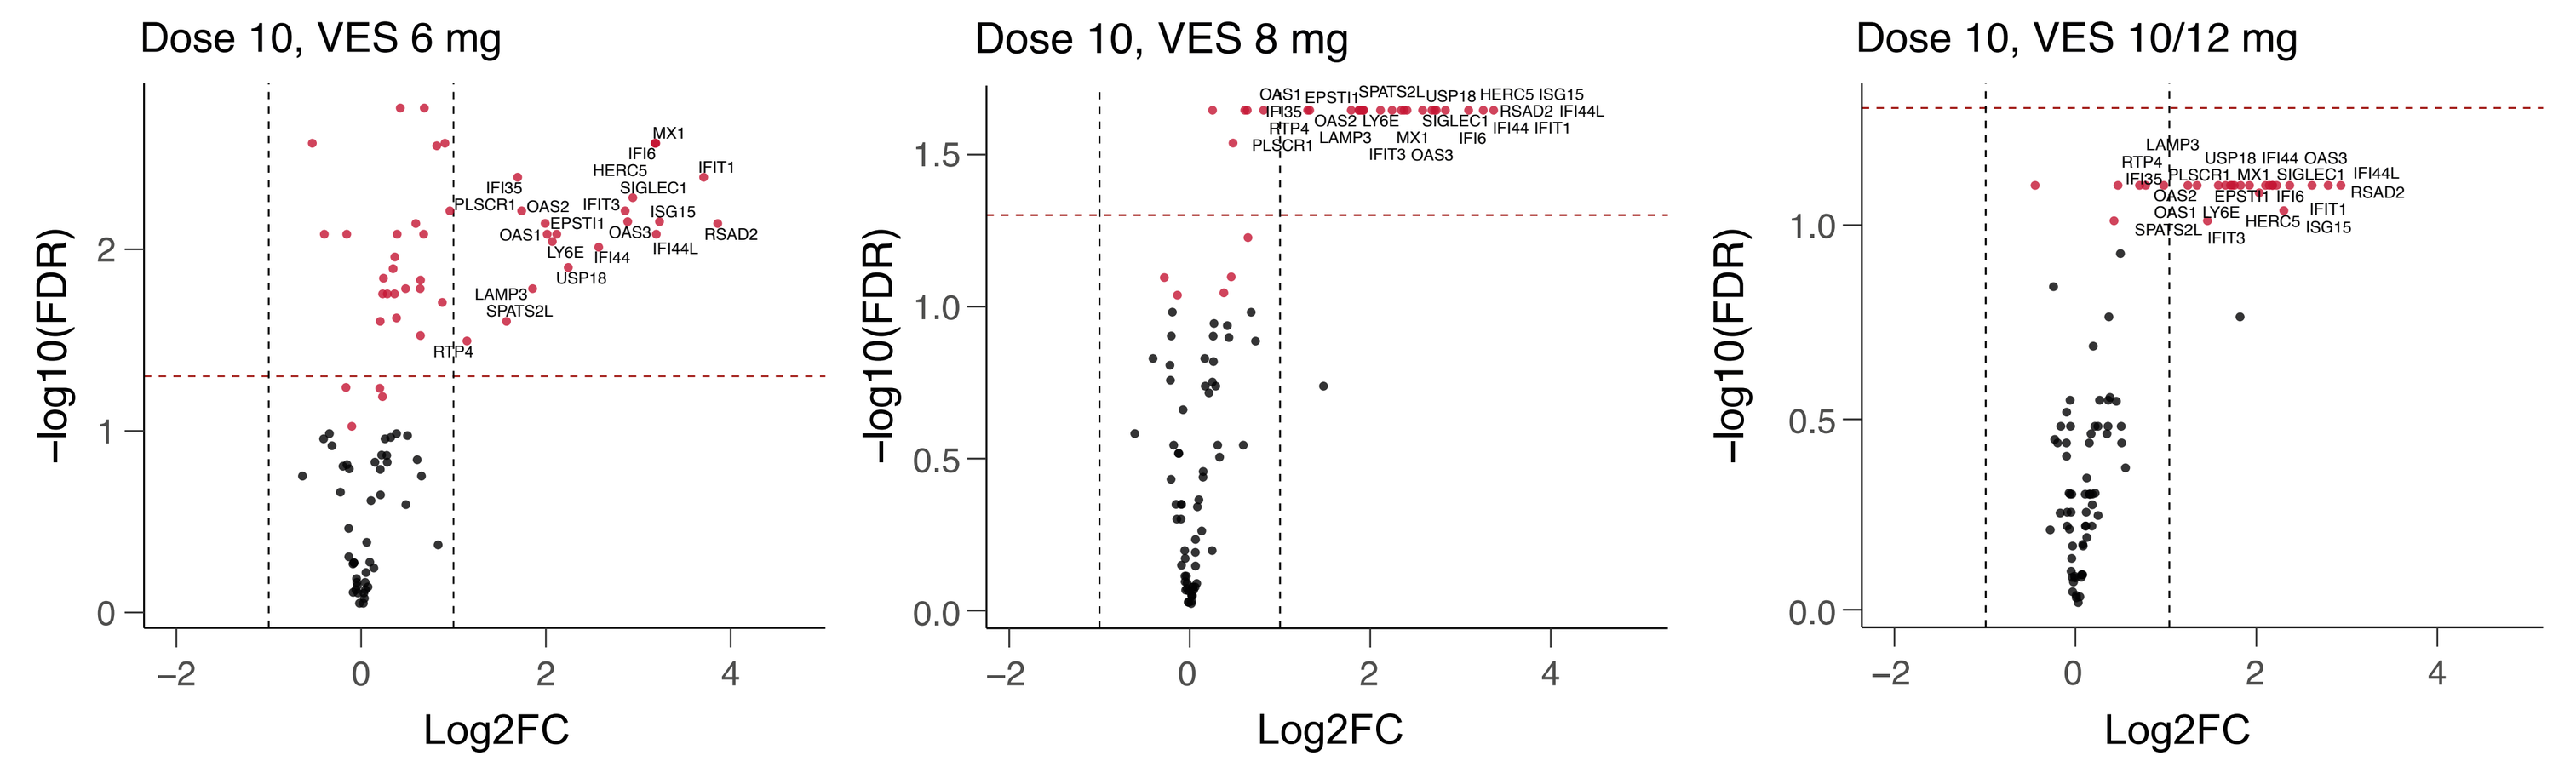
Dose 10, VES 6 mg
Dose 10, VES 8 mg
Dose 10, VES 10/12 mg
−log10(FDR)
Log2FC

Significance denoted at FDR < 0.1; black dashed lines denote abs(log2FC) = 1; black circles denote non-significant genes; red dashed lines denote FDR < 0.05; no target gene with FDR < 0.05 in VES 10/12 mg. DEG, differentially expressed gene; FC, fold change; FDR, false discovery rate; VES, vesatolimod.

- The highest number of DEGs (FDR < 0.1), including those associated with activation and inhibition, was observed with VES 6 mg (49 genes), followed by VES 8 mg (31 genes) and 10/12 mg (27 genes)
 - — The increased upregulation of activators was most noticeable with VES 6 mg (**Figure 6**)

Figure 7. Twenty-five DEGs Were Common Among VES Concentrations Post-Dose 10

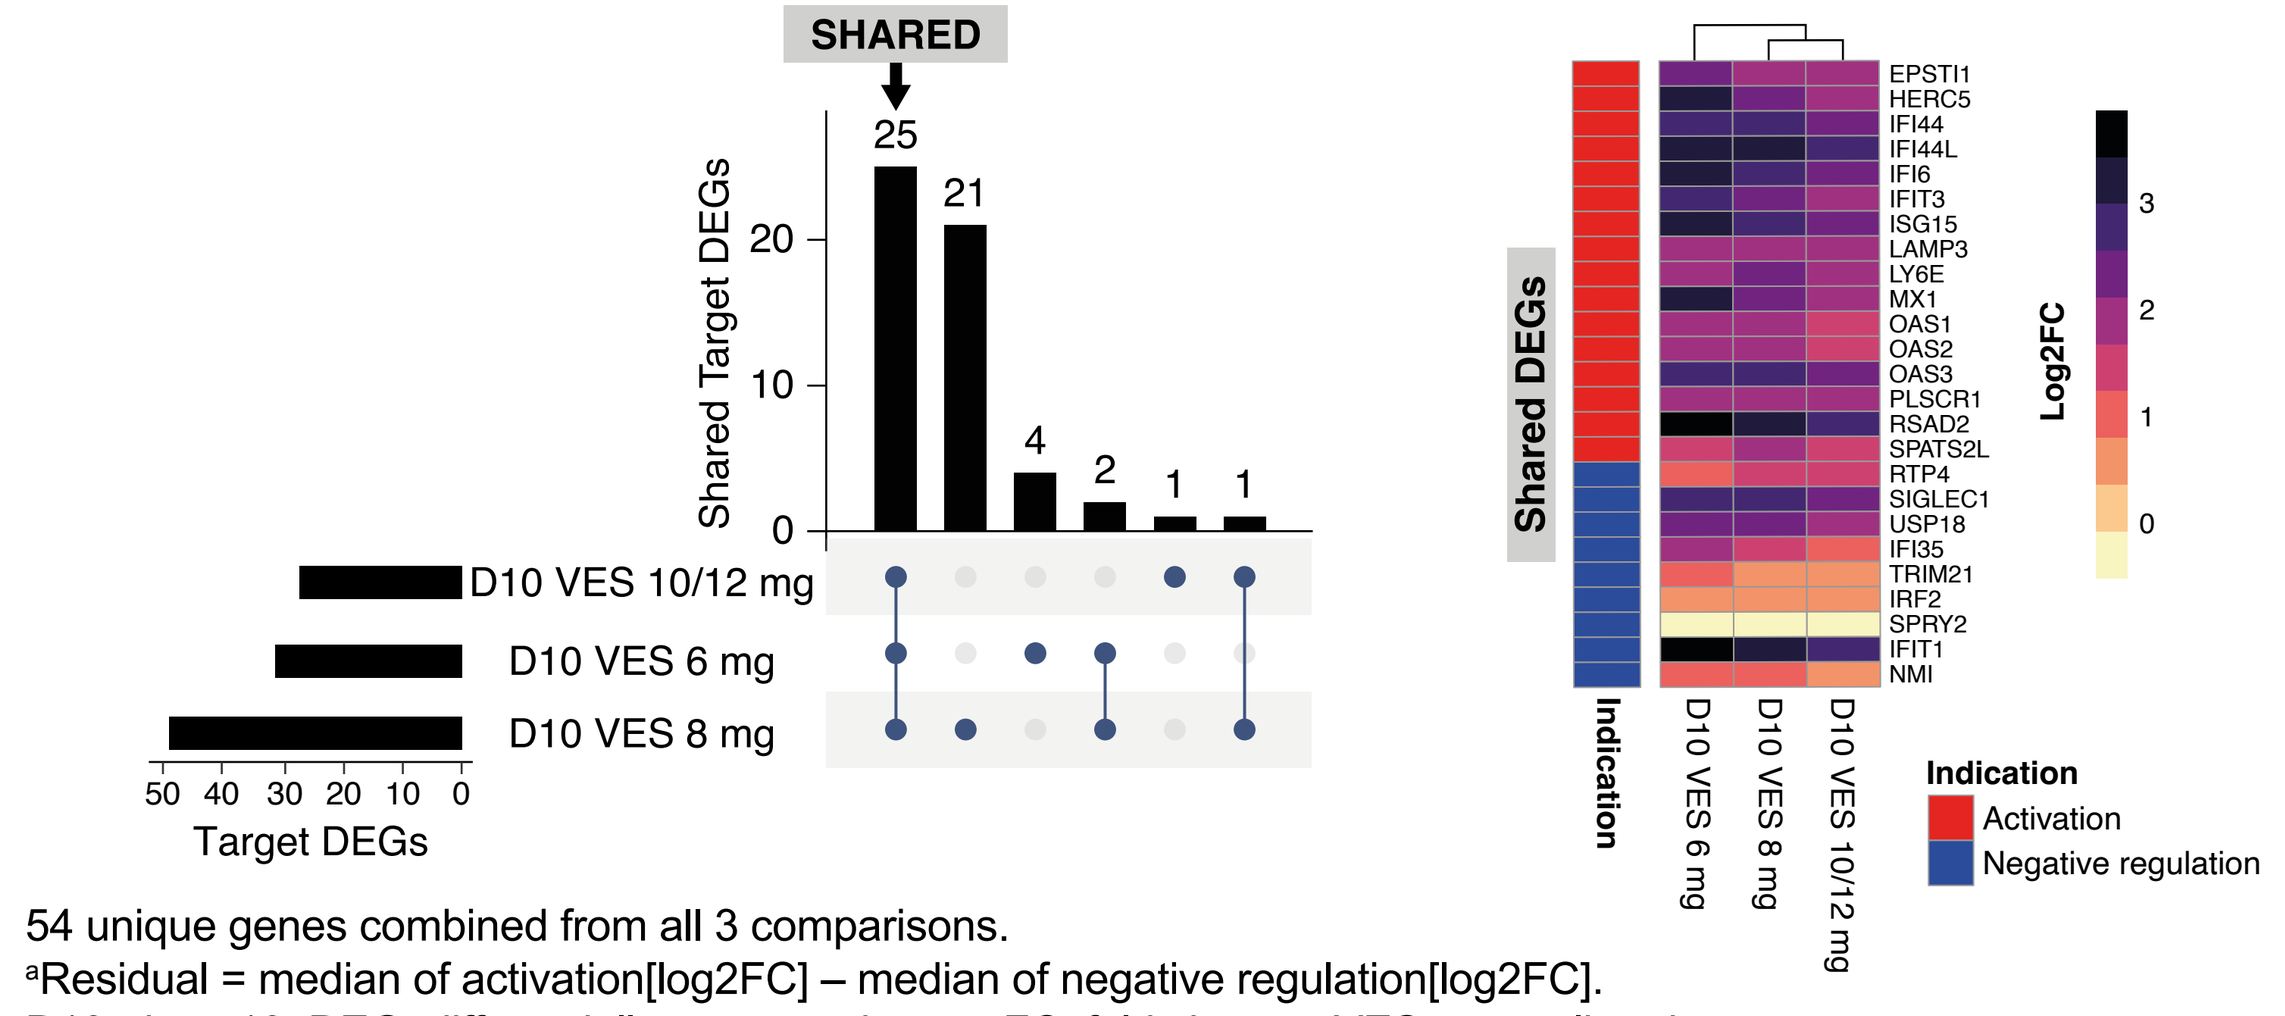
SHARED
Shared Target DEGs
Target DEGs
D10 VES 10/12 mg
D10 VES 6 mg
D10 VES 8 mg
Shared DEGs
Indication
Log2FC
Activation
Negative regulation

Residual Response[a] (54 Unique DEGs)

VES Concentration	Residual
8 mg	1.82
6 mg	1.66
10/12 mg	1.45

54 unique genes combined from all 3 comparisons.
[a]Residual = median of activation[log2FC] – median of negative regulation[log2FC].
D10, dose 10; DEG, differentially expressed gene; FC, fold change; VES, vesatolimod.

- Compared with the first dose, residual values after the 10th dose were more similar among VES concentrations and highest with 8 mg (**Figure 7**)

Figure 8. DEGs Are Regulated to Different Extents by VES Concentrations and Dose

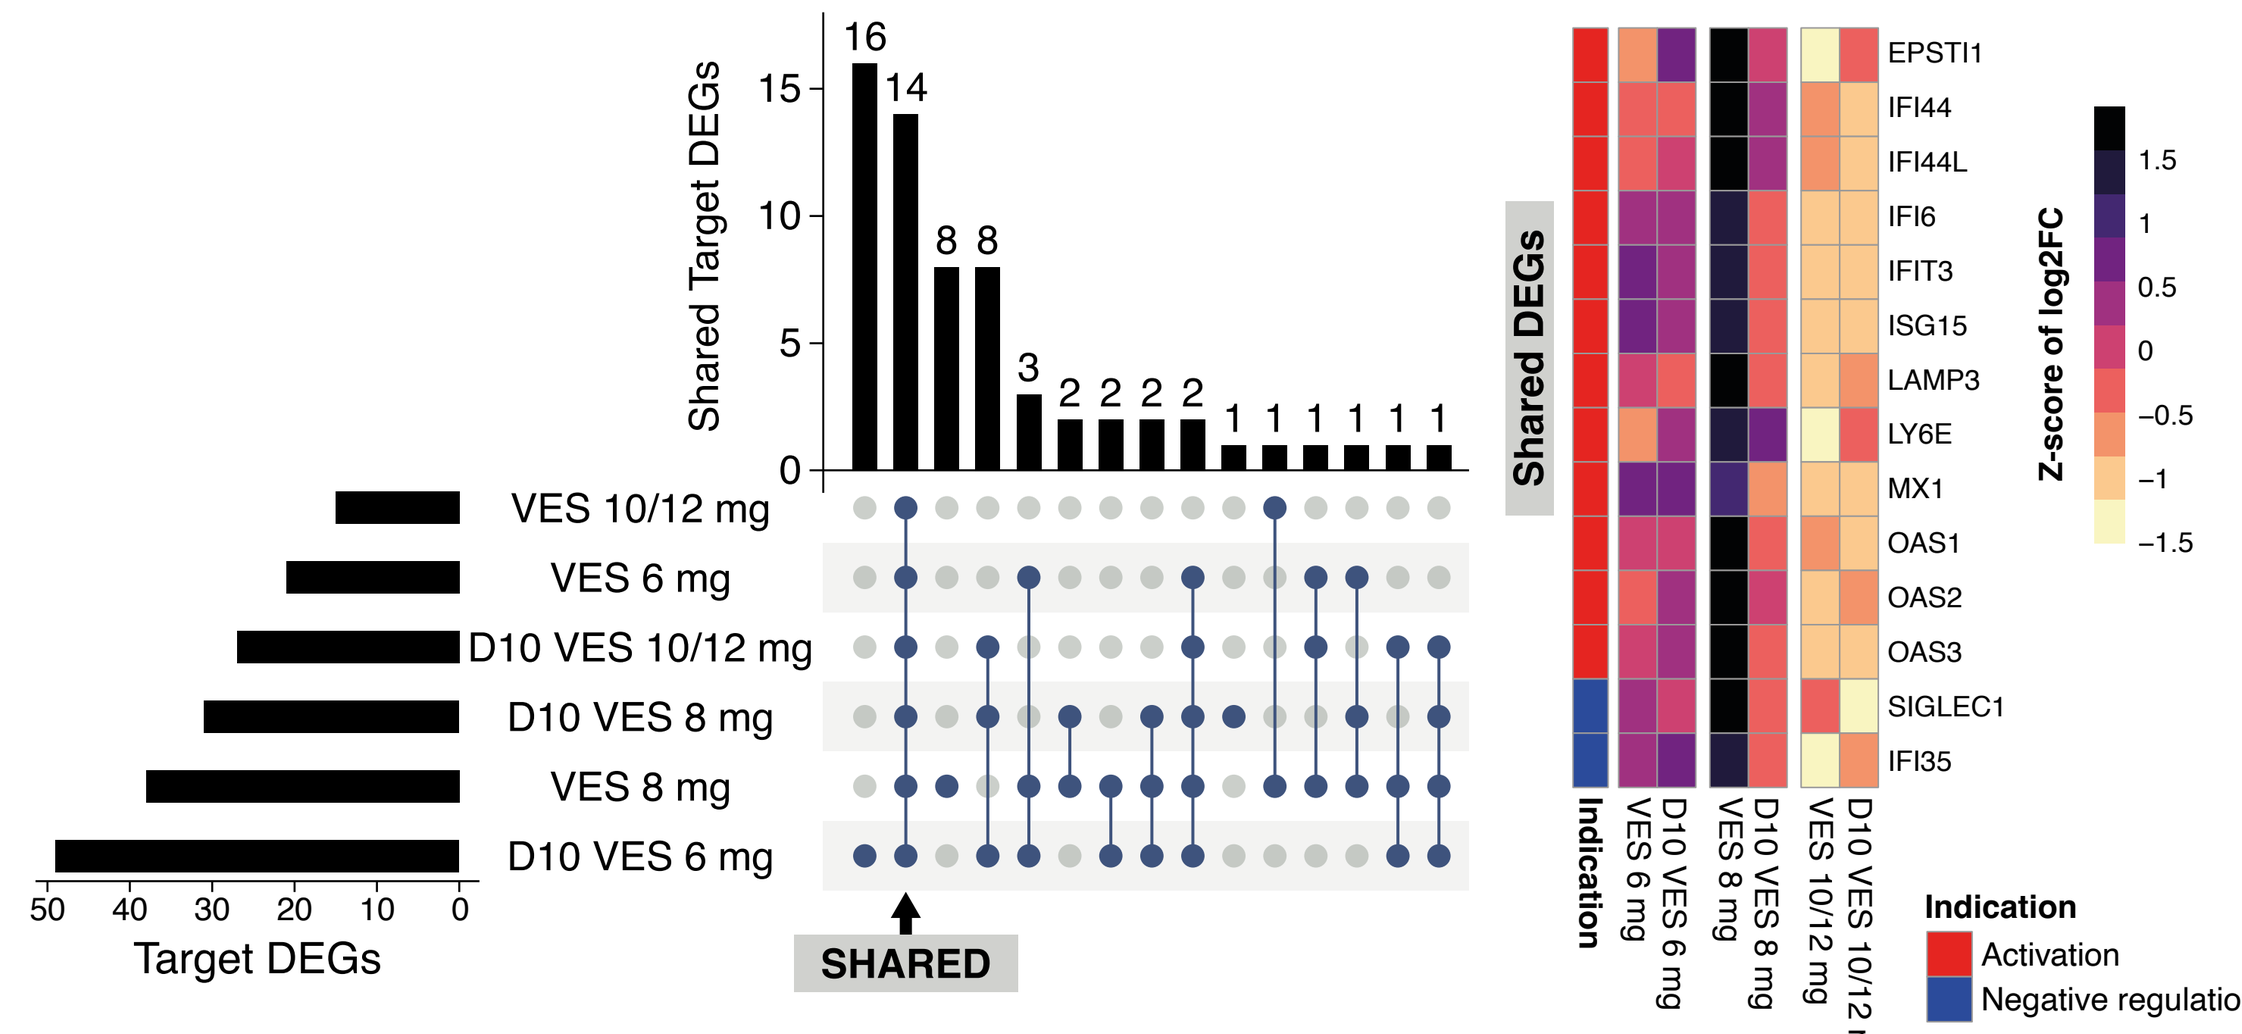
Shared Target DEGs
Target DEGs
SHARED
VES 10/12 mg
VES 6 mg
D10 VES 10/12 mg
D10 VES 8 mg
VES 8 mg
D10 VES 6 mg
Shared DEGs
Z-score of log2FC
Indication
Activation
Negative regulation

Residual Response[a] (14 Shared DEGs)

VES Concentration	Dose Order	Residual
8 mg	1	0.46
6 mg	10	0.40
10/12 mg	10	0.35
8 mg	10	0.34
10/12 mg	1	0.26
6 mg	1	0.24

[a]Residual = median of activation[log2FC] – median of negative regulation[log2FC].
D10, dose 10; DEG, differentially expressed gene; FC, fold change; VES, vesatolimod.

- Over multiple doses, upregulation of type I IFN response to VES was highest after the first dose at 8 mg (**Figure 8**)
- However, VES 6 mg induced the greatest increase in type 1 IFN response after the 10th dose (**Figure 8**)

Figure 9. Induction of Type I IFN Response to VES Decreased Over Multiple Doses at 8 mg, but Was More Sustained at 6 mg

- Induction of ISGs diminished in the 8 mg concentration group by dose 10, except for LY6E (**Figure 9**)
- 10/12 mg maintained relatively similar induction levels between doses but exhibited the smallest increase in fold change

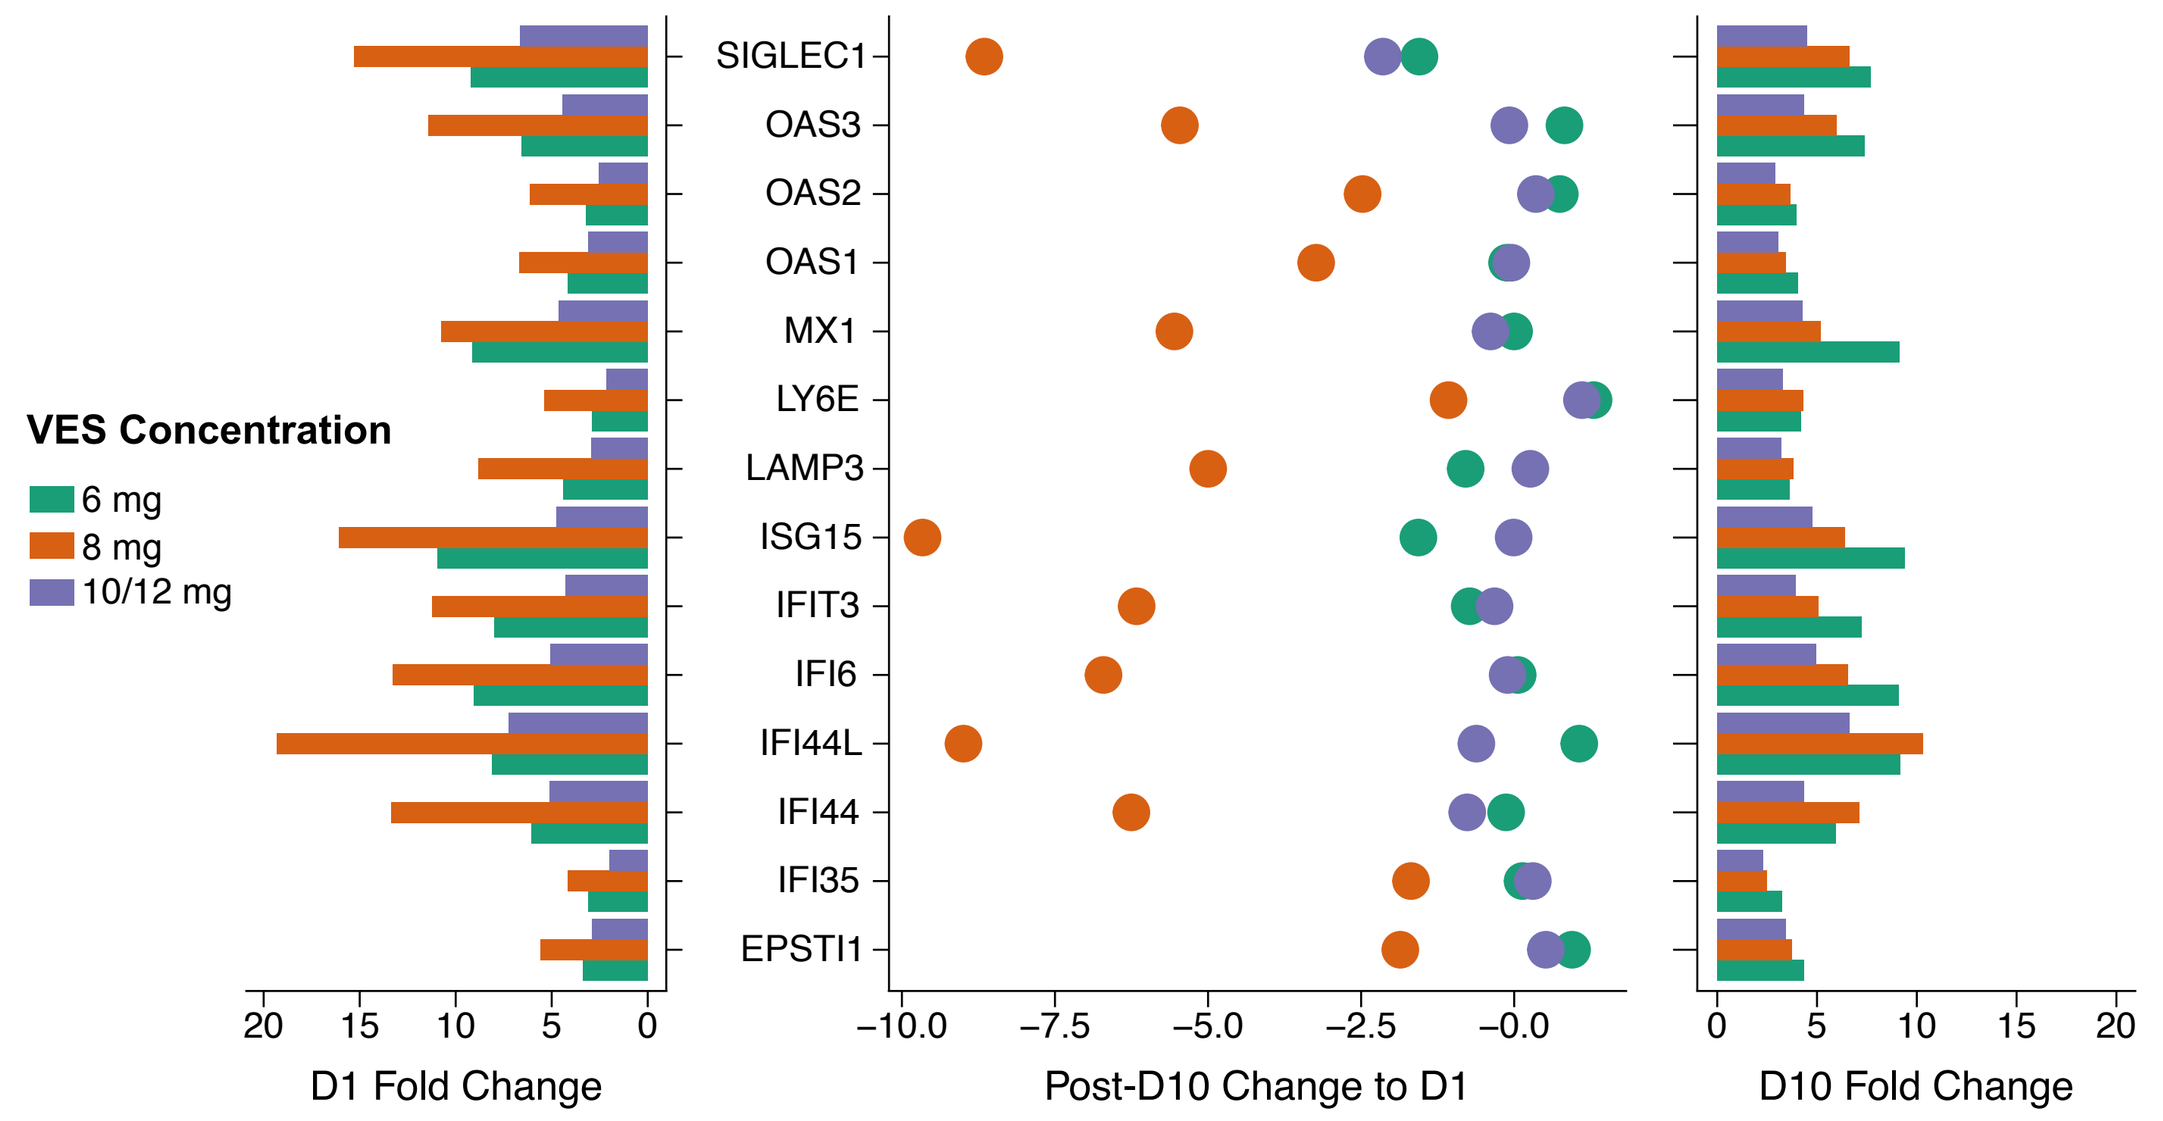
VES Concentration
6 mg
8 mg
10/12 mg
SIGLEC1
OAS3
OAS2
OAS1
MX1
LY6E
LAMP3
ISG15
IFIT3
IFI6
IFI44L
IFI44
IFI35
EPSTI1
D1 Fold Change
Post-D10 Change to D1
D10 Fold Change

Fold change: 24 hours post-dose – pre-dose; post-D10 change to D1 = D10 fold change – D1 fold change.
D1, dose 1; D10, dose 10; VES, vesatolimod.

Conference on Retroviruses and Opportunistic Infections (CROI); March 3-6, 2024; Denver, CO, USA