LOWER ADCC AFTER 26 WEEKS OF PEG-IFN-α2b AND TWO bNAbs IN OTHERWISE SUPPRESSED HIV-1+ INDIVIDUALS

THE WISTAR INSTITUTE — TODAY'S DISCOVERIES – TOMORROW'S CURES

BEAT-HIV DELANEY COLLABORATORY

Emmanouil Papasavvas, PhD, Senior Staff Scientist,
The Wistar Institute,
3601 Spruce Street
Philadelphia, PA 19104
Email: manolisp@wistar.org

Emmanouil Papasavvas[1], Jessicamarie Morris[1], Brian Ross[1], Matthew Fair[1], Livio Azzoni[1], Karam Mounzer[2], Jay R. Kostman[3], Pablo Tebas[4], Luis J. Montaner[1]

[1]The Wistar Institute, Philadelphia, PA, USA; [2]Jonathan Lax Immune Disorders Treatment Center, Philadelphia Field Initiating Group for HIV-1 Trials, Philadelphia, PA, USA; [3]John Bell Health Center, Philadelphia Field Initiating Group for HIV-1 Trials, Philadelphia, PA, USA; [4]Department of Medicine, University of Pennsylvania, Philadelphia, PA, USA

Abstract

Background: Previous studies suggested that pegylated interferon α2b (peg-IFN-α2b) and the broadly neutralizing antibodies (bNAbs) 3BNC117 and 10-1074 may contribute to cure-related strategies. We evaluated the effect of a 26-week immunotherapy course with peg-IFN-α2b+bNAbs in the cytotoxic function and activation of natural killer (NK) cell subsets in persons with HIV infection (PWH) that participated in the BEAT 2 study (NCT03588715).

Methods: Fourteen PWH receiving suppressive antiretroviral therapy (ART, <50 HIV-1 copies/ml) underwent ART interruption (ATI) while receiving a 26-week immunotherapy course of peg-IFN-α2b+bNAbs. Peripheral blood mononuclear cells (PBMC) were collected prior to ATI/immunotherapy (ART alone, time-point 1), on ART+4 weeks peg-IFN-α2b (time-point 2), and on ATI+26 weeks peg-IFN-α2b+bNAbs (time-point 3). Fresh PBMC were used in ^{51}Cr release assays for assessment of antibody-dependent cell-mediated cytotoxicity (ADCC) against NK-resistant lymphoblastic target cell line prior and after in vitro stimulation with gp120, and for direct cytotoxicity against MHC-cell null cancer target cell line prior and after in vitro stimulation with IFN-α. Cryopreserved PBMC were used for immunophenotypic characterization by flow cytometry of NK cell subsets and of markers associated with NK activation, inhibition or maturation (e.g. CD38, NKp46, NKG2A, Siglec 7, Siglec 9, CD57). Statistics were performed by JMP 15.

Results: Immunotherapy did not affect IFN-α-induced NK direct cytotoxicity but resulted in a decrease in gp120-mediated ADDC. Reduced ADCC was observed together with an increase in the cytokine producing $CD56^{hi}$ and in $CD56^{lo/+}CD16^{-}$ % of $CD56^{+}$ NK cells, and a decrease in the cytotoxic $CD56^{lo/+}CD16^{+}$ % of $CD56^{+}$, suggesting that decrease in the expression of Fc receptor CD16 on NK could be associated with lower ADCC function. These findings were supported by a negative correlation between ADCC and $CD56^{lo/+}CD16^{-}$ % of lymphocytes after IFN-α immunotherapy (end of step 2). Finally, the gp120-induced ADCC decrease was observed together with a decrease in the maturation/cytotoxicity marker CD57 in $CD56^{lo/+}CD16^{+}$ NK cells, despite an increase in activation (CD38, NKp46) and inhibition (NKG2A, Siglec 7) markers.

Conclusion: In PWH, combined immunotherapy with peg-IFN-α2b+bNAbs resulted in no effect on IFN-α-induced NK direct cytotoxicity and an unexpected decrease in gp120-induced ADCC and in circulating $CD16^{+}$ NK cell subsets.

Introduction

In our previous studies NCT00594880 **(1-3)** and NCT01935089 **(4)** we showed that pegylated interferon (peg-IFN-α2a and peg-IFN-α2b respectively) can achieve sustained virological control in patients with HIV during antiretroviral therapy interruption (ATI). Pillar-Mendoza et al. **(5)** have demonstrated the effectiveness of combining two broadly neutralizing antibodies (bNabs), 3BNC117 and 10-1074, in maintaining viral suppression during analytical treatment interruption in participants with sensitive virus at baseline. These data suggest that pegylated interferon and the bNAbs 3BNC117 and 10-1074 may contribute to cure-related strategies. We conducted the NCT03588715 study to test whether the combination of bNabs and peg-IFN-α2b, when administered in together with antiretroviral therapy, would have a significant impact on achieving durable suppression off ART together with a reduction of the HIV reservoir. Here we present the results of the evaluation of the effect of a 26-week immunotherapy course with peg-IFN-α2b+bNAbs in the cytotoxic function and activation of natural killer (NK) cell subsets in persons with HIV infection (PWH) that participated in this study.

Subjects, Materials & Methods

Study participants

A total of 14 PWH participated in the study. All participants were adults living with HIV and well-controlled viral replication with antiretroviral therapy (ART) for more than 6 months, with a current $CD4^{+}$ T cell count ≥450 cells/mm^3. Participants underwent ART interruption (ATI) while receiving a 26-week immunotherapy course of peg-IFN-α2b+bNAbs.

Methods

Peripheral blood mononuclear cells (PBMC) were collected prior to ATI/immunotherapy (ART alone, time-point 1), on ART+4 weeks peg-IFN-α2b (time-point 2), and on ATI+26 weeks peg-IFN-α2b+bNAbs (time-point 3) (**Figure 1**). Assays performed:

Antibody-dependent cell-mediated cytotoxicity (ADCC)

Fresh PBMC were used in ^{51}Cr release assays for ADCC against NK-resistant lymphoblastic target cell line prior and after in vitro stimulation with gp120. Percent lysis was determined by the following formula: [(experimental counts-spontaneous released counts)/(total counts-spontaneous released counts)] x 100. Results were expressed as area under the curve (AUC) for effector to target (E:T) ratios of 50:1, 25:1, 12.5:1 and 6.25:1 for both constitutive and gp120-induced ADDC. Data were normalized by number of NK.

Direct cytotoxicity

Fresh PBMC were used in ^{51}Cr release assays for direct cytotoxicity against MHC-cell null cancer target cell line prior and after in vitro stimulation with IFN-α. Percent lysis was determined by the following formula: [(experimental counts-spontaneous released counts)/(total counts-spontaneous released counts)] x 100. Results were expressed as area under the curve (AUC) for effector to target (E:T) ratios of 50:1, 25:1, 12.5:1 and 6.25:1 for both constitutive and in vitro IFN-α-induced NK function. Data were normalized by number of NK.

Immunophenotypic characterization by flow cytometry

Cryopreserved PBMC were used for immunophenotypic characterization by flow cytometry of NK cell subsets and of markers associated with NK activation, inhibition or maturation (e.g. CD38, NKp46, NKG2A, Siglec 7, Siglec 9, CD57). Results were expressed as mean fluorescent intensity (MFI), and percent (%) positive.

Statistical analysis

Data were described as medians, 25th and 75th percentiles. Variable distributions were analyzed for normality using the Shapiro-Wilk W test (p>0.05). Depending on data distribution, between time points comparisons were performed using non-parametric Wilcoxon Sign-Rank test or paired t-tests. P-values <0.05 were considered statistically significant. All statistics were performed with JMP Pro15 (SAS Institute, Cary, NC).

1 Study schema

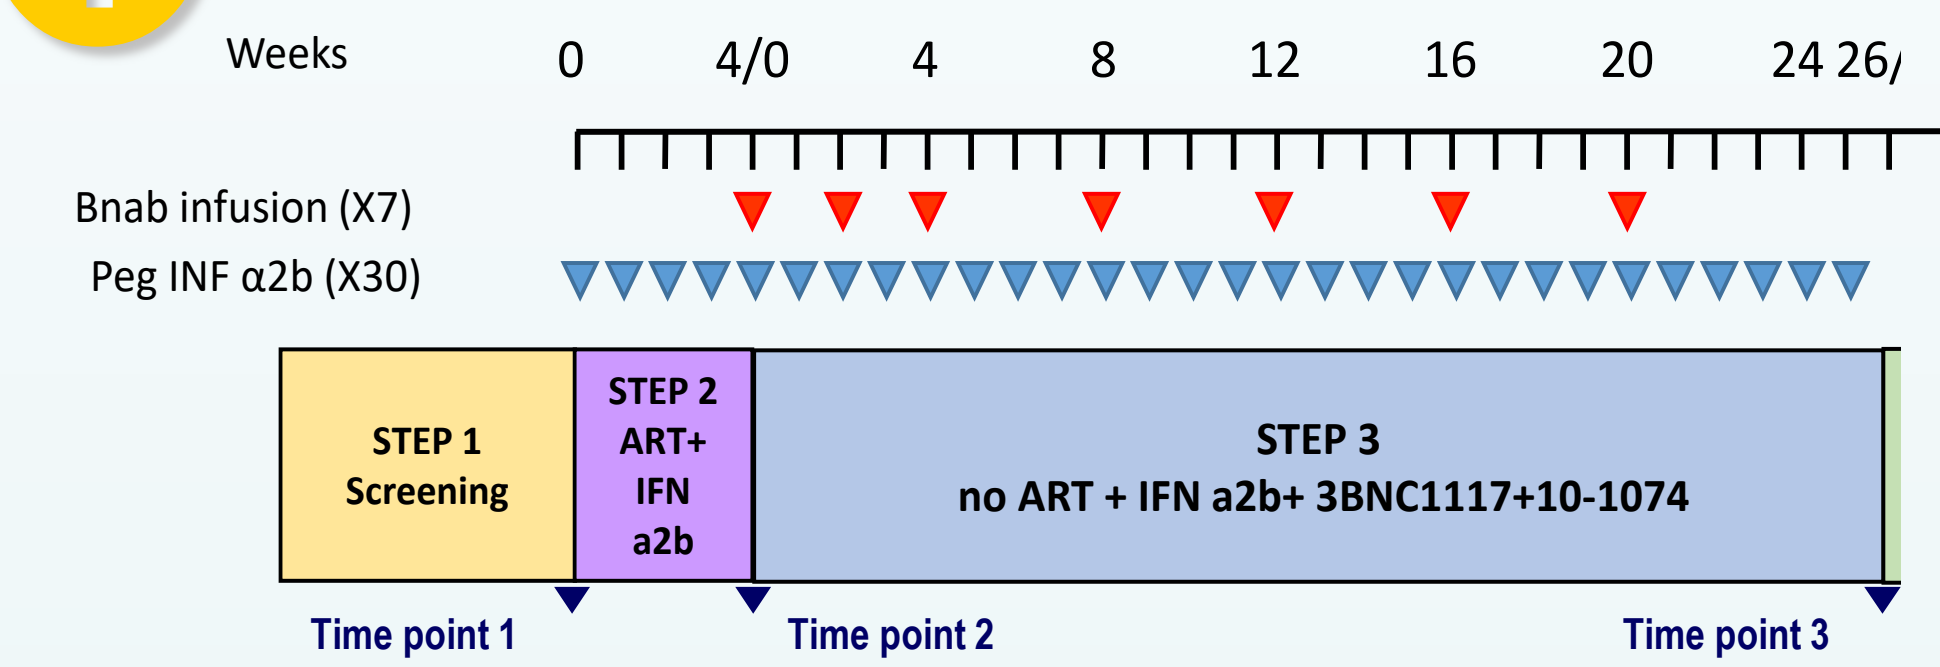

Weeks
0
4/0
4
8
12
16
20
24 26
Bnab infusion (X7)
Peg INF α2b (X30)
STEP 1 Screening
STEP 2 ART+ IFN a2b
STEP 3 no ART + IFN a2b+ 3BNC1117+10-1074
Time point 1
Time point 2
Time point 3

2 No effect on intact HIV proviruses

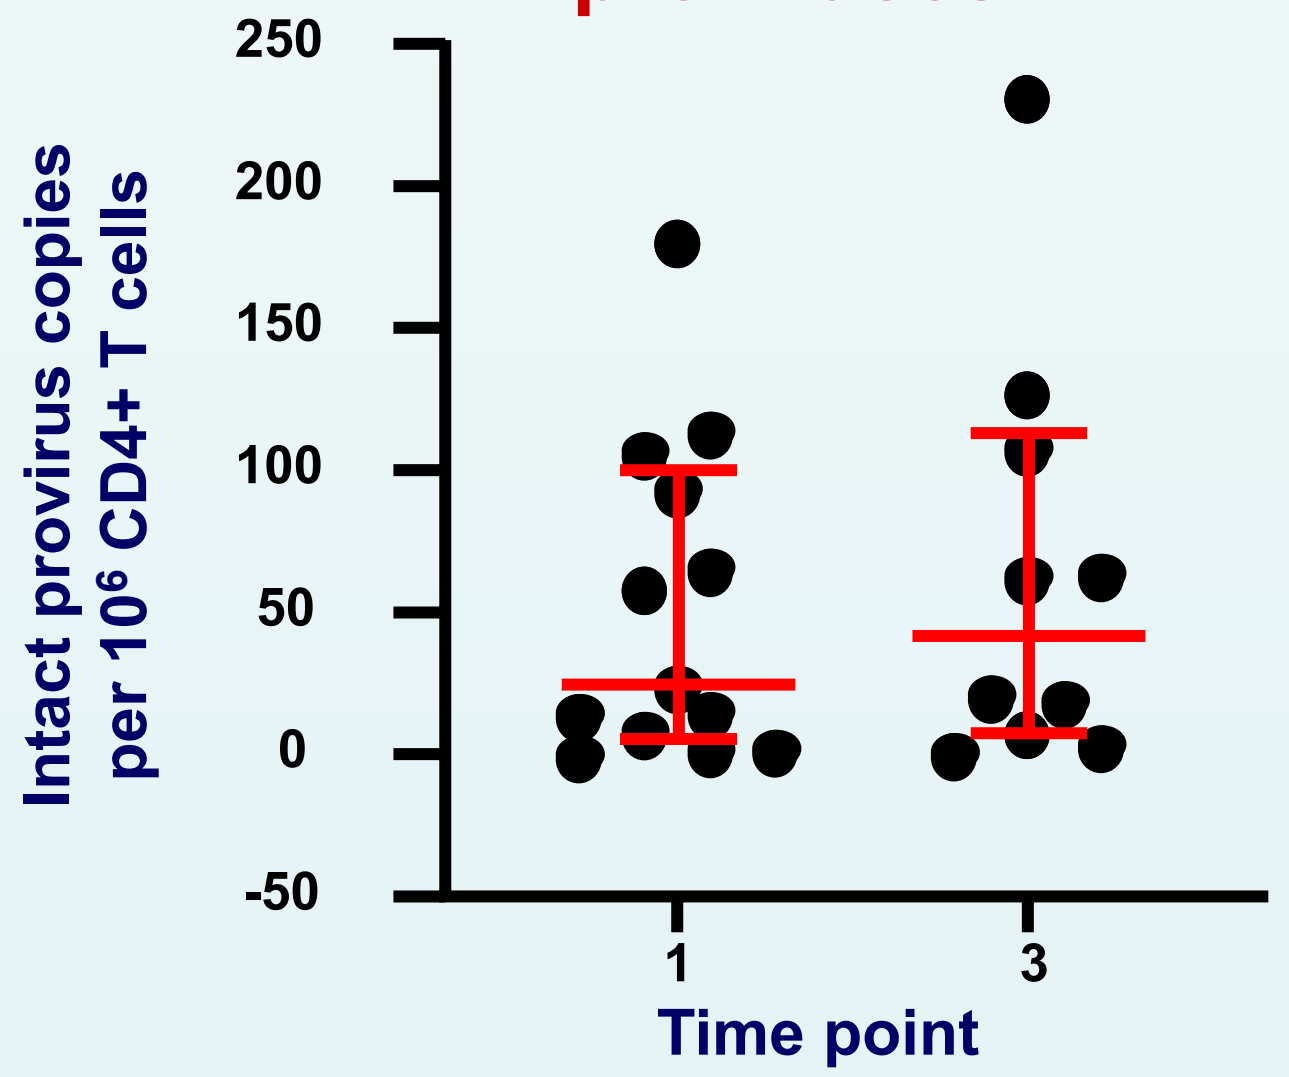

Intact provirus copies per 10⁶ CD4+ T cells
250
200
150
100
50
0
-50
1
3
Time point

3 Decrease in NK of maturation/cytotoxicity markers in contrast to increase in activation markers

Variable	Function	Time point 1 Median (25-75 IQR)	Time point 3 Median (25-75 IQR)	P
CD57+/CD56lo/CD16+ % of CD56lo/CD16+	Maturation/cytotoxicity marker	77.98 (70.55-85.05)	61.49 (48.2-72.6)	0.0098
CD38+/CD56lo/CD16+ % of CD56lo/CD16+	Activation marker	71.73 (64.59-87.47)	97.66 (95.7-99.4)	0.0039
MFI of CD38 on CD56lo/CD16+	Activation marker	1817 (1322-3732.5)	10012.5 (6992-12109.25)	0.0195
NKp46+/CD56lo/CD16+ % of CD56lo/CD16+	Activation marker	79.97 (64.27-85.75)	91.61 (82.77-95.39)	0.0371
MFI of NKp46 on CD56lo/CD16+	Activation marker	935 (579.75-1082.25)	1535.5 (1191.75-2478)	0.0098
NKG2A+/CD56lo/CD16+ % of CD56lo/CD16+	Inhibition marker	28.67 (15.38-45.22)	44.36 (37.87-62.11)	0.0059
MFI of NKG2A on CD56lo/CD16+	Inhibition marker	117.5 (89.47-179.5)	158 (143-230.75)	0.0137
MFI of Siglec7 on CD56lo/CD16+	Inhibition marker	3736.5 (1868.25-6089.25)	3058.5 (1584.75-3912)	0.0371
Siglec9+/CD56lo/CD16+ % of CD56lo/CD16+	Inhibition marker	31.84 (26.11-43.27)	14.33 (6.42-22.29)	0.002
MFI of Siglec9 on CD56lo/CD16+	Inhibition marker	131.5 (102.17-196.5)	58.4 (50.42-77.52)	0.002

4 No effect on total NK in contrast to a decrease in $CD16^{+}$ NK cell subsets

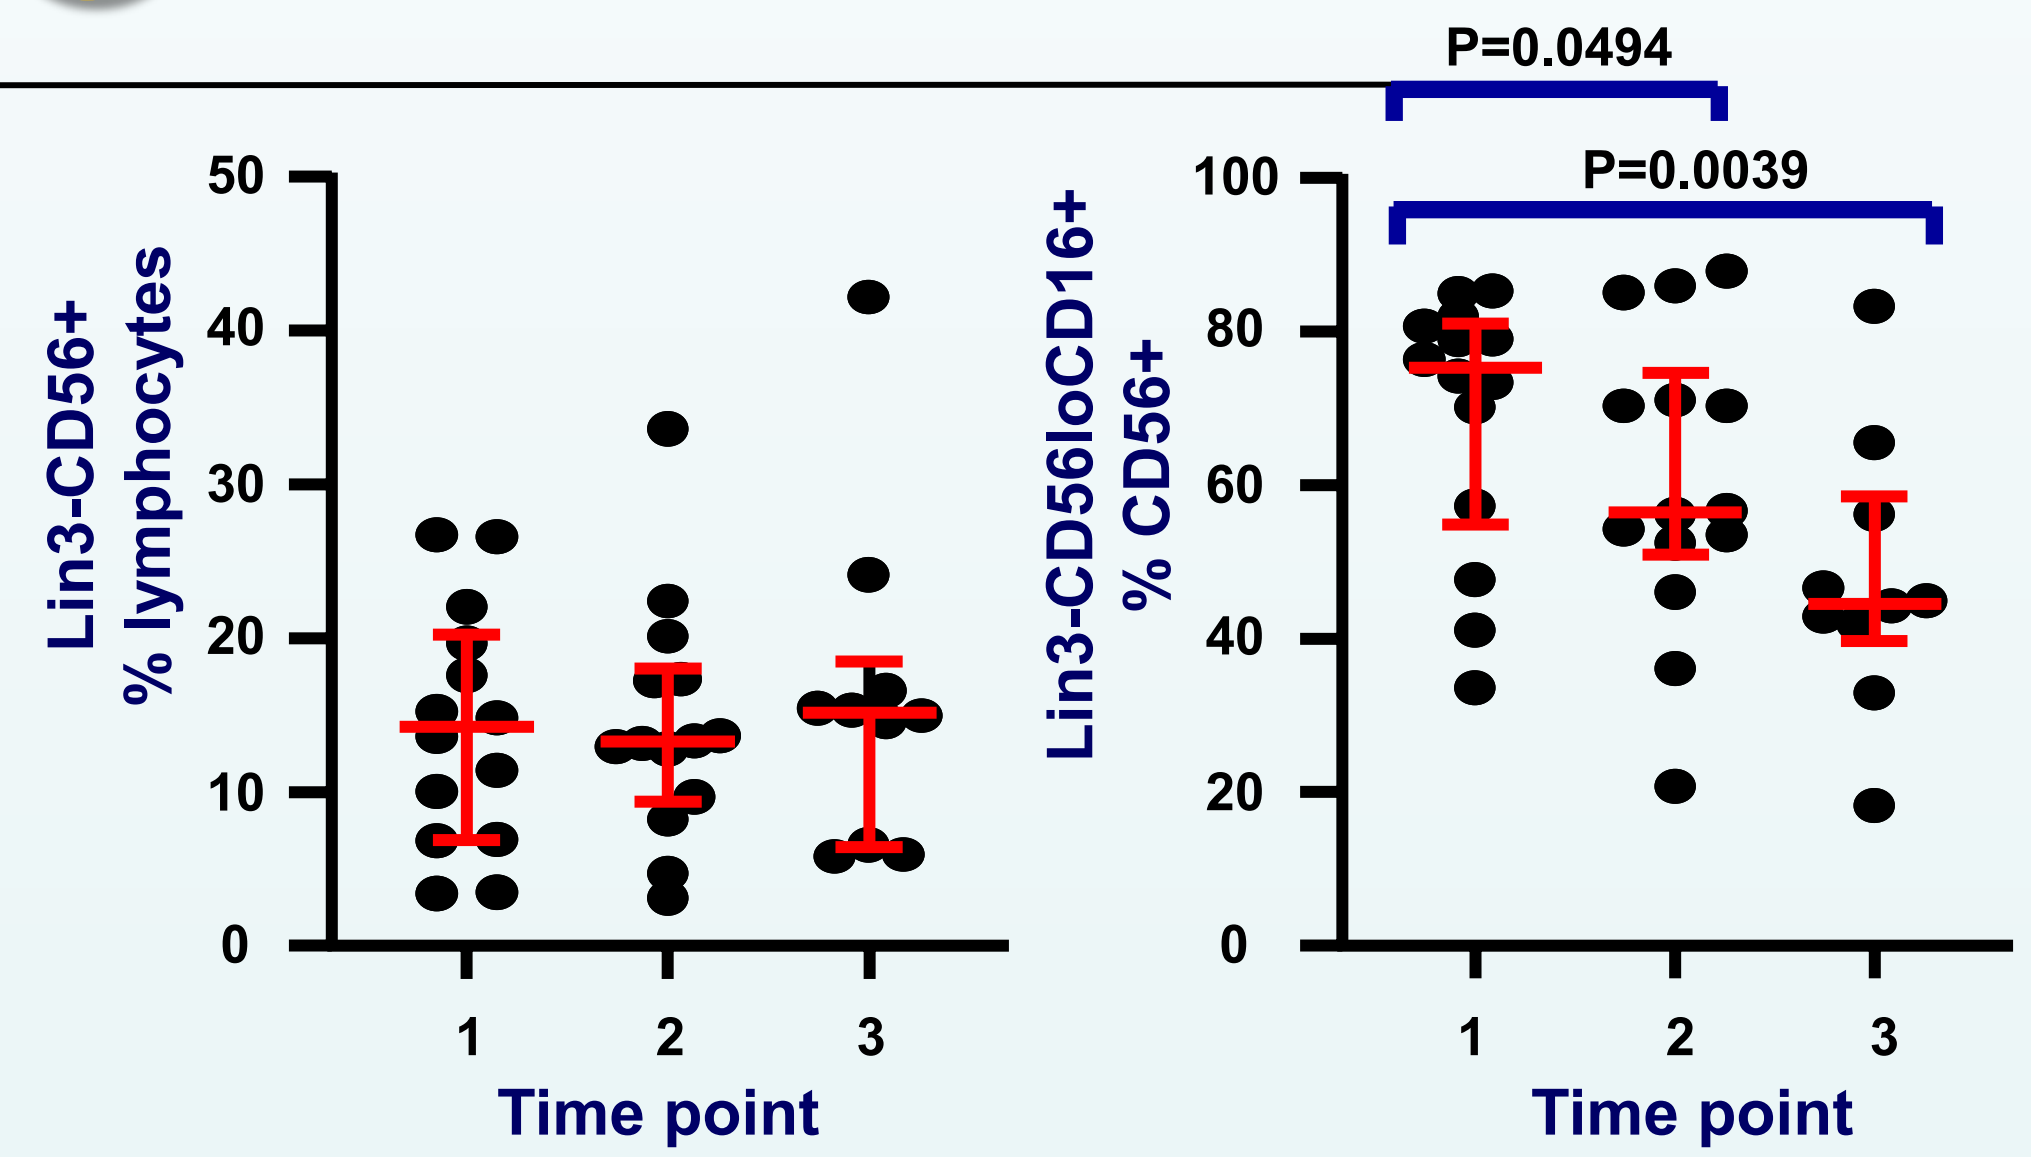

Lin3-CD56+ % lymphocytes
50
40
30
20
10
0
1
2
3
Time point
P=0.0494
P=0.0039
Lin3-CD56loCD16+ % CD56+
100
80
60
40
20
0
1
2
3
Time point

5 No effect on IFN-α-induced NK direct cytotoxicity in contrast to a decrease in gp120-induced ADCC

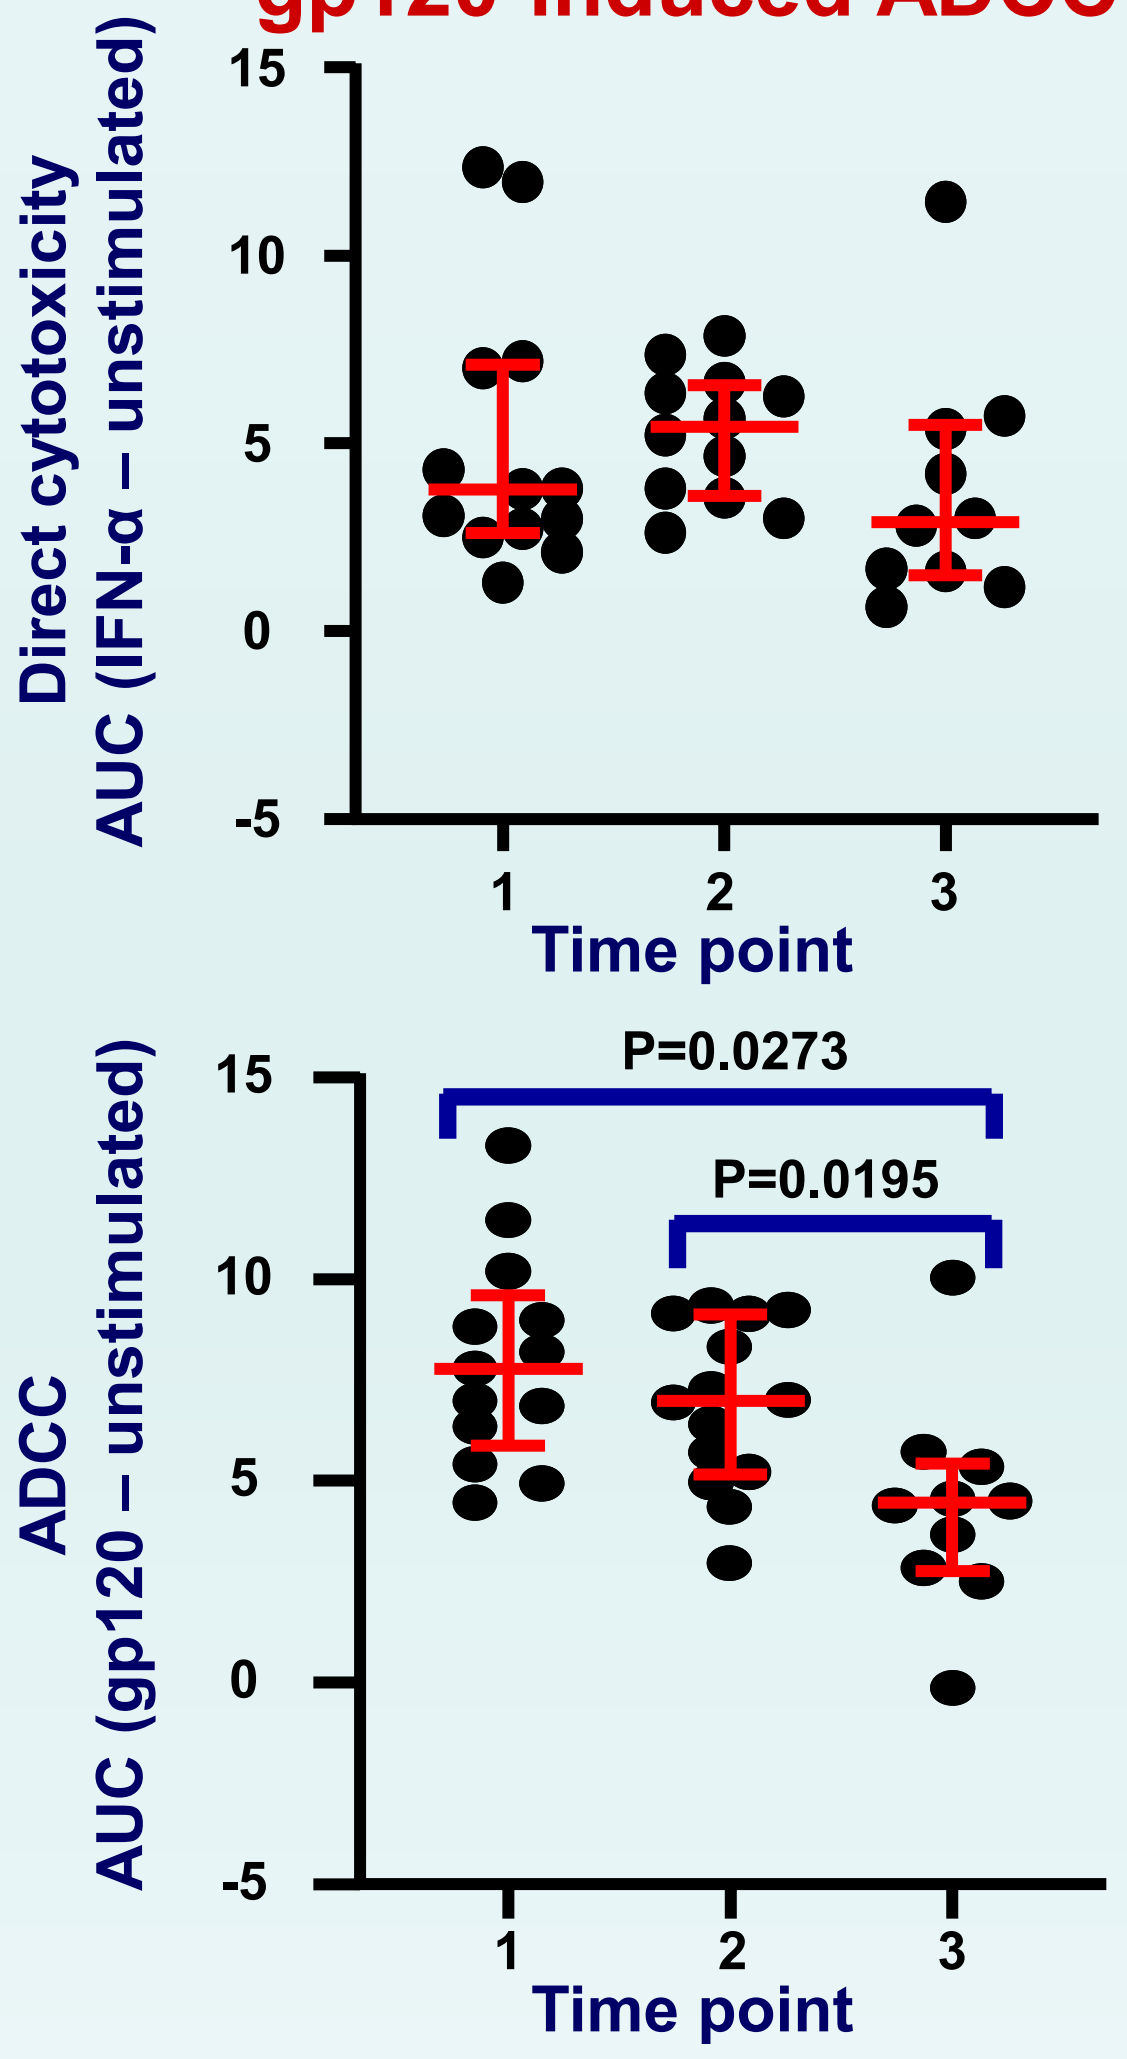

Direct cytotoxicity AUC (IFN-α – unstimulated)
15
10
5
0
-5
1
2
3
Time point
P=0.0273
P=0.0195
ADCC AUC (gp120 – unstimulated)
15
10
5
0
-5
1
2
3
Time point

Conclusions

In PWH, combined immunotherapy with peg-IFN-α2b+bNAbs on ART resulted in:

- No effect in HIV proviruses levels
- Increase in NK activation
- Decrease in maturation/cytotoxicity markers
- Reduction of NK functionality as shown by:
 - the decrease in circulating $CD16^{+}$ NK cell subsets
 - the decrease in gp120-induced ADCC
 - the lack of effect on IFN-α-induced NK direct cytotoxicity

Interpretation

The cause for the reduced NK functionality was clearly associated with the presence of immunotherapy before bNAb infusions. It remains to be determined if other preclinical immunotherapy strategies administered on ART and associated with IFN-α induction could also result in a reductions of NK ADCC function.

References

1. Azzoni L, Foulkes AS, Papasavvas E, et al. J Infect Dis 2013;208(2):363.
2. Tomescu C, Tebas P, Montaner LJ. AIDS 2017;31(5):613-622.
3. Papasavvas E, Azzoni L, Ross BN, et al. AIDS 2021;35(12):2051-2054.
4. Papasavvas E, Azzoni L, Pagliuzza A, et al. AIDS Res Hum Retroviruses 2021;37(6):433-443.
5. Mendoza P, Gruell H, Nogueira L, et al. Nature 2018;561(7724):479-484.

Acknowledgements

We would like to thank the study participants and their providers.

Support:

NIAID grant BEAT-HIV Delaney Collaboratory UM1AI126620, co-funded by NIAID, NIMH, NINDS, and NIDA; Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA; Philadelphia FIGHT; The Philadelphia Foundation (Robert I. Jacobs Fund); the Penn Center for AIDS Research (P30 AI 045008); Dr. Luis J. Montaner is supported by the Herbert Kean, M.D., Family Professorship