433

A Placebo-Controlled Randomized Trial of the HTI Immunogen Vaccine and Vesatolimod

Beatriz Mothe,[1*] Adrià Curran,[2] Juan Carlos López Bernardo de Quirós,[3] Julen Cadiñanos,[4] Ignacio de los Santos,[5] Juan Ambrosioni,[6] Arkaitz Imaz,[7] Santiago Moreno,[8] Pere Domingo,[9] Yanhui Cai,[10] Romas Geleziunas,[10] Devi SenGupta,[10] Ian McGowan,[11] Christian Brander,[1,11,12] Jose Ramon Arribas,[4] for the AELIX-003 Research Group

[1]Infectious Diseases Department, IrsiCaixa AIDS Research Institute, HUGTiP, CIBERINFEC, Barcelona, Spain; [2]Hospital Universitari Vall d'Hebron, Vall d'Hebron Institut de Recerca, Barcelona; [3]Hospital General Universitario Gregorio Marañón, Madrid, Spain; [4]Hospital Universitario La Paz, CIBERINFEC, Madrid; [5]Hospital Universitario de La Princesa, CIBERINFEC, Madrid; [6]Hospital Clínic de Barcelona; [7]Institut d'Investigació Biomèdica de Bellvitge (IDIBELL), Hospital Universitari de Bellvitge, L'Hospitalet de Llobregat, Barcelona; [8]Hospital Ramón y Cajal, Madrid; [9]Hospital de Sant Pau, Barcelona; [10]Gilead Sciences, Inc, Foster City, CA; [11]AELIX Therapeutics SL, Barcelona; [12]ICREA, Barcelona

*Presenting author.

AELIX Therapeutics SL
Parc Científic de Barcelona
Carrer Baldiri Reixac 4-8
08028 Barcelona, Spain
+34 93 403 13 39
info@aelixtherapeutics.com

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Background

- Therapeutic vaccines designed to enhance HIV-specific T-cell immunity have been postulated to be a key component of any HIV cure strategy
- HIVACAT T-cell immunogen (HTI) is a novel HIV vaccine immunogen designed to elicit cellular immune responses to HIV regions within group-specific antigen (Gag), polymerase (Pol), viral infectivity factor (Vif), and negative regulatory factor (Nef) proteins that are associated with viral control in humans[1]
- In people with HIV (PWH) treated early with antiretroviral therapy (ART), the HIV-specific T cell response induced by a combination of DNA.HTI (D), MVA.HTI (M), and ChAdOx1.HTI (C) vaccines was associated with longer time off ART during analytical treatment interruption (ATI)[2]
- Vesatolimod (VES) is a toll-like receptor 7 (TLR7) agonist under investigation as part of an HIV cure strategy; TLR7 agonists activate immune responses and potentially lead to improved HIV control in combination with other agents[3-5]
- We conducted a Phase 2a study (AELIX-003 [NCT04364035]) of ChAdOx1.HTI and MVA.HTI vaccines (referenced collectively as CCMM) with VES in adults who received ART within the first 6 months of HIV acquisition

Objectives

- To evaluate the safety, immunogenicity, and efficacy of CCMM in combination with the TLR7 agonist VES in early-treated PWH

Methods

Figure 1. AELIX-003 Study Design

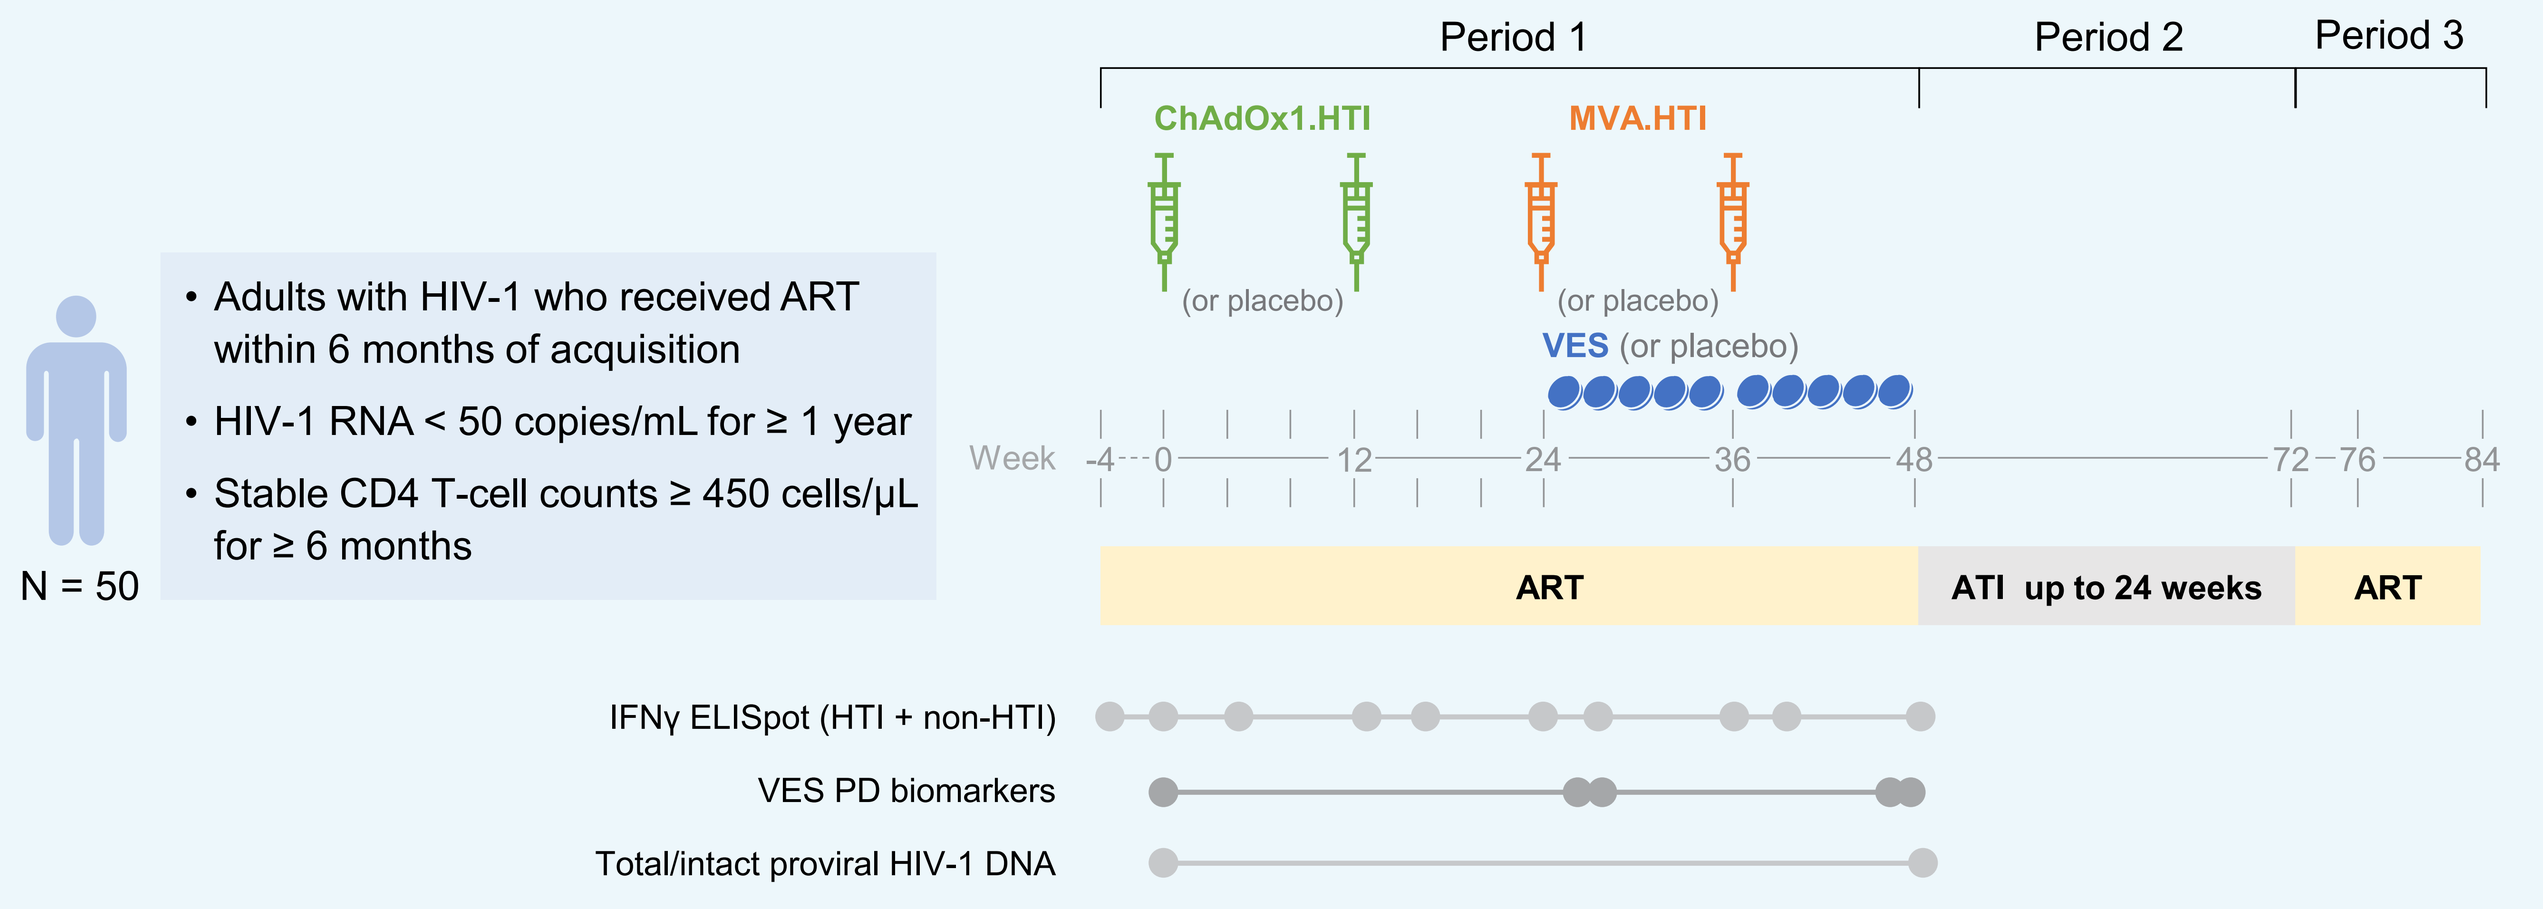

Period 1
Period 2
Period 3
ChAdOx1.HTI
MVA.HTI
(or placebo)
(or placebo)
VES (or placebo)
• Adults with HIV-1 who received ART within 6 months of acquisition
• HIV-1 RNA < 50 copies/mL for ≥ 1 year
• Stable CD4 T-cell counts ≥ 450 cells/μL for ≥ 6 months
N = 50
Week -4 0 12 24 36 48 72 76 84
ART
ATI up to 24 weeks
ART
IFNγ ELISpot (HTI + non-HTI)
VES PD biomarkers
Total/intact proviral HIV-1 DNA

ELISpot = enzyme-linked immunosorbent spot; HLA = human leukocyte antigen; IFNγ = interferon-γ; PD = pharmacodynamics.

- Double-blind, randomized (2:1) trial conducted at 9 sites in Spain **(Figure 1)**
- Participants were randomized 2:1 to receive CCMM and oral VES 6 mg or placebo while continuing ART
 - Criteria for ART resumption during ATI: plasma viral load (pVL) > 100,000 copies/mL, pVL > 10,000 copies/mL for 8 consecutive weeks, or CD4 < 350 cells/μL
- Randomization was stratified based on HLA type with potential for superior viral control (HLA-B27 [any subtype], HLA-B57 [any subtype], HLA-B58:01, HLA-B15:16, and/or HLA-B15:17)

Endpoints

Primary: proportion of participants developing solicited Grade 3 or 4 local or systemics reactions in the 7 days after receiving CCMM + VES, or developing treatment-emergent serious adverse events during Period 1

Secondary and Exploratory:

- Proportion of participants with T-cell responses, including breadth and magnitude, to HTI-encoded regions as determined by IFNγ ELISpot (Period 1)
- Changes from baseline (BL) in serum/plasma cytokines, gene expression (including IFN-stimulated genes), and immune cell phenotype/activation in peripheral blood
- Change from BL in peripheral viral reservoir at Week 48
- Proportion of participants with pVL < 50 or < 2000 copies/mL at 12 and 24 weeks after ATI start; proportion who remain off ART at 12 and 24 weeks after start of ATI; time to pVL ≥ 50 or ≥ 10,000 copies/mL during ATI; and time to ART resumption (Period 2)

Results

Study Participant Disposition, Demographics, and Baseline Characteristics (Table 1)

- A total of 50 participants were randomized; 47 (placebo: n = 17; CCMM + VES: n = 30) entered the ATI phase and restarted ART
- Of these 47 participants, 46 (placebo: n = 17; CCMM + VES: n = 30) completed the study
 - 1 in the placebo group discontinued (personal decision)

Table 1. Baseline Characteristics

Category		Placebo n = 17	CCMM + VES n = 33	Total N = 50
Age, years		37 (21-59)	38 (24-55)	36 (21-59)
Male at birth, n (%)		17 (100)	33 (100)	50 (100)
BMI, kg/m²		23.7 (19-33)	23 (16-44)	23.3 (16-44)
Time from HIV transmission to ART initiation, days		86 (16-167)	61 (7-170)	67 (7-170)
Fiebig stage at ART initiation, n (%)[6]	I	0	2 (6.1)	2 (4.0)
	II	1 (5.9)	3 (9.1)	4 (8.0)
	III	0	3 (9.1)	3 (6.0)
	IV	3 (17.6)	3 (9.1)	6 (12)
	V	6 (35.3)	12 (36.4)	18 (36.0)
	VI	5 (29.4)	7 (21.2)	12 (24.0)
	Missing	2 (11.8)	3 (9.1)	5 (10.0)
Pre-ART pVL, log copies/mL		4.9 (1.9-7)	5.2 (2.5-7)	5.2 (1.9-7)
Current ART, n (%)	BIC/TAF/FTC	10 (58.8)	15 (45.5)	25 (50.0)
	DTG/ABC/3TC	5 (29.4)	13 (39.4)	18 (36.0)
	DTG + TDF or TAF/FTC	1 (5.9)	5 (15.1)	6 (12)
	RPV/TAF/FTC	1 (5.9)	0	1 (2)
Time on ART, months		43 (17-116)	41 (16-132)	42 (16-132)
Absolute CD4, cells/μL		831 (534-1,333)	882 (451-1,600)	872 (451-1,600)
CD4/CD8 ratio		1.2 (1-2)	1.2 (1-2)	1.2 (1-2)
Beneficial HLA alleles, n (%)[a]		4 (23.5)	6 (18.2)	10 (20)

Data are median (minimum-maximum) except where specified. [a]HLA-B27 (any subtype), HLA-B57 (any subtype), HLA-B58:01, and HLA-B15:16 and/or HLA-B15:17. 3TC = lamivudine; ABC = abacavir; BIC = bictegravir; BMI = body mass index; DTG = dolutegravir; FTC = emtricitabine; ITT = intention to treat; RPV = rilpivirine; TAF = tenofovir alafenamide fumarate; TDF = tenofovir disoproxil fumarate.

Safety of CCMM + VES 6 mg x 10 Doses

- 1 participant experienced a serious adverse event (acute cholangitis) at Week 46 that was considered by the investigator to be unrelated to study drugs
- There were no cases of cytokine-release syndrome
- Safety of the ATI:
 - There were no cases of clinically relevant acute retroviral syndrome prompting early ART resumption
 - 8 participants had mild COVID-19 infection, which did not trigger early ART resumption
 - 1 participant resumed ART at Week 18 of ATI due to an acute hepatitis B infection
- All participants achieved pVL < 50 copies/mL after ART resumption

HTI Vaccine Immunogenicity (Preliminary Data: n = 13 placebo and n = 21 CCMM + VES; Figure 2)

- Participants receiving CCMM + VES showed broad and strong HTI-specific responses
- At ATI, close to 50% of all HIV T cells were specific to HTI targets, with no specific patterns of immunodominance across the HIV subproteins covered by the HTI immunogen

Figure 2. HTI Immunogenicity Profile in Placebo and CCMM + VES Recipients

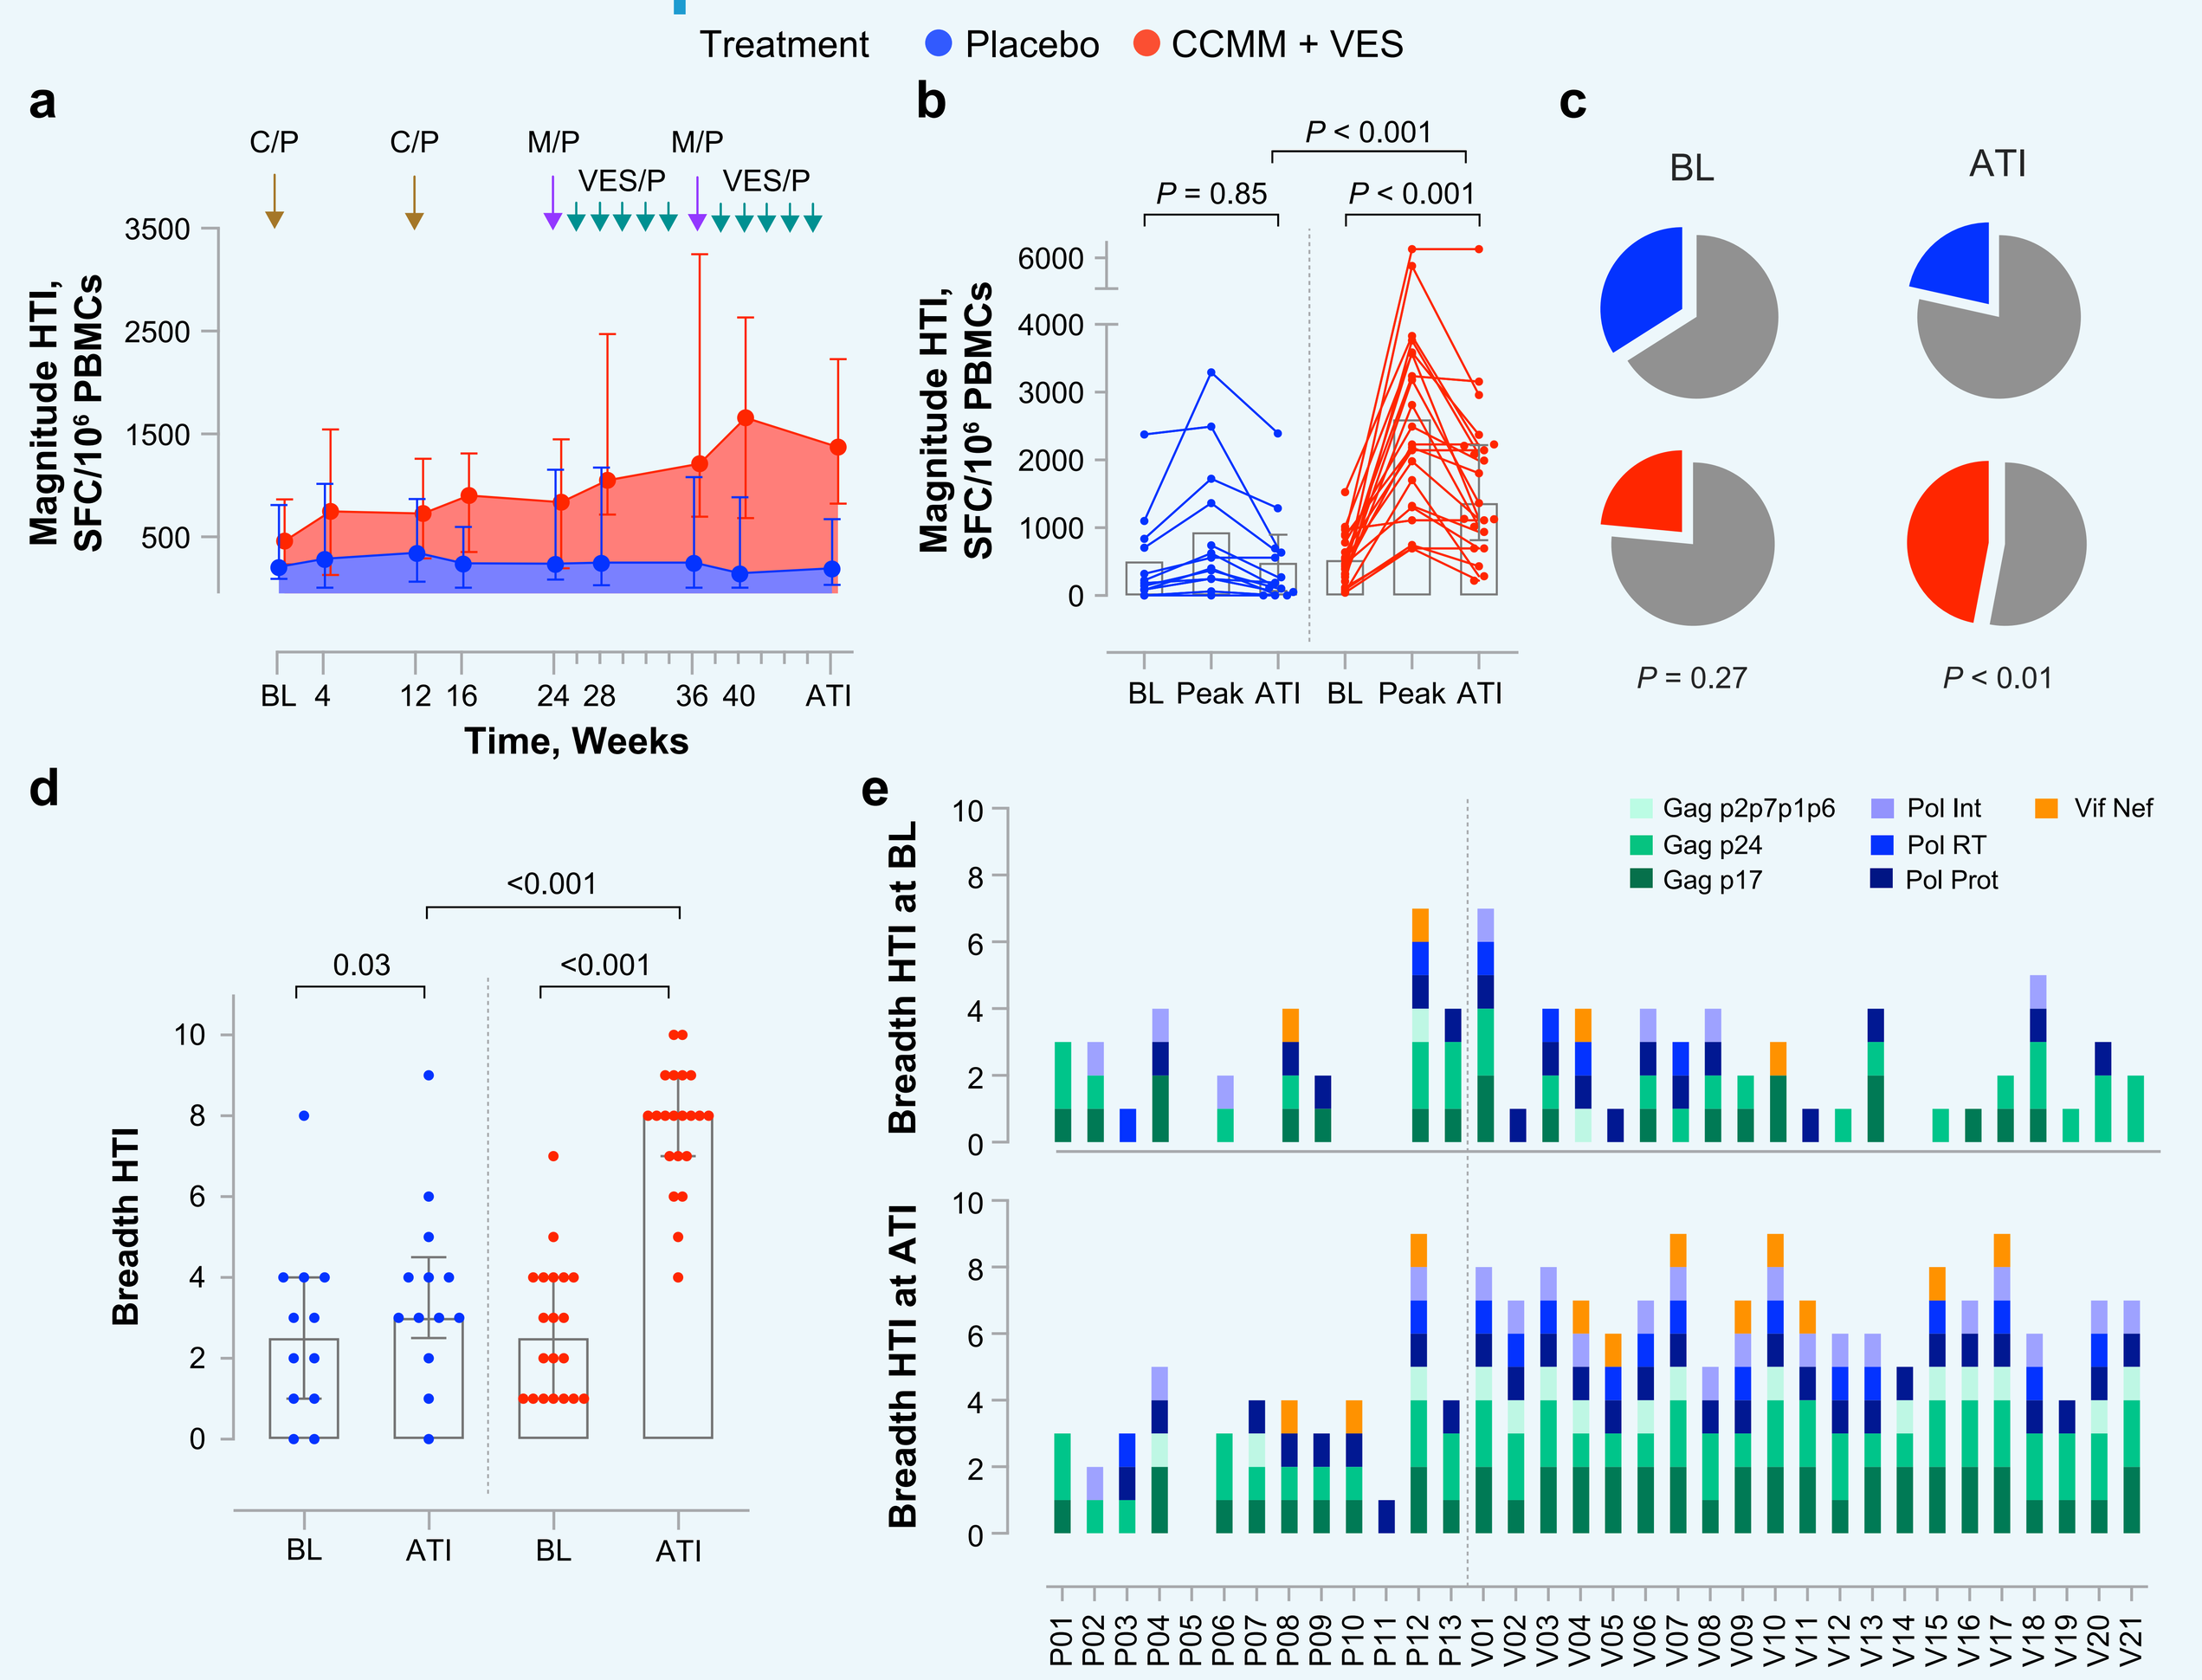

Treatment ● Placebo ● CCMM + VES
a
C/P C/P M/P M/P VES/P VES/P
Magnitude HTI, SFC/10⁶ PBMCs
Time, Weeks
b
P = 0.85
P < 0.001
P < 0.001
BL Peak ATI BL Peak ATI
c
BL ATI
P = 0.27
P < 0.01
d
Breadth HTI
0.03
<0.001
<0.001
BL ATI BL ATI
e
Breadth HTI at BL
Breadth HTI at ATI
Gag p2p7p1p6
Gag p24
Gag p17
Pol Int
Pol RT
Pol Prot
Vif Nef

Total HIV-1- and HTI-specific T cells were assessed by IFN-γ-detecting ELISpot assay, using 10 peptide pools covering HTI. **(a)** Median (interquartile range [IQR]) magnitude of response (sum of spot-forming cells [SFCs]/10⁶ peripheral blood mononuclear cells (PBMCs) for 10 HTI pools). **(b)** Individual and median magnitudes of response (sum of SFCs/10⁶ PBMCs for HTI pool) for placebo (blue) and CCMM + VES (red) recipients at BL, peak immunogenicity time point, and Week 48 (ATI start). **(c)** Average distribution of total HIV-1 T cells according to specificity. HTI-specific responses for placebo *(blue)* and CCMM + VES *(red)* recipients and non-HTI HIV-1 specific responses *(gray)* are shown. **(d)** Cumulative breadth (median and IQR) of vaccine-elicited responses. **(e)** Cumulative distribution of HTI-specific responses within the different HIV-1 subproteins at BL and start of ATI for each placebo (P01-P13) and CCMM + VES (V01-V21) recipient. Mann-Whitney and Wilcoxon test was used to compare between groups and between 2 different time points within same participant, respectively. Int = integrase; Prot = protease; RT = reverse transcriptase.

VES Pharmacodynamic Biomarkers (Figure 3)

- VES consistently increased the production of acute phase cytokines (IFNα and interleukin-1 receptor antagonist [IL-1RA]) and chemokines (IFNγ-inducible protein-10 [IP-10] and IFN-inducible T-cell-α chemoattractant [ITAC]) 24 hours postdosing in CCMM + VES recipients

Figure 3. PD of VES in Placebo and CCMM + VES Recipients

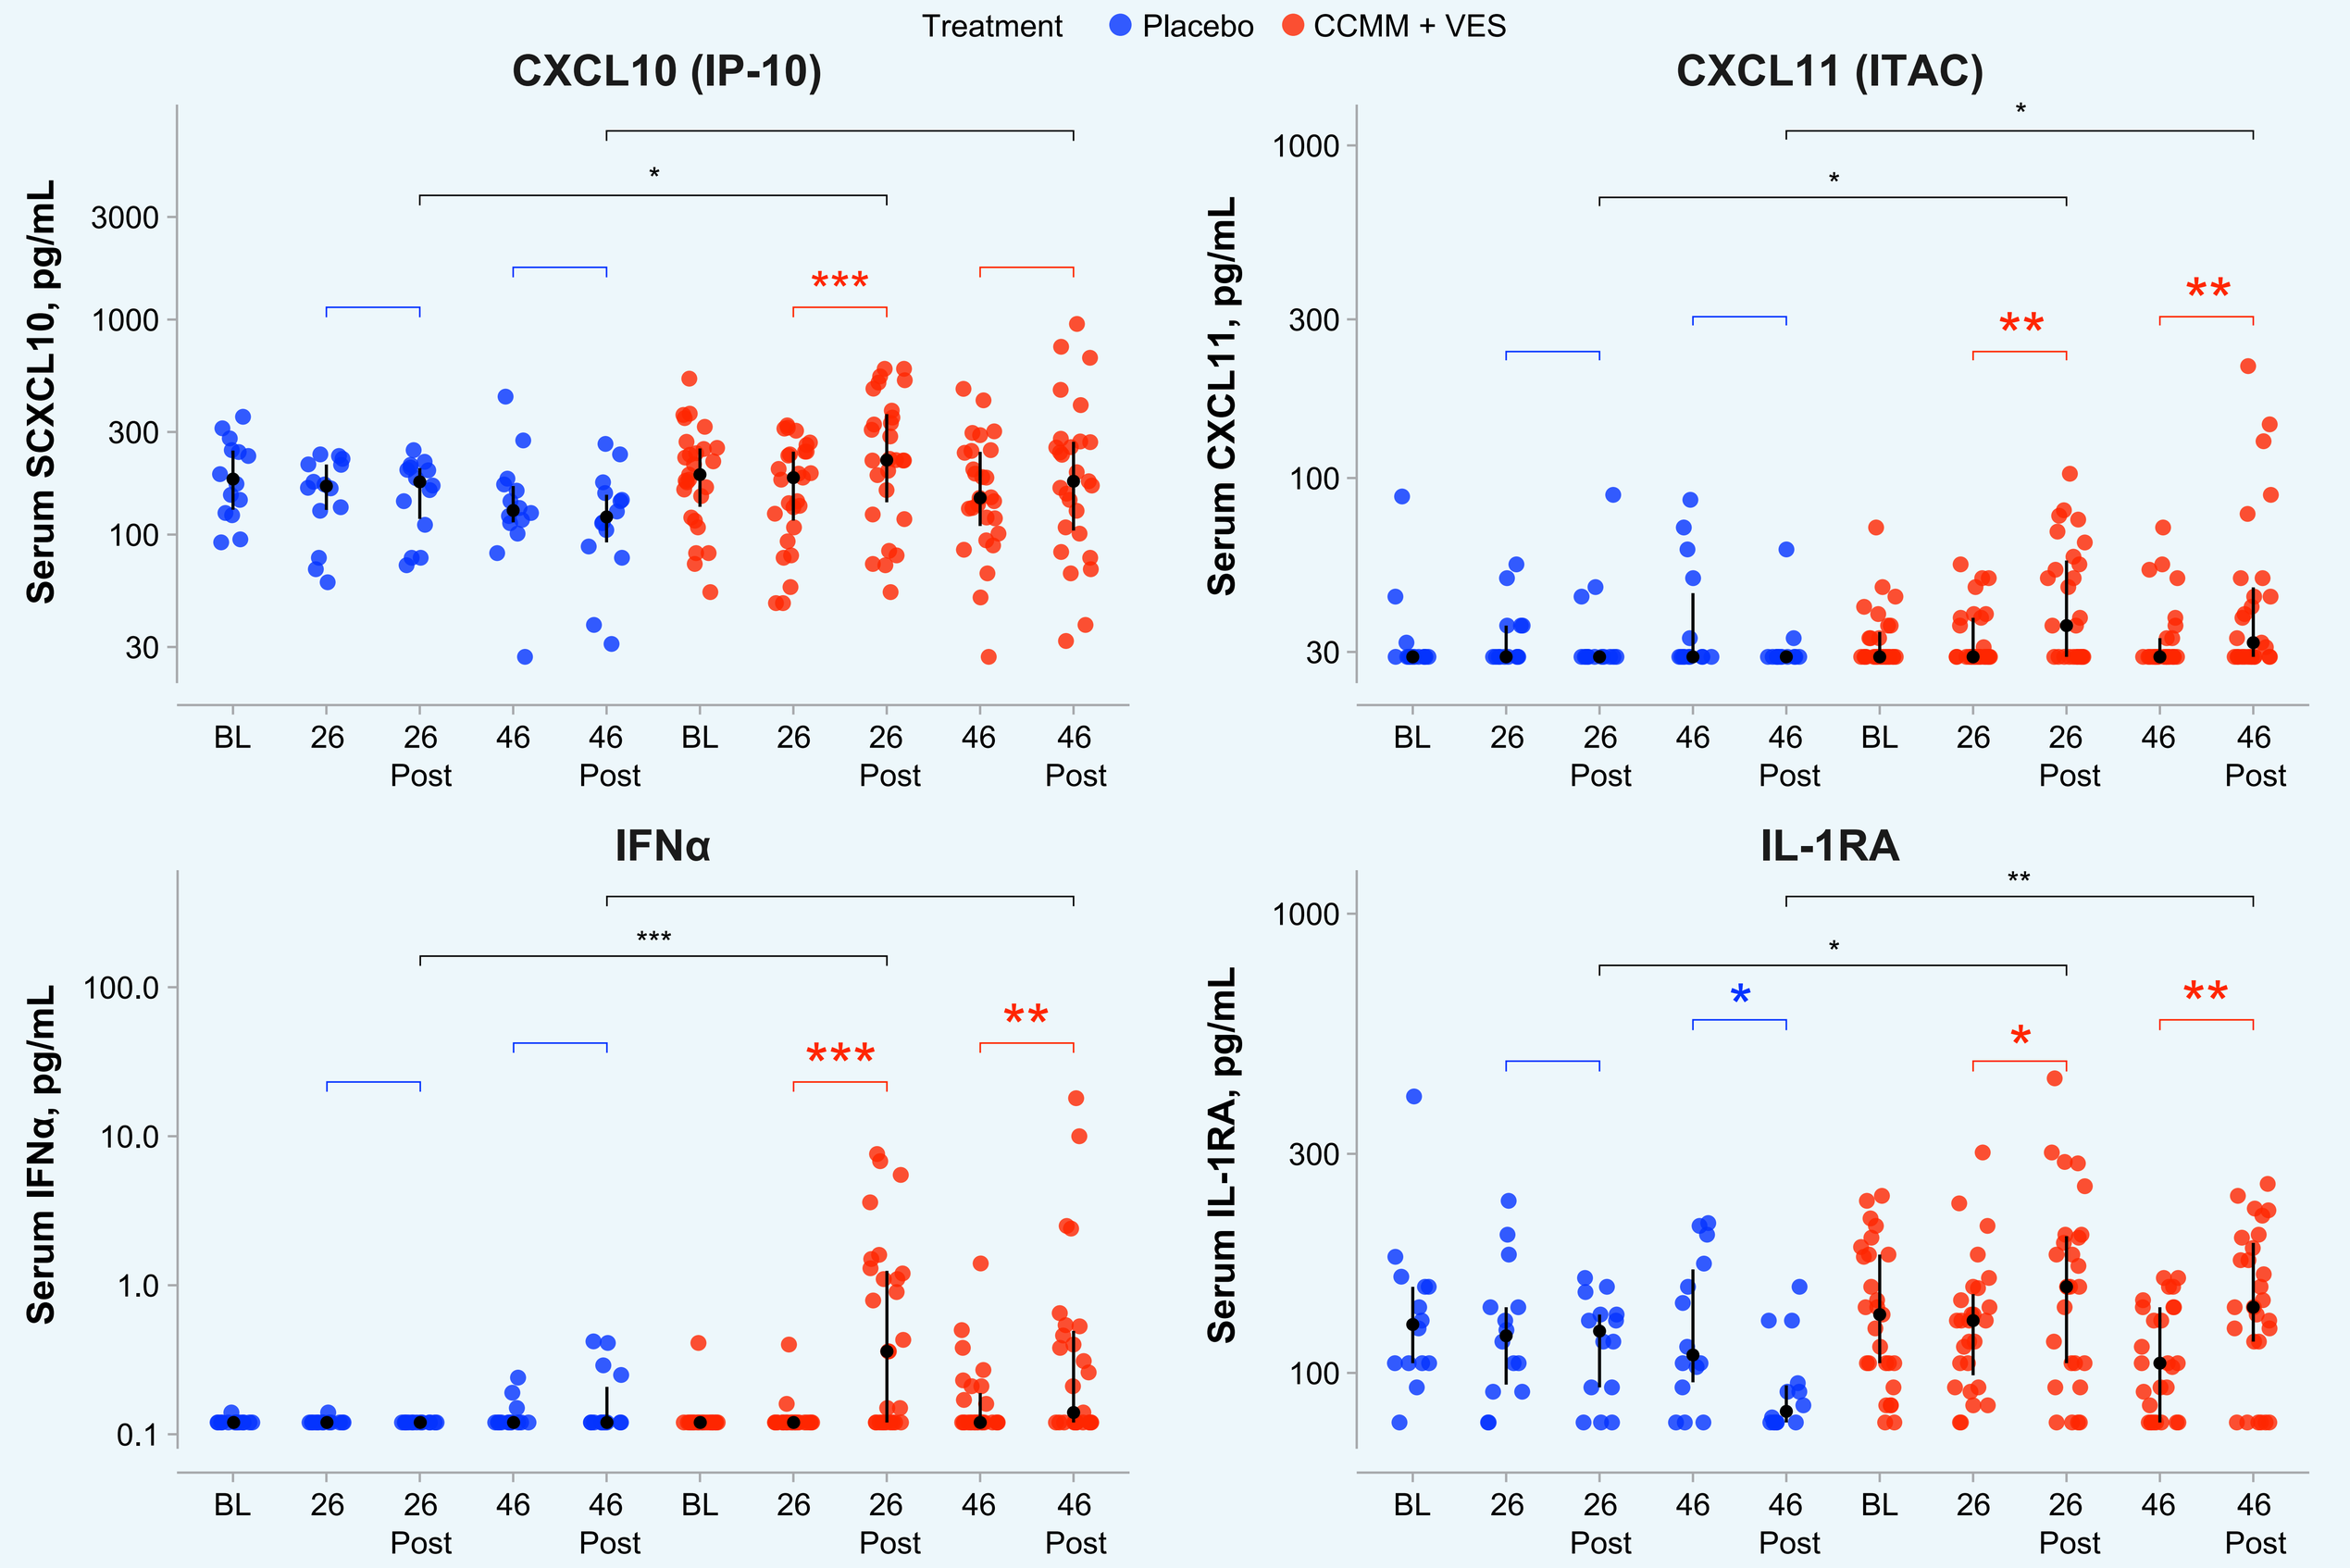

Treatment ● Placebo ● CCMM + VES
CXCL10 (IP-10)
CXCL11 (ITAC)
IFNα
IL-1RA
Serum SCXCL10, pg/mL
Serum CXCL11, pg/mL
Serum IFNα, pg/mL
Serum IL-1RA, pg/mL
BL 26 26 Post 46 46 Post

Serum levels were measured by single-molecule array assay for IFNα, and multiplex cytokine assay for ITAC, IP-10, and IL-1RA at BL, before and 24 hours after 2nd VES/placebo dose (Week 26), and before and 24 hours after 10th (last) VES/placebo dose (Week 46). *Colored-coded dots* represent data collected from individual participants. Median and IQR are shown with *black dots* and *lines*. Wilcoxon test was used to compare between groups or 2 different timepoints. *$P \le 0.05$, **$P \le 0.01$, ***$P \le 0.001$. CXCL = C-X-C motif chemokine ligand.

Viral Reservoir (Figure 4)

- No differences in the reduction of total (integrated) or intact (replication competent) proviral HIV-1 DNA were observed between arms **(Figure 4)**

Figure 4. HIV-1 Reservoir Decay in Placebo and CCMM + VES Recipients

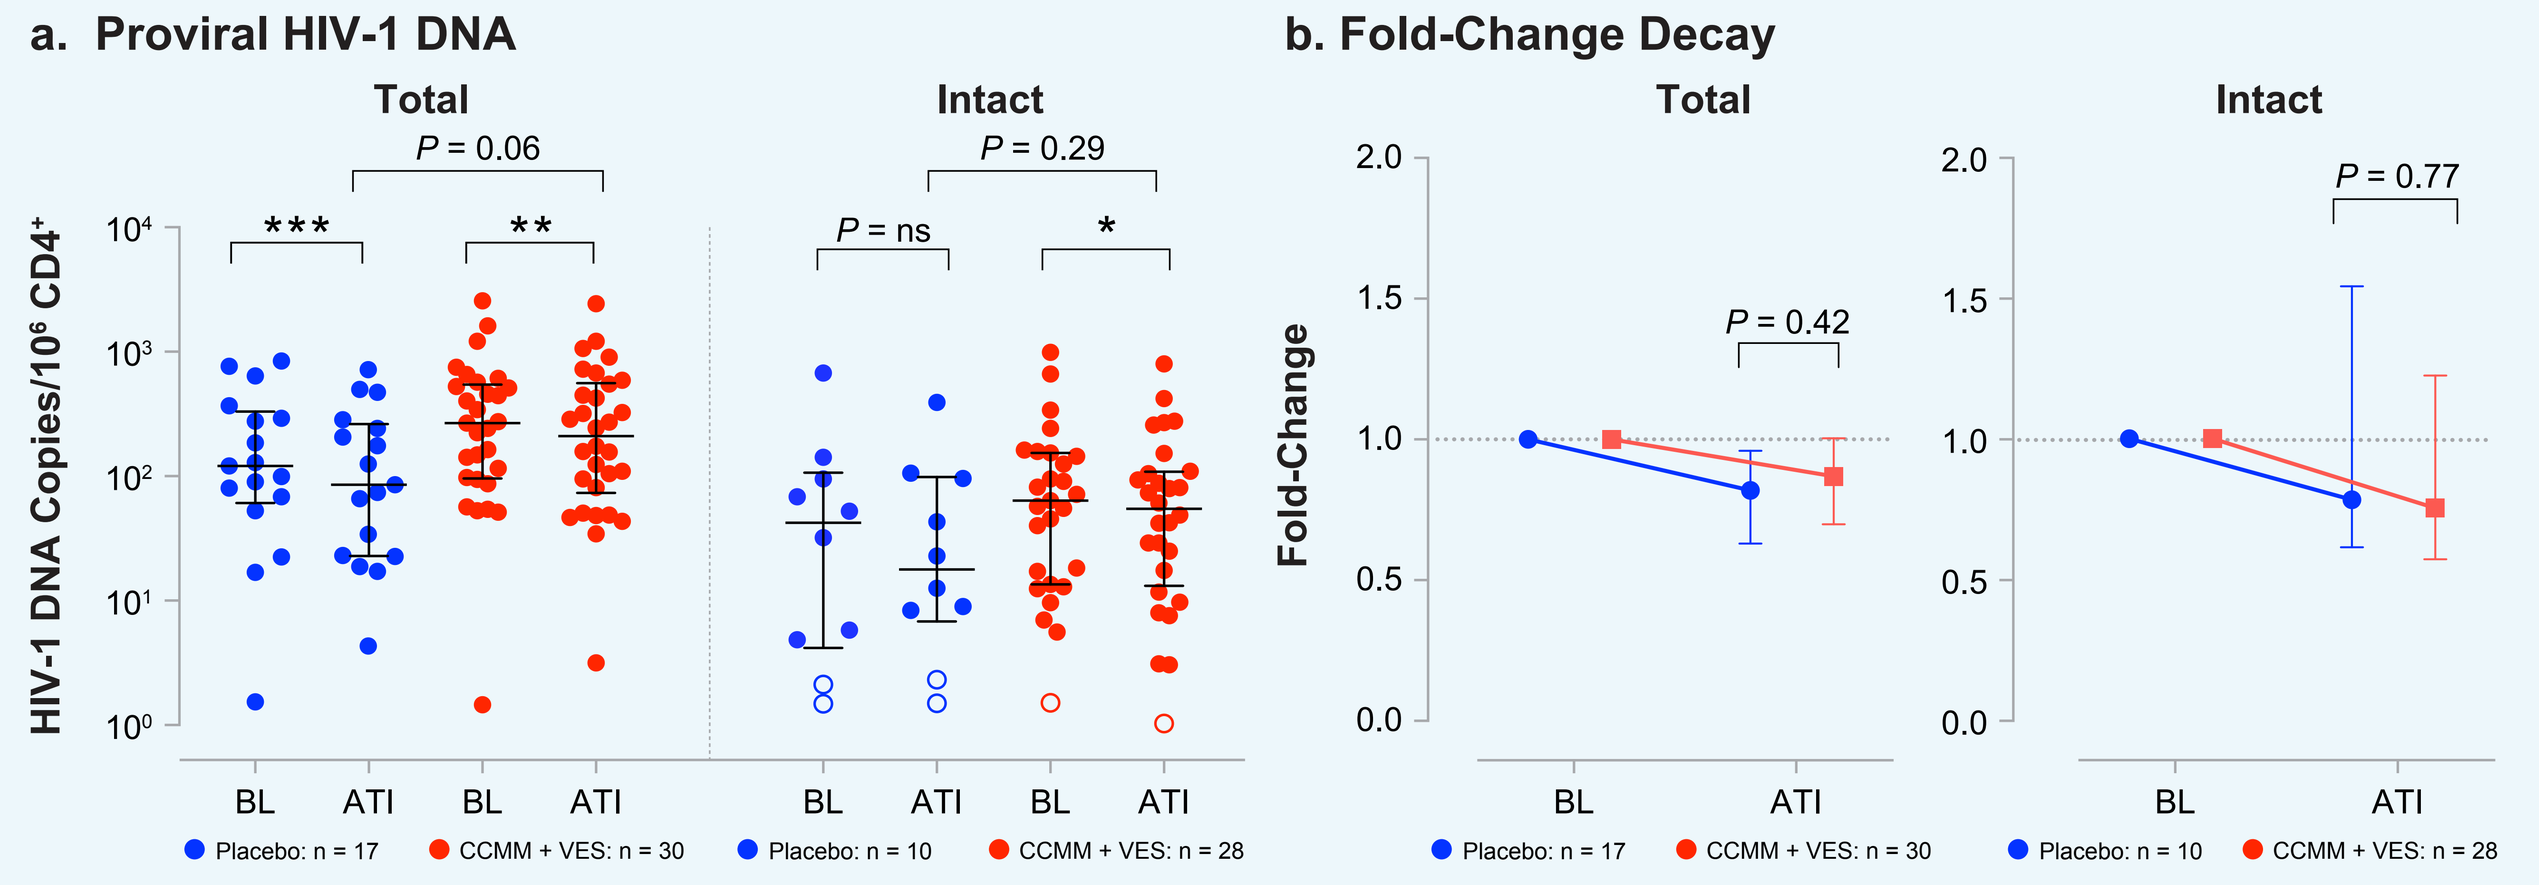

a. Proviral HIV-1 DNA
Total
Intact
P = 0.06
P = 0.29
P = ns
HIV-1 DNA Copies/10⁶ CD4⁺
BL ATI BL ATI
● Placebo: n = 17 ● CCMM + VES: n = 30 ● Placebo: n = 10 ● CCMM + VES: n = 28
b. Fold-Change Decay
Total
Intact
P = 0.42
P = 0.77
Fold-Change
BL ATI

(a) Comparison between levels of total and intact proviral HIV-1 DNA at BL and ATI in placebo *(blue)* and CCMM + VES *(red)* recipients. Participants with undetectable reservoir are shown in *open circles*. **(b)** Fold-change decay for total and intact proviral HIV-1 DNA by treatment group from BL to ATI start. Median and IQR are represented. Mann-Whitney and Wilcoxon tests were used to compare between groups and between 2 different time points within same participant, respectively. ns = not significant. *$P < 0.05$; **$P < 0.01$; ***$P < 0.001$.

ATI Period (Figure 5)

- 10 of 30 participants (33%) in the active arm remained off ART for 24 weeks compared with 4 of 17 placebo recipients (24%)
- Reasons for resuming ART before 24 weeks of ATI were pVL ≥ 100,000 copies/mL (16 CCMM + VES; 8 placebo) and pVL ≥ 10,000-100,000 copies/mL for 8 consecutive weeks (2 CCMM + VES; 5 placebo)
 - 1 CCMM + VES participant voluntarily resumed ART at Week 23 of ATI and 1 CCMM + VES participant at Week 18 due to hepatitis B infection

Figure 5. HIV-1 pVL and Percentage of Participants Remaining ART-Free During 24-Week ATI

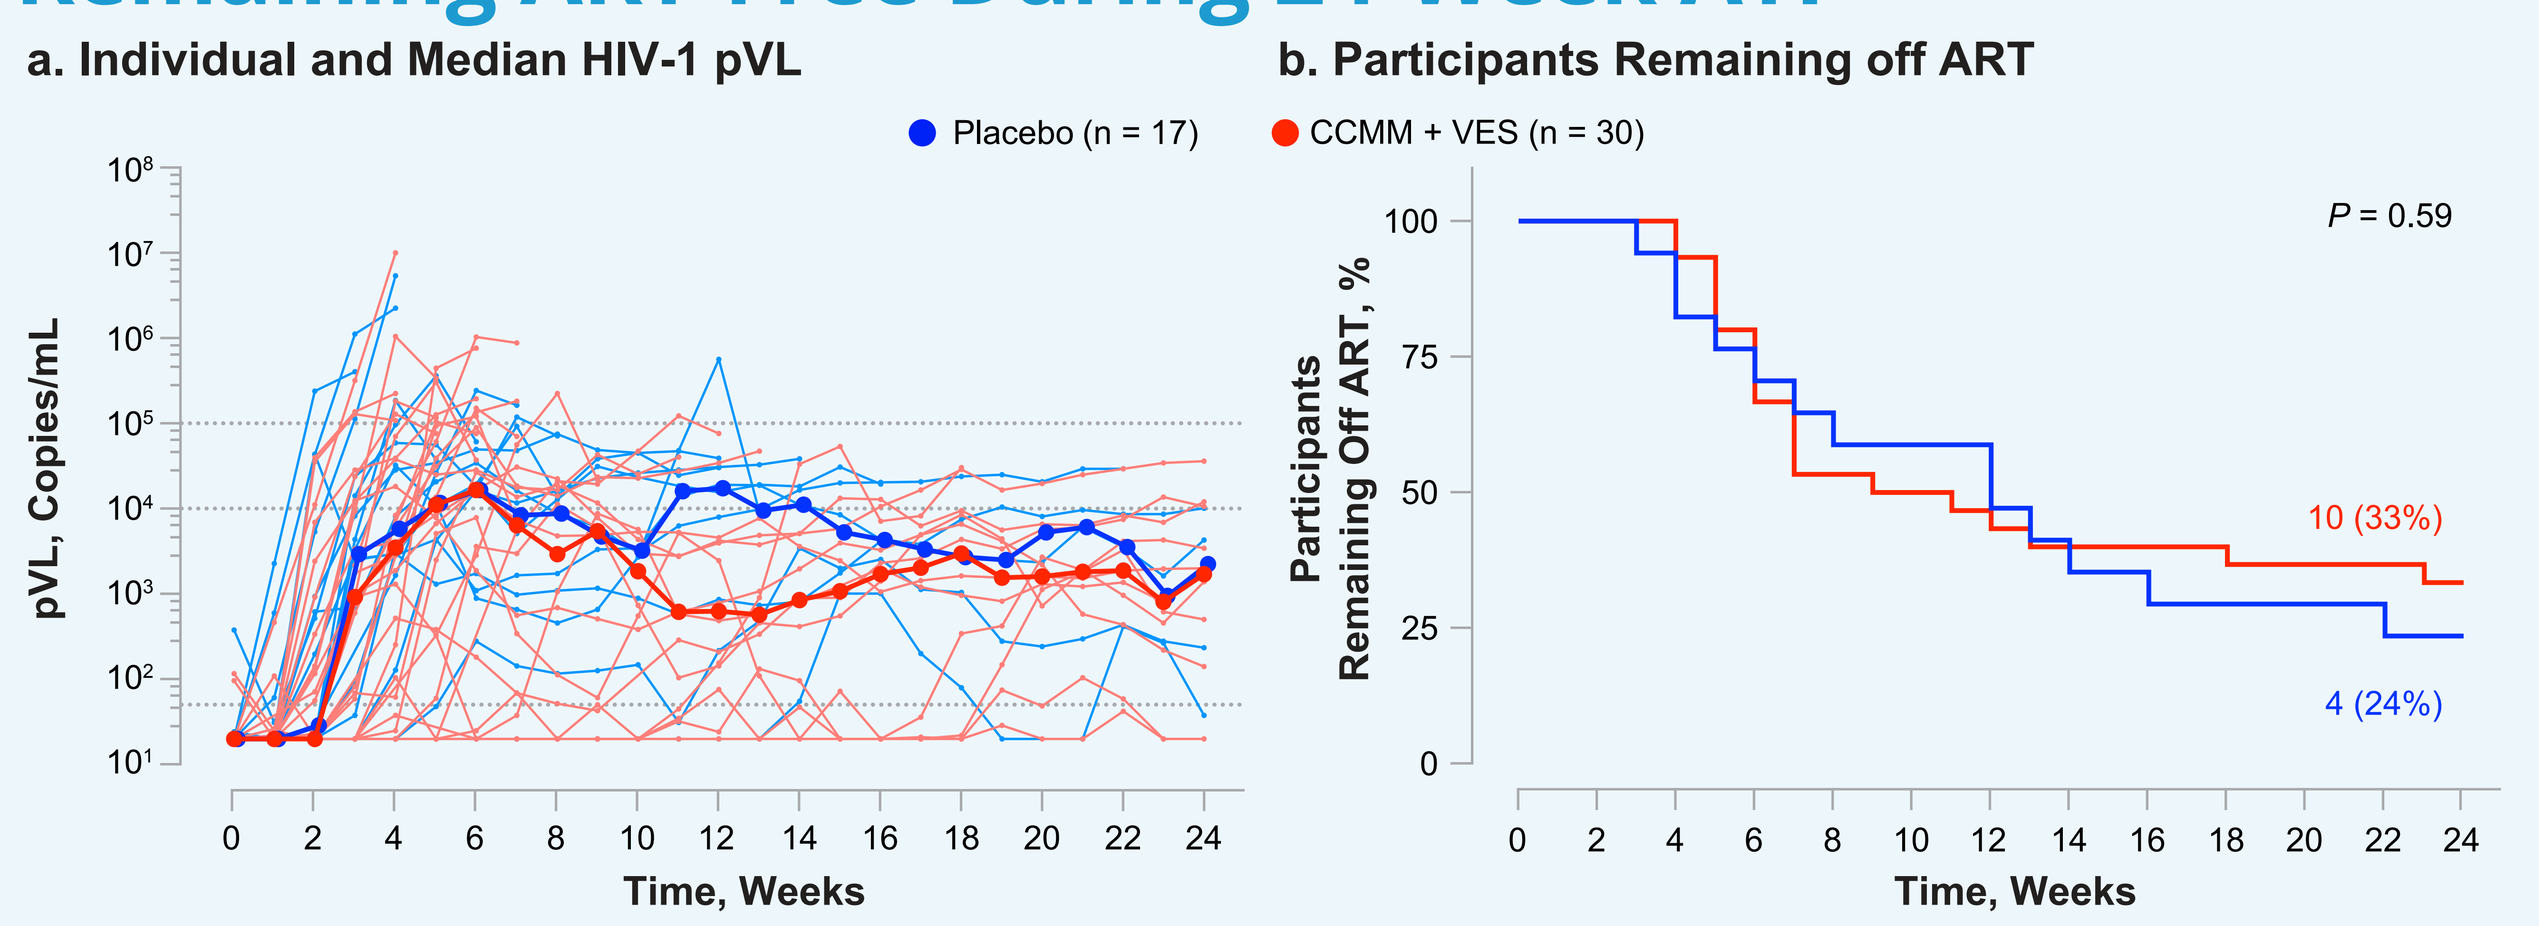

a. Individual and Median HIV-1 pVL
● Placebo (n = 17)
pVL, Copies/mL
Time, Weeks
b. Participants Remaining off ART
● CCMM + VES (n = 30)
P = 0.59
10 (33%)
4 (24%)
Participants Remaining Off ART, %
Time, Weeks

(a) Individual and median HIV-1 pVL during the 24 weeks of ATI, shown for all placebo *(blue)* and CCMM + VES *(red)* recipients (n = 47). *Lines* are interrupted at week of ART resumption. *Dotted lines* represent detection limit and 2 different virologic thresholds for ART resumption (10,000 and 100,000 HIV-1 RNA copies/mL, respectively). **(b)** Proportions of participants in placebo and CCMM + VES arms remaining off ART following treatment interruption. Stratified log-rank test was performed adjusting for stratification factor potential for superior viral control.

Preliminary Correlate Analyses (Figure 6)

- In the CCMM + VES group, higher pre-ART pVL correlated with worse viral outcomes (shorter time to viral rebound > 50 and > 10,000 copies/mL, and higher pVL at the end of ATI)
- In the placebo group, lower pre-intervention viral reservoir size was associated with better viral outcomes (longer time off ART and lower pVL at the end of ATI)
- In the CCMM + VES group, higher magnitude and breadth of vaccine response correlated with better viral outcomes (longer time to viral rebound > 50 or > 10,000 copies/mL, and longer time off ART, respectively)

Figure 6. Subgroup Analysis of Correlates Associated With Viral Kinetics During ART (Interim Analysis)

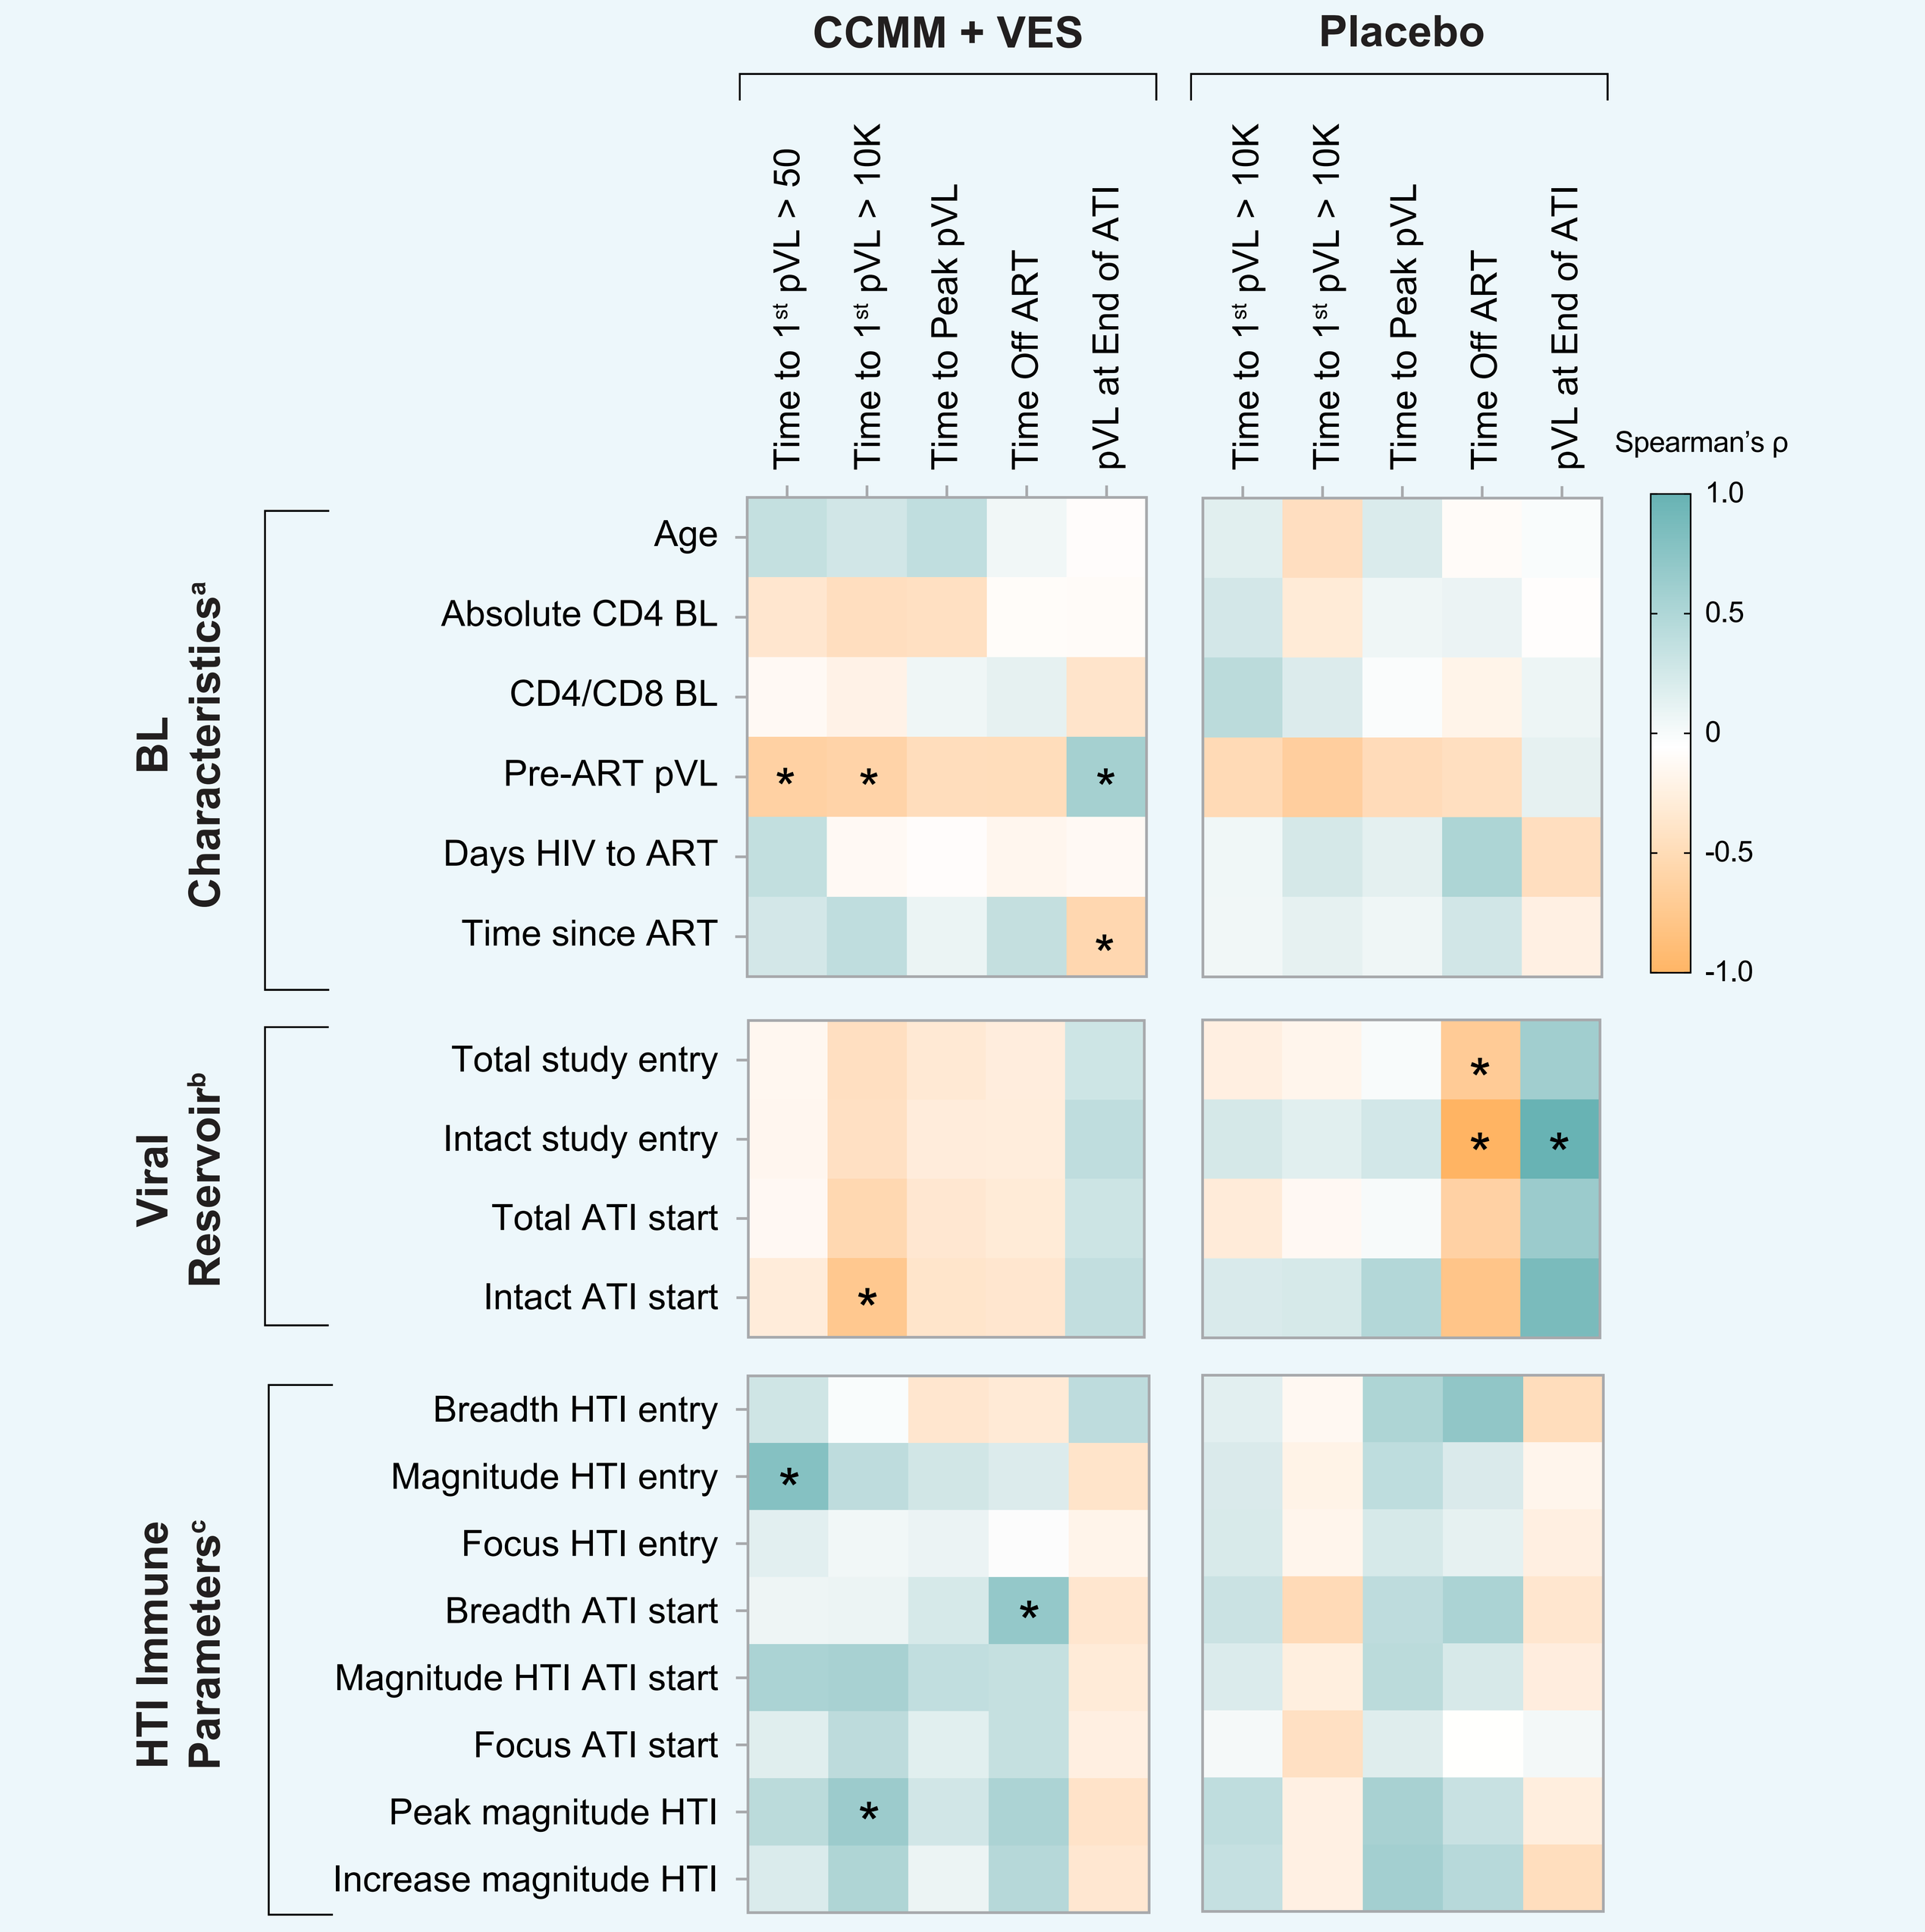

CCMM + VES
Placebo
Time to 1st pVL > 50
Time to 1st pVL > 10K
Time to Peak pVL
Time Off ART
pVL at End of ATI
Spearman's ρ
BL Characteristics[a]
Age
Absolute CD4 BL
CD4/CD8 BL
Pre-ART pVL
Days HIV to ART
Time since ART
Viral Reservoir[b]
Total study entry
Intact study entry
Total ATI start
Intact ATI start
HTI Immune Parameters[c]
Breadth HTI entry
Magnitude HTI entry
Focus HTI entry
Breadth ATI start
Magnitude HTI ATI start
Focus ATI start
Peak magnitude HTI
Increase magnitude HTI

[a]Placebo: n = 17; CCMM + VES: n = 33; [b]For total HIV-1 DNA: placebo: n = 17, and CCMM + VES: n = 30; for intact HIV-1 DNA: placebo: n = 10, and CCMM + VES: n = 28; [c]Placebo: n = 13; CCMM + VES: n = 21. Spearman's ρ is used for correlations. All tests are 2-sided, unadjusted for multiple comparisons, with 5% error rate. Significant correlations are shown by * when $P < 0.05$.

Conclusions

- **The combination of the HTI vaccine and VES as part of an HIV cure strategy in early-treated PWH was safe and well tolerated**
- **Preliminary immune data suggest that HTI vaccines given in combination with VES induce high levels of HTI-specific T-cell responses; VES consistently induced PD response over multiple doses in combination therapy with HTI vaccine**
- **A lower pre-intervention reservoir was associated with better viral outcomes during the ATI, but only in the placebo group**
- **The magnitude and breadth of vaccine response correlated with better viral outcomes**
- **The impact of VES on antiviral activity of HTI-specific T cells and virus control remains to be investigated**

References: 1. Mothe B, et al. J Transl Med 2015:13;60. **2.** Bailón L, et al. Nat Med 2022;28:2611-21. **3.** Borducchi E, et al. Nature 2016;540:284-7. **4.** Riddler SA, et al. Clin Infect Dis 2021;72:e815-24. **5.** SenGupta D, et al. Sci Transl Med 2021;13:eabg3071. **6.** Fiebig E, et al. AIDS 2003;17:1871-9.

Acknowledgments: Special thanks to all the volunteers participating in this study, and all the site investigators for their perseverance and dedication to the study. The authors thank Elena Vendrame, Yuan Tian, and Michelle Frankot of Gilead Sciences, Inc. for scientific oversight and writing support. Editing and production assistance were provided by BioScience Communications, New York, NY, funded by Gilead.

AELIX-003 Research Group: **La Paz:** Julen Cadiñanos, Juan Gonzalez, Jose-Ramón Arribas; **H. Gregorio Marañón:** Juan Carlos Lopez Bernaldo de Quirós, Leire Perez, Juan Berenguer, Chiara Fanciulli; **H. La Princesa:** Ignacio de los Santos, Lucio Jesús García-Fraile, Gina Paola Mejía; **H. Ramón y Cajal:** Santiago Moreno; **HUGTiP:** Lucía Bailón, Susana Benet, Francisco Pérez, Yovannina Alarcón-Soto, José Moltó, Beatriz Mothe; **H. Clínic:** Juan Ambrosiono, Josep-Maria Miró-Moreno; **H. Sant Pau:** Pere Domingo; **H. Bellvitge:** Sofia Scevola, Arkaitz Imaz; **Vall d'Hebrón:** Paula Suanzez, Jordi Navarro, Vicenç Falcó, Adrià Curran.

This study was funded by AELIX Therapeutics and Gilead Sciences, Inc.

Disclosures: B. Mothe: HTI co-inventor; consultancy fees: AELIX; advisory and speaker fees: Gilead, Janssen, ViiV; **A. Curran:** advisory and speaker fees, and grant support: Gilead, Janssen, MSD, ViiV Healthcare; **J.C. López Bernardo de Quirós**: speaker fees and grant support: Gilead, MSD; **J. Cadiñanos:** speaker fees and grant support: Gilead, MSD; **I. de los Santos:** none; **J. Ambrosioni:** advisory board and speaker fees, and grant support: Gilead, Janssen, MSD, ViiV; data and safety monitoring board member: Grifols, HIPRA; employee: European AIDS Clinical Society (Guidelines Coordinator); **A.I. Vacas:** advisory and speaker fees, and grant support: Gilead, Janssen, MSD, Theratechnologies, ViiV; **S. Moreno:** speaking fees and research grants: Gilead, Janssen Cilag, MSD, ViiV; **P. Domingo:** lecture and advisory board fees: ViiV Healthcare, MSD, GSK, Roche, Theratechnologies, Janssen & Cilag, Gilead; **Y. Cai, R. Geleziunas, D. SenGupta:** employees and stockholders of Gilead; **I. McGowan:** employee of AELIX; **C. Brander:** cofounder, shareholder, and CSO of AELIX; **J.R. Arribas:** advisory and speaker fees, and grant support: Viiv, Janssen, Gilead, MSD, AELIX.

Presented at CROI 2023, 19-23 February 2023, Seattle, WA

© 2023. All rights reserved.