HIV Gag x CD3 soluble TCR bispecific: safety and activity in a first in human trial

436 LB

Linos Vandekerckhove[1], Julie Fox[2], Borja Mora-Peris[3], Jordi Navarro[4], Sabine Allard[5], Alison Uriel[6], Santiago Moreno[7], Marta Boffito[8], Frank Post[9], Vicente Estrada[10], Beatriz Mothe[11], Yuan Yuan[12], Adel Benlahrech[13], Lucy Dorrell[13], Sarah Fidler[3]

[1]HIV Cure Research Center, Ghent University & Ghent University Hospital, Belgium; [2]King's College London, UK; [3]Imperial College London, UK; [4]Hospital Universitari Vall d'Hebron, Barcelona, Spain; [5]Vrije Universiteit Brussel, Universitair Ziekenhuis Brussel, Brussels, Belgium; [6]North Manchester General Hospital, Manchester, UK; [7]Hospital Ramon y Cajal, Madrid, Spain; [8]Chelsea and Westminster Hospital, London, UK; [9]King's College Hospital, London, UK; [10]Hospital Clínico San Carlos, Madrid, Spain; [11]Irsicaixa Institute for AIDS Research, Badalona, Spain; [12]Immunocore , Rockville MD, USA; [13]Immunocore Ltd, Abingdon, UK

Research collaborators: Praveen K Singh[13], Haseeb Rahman[13], Zoë Wallace[13], Agatha Treveil[13], Kathryn Lamming[13], Andrew D Whale[13]

IMC-M113V is a soluble TCR bispecific protein targeting HIV Gag

- A functional cure for HIV requires interventions that can eliminate long-lived reservoirs of integrated proviruses in CD4+ T cells.
- Immune-mobilising monoclonal TCR against viruses (ImmTAV®) are soluble bispecific proteins designed to redirect polyclonal T cells, irrespective of specificity, to kill virus-infected cells[1].
- IMC-M113V (Gag x CD3) redirects effector T cells to kill HIV-infected cells presenting a specific HLA-bound Gag-derived peptide (Gag_{77-85}) (Figure 1)[2,3].
- IMC-M113V is active at picomolar drug concentrations *in vitro* (Figure 2) and *ex vivo*[2], and therefore has the potential to reduce or eliminate HIV reservoirs in people living with HIV (PLWH).

Figure 1a. ImmTAV structure

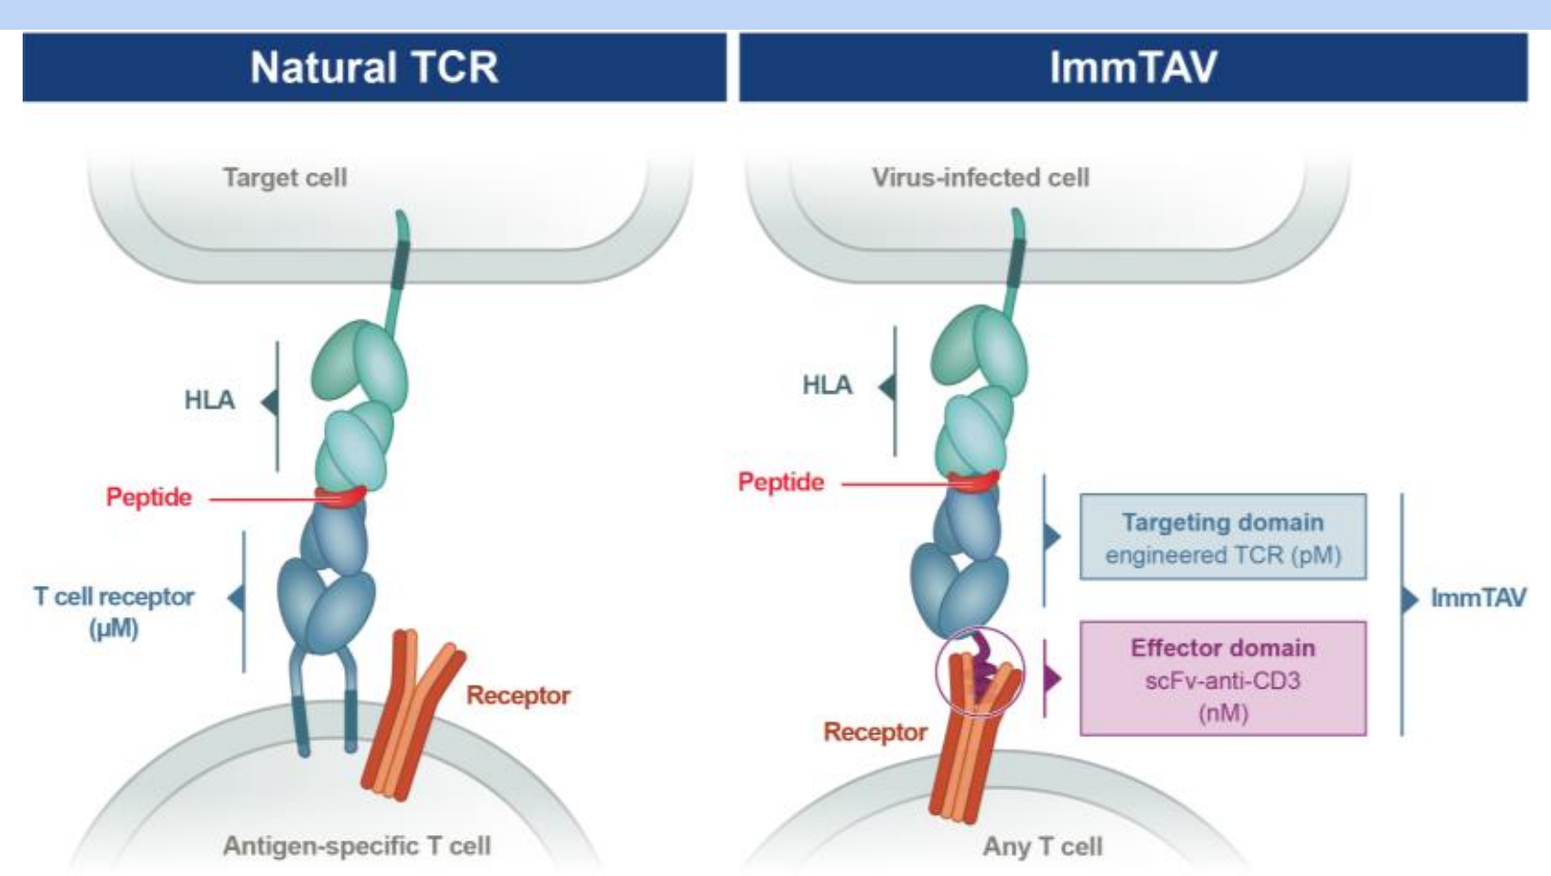
Natural TCR
ImmTAV
Target cell
Virus-infected cell
HLA
Peptide
T cell receptor (µM)
Receptor
Antigen-specific T cell
Targeting domain engineered TCR (pM)
Effector domain scFv-anti-CD3 (nM)
ImmTAV
Any T cell

Figure 1b. Mechanism of action of IMC-M113V (Gag x CD3)

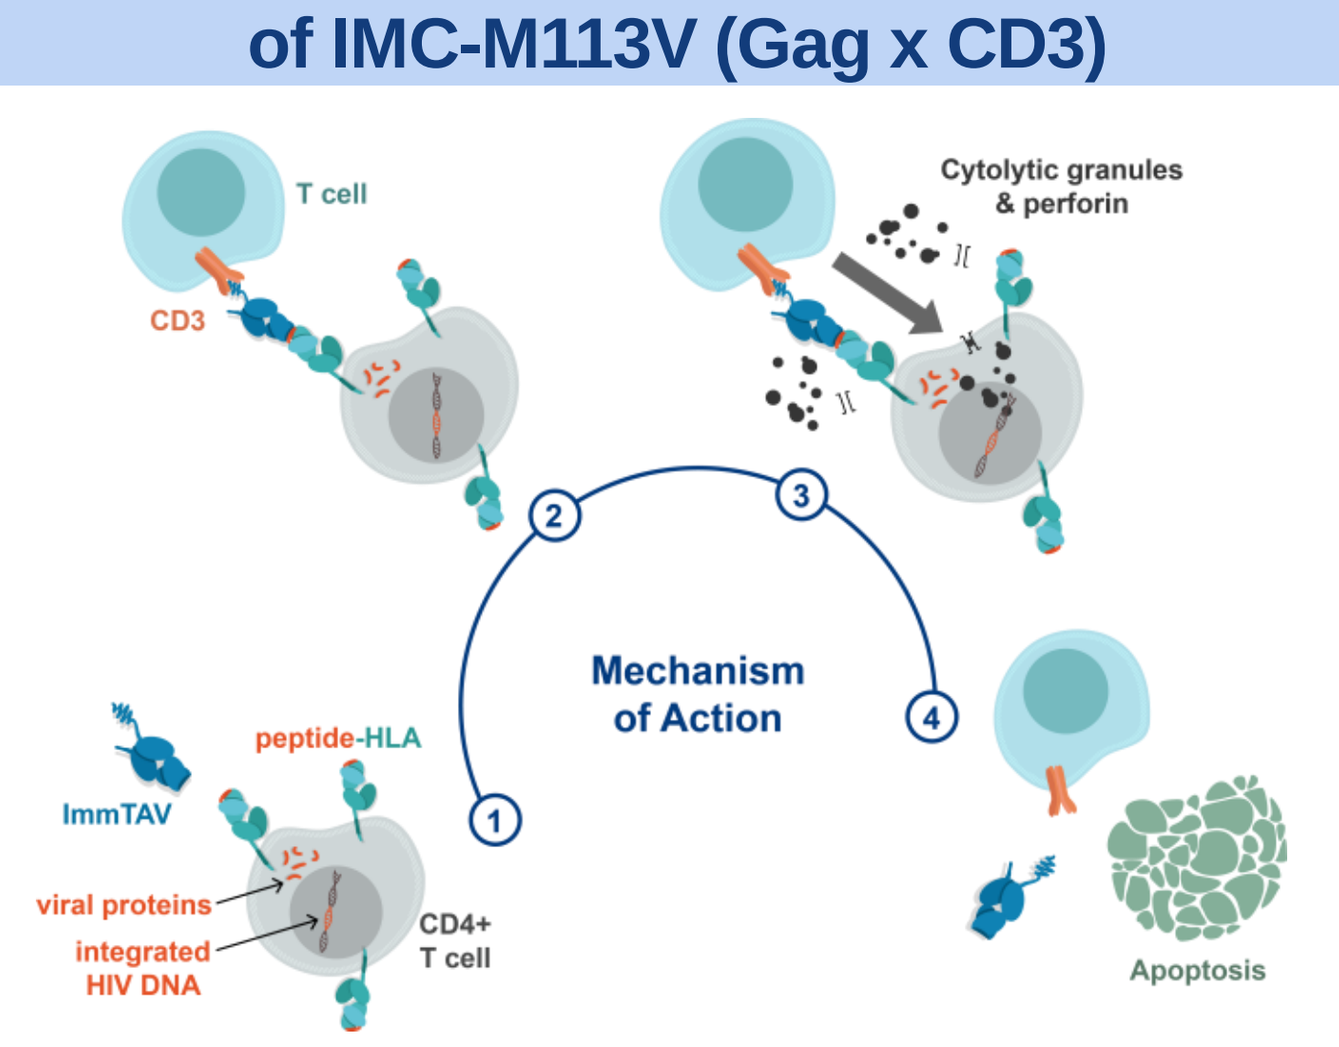
T cell
CD3
Cytolytic granules & perforin
Mechanism of Action
ImmTAV
peptide-HLA
viral proteins
integrated HIV DNA
CD4+ T cell
Apoptosis
1
2
3
4

IMC-M113V redirects any effector T cell to kill HIV-infected cells

1. IMC-M113V targeting domain engages pHLA on HIV Gag-positive CD4+ T cell
2. IMC-M113V effector domain engages CD3 on passing T cell
3. Activated T cell releases pro-inflammatory cytokines and lytic molecules
4. Activated T cell triggers apoptosis of infected CD4+ T cell

Blood biomarkers:
↓ Absolute lymphocyte count
↑ IL-6, CXCL10, IFN-γ etc

Figure 2. IMC-M113V potency demonstrated in preclinical studies

IMC-M113V has picomolar potency against HIV-infected cells *in vitro*

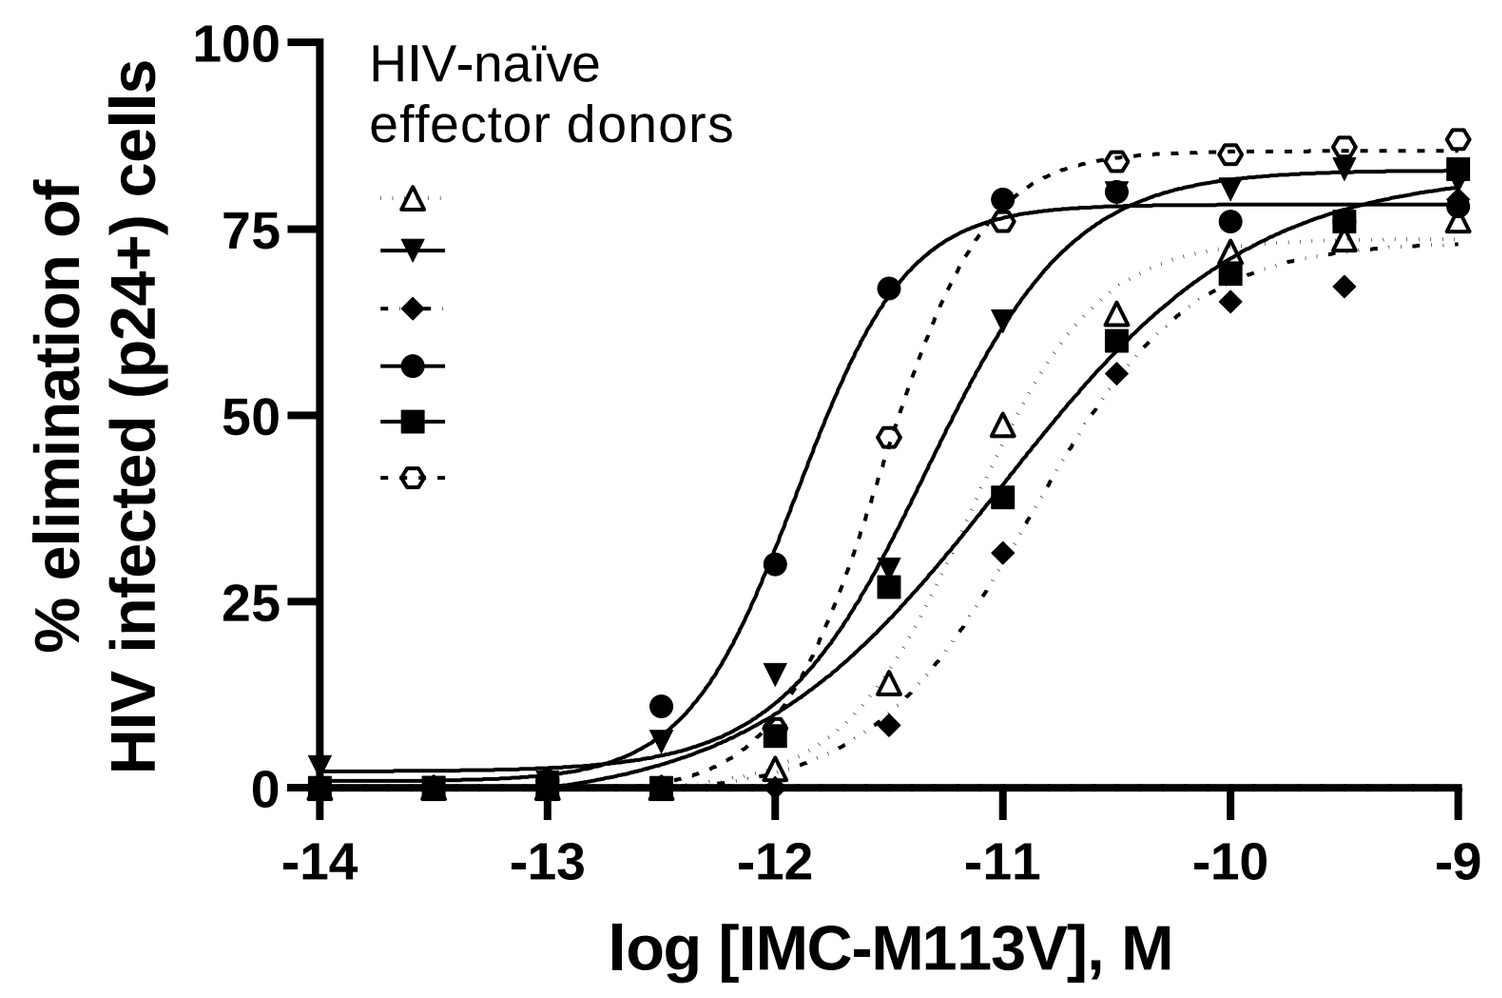
% elimination of HIV infected (p24+) cells
HIV-naïve effector donors
100
75
50
25
0
-14
-13
-12
-11
-10
-9
log [IMC-M113V], M

Targets: HIV-infected CD4+ cell line, transduced with HLA-A*02:01
Effectors: CD8+ T cells from HIV-naïve donors (n = 6)
ImmTAV: Increasing concentration of IMC-M113V
Co-culture: 7 days

IMC-M113V is active against known Gag_{77-85} epitope variants

Target sequence	$t_{1/2}$ (h)[a]	Relative EC_{50}[b]
SLYNTVATL	**3.71**	**1.0**
SLFNTVATL	3.79	2.28
SLYNTVAVL	2.03	1.68
SLYNTIATL	3.51	0.6
SLFNTVAVL	2.25	6.28
SLFNTIATL	3.06	1.19
SLFNTIAVL	1.78	2.92
SLYNTIAVL	1.81	0.77

[a] pHLA stability at 37ºC

[b] EC_{50} values derived from IFN-γ Elispot assay: HIV-naïve donor PBMC were redirected by IMC-M113V against T2 cells pulsed with variant peptides. Relative EC_{50} = EC_{50} of variant/ EC_{50} of cognate epitope, SLYNTVATL

Methods

- STRIVE (Soluble T cell Receptors In Virus Eradication) is an open-label Phase 1/2 study evaluating IMC-M113V in HLA-A*02:01-positive PLWH receiving suppressive antiretroviral therapy (ART).
- 12 subjects enrolled in a single ascending dose study, designed to identify safe and biologically active doses for evaluation in multiple dose schedules (Figure 3).
- Duration of ART was limited to 7 years to enable targeting of active reservoirs[4].
- Participants received antipyretics and antihistamines as premedication and underwent extended monitoring for dosing visits to mitigate risk of cytokine release syndrome[5,6].
- A ≥4-fold rise in serum IL-6 at 8-24 hours post-dose was prespecified as indicative of pharmacodynamic (PD) activity.
- Gag_{77-85} sequence (IMC-M113V target) was determined pre-dose by next-generation sequencing (Illumina Miseq) of cell-associated HIV RNA (CA-RNA).

Figure 3. Single ascending dose study schema

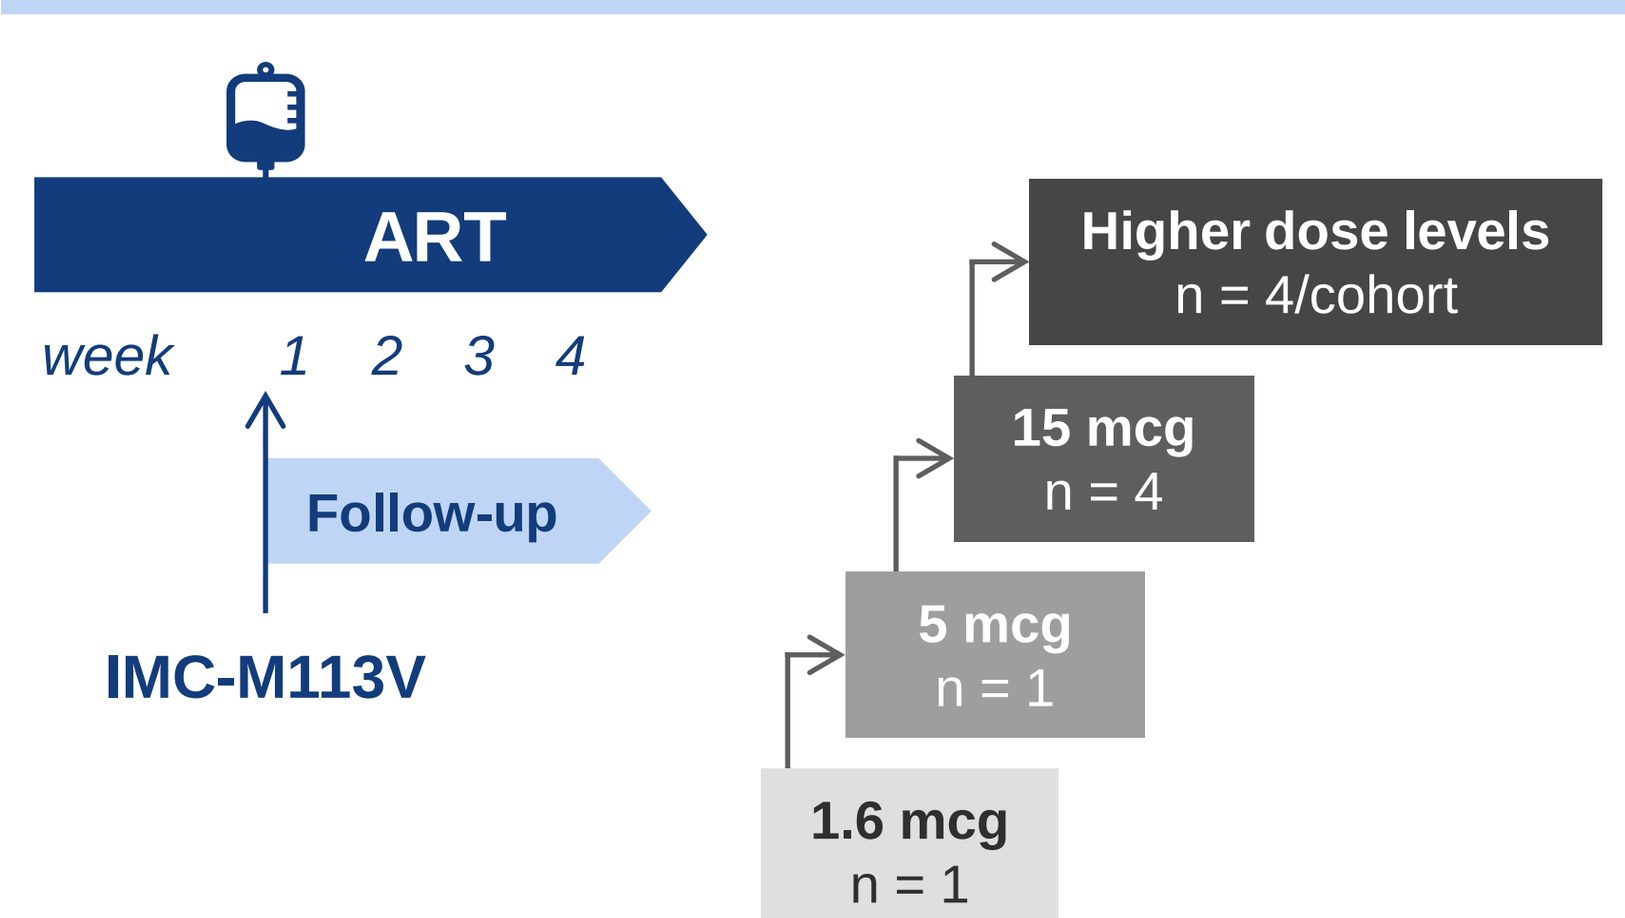
ART
week 1 2 3 4
Follow-up
IMC-M113V
1.6 mcg
n = 1
5 mcg
n = 1
15 mcg
n = 4
Higher dose levels
n = 4/cohort

Key eligibility criteria

- HLA-A*02:01+ PLWH
- Age 18-65
- On ART 1-7 years
- Viral load < 50 c/ml
- CD4 count > 500 cells/µl
- CD4 nadir > 200 cells/µl

Participants were allocated sequentially to ascending dose cohorts: starting dose was based on the minimum anticipated biological effect level; dose increments were ≤ 3-fold. Minimum number of subjects per cohort is indicated. Study design allowed for enrolment of higher dose cohorts (up to 7 in total).

IMC-M113V was given as a single IV infusion of IMC-M113V on Day 1. Subjects were followed weekly to Day 29.

Results

Results 1. IMC-M113V was well tolerated and not associated with cytokine release syndrome at doses ≤15 mcg

92 pre-screened for HLA
↓
35 HLA-A*02:01+
↓
16 completed screening for Part 1
↓
3 SAD cohorts completed (n = 6)
↓
Cohort 3 (15 mcg) expanded from 4 to 10 participants
↓
12/12 completed follow-up

- Subjects enrolled at sites in UK, Belgium and Spain
- Doses were well tolerated; no dose-limiting toxicities
- No cytokine release syndrome of any grade
- No neurotoxicity
- No SAEs
- No Grade ≥ 3 hematology, chemistry or liver function test abnormalities
- HIV viral load remained < 50 c/ml through dosing and follow-up
- CD4 cell counts remained within normal range

Results 2. Gag_{77-85} sequences obtained from cell-associated RNA pre-dose indicate the presence of druggable, transcriptionally active proviruses

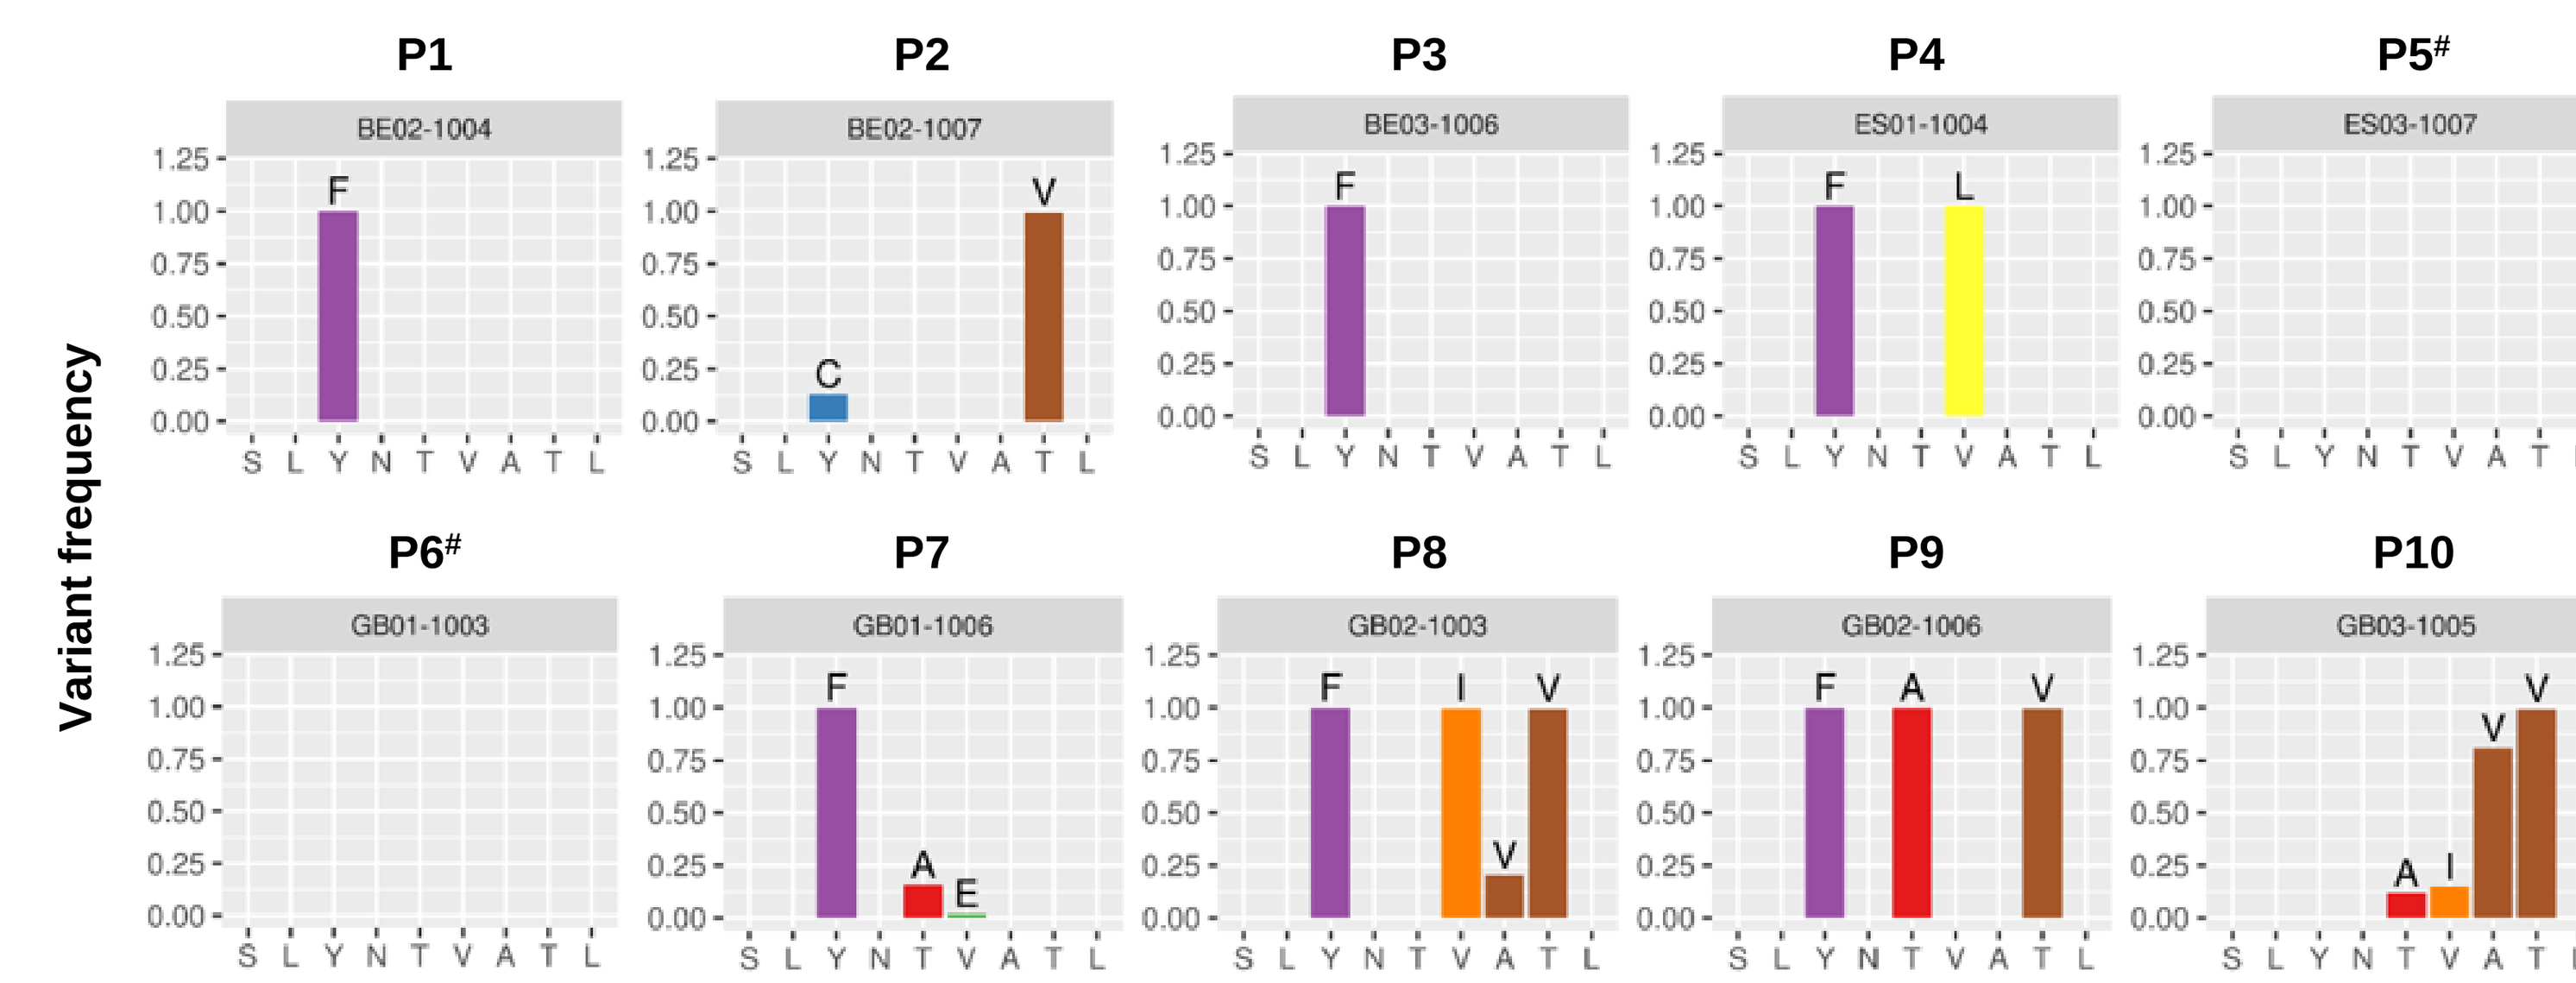
Variant frequency
P1
BE02-1004
P2
BE02-1007
P3
BE03-1006
P4
ES01-1004
P5#
ES03-1007
P6#
GB01-1003
P7
GB01-1006
P8
GB02-1003
P9
GB02-1006
P10
GB03-1005
S L Y N T V A T L

- Anticipated variability in Gag_{77-85} observed in study participants
- Baseline viral sequencing will inform clinical and antiviral signals

Nextera XT Libraries sequenced on Illumina MiSeq Platform, created from targeted amplification of CA-RNA Gag from purified CD4+ cells isolated pre-dose from the ten participants (P1-10) in cohort 3 (15 mcg). # Gag_{77-85} sequence matched cognate (SLYNTVATL) in participants P5 and P6.

Results 3. Tolerable and biologically active dose identified in SAD study

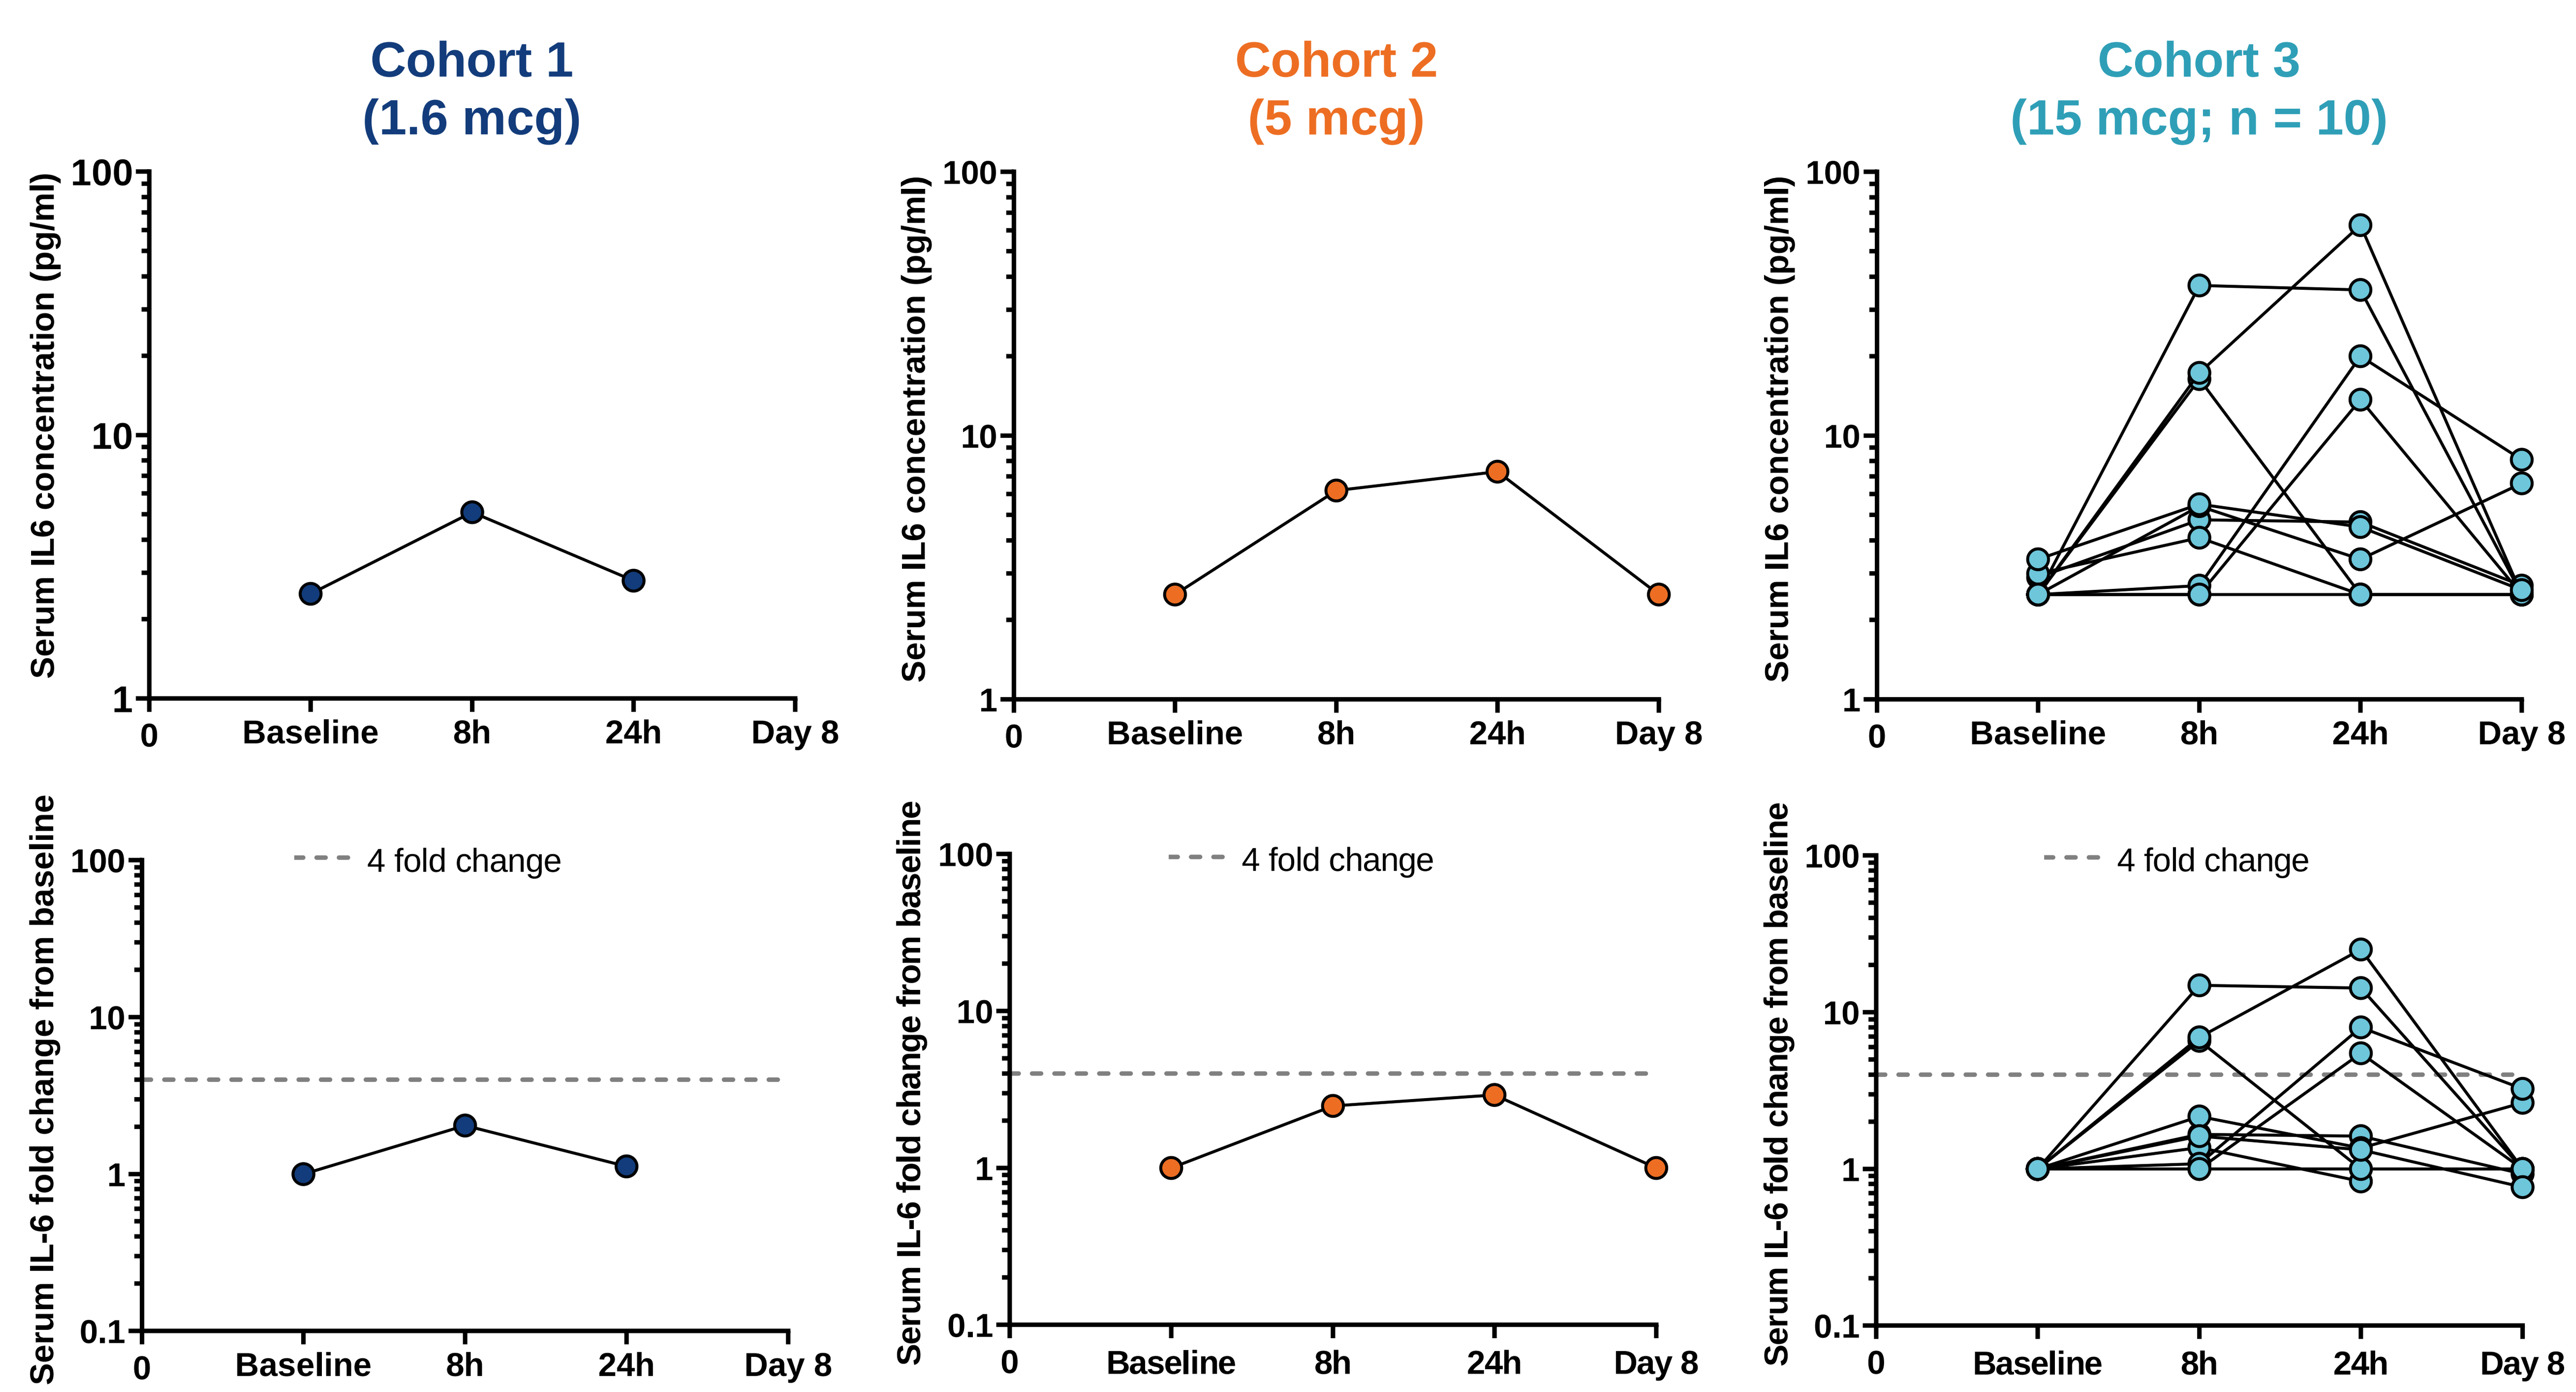
Cohort 1
(1.6 mcg)
Cohort 2
(5 mcg)
Cohort 3
(15 mcg; n = 10)
Serum IL6 concentration (pg/ml)
Serum IL-6 fold change from baseline
4 fold change
Baseline
8h
24h
Day 8

- 15 mcg dose fulfilled criteria to stop escalation in SAD and open MAD study

Results 4: *In vitro* assays confirm that IL-6 induction is target-dependent

Whole blood off-target cytokine release assay: IMC-M113V at ≤ 30 nM (~11,250 mcg) does not induce IL-6

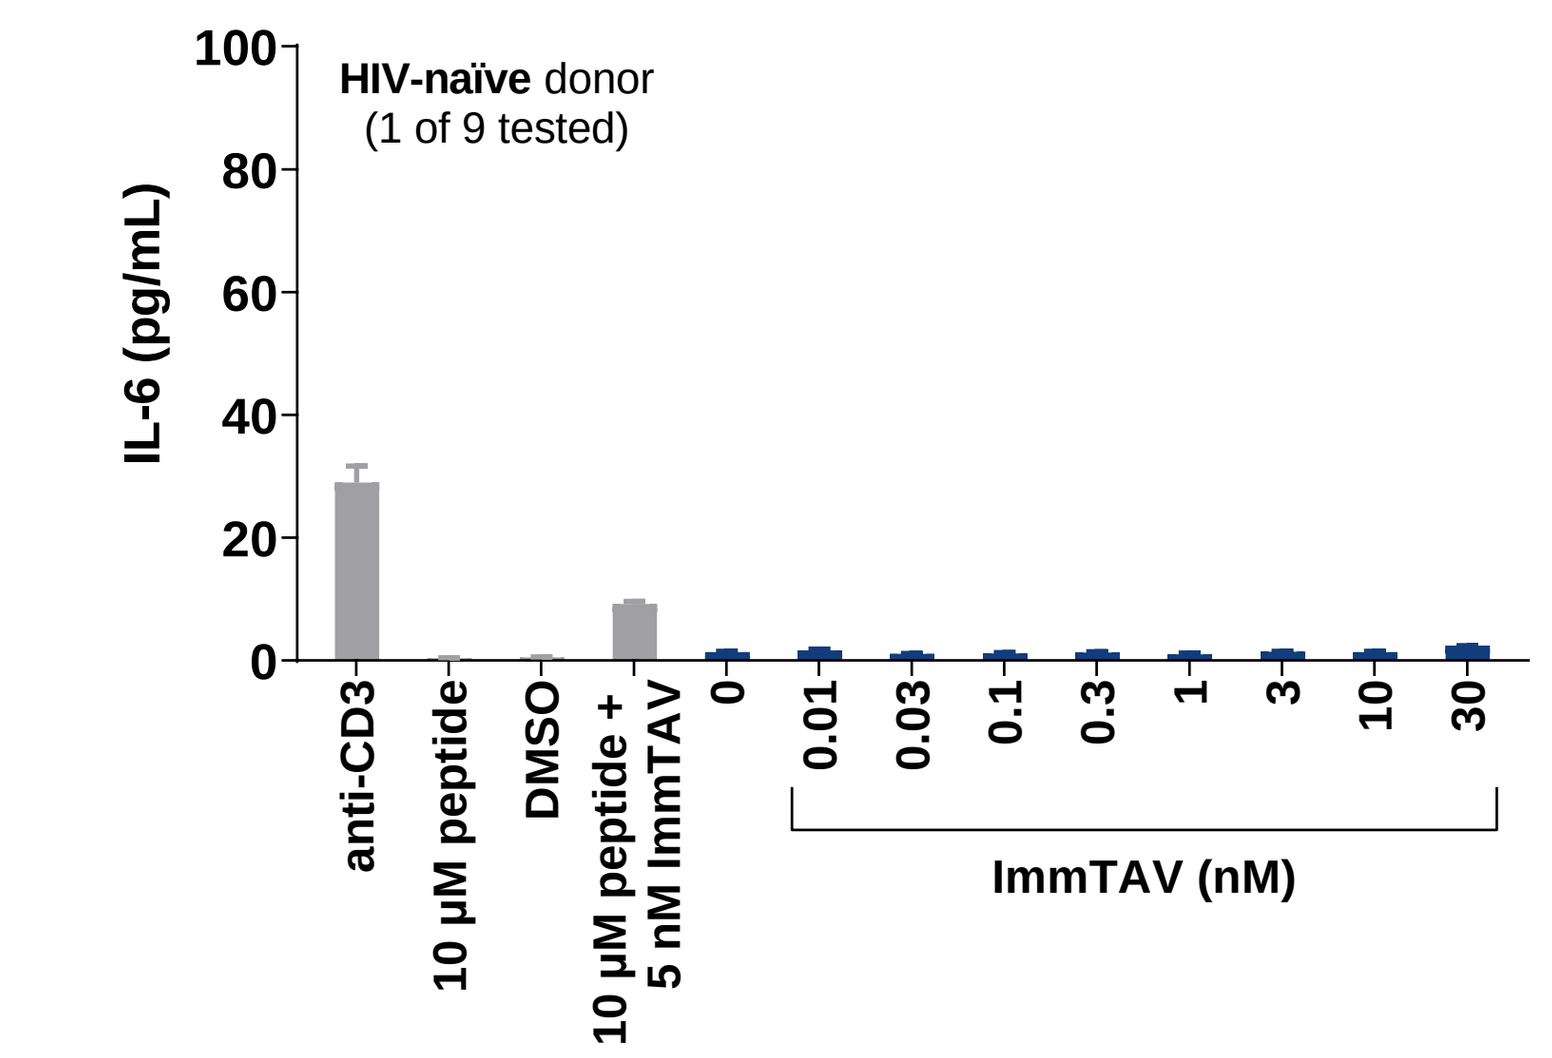
IL-6 (pg/mL)
HIV-naïve donor
(1 of 9 tested)
anti-CD3
10 µM peptide
DMSO
10 µM peptide + 5 nM ImmTAV
0
0.01
0.03
0.1
0.3
1
3
10
30
ImmTAV (nM)

Whole blood spiked with IMC-M113V for 4h at 37°C; IL-6 quantified by MSD immunoassay

On-target T cell redirection assay: 40 pM IMC-M113V (=15 mcg dose) elicits IL-6 only when HIV target peptide present

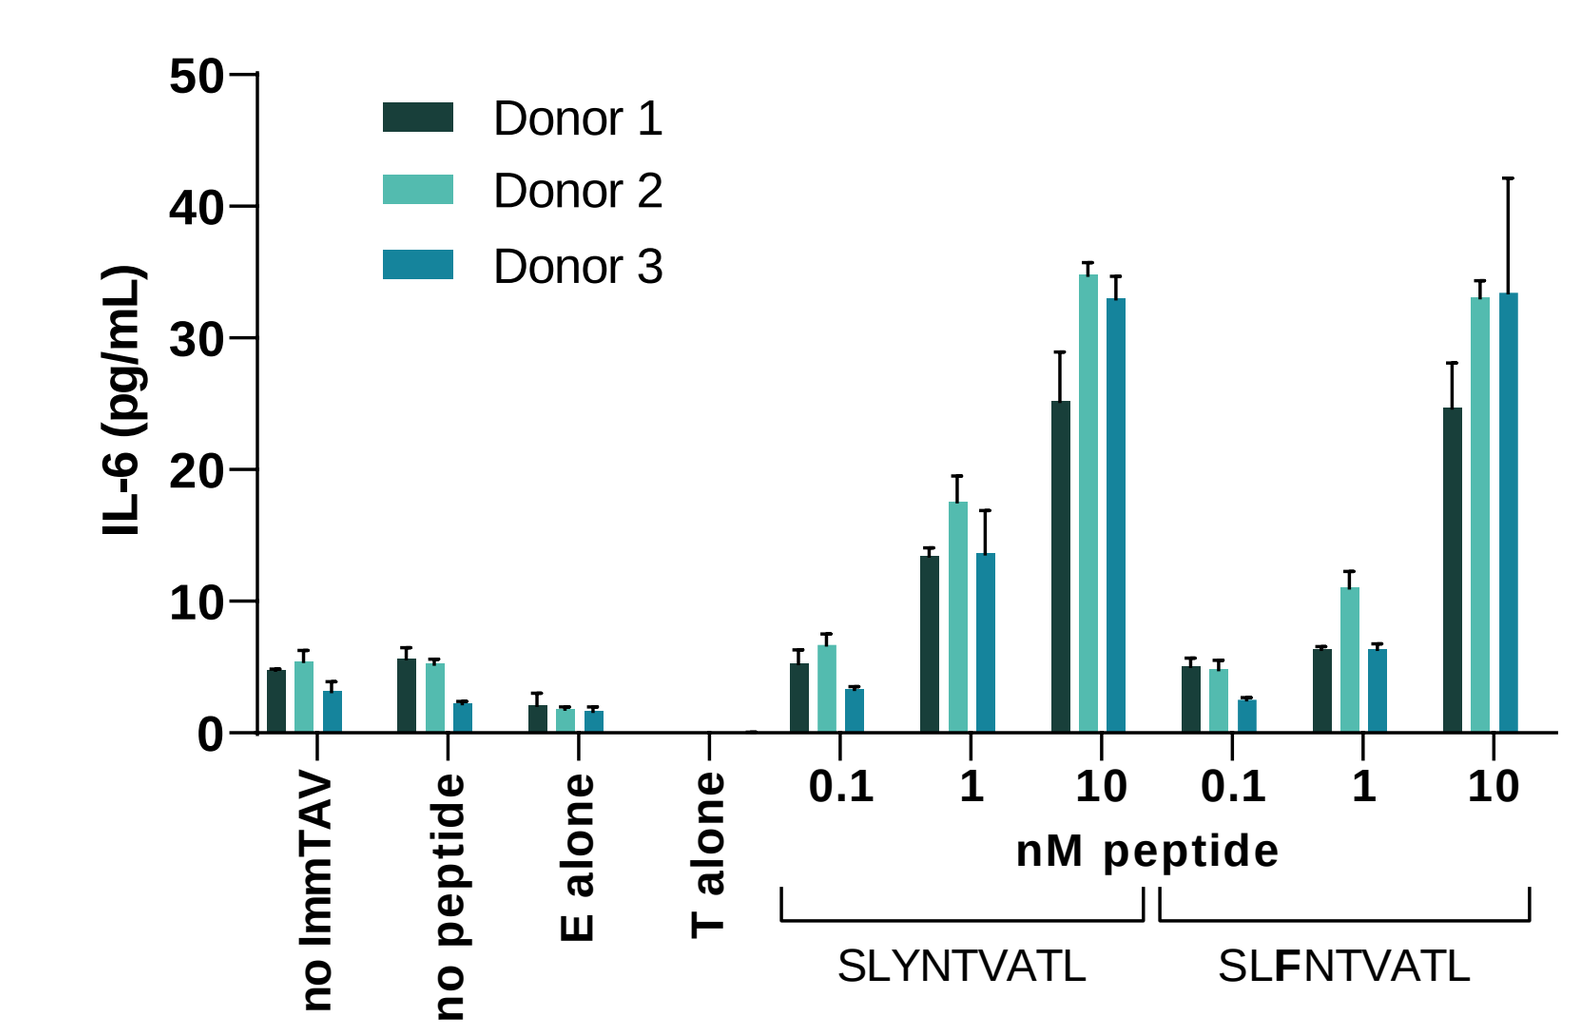
IL-6 (pg/mL)
Donor 1
Donor 2
Donor 3
no ImmTAV
no peptide
E alone
T alone
0.1
1
10
nM peptide
SLYNTVATL
SLFNTVATL

Peptide-pulsed T2 cells (T) cultured with PBMC (E) from HIV-naïve donors overnight; IL-6 assay as per (A). Both the cognate peptide (SLYNTVATL) and major variant (SLFNTVATL) elicited an IL-6 response. No IL-6 detected with HLA-A*02:01-negative targets (not shown)

Conclusions

- IMC-M113V is, to our knowledge, the first TCR bispecific biologic targeting HIV to enter clinical evaluation.
- Single doses ranging from 1.6 – 15 mcg were well tolerated and not associated with cytokine release syndrome, neurotoxicity or any serious adverse event.
- Biological activity (IL-6) consistent with the mechanism of action was observed at the 15 mcg dose.
 - Further analysis of the relationship between IL-6 response and target expression (determined by baseline viral sequence and CA-RNA copies), is ongoing.
- This study provides the first human safety and PD impact of this novel mechanism.
- A multiple ascending dose study will evaluate safety, together with the potential for IMC-M113V to reduce HIV reservoirs and enable post-treatment viral control (now enrolling, Eudract no. 2021-002008-11).

References

1. Wallace Z, Singh PK, Dorrell L. J Virus Erad. 2022;8:100082
2. Yang H, Buisson S, Bossi G et al. Mol Ther. 2016;24:1913-1925
3. Wallace Z, Kopycinski J, Yang H, et al. Sci Rep. 2022;12:18366
4. Astorga-Gamaza A, Buzon M. Curr Opin HIV AIDS 2021;16:193-199
5. Nathan P, Hassel JC, Rutkowski P et al. N Engl J Med. 2021;385:1196-1206
6. Carvajal RD, Nathan P, Sacco JJ et al. J Clin Oncol. 2022;40:1939-1948

Corresponding author email: linos.Vandekerckhove@ugent.be

Disclaimers: IMC-M113V is an investigational product for which safety and efficacy have not been established. Copies of this poster are for personal use only and may not be reproduced without permission from CROI and the author of this poster.

IMMUNOCORE