NK CELL BIOMARKERS THAT PREDICT TIME-TO-REBOUND IN HIV+ INDIVIDUALS UNDERGOING ATI

00337

Ashley F. George[1,2], Tongcui Ma[1,2], Min-Gyoung Shin[3], Reuben Thomas[3], Mauricio Montano[1], Satish Pillai[4], Martin Tolstrup[5], Ole S. Søgaard[5], Jonathan Z. Li[6], Davey M. Smith[7], Steven G. Deeks[8], Katherine S. Pollard[3], Warner C. Greene[1], Nadia R. Roan[1,2]

[1]Gladstone Institute of Virology, San Francisco, CA, USA; [2]Department of Urology, University of California, San Francisco, CA, USA; [3]Gladstone Institutes, San Francisco, CA, USA; [4]Vitalant Research Institute, San Francisco, California; Department of Laboratory Medicine, University of California, San Francisco, California.; [5]Departments of Infectious Diseases and Clinical Medicine, Aarhus University Hospital, Aarhus N, Denmark; [6]Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [7]University of California, San Diego, San Diego, CA.; [8]Division of HIV, Infectious Diseases and Global Medicine, University of California San Francisco, San Francisco, CA, USA

BACKGROUND

- Most ART-suppressed people with HIV exhibit viral rebound within weeks of analytical treatment interruption (ATI), but some can control replication for prolonged periods of time.
- The mechanisms mediating control upon ATI remain poorly understood, and may involve Natural Killer (NK) cells, potent innate immune effector cells.
- NK cells are also capable of mounting “memory” recall responses, and are critical for controlling viral infections including HIV.

STUDY QUESTIONS

1) Do any NK features associate with ART initiation time (acute vs. chronic infection)?
2) Do any NK features associate with reservoir size or time-to-rebound?

METHODS

(1)

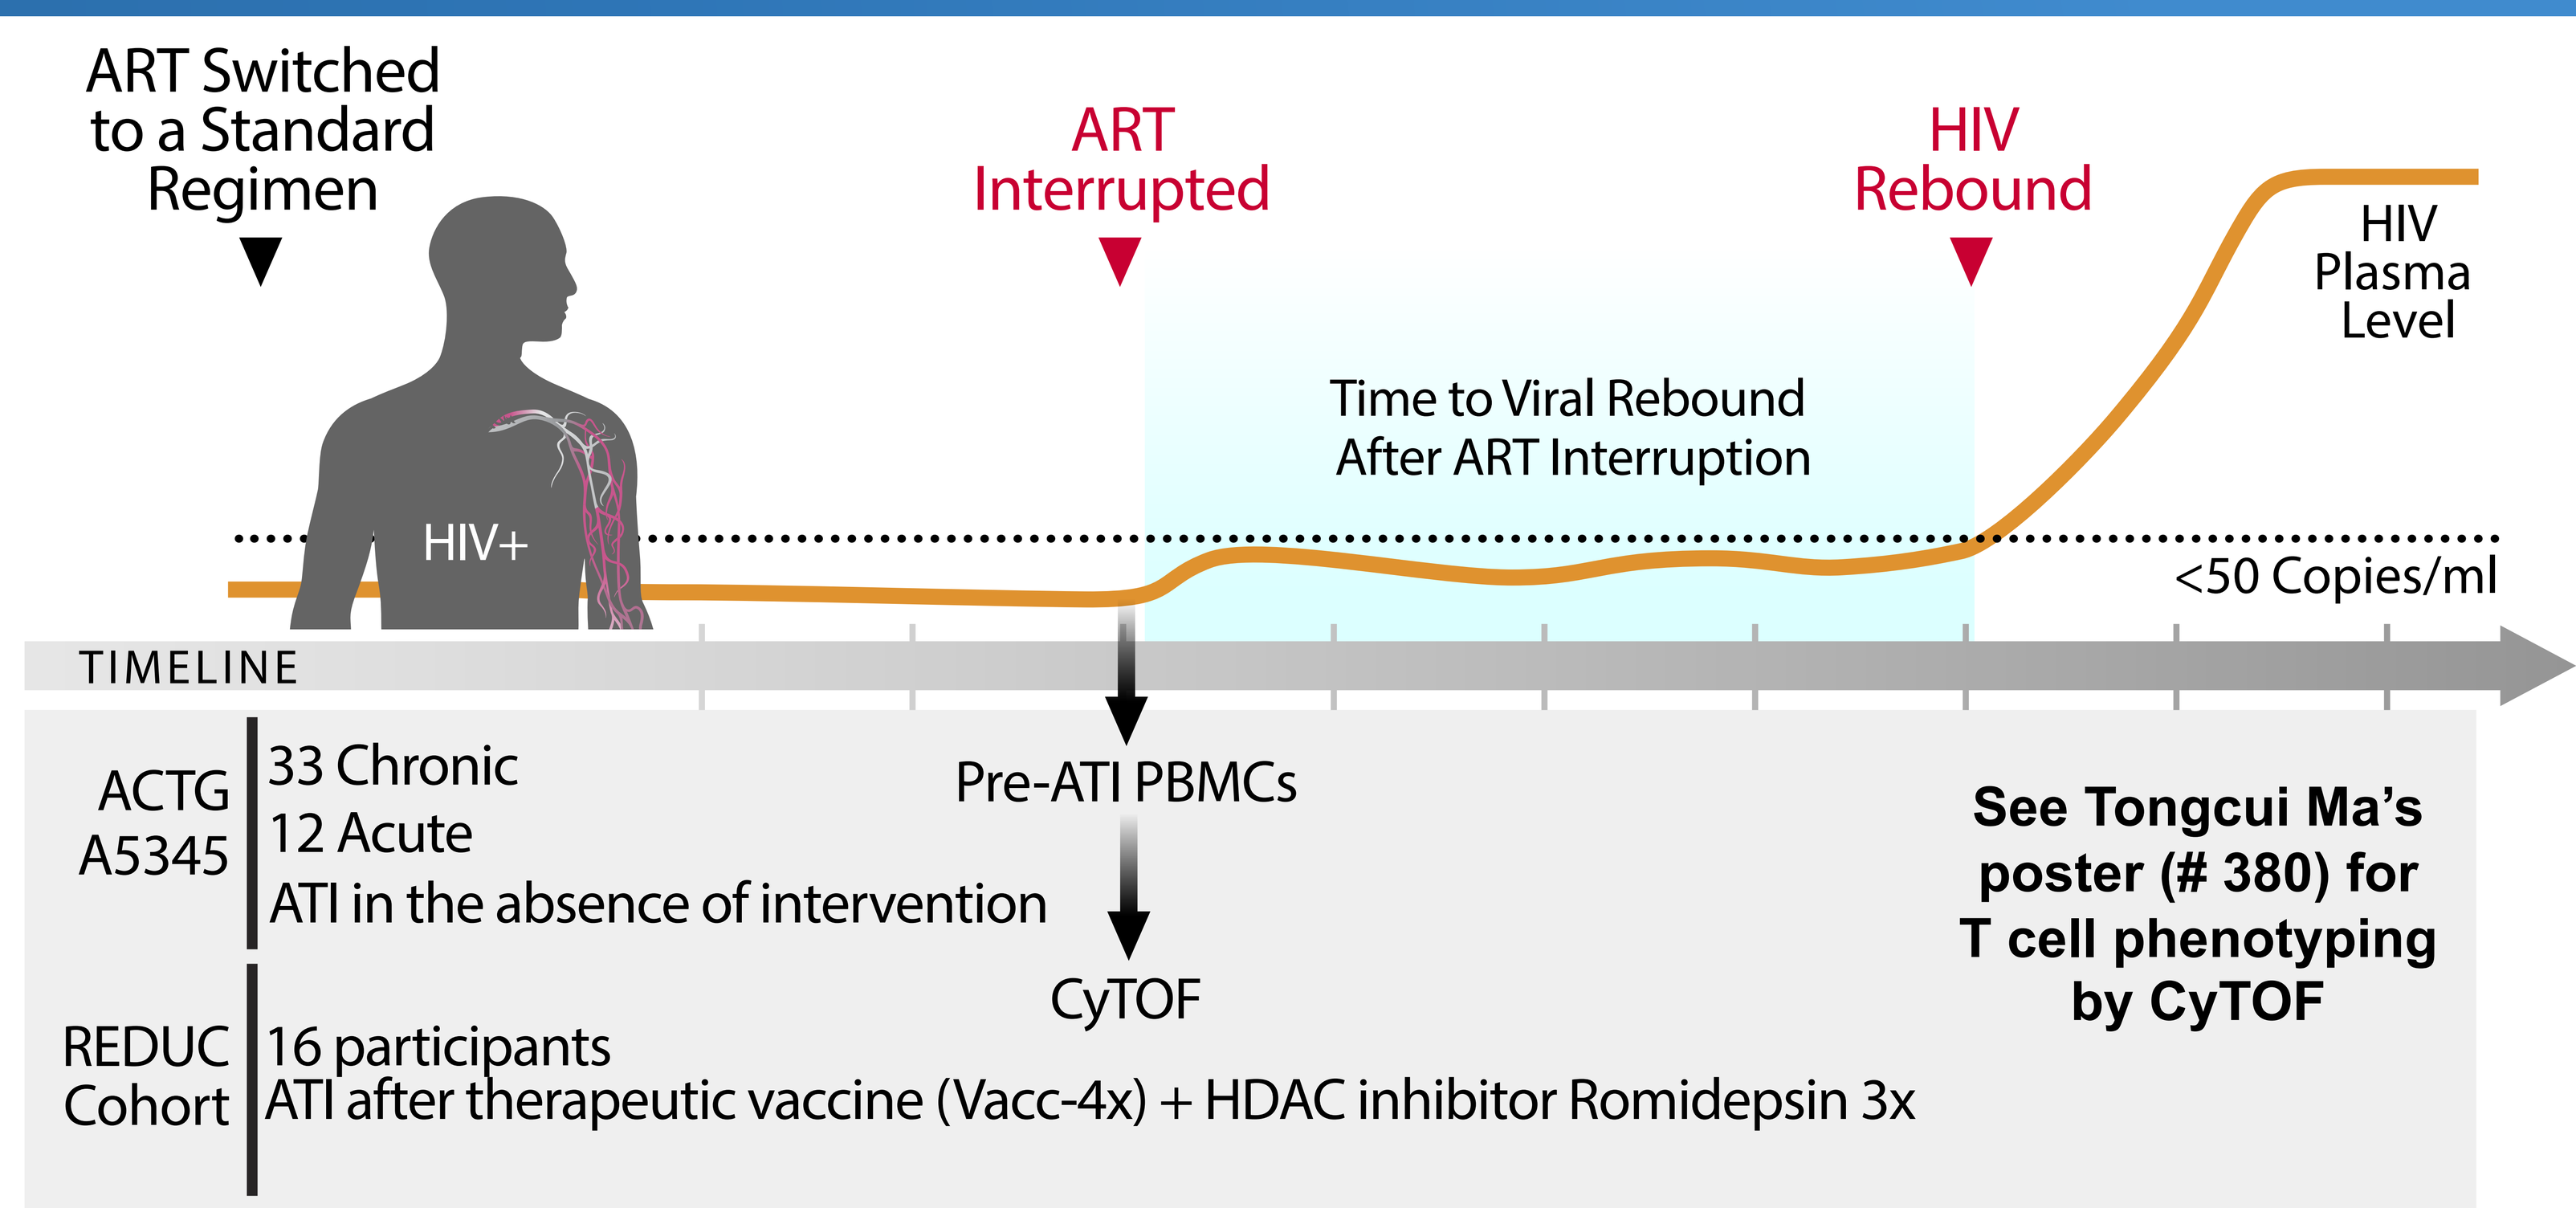
ART Switched to a Standard Regimen
ART Interrupted
HIV Rebound
HIV Plasma Level
Time to Viral Rebound After ART Interruption
HIV+
<50 Copies/ml
TIMELINE
ACTG A5345
33 Chronic
12 Acute
ATI in the absence of intervention
Pre-ATI PBMCs
CyTOF
REDUC Cohort
16 participants
ATI after therapeutic vaccine (Vacc-4x) + HDAC inhibitor Romidepsin 3x
See Tongcui Ma’s poster (# 380) for T cell phenotyping by CyTOF

- We analyzed samples from two cohorts (Fig. 1, above): the ACTG A5345 cohort, which consisted of 33 chronic- and 12 acute-treated participants that underwent ATI in the absence of intervention, and the REDUC cohort, which consisted of 17 participants that received therapeutic vaccination (Vacc4x) with the HDAC inhibitor Romidepsin prior to ATI.
- We phenotyped pre-ATI PBMCs using an NK cell CyTOF panel (Table 1, below) that includes markers of differentiation and activation states, homing receptors, inhibitory and activating receptors, and intracellular factors.
- We performed unsupervised clustering analyses to define NK cell clusters, optimized the number of clusters using a leave-one-out cross-validation model, and looked for associations with time-to-rebound, HIV reservoir size as determined by IPDA, and timing of treatment initiation (chronic vs. acute).

Table 1

Lineage Markers	Phenotyping Markers			
	Activating Receptors:	Inhibitory Receptors:	Differentiation State:	Homing Receptors
CD3	2B4	TIM-3	CD57	CXCR6
CD4	NKG2D	LAG-3	CD25	CXCR5
CD8	NKp46	TIGIT	CD38	CD69
CD14	NTB-A	PD-1	CD2	CD49a
CD19	NKp30	LILRB1	CD62L	
CD33	NKB1	KIR2DL1	NKG2C	
CD7	DNAM-1	Siglec-7	Ki67	
CD56	HLA-DR	NKG2A	TCF1	
CD16		KIR2DL2/3		

FUNDING: NIH K99 HD110319 and Gilead HIV Cure Mentored Scientist Award UCSF AIDS Research Institute (AFG); **P01 AI131374**, R01 AI147777, UM1 AI164559, UM1 AI164567, R01 DK131526, P01 AI69606, R21 AI172060 (NRR)

RESULTS

(2)

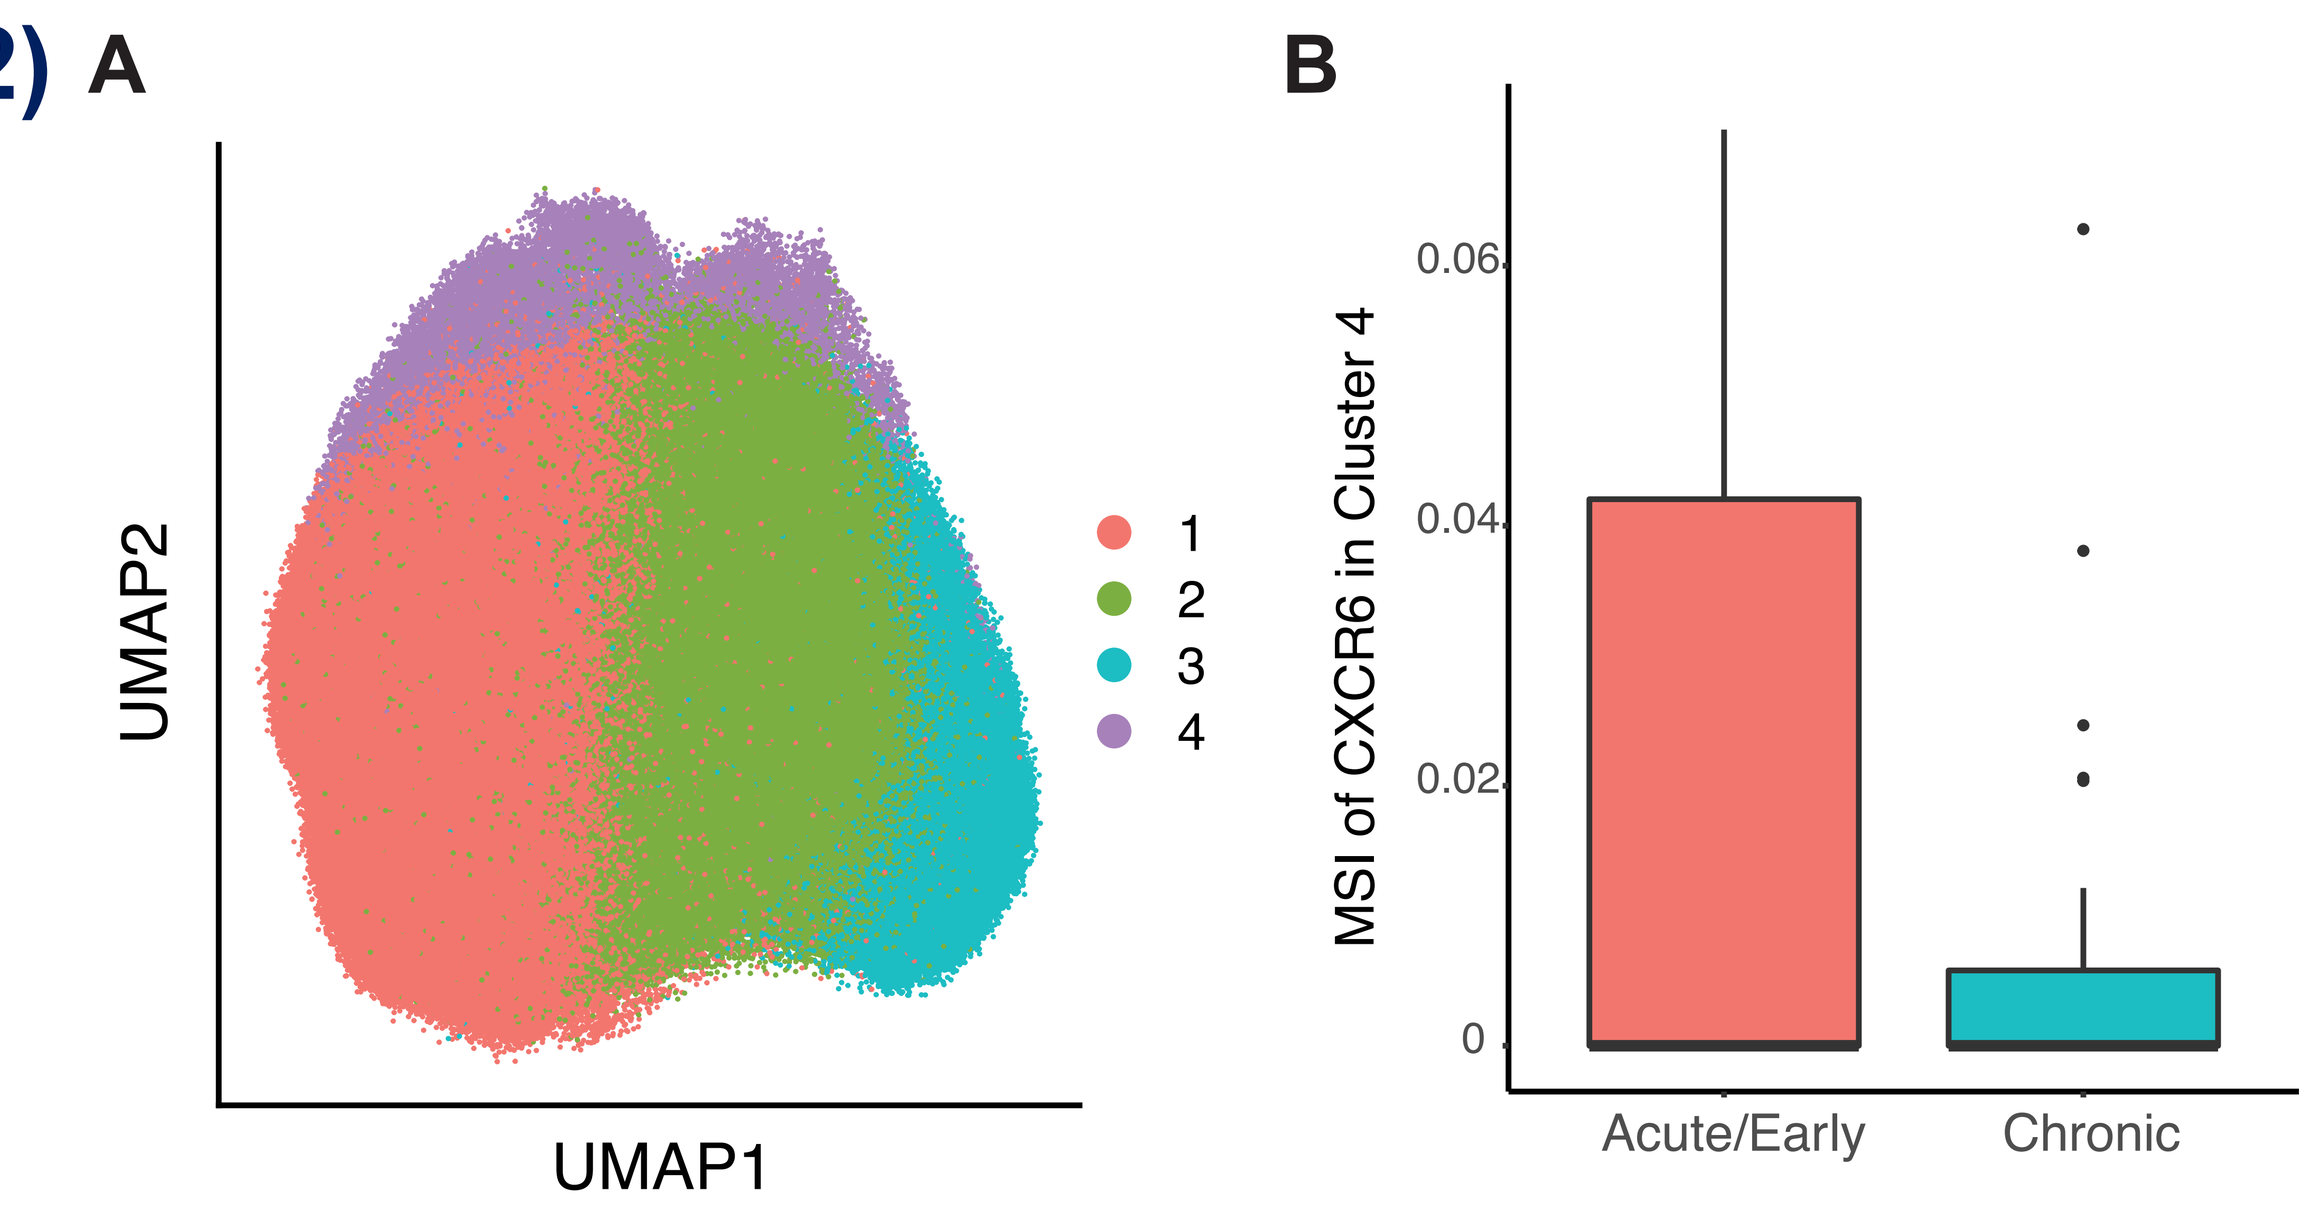
A
UMAP2
UMAP1
1
2
3
4
B
MSI of CXCR6 in Cluster 4
Acute/Early
Chronic

A cluster of NK cells (cluster 4) from ACTG A5345 is more likely to express memory NK cell antigen CXCR6 when the individuals were acute- vs. chronic-treated.

A) NK cell clustering of CyTOF data from ART-suppressed individuals from the ACTG A5345 cohort identified 4 clusters. This cohort consisted of 33 chronic- and 12 acute-treated participants that subsequently underwent ATI.

B) Acute-treated participants were more likely to have CXCR6-expressing NK cells in Cluster 4 ($p<0.01$). CXCR6 is a marker of memory NK cells.

(3)

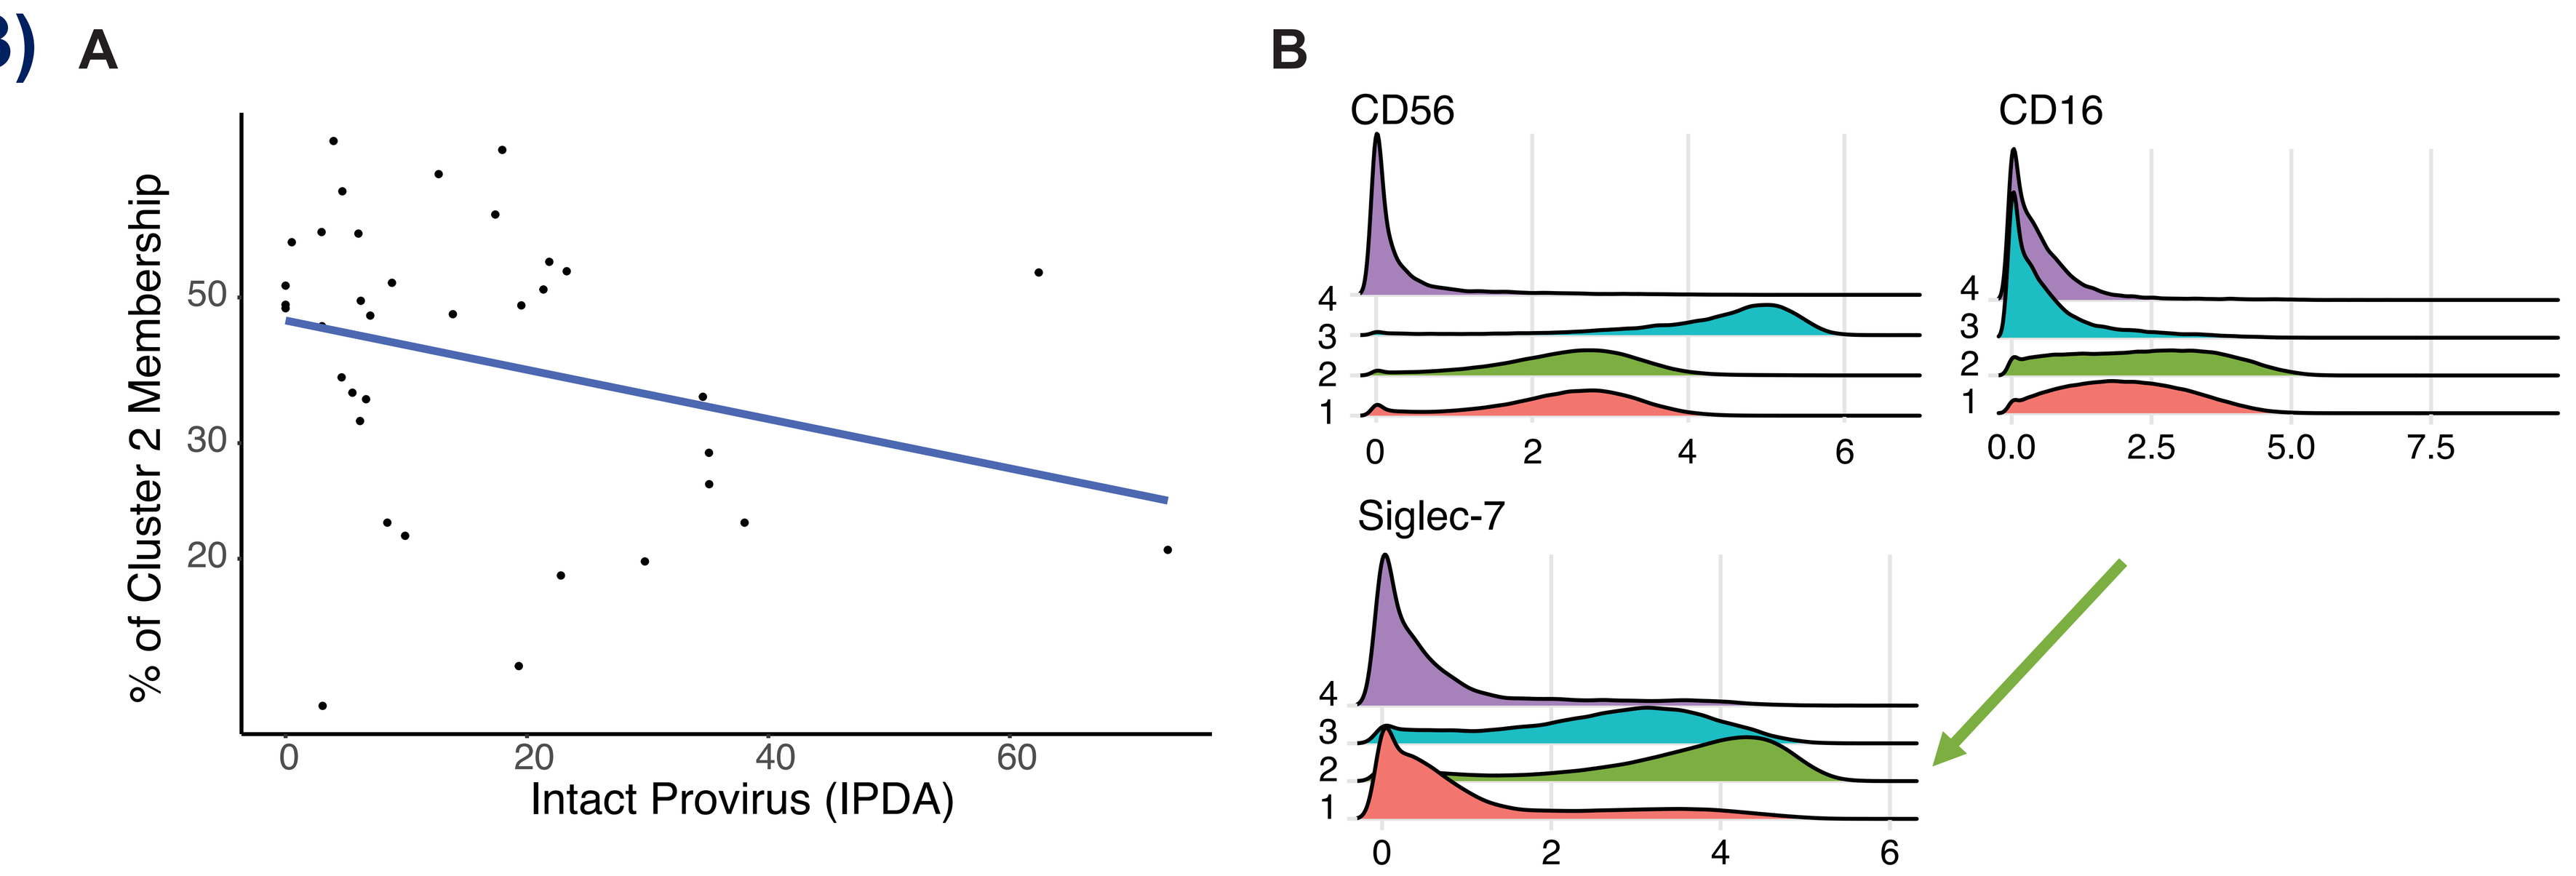
A
% of Cluster 2 Membership
Intact Provirus (IPDA)
B
CD56
CD16
Siglec-7

Siglec-7+ CD56dimCD16+ NK cells are associated with a lower frequency of intact provirus in ACTG A5345.

A) NK cell cluster 2 (in green) was associated with a lower frequency of intact provirus as measured by IPDA.

B) Cluster 2 is comprised primarily of CD56dimCD16+ NK cells, and expresses high levels of the inhibitory receptor Siglec-7, which is associated with functional NK responses.

(4)

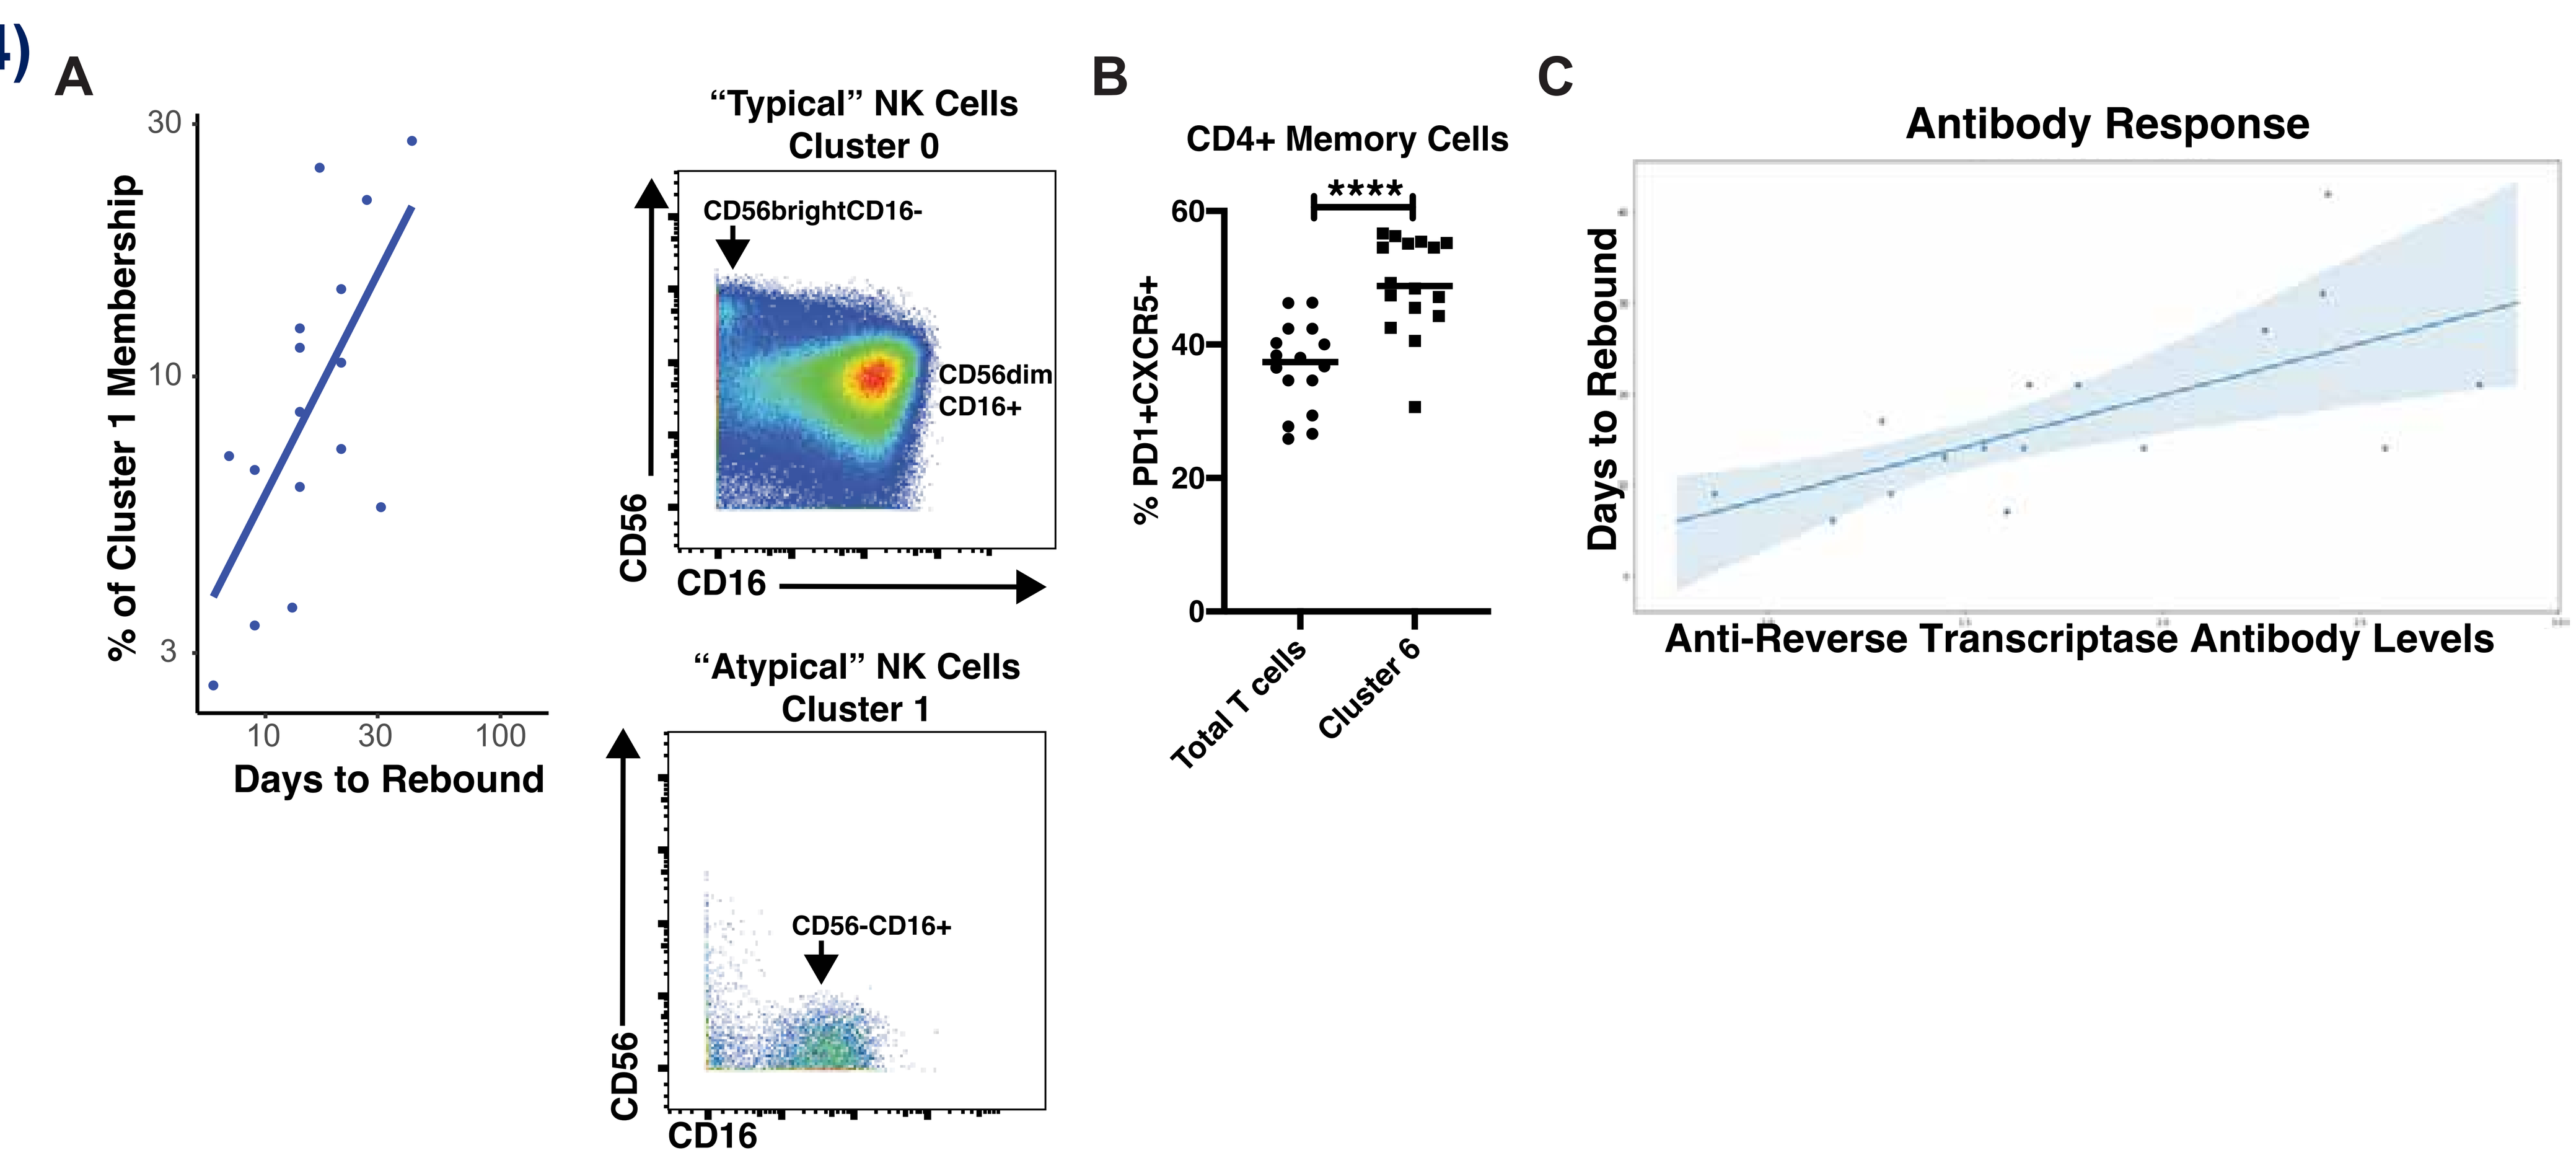
A
% of Cluster 1 Membership
Days to Rebound
“Typical” NK Cells
Cluster 0
CD56brightCD16-
CD56dim
CD16+
CD56
CD16
“Atypical” NK Cells
Cluster 1
CD56-CD16+
B
CD4+ Memory Cells
% PD1+CXCR5+

Total T cells
Cluster 6
C
Antibody Response
Days to Rebound
Anti-Reverse Transcriptase Antibody Levels

NK, T cell, and antibody features are associated with increased time-to-rebound in REDUC.

Increased time-to-rebound in the REDUC cohort was associated with increased frequencies of:

A) “Atypical” CD56-CD16+ NK cells, B) CD4+ Tfh cells, and C) anti HIV RT antibody levels.

From prior studies (Bradley et al., 2018 *Cell*): CD56(high)CD16- and CD56(dim)CD16+ NK cells can reduce CD4+ Tfh availability and decrease B cell antibody responses. By contrast, by being more dysfunctional, “atypical” CD56-CD16+ NK cells poorly regulate CD4+ Tfh responses and associate with lower bnAb levels.

CONCLUSIONS / PERSPECTIVES

- Acute-treated participants in ACTG A5345 were more likely to have a specific subset of NK cells expressing high levels of the memory NK cell antigen CXCR6. Therefore, acute-treated individuals, which exhibit longer time-to-rebound after ATI (Li et al., 2022 *Clin Infect Dis*), may retain a more functional memory NK cell response which can help control viremia.
- A cluster of CD56^{dim}CD16+ Siglec-7+ NK cells was associated with a lower frequency of intact provirus in ACTG A5345. As increased expression of Siglec-7 is associated with improved functional NK responses (Brunetta et al., 2009 *Blood*; Shao et al., 2016 *Scand J Immunol*; Zhao et al., 2020 *PLoS ONE*), these cells may help eliminate infected cells harboring genome-intact provirus.
- Increased time-to-rebound in the REDUC cohort was associated with increased frequencies of “atypical” CD56-CD16+ NK cells, CD4+ Tfh cells, and anti-HIV RT antibody levels. These NK cells, which have been associated with bnAb production (Bradley et al., 2018 *Cell)*, may play a role in enabling a robust Tfh-mediated antibody response to control HIV replication upon ART interruption.

Thanks to all the participants and ACTG!!!