520

Impact of Baseline Factors on Virologic Response to bNAb VH3810109 (N6LS) in BANNER

Peter Leone,[1] Margaret Gartland,[1] Judah Abberbock,[2] Paul Wannamaker,[1] Beta Win,[3] Mark Baker,[3] Viviana Wilches,[2] Max Lataillade,[4] Jan Losos[1]

[1]ViiV Healthcare, Durham, NC, USA; [2]GSK, Collegeville, PA, USA; [3]GSK, Brentford, UK; [4]ViiV Healthcare, Branford, CT, USA

Introduction

- Broadly neutralizing antibodies (bNAbs) are being developed for long-acting HIV-1 therapy[1-9]
- VH3810109 (N6LS) is a CD4-bs antibody with broad and potent neutralization activity in vitro targeting the CD4 binding site of the HIV-1 envelope protein and is currently being evaluated in the phase 2a BANNER study
- Here we report the impact of baseline viral and participant factors on maximum viral load (VL) decline and time to virologic rebound after infusion of VH3810109 in part 1 of the BANNER study

Methods

- BANNER is a randomized, open-label, 2-part, multicenter study in ART-naive viremic (VL ≥5000 c/mL) adults to evaluate antiviral activity, safety, and pharmacokinetics (PK) of VH3810109
- In part 1, VH3810109 was evaluated during monotherapy after a single IV infusion of 40 mg/kg or 280 mg (~4 mg/kg) followed by 48 weeks of standard-of-care (SOC) ART (Figure 1)
 - Monotherapy duration was determined by either virologic non-response (VL decline <0.5 $\log_{10}$ c/mL by Day 11) or rebound (VL ≥1.0 $\log_{10}$ c/mL increase over nadir or return to <0.5 $\log_{10}$ c/mL from baseline)
 - Antibody sensitivity of pre-dose and rebound viruses was determined retrospectively using the PhenoSense® mAb assay (Monogram Biosciences, South San Francisco, CA); IC_{80} value of 50 µg/mL represents the highest concentration tested
 - At the end of part 1, a planned interim analysis was performed to evaluate virologic response, safety, and PK from the monotherapy and ongoing SOC periods in part 1 and to determine whether to proceed to a similarly designed part 2 as well as to determine dose selection for part 2
- Primary endpoints were plasma HIV-1 RNA maximum change from baseline during monotherapy and safety parameters

Figure 1. Study Design

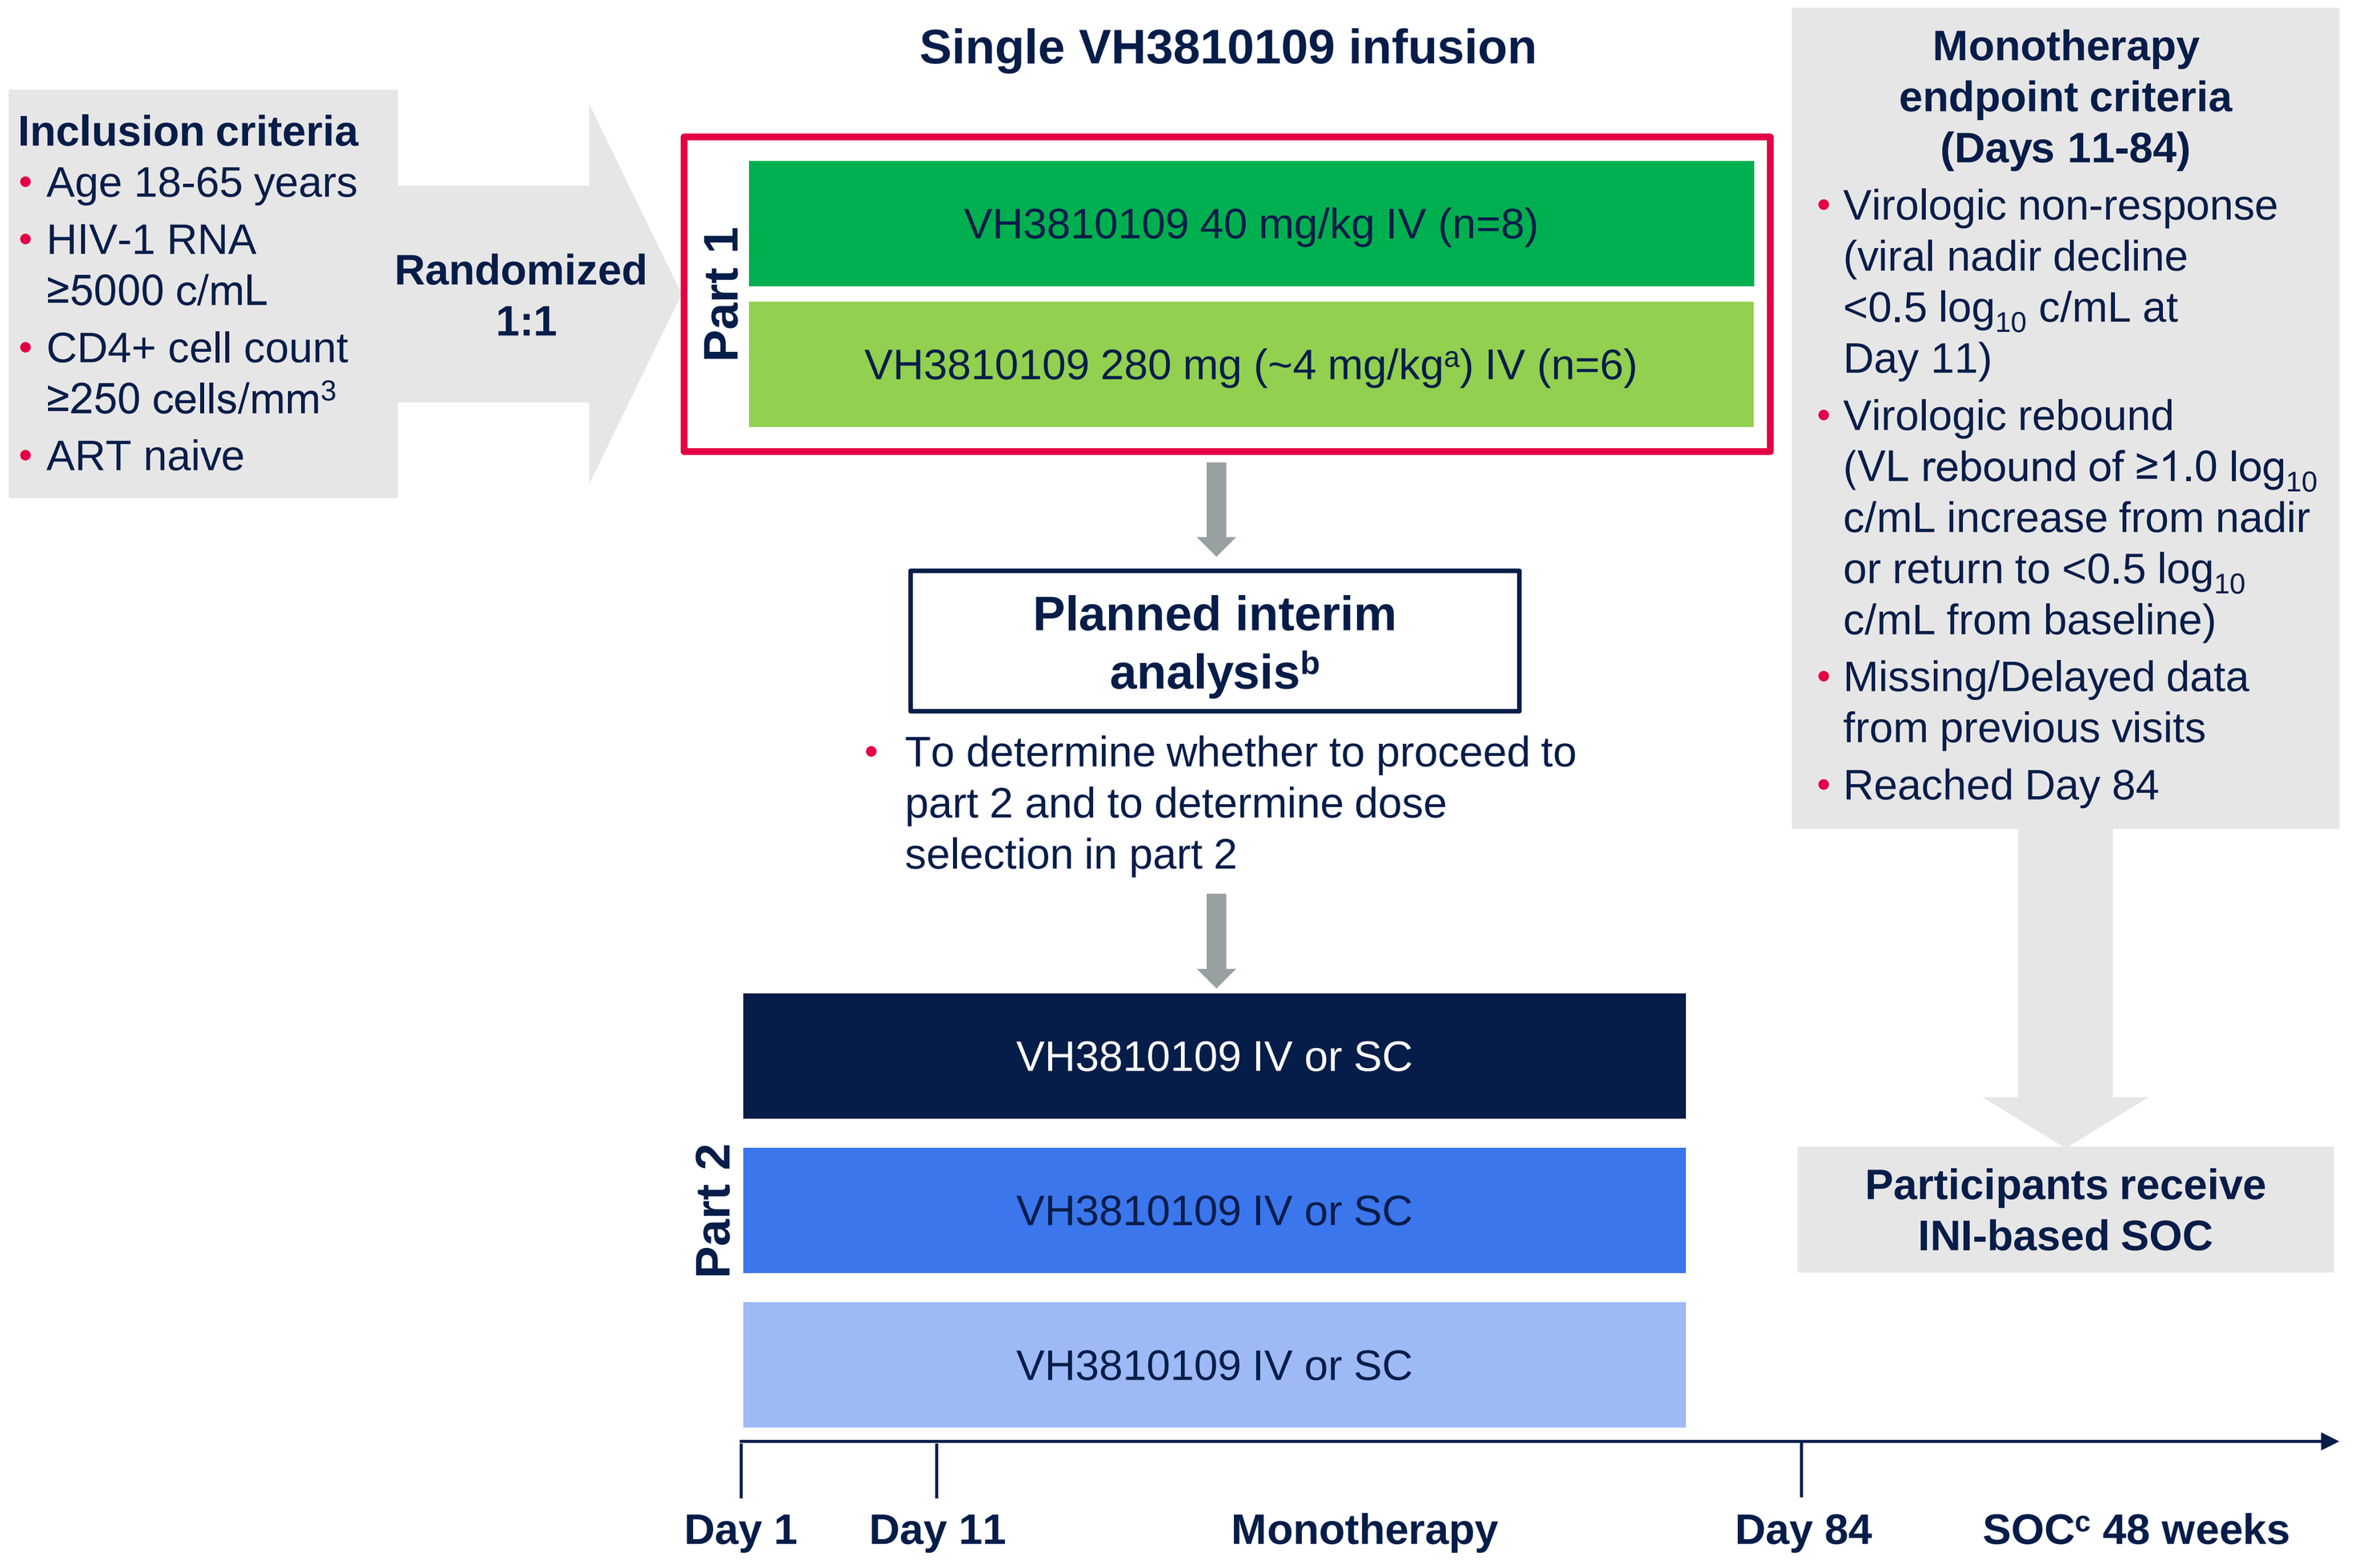
Single VH3810109 infusion
Inclusion criteria
Age 18-65 years
HIV-1 RNA ≥5000 c/mL
CD4+ cell count ≥250 cells/mm3
ART naive
Randomized 1:1
Part 1
VH3810109 40 mg/kg IV (n=8)
VH3810109 280 mg (~4 mg/kga) IV (n=6)
Planned interim analysisb
To determine whether to proceed to part 2 and to determine dose selection in part 2
Monotherapy endpoint criteria (Days 11-84)
Virologic non-response (viral nadir decline <0.5 log10 c/mL at Day 11)
Virologic rebound (VL rebound of ≥1.0 log10 c/mL increase from nadir or return to <0.5 log10 c/mL from baseline)
Missing/Delayed data from previous visits
Reached Day 84
Part 2
VH3810109 IV or SC
VH3810109 IV or SC
VH3810109 IV or SC
Participants receive INI-based SOC
Day 1
Day 11
Monotherapy
Day 84
SOCc 48 weeks

[a]For a 70-kg individual. [b]A planned interim analysis was performed to evaluate virologic response, safety, and PK from the monotherapy and ongoing SOC periods in part 1. [c]An SOC integrase inhibitor–based regimen (DTG/3TC) was provided at the end of the monotherapy periods in parts 1 and 2.

- VH3810109 at doses of 40 mg/kg and 280 mg was well tolerated and showed robust antiviral efficacy in people living with HIV
- Baseline viral sensitivity to VH3810109 and CD4+ cell count correlated with magnitude and duration of antiviral response

Results

Participants

- 14 participants enrolled in Part 1; 13 were male, 11 were of White/Caucasian/European heritage, and 10 were of Latinx ethnicity
 - Median (range) baseline VL was 4.31 (3.13-5.24) $\log_{10}$ c/mL, and median (range) baseline CD4+ cell count was 369 (190-700) cells/mm³

Virologic Response

- Virologic response was observed in 13 participants; median (range) viral nadir from baseline was −1.72 (−2.60 to −0.60) and −1.18 (−2.18 to −0.30) $\log_{10}$ c/mL for 40 mg/kg and 280 mg, respectively (Figure 2)

Figure 2. Virologic Response After VH3810109 Infusion

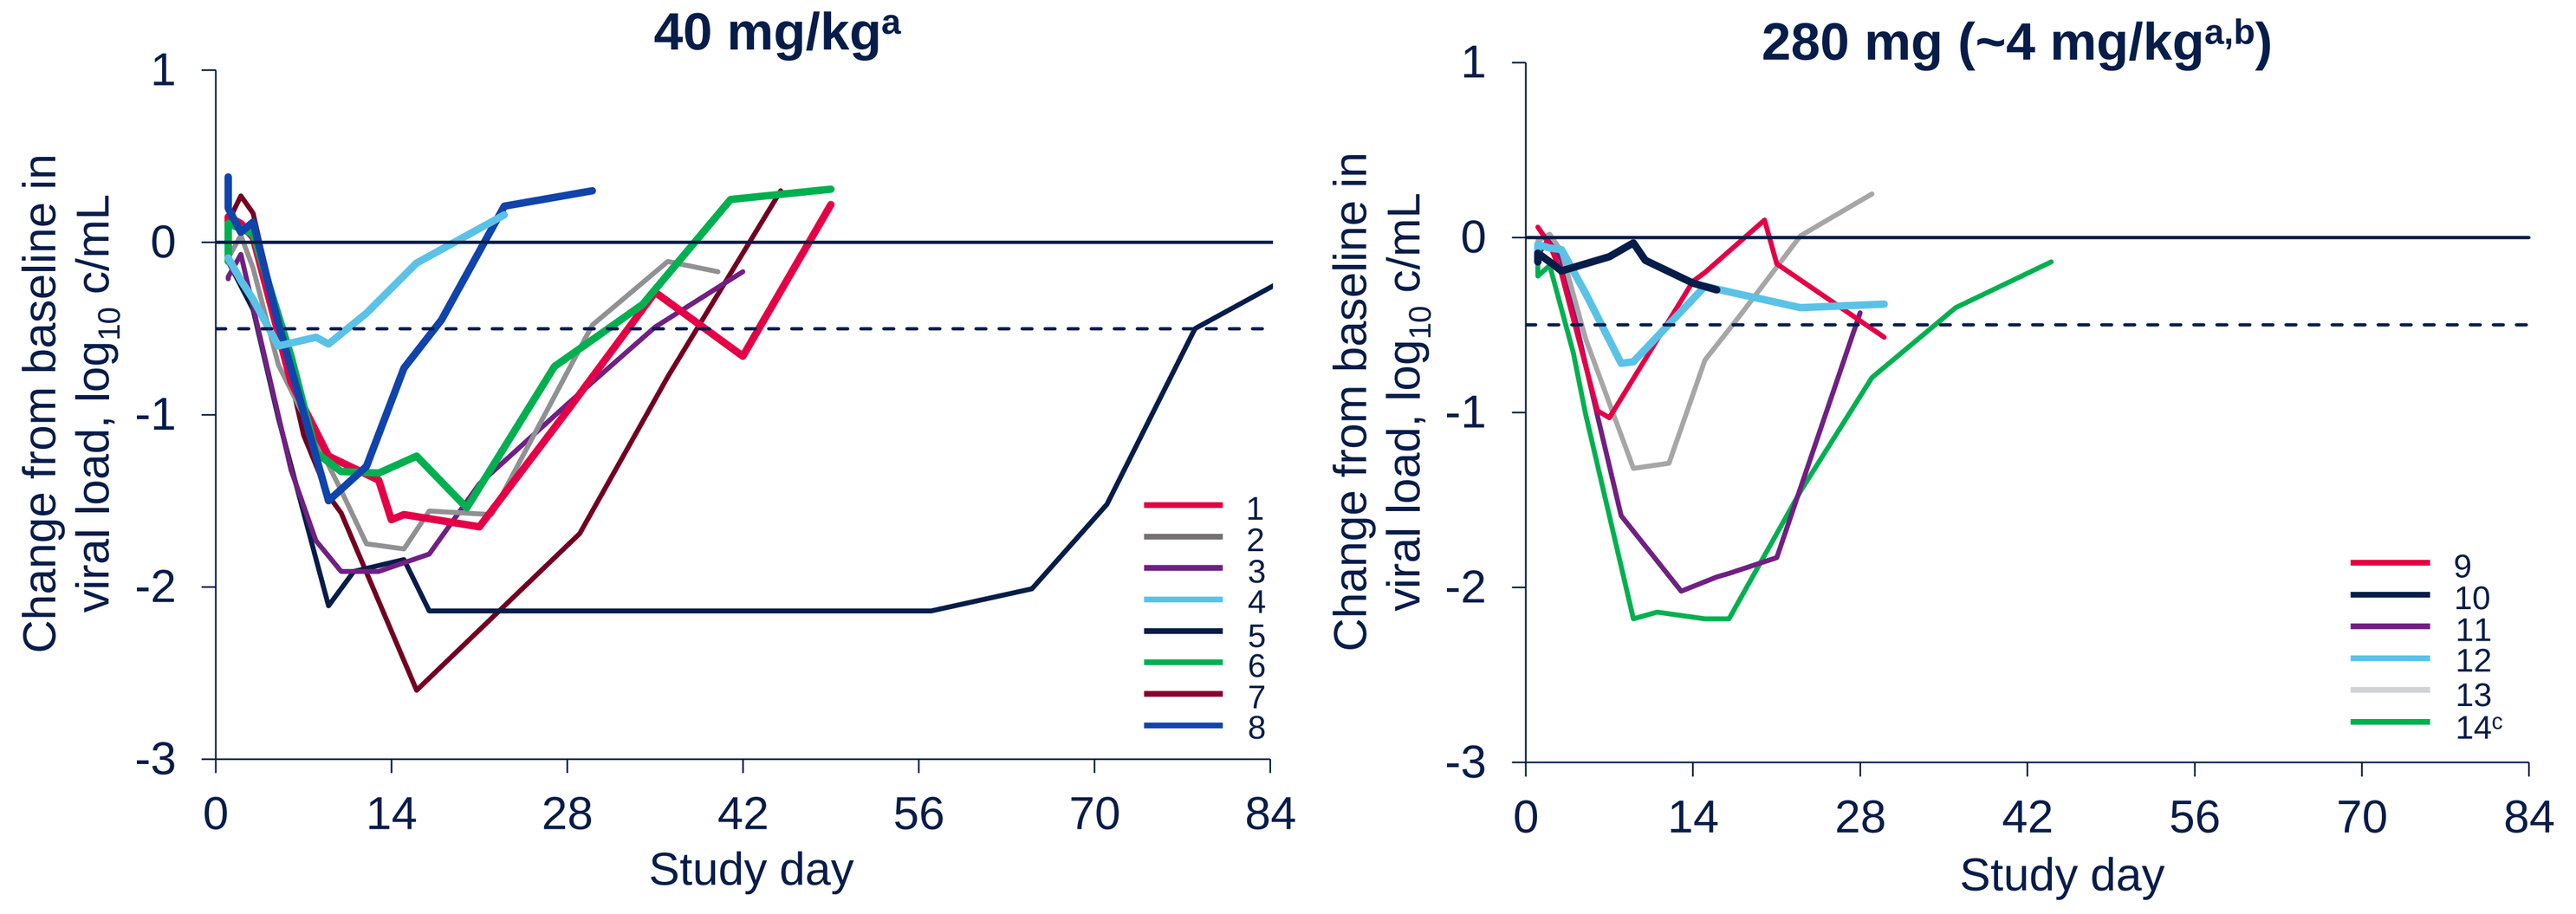
40 mg/kga
280 mg (~4 mg/kga,b)
Change from baseline in viral load, log10 c/mL
Study day
0
14
28
42
56
70
84
1
2
3
4
5
6
7
8
9
10
11
12
13
14c

Viral dynamic measures	VH3810109 40 mg/kg IV (n=8)	VH3810109 280 mg IV (~4 mg/kg[b]; n=6)
Median (range) VH3810109 IC_{80} of pre-dose virus, µg/mL	0.51 (0.09-1.51)	0.36 (0.29-50)
Median (range) VH3810109 IC_{80} of rebound virus, µg/mL	3.17 (1.73-7.27) [n=7]	1.21 (0.51-50)
Median (range) viral nadir from baseline, $\log_{10}$ c/mL	−1.72 (−0.60, −2.60)	−1.18 (−0.30, −2.18)
Median (range) time to viral nadir, days	16 (5-21)	9 (7-16)
Maximum viral nadir from baseline, $\log_{10}$ c/mL	−2.60	−2.18
Median (range) time to viral rebound among responders, days	35 (12-78) [n=8]	18 (14-29) [n=5]

Solid line represents no change from baseline and dashed line represents virologic non-response (viral nadir decline <0.5 $\log_{10}$ c/mL at Day 11). [a]Each line represents an individual participant. [b]For a 70-kg individual. [c]Participant 14 is the only female participant in the study.

- In a post hoc analysis, baseline VH3810109 IC_{80} and CD4+ cell count were moderately correlated with maximum VL decline and time to viral rebound in both treatment groups (Figures 3-4)
 - The 2 participants with the highest baseline IC_{80}s (1.51 and >50 µg/mL) had the smallest VL declines (0.60 and 0.30 $\log_{10}$ c/mL) and the shortest times to rebound (12 days and virologic non-response)
 - The longest time to rebound (78 days) was observed in the participant with the lowest baseline IC_{80} (0.09 µg/mL)
- A weak correlation between lower baseline $\log_{10}$ HIV-1 RNA and increased virologic response was apparent only in the 280 mg dose group

Figure 3. Maximum VL Decline and Time to Rebound by (A, B) Baseline VH3810109 IC_{80}, (C, D) Baseline VL, and (E, F) Baseline CD4+ Cell Count

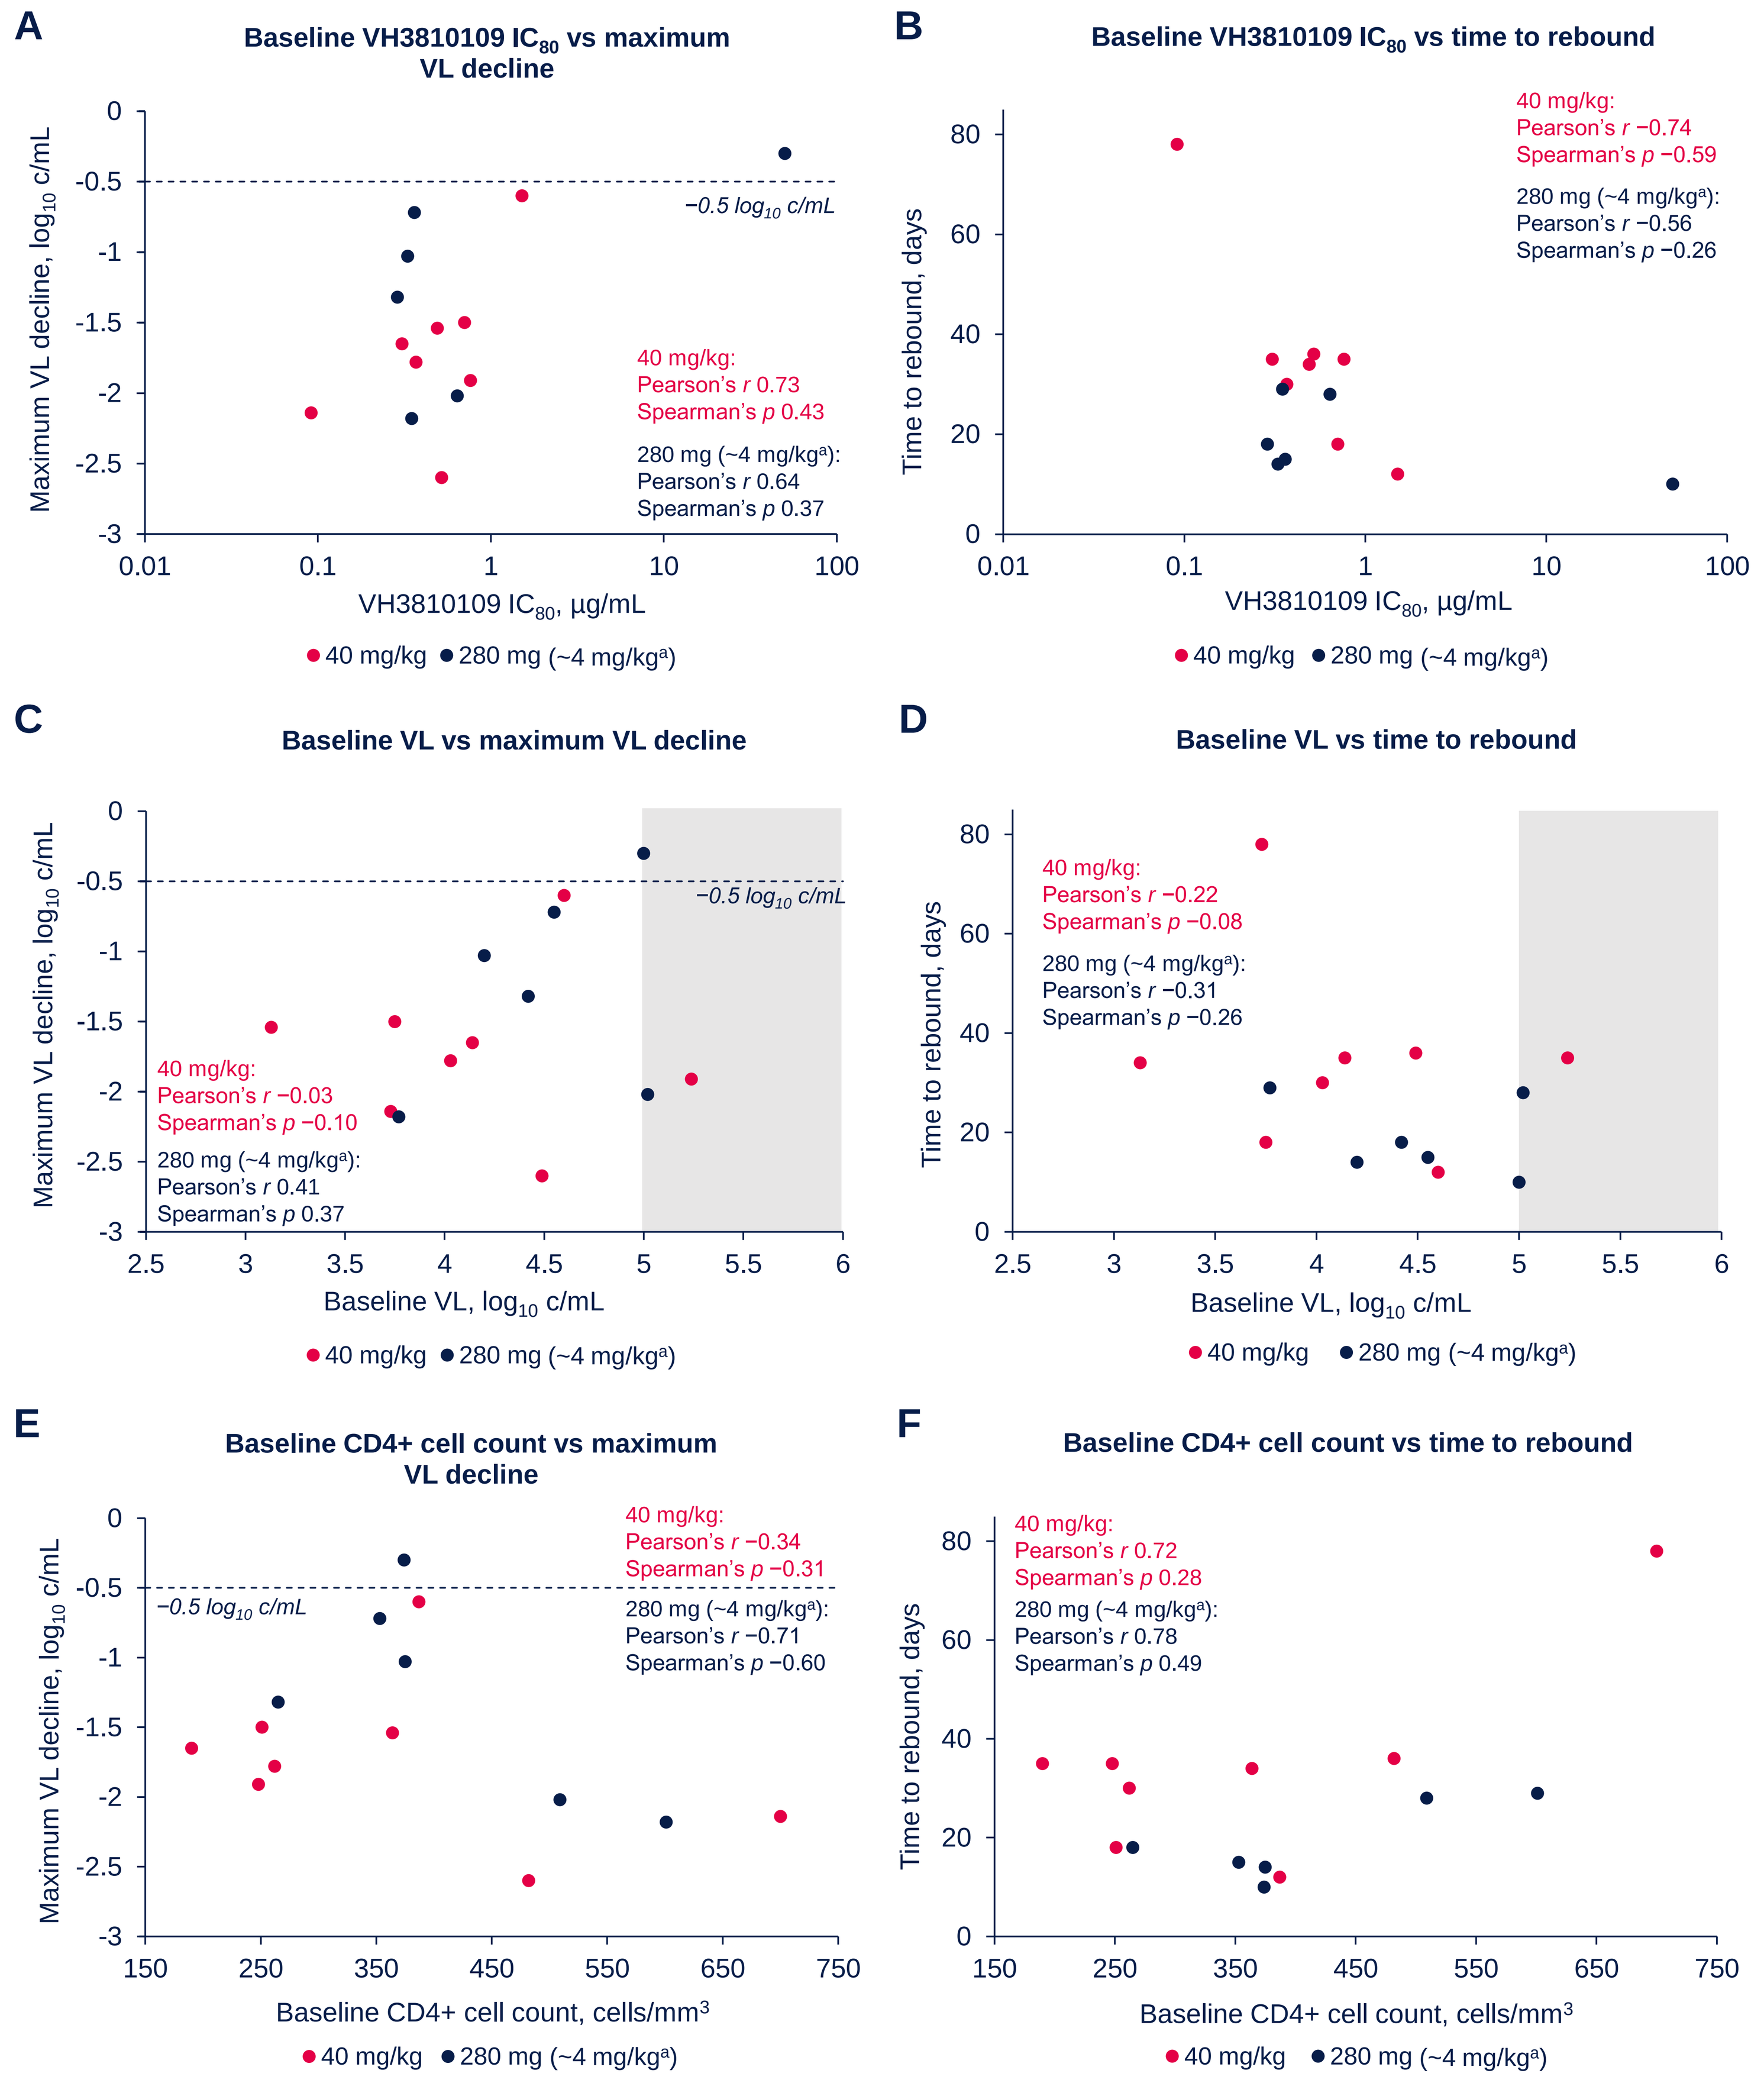
A
Baseline VH3810109 IC80 vs maximum VL decline
Maximum VL decline, log10 c/mL
VH3810109 IC80, µg/mL
−0.5 log10 c/mL
40 mg/kg:
Pearson's r 0.73
Spearman's p 0.43
280 mg (~4 mg/kga):
Pearson's r 0.64
Spearman's p 0.37
B
Baseline VH3810109 IC80 vs time to rebound
Time to rebound, days
40 mg/kg:
Pearson's r −0.74
Spearman's p −0.59
280 mg (~4 mg/kga):
Pearson's r −0.56
Spearman's p −0.26
C
Baseline VL vs maximum VL decline
Baseline VL, log10 c/mL
40 mg/kg:
Pearson's r −0.03
Spearman's p −0.10
280 mg (~4 mg/kga):
Pearson's r 0.41
Spearman's p 0.37
D
Baseline VL vs time to rebound
40 mg/kg:
Pearson's r −0.22
Spearman's p −0.08
280 mg (~4 mg/kga):
Pearson's r −0.31
Spearman's p −0.26
E
Baseline CD4+ cell count vs maximum VL decline
Baseline CD4+ cell count, cells/mm3
40 mg/kg:
Pearson's r −0.34
Spearman's p −0.31
280 mg (~4 mg/kga):
Pearson's r −0.71
Spearman's p −0.60
F
Baseline CD4+ cell count vs time to rebound
40 mg/kg:
Pearson's r 0.72
Spearman's p 0.28
280 mg (~4 mg/kga):
Pearson's r 0.78
Spearman's p 0.49
40 mg/kg
280 mg (~4 mg/kga)

Each dot represents an individual participant. Horizontal dashed line represents virologic non-response (viral nadir decline <0.5 $\log_{10}$ c/mL at Day 11). Gray shading in panels C and D indicates participants with baseline VL >100,000 c/mL. [a]For a 70-kg individual.

Figure 4. Post Hoc Analysis of VH3810109 Sensitivity, Plasma VL, and CD4+ Cell Count vs Maximum VL Decline and Time to Rebound for (A) 40 mg/kg and (B) 280 mg (~4 mg/kg[a]) Sorted by Greatest Maximum VL Decline

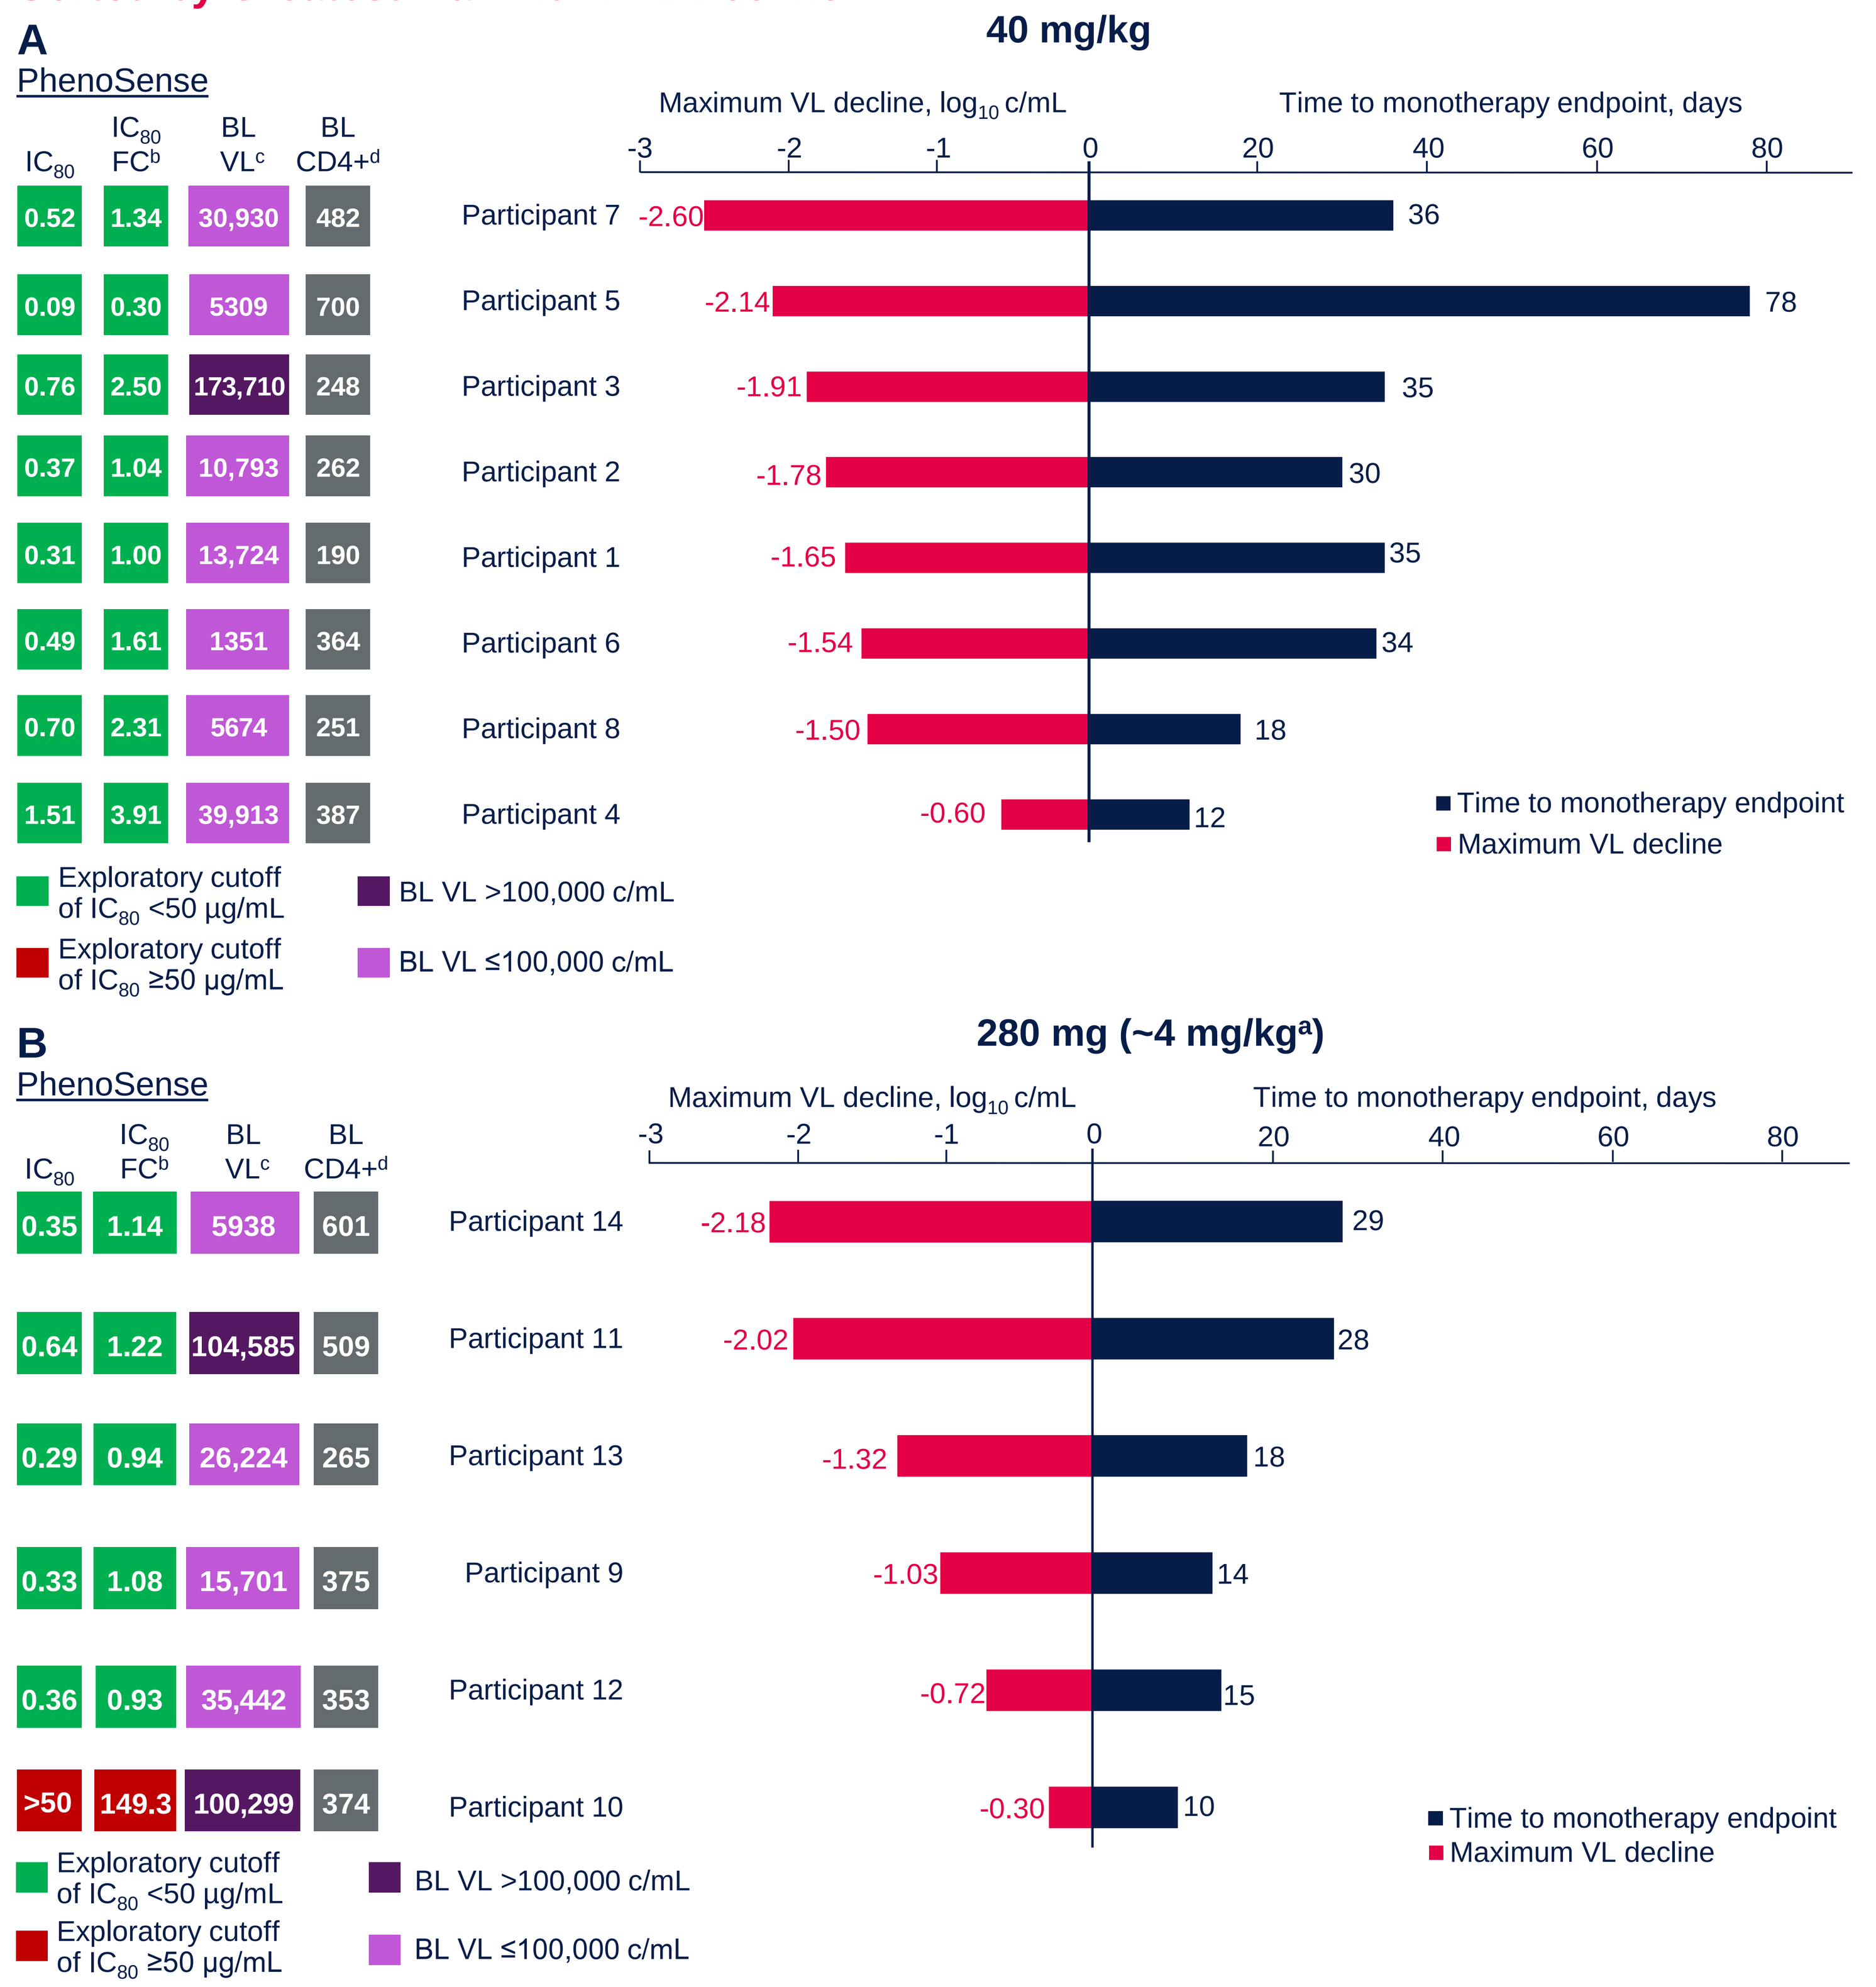
A
40 mg/kg
PhenoSense
IC80
IC80 FCb
BL VLc
BL CD4+d
Maximum VL decline, log10 c/mL
Time to monotherapy endpoint, days
0.52 1.34 30,930 482 Participant 7 -2.60 36
0.09 0.30 5309 700 Participant 5 -2.14 78
0.76 2.50 173,710 248 Participant 3 -1.91 35
0.37 1.04 10,793 262 Participant 2 -1.78 30
0.31 1.00 13,724 190 Participant 1 -1.65 35
0.49 1.61 1351 364 Participant 6 -1.54 34
0.70 2.31 5674 251 Participant 8 -1.50 18
1.51 3.91 39,913 387 Participant 4 -0.60 12
Time to monotherapy endpoint
Maximum VL decline
Exploratory cutoff of IC80 <50 µg/mL
Exploratory cutoff of IC80 ≥50 µg/mL
BL VL >100,000 c/mL
BL VL ≤100,000 c/mL
B
280 mg (~4 mg/kga)
0.35 1.14 5938 601 Participant 14 -2.18 29
0.64 1.22 104,585 509 Participant 11 -2.02 28
0.29 0.94 26,224 265 Participant 13 -1.32 18
0.33 1.08 15,701 375 Participant 9 -1.03 14
0.36 0.93 35,442 353 Participant 12 -0.72 15
>50 149.3 100,299 374 Participant 10 -0.30 10

Monotherapy continued until ~Day 11 for all participants, regardless of virologic response. [a]For a 70-kg individual. [b]IC_{80} fold-change normalized to an in-assay reference virus. [c]Copies/mL. [d]Cells/mm³.

Safety

- Overall, 35 adverse events (AEs) were reported by 9 of 14 participants (40 mg/kg group, n=6; 280 mg group, n=3), with no grade ≥3 AEs or serious AEs reported
- All drug-related AEs and injection site reactions were grade 1

Conclusions

- In BANNER part 1, a single IV infusion of VH3810109 was well tolerated, with few drug-related AEs and robust antiviral efficacy at both doses studied
- In this small study, baseline viral sensitivity to VH3810109 and baseline CD4+ cell count correlated with magnitude and duration of antiviral response
- However, antiviral response to VH3810109 may, in addition, be multifactorial and several factors may impact virologic outcome, including pre-treatment VL, serum antibody concentration, and an individual's inherent control of viral replication
- BANNER part 2 is ongoing to evaluate alternate dosing options and modalities for VH3810109

Acknowledgments: We thank the study participants, their families and caregivers, investigators and site staff who participated in the study, and the ViiV Healthcare and GSK study team members. We would also like to acknowledge the contributions of Darren Bentley. Editorial assistance and graphic design support for this poster were provided under the direction of the authors by MedThink SciCom and funded by ViiV Healthcare.

References: **1.** Bar et al. *N Engl J Med*. 2016;375:2037-2050. **2.** Caskey. *Curr Opin HIV AIDS*. 2020;15:49-55. **3.** Doria-Rose et al. *J Virol*. 2015;90:76-91. **4.** Kwon et al. *J Virol*. 2016;90:5899-5914. **5.** Mouquet et al. *Proc Natl Acad Sci U S A*. 2012;109:E3268-E3277. **6.** Scheid et al. *Science*. 2011;333:1633-1637. **7.** Walker et al. *Nature*. 2011;477:466-470. **8.** Kwon et al. *Retrovirology*. 2012;9(suppl 2):O34. **9.** Huang et al. *Immunity.* 2016;45:1108-1121.

Conference on Retroviruses and Opportunistic Infections; February 19-22, 2023; Virtual and Seattle, WA

Presenting author: Peter Leone; peter.a.leone@viivhealthcare.com