Predictors of Post-Switch Viremia in Patients on Injectable Cabotegravir/Rilpivirine

#516

Stephanie Kenney[1], Nimish Patel[1], Lucas Hill[1]

[1]University of California San Diego Owen Clinic, San Diego, California, USA

UC San Diego Health

Background

- The introduction of long-acting injectable (LAI) cabotegravir/rilpivirine (CAB/RPV) has provided a highly efficacious alternative to daily oral medication for people with HIV (PWH) maintaining viral suppression rates equivalent to standard oral antiretrovrial therapy.[1]
- In the phase 3 clinical trials, predictors of virologic failure included baseline rilpivirine resistance, higher BMI, and HIV subtype A1/A6.[2]
- The frequency of virologic blips ranged from 3-13% and persistent low-level viremia 0-6% in the phase 3 clinical trials and did not predict virologic failure.[3]
- We aimed to evaluate predictors of detectable HIV viremia after switching to LAI CAB/RPV in the real-world setting as well as quantify the frequency of blips and persistent low-level viremia.

Methods

- A retrospective cohort study was performed among adult PWH receiving care at the University of California San Diego Owen Clinic.
- Inclusion criteria included treatment with LAI CAB/RPV for at least 3 months, availability of both pre- and on-treatment HIV RNA values, a HIV viral load of <200 copies/mL (cpm) at the time of switch to CAB/RPV, and at least one HIV RNA value collected greater than 21 days after start of LAI CAB/RPV.
- Pre- and post-switch viral load trends were categorized into 4 categories with pre-switch viral loads evaluated one year prior to switch and all post-switch viral loads evaluated:
 - Blip - at least one viral load 20-199cpm followed by the next viral load <20cpm
 - Persistent low-level viremia (pLLV) - at least 2 consecutive viral loads 20-199cpm
 - High viral load - at least one viral load ≥ 200cpm
 - Continuously suppressed - all viral loads <20cpm
- Demographics and laboratory values were extracted from the electronic medical record and the incidence of on treatment HIV viral loads greater than 20, 50, and 200cpm were evaluated both in those that were undetectable at the time of switch (<20cpm and <50cpm) as well as the entire cohort. Follow up time was calculated as the number of days from starting LAI CAB/RPV to the most recent HIV RNA.
- Univariable analyses were performed using Student's t-test, Mann Whitney U test, or chi-square. Multivariable logistic regression analyses were performed and variables were removed in a backwards, stepwise fashion.

Results

- Among 144 PWH, mean follow up after switching to LAI CAB/RPV was 278 days
- After switching to LAI CAB/RPV 59.7%, 81.9%, and 97.2% maintained all viral loads <20cpm, <50cpm, and <200cpm respectively

Table 1: Comparison of those with and without at least one HIV RNA≥20cpm after switching to LAI CAB/RPV

	Total (n=144)	At least one VL ≥20cpm (n=58)	All VL <20cpm (n=86)	p-value
Median Age (IQR)	44 (34-54)	45 (33-53)	41 (34-54)	0.99
Race (%)				0.51
White	62 (43.1)	27 (46.6)	35 (40.7)	
Black	27 (18.8)	13 (22.4)	14 (16.3)	
Asian	4 (2.8)	1 (1.7)	3 (3.5)	
Mixed/Other	48 (33.3)	16 (27.6)	32 (37.2)	
Unknown	3 (2.1)	1 (1.7)	2 (2.3)	
Ethnicity (%)				0.46
Hispanic	58 (40.3)	27 (46.6)	31 (36.0)	
Non-Hispanic	84 (58.3)	31 (53.4)	53 (61.6)	
Other/unknown	2 (1.4)	0 (0.0)	2 (2.3)	
SAB (%)				
Female	16 (11.1)	5 (8.6)	11 (12.8)	0.43
Gender identity (%)				0.51
Female	15 (10.4)	4 (6.9)	11 (12.8)	
Male	127 (88.2)	53 (91.4)	74 (86.0)	
Trans/non-binary	2 (1.4)	1 (1.7)	1 (1.2)	
CAB/RPV regimen (%)				0.67
Once monthly only	3 (2.1)	1 (1.7)	2 (2.3)	
Every 2 months only	73 (50.7)	27 (46.6)	46 (53.5)	
Switched from every 1 month to 2 month	68 (47.2)	30 (51.7)	38 (44.2)	
Completed oral lead-in (%)	125 (86.8)	49 (84.5)	76 (88.4)	0.50
Late injection or used oral bridge (%)	15 (10.4)	5 (8.6)	10 (11.6)	0.56
Median CD4 count (IQR)	745 (520-941)	809 (554-961)	680 (504-923)	0.08
Median BMI (IQR)	26.9 (24.3-30.2)	27.4 (24.6-31.7)	26.4 (24.2-30.0)	0.33
Pre-switch category (%)				**0.02**
Blip	26 (18.1)	12 (20.7)	14 (16.3)	
pLLV	26 (18.1)	17 (29.3)	9 (10.5)	
High viral load	6 (4.2)	2 (3.4)	4 (4.7)	
Always<20cpm	86 (59.7)	27 (46.6)	59 (68.6)	

IQR=interquartile range, SAB=sex assigned at birth, CAB=cabotegravir, RPV=rilpivirine, BMI=body mass index, pLLV=persistent low-level viremia

- 4 patients with viral load increase to ≥200cpm after switching to LAI CAB/RPV, 3 of which re-suppressed without intervention and one developed virologic failure.
- In the multivariable regression model, having pLLV pre-switch was associated with having at least one viral load ≥20cpm post switch [OR 3.55 (1.54-8.66), p=0.005]. When performing the analysis using a VL cut-off of 50cpm for all outcomes, results were similar in that pLLV was still a significant predictor of having a post-switch HIV VL>50cpm.
- Frequency of HIV RNA testing was similar in those with and without post switch viral loads ≥20cpm (5.7 tests/year of follow up vs. 5.3 tests/year of follow up, p=0.40).

Results (cont'd)

Figure 1: Time to event analyses comparing probability of HIV RNA remaining <20cpm in those with viral load <20cpm at time of switch

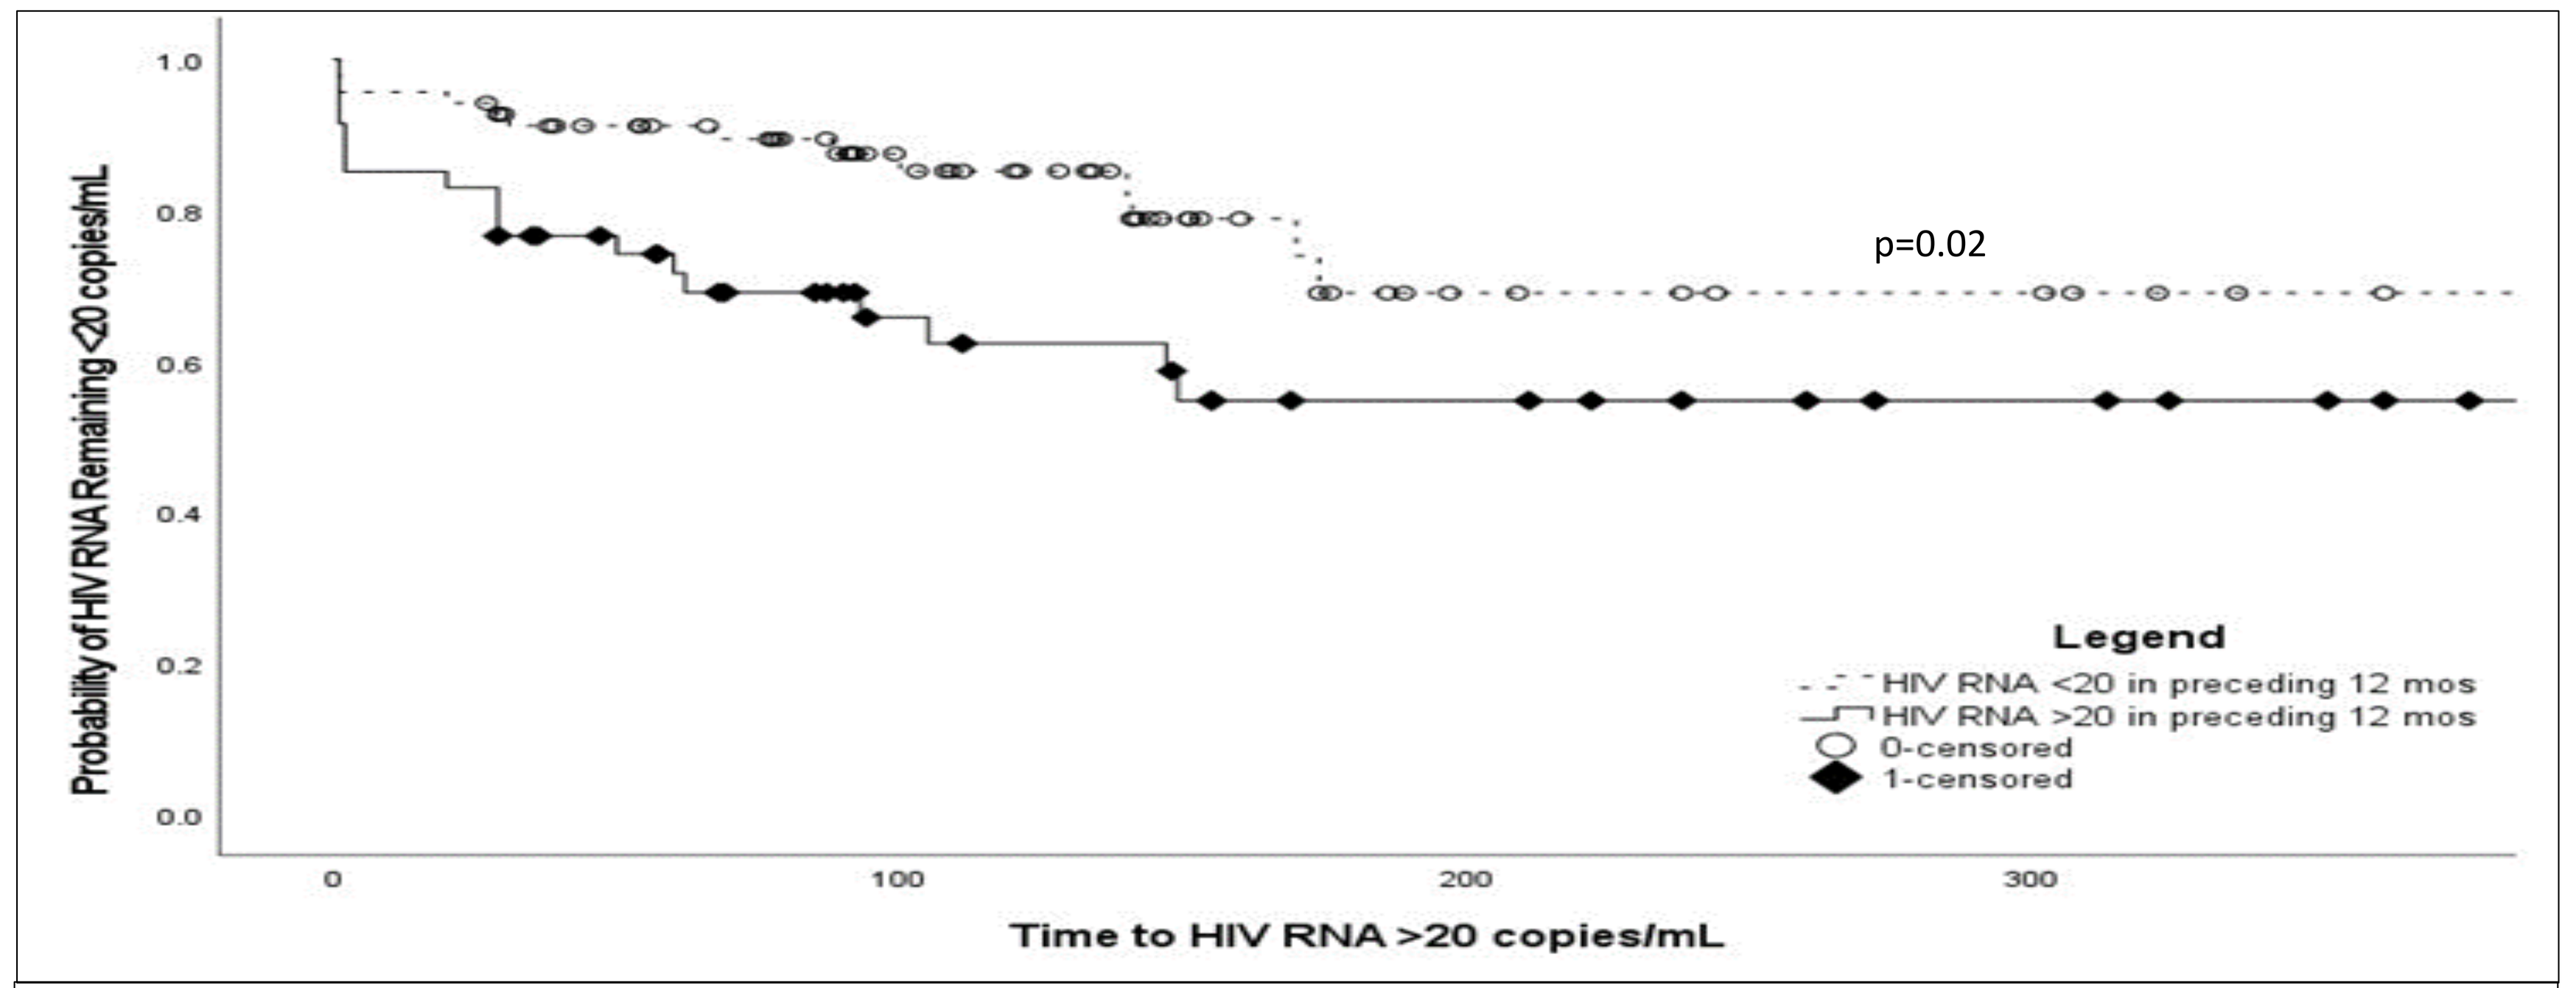
Probability of HIV RNA Remaining <20 copies/mL
1.0
0.8
0.6
0.4
0.2
0.0
0
100
200
300
Time to HIV RNA >20 copies/mL
p=0.02
Legend
HIV RNA <20 in preceding 12 mos
HIV RNA >20 in preceding 12 mos
0-censored
1-censored

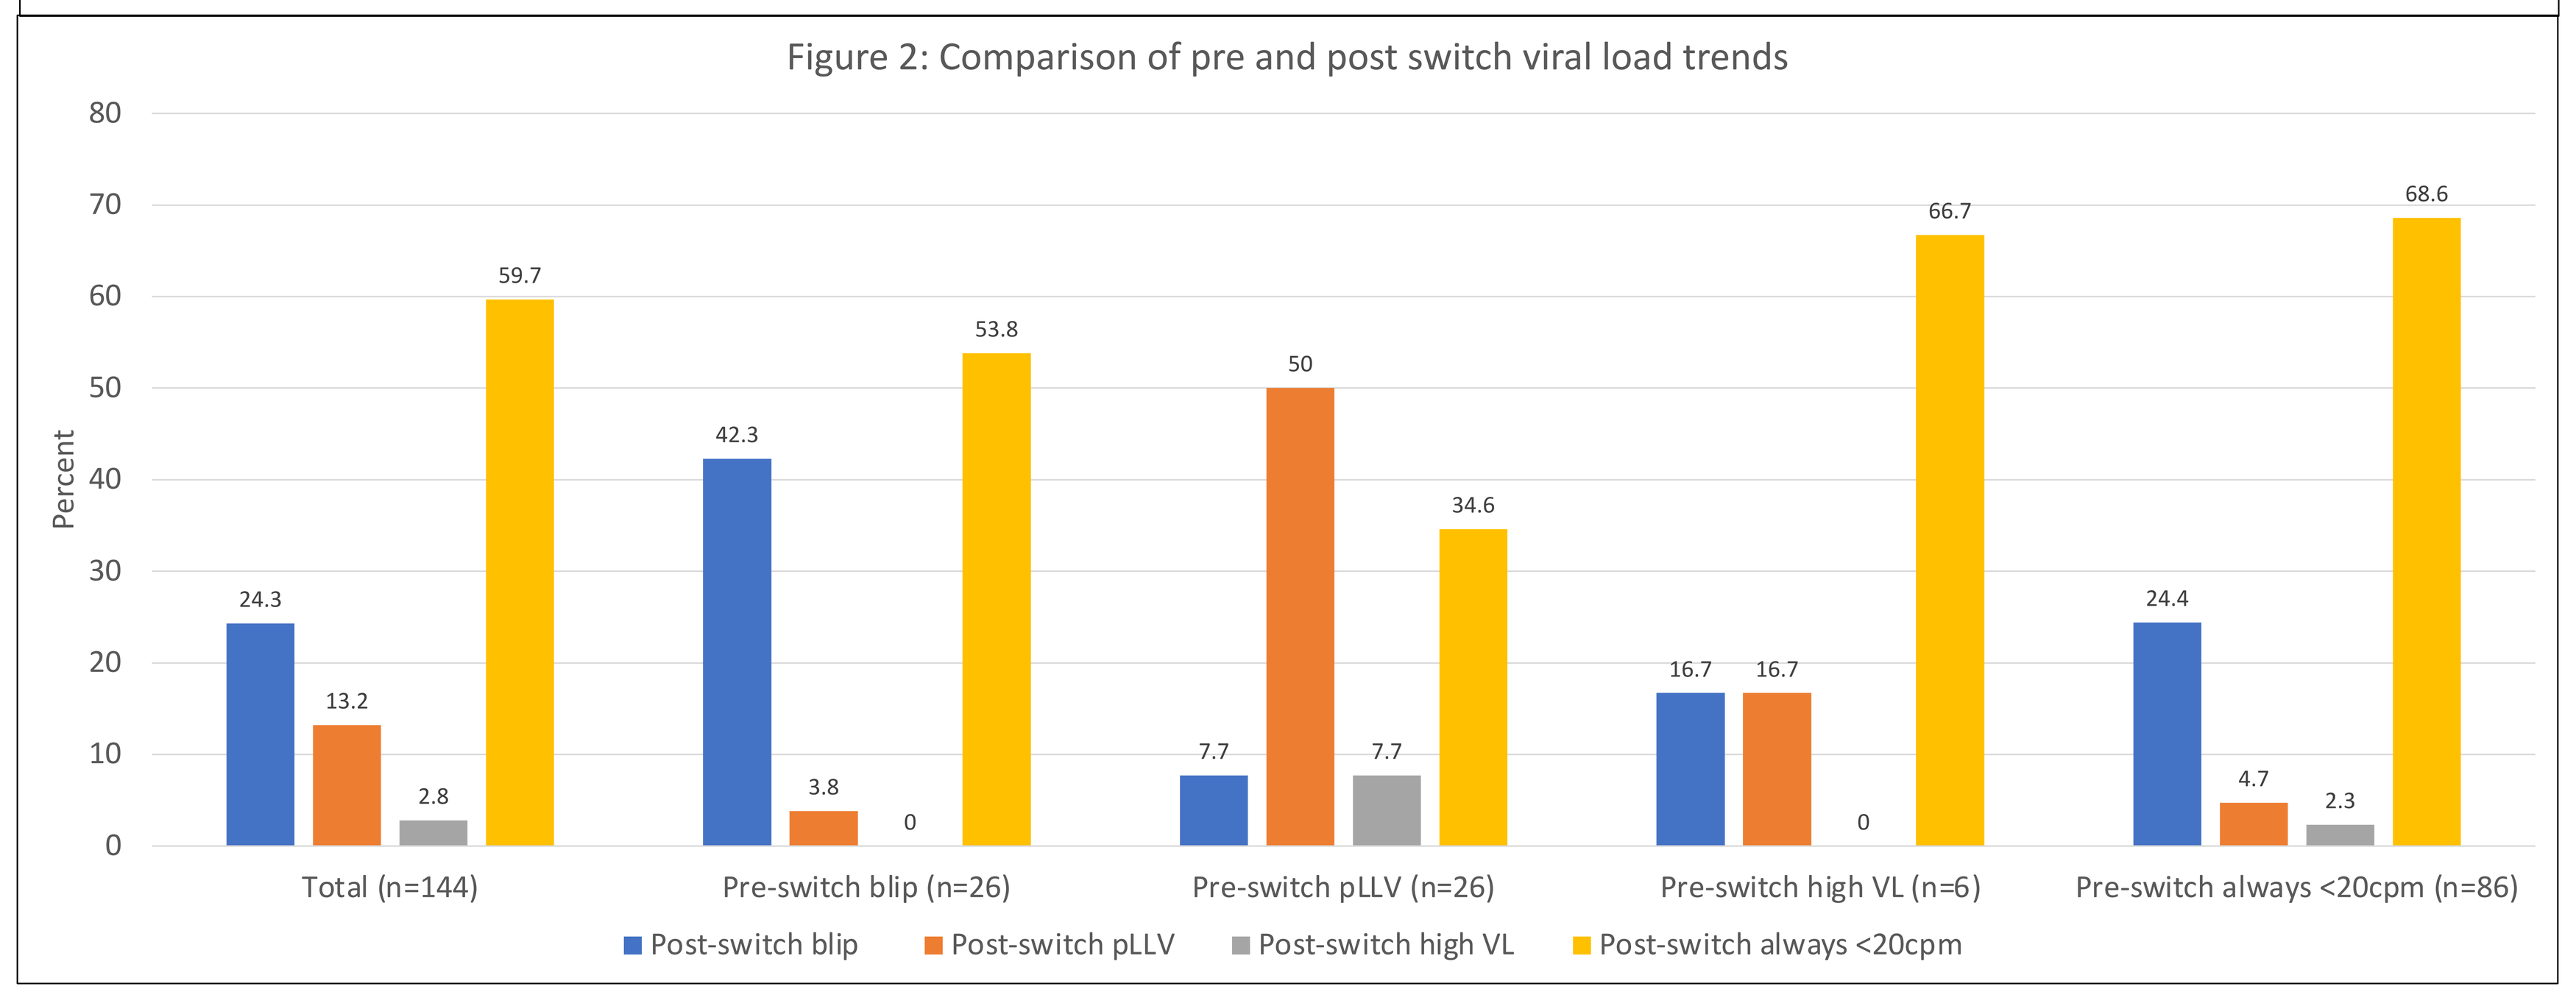
Figure 2: Comparison of pre and post switch viral load trends
Percent
80
70
60
50
40
30
20
10
0
24.3
13.2
2.8
59.7
42.3
3.8
0
53.8
7.7
50
7.7
34.6
16.7
16.7
0
66.7
24.4
4.7
2.3
68.6
Total (n=144)
Pre-switch blip (n=26)
Pre-switch pLLV (n=26)
Pre-switch high VL (n=6)
Pre-switch always <20cpm (n=86)
Post-switch blip
Post-switch pLLV
Post-switch high VL
Post-switch always <20cpm

Discussion

- Patients with pLLV or at least one detectable HIV RNA in the year before switching to LAI CAB/RPV were more likely to have detectable HIV RNA after switching despite the benefit of improved adherence with LAI CAB/RPV.
- Having a higher BMI or receiving injections outside of the dosing window was not associated with having detectable HIV viral loads, however the majority of patients who received injections outside the dosing window used an oral bridge or received their injection only 1 day late.
- Despite the frequency of blips or persistent viremia observed after switching to LAI CAB/RPV, we observed only one virologic failure in our cohort.

References

1. Swindells S, Andrade-Villanueva J-F, Richmond G, et al. Long-acting cabotegravir and rilpivirine for maintenance of HIV-1 suppression. *N Engl J Med.* 2020;382:1112-1123.
2. Cutrell A, Schapiro J, Perno C, et al. Exploring predictors of HIV-1 virologic failure to long-acting cabotegravir and rilpivirine: a multivariable analysis. *AIDS.* 2021;35:1333-1342.
3. Talarico C, Wu S, Upadhyay O, et al. HIV-1 RNA blips and low-level replication during phase III/IIIb cabotegravir+rilpivirine long-acting studies are similar to oral 3-drug therapy and not associated with week 48 virologic outcome. *Open Fourm Infec Dis.* 2020;7:S540-S531.