PREEXISTING HOST EPIGENETIC STATES ASSOCIATED WITH HIV REBOUND KINETICS

383

Michael J. Corley[1], Nathalia Mantovani[2], Leila B Giron[4], Alina Pang[1], Juliana Galinskas[2], Danilo Dias[2], Paulo A Ferreira[2], Gisele Gosuen[2], James R Hunter[2], Andrea Savarino[3], Ricardo S. Diaz[2*], Lishomwa C. Ndhlovu[1*]

[1]Division of Infectious Diseases, Department of Medicine, Weill Cornell Medicine, New York City NY, USA [2]Laboratório de Retrovirologia, Escola Paulista de Medicina, Universidade Federal de São Paulo, Sao Paulo, Brazil [3]Department of Infectious Diseases, Italian Institute of Health, Rome, 00161, Italy. [4]The Wistar Institute, Philadelphia, PA, USA

BACKGROUND

- Viral rebound with variable kinetics consistently occurs in people living with HIV (PLWH) after the discontinuation of ART during analytical treatment interruptions (ATI).
- Whether preexisting host DNAm states in bulk peripheral blood mononuclear cells (PBMCs) of PLWH participating in diverse curative interventions can reveal novel preexisting host epigenetic signatures related to time to viral rebound remains understudied.

METHODS

Longitudinal Evaluation of Host Epigenetic states among people living with HIV undergoing Multimodal Curative Interventions

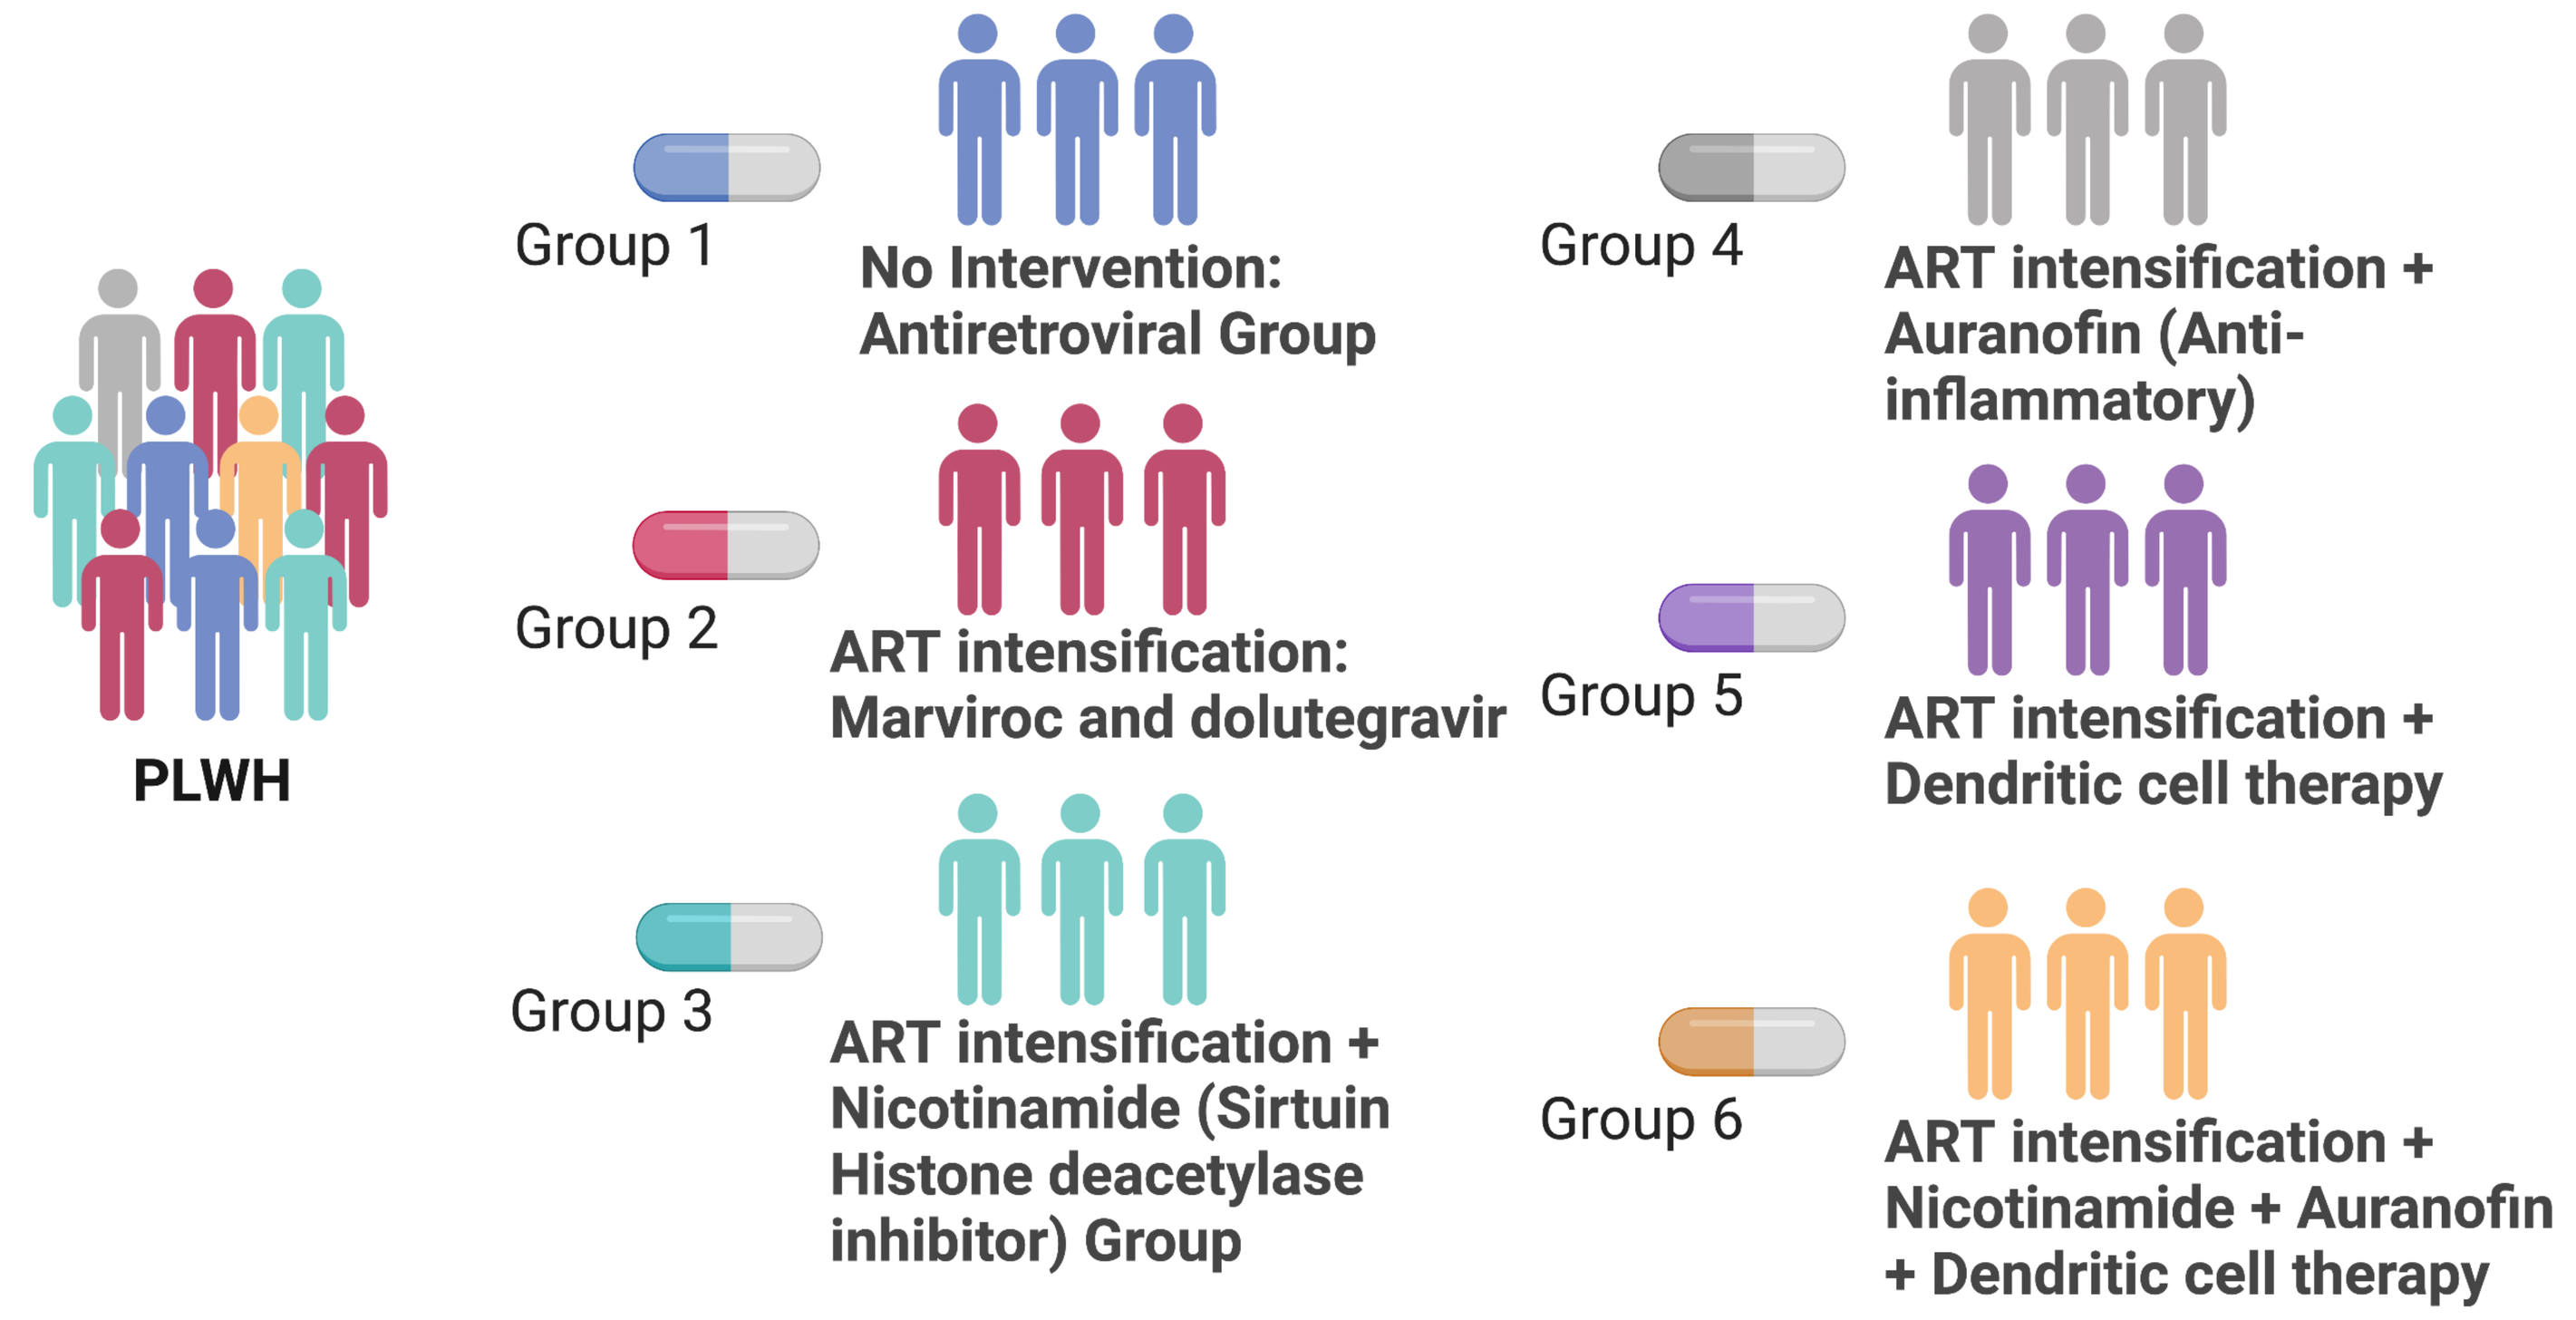
PLWH
Group 1
No Intervention: Antiretroviral Group
Group 2
ART intensification: Marviroc and dolutegravir
Group 3
ART intensification + Nicotinamide (Sirtuin Histone deacetylase inhibitor) Group
Group 4
ART intensification + Auranofin (Anti-inflammatory)
Group 5
ART intensification + Dendritic cell therapy
Group 6
ART intensification + Nicotinamide + Auranofin + Dendritic cell therapy

Figure 1. Overview of 30 male PLWH on ART who participated in combination interventions designed for eradicating residual plasma viremia and decreasing HIV reservoirs (NCT02961829). Genome-wide DNAm profiling of PBMCs was obtained at study entry from 26 male PLWH on ART who participated in an ATI following combination interventions. *Group 3 included the São Paulo Patient that rebounded at 511 days following analytical treatment interruption.

ID	Group	Age	Sex	CD4 Baseline (cell/uL)	Total HIV DNA (copies/10^6cell)	Time Rebound After Interruption (Days)
P1	G1	28	Male	541	2.42	35
P2	G1	47	Male	788	3.21	20
P3	G1	51	Male	1067	1.49	22
P4	G1	51	Male	775	4.60	34
P5	G1	52	Male	1585	0.51	21
P6	G2	26	Male	1287	0.67	41
P7	G2	57	Male	1202	4.71	102
P8	G2	43	Male	950	6.50	21
P9	G2	38	Male	921	0.18	20
P10	G2	33	Male	NA	NA	62
P11	G3	38	Male	762	2.93	14
P12	G3	25	Male	1231	4.84	22
P13	G3	49	Male	964	4.70	34
P14	G3	32	Male	720	0.89	511
P15	G3	33	Male	646	0.98	24
P16	G4	39	Male	1274	0.26	51
P17	G4	46	Male	1335	7.84	70
P18	G4	26	Male	NA	NA	20
P19	G5	30	Male	1053	27.12	21
P20	G5	52	Male	1617	7.70	24
P21	G5	24	Male	693	13.00	21
P22	G5	42	Male	1591	21.37	21
P23	G6	23	Male	839	11.71	21
P24	G6	42	Male	1265	33.31	21
P25	G6	47	Male	677	0.31	33
P26	G6	58	Male	1001	98.30	35

RESULTS

Host immune cell DNA methylation states at baseline study entry prior to intervention associate with time to viral rebound following analytical treatment interruption.

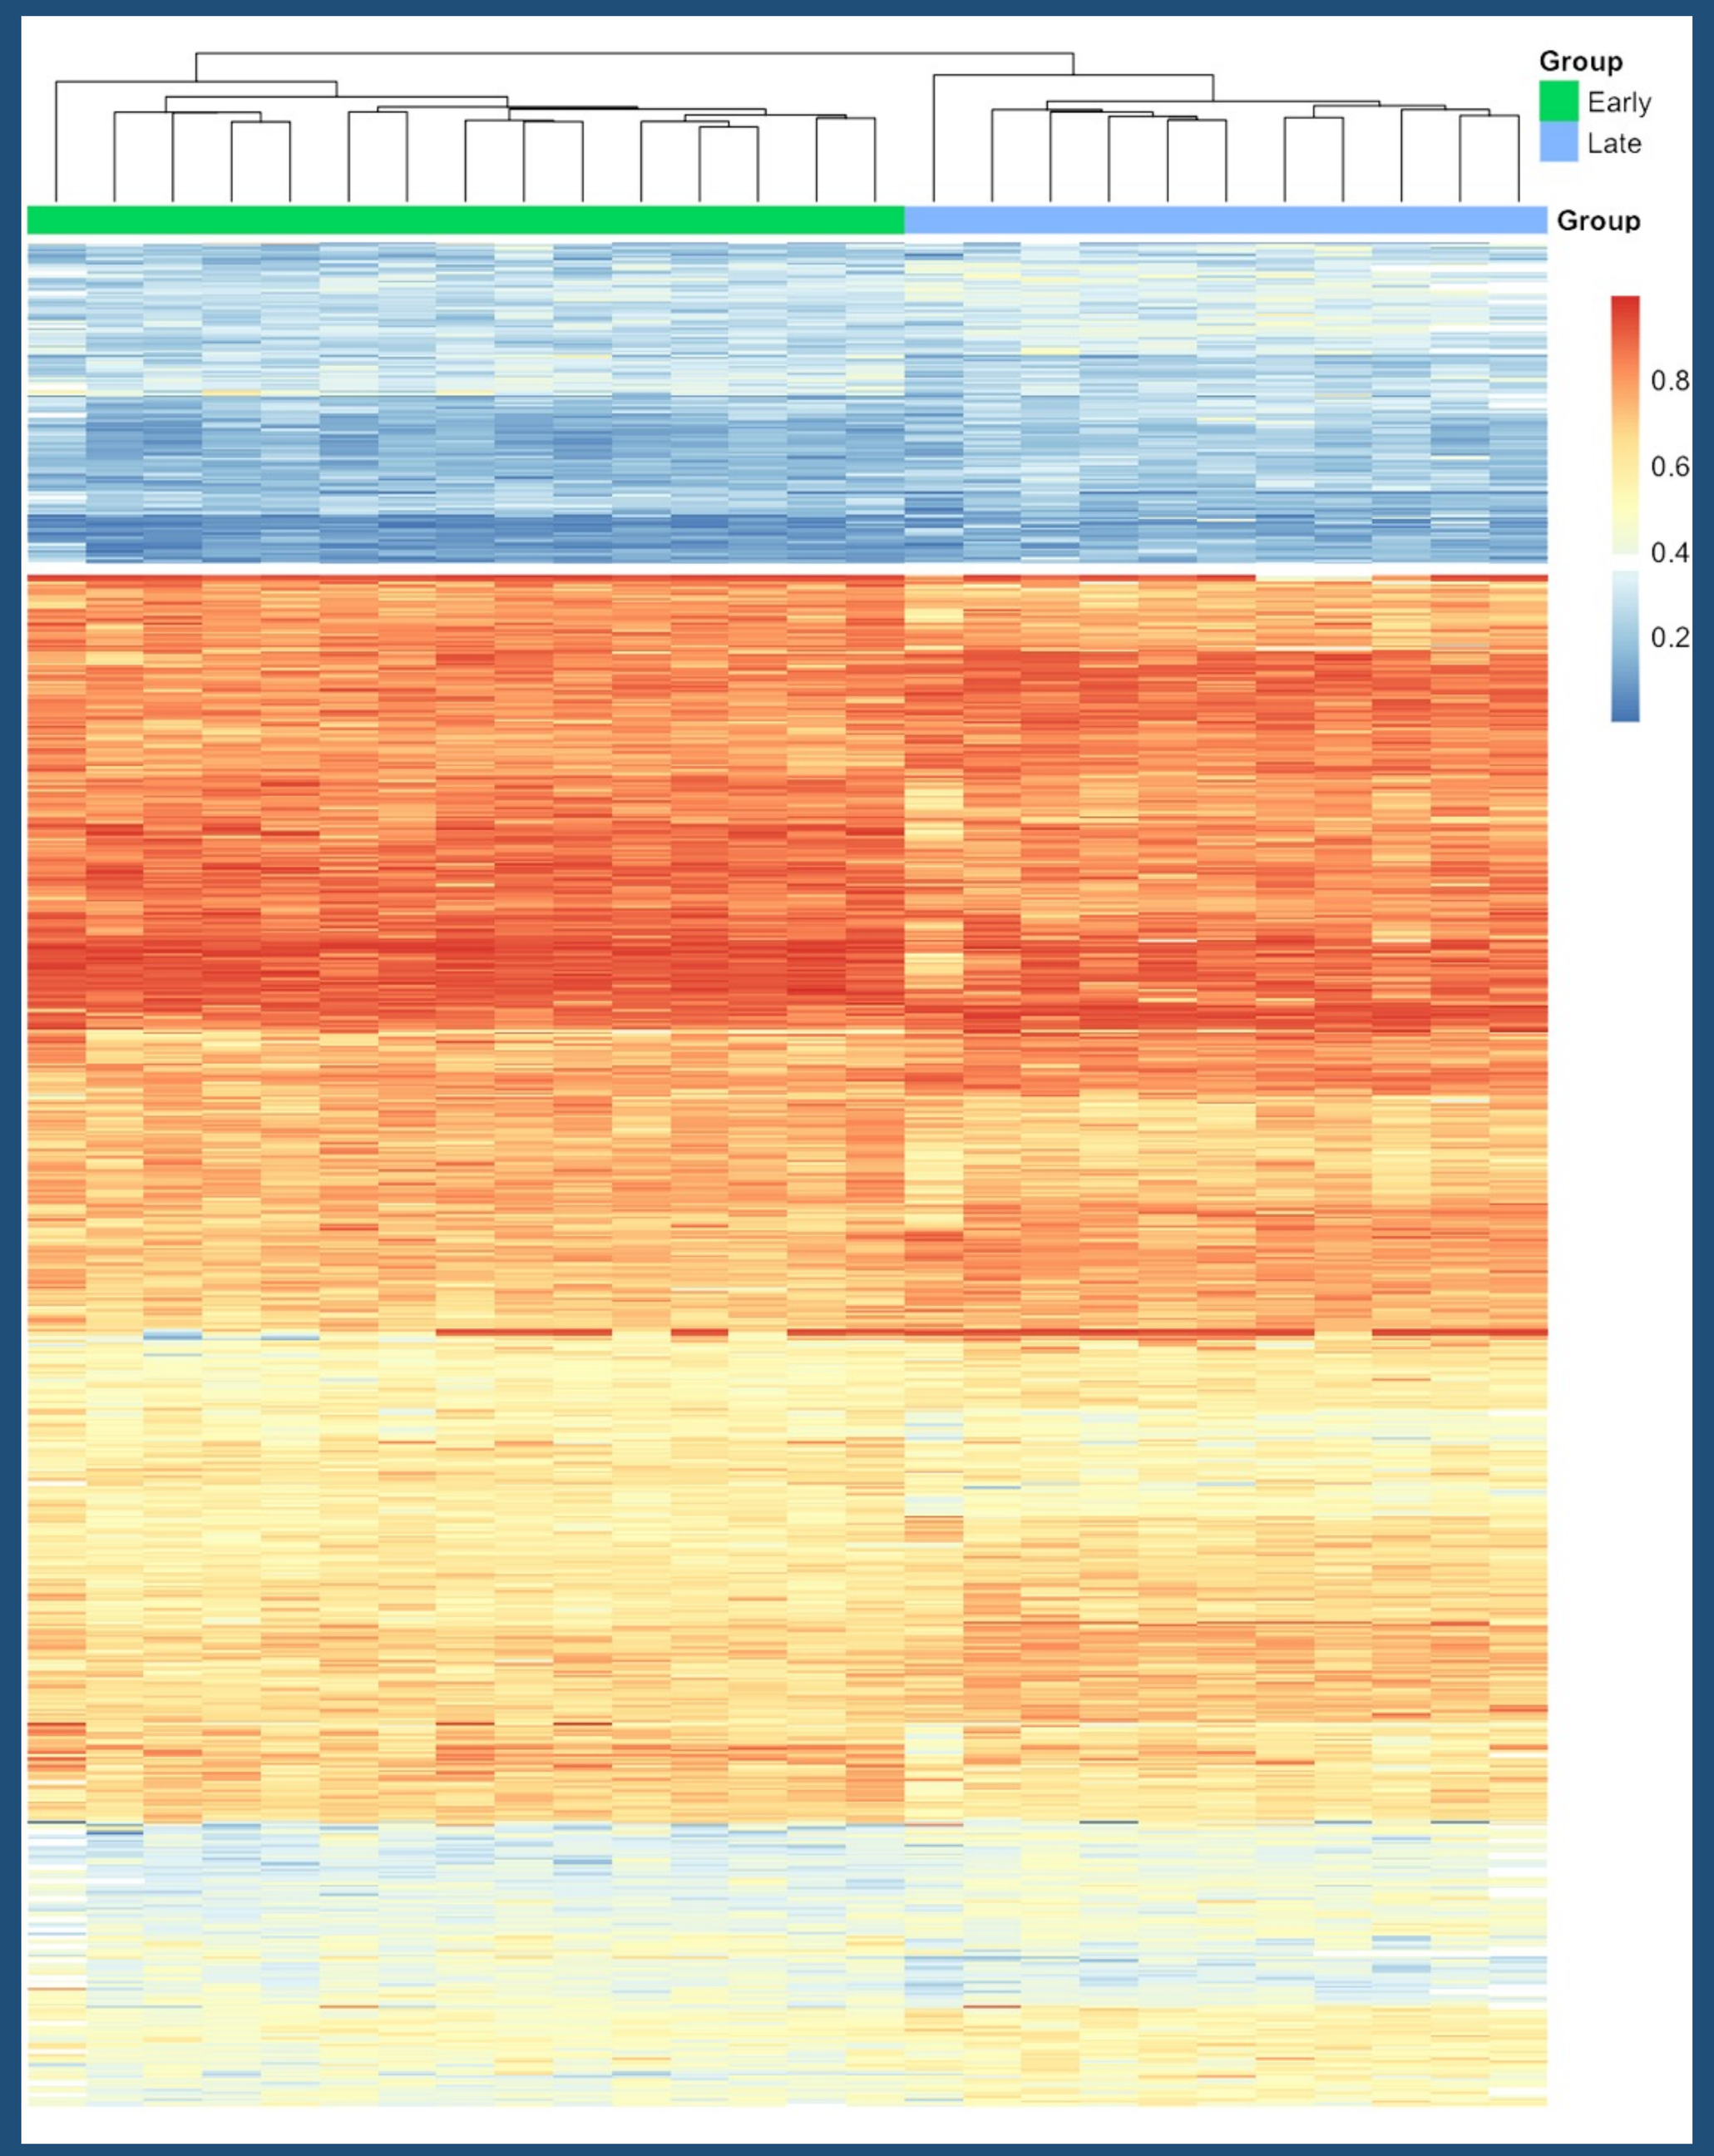
Group
Early
Late

Figure 2. At baseline study entry visit prior to intervention, we identified 2,694 differentially methylated loci (DML) (Δβ-value > |0.05|; P < 0.05) comparing early compared to late rebounding participants.

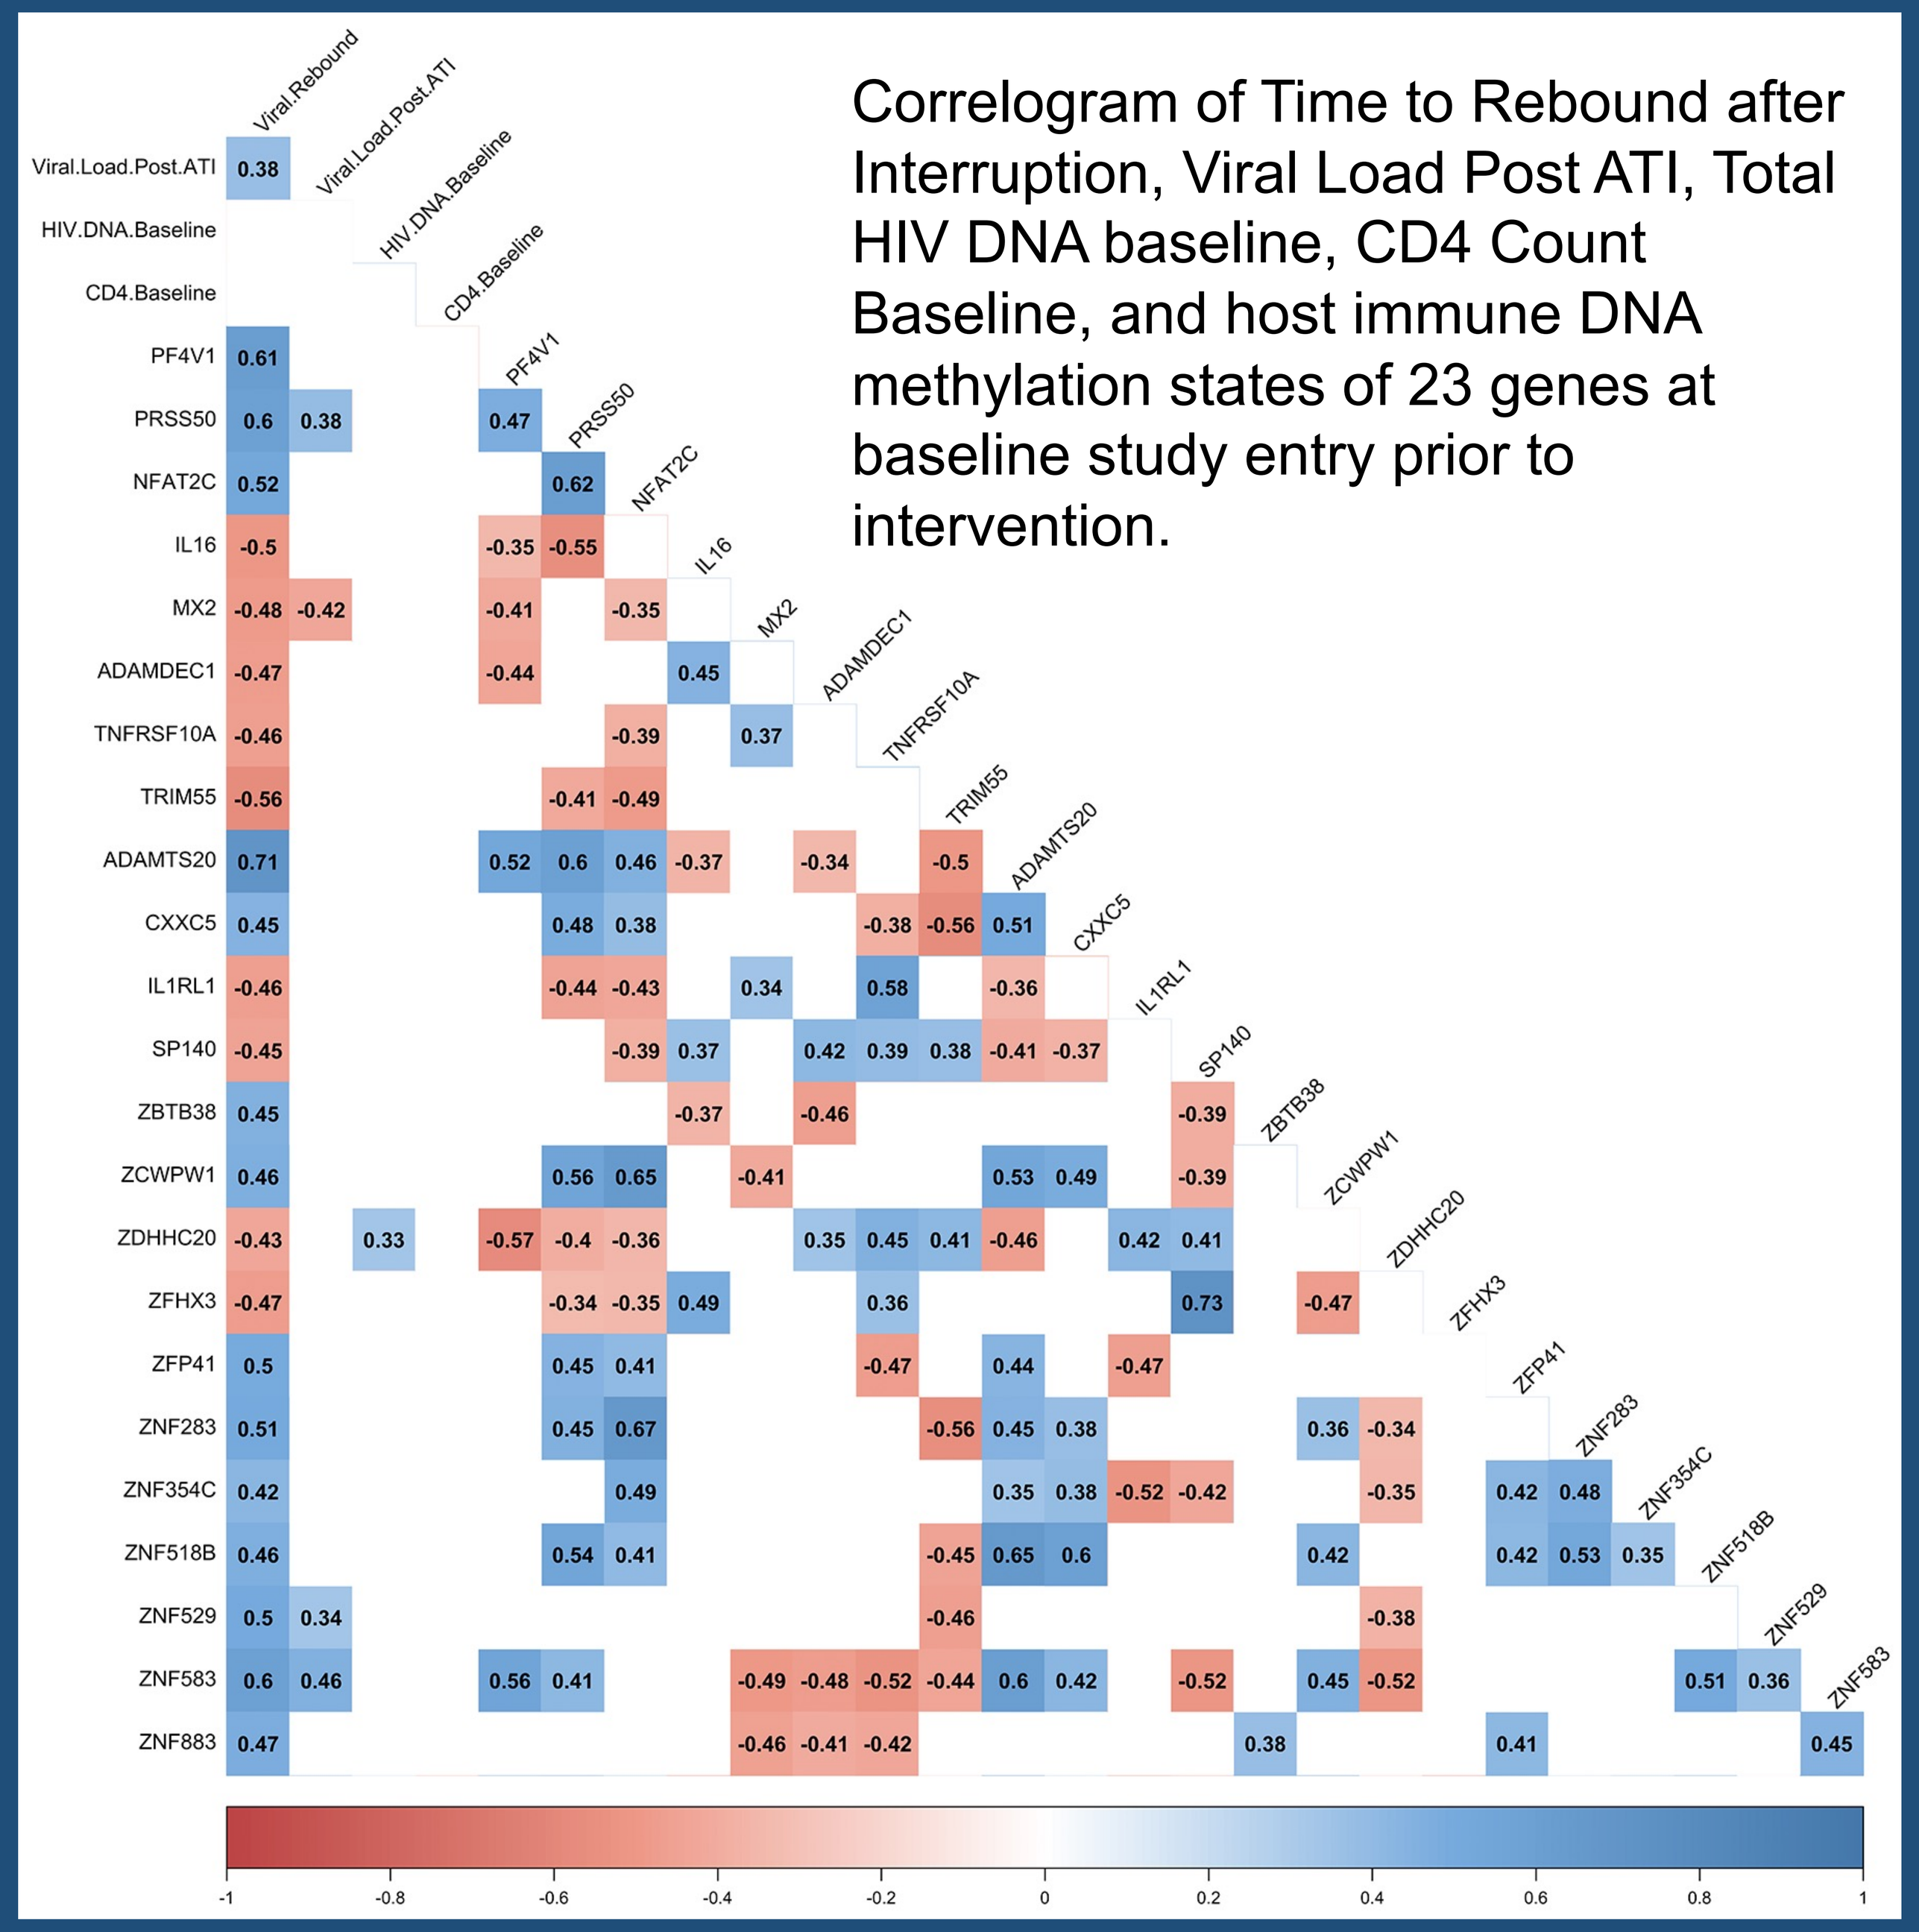
Correlogram of Time to Rebound after Interruption, Viral Load Post ATI, Total HIV DNA baseline, CD4 Count Baseline, and host immune DNA methylation states of 23 genes at baseline study entry prior to intervention.

Figure 3. Among the top DML associated with rebound group were genes including *ADAMTS20* involved in HIV life cycle, *MX2* antiviral protein against HIV, epigenetic regulator of plasmacytoid dendritic cells *CXXC5*, and chemokine gene *PF4V1*. DNAm states of *PRSS50*, *MX2*, *ZNF529*, and *ZNF583* significantly associated with viral load post-ATI

No significant differences in participants age, total HIV DNA, and CD4 count comparing early versus late rebound.

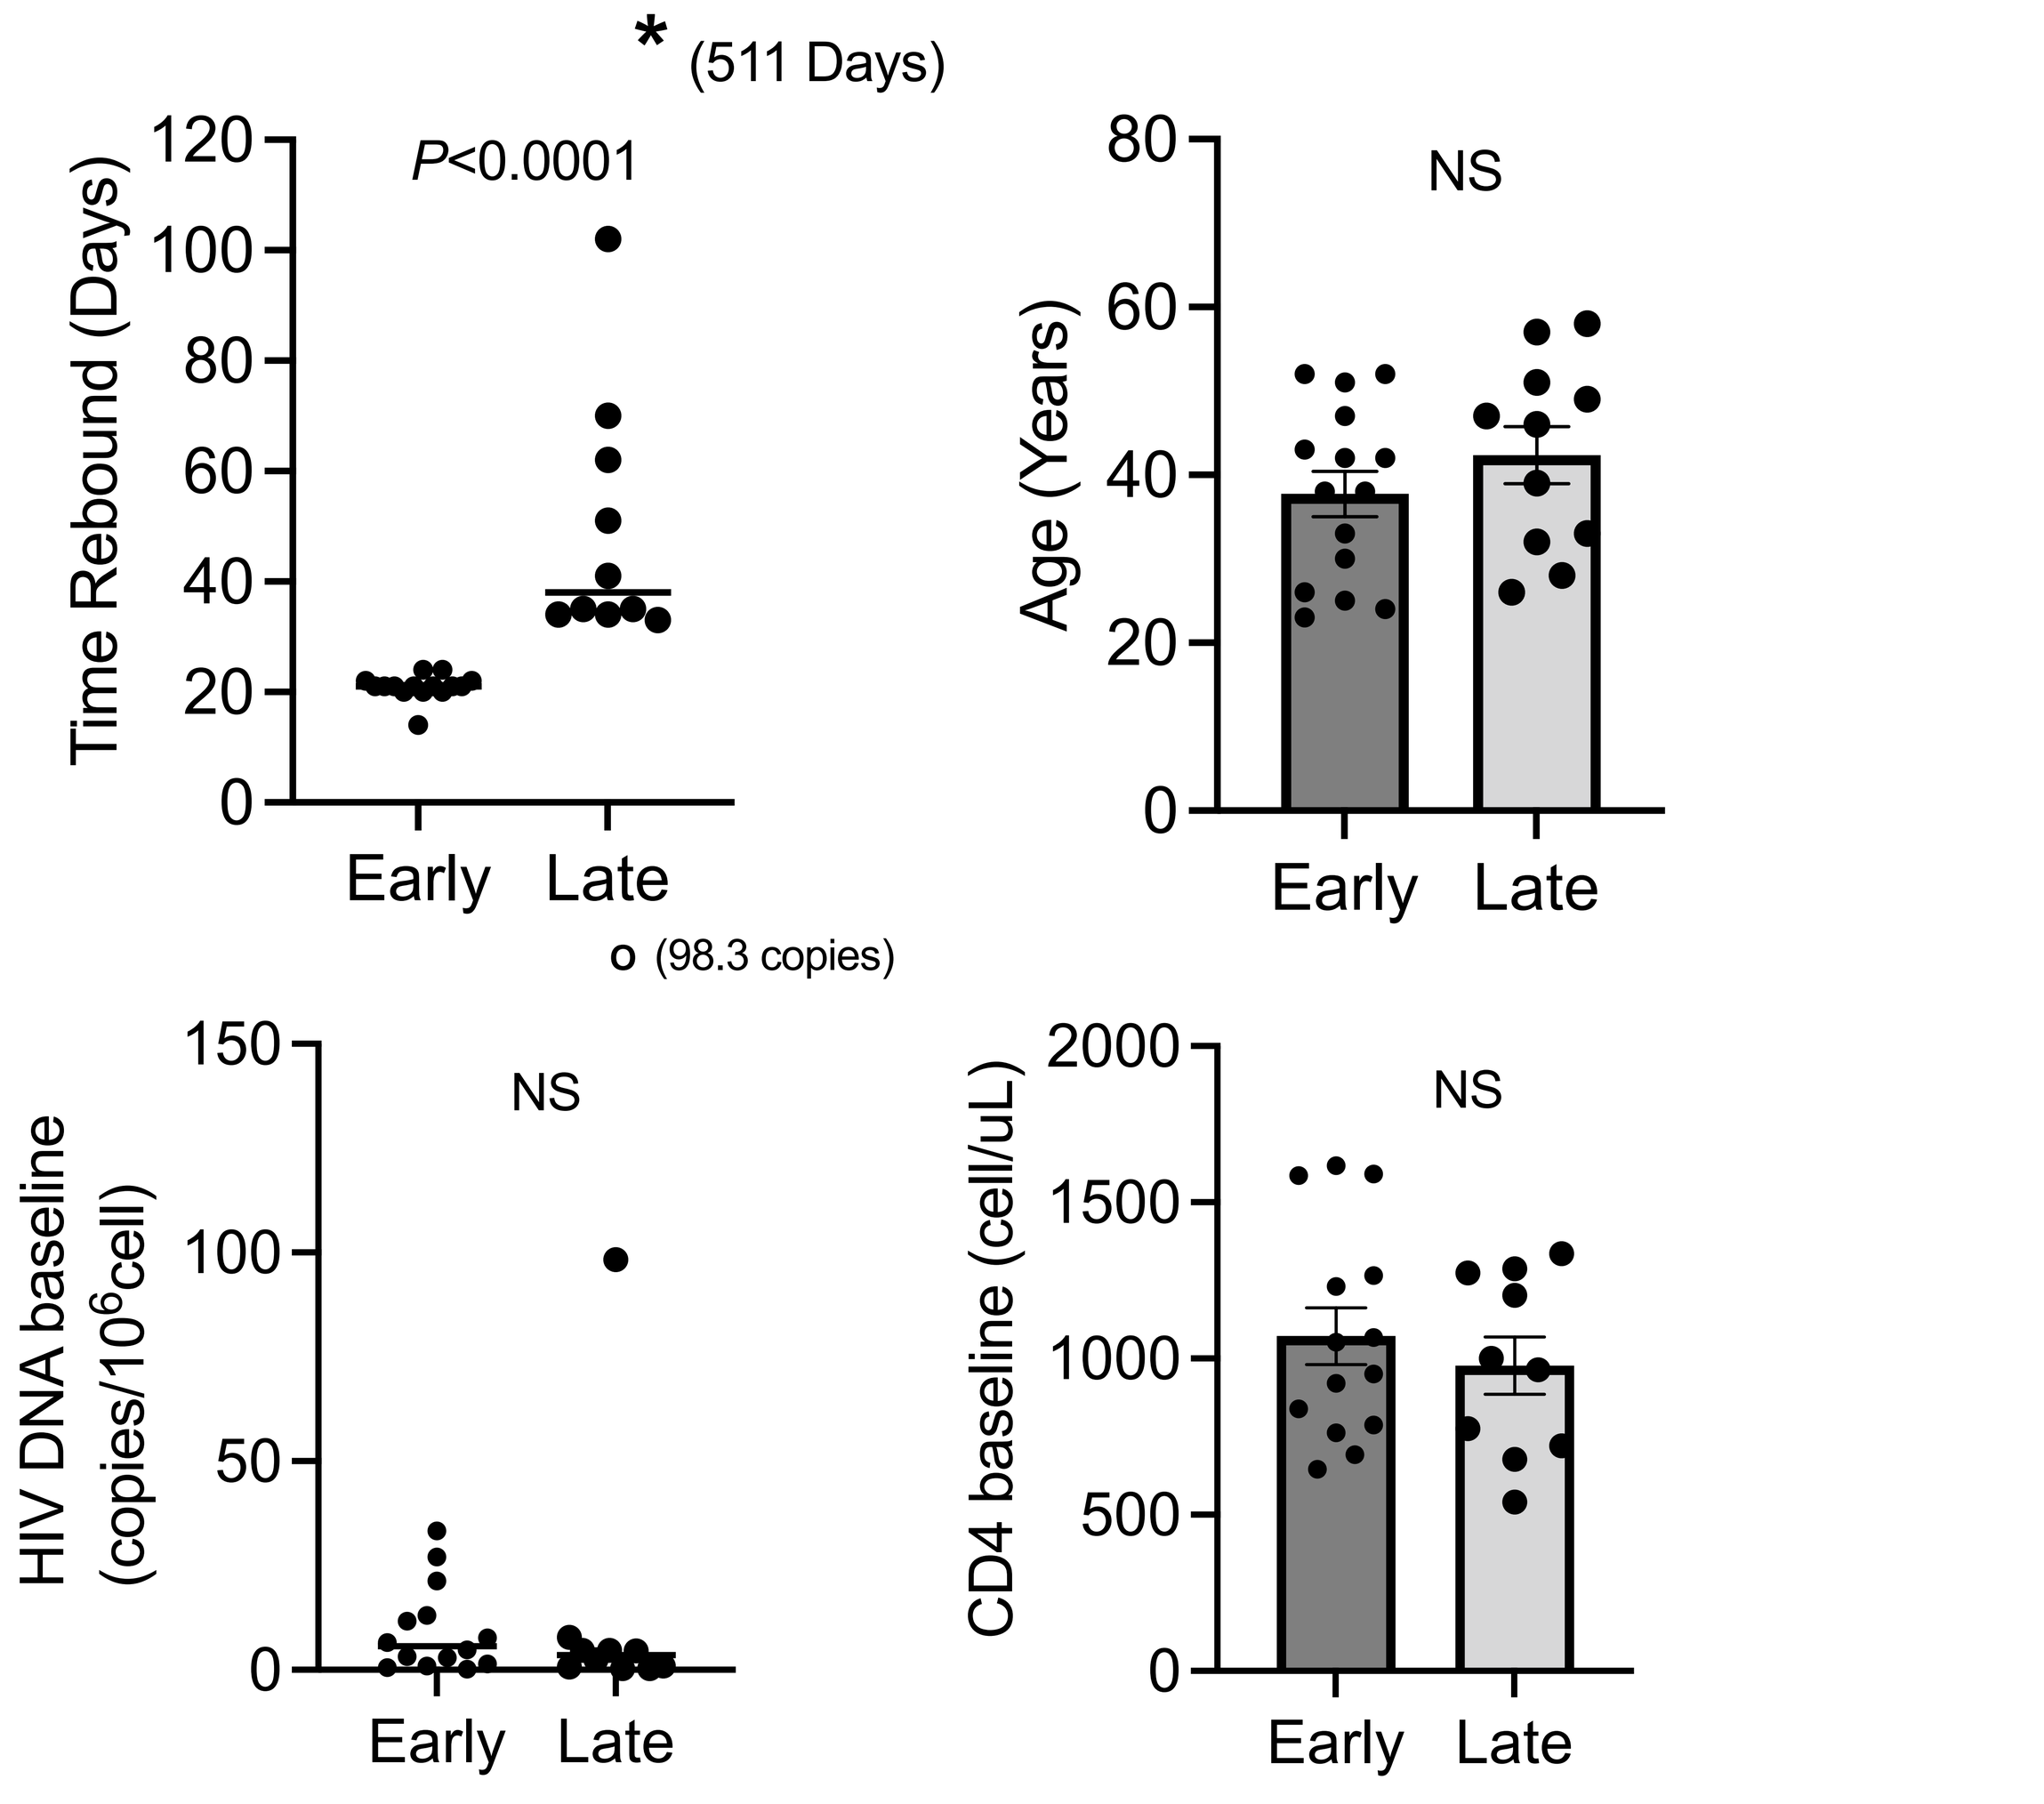
* (511 Days)
P<0.0001
Time Rebound (Days)
Early
Late
o (98.3 copies)
NS
Age (Years)
HIV DNA baseline (copies/10⁶cell)
CD4 baseline (cell/uL)

Figure 4. Comparisons of early and late rebounding participants.

Participation in multimodal curative interventions does not accelerate epigenetic biomarkers of biological aging and mortality risks.

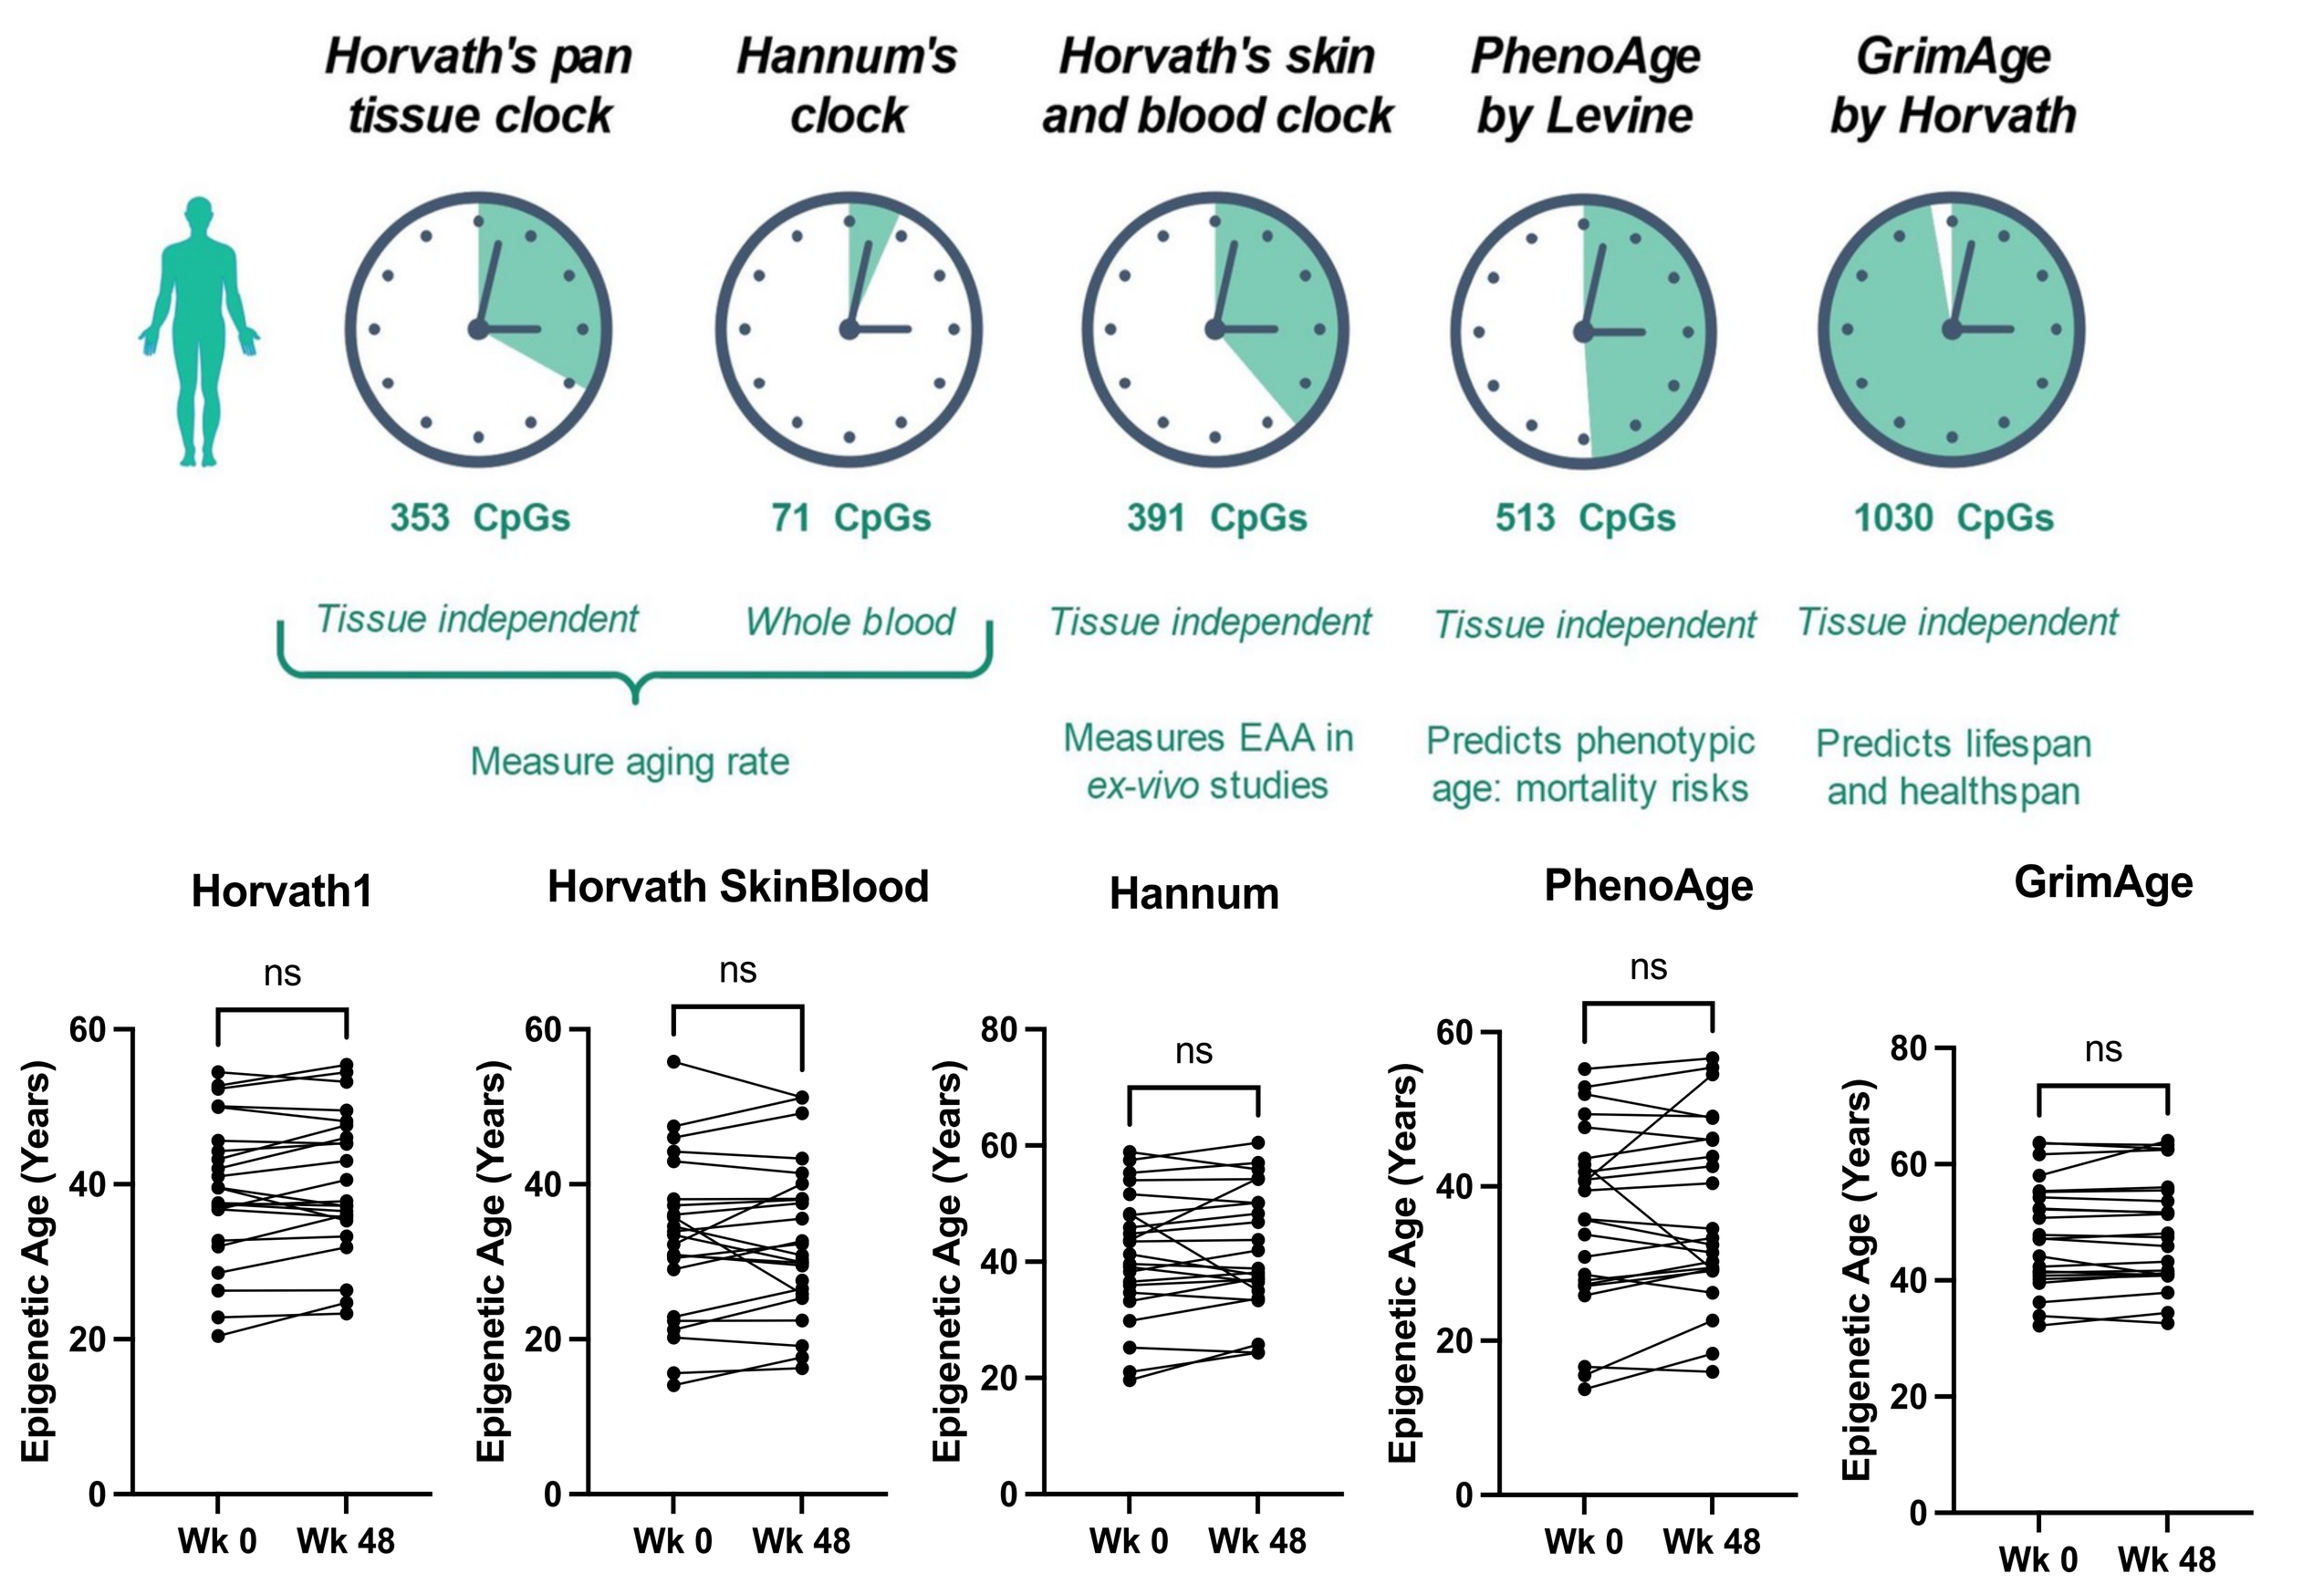
Horvath's pan tissue clock
353 CpGs
Tissue independent
Hannum's clock
71 CpGs
Whole blood
Measure aging rate
Horvath's skin and blood clock
391 CpGs
Tissue independent
Measures EAA in ex-vivo studies
PhenoAge by Levine
513 CpGs
Tissue independent
Predicts phenotypic age: mortality risks
GrimAge by Horvath
1030 CpGs
Tissue independent
Predicts lifespan and healthspan
Horvath1
Horvath SkinBlood
Hannum
PhenoAge
GrimAge
ns
Epigenetic Age (Years)
Wk 0
Wk 48

Figure 5. Comparison of epigenetic age estimates based on 5 epigenetic clocks of participants before and 48 weeks following interventions for groups 2-6. ns: Not significant, Paired T-test.

The São Paulo participant who had long-term ART-free virological control (72 weeks off ART following ATI) exhibited a reversal of epigenetic age at weeks 24 and 48.

Male, 35 y.o., MSM. Diagnosed 2012,
started ART two months later. VL at ART initiation: 20,221 copies/mL.
ART: AZT (TDF)/3TC+EFV.
Study entry: CD4 count: 720 cells/mm3

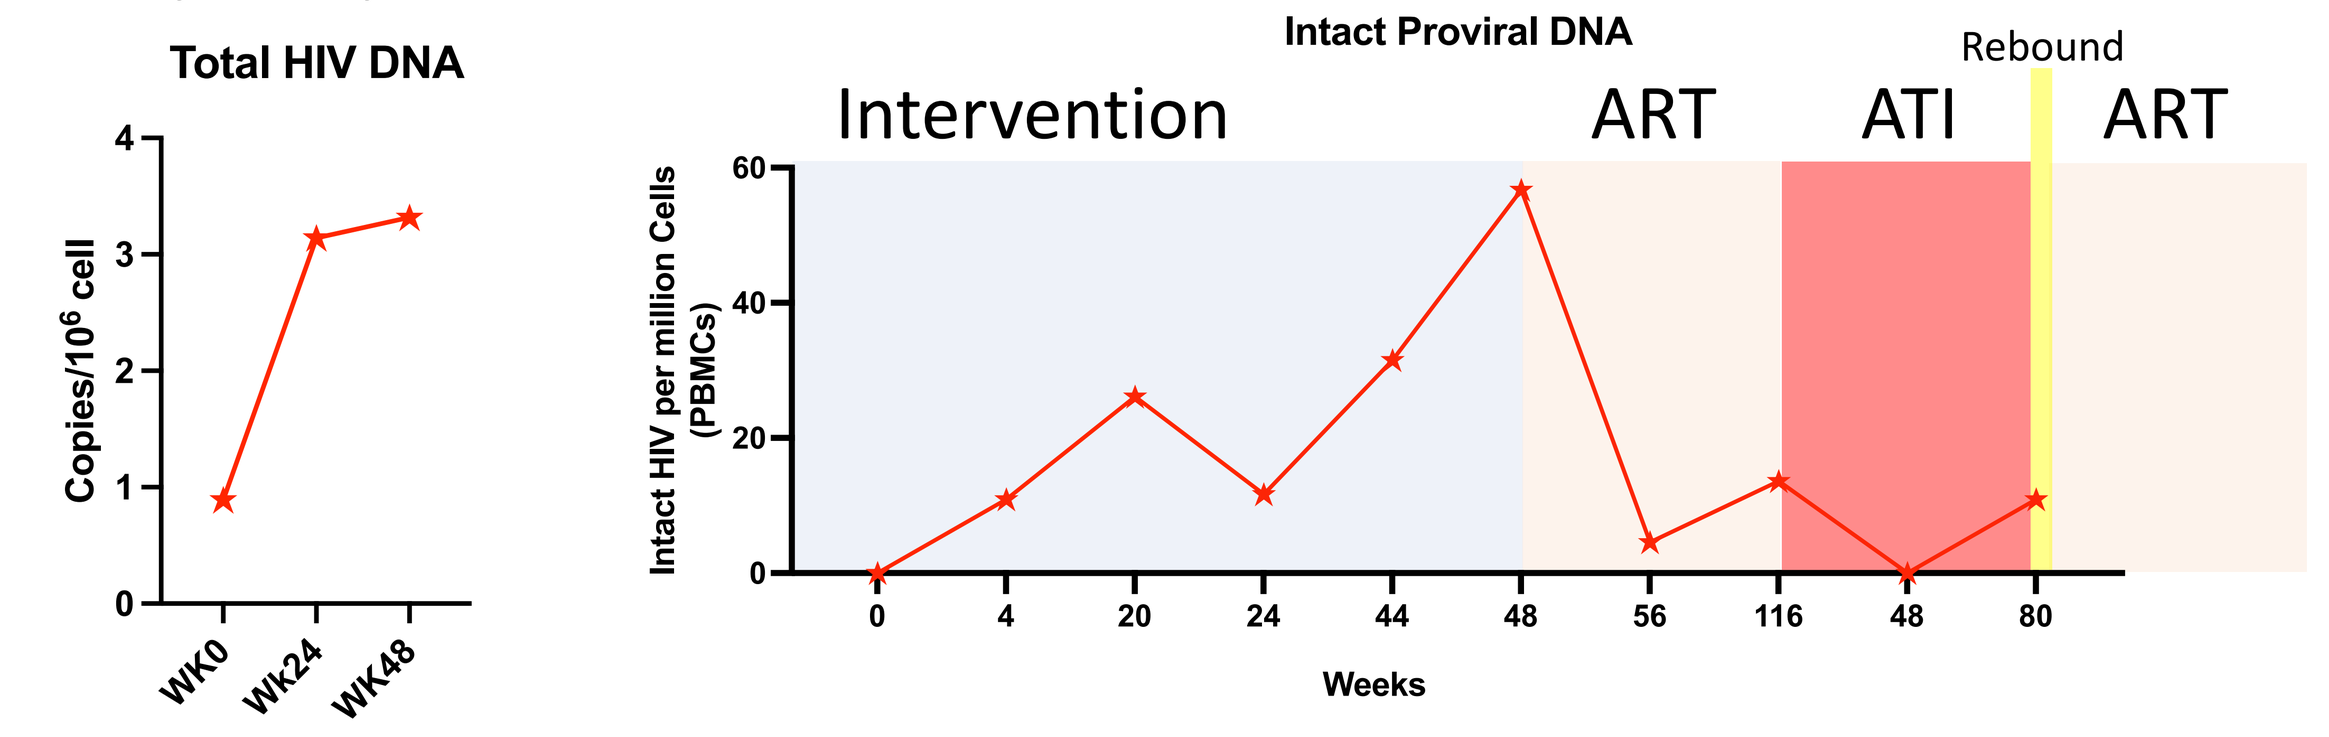
Total HIV DNA
Copies/10⁶ cell
WK0
Wk24
WK48
Intact Proviral DNA
Intervention
ART
ATI
Rebound
ART
Intact HIV per million Cells (PBMCs)
Weeks

Figure 6. Total HIV DNA and Intact Proviral DNA measures from PBMCs.

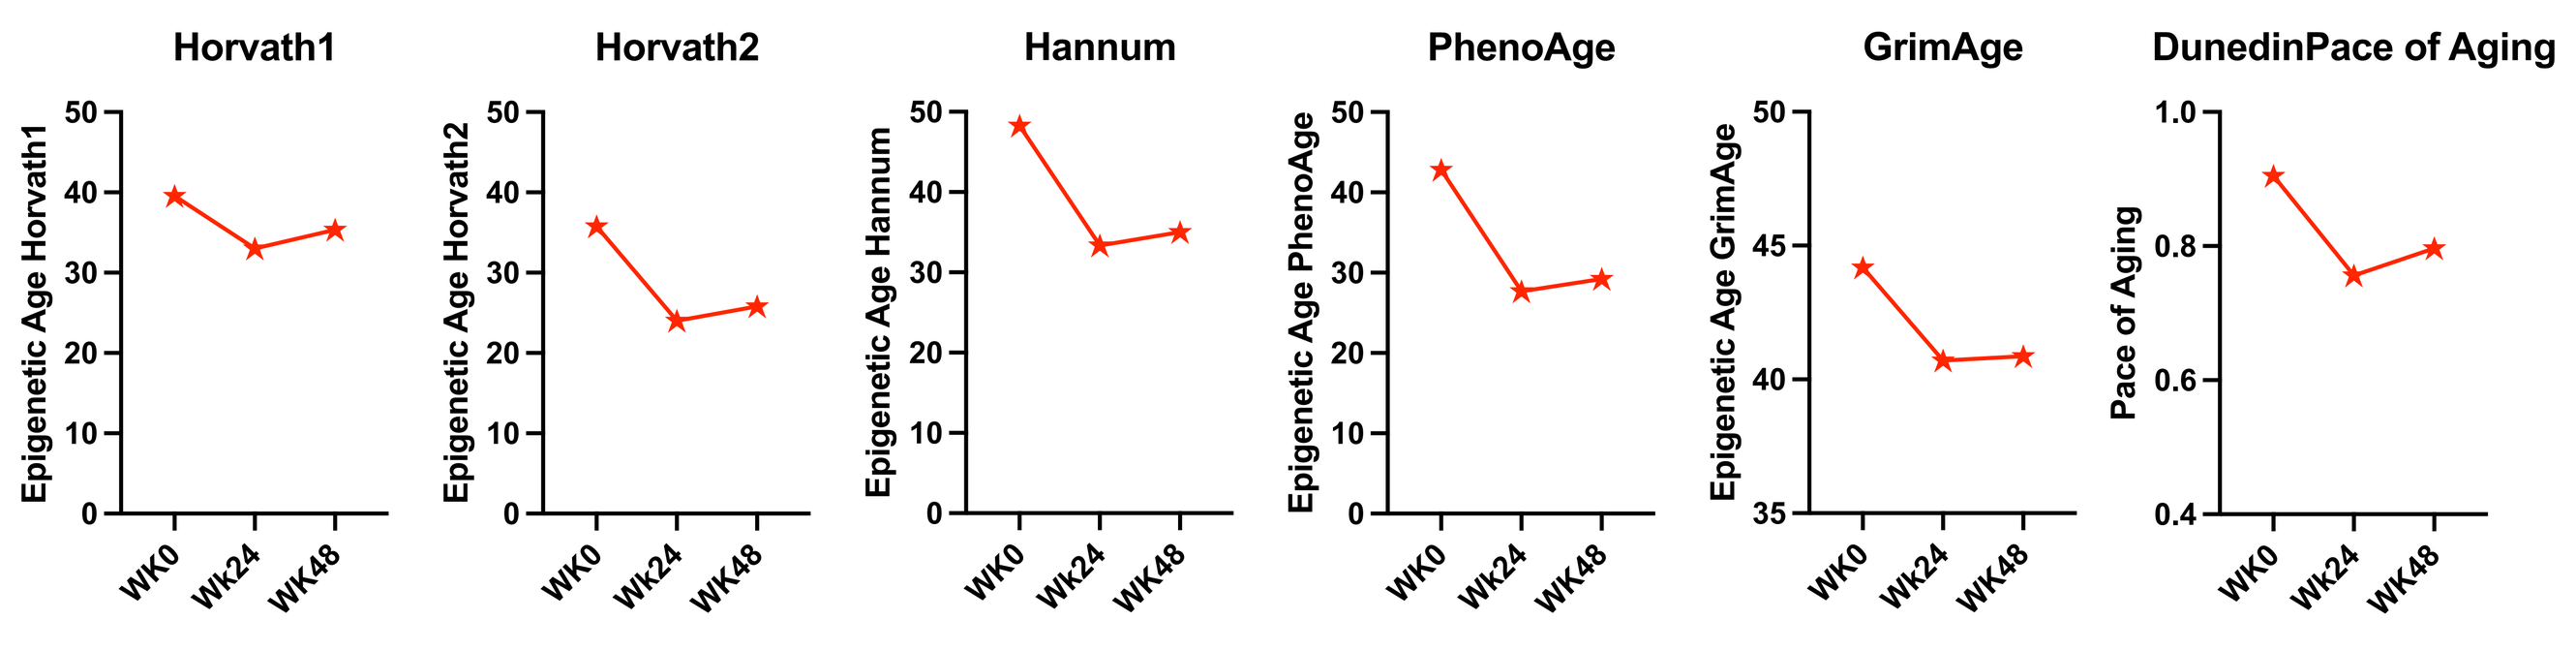
Horvath1
Horvath2
Hannum
PhenoAge
GrimAge
DunedinPace of Aging
Pace of Aging

Figure 7. Longitudinal epigenetic age estimates of São Paulo participant at Week 0, 24, and 48 using 6 different epigenetic clocks.

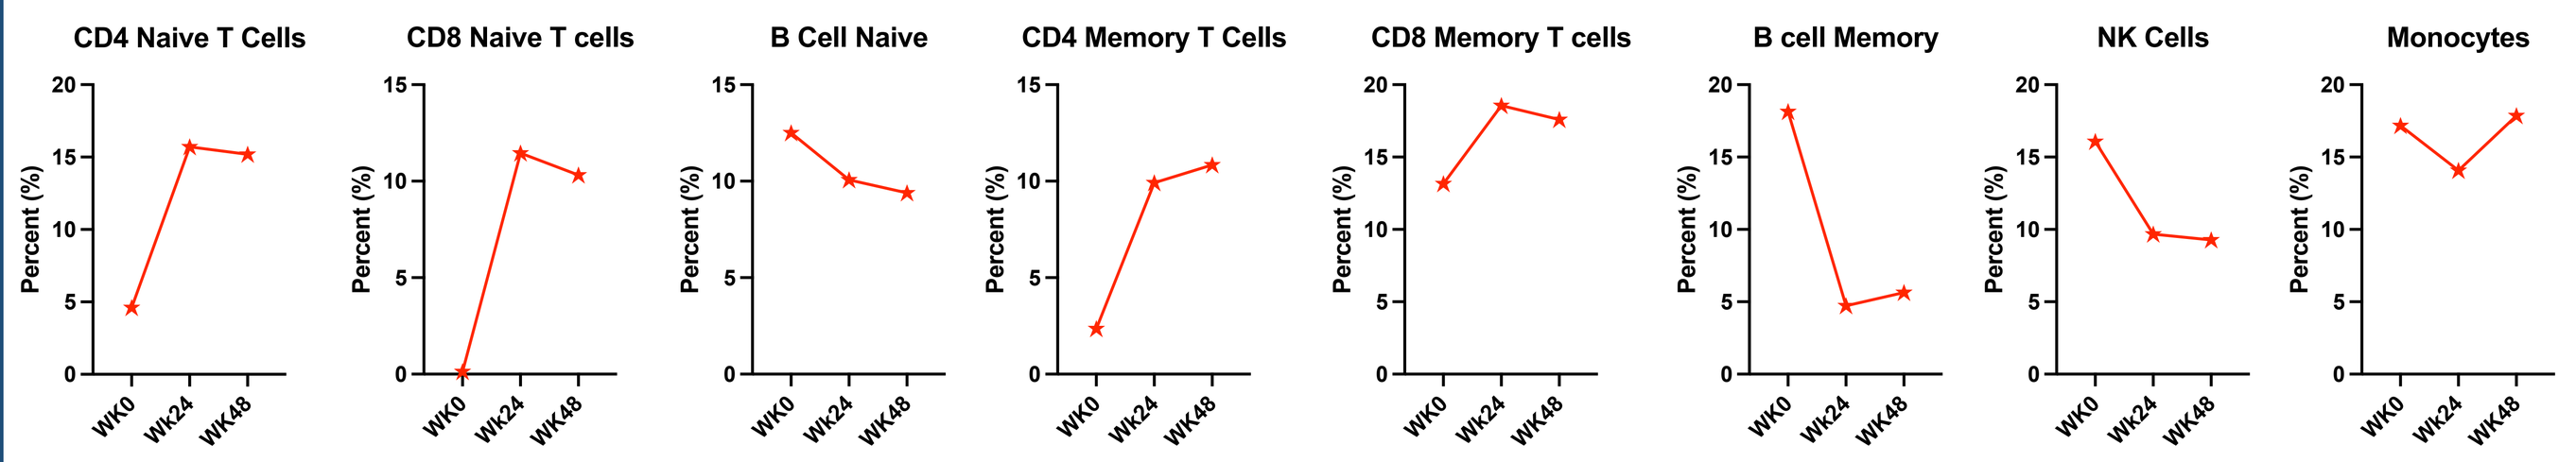
CD4 Naive T Cells
CD8 Naive T cells
B Cell Naive
CD4 Memory T Cells
CD8 Memory T cells
B cell Memory
NK Cells
Monocytes
Percent (%)

Figure 8. Longitudinal DNA methylation-based cell deconvolution of São Paulo participant at Week 0, 24, and 48 using an enhanced reference-based deconvolution algorithm of blood DNA methylation.

CONCLUSIONS

- Fixed host epigenetic features of innate and adaptive immune cells in PLWH may predetermine varying viral rebound kinetics during analytical treatment interruption.
- These findings also suggest that an optimal host epigenetic landscape may exist to control HIV expression and silencing.
- Epigenetic data from the São Paulo patient suggest that potential interventions decreasing/reversing epigenetic age may be a reliable biomarker of viral rebound after treatment interruption and long-term ART-free virological control.
- Findings suggest that multimodal curative interventions involving ART intensification and immune-based approaches in PLWH do not increase/accelerate epigenetic biomarkers of aging, mortality risk, and pace of aging.

ACKNOWLEDGEMENTS

We would like to thank the study participants who committed so much time for this study.

Weill Cornell Medicine