PROJECTED IMPACT OF EXPANDED LONG-ACTING INJECTABLE PREP USE AMONG MEN WHO HAVE SEX WITH MEN ON LOCAL HIV EPIDEMICS

Ruchita Balasubramanian, Parastu Kasaie PhD, Melissa Schnure ScM, David Dowdy MD PhD, Maunank Shah MD MHS, Anthony Todd Fojo MD MHS

Poster Number: 00916

Background

- **The Ending the HIV Epidemic (EHE)** initiative aims to reduce HIV incidence in the US by 90% over a decade. Pre-exposure prophylaxis (PrEP) is a key component in this effort.
- Long-acting injectable (LAI) PrEP reduces the risk of HIV acquisition more than oral PrEP but its potential to impact local HIV epidemics remains unclear.

Methods

- We extended the Johns Hopkins HIV Economic Epidemiological Model **(JHEEM),** a dynamic model of HIV transmission in 32 high priority urban areas in the US[1].

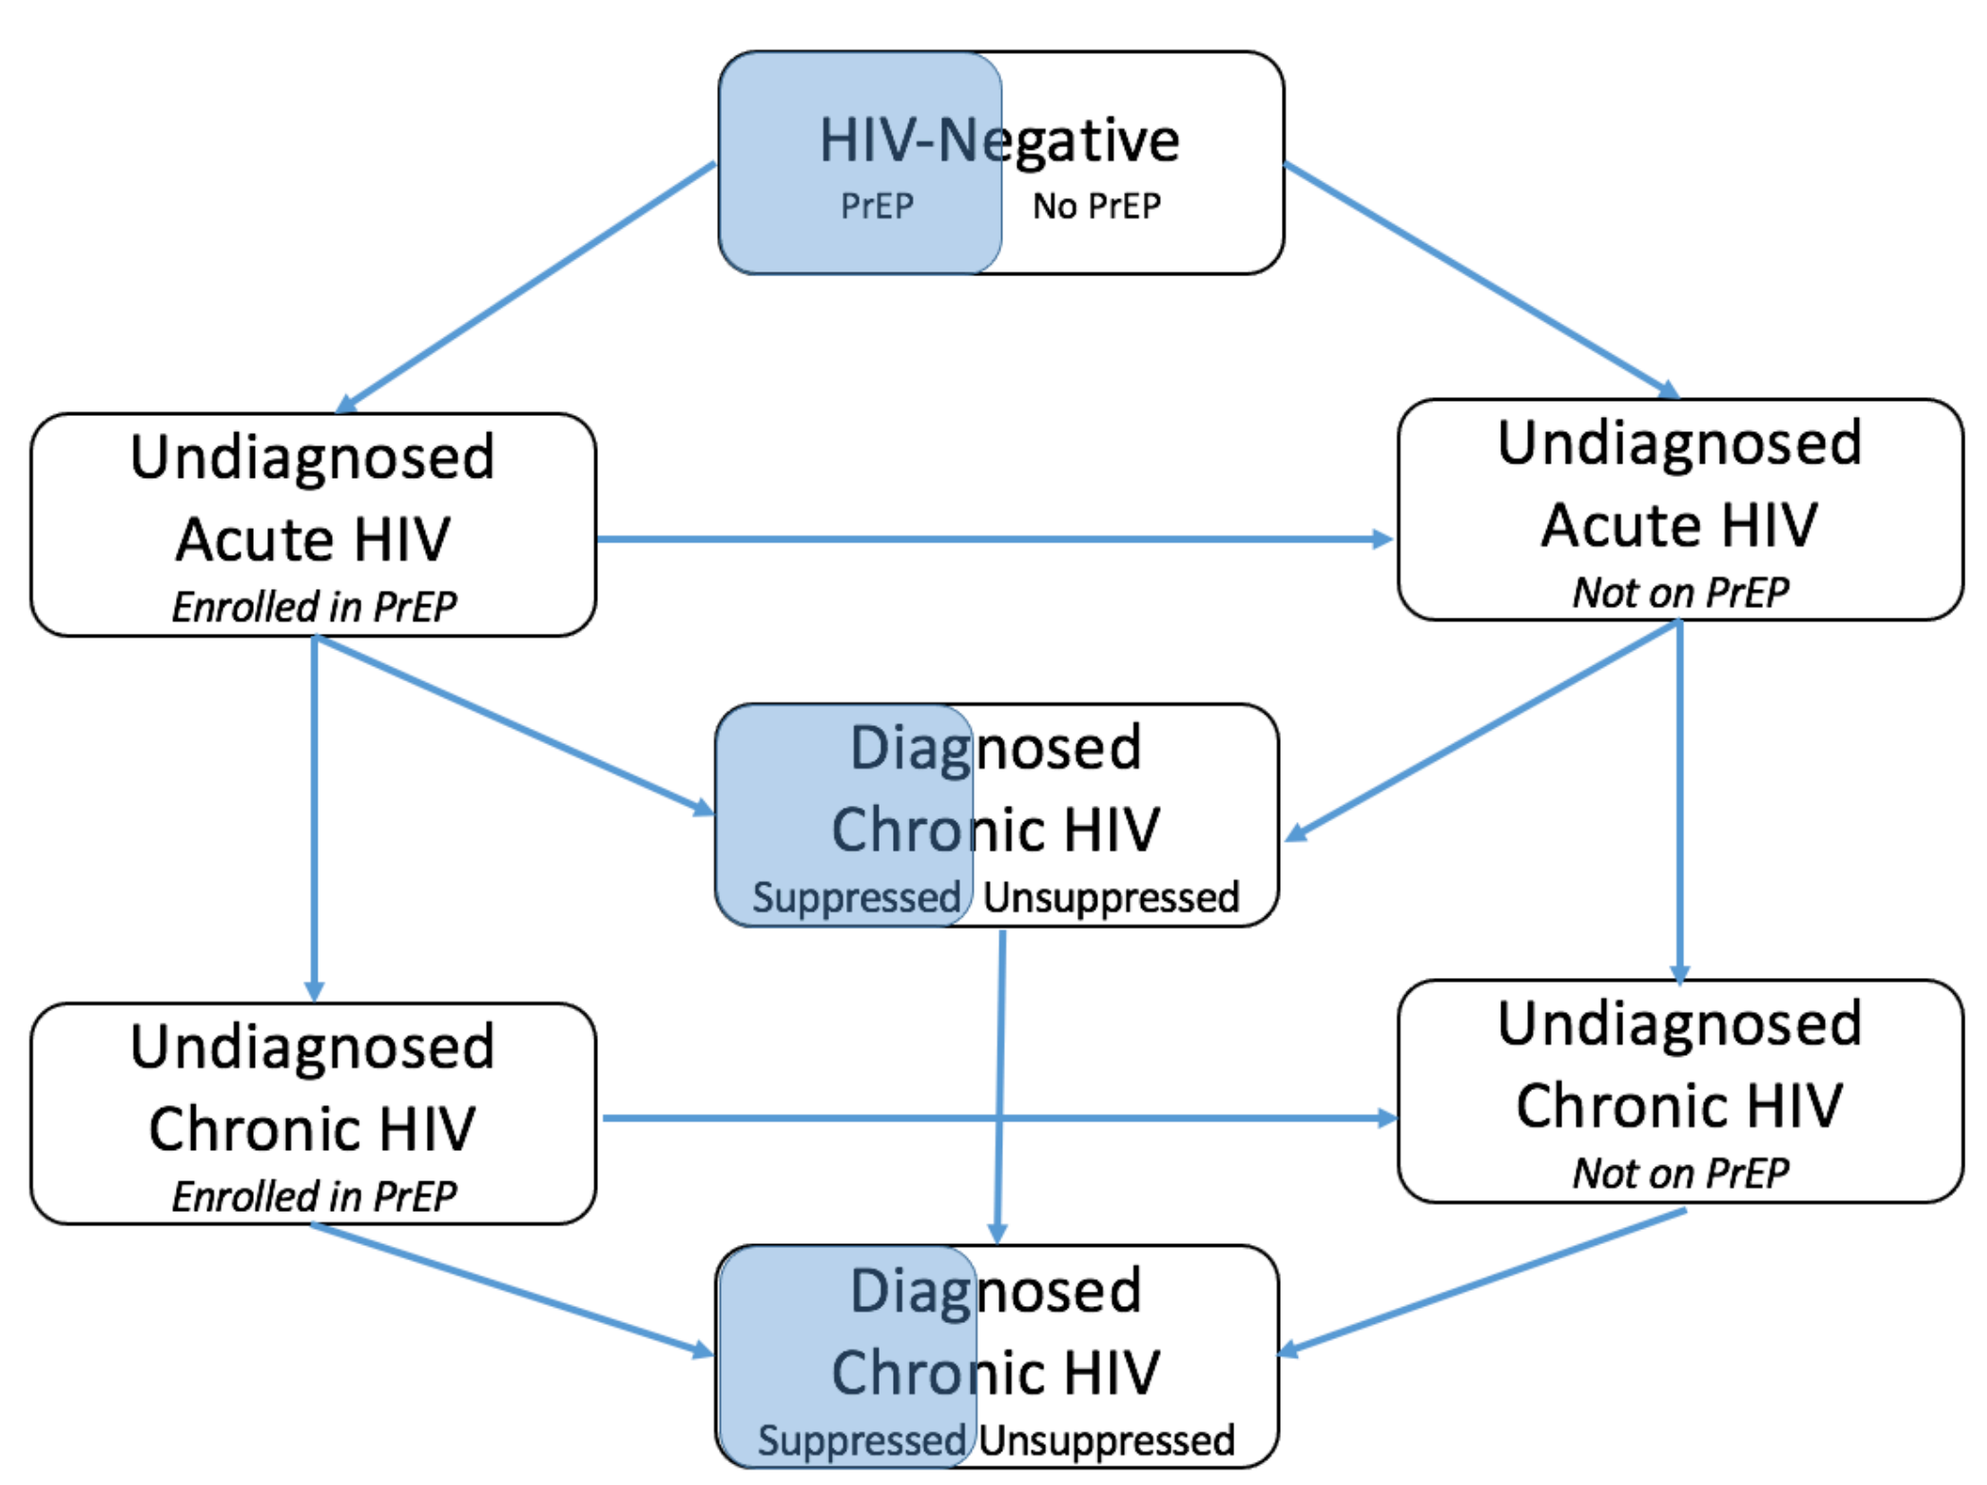
HIV-Negative
PrEP
No PrEP
Undiagnosed
Acute HIV
Enrolled in PrEP
Undiagnosed
Acute HIV
Not on PrEP
Diagnosed
Chronic HIV
Suppressed
Unsuppressed
Undiagnosed
Chronic HIV
Enrolled in PrEP
Undiagnosed
Chronic HIV
Not on PrEP
Diagnosed
Chronic HIV
Suppressed
Unsuppressed

- In each of the 32 cities among men who have sex with men (MSM), we leveraged **JHEEM** to run 1,000 simulations under various interventions testing an expansion of PrEP uptake.

 -Baseline: Oral PrEP at current levels

 -Alternatives:

 --Converting baseline Oral PrEP to LAI PrEP

 --Increasing overall PrEP uptake with either LAI PrEP or Oral PrEP over a baseline of either Oral PrEP or LAI PrEP

 --Uptake levels of 10% or 25% considered

Results

Reduction in HIV Incidence among MSM from 2020-2030 Across Different Levels of Oral and Injectable PrEP in US Cities

Additional Uptake	0%	0%	10%	10%	10%	25%	25%	25%
PrEP Type: Oral or Long-Acting-Injectable (LAI)	NA	NA	All Oral	All LAI	All LAI	All Oral	All LAI	All LAI
Baseline PrEP Composition	Baseline Oral	Baseline LAI	Baseline Oral	Baseline Oral	Baseline LAI	Baseline Oral	Baseline Oral	Baseline LAI
Atlanta-Sandy Springs-Alpharetta	9%	10%	20%	22%	23%	34%	39%	40%
Washington-Arlington-Alexandria	37%	39%	47%	49%	50%	59%	62%	64%
San Francisco-Oakland-Berkeley	35%	38%	48%	50%	52%	62%	66%	67%
San Diego-Chula Vista-Carlsbad	35%	37%	47%	49%	50%	60%	63%	64%
Seattle-Tacoma-Bellevue	15%	22%	40%	45%	48%	63%	69%	70%
Austin-Round Rock-Georgetown	34%	37%	47%	50%	52%	62%	66%	67%
Cleveland-Elyria	-2%	0%	24%	28%	28%	49%	56%	56%
Total	19%	22%	33%	36%	38%	50%	55%	56%

Washington D.C.

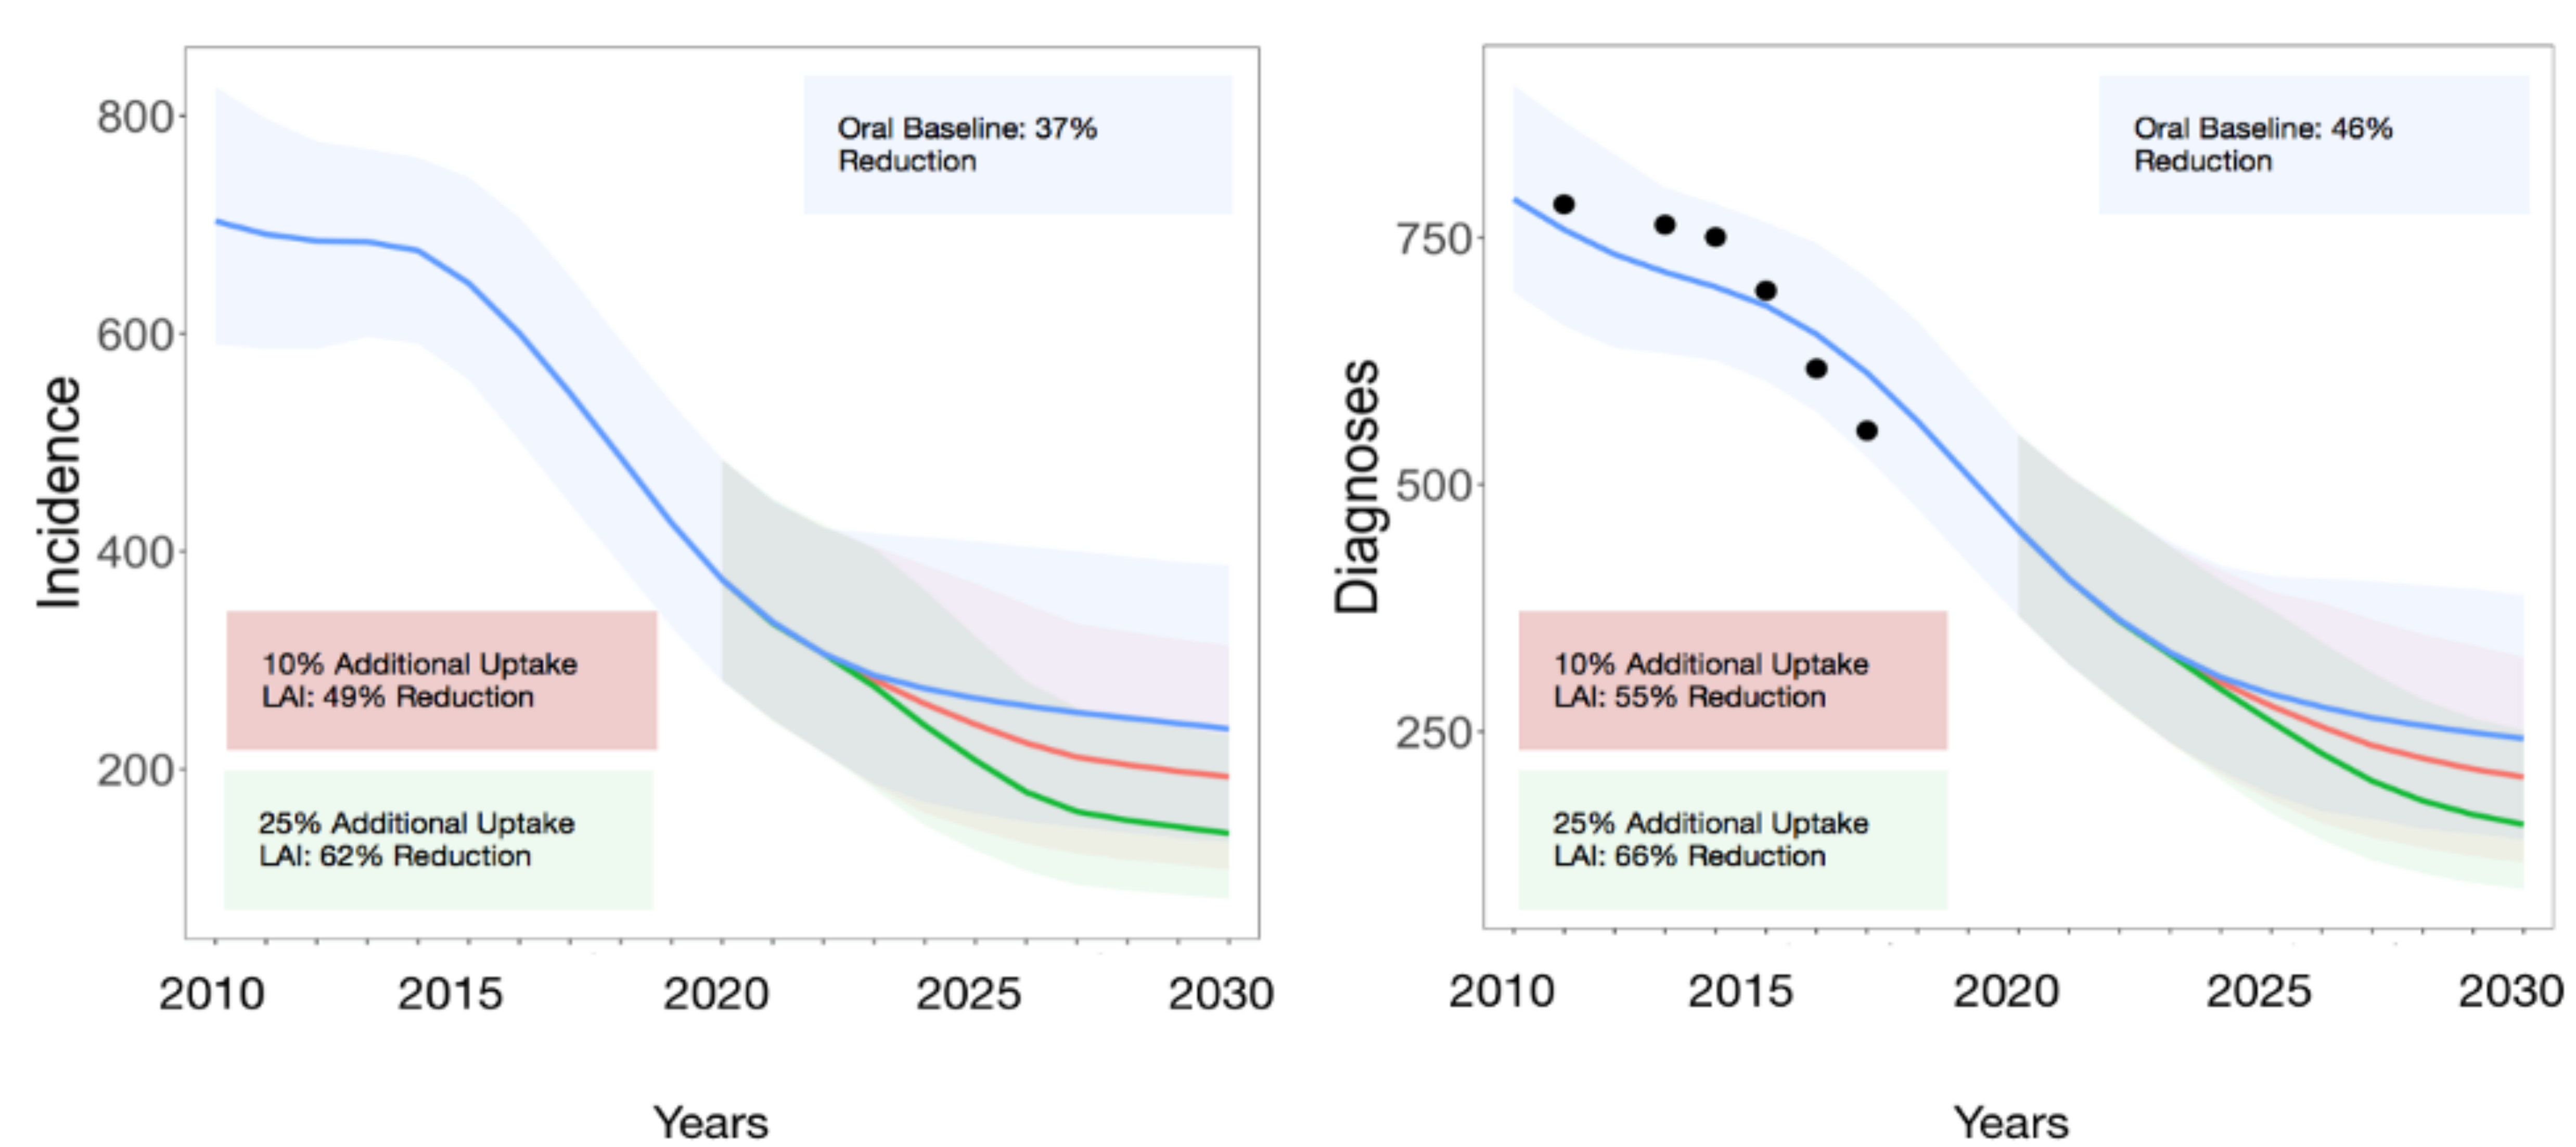
Incidence
800
600
400
200
Oral Baseline: 37% Reduction
10% Additional Uptake LAI: 49% Reduction
25% Additional Uptake LAI: 62% Reduction
2010
2015
2020
2025
2030
Years
Diagnoses
750
500
250
Oral Baseline: 46% Reduction
10% Additional Uptake LAI: 55% Reduction
25% Additional Uptake LAI: 66% Reduction
2010
2015
2020
2025
2030
Years

Key Findings

- In the absence of any intervention, we projected a 19% reduction (95% Credible Interval, CrI 1-36%) in HIV incidence among MSM from 2020-2030 across all 32 cities.
- A greater reduction in HIV incidence was observed between uptake levels as opposed to between LAI and Oral PrEP
- Reductions in incidence after introducing LAI PrEP driven primarily by increased uptake/sustained usage rather than increased efficacy.
- Greatest potential impact of LAI PrEP is in expansion of total PrEP uptake in conjunction with oral PrEP.
- Since potential effects vary by city, strategies to expand PrEP use should account for local dynamics.

Funding

- This work was supported by grants from the National Institutes of Health [K08-MH118094 to A.T.F., K01-AI138853 to P.K., T32-HL007024, and P30-AI094189]. The funders had no role in study's design, conduct, or reporting.

References

- 1. Anthony Todd Fojo, Melissa Schnure, Parastu Kasaie, David W. Dowdy, and Maunank Shah. What will it take to end HIV in the united states? Annals of Internal Medicine, 174(11):1542{1553, 2021.