Poster Number **00798**

URBAN VERSUS RURAL ACHIEVEMENTS OF 90:90:90 TARGETS IN 12 AFRICAN COUNTRIES

Suhani S. Patel[1], Mansoor Farahani[1,2], Rachel Bray[1,2], Jessica Justman[1,2], Wafaa M. El-Sadr [1,2]
[1] Columbia University, Mailman School of Public Health, Department of Epidemiology [2]ICAP at Columbia University

BACKGROUND

The relationship between the HIV epidemic and urbanicity is complex, and the HIV urban-rural characteristics are not fully understood. To achieve the UNAIDS 90-90-90 targets and ultimately end the epidemic, it is critical to discover potential geographical areas of concern. One such method is to assess the epidemic in rural vs. urban areas. Several differences between rural and urban communities including socio-demographic factors and access to healthcare exist, which can impact the outcome of HIV interventions. Currently, there is limited information in the literature on the HIV epidemic characteristics in rural versus urban areas.

In this analysis, we compared the HIV epidemic characteristics and the progress toward the UNAIDS 90-90-90 targets in urban versus rural populations among adults ages 15-64 years in 12 African countries.

METHODS

Data were obtained from the Population-Based HIV Impact Assessment (PHIA) Surveys in Lesotho*, Eswatini, Zambia, Zimbabwe, Malawi, Namibia, Uganda, Tanzania, Rwanda, Cameroon, Ethiopia**, and Cote d'Ivoire (2015-2019).

Consenting participants from randomly selected households provided demographic and clinical information and whole blood for household HIV testing per national guidelines.

Estimates of on-treatment status (2nd 90) were based on self-report and antiretroviral detection in blood. Viral load suppression (VLS) (3rd 90) was defined as HIV RNA <1000 copies/ml.

In this analysis, we applied multilevel logistic regression models using survey weights. Weighted variances were estimated using Rao-Scott Chi-Square tests via jackknife series.

* In Lesotho survey, areas with moderate population density, or a lesser extent of economic activities or infrastructure was considered as pre-urban areas. However, these areas were included as urban areas in this analysis for consistency.

** The PHIA survey in Ethiopia was only in urban area. For the purpose of this analysis, an urban area was defined as large urban areas having a population of more than 50,000 residents, while rural areas was defined as a small urban area with a population of 50,000 or less.

Awareness of HIV status was greater in urban than rural Tanzania and Zambia. In contrast, Awareness of HIV status was greater in rural than urban Eswatini, Malawi, and Namibia.

Urban areas compared to rural areas share a disproportionate burden of HIV.

RESULTS

- **Prevalence:** The HIV prevalence in rural areas ranged from 2.3% (95% CI: 1.8-2.8) in Cote d'Ivoire to 26.0% (95% CI: 24.8%-27.2%) in Eswatini, and in urban areas from 2.8% (95% CI: 2.3%-3.4%) to 29.8% (95% CI: 25.5%-34.0%) in Eswatini. The HIV prevalence was statistically significantly higher in urban areas than rural areas in the following six countries: Malawi, Tanzania, Uganda, Zambia, and Rwanda. In contrast, the HIV prevalence in Namibia rural areas was higher than in urban areas.
- **Gender:** In all these countries, HIV prevalence was higher among women than men in urban and rural areas.
- **Age:** In all these countries, HIV prevalence in both urban and rural areas was significantly larger in the 35-44 years, 45-54 years, age groups.
- **Wealth index:** Among people living in a household with a wealth index below the 60th percentile, HIV prevalence was significantly higher in urban than rural areas in Eswatini, Lesotho, Tanzania, and Zambia.
- **Education:** Among individuals with below secondary education, the prevalence of HIV is generally higher in urban locations than in rural.
- **Marital Status:** In all these countries, the HIV prevalence was higher among those divorced, separated, or widowed, whether they lived in urban or rural areas.
- **90-90-90 Targets:** Awareness of HIV status was greater in rural than urban Tanzania and Zambia (Figure 1). In contrast, Awareness of HIV status was greater in urban than rural Eswatini, Malawi, and Namibia. (Figure 1) Awareness ranged from 55.1% (95% CI: 50.4-59.8) in rural Cameroon to 88.6 (95% CI: 87.2-90.0) in rural Eswatini. (Figure 1) We found no significant differences between rural and urban areas when comparing antiretroviral uptake and viral suppression. (Figure 1)

Figure 1.
UNAIDS 90-90-90 Targets by Rural vs. Urban Regions

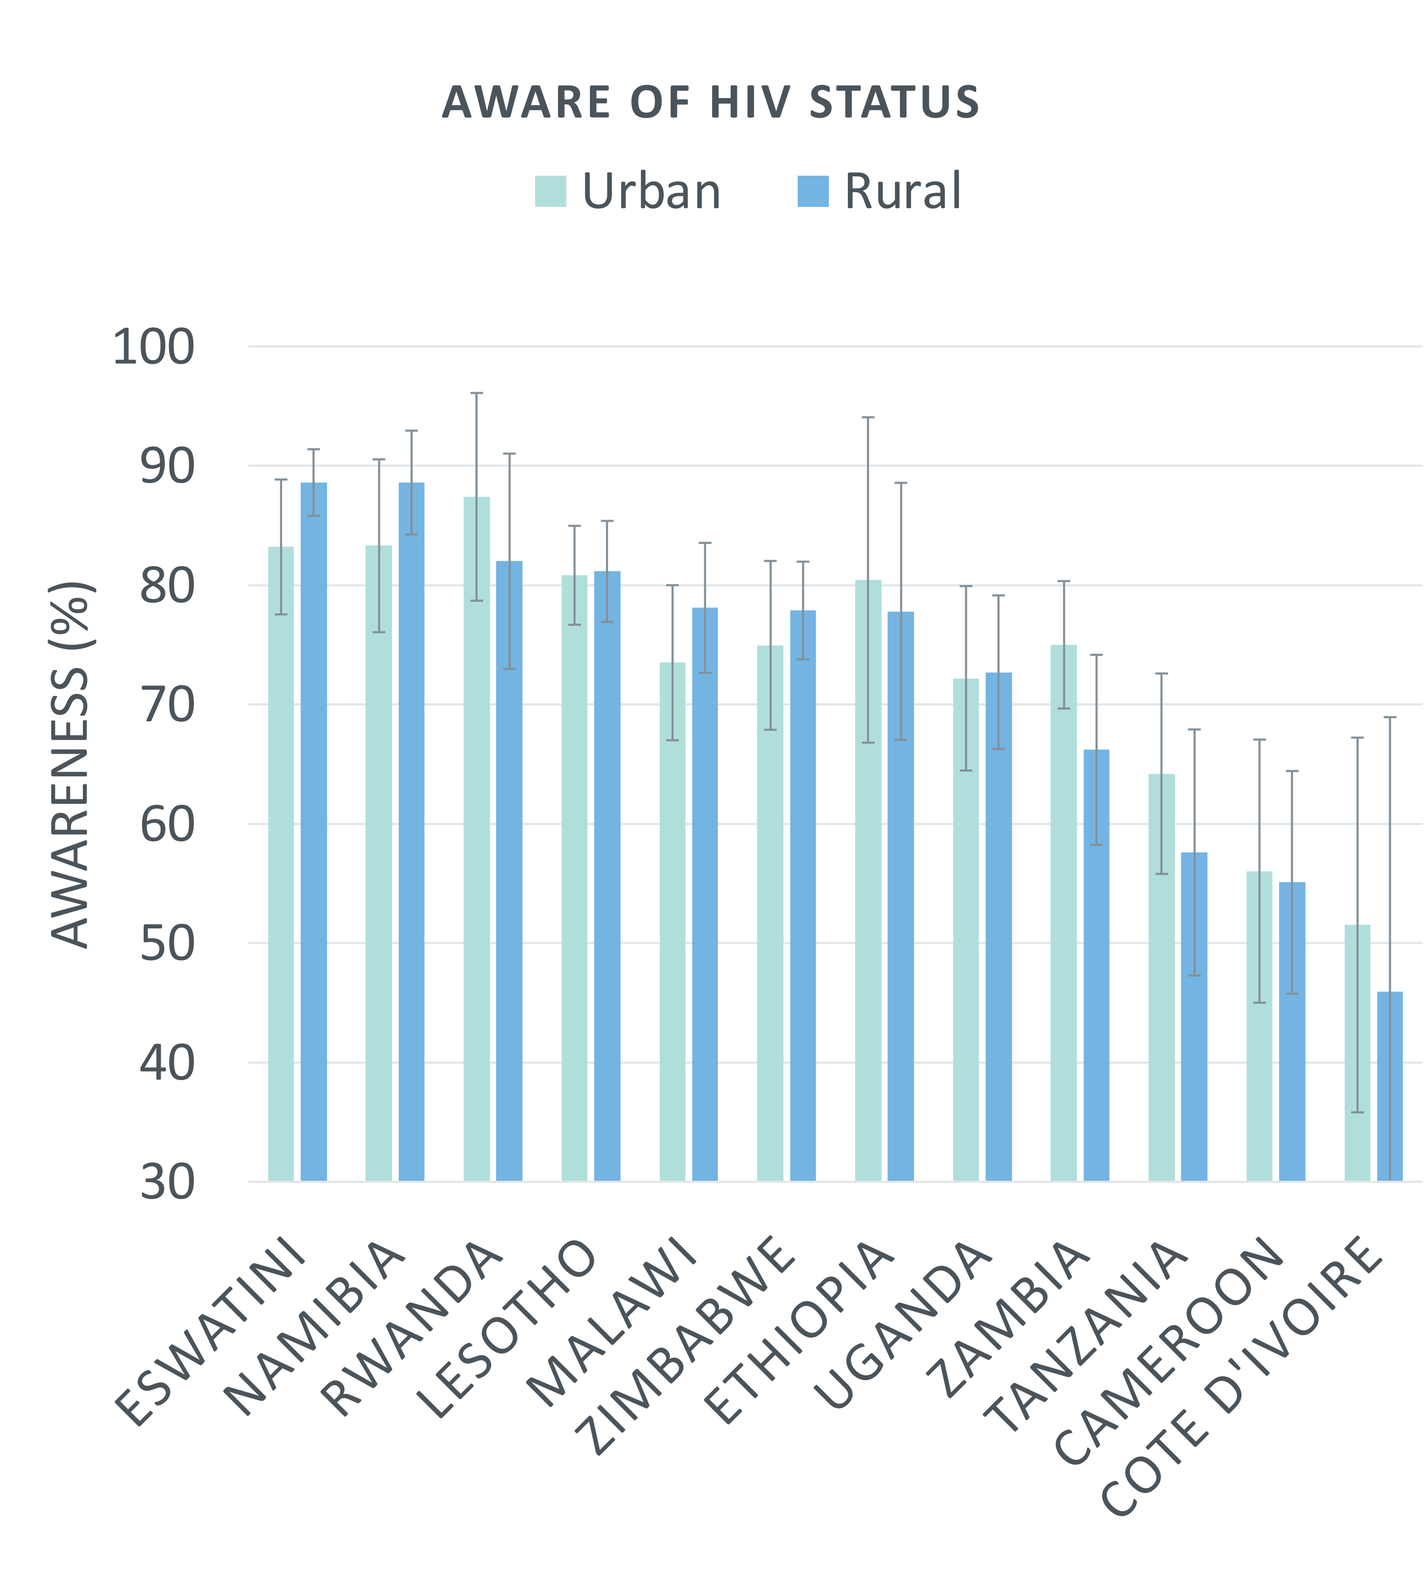
AWARE OF HIV STATUS
Urban
Rural
AWARENESS (%)
100
90
80
70
60
50
40
30
ESWATINI
NAMIBIA
RWANDA
LESOTHO
MALAWI
ZIMBABWE
ETHIOPIA
UGANDA
ZAMBIA
TANZANIA
CAMEROON
COTE D'IVOIRE

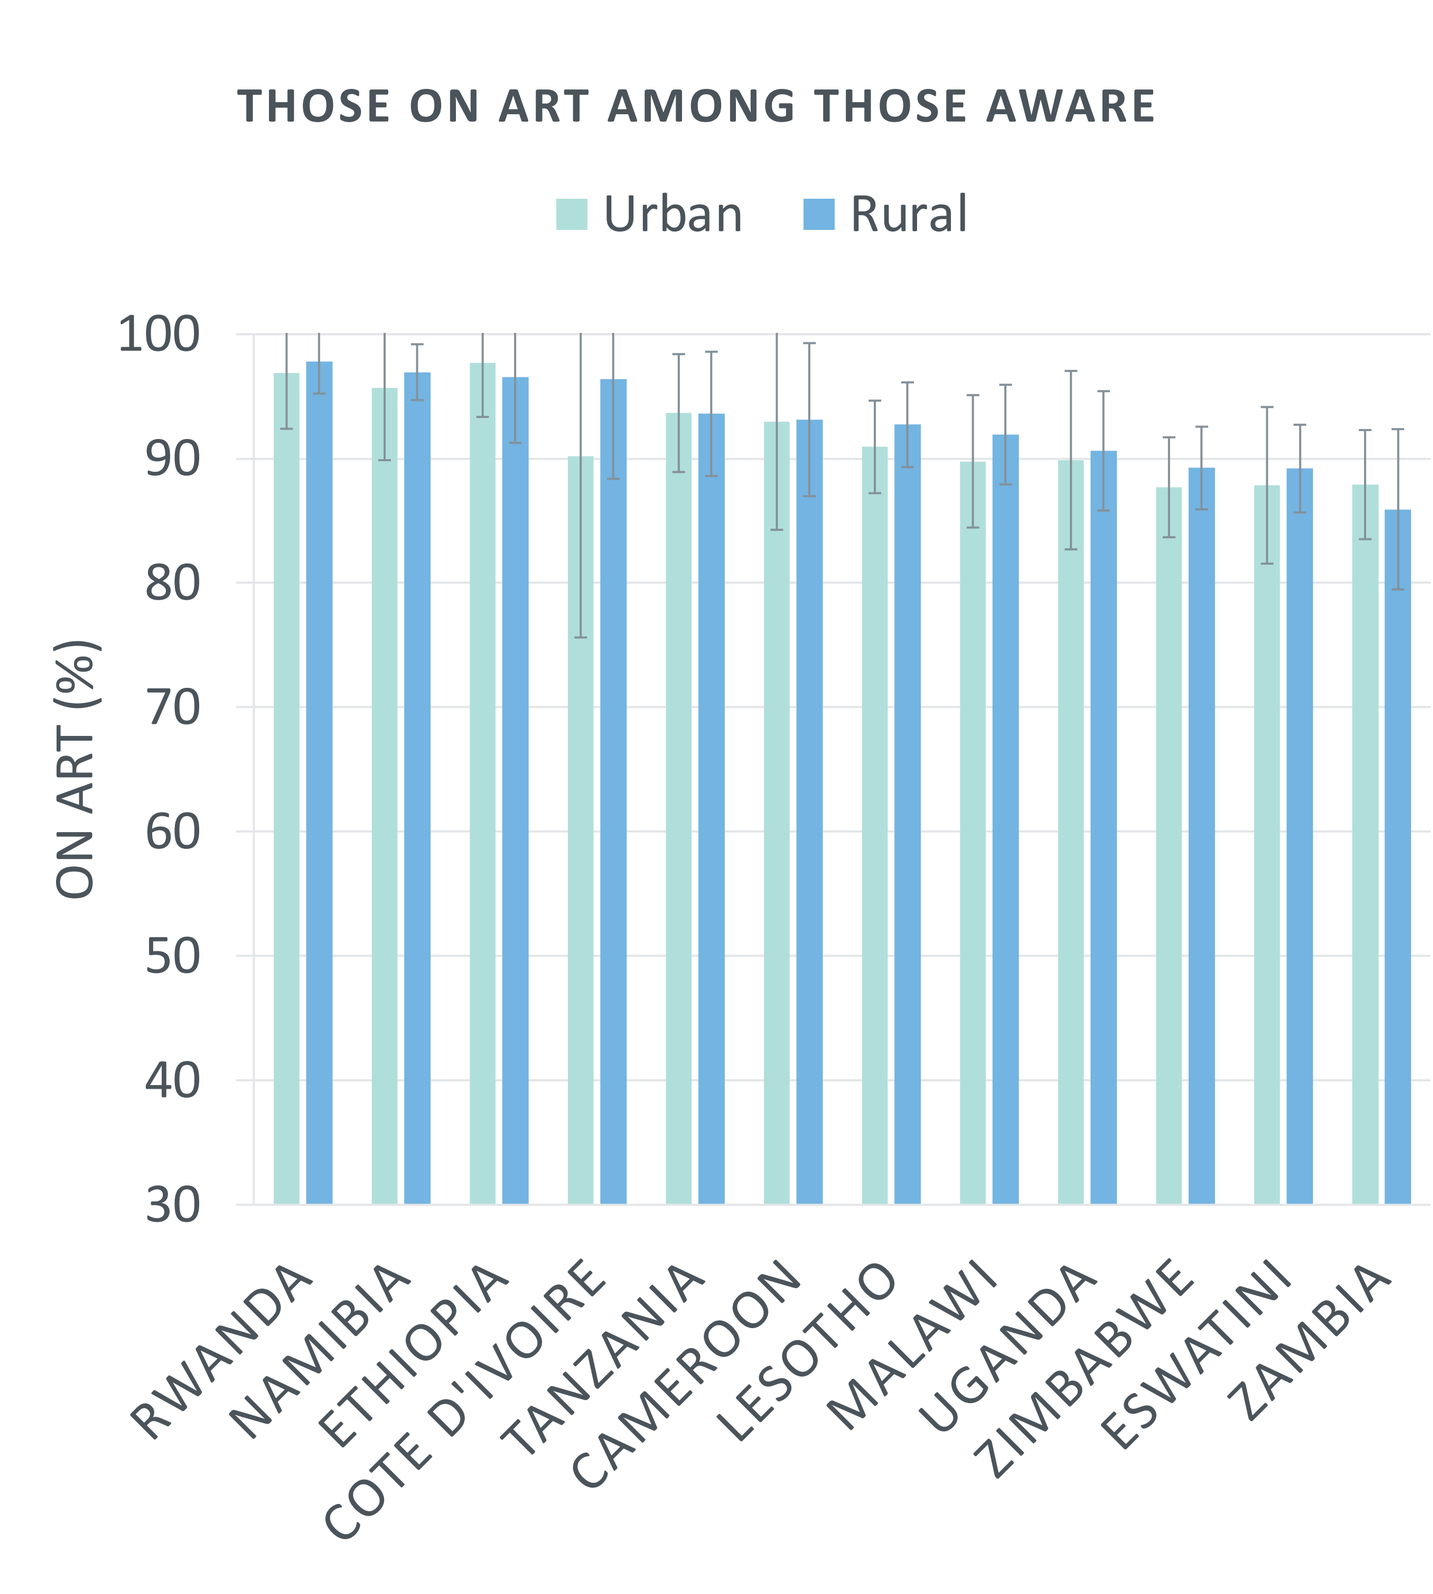
THOSE ON ART AMONG THOSE AWARE
Urban
Rural
ON ART (%)
100
90
80
70
60
50
40
30
RWANDA
NAMIBIA
ETHIOPIA
COTE D'IVOIRE
TANZANIA
CAMEROON
LESOTHO
MALAWI
UGANDA
ZIMBABWE
ESWATINI
ZAMBIA

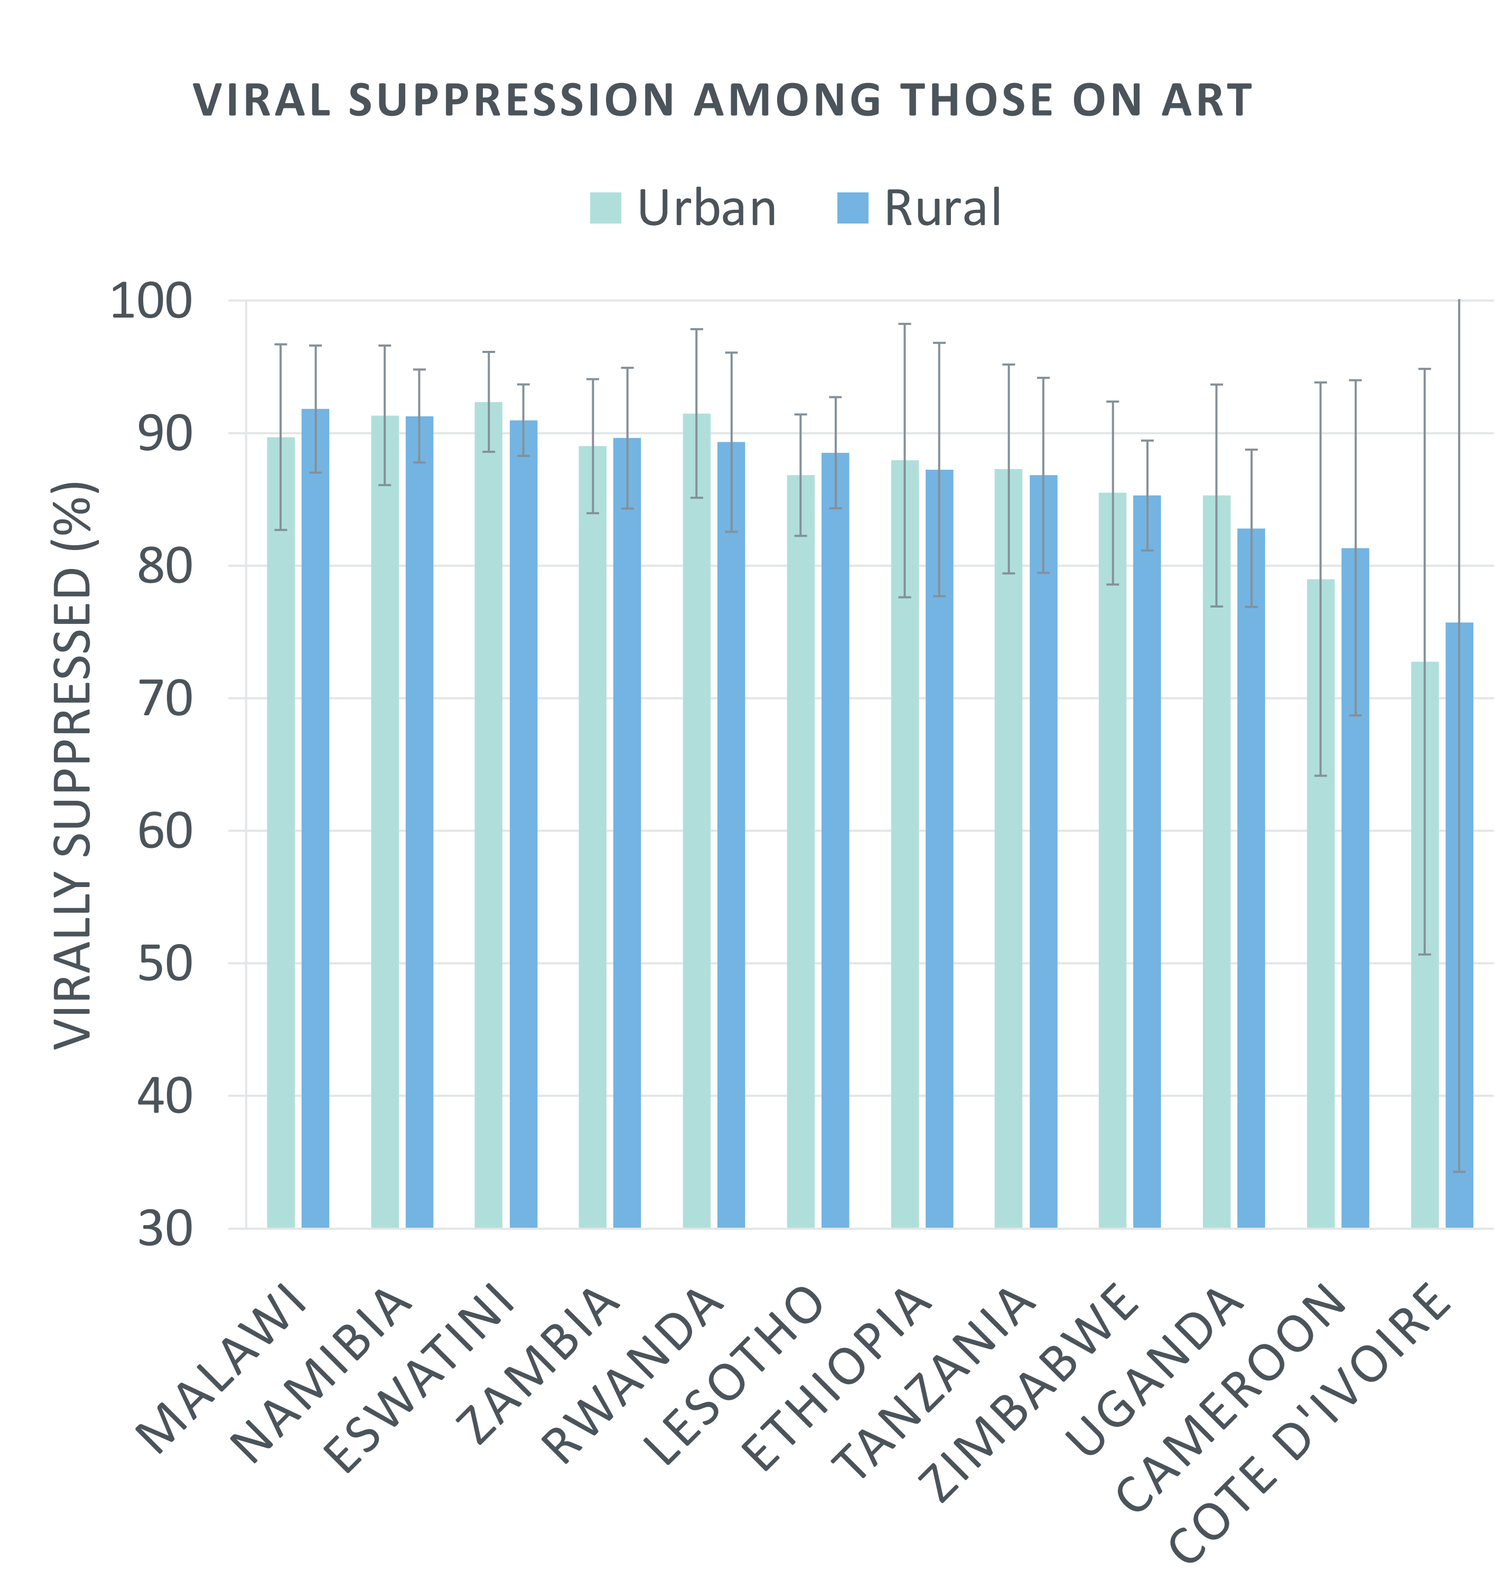
VIRAL SUPPRESSION AMONG THOSE ON ART
Urban
Rural
VIRALLY SUPPRESSED (%)
100
90
80
70
60
50
40
30
MALAWI
NAMIBIA
ESWATINI
ZAMBIA
RWANDA
LESOTHO
ETHIOPIA
TANZANIA
ZIMBABWE
UGANDA
CAMEROON
COTE D'IVOIRE

Table 1.
Prevalence of HIV by Selected Variables and Progress Toward 90-90-90 Targets in Urban vs. Rural Regions in 12 African Countries

	Cameroon		Cote d'Ivoire		Eswatini		Ethiopia		Lesotho		Malawi	
	Rural	Urban	Rural	Urban	Rural	Urban	Small urban	Large urban	Rural	Urban	Rural	Urban
Prevalence (national level):	3.5	3.8	2.3**	3.1**	26.0*	29.8*	3.2	2.8	24.9	26.3	9.7***	14.2***
gender:												
male	2.5	2.2	1.4	1.8	19.3*	23.5*	2.0	1.9	20.3	21.3	7.9***	10.8***
female	4.5*	5.5	3.3**	4.3**	31.6	34.9	4.4	3.8	29.8	30.9	11.3***	17.7***
age:												
15-24 years	1.2	1.2	0.2***	0.8***	8.5*	11.1*	0.9	0.6	6.1***	8.5***	2.3**	3.4**
25-34 years	3.9	3.4	2.1	2.4	34.1	29.9	2.8	2.4	26.3	28.1	10.6***	16.1***
35-44 years	5.5*	7.4*	3.2***	6.4***	48.3	49.9	6.1	6.4	44.0	44.2	18.1***	27.8***
45-54 years	5.8	7.2	4.2	5.2	41.5	44.1	6.2	5.9	41.6	40.9	17.0***	33.3***
65+ years	4.2	5.1	5.1	7.7	20.2	22.5	4.7***	1.9***	32.6	30.9	13.0***	20.9***
wealth:												
< 60th Percentile	3.2*	3.9*	3.1	3.3	26.1***	35.6***	3.8	3.0	26.2***	32.7***	10.0	11.9
≥ 60th Percentile	2.1*	4.5*	0.0	3.0	28.1**	46.1**	3.5	3.0	26.6	21.5	11.3	11.9
education:												
below secondary	3.6***	5.3***	2.6***	3.9***	30.3***	42.3***	5.0	4.0	36.3	41.1	10.2***	17.5***
secondary and above	3.5	3.3	0.1*	0.9*	23.4	26.3	2.0	1.5	24.0*	25.8*	7.4***	11.6***
marital status:												
married, or living together					34.3	34.5	3.2	2.4	27.5**	30.7**	10.2***	17.2***
divorced, separated, or widowed	N/A		N/A		41.8	45.9	10.2	9.6	53.9	54.9	25.1***	37.8***
never-married					16.5***	23.4***	1.1	0.8	10.3	12.0	1.9***	3.6***
pregnancy status:												
pregnant	3.3	3.5	2.0	3.1	34.1	37.9	2.6	1.7	20.5	28.1	7.8	13.1
not pregnant	4.6*	5.6*	3.4*	4.4*	31.5	34.9	4.5	4.0	30.2	31.0	11.6***	18.1***
90-90-90 targets:												
Awareness	55.1	56.0	45.9	51.5	88.6***	83.2***	77.8	80.4	81.2	80.8	78.1**	73.5**
On ART Treatment	93.1	93.0	96.4	90.2	89.2	87.9	96.6	97.7	92.7	90.9	91.9	89.8
Virally Suppressed	81.4	79.0	75.7	72.8	91.0	92.4	87.3	87.9	88.5	86.8	91.8	89.7

	Namibia		Tanzania		Uganda		Zambia		Zimbabwe		Rwanda	
	Rural	Urban	Rural	Urban	Rural	Urban	Rural	Urban	Rural	Urban	Rural	Urban
Prevalence (national level):	15.0***	10.8***	4.2***	6.0***	5.8***	7.5***	9.2***	15.3***	13.1**	14.4**	2.5***	4.8***
gender:												
male	10.7***	8.2***	3.4	3.4	4.7***	4.6***	7.4***	11.6***	11.4	12.3	2.0***	3.2***
female	19.0***	13.2***	5.0***	8.3***	6.7***	9.8***	11.1***	18.7***	14.7**	16.2**	3.0***	6.5***
age:												
15-24 years	4.6**	3.4**	1.2	1.6	1.7***	3.0***	3.0***	4.6***	3.9**	5.4**	0.7***	1.6***
25-34 years	13.9***	7.5***	4.6*	5.8*	6.9*	8.5*	9.2***	16.2***	14.6	13.2	2.1***	3.5***
35-44 years	27.4***	20.6***	8.1***	10.9***	10.4**	13.6**	16.9***	29.9***	25.1	25.0	3.8***	8.2***
45-54 years	27.6**	22.5**	7.2***	13.0***	10.5***	16.6***	17.3***	32.5***	25.3*	29.5*	5.4***	13.6***
65+ years	18.4***	12.2***	3.4***	8.4***	7.7	8.8	12.4***	24.5***	10.2***	17.6***	4.4***	11.6***
wealth:												
< 60th Percentile	15.8	15.8	4.2***	9.0***	5.2**	7.1**	8.7***	17.6***	13.1**	17.5**	2.5***	6.2***
≥ 60th Percentile	17.6	19.7	3.9***	10.1***	4.3	4.3	8.0	0.0	14.1**	21.6**	2.7	4.5
education:												
below secondary	13.5**	18.8**	4.5***	8.3***	7.7***	14.0***	9.0***	17.7***	11.5***	20.8***	2.9***	7.3***
secondary and above	15.2***	10.4***	2.5	2.4	6.0***	10.1***	9.0***	15.7***	13.3***	15.8***	1.4***	2.6***
marital status:												
married, or living together	17.7***	12.1***	4.0***	6.4***	6.2***	7.7***	9.7***	19.0***	14.2	15.5	2.6***	5.3***
divorced, separated, or widowed	24.7	20.3	10.3***	14.2***	13.8***	19.8***	26.3***	41.7***	26.2***	33.0***	7.7***	18.5***
never-married	12.4***	8.7***	1.3***	2.4***	1.3**	2.2**	3.7***	5.5***	4.1**	5.6**	1.1***	2.2***
pregnancy status:												
pregnant	14.7	9.8	3.3***	7.7***	5.1	7.4	5.6***	14.8***	11.2	9.0	1.7***	4.8***
not pregnant	19.3***	13.3***	5.1***	8.4***	6.8***	9.9***	11.7***	19.1***	14.9**	16.6**	3.1***	6.7***
90-90-90 targets:												
Awareness	88.6***	83.3***	57.6**	64.2**	72.7	72.2	66.2***	75.0***	77.9	75.0	82.0*	87.4*
On ART Treatment	97.0	95.7	93.6	93.7	90.6	89.9	85.9	87.9	89.2	87.7	97.8	96.9
Virally Suppressed	91.3	91.3	86.8	87.3	82.8	85.3	89.6	89.0	85.3	85.5	89.3	91.5

Note: In Lesotho survey, areas with moderate population density, or a lesser extent of economic activities or infrastructure was considered as pre-urban areas. However, these areas were included as urban areas in this analysis for consistency. The PHIA survey in Ethiopia was only in urban area. For the purpose of this analysis, an urban area was defined as large urban areas having a population of more than 50,000 residents, while rural areas was defined as a small urban area with a population of 50,000 or less.

***p<0.01 **p<0.05 *p<0.1

CONCLUSIONS

Our results indicate that these countries may benefit from targeted intervention to improve people's awareness of their HIV status (1st 90)-- the critical first step in the HIV care and treatment cascade.

The findings from these countries with generalized epidemics could inform public health planners and policymakers.

While rural areas have many challenges in accessing care, the health system in urban areas has a significantly higher HIV burden. With the rapid rise in urbanization in many African countries, we must continue monitoring and enhancing HIV prevention and treatment services in urban areas.

This research was supported by the United States President's Emergency Plan for AIDS Relief (PEPFAR) through the U.S. Centers for Disease Control and Prevention (CDC) under the terms of Cooperative Agreement Number NU2GGH001226. The contents are solely the responsibility of ICAP and do not necessarily reflect the views of the United States Government.

A major global health organization with projects in more than 30 countries, ICAP at Columbia University works to transform the health of populations through innovation, science, and global collaboration. Since its founding in 2003 at the Columbia Mailman School of Public Health, more than 2.6 million people have received HIV care through ICAP-supported programs. Learn more online at icap.columbia.edu