Adolescent and Parent Experiences with Long-acting Injectables in the MOCHA Study

00739

Elizabeth D. Lowenthal[1,2], Jennifer Chapman[2], Katherine Calabrese[3], Ryan Milligan[4], Allison Agwu[5], Andres Camacho-Gonzalez[6], Christiana Smith-Anderson[7], Jared Kneebone[8], Cindy McCoig[9], Conn M. Harrington[10], Rodica Van Solingen-Ristea[11], Adeola Adeyeye[12], Jack Moye[13], Carolyn Bolton Moore[14], Aditya H. Gaur[15]

[1]University of Pennsylvania, Philadelphia, PA, USA, [2]Children's Hospital of Philadelphia, Philadelphia, PA, USA, [3]FHI 360, Durham, NC, USA, [4]Frontier Science and Technology Research Foundation, Inc, Brookline, MA, USA, [5]The Johns Hopkins University School of Medicine, Baltimore, MD, USA, [6]Emory University, Atlanta, GA, USA, [7]University of Colorado, Aurora, CO, USA, [8]Frontier Science and Technology Research Foundation, Inc, Buffalo, NY, USA, [9]ViiV Healthcare, Madrid, Spain, [10]ViiV Healthcare, Research Triangle Park, NC, USA, [11]Janssen, Beerse, Belgium, [12]National Institutes of Health, Rockville, MD, USA, [13]National Institutes of Health, Bethesda, MD, USA, [14]Centre for Infectious Disease Research in Zambia, Lusaka, Zambia, [15]St Jude Children's Research Hospital, Memphis, TN, USA

BACKGROUND

- The ongoing More Options for Children and Adolescents Study (MOCHA; Clinicaltrials.gov NCT03497676) is a phase I/II study, the first to examine use of long-acting injectable (LA) antiretrovirals (cabotegravir (CAB) and rilpivirine (RPV)) in adolescents living with HIV (age 12-<18 years)
- Adolescents frequently struggle with adherence to medications, making LA a potentially attractive option for this age group
- MOCHA participants are the first virologically suppressed adolescents to access LA antiretrovirals (ARV)

METHODS

To assess LA acceptability issues of importance to adolescents, we used a mixed-methods approach among participants in this Phase I/II multi-center study including:

- Questionnaires completed by all adolescents at study entry (N=23) and after receipt of 3 injectable doses (CAB (N=8) or RPV (N=13)): included 1) Reasons for choosing the LA ARV; 2) Perceptions of injections; 3) Health-related quality of life (PedsQL™)
- In-depth telephone interviews with English-speaking adolescent participants (N=11) and their parents/caregivers ("parents"; N=10: 7 mothers, 1 father and 2 grandmothers)
 *1 mother had two enrolled adolescents
- Coding and thematic analysis using the consolidated framework for implementation research (CFIR)

RESULTS

TABLE 1. Characteristics of Cohort

Characteristic	All Adolescent Participants	Interviewed Adolescents
	N=23*	N=11
Age	Median: 16 Range: 12-17	Median: 15 Range: 12-17
Female Sex	10 (43.5%)	5 (45.5%)
Cohort		
CAB	8 (34.8%)	6 (54.5%)
RPV	15 (65.2%)	5 (45.5%)
Study Site		
Emory	8 (34.8%)	3 (27.3%)
Johns Hopkins	5 (21.7%)	3 (27.3%)
Lurie Children's	1 (4.3%)	1 (9.1%)
St. Jude	1 (4.3%)	0
Texas Children's	2 (8.7%)	0
U. Colorado	5 (21.7%)	3 (27.3%)
USC	1 (4.3%)	1 (9.1%)

*1 discontinued study drug after the entry visit due to hypersensitivity reaction to the first oral dose. 1 participant declined injections after needle insertion, but prior to receiving the first IM dose. Both were in the RPV group.

Themes related to not having to remember to take pills/avoiding the stress of monitoring adolescents' daily medication-taking and not having to worry about hiding pills from peers dominated discussions of the relative advantage of LA versus pill-based regimens.

Concerns regarding long-term use of LA antiretrovirals included questions about ability to maintain a routine injection schedule when busy with school, extracurricular activities and work, particularly for those planning to move to college.

- Overall, perceptions of injections were favorable
- Health-related QoL was similar before vs. after initiation of LA
- Of 21 adolescents who received 3 study injections, 90.5% (19/21) reported wanting to receive LA ARV even after the study ended (15/21 (71.4%) definitely, and 4/21 (19.1%) probably).

TABLE 2. Questionnaire Responses- Reasons for Choosing LA ARV (N=22*)

Prompt: *You have chosen to be in a study where you will get medicine shots. We call these shots long-acting injectable medicines. We want to understand your reason or reasons for wanting to try the long-acting injectable medicines.*	Reason for wanting to try LA ARV**	Primary reason for wanting to try LA ARV
Interested in research of new treatments	18 (81.8%)	6 (27.3%)
Someone in my clinic suggested	12 (54.5%)	0
Someone in my family suggested	7 (31.8%)	0
I do not like the way my medicine makes me feel	2 (9.1%)	0
Current medicine may cause future problems	2 (9.1%)	0
Difficult to take current medicines regularly	11 (50%)	4 (18.2%)
Hope to take medicine without daily pills	22 (100%)	12 (54.5%)

*One participant withdrew from the study before obtaining an injection and did not complete the survey
**Participants indicated all that were applicable to them

BOX 1
Example Quotes:
1. *"Parents are trying to give them more independence, but we don't know for sure if they're actually taking their meds or when they're skipping them. Versus being able to say, hey, you gotta go to the clinic and get your shot today. And then, knowing they go and get their shot. And just feeling good that they have that independence, but also, were not missing that medicine."* (parent of 12-year-old)

2. *"Even when I was telling him that I don't want him to get it, he was like, 'oh, mommy, please, please, please, let me get it, let me get it, I just want to get out of this medication…'"* (parent of 14-year-old)

3. *"(after the loading dose) I couldn't walk for a while because it would hurt, and I would start to limp… ..And it felt like my butt was gonna fall off." " The second dose was fine… ..(it hurt) for around five minutes, ten minutes… ..but the second time was just two milliliters. So, it was less than the first time." (17-year-old female)*

TABLE 3. CFIR Domains and Codes Featuring Prominently in Qualitative Data

CFIR DOMAIN	EXAMPLE CODES	SUMMARY OF REPRESENTATION OF CODES IN DATA
CHARACTERISTICS OF INJECTION	RELATIVE ADVANTAGE [1]	Perception of advantage of implementing LA ART vs. pill-based treatment
	ADAPTABILITY	Degree to which LA ART can be adapted or refined to meet the needs of adolescents
	COMPLEXITY	Perceived difficulty of adolescents accessing and maintaining LA ART
INNER SETTING	COMPATIBILITY	Degree of fit of LA ART with stakeholders' norms, values and systems
	TENSION FOR CHANGE[1, 2]	Degree to which stakeholders consider current situation in need of change
OUTER SETTING	PATIENT NEEDS & RESOURCES	Extent to which adolescent patients' needs, barriers and facilitators are known and prioritized by implementer
	COSMOPOLITANISM	Degree to which implementer is networked with complementary organizations
INDIVIDUAL CHARACTERISTICS	SELF-EFFICACY[1]	Individual belief in own/adolescent's capacity to execute course of action to achieve implementation goals
	INDIVIDUAL STAGE OF CHANGE[2]	Characterization of phase an individual is in towards accepting and maintaining LA ART
PROCESS	PLANNING & ENGAGING	Development of plan and involvement of appropriate individuals in intervention
	EXECUTING	Ability to carry out the implementation according to plan
	REFLECTING & EVALUATING[3]	Feedback about adolescents' experience of receiving LA ART

1,2,3- numbers correspond with codes represented by example quotes in Box 1

CONCLUSIONS

- The first adolescents to access LA ARV through the MOCHA Study and their parents found the formulation to be acceptable.
- Adolescent-specific concerns such as differences of opinion between adolescents and their consenting parent regarding whether or not they should initiate LA ARV arose. However, most dyads indicated that the relative advantage of avoiding pill-taking for long-term therapy would outweigh potential disadvantages of making the switch.

ACKNOWLEDGEMENTS

The study team would like to thank all participating adolescents, their parents and caregivers, and all staff at the study sites who assisted with making this work possible. We would also like to acknowledge the following individuals for their contributions towards the study and this abstract: Shawn Ward, Pearl Samson, Jenny Huang, Barbara Heckman, and Kelsey Simon.

For more information, visit impaactnetwork.org and follow us:

Facebook: IMPAACTNetwork | Twitter: @IMPAACTNetwork | LinkedIn: IMPAACTNetwork

Overall support for the International Maternal Pediatric Adolescent AIDS Clinical Trials Network (IMPAACT) was provided by the National Institute of Allergy and Infectious Diseases (NIAID) with co-funding from the Eunice Kennedy Shriver National Institute of Child Health and Human Development (NICHD) and the National Institute of Mental Health (NIMH), all components of the National Institutes of Health (NIH), under Award Numbers UM1AI068632-15 (IMPAACT LOC), UM1AI068616-15 (IMPAACT SDMC) and UM1AI106716-09 (IMPAACT LC), and by NICHD contract number HHSN275201800001I. The content is solely the responsibility of the authors and does not necessarily represent the official views of the NIH.each o

Presented at the 2022 Conference on Retroviruses and Opportunistic Infections (CROI)
12-16 February 2022