SAFETY AND PK OF LONG-ACTING CABOTEGRAVIR AND RILPIVIRINE IN ADOLESCENTS

00738

Carolyn Bolton Moore[1], Edmund Capparelli[2], Katherine Calabrese[3], Brookie M. Best[2], Shawn Ward[4], Cindy McCoig[5], Herta Crauwels[6], Allison Agwu[7], Pearl Samson[4], Barbara Heckman[4], Susan L. Ford[8], Adeola Adeyeye[9], Andres Camacho-Gonzalez[10], Jack Moye[11], Aditya H. Gaur[12]

[1]Centre for Infectious Disease Research in Zambia, Lusaka, Zambia, [2]University of California San Diego, La Jolla, CA, United States, [3]FHI 360, Durham, NC, United States, [4]Frontier Science & Technology Research Foundation, Inc, Amherst, NY, United States, [5]ViiV Healthcare, Collegeville, PA, United States, [6]Janssen Research & Development, Beerse, Belgium, [7]The Johns Hopkins University School of Medicine, Baltimore, MD, United States, [8]GlaxoSmithKline, Research Triangle Park, NC, United States, [9]National Institute of Allergy and Infectious Diseases, Baltimore, MD, United States, [10]Emory University, Atlanta, GA, United States, [11]National Institute of Child Health and Human Development, Bethesda, MD, United States, [12]St Jude Children's Research Hospital, Memphis, TN, United States

Long-acting cabotegravir and rilpivirine, when given individually, in conjunction with background ART, were well tolerated in adolescents and achieved drug concentrations similar to those seen in adults.

BACKGROUND

- Long-acting cabotegravir (CAB-LA) plus long-acting rilpivirine (RPV-LA) are approved for maintenance of viral suppression in adults with HIV-1 infection
- IMPAACT 2017 (MOCHA) is a phase I/II non-comparative, open-label study to confirm the dose and evaluate safety, tolerability, acceptability, and pharmacokinetics (PK) of oral (PO) CAB, CAB-LA, and RPV-LA in adolescents, ≥12 to <18 years, living with HIV-1

METHODS

- Virologically suppressed (<50 copies/mL) adolescents with HIV-1 on stable combination antiretroviral therapy (ART) were enrolled into Cohort 1C (CAB) or Cohort 1R (RPV) based on background ART (background ART was continued throughout Cohort 1)
- Doses: 4 weeks of oral lead-in with CAB (30mg once daily) or RPV (25mg once daily), then CAB-LA (600mg/3mL at Week 4 and 400mg/2mL at Weeks 8 and 12) or RPV-LA (900mg/3mL at Week 4 and 600mg/2mL at Weeks 8 and 12) by gluteal intramuscular (IM) injection
- PK samples drawn at: Week 2 (oral dosing) and Weeks 4, 5, 6, 8, 12, 13, 14 and 16 (LA dosing)

TABLE 1. Baseline Characteristics (All-Treated Population)

	Cohort 1C (N=8)	Cohort 1R	Total (N=23)
Age in Years; Median (Q1,Q3)	14.5 (13.5, 17.0)	17.0 (15.0, 17.0)	16.0 (14.0,17.0)
Age; n (%)			
12 years	1 (12.5%)	1 (6.7%)	2 (8.7%)
13 years	1 (12.5%)	0	1 (4.3%)
14 years	2 (25.0%)	2 (13.3%)	4 (17.4%)
15 years	1 (12.5%)	1 (6.7%)	2 (8.7%)
16 years	0	3 (20.0%)	3 (13.0%)
17 years	3 (37.5%)	8 (53.3%)	11 (47.8%)
Sex at Birth; F/M, n (%)	2 (25.0%) / 6 (75.0%)	8 (53.3%) / 7 (46.7%)	10 (43.5%) / 13 (56.5%)
Race; n (%)			
Asian	1 (12.5%)	0	1 (4.3%)
Black/ African American	7 (87.5%)	11 (73.3%)	18 (78.3%)
White	0	4 (26.7%)	4 (17.4%)
Ethnicity; n (%)			
Hispanic or Latino	0	3 (20.0%)	3 (13.0%)
Weight in kg Median (Q1, Q3)	57.2 (44.2, 71.7)	63.0 (54.0, 75.0)	63 (47.7, 72.4)

Figure 1: Observed preliminary median (5th, 95th percentile) concentration-time data in adolescents (MOCHA) compared to pivotal Phase 3 Studies ATLAS and FLAIR in adults following oral lead in and 3 x monthly injections (CAB left panel, RPV right panel)

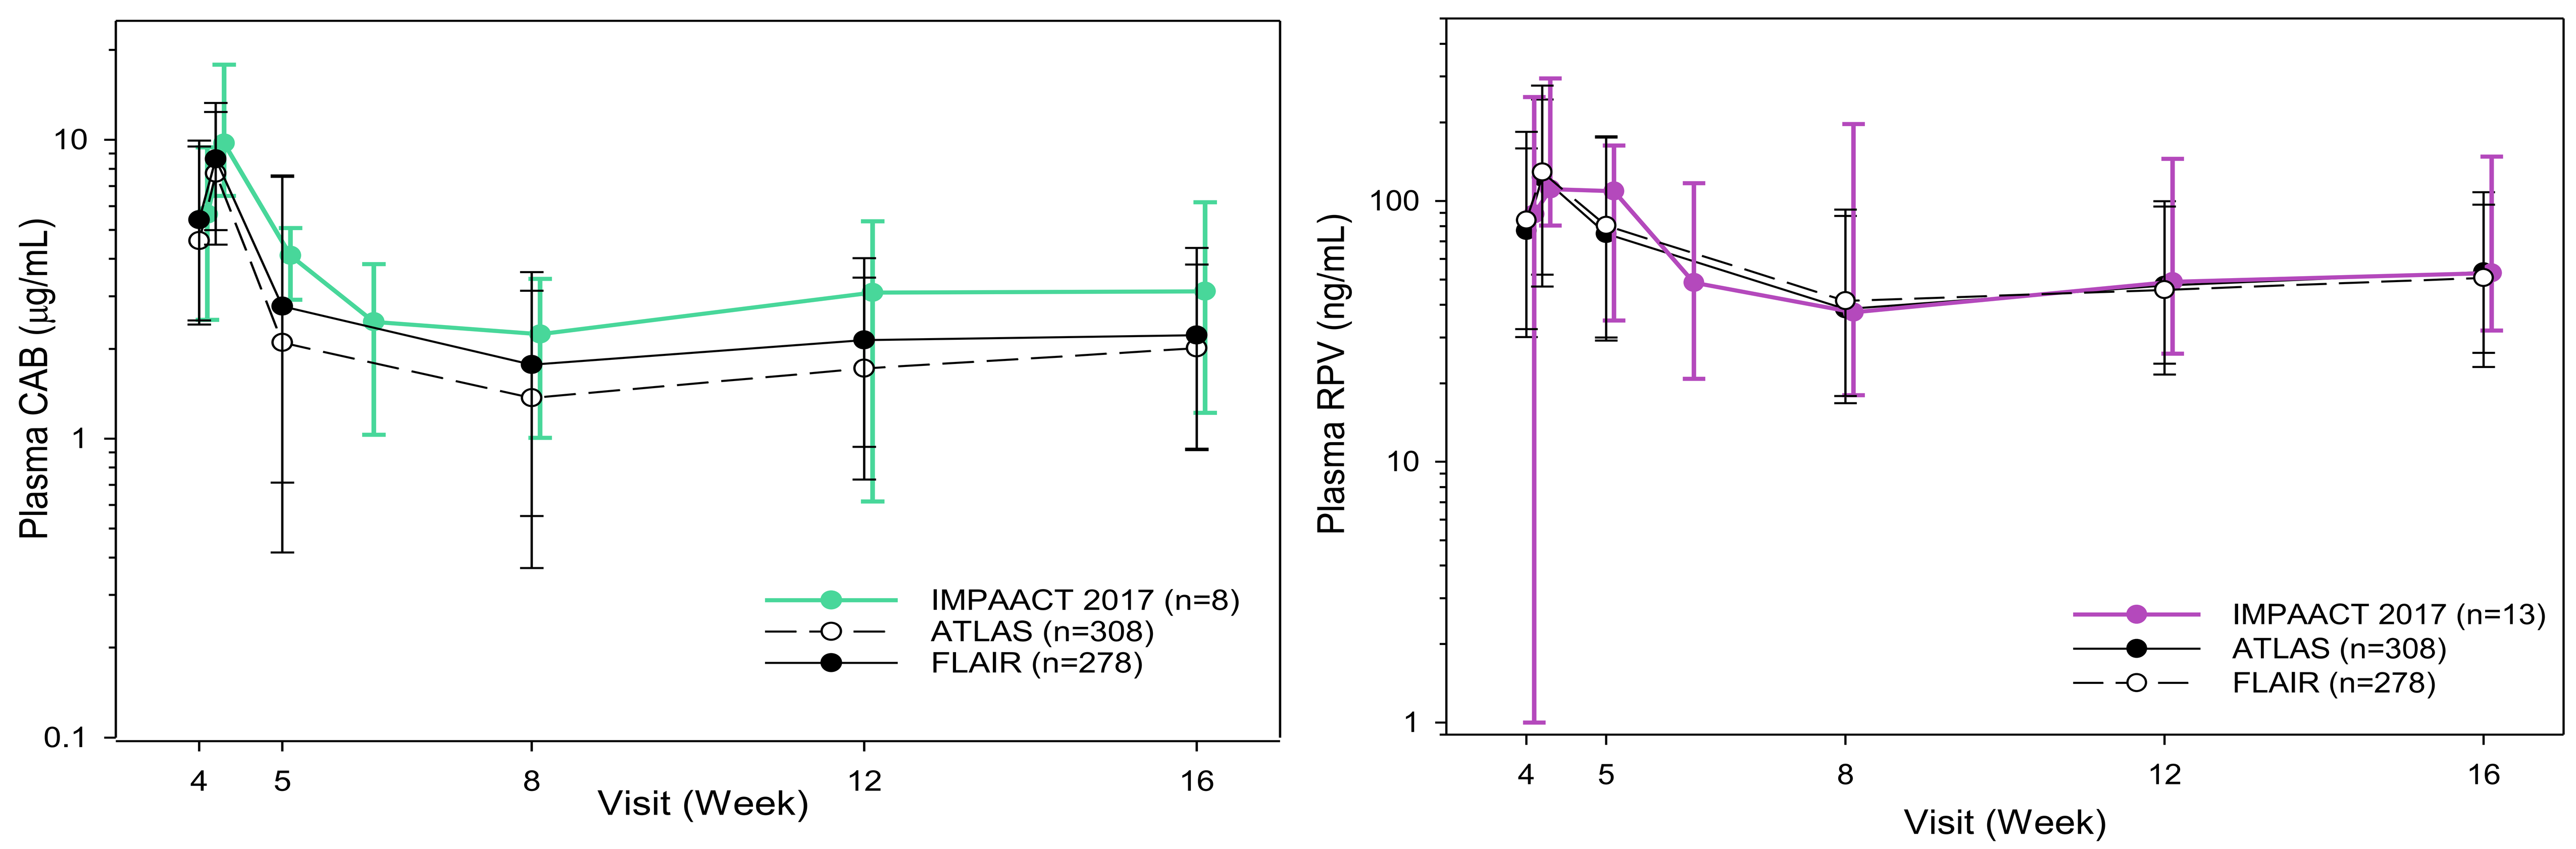
Plasma CAB (µg/mL)
10
1
0.1
4
5
8
12
16
Visit (Week)
IMPAACT 2017 (n=8)
ATLAS (n=308)
FLAIR (n=278)
Plasma RPV (ng/mL)
100
10
1
Visit (Week)
IMPAACT 2017 (n=13)
ATLAS (n=308)
FLAIR (n=278)

RESULTS

- Data per freeze date (10 Sept 2021): 23 participants enrolled (Table 1)
 - Cohort 1R: two premature treatment discontinuations; one due to hypersensitivity (after 1st oral dose, no systemic symptoms) and one due to pain with needle insertion prior to receiving first IM injection
- All participants to date were virally suppressed at Week 16 (Table 2)
- Injection site reactions were grade 1 or 2; none led to treatment discontinuation. One Grade 3 drug-related AE per cohort (Table 3 - 1C: insomnia [day 41], 1R: hypersensitivity [day 1; led to study withdrawal])
- Median (range) PK parameters met study targets with Q4 dosing (Figures 1 and 2)
 - Cohort 1C: *Week 2 Oral CAB* AUC_{0-T}: 160 (94.3-325) mcg*h/mL (target median 46-277)
 Week 16 IM CAB trough: 3.11 (range 1.22-6.19) mcg/mL (target median 0.71-6.7)
 - Cohort 1R: *Week 16 IM RPV* trough: 52.9 (range 31.9-148) ng/mL (target median 25-100)

Table 2: Viral Suppression through Week 16 (All-Treated Population)

Analysis Visit	HIV-1 RNA <50 copies/mL; n (%)	HIV-1 RNA ≥50 copies/mL; n (%)	Total; n (%)
Cohort 1C (N=8)			
Baseline	8 (100.0)	0	8 (100.0)
Week 2	8 (100.0)	0	8 (100.0)
Week 4b	8 (100.0)	0	8 (100.0)
Week 8	7 (87.5)	1 (12.5)	8 (100.0)
Week 12	8 (100.0)	0	8 (100.0)
Week 16	7 (100.0)	0	7 (100.0)
Cohort 1R (N=15)			
Baseline	14 (93.3)	1 (6.7)	15 (100.0)
Week 2	14 (100.0)	0	14 (100.0)
Week 4b	13 (92.9)	1 (7.1)	14 (100.0)
Week 8	13 (100.0)	0	13 (100.0)
Week 12	13 (100.0)	0	13 (100.0)
Week 16	13 (100.0)	0	13 (100.0)

Table 3: Drug-related Adverse Events through Week 16 (Evaluable Population)

System Organ Class	Cohort 1C (N=8)			Cohort 1R (N=13)		
Grade*; n (%)	1	2	3	1	2	3
Number of participants with ≥ one adverse events	1 (12.5)	3 (37.5)	1 (12.5)	5 (38.5)	3 (23.1)	1 (7.7)
Diarrhoea	1 (12.5)	0	0	0	0	0
Nausea	0	0	0	1 (7.7)	0	0
General disorders	2 (25.0)	3 (37.5)	0	5 (38.5)	3 (23.1)	0
Injection site hypoaesthesia	0	0	0	1 (7.7)	0	0
Injection site nodule	0	0	0	1 (7.7)	0	0
Injection site pain	2 (25.0)	3 (37.5)	0	5 (38.5)	3 (23.1)	0
Injection site swelling	0	0	0	1 (7.7)	0	0
Immune system disorders	0	0	0	0	0	1 (7.7)
Drug hypersensitivity	0	0	0	0	0	1 (7.7)
Decreased appetite	1 (12.5)	0	0	0	0	0
Dizziness	0	0	0	1 (7.7)	0	0
Headache	1 (12.5)	0	0	0	0	0
Insomnia	0	0	1 (12.5)	1 (7.7)	0	0
Papular rash	0	0	0	1 (7.7)	0	0

*Grade 1 = Mild, 2 = Moderate, 3 = Severe, 4 = Potentially Life-Threatening, 5 = Death; No Grade 4 or 5 AEs were reported in either Cohort.

CONCLUSIONS

- IM administration of CAB-LA or RPV-LA in adolescents achieved target exposure concentrations consistent with predictions and comparable to those observed in adults receiving monthly intra-muscular dose regimens.
- No new or unanticipated safety concerns were identified.

ACKNOWLEDGEMENTS

We acknowledge the following individuals for their contributions towards the study and abstract: Mark Marzinke, Elizabeth Lowenthal, Jared Kneebone, Jenny Huang, Michael Whitton, Sarah Buisson, and the site PIs. We would also like to acknowledge all participants, their families, the study staff at the many sites, and the study sponsors DAIDS, and our supporting partners ViiV and Janssen.

For more information, visit impaactnetwork.org and follow us:

Facebook: IMPAACTNetwork | Twitter: @IMPAACTNetwork | LinkedIn: IMPAACTNetwork

Overall support for the International Maternal Pediatric Adolescent AIDS Clinical Trials Network (IMPAACT) was provided by the National Institute of Allergy and Infectious Diseases (NIAID) with co-funding from the Eunice Kennedy Shriver National Institute of Child Health and Human Development (NICHD) and the National Institute of Mental Health (NIMH), all components of the National Institutes of Health (NIH), under Award Numbers UM1AI068632-15 (IMPAACT LOC), UM1AI068616-15 (IMPAACT SDMC) and UM1AI106716-09 (IMPAACT LC), and by NICHD contract number HHSN275201800001I. The content is solely the responsibility of the authors and does not necessarily represent the official views of the NIH.

Presented at the 2022 Conference on Retroviruses and Opportunistic Infections (CROI) 12-16 February 2022