00545

A LONG-ACTING TENOFOVIR PRODRUG SUPPRESSES HBV REPLICATION FOR OVER THREE MONTHS

Srijanee Das[1], Weimin Wang[1], Denise A. Cobb[1], Murali Ganesan[1], Franchesca G. Fonseca[2], Mohammad Ullah Nayan[1], Suyash Deodhar[1], Brandon Hanson[1], Edward Makarov[1], Howard E. Gendelman[1], Natalia A. Osna[1], Benson J. Edagwa[1] and Larisa Y. Poluektova[1]

[1]University of Nebraska Medical Center, Omaha, NE, United States, [2]University of Nebraska, Omaha, NE, United States

BACKGROUND

Tenofovir (TFV) prodrugs (TFV alafenamide, TAF and TFV disoproxil fumarate, TDF) are recommended for the treatment of chronic hepatitis B (HBV) in patients with HIV co-infection. However, TAF and TDF exhibit short half-lives and therefore require frequent administration. Consequently, this has resulted in treatment failures mainly due to patient non-adherence. To this end, we transformed TFV into a long-acting prodrug formulation (NM1TFV) and demonstrated sustained active diphosphate metabolite (TFV-DP) levels in key HIV and HBV target cells and tissues of Sprague Dawley rats for up to two months [Nat Commun 12,5458(2021)]. We now demonstrate that a single intramuscular dose of NM1TFV provides sustained efficacy in HBV-infected chimeric liver humanized mice and HBV transgenic Tg05 mice.

METHODS

A lipophilic TFV prodrug, M1TFV, was synthesized and nanoformulated into stable poloxamer 407 stabilized aqueous nanocrystals (NM1TFV) by high pressure homogenization. Solid drug nanocrystals of TAF (NTAF) were produced and used as controls. Formulation efficacy was evaluated in two mouse models (HBV-infected humanized liver TK-NOG mice and HBV transgenic Tg05 mice) following a single intramuscular injection of 168 mg/kg TFV equivalents of either NM1TFV or NTAF. HBV DNA levels in peripheral blood were assessed biweekly for 12 weeks. HBV markers HBcAg and HBsAg were evaluated on stained liver sections of TK-NOG mice. Drug levels were quantified by mass spectrometry.

RESULTS

HUMANIZED TK-NOG MICE

Liver chimeric ***TK-NOG mouse*** model has a reconstituted 'humanized liver,' engineered in highly immune-deficient mice by conditional depletion of mouse hepatocytes to permit repopulation by human hepatocytes. The rate of chimerism correlates with serum human albumin levels. (Hasegawa M,2011)

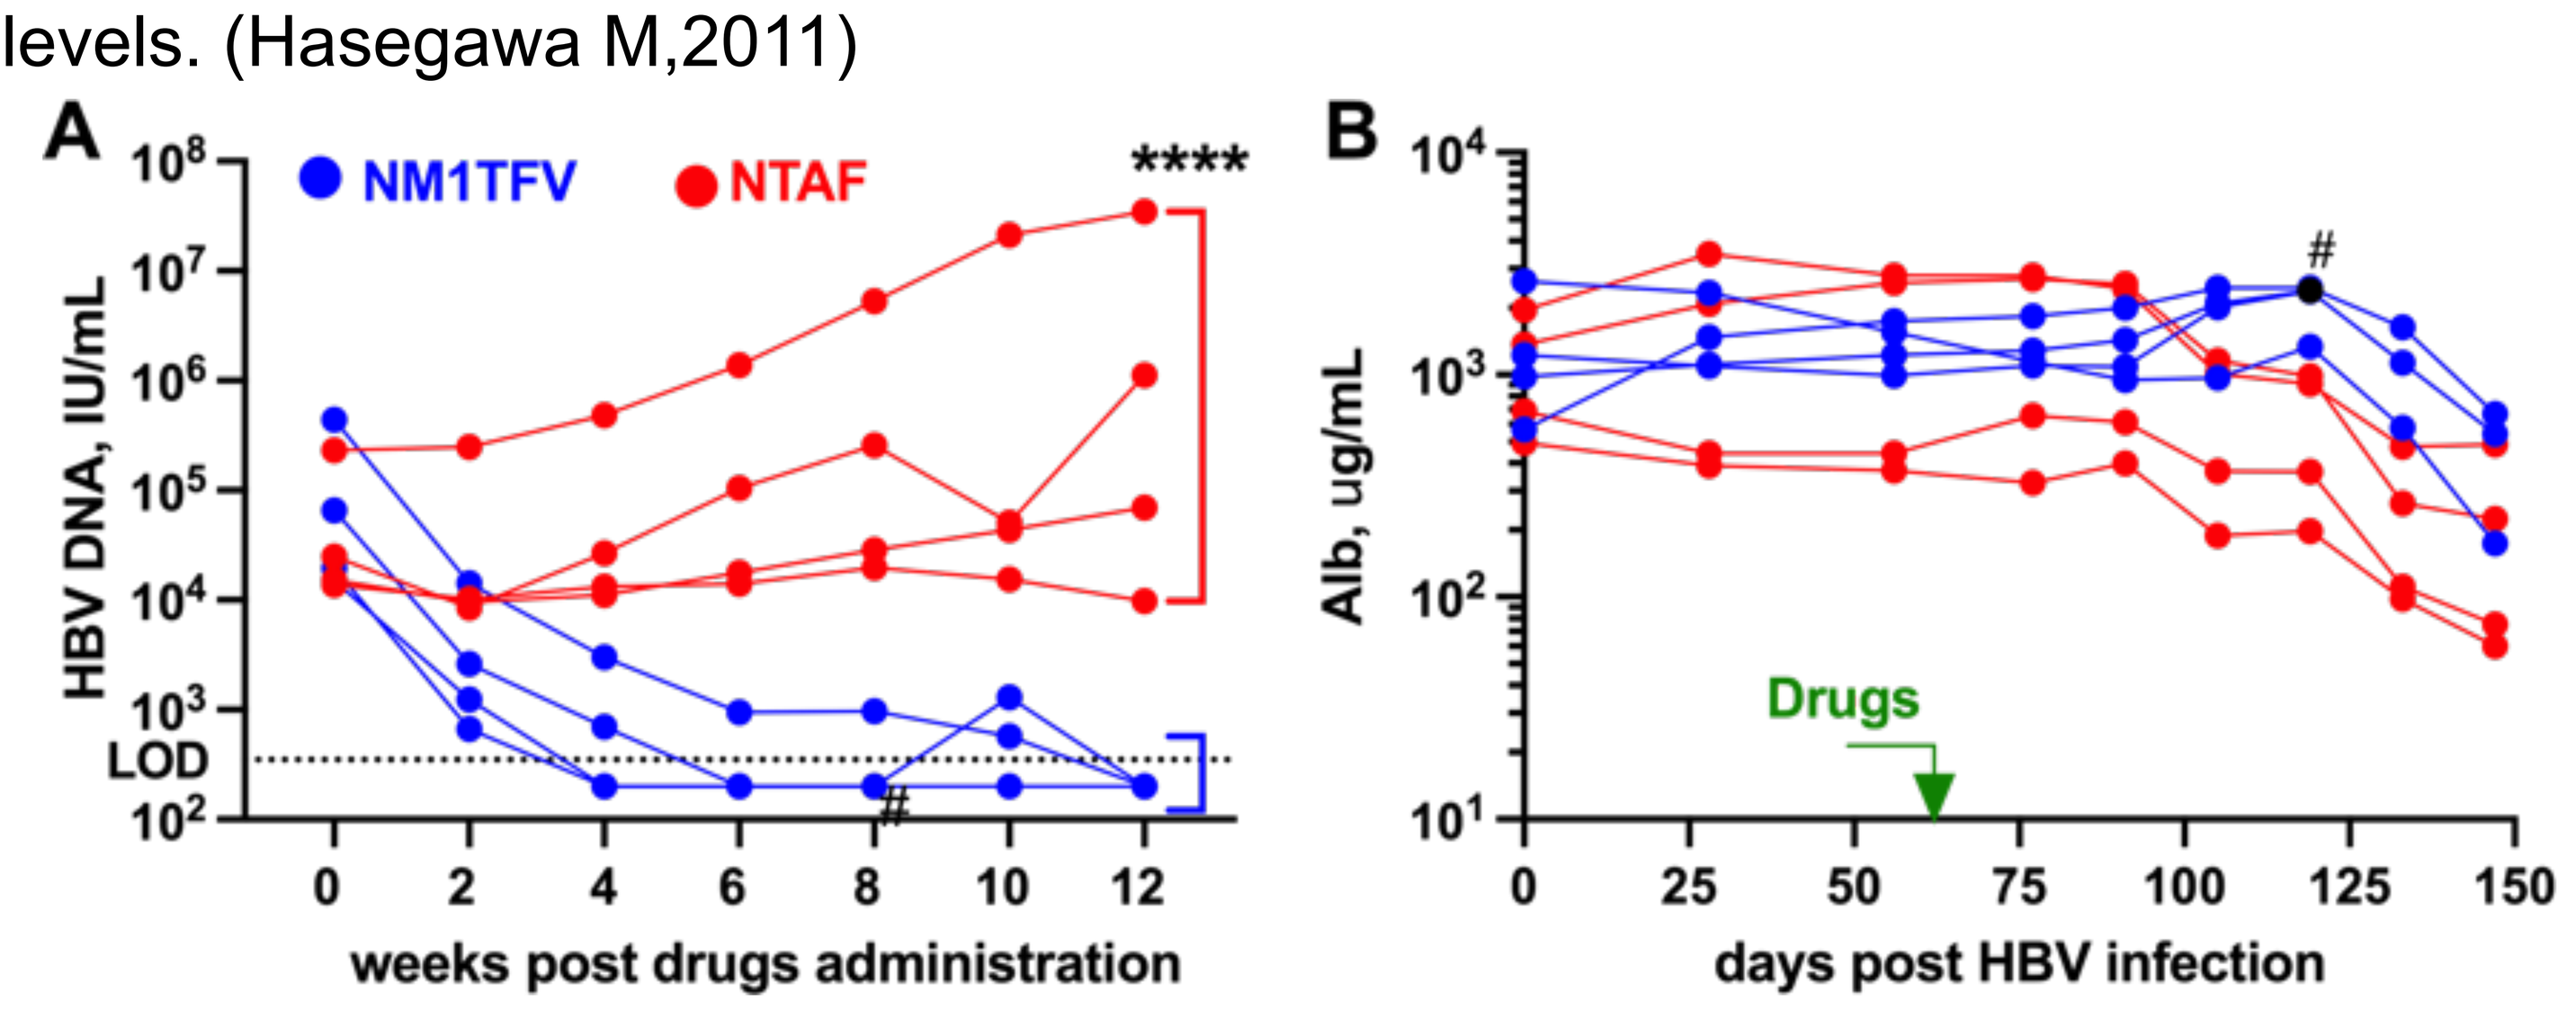
A
NM1TFV
NTAF

HBV DNA, IU/mL
LOD
weeks post drugs administration
B
Alb, ug/mL
Drugs
#
days post HBV infection

Figure 1. Suppression of HBV replication in humanized TK-NOG mice. (A) The dynamics of HBV DNA viral load in peripheral blood. NM1TFV suppressed viral replication to below the levels of detection (LOD, 350 UI/mL) over 3 months. **(B)** The levels of human albumin (hAlb) in peripheral blood. Control of HBV replication was not related to the partial loss of human hepatocytes but linked to treatment. NTAF was not able to control HBV replication. # - lost mouse but not related to treatment. **** - P<0.0001 by one-way ANOVA between effects of NM1TFV and NTAF.

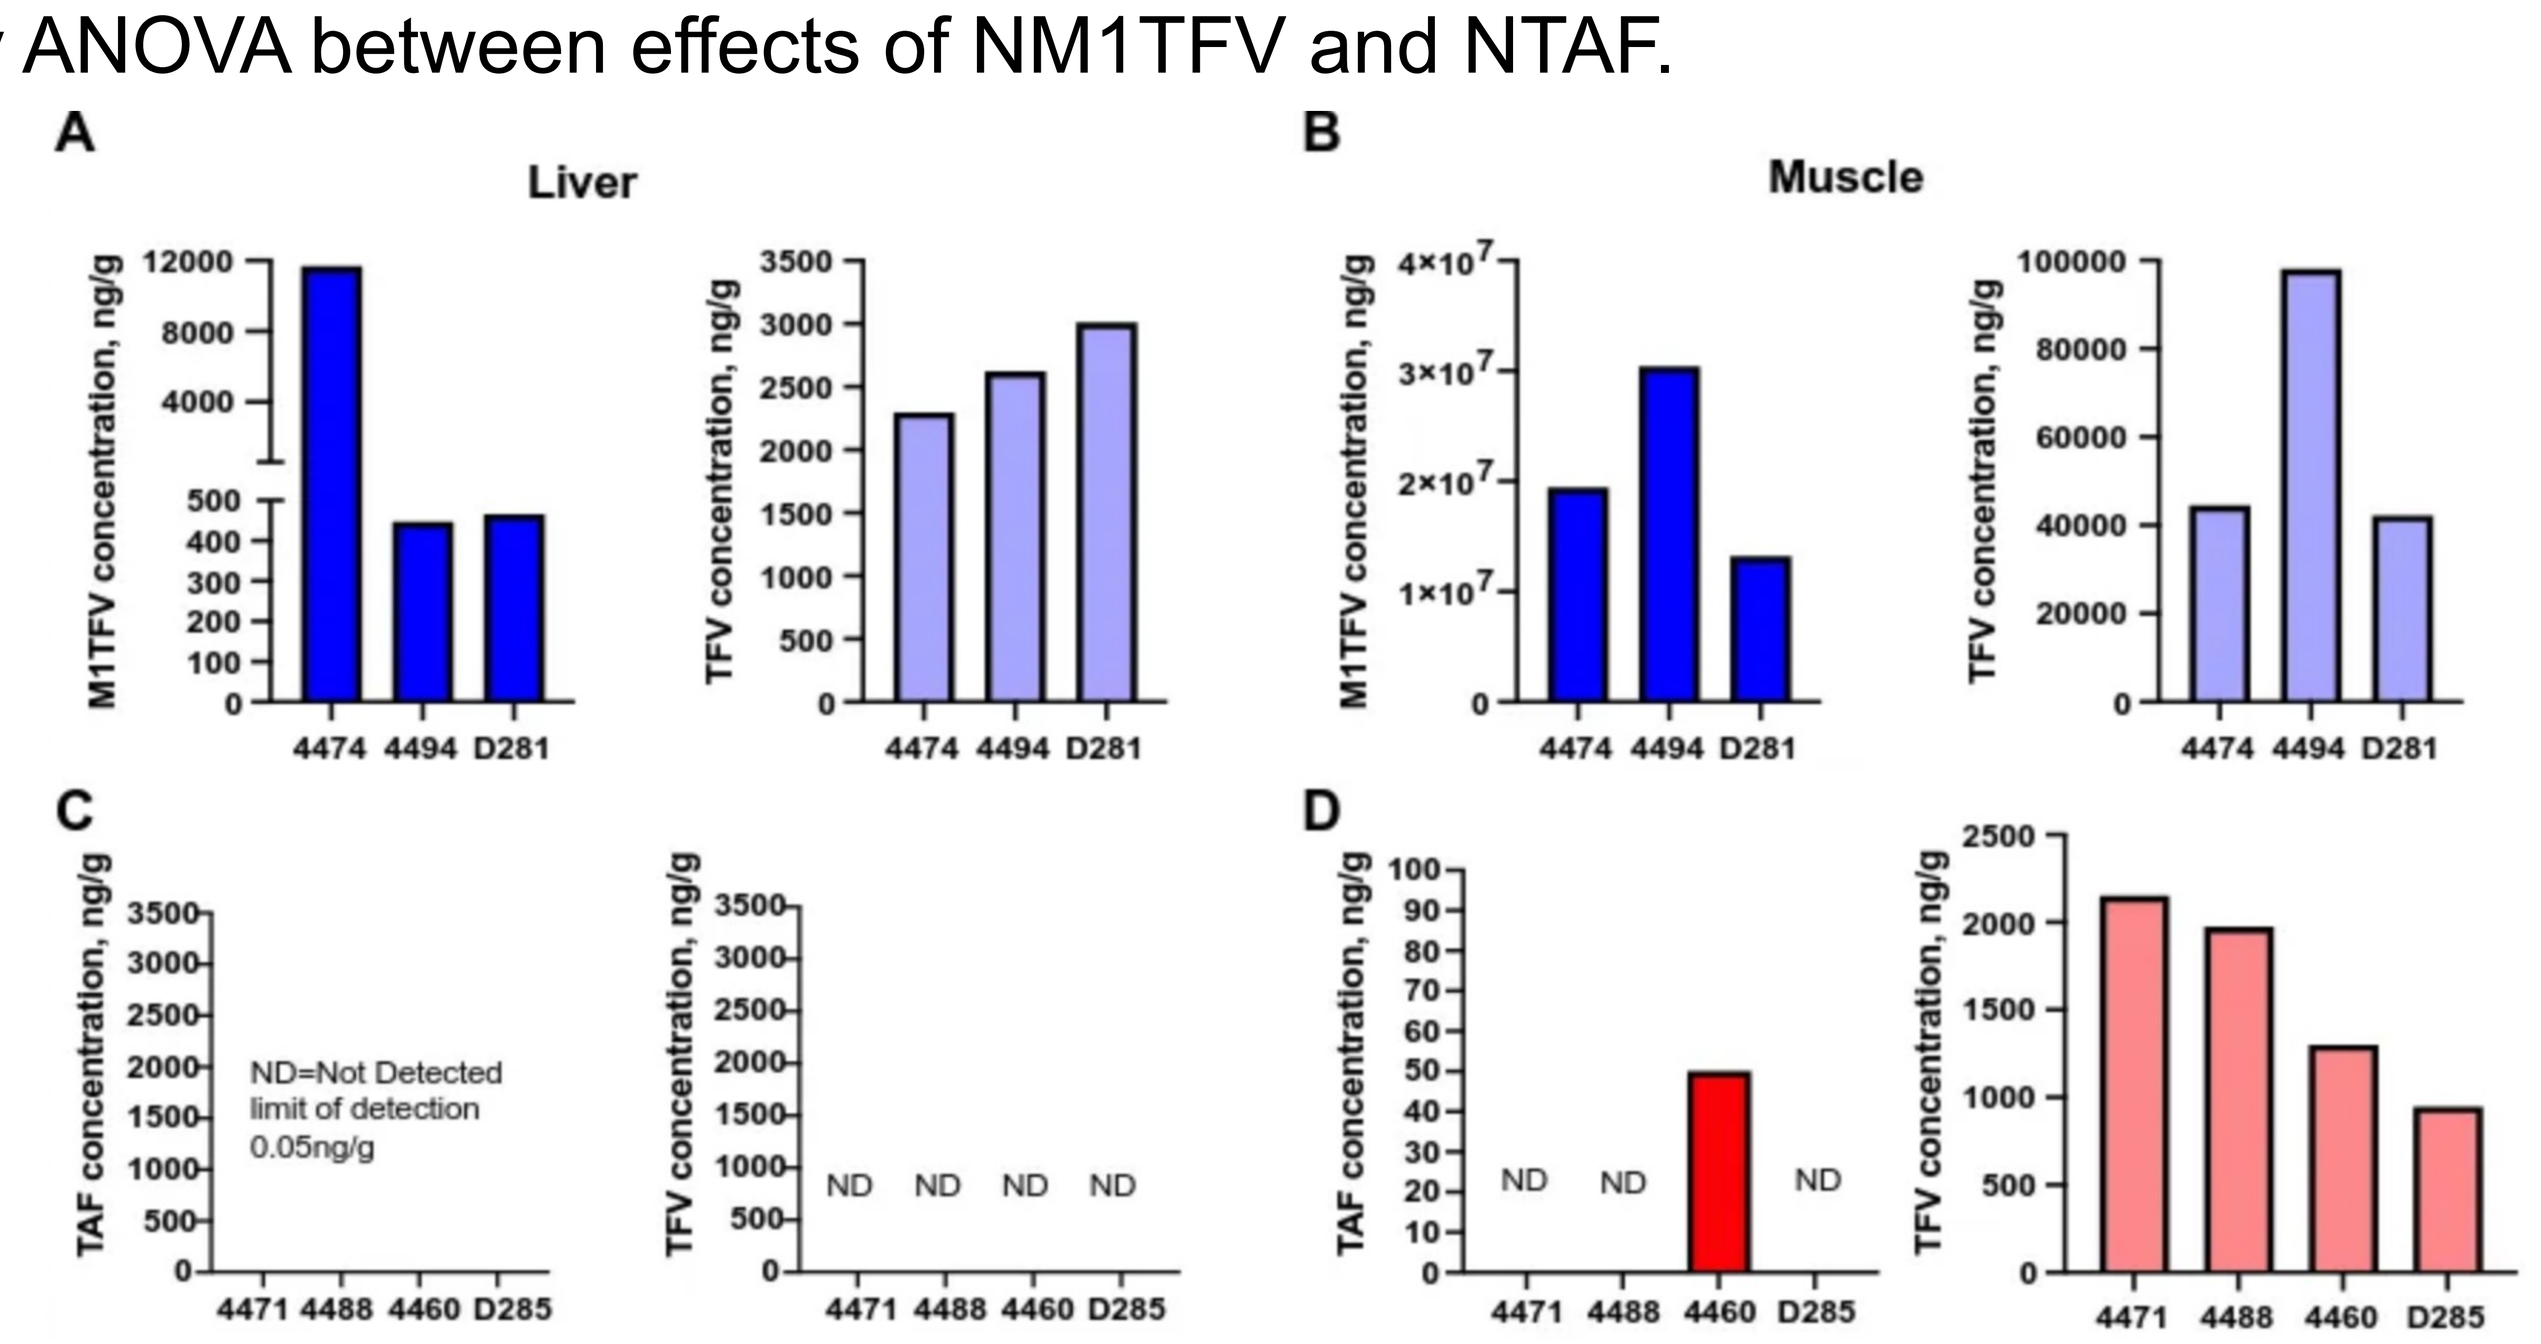
A
Liver
B
Muscle
M1TFV concentration, ng/g
TFV concentration, ng/g
4474 4494 D281
C
D
TAF concentration, ng/g
ND=Not Detected
limit of detection
0.05ng/g
ND ND ND ND
4471 4488 4460 D285

Figure 2. Tissue prodrug and TFV concentrations. (**A** and **B**) Significant amounts of M1TFV prodrug and TFV were quantified in the livers and injection sites of NM1TFV treated animals. (**C**) The amount of drug in the liver for TAF treated animals was below the limit of quantitation at 12 weeks. However, TAF treated animals exhibited detectable drug levels at the muscle injection site (**D**).

A nanoformulated TFV prodrug suppresses HBV DNA in humanized and transgenic mice for three months.

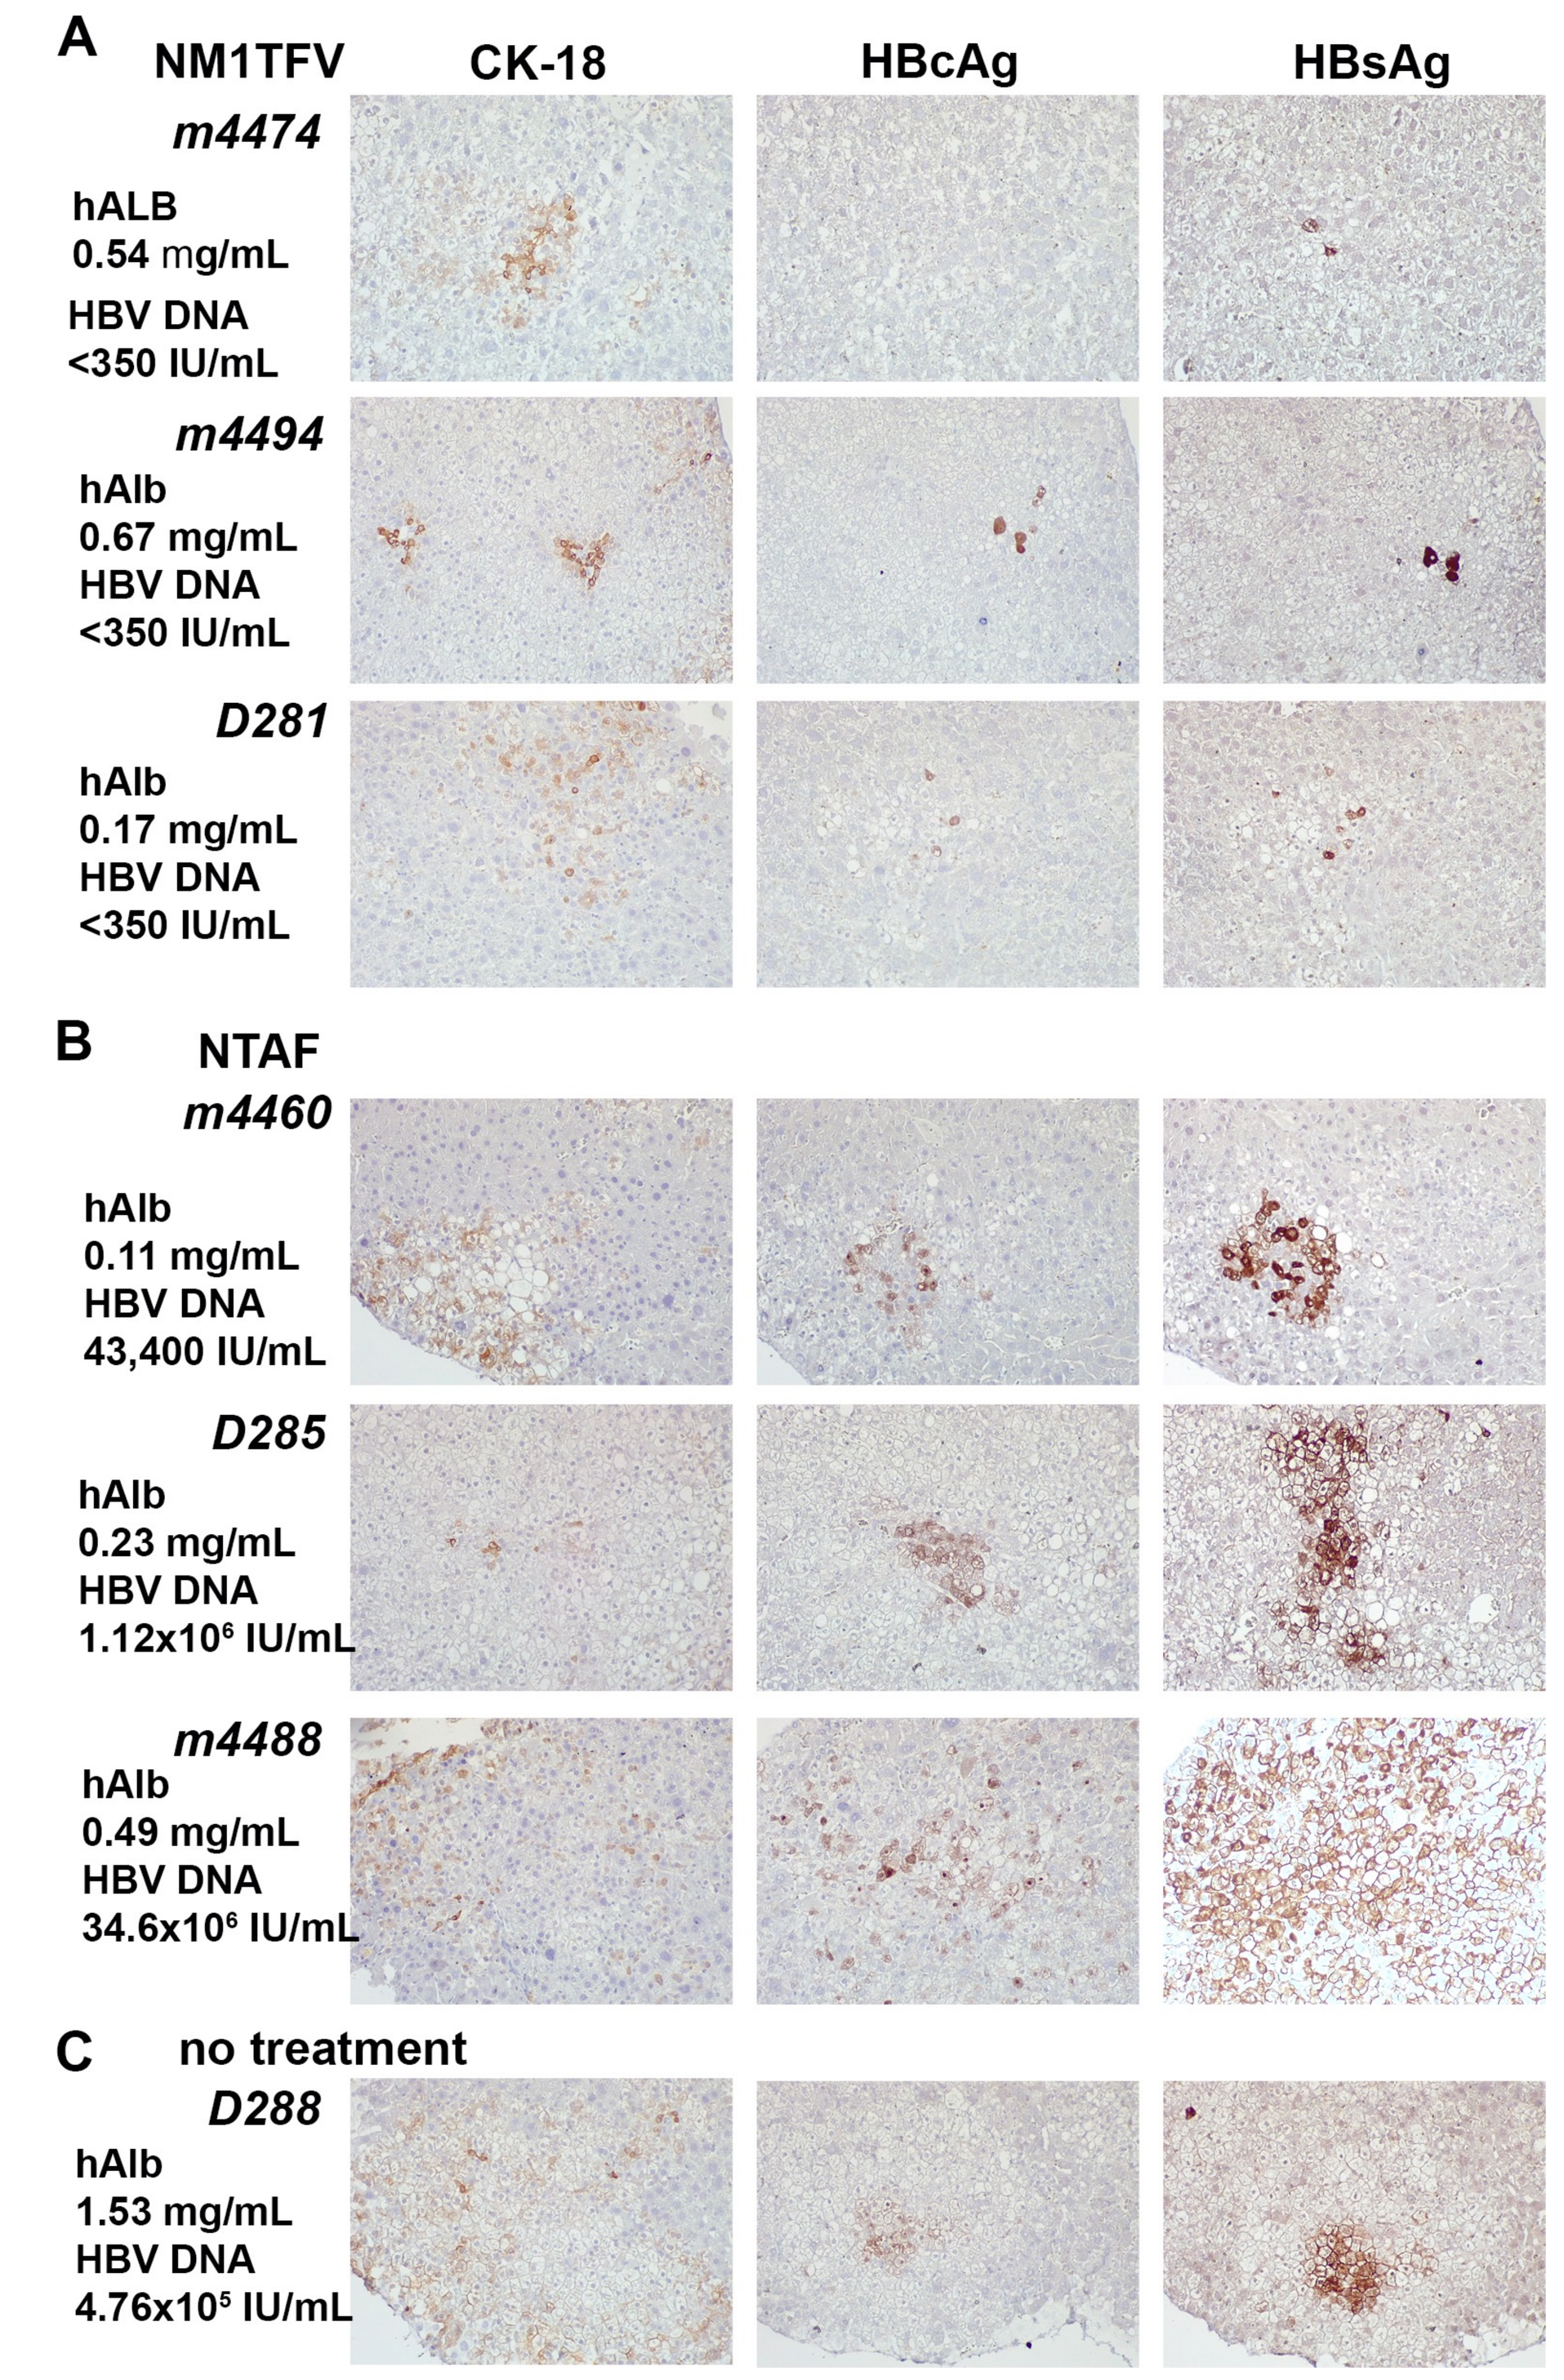
A NM1TFV
CK-18
HBcAg
HBsAg
m4474
hALB 0.54 mg/mL
HBV DNA <350 IU/mL
m4494
hAlb 0.67 mg/mL
HBV DNA <350 IU/mL
D281
hAlb 0.17 mg/mL
HBV DNA <350 IU/mL
B NTAF
m4460
hAlb 0.11 mg/mL
HBV DNA 43,400 IU/mL
D285
hAlb 0.23 mg/mL
HBV DNA 1.12x10⁶ IU/mL
m4488
hAlb 0.49 mg/mL
HBV DNA 34.6x10⁶ IU/mL
C no treatment
D288
hAlb 1.53 mg/mL
HBV DNA 4.76x10⁵ IU/mL

Figure 3. Immunohistological evaluation of chimeric liver samples from NM1TFV, NTAF and control mice at three months post drug administration. The liver tissue samples were fixed, embedded in paraffin and 5 microns serial sections were stained for human cytokeratin-18 (CK-18) to identify areas of human hepatocytes, HBcAg and HBsAg to confirm the presence of ongoing viral replication. The positive signal was visualized with DAB (brown). Sections were counterstained with hematoxylin. The levels of human albumin and HBV DNA are shown in left column for each mouse. **(A)** Liver tissue samples for NM1TFV treated mice exhibited limited numbers of viral protein positive cells and did not have detectable levels of virus in the blood (<350 UI/mL). **(B)** NTAF treated mice exhibited significant numbers of viral protein positive cells that corresponded to the peripheral viral load shown for each mouse. **(C)** Untreated animal contained readily detectable infected cells. All images were captured under original magnification 100×.

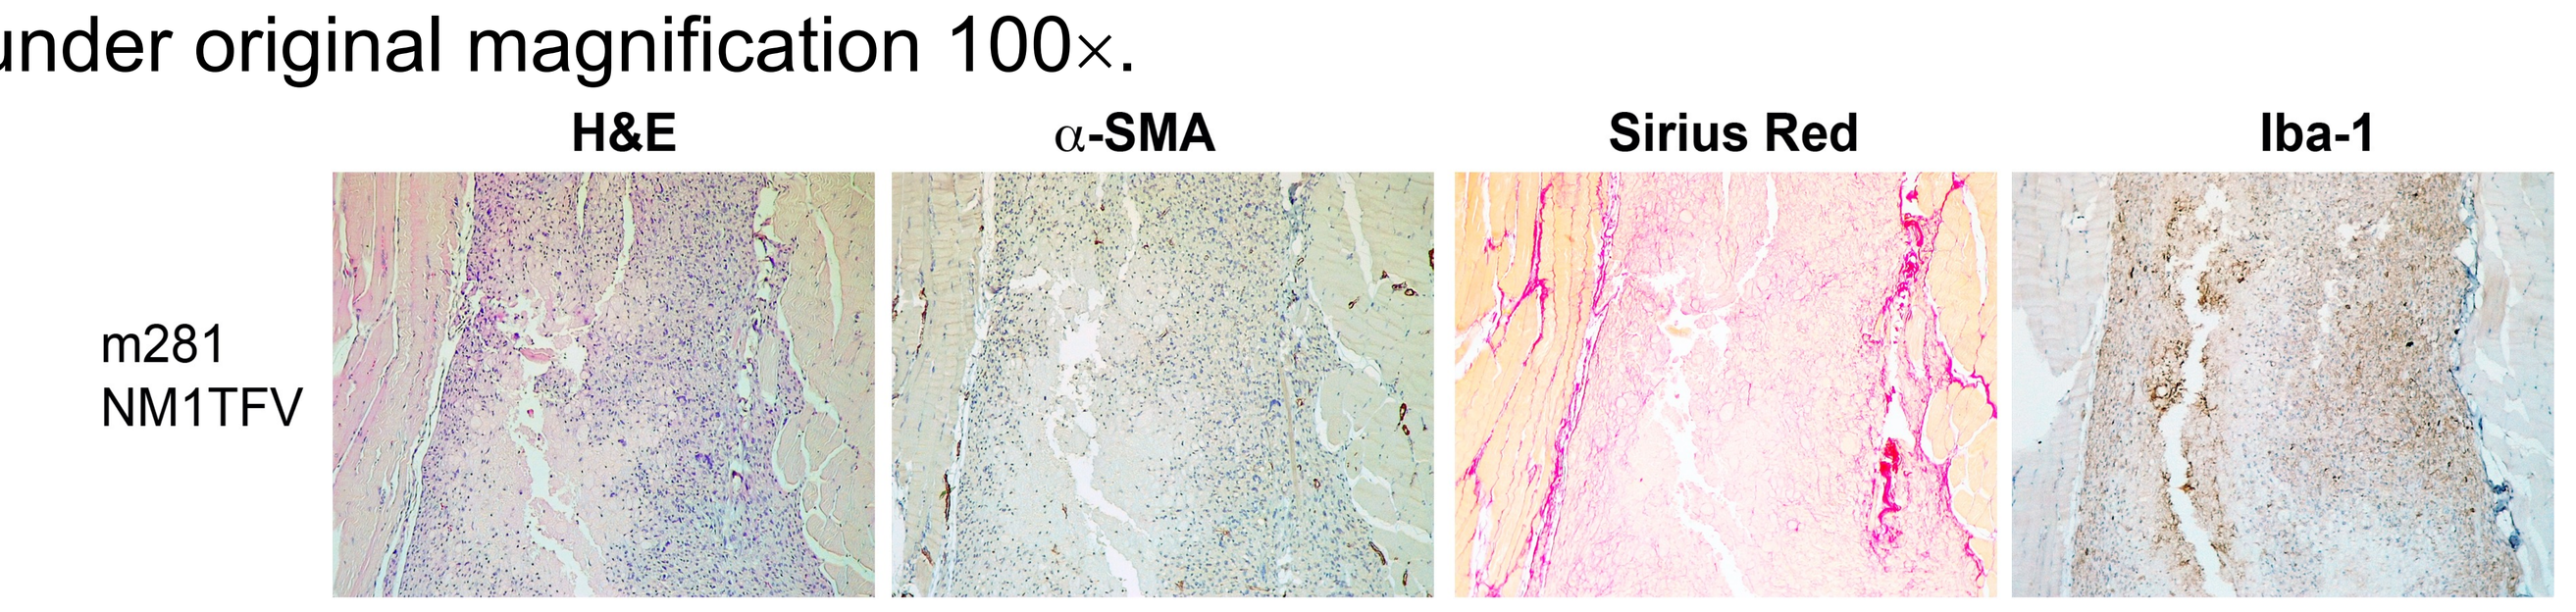
H&E
α-SMA
Sirius Red
Iba-1
m281
NM1TFV

Figure 4. Injection site evaluation following intramuscular administration of NM1TFV nanocrystals. A primary injection site drug depot was sustained for NM1TFV but not for NTAF. Injection site samples were stained for presence of fibrotic demarcation of foreign material (Sirius Red) and infiltrating macrophages (Iba-1). There was no evidence of muscle degeneration, basophils or eosinophils, cellulitis, or abscesses. Formation of small vessels was detected by alpha-smooth muscle actin (a-SMA) staining. All images were captured under original magnification 100×.

HBV TRANSGENIC Tg05 MICE

Transgenic Tg05 mice express the 1.3 genome length transgene of HBV wild-type viral genome and show very high HBV replication and gene expression levels in the liver and kidney tissues, producing circulating virions in the blood. The estimated 10^7 to 10^8 viral genomes detectable per ml of serum in these animals compares favorably with the virus concentration present in the sera of chronically infected humans. (Guidotti LG, 1995)

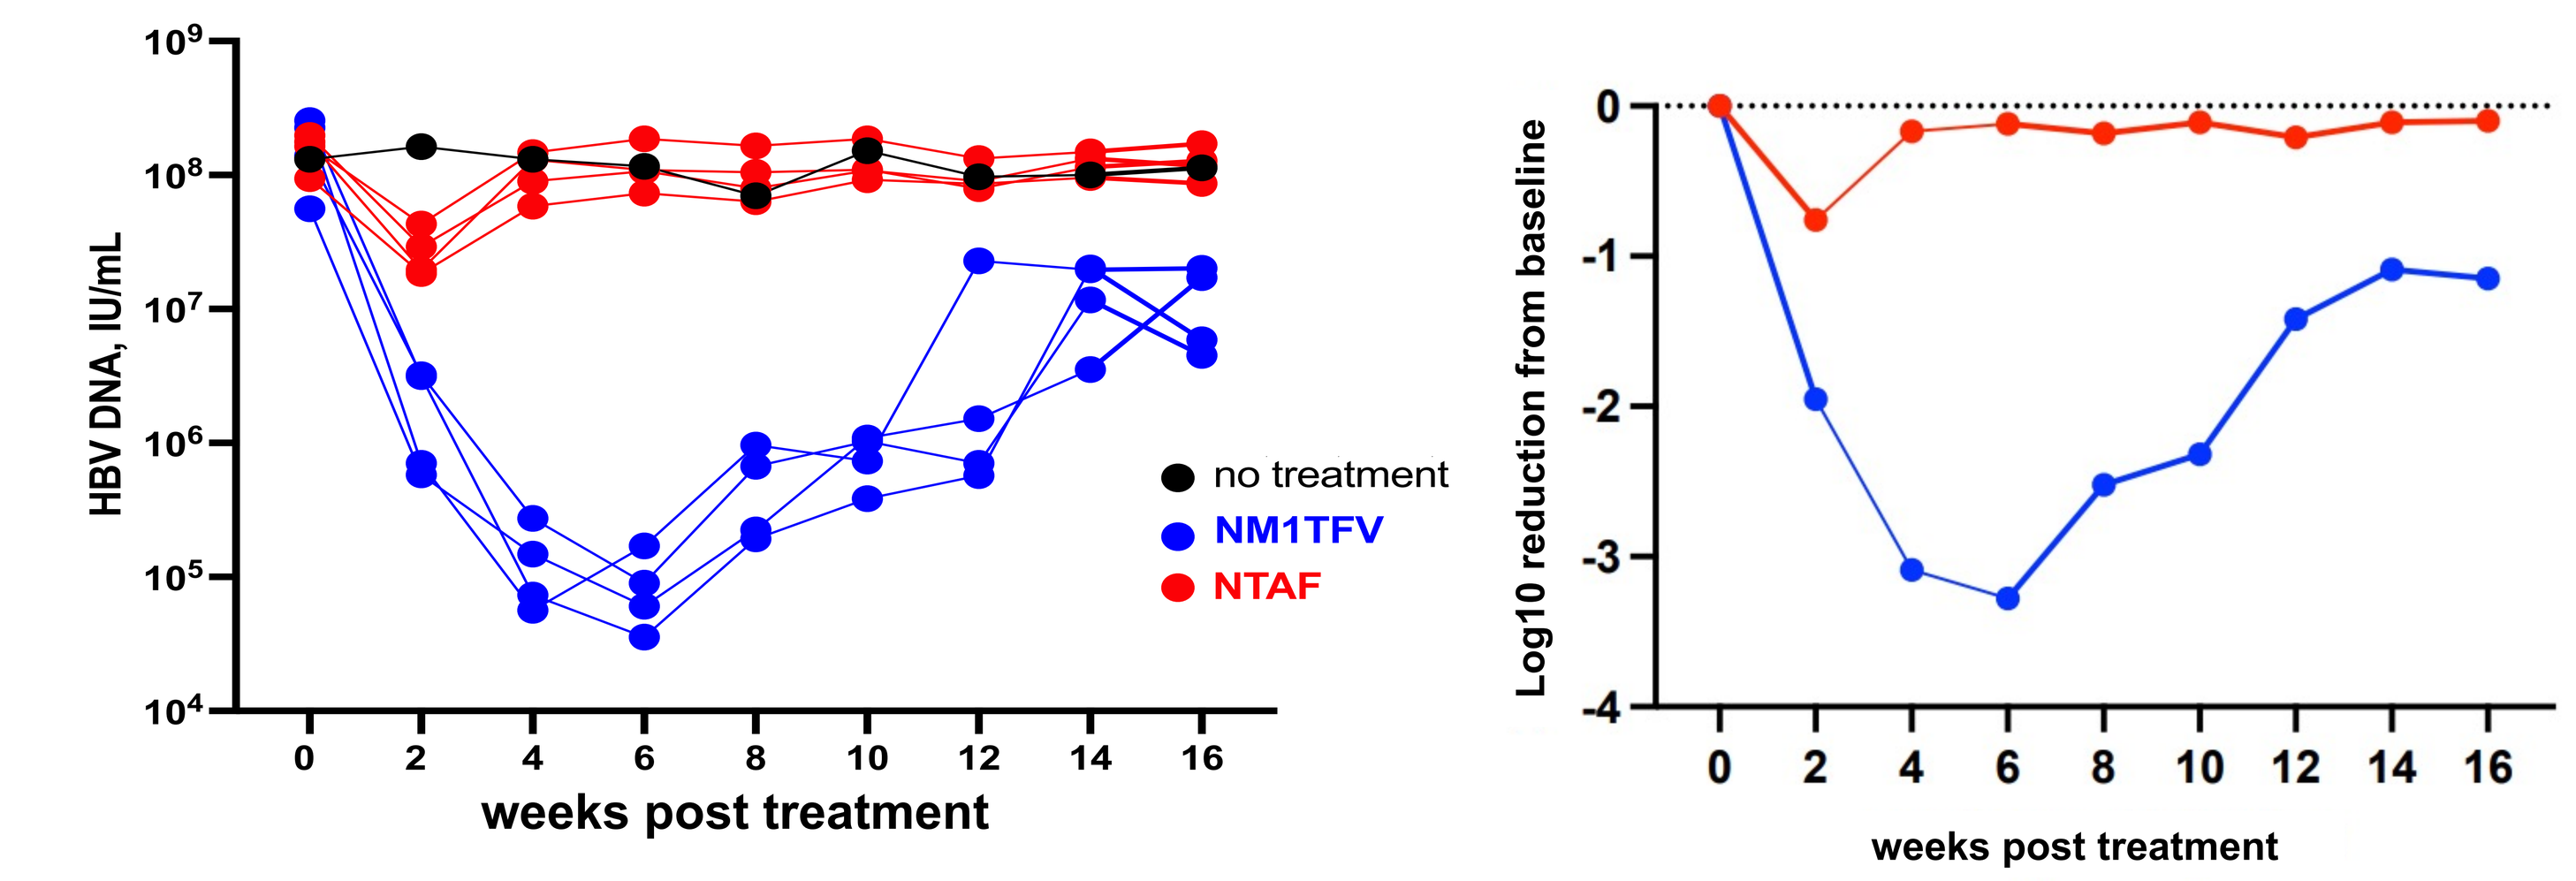
HBV DNA, IU/mL
no treatment
NM1TFV
NTAF
weeks post treatment
Log10 reduction from baseline
weeks post treatment

Figure 5. Suppression of HBV replication in HBV transgenic Tg05 mice. The dynamics of HBV DNA viral load in peripheral blood. NM1TFV suppressed viral replication from baseline levels by several log10 over the 16 weeks experiment duration. By contrast, NTAF exhibited minimal effect.

Table 1. Efficacy in HBV transgenic mice

Treatment / weeks	log10 reduction in serum HBV DNA levels from baseline							
	2	4	6	8	10	12	14	16
NM1TFV	-1.95	-3.09	-3.28	-2.52	-2.32	-1.42	-1.09	-1.15
NTAF	-0.76	-0.17	-0.12	-0.18	-0.11	-0.21	-0.11	-0.10

CONCLUSIONS

- A single intramuscular injection of NM1TFV to HBV infected humanized TK-NOG mice suppress viral DNA in peripheral blood and liver for three months.
- Levels of HBcAg and HBsAg were suppressed over three months in NM1TFV treated mice.
- NM1TFV formulations were well tolerated. No evidence of muscle degeneration, basophil or eosinophil infiltration, cellulitis or abscesses was observed at the injection site.
- NM1TFV not NTAF demonstrated sustained HBV DNA suppression in transgenic Tg05 mice.
- We posit that NM1TFV can be developed as a therapy for chronic HBV infection.

FUNDING SOURCES

This research was supported by the University of Nebraska Foundation, which includes donations from the Carol Swarts, M.D. Emerging Neuroscience Research Laboratory, the Margaret R. Larson Professorship, the Frances and Louie Blumkin Endowment, and the Harriet Singer Endowment; the Vice Chancellor's Office of the University of Nebraska Medical Center for Core Facilities; Nickolus Badami Fellowship and the National Institutes of Health grants R01AI145542-01A1, R01AI158160-01A1, 1R56 AI138613-01A1, P01 DA028555, and P30 MH062261 (to B.E. and L.P.)