00518

IMPACT OF NASH ON THE SURVIVAL OF PEOPLE LIVING WITH HIV

Juan Macias[1,4], Antonio Rivero-Juarez[2,4], Alejandro Gonzalez-Serna[1,4], Mario Frías[2,4], Marta Santos[1,4], Ignacio Perez-Valero[2,4], Anaïs Corma-Gomez[1,4], Antonio Rivero[2,4], Miguel Garcia-Deltoro[3], Luis M Real[1,4], Juan A. Pineda[1,4]; GEHEP group from the Sociedad Española de Enfermedades Infecciosas y Microbiología (SEIMC).

1 Hospital Universitario Virgen de Valme, Seville, 2 Hospital Universitario Reina Sofia, Cordoba, 3 Hospital General de Valencia, Valencia, Spain. 4 CIBERINFEC

BACKGROUND

Non-alcoholic fatty liver disease (NAFLD) is an increasing concern for PLWH. However, information on the impact of NAFLD on the prognosis of PLWH is lacking. Because of these, we investigated the influence of non-alcoholic steatohepatitis (NASH) with fibrosis on the overall mortality in PLWH.

METHODS

- PLWH followed in three Spanish centers were included in a prospective cohort at the date when a vibration-controlled transient elastography (VCTE) evaluation, including controlled attenuation parameter (CAP) measurement, was conducted for the first time.
- Clinical visits were scheduled, at least, every 6 months.
- Survival data was recorded, and the causes of death were centrally monitored.
- The risk of all-cause of death were evaluated applying time-to-event analyses.
- NAFLD was defined as steatosis (CAP ≥248 dB/m) without any other liver disease.
- Cirrhosis was defined as liver stiffness measurement (LSM) ≥10.3 kPa for NAFLD (Wong. Hepatology 2010).
- The FibroScan-AST (FAST) score (Newsome. Lancet Gastroenterol Hepatol 2020), which includes AST, CAP and LSM was calculated. The FAST score is aimed to identify HIV-negative people with NASH and significant fibrosis.
- PLWH without other concomitant causes of liver disease (HCV exposure, HBV active infection, or alcohol intake ≥50 g/day) were selected for the elaboration of predictive models

Among PLWH without concomitant causes of liver disease other than NAFLD, liver stiffness predicts survival similarly to the FAST score

CONCLUSIONS

- Among PLWH, the presence of NAFLD, as a whole, is not associated with survival.
- However, for PLWH without concomitant liver diseases other than NAFLD, liver fibrosis increases the risk of death.
- Liver stiffness and the FAST score are similar predictors of survival in this setting.

RESULTS

Baseline characteristics (n=1810)

Characteristic	
Age, years*	47 (42-52)
Male sex, n (%)	1436 (79)
PWID, n (%)	1047 (58)
AIDS, n (%)	614 (34)
Nadir CD4, cells/mL*	191 (50-370)
CD4 cell counts, cells/mL*	517 (320-734)
HIV RNA ≤50 copies/mL, n (%)	1642 (91)
Anti-VHC (+), n (%)	1082 (60)
Fasting plasma glucose, mg/dL*	93 (85-102)
Cholesterol, mg/dL*	176 (150-202)
HDL cholesterol, mg/dL*	44 (36-56)
Triglycerides, mg/dL*	120 (85-171)
Liver stiffness, kPa*	6.2 (4.8-9.5)
CAP, dB/m*	228 (194-269)

*Median (Q1-Q3)

Causes of death

Deaths: 134 (7.4%).
Median follow-up time: 74 (23-102) months.
Losses to follow-up: 163 (9%).

Causa de muerte	N (%)
AIDS	15 (11)
Liver disease	36 (27)
Cancer	38 (28)
Cardiovascular disease	12 (9)
Other causes*	33 (25)

* Non-AIDS infections: 10 (7.5%); Overdose: 8 (6%); Chronic lung disease: 7 (5.2%); Accidental: 1 (0,7%); Unknown cause: 7 (5.2%)

Survival according to steatosis grade PLWH without other concomitant causes of liver disease (n=687)

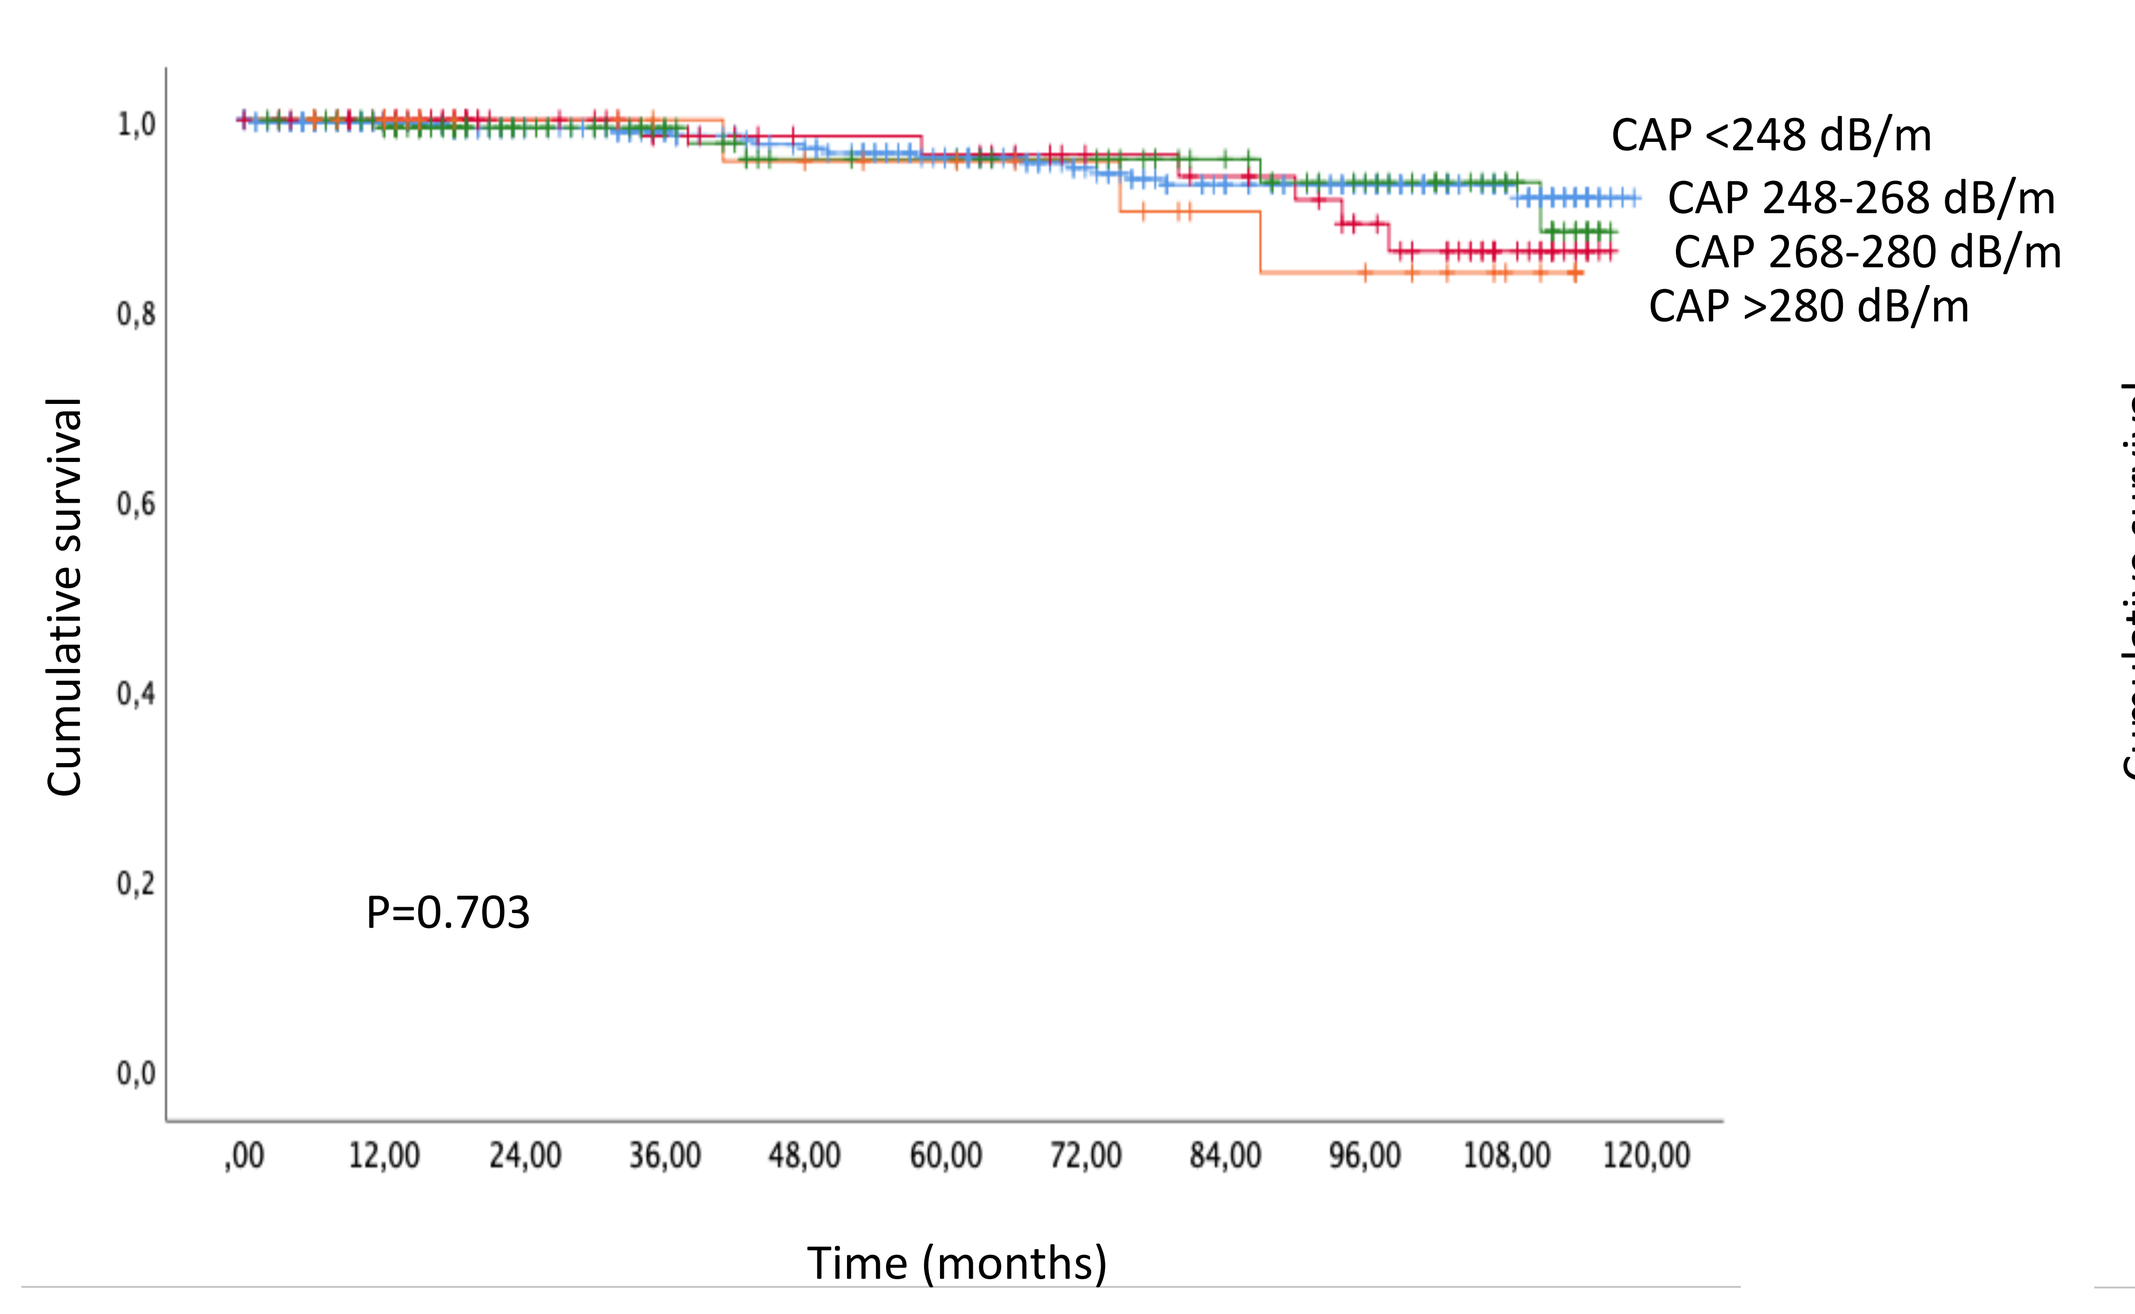
CAP <248 dB/m
CAP 248-268 dB/m
CAP 268-280 dB/m
CAP >280 dB/m
P=0.703
Cumulative survival
Time (months)

Survival according to liver stiffness PLWH without other concomitant causes of liver disease (n=687)

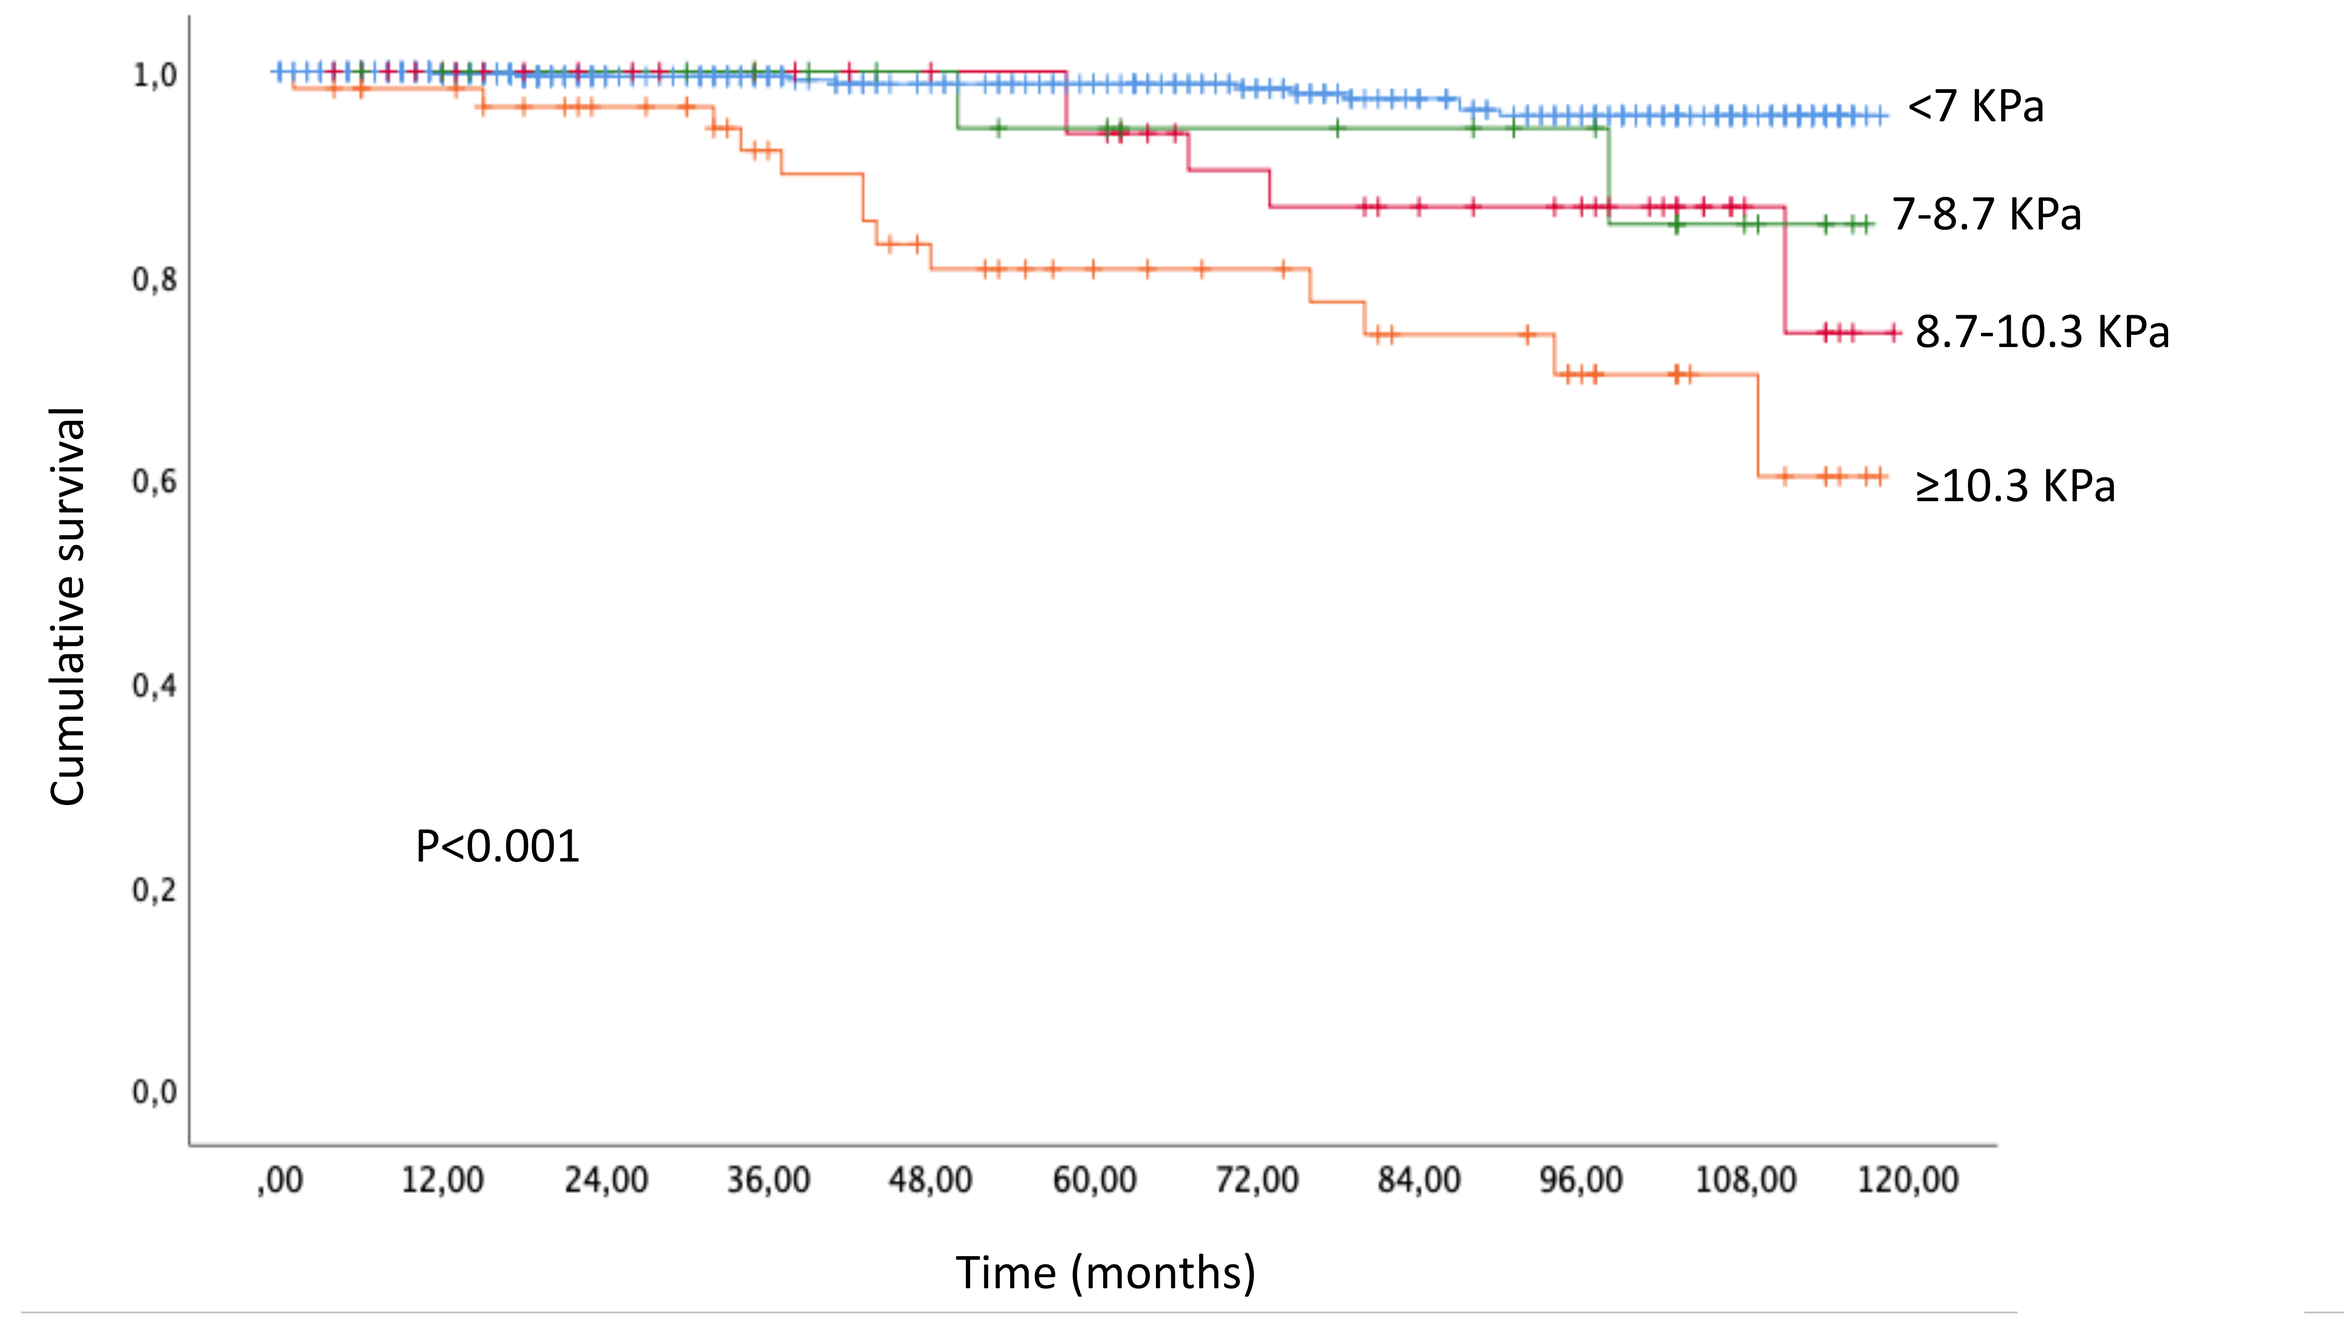
<7 KPa
7-8.7 KPa
8.7-10.3 KPa
≥10.3 KPa
P<0.001
Cumulative survival
Time (months)

Survival according to FAST score PLWH without other concomitant causes of liver disease (n=687)

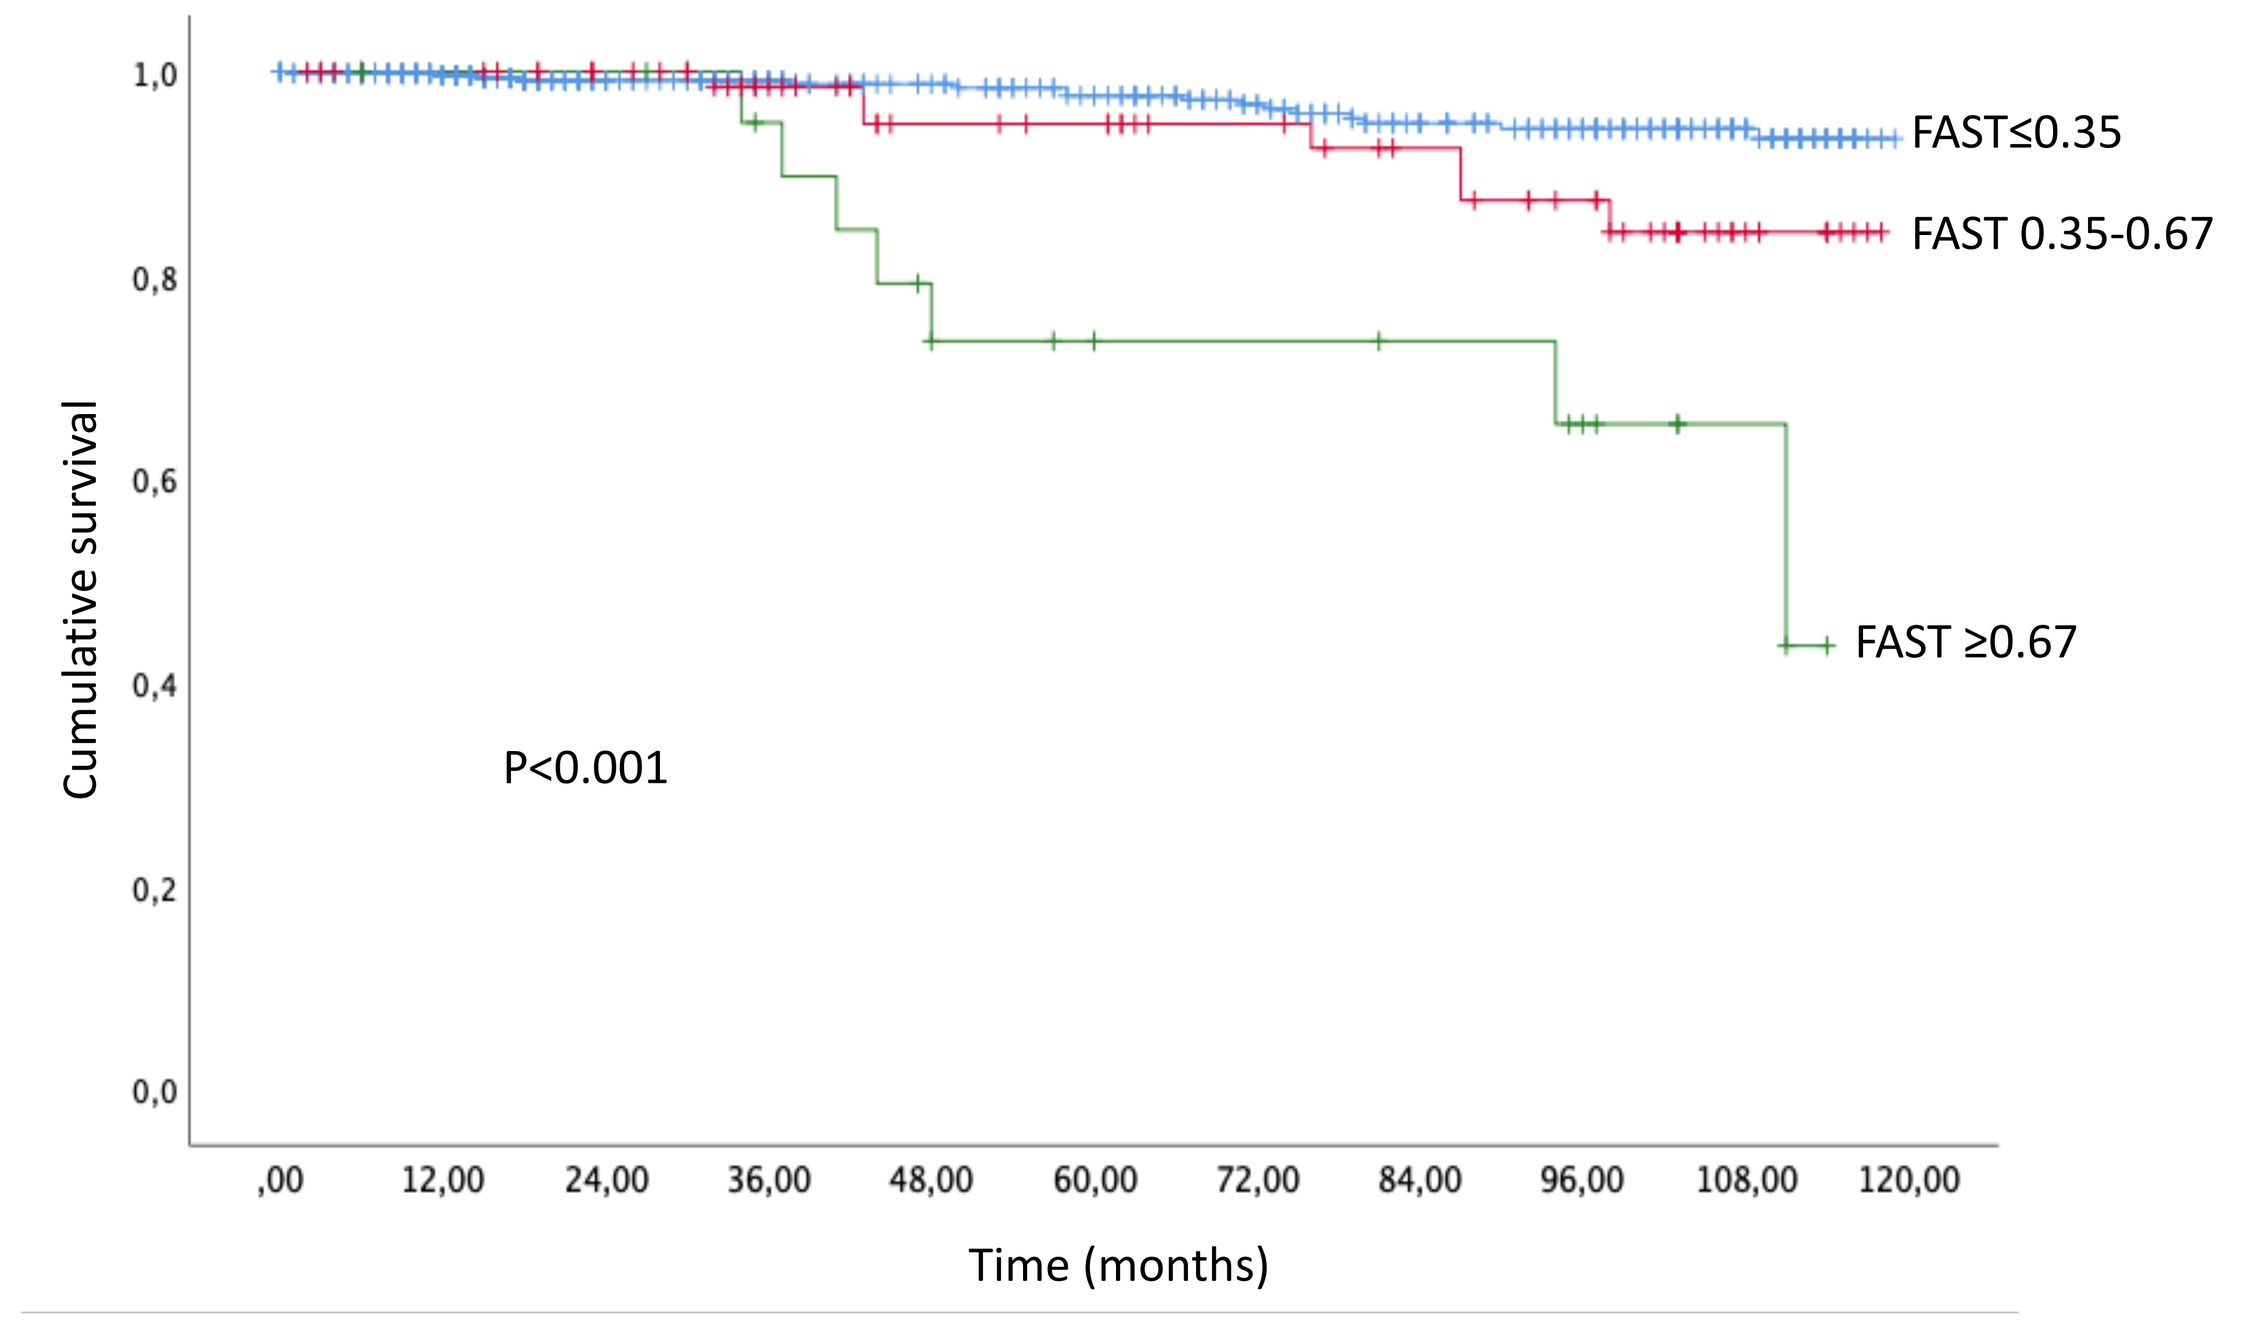
FAST≤0.35
FAST 0.35-0.67
FAST ≥0.67
P<0.001
Cumulative survival
Time (months)

Survival according to steatosis grade (n=18010)

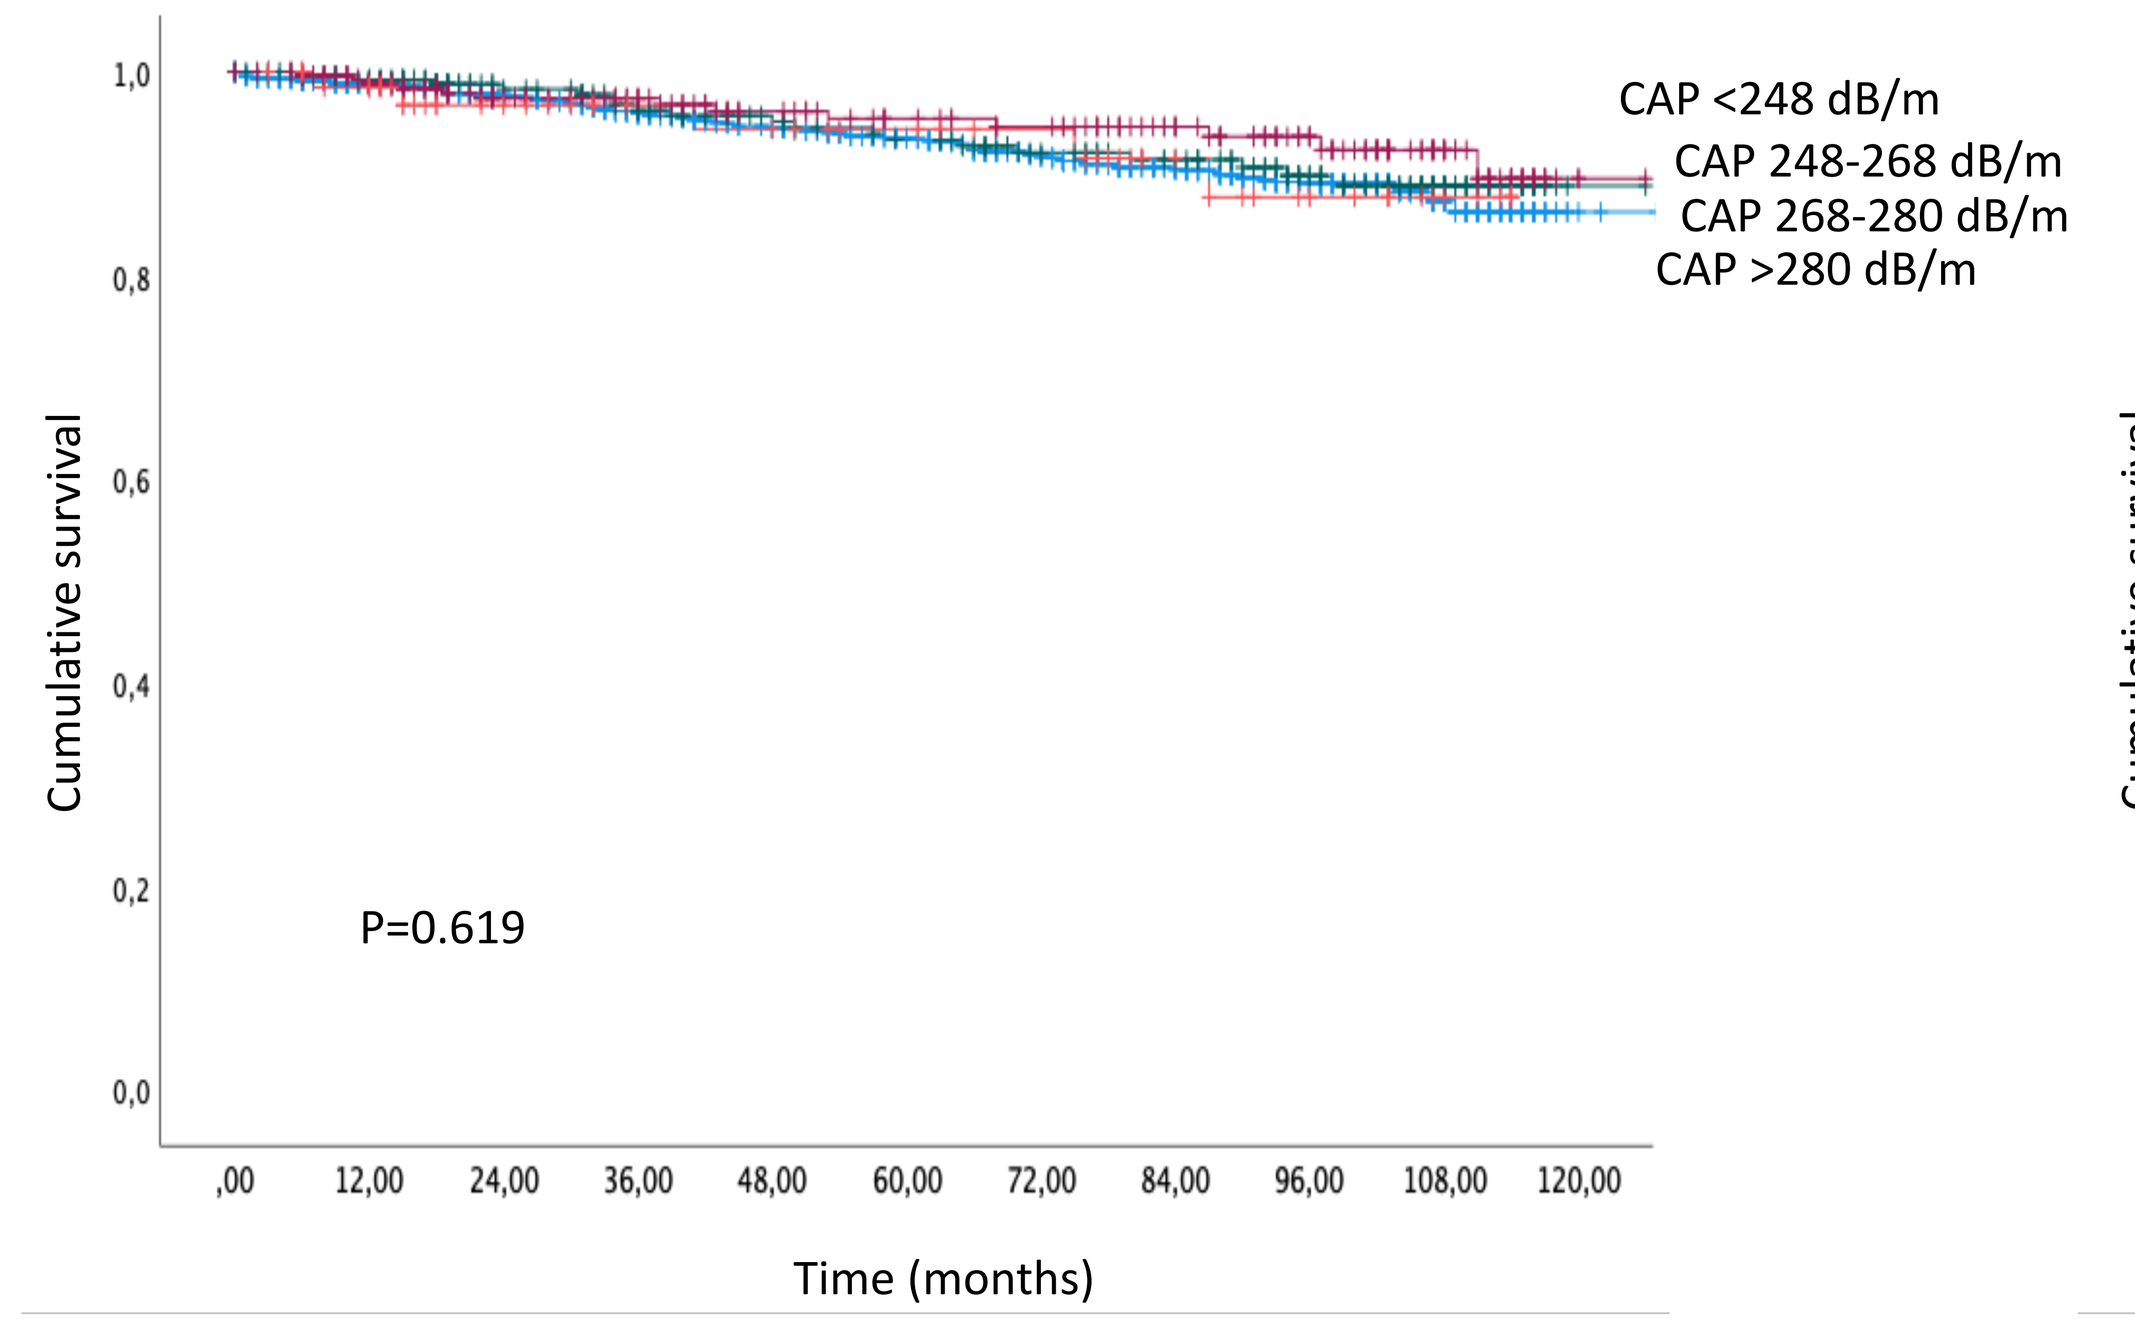
CAP <248 dB/m
CAP 248-268 dB/m
CAP 268-280 dB/m
CAP >280 dB/m
P=0.619
Cumulative survival
Time (months)

Survival according to liver stiffness (n=1810)

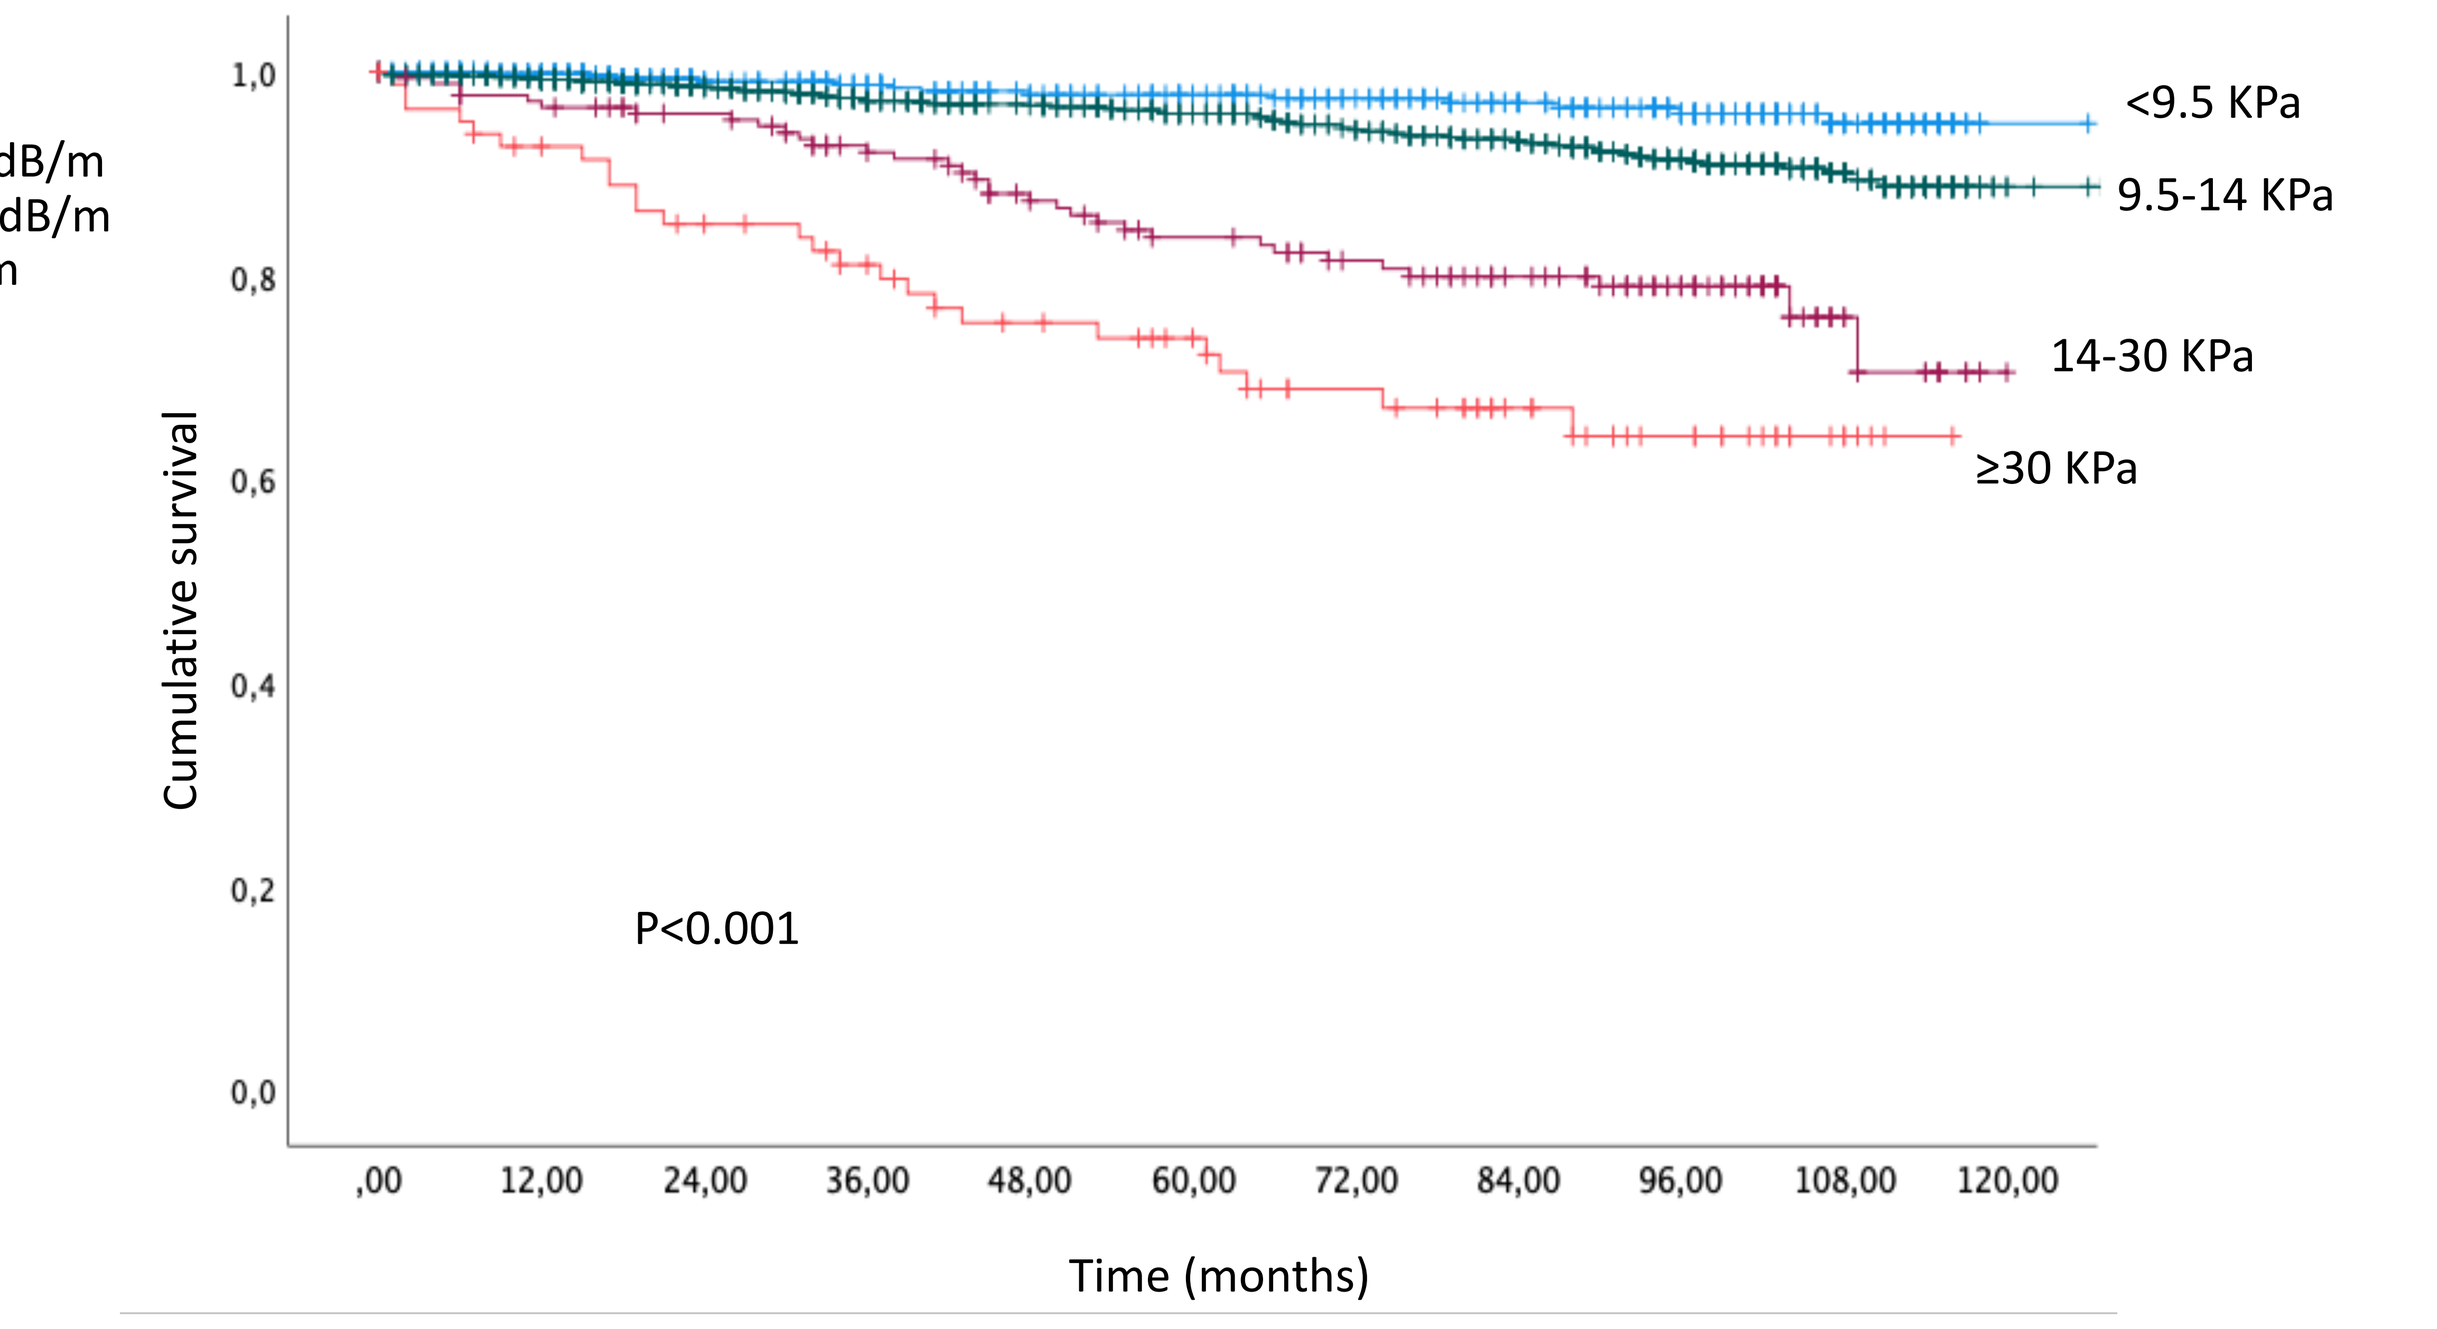
<9.5 KPa
9.5-14 KPa
14-30 KPa
≥30 KPa
P<0.001
Cumulative survival
Time (months)

Survival according to FAST score (n=1325)

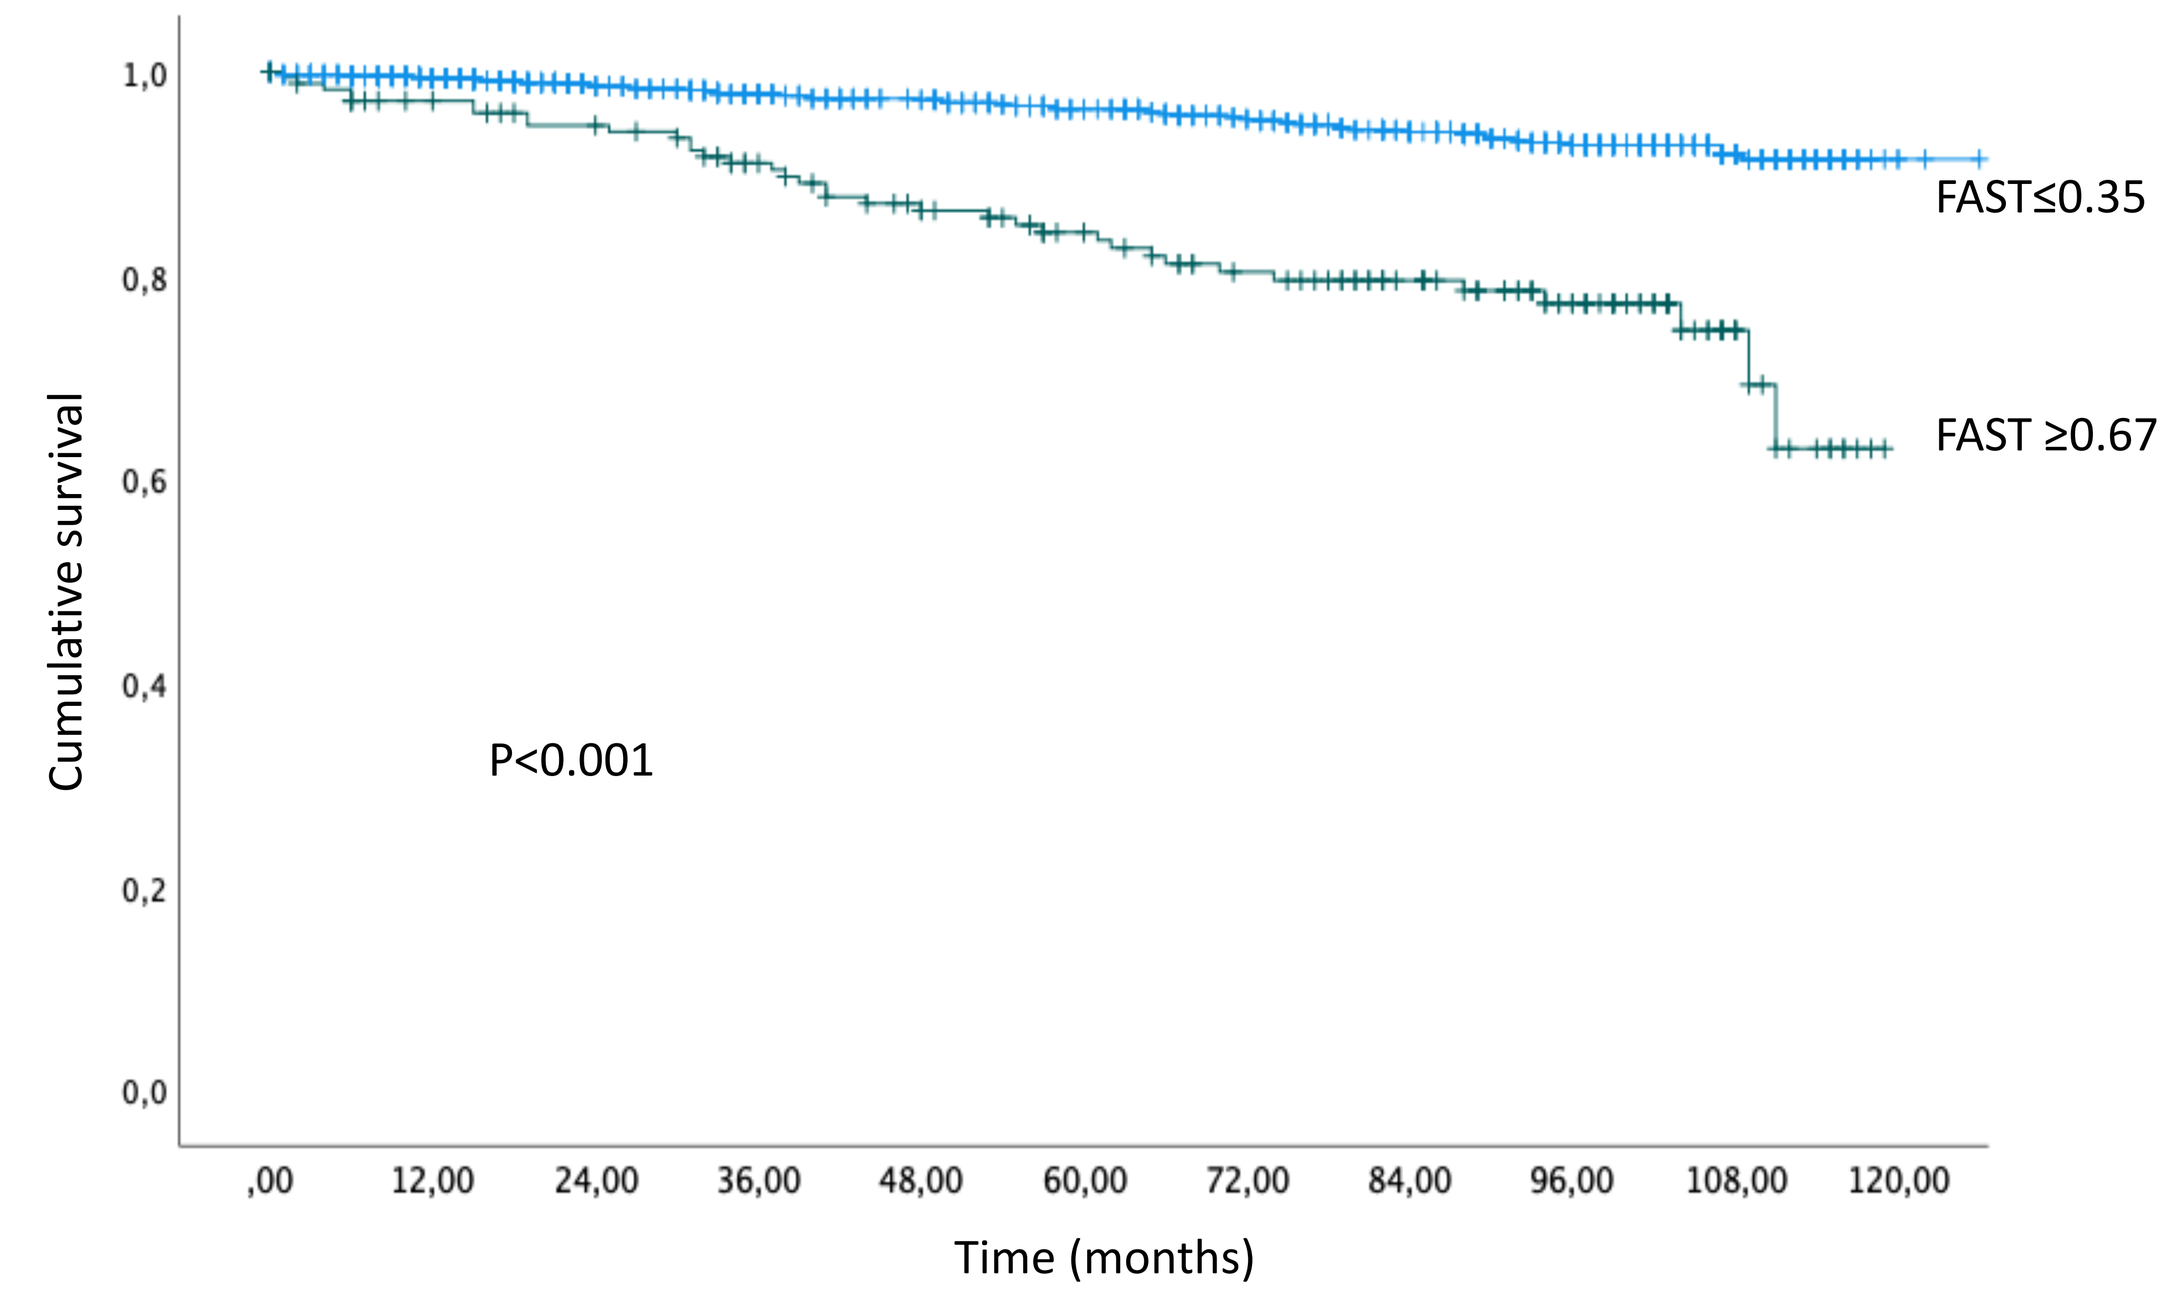
FAST≤0.35
FAST ≥0.67
P<0.001
Cumulative survival
Time (months)

Risk factors for overall mortality among PLWH without other concomitant causes of liver disease. Model including liver stiffness

Variables at baseline	Crude HR	95% CI HR		P	Adjusted HR	95% CI HR		P
Male sex	1,1	0,44	2,6	0,869	0,94	0,36	2,46	0,901
Age, years	1,07	1,04	1,09	<0,001	1,09	1,05	1,12	<0,001
Diabetes mellitus	5,3	2,01	13,9	0,001	4,7	1,5	14,4	0,007
Hypertension	1,46	0,9	2,38	0,125	0,88	0,25	3,1	0,837
CD4 counts, cells/ml	0,998	0,997	0,999	0,098	1	,999	1,001	0,828
HIV RNA <50 cop/ml	0,28	0,12	0,67	0,004	0,125	0,045	0,346	<0,001
Tobacco smoking	1,12	0,56	2,33	0,775	-	-	-	-
Liver stiffness, KPa	**1,05**	**1,03**	**1,07**	**<0,001**	**1,06**	**1,03**	**1,09**	**<0,001**

Cox regression, adjusted by age and sex, enter method. HR: Hazard ratio. 95% CI: 95% Confidence interval

Risk factors for overall mortality among PLWH without other concomitant causes of liver disease. Model including FAST score

Variables at baseline	Crude HR	95% CI HR		P	Adjusted HR	95% CI HR		P
Male sex	1,1	0,44	2,6	0,869	1,14	0,44	3,03	0,770
Age, years	1,07	1,04	1,09	<0,001	1,09	1,06	1,13	<0,001
Diabetes mellitus	5,3	2,01	13,9	0,001	3,96	1,24	12,7	0,020
Hypertension	1,46	0,9	2,38	0,125	1,1	0,34	3,32	0,913
CD4 counts, cells/ml	0,998	0,997	0,999	0,098	0,999	0,998	1	0,440
HIV RNA <50 cop/ml	0,28	0,12	0,67	0,004	0,16	0,061	0,43	<0,001
Tobacco smoking	1,12	0,56	2,33	0,775	-	-	-	-
FAST score ≥0.67	**2,08**	**1,55**	**2,78**	**<0,001**	**1,74**	**1,21**	**2,5**	**0,003**

Cox regression, adjusted by age and sex, enter method. HR: Hazard ratio. 95% CI: 95% Confidence interval

Prognostic yield of the models

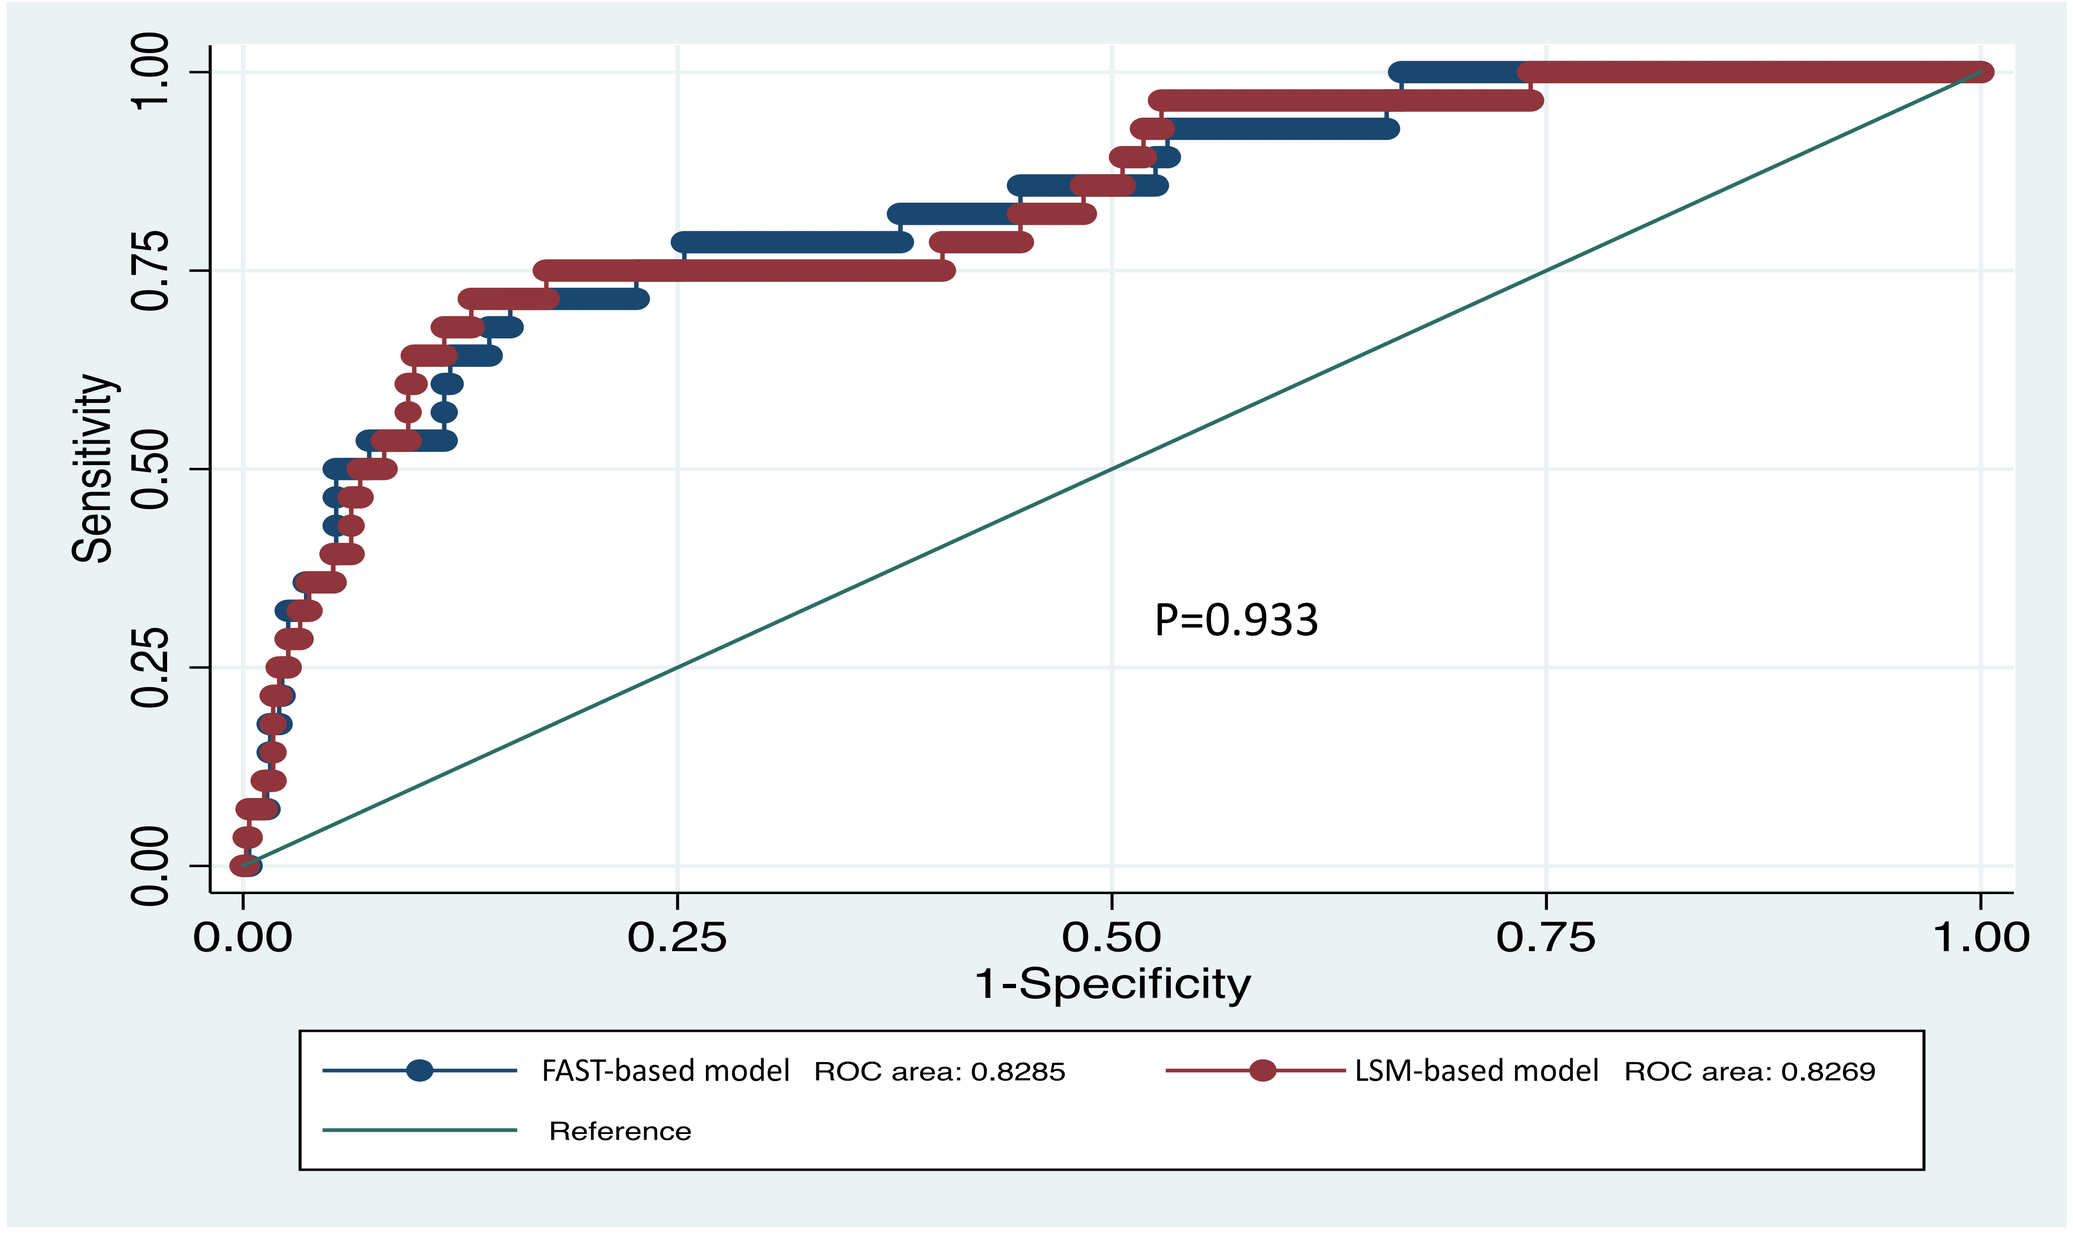
Sensitivity
1-Specificity
P=0.933
FAST-based model ROC area: 0.8285
LSM-based model ROC area: 0.8269
Reference

ADDITIONAL KEY INFORMATION: Funding: This work has been partially funded by the Fondo de Investigaciones Sanitarias (grant no: PI15/01124). **Contact information:** Dr. J. Macías. Unit of Infectious Diseases and Microbiology, Hospital Universitario Virgen de Valme, Avda Bellavista s/n. 41014-Seville, Spain. email: juan.macias.sanchez@gmail.com