GILEAD
Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Absence of Cross-Resistance to Lenacapavir in HIV Entry Inhibitor-Resistant Isolates

508

Nicolas Margot, Vidula Naik, Martin S. Rhee, Christian Callebaut — Gilead Sciences, Inc., Foster City, CA

Introduction

Lenacapavir Targets Multiple Stages of HIV Replication Cycle[1,2]

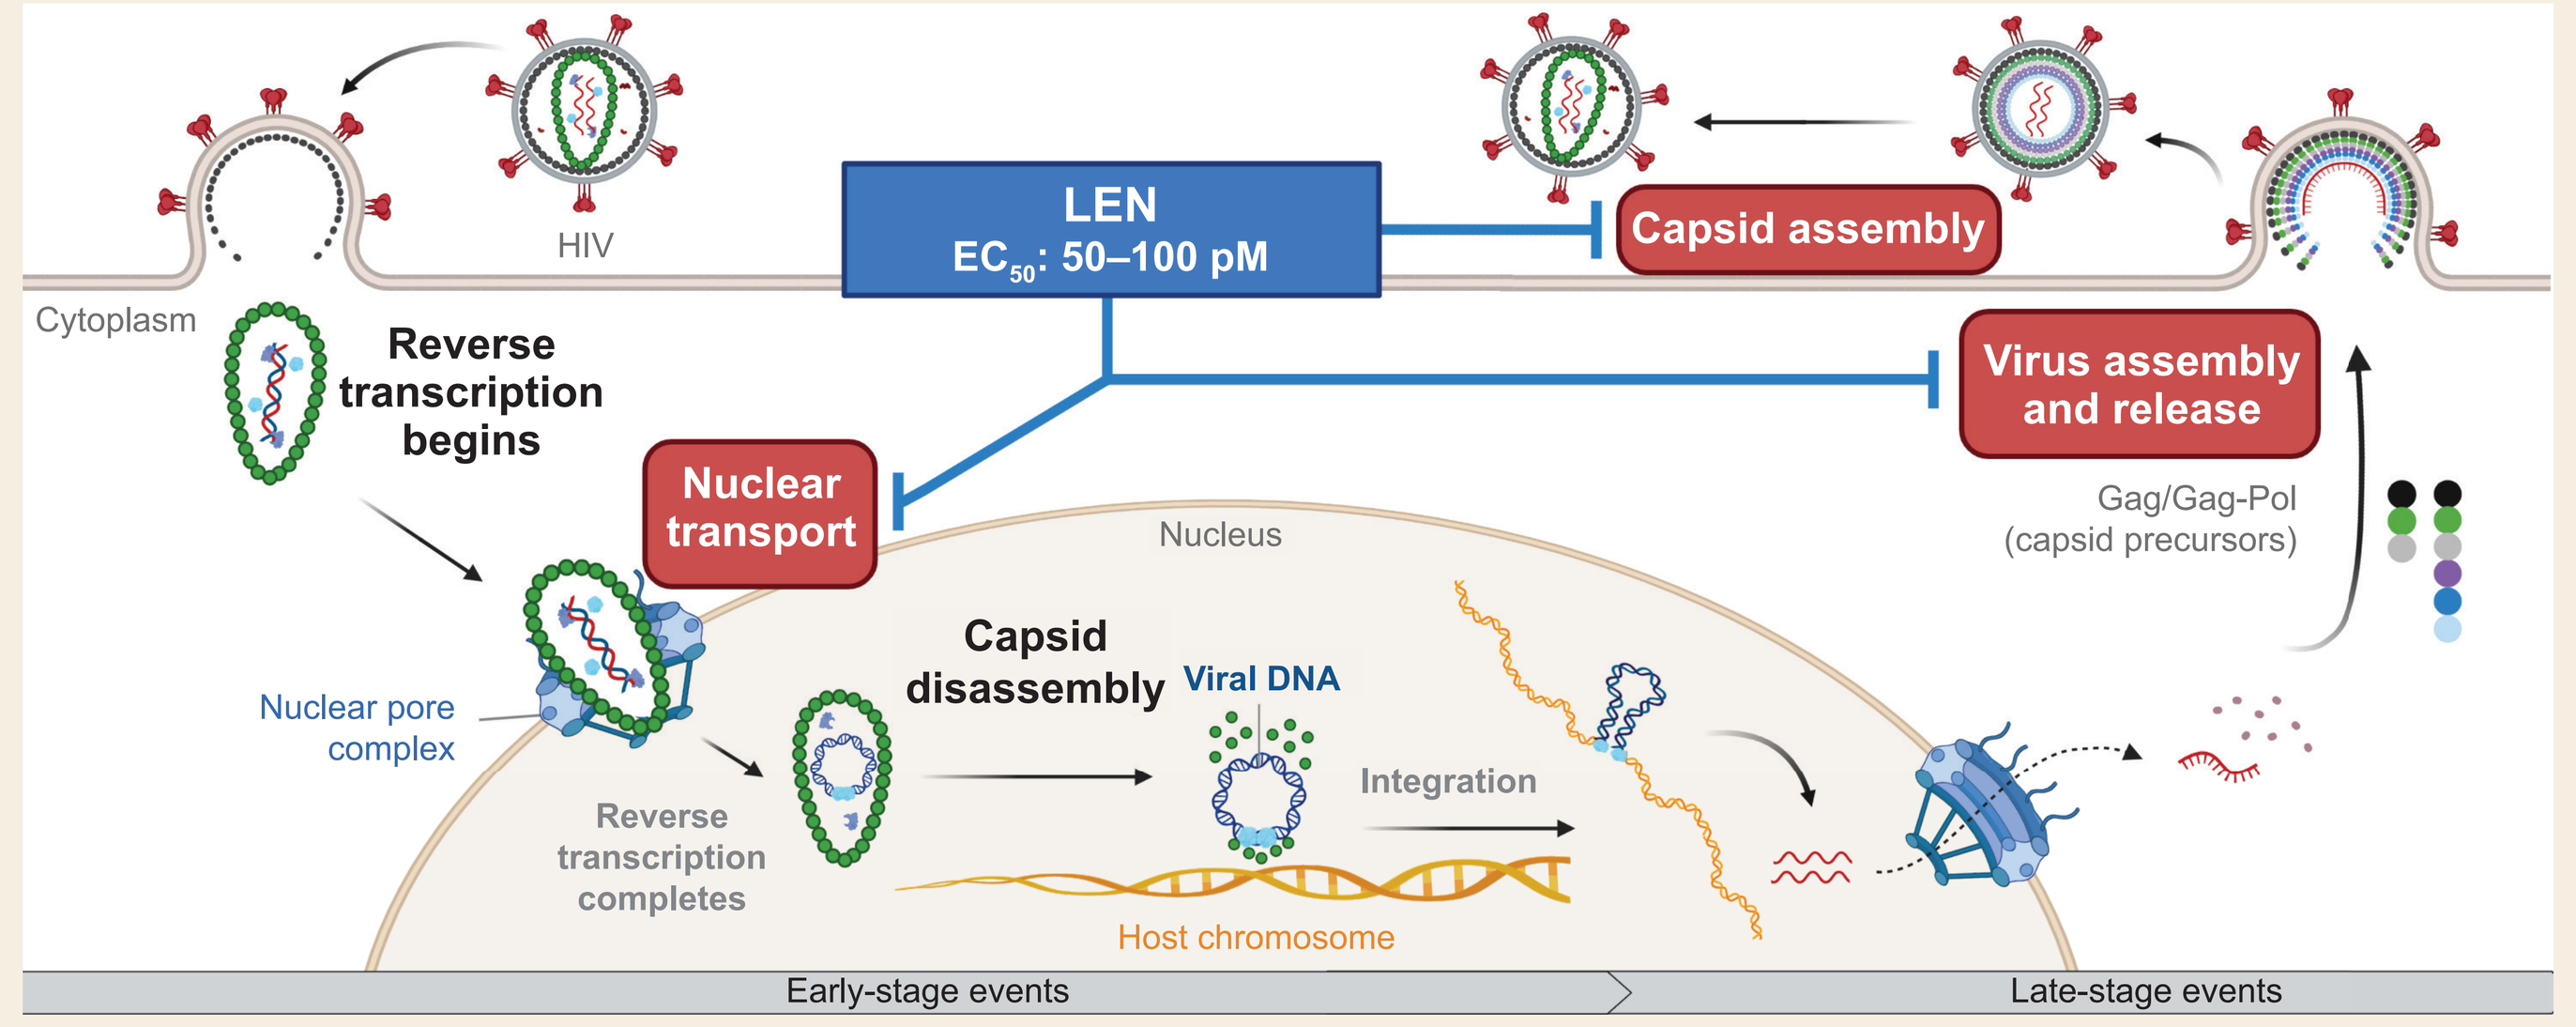
HIV
LEN
EC_{50}: 50–100 pM
Capsid assembly
Cytoplasm
Reverse transcription begins
Virus assembly and release
Nuclear transport
Nucleus
Gag/Gag-Pol (capsid precursors)
Capsid disassembly
Viral DNA
Nuclear pore complex
Integration
Reverse transcription completes
Host chromosome
Early-stage events
Late-stage events

EC_{50}, half-maximal effective concentration; Gag, group antigens; LEN, lenacapavir; Pol, polymerase.

Lenacapavir: Long-Acting Inhibitor of HIV-1 Capsid

- Fully active against HIV with resistance to existing classes of antivirals: nucleos(t)ide reverse transcriptase inhibitors (NRTIs), non-NRTIs (NNRTIs), protease inhibitors (PIs), integrase strand transfer inhibitors (INSTIs), and maturation inhibitors (MIs)[1,3,4]
- The pharmacokinetics of subcutaneous (SC) LEN support its use every 6 months (Q6M)[5]
- Potent antiviral activity in people with HIV (PWH)
 - In a Phase 1 proof-of-concept study, there was an up to 2.3-$\log_{10}$ HIV-1 RNA decline after 9 days of single-dose monotherapy[6]
 - In the Phase 2 CALIBRATE study in treatment-naïve PWH, there was a high rate of viral suppression (94%) at Week 28 when given SC or orally in combination with emtricitabine/tenofovir alafenamide, which was maintained through Week 54[7,8]
 - In the Phase 2/3 CAPELLA study (NCT04150068) in viremic, heavily treatment-experienced PWH with multidrug resistance (MDR), there was a high rate of viral suppression (81%) at Week 26 in combination with an optimized background regimen (OBR), which was maintained through Week 52[9-11]

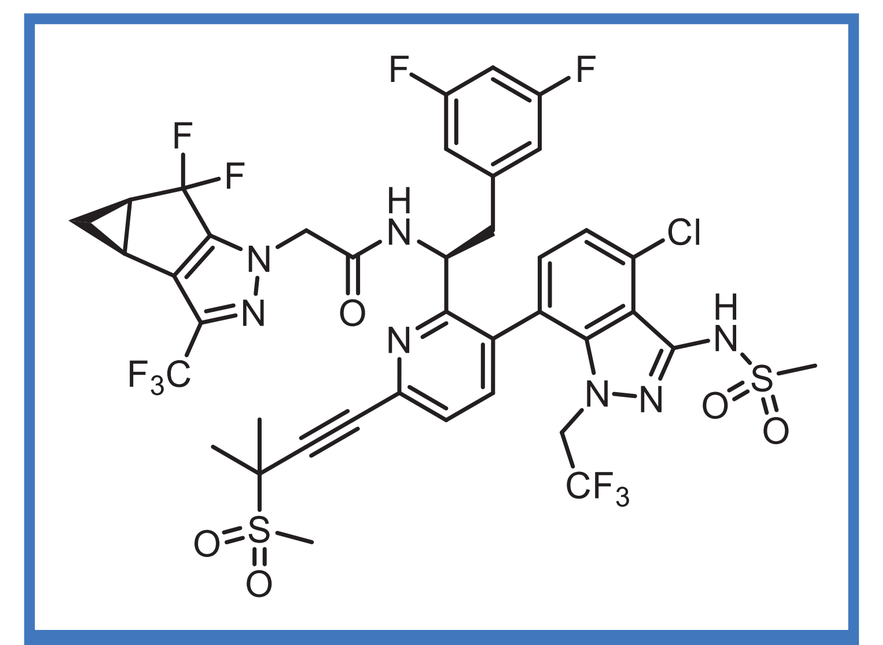

Lenacapavir

CAPELLA Baseline Class Resistance
4 Main ARV Classes

Resistance Within Classes				Participants, n (%)		
NRTI*	NNRTI	PI	INSTI	Cohort 1: n=36	Cohort 2: n=36	All: N=72
✓	✓	✓	✓	17 (47)	16 (44)	33 (46)
✓	✓	✓		9 (25)	13 (36)	22 (31)
✓	✓		✓	8 (22)	5 (14)	13 (18)
✓		✓	✓	2 (6)	0	2 (3)
	✓	✓	✓	0	1 (3)	1 (1)
	✓		✓	0	1 (3)	1 (1)

*M184V/I alone was not sufficient to fulfill NRTI resistance criteria in this study. ARV, antiretroviral.

- Entry criteria: resistance to ≥3 of 4 main ARV classes and ≥2 ARVs within class

Objectives

- To evaluate in vitro susceptibility to entry inhibitors (EIs) and LEN in HIV samples from heavily treatment-experienced PWH with MDR in the CAPELLA study to assess cross-resistance to LEN
- To assess clinical response to LEN in heavily treatment-experienced PWH with MDR in the CAPELLA study in relation to their EI susceptibility

Methods

HIV-1 EIs Prevent HIV Entry Into Target Cells

Enfuvirtide (ENF, T-20)
- Synthetic peptide HIV-1 fusion inhibitor
- ENF binding to HIV-1 gp41 envelope protein prevents HIV fusion with $CD4^+$ target cells

Fostemsavir (FTR)
- Small molecule HIV-1 attachment inhibitor; prodrug of temsavir
- Temsavir binding to HIV-1 gp120 envelope protein prevents HIV attachment to CD4 receptor

Ibalizumab (IBA)
- Monoclonal antibody to CD4 receptor
- IBA binding to CD4 receptor prevents HIV-1 gp120 envelope protein from binding to CD4

Maraviroc (MVC)
- Small molecule HIV-1 C-C chemokine receptor type 5 (CCR5) coreceptor antagonist
- MVC binding to CCR5 prevents HIV-1 gp120 envelope protein from binding to CCR5

Assays for Baseline Susceptibility[12,13]
HIV-1 Entry Inhibitors

- Resistance assays (Monogram Biosciences Inc., South San Francisco, CA):
 - PhenoSense® Entry: phenotypic assay to assess susceptibility to ENF, FTR, and IBA
 - Trofile®: viral coreceptor tropism (CCR5 or C-X-C chemokine receptor type 4 [X4]) assay to assess MVC susceptibility
 - Envelope gene from clinical samples cloned into resistance test vector

Susceptibility Assessment

	ENF	FTR	IBA	MVC
Source	Monogram	Literature[12]	Literature[13]	Monogram
Susceptibility criteria	EC_{50}	EC_{50}	% maximal inhibition	Coreceptor tropism
Determinant/cutoff	≤0.25 µM	<0.1 µM	>83%	CCR5

 - Overall susceptibility score (OSS) set to 1 (criteria met, susceptible) or 0 (criteria not met, nonsusceptible/resistant)

Lenacapavir

- HIV-1 Gag-Pro PhenoSense assay (Monogram): Gag-protease fragment from clinical samples cloned into resistance test vector (assay currently for research use only)

CAPELLA Study Design[9,11]

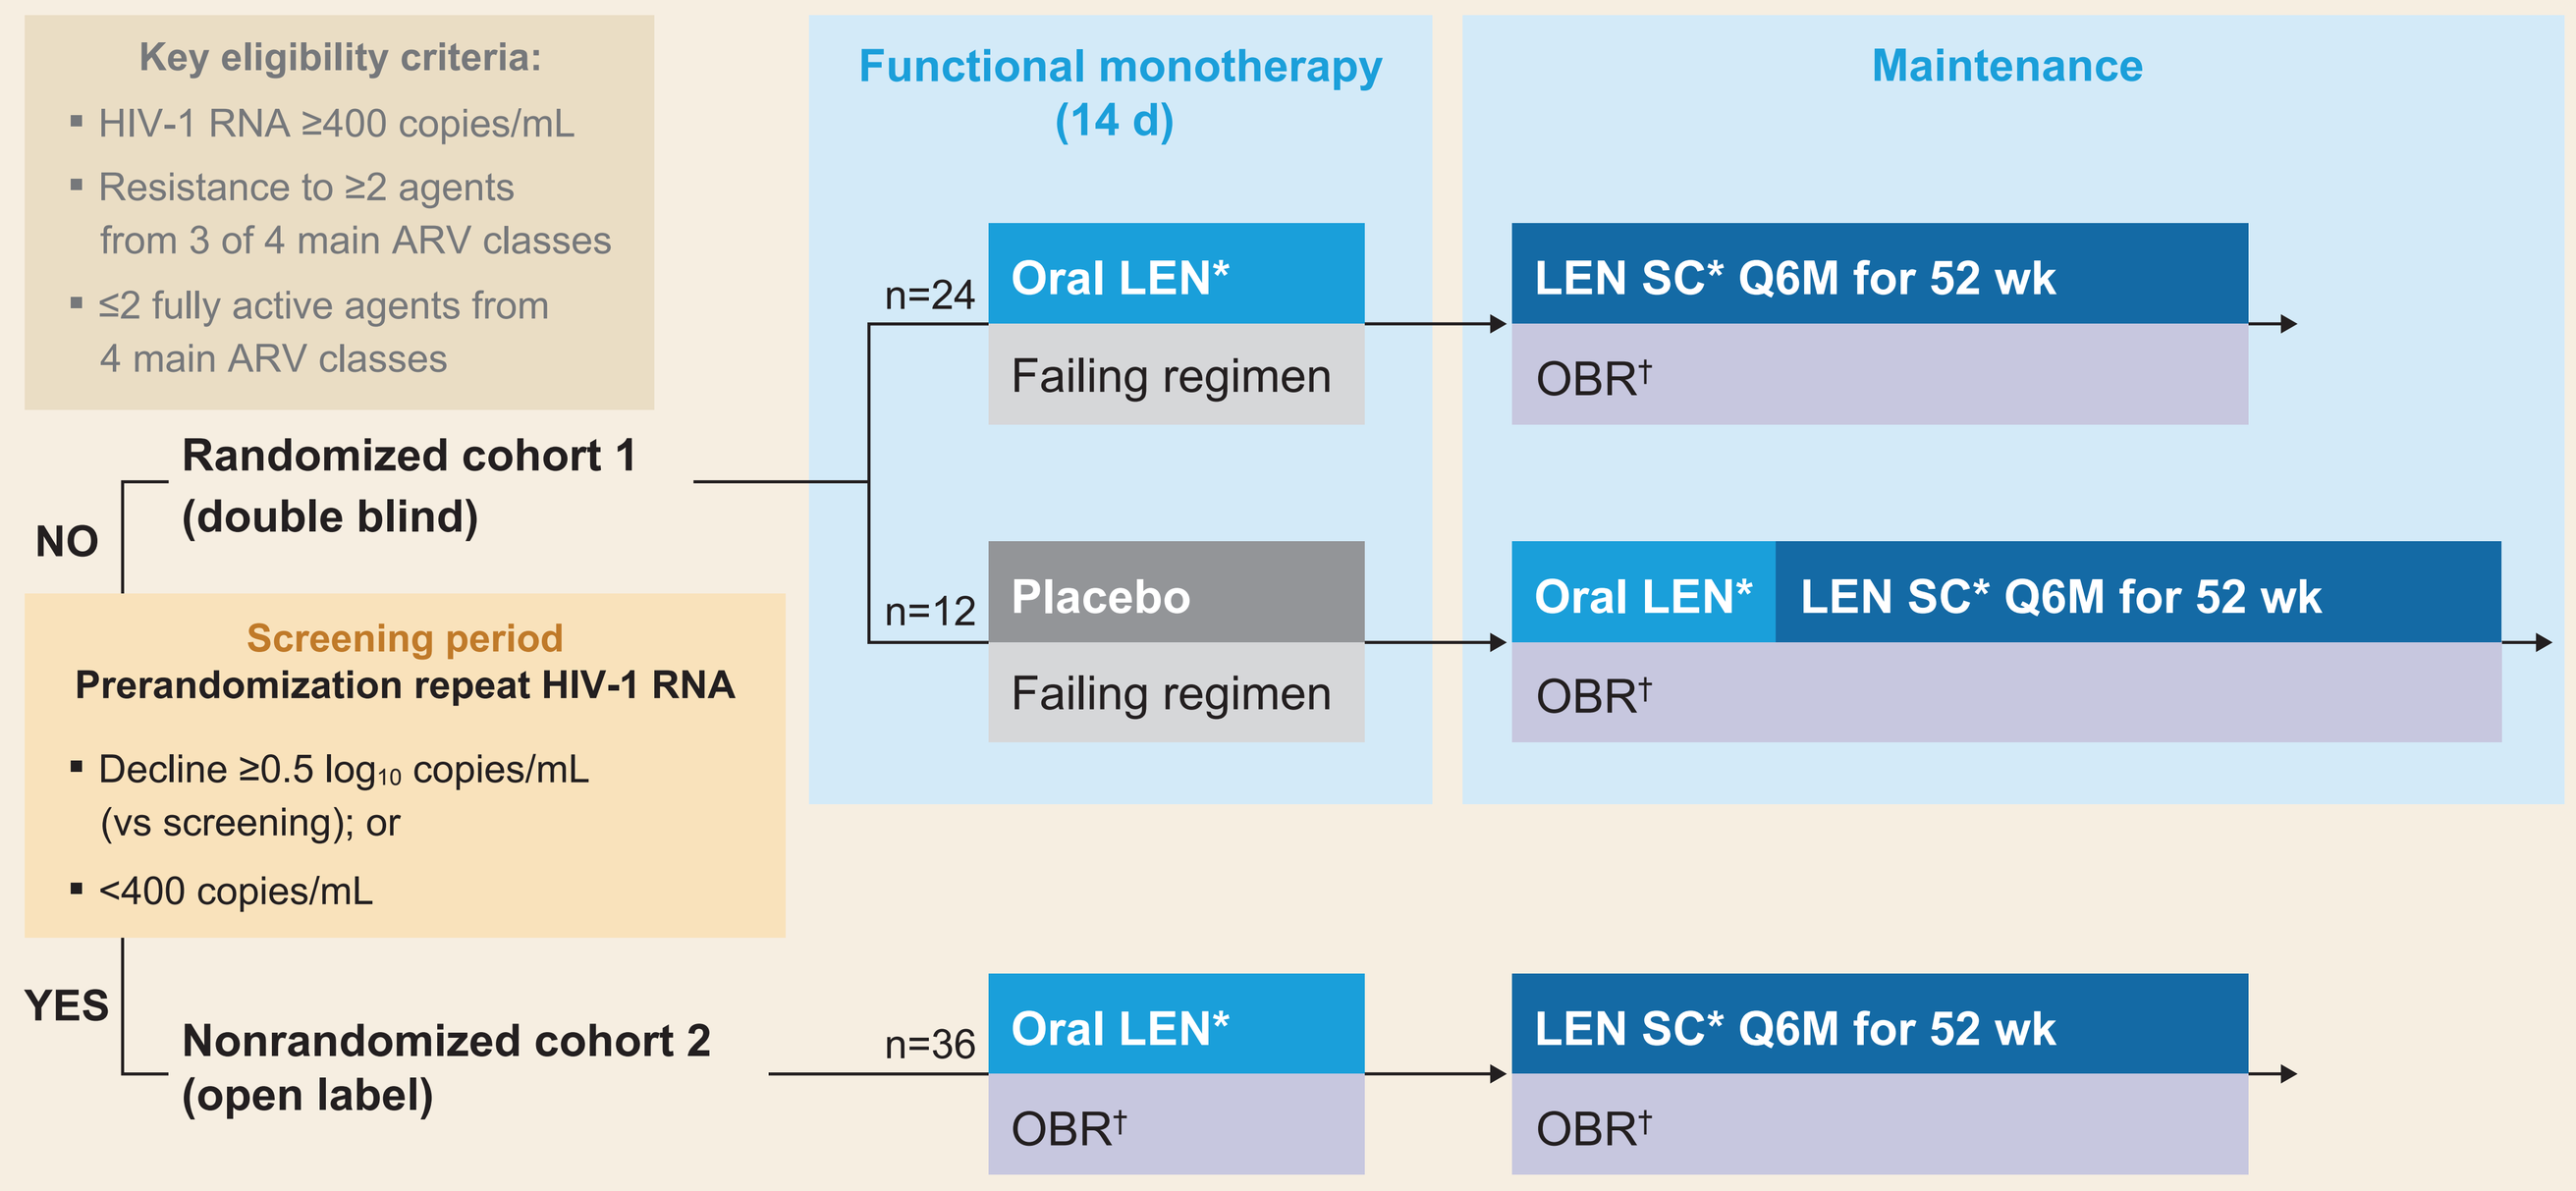
Key eligibility criteria:
HIV-1 RNA ≥400 copies/mL
Resistance to ≥2 agents from 3 of 4 main ARV classes
≤2 fully active agents from 4 main ARV classes
Functional monotherapy (14 d)
Maintenance
n=24
Oral LEN*
Failing regimen
LEN SC* Q6M for 52 wk
OBR†
Randomized cohort 1 (double blind)
NO
n=12
Placebo
Failing regimen
Oral LEN*
LEN SC* Q6M for 52 wk
OBR†
Screening period
Prerandomization repeat HIV-1 RNA
Decline ≥0.5 $\log_{10}$ copies/mL (vs screening); or
<400 copies/mL
YES
Nonrandomized cohort 2 (open label)
n=36
Oral LEN*
OBR†
LEN SC* Q6M for 52 wk
OBR†

*Oral LEN administered as 600 mg on Days 1 and 2, and 300 mg on Day 8 (600 mg on Days 15 and 16, and 300 mg on Day 22 for placebo participants); SC LEN administered as 927 mg (2 x 1.5 mL) in abdomen on Day 15; †Investigational agents, eg, FTR, were allowed; atazanavir (ATV), ATV/cobicistat, ATV/ritonavir, efavirenz, entecavir, nevirapine, and tipranavir were not allowed.

Results

Phenotypic Susceptibility to EIs*

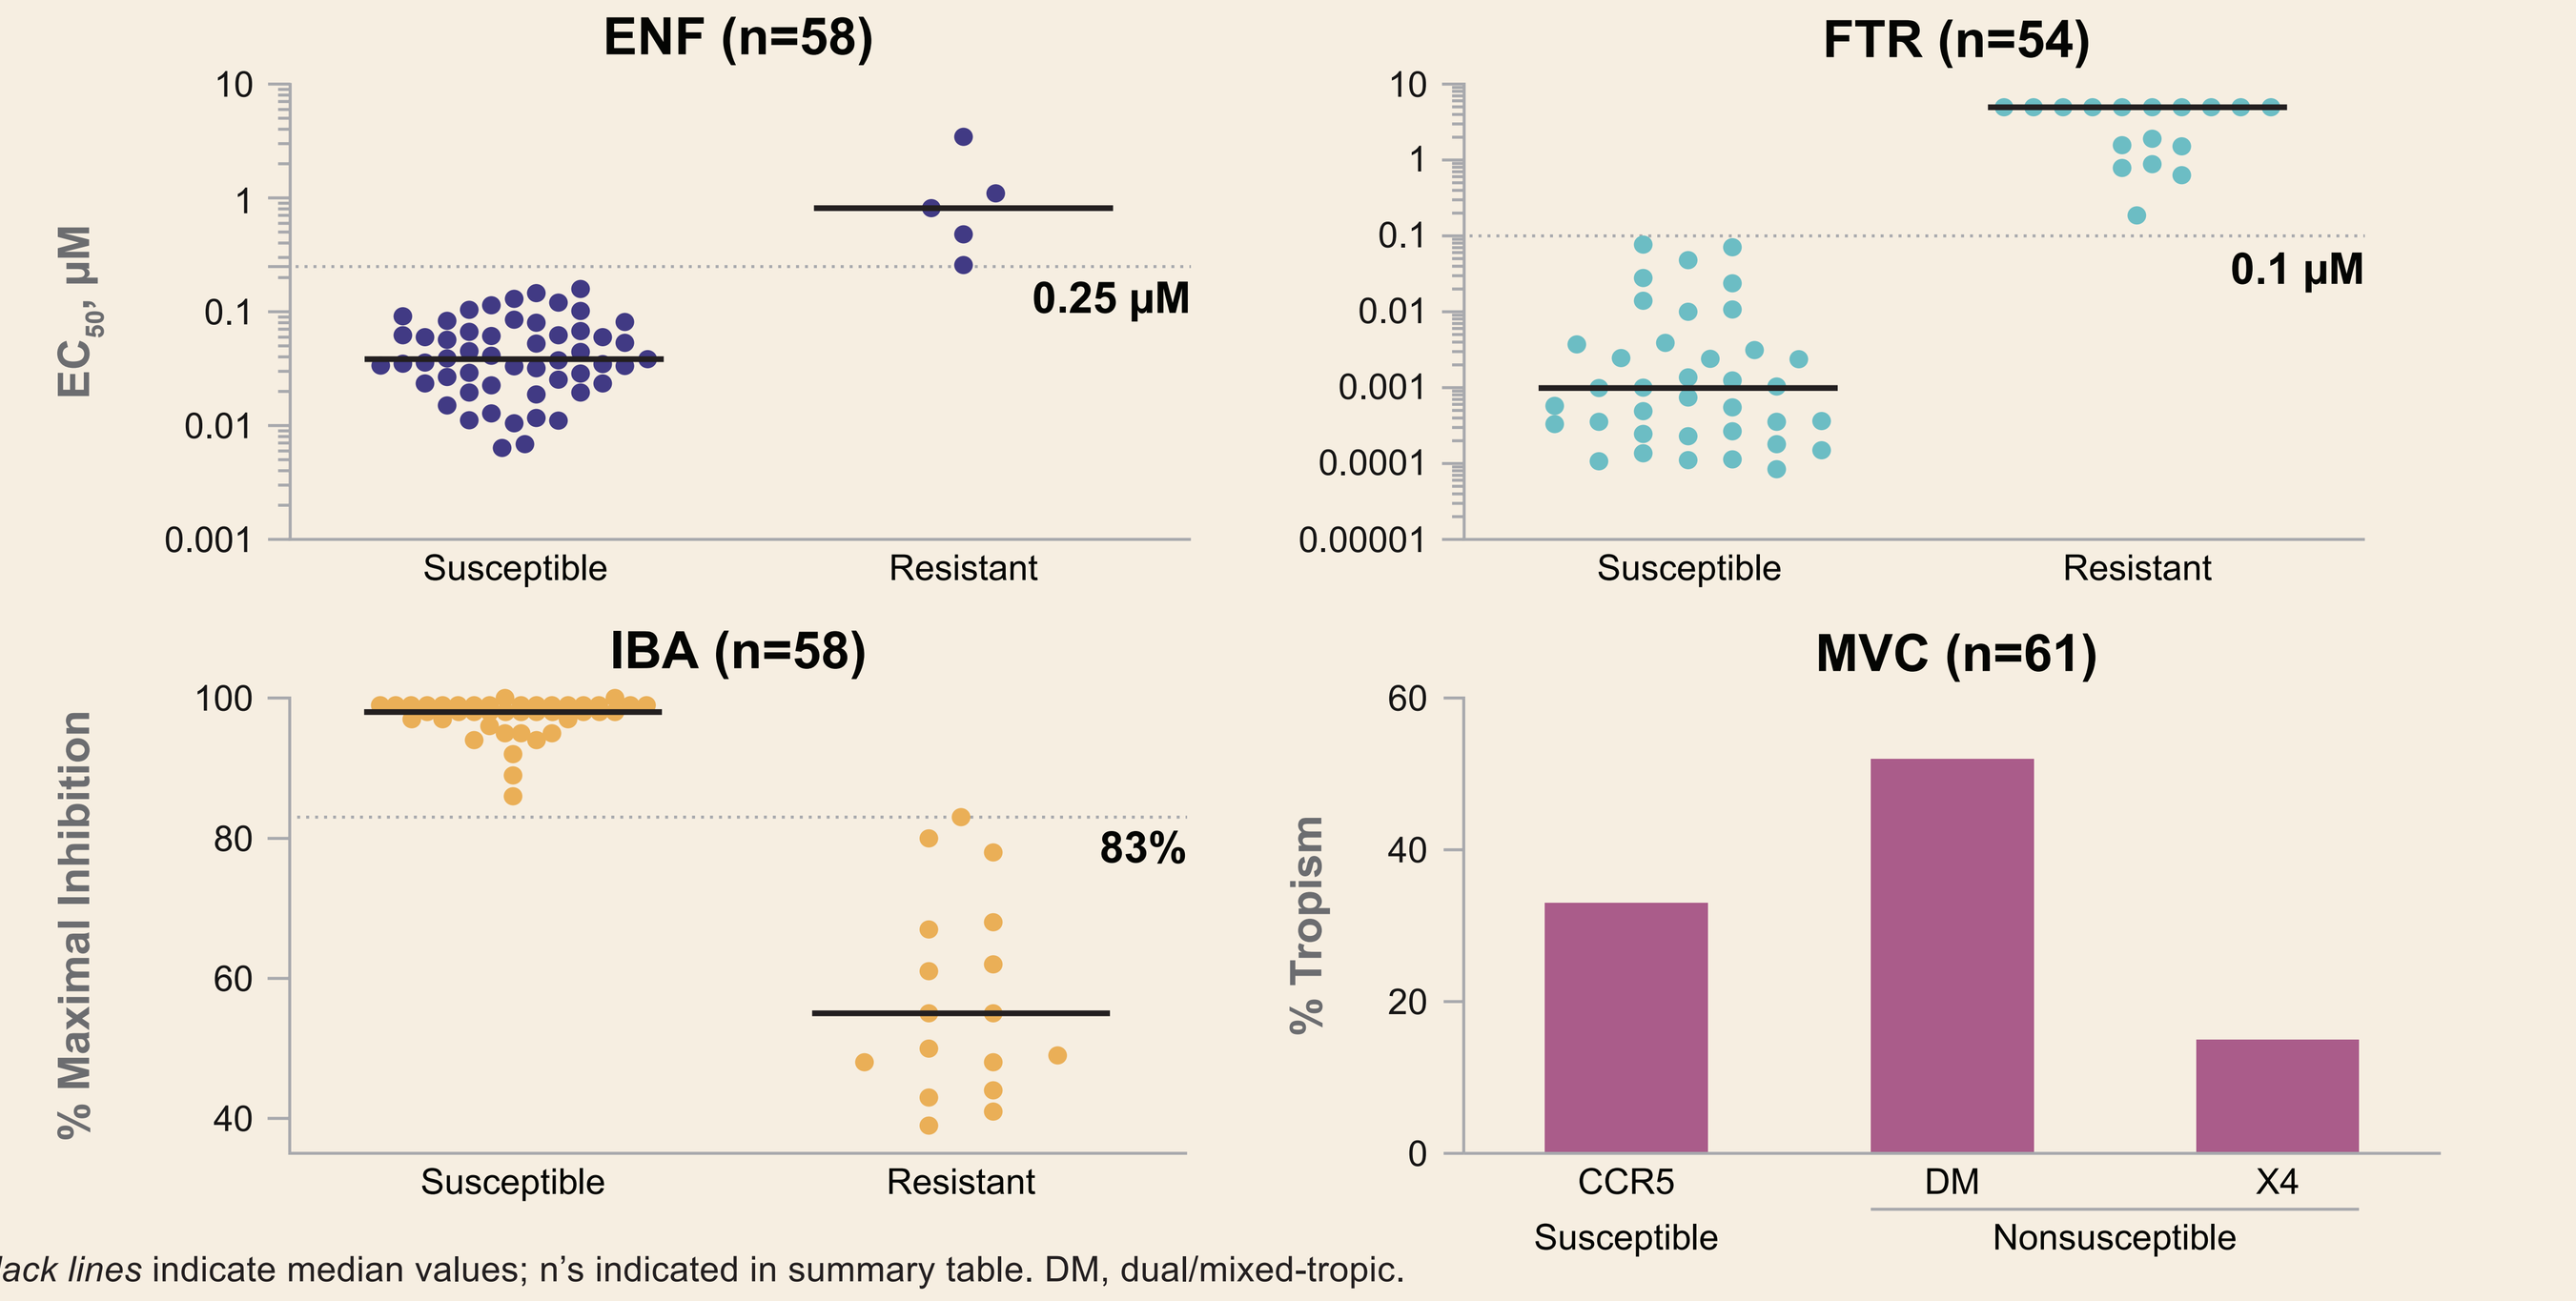
ENF (n=58)
EC_{50}, µM
10
1
0.1
0.01
0.001
0.25 µM
Susceptible
Resistant
FTR (n=54)
10
1
0.1
0.01
0.001
0.0001
0.00001
0.1 µM
Susceptible
Resistant
IBA (n=58)
% Maximal Inhibition
100
80
60
40
83%
Susceptible
Resistant
MVC (n=61)
% Tropism
60
40
20
0
CCR5
DM
X4
Susceptible
Nonsusceptible

**Black lines* indicate median values; n's indicated in summary table. DM, dual/mixed-tropic.

Summary of Baseline EI Susceptibility

Participants With Nonsusceptible Virus/Participants With Data (%)*

EI	Cohort 1 n=36	Cohort 2 n=36	All Participants N=72
ENF	5/33 (15)	0/25 (0)	5/58 (9)
FTR	10/33 (30)	7/21 (33)	17/54 (31)
IBA	11/33 (33)	6/25 (24)	17/58 (29)
MVC	27/35 (77)	14/26 (54)	41/61 (67)

*Nonsusceptibility to EIs was associated with OSS of 0.

- Nonsusceptibility to EIs at baseline was common in heavily treatment-experienced participants

Baseline Susceptibility to LEN by EI Susceptibility*

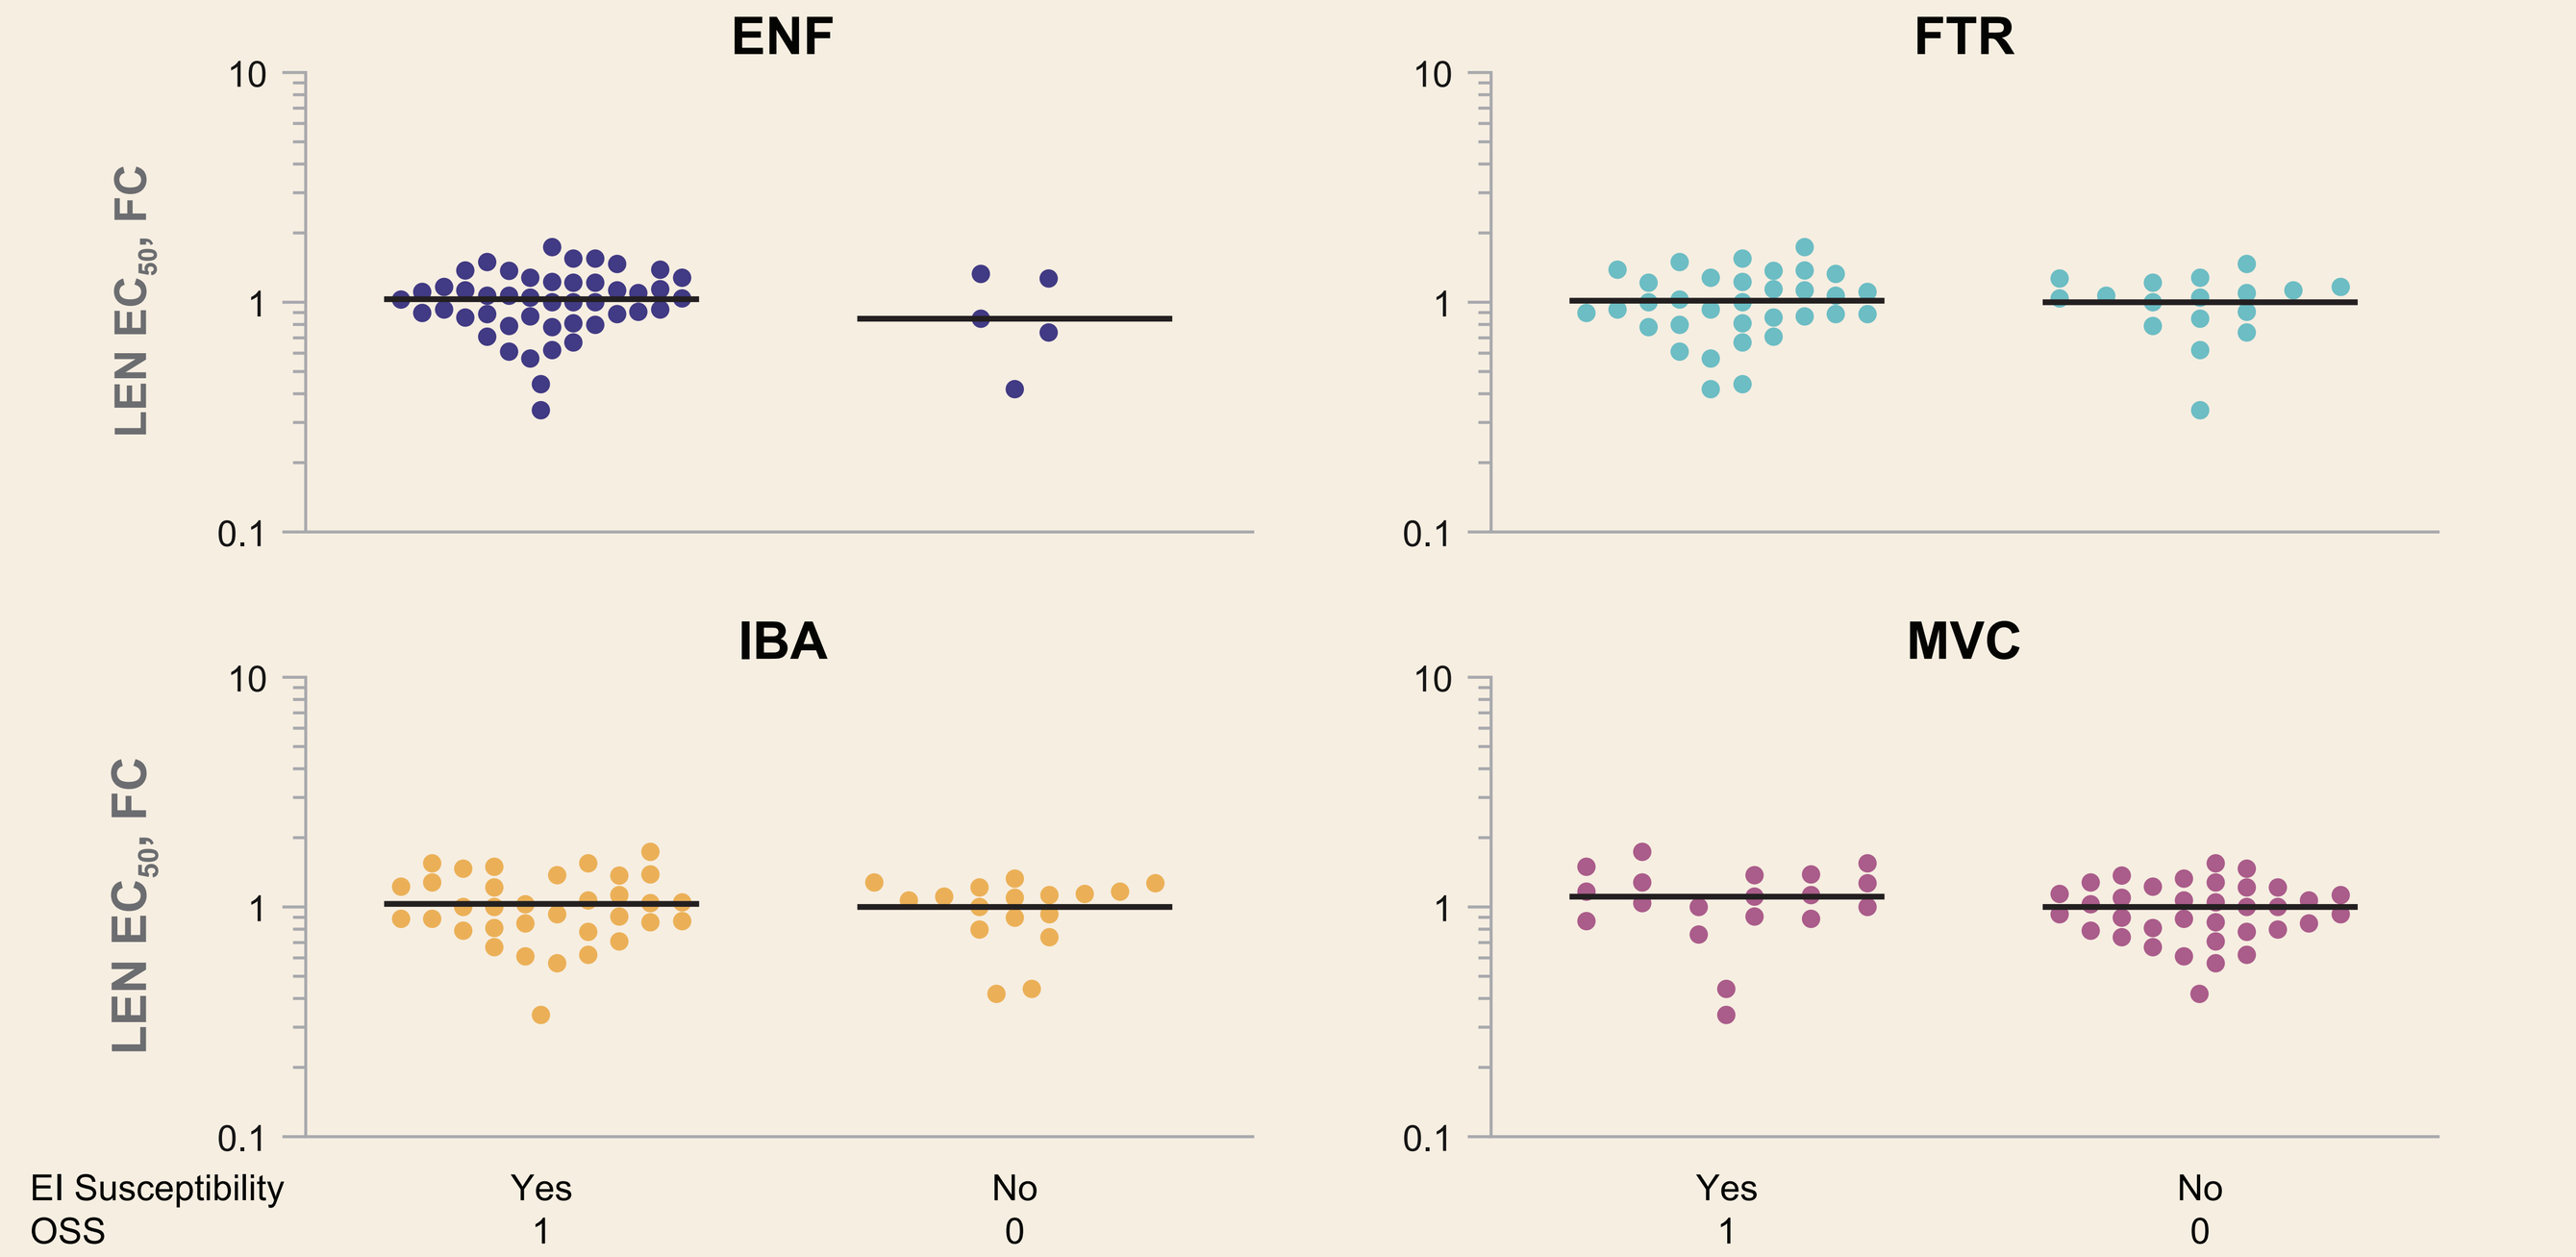
ENF
FTR
LEN EC_{50}, FC
10
1
0.1
IBA
MVC
EI Susceptibility
OSS
Yes
1
No
0

**Black lines* indicate mean LEN EC_{50} fold change (FC).

Summary of LEN FC by EI Susceptibility

	LEN Susceptibility by EI Susceptibility			
	Samples Susceptible to EI		Samples Not Susceptible to EI	
EI	n	Mean LEN FC (range)	n	Mean LEN FC (range)
ENF	46	1.0 (0.34–1.74)	5	0.9 (0.42–1.33)
FTR	33	1.0 (0.42–1.74)	17	1.0 (0.34–1.47)
IBA	34	1.0 (0.34–1.74)	17	1.0 (0.42–1.33)
MVC	19	1.1 (0.34–1.74)	35	1.0 (0.42–1.55)

- Susceptibility to LEN was unaffected by EI susceptibility
 - Mean LEN FCs were similar to wild-type control across all EI phenotypes
- LEN susceptibility was not affected by envelope-associated resistance or viral tropism

Baseline EI Susceptibility by Week 26 Treatment Outcome
Treatment Success vs Failure*

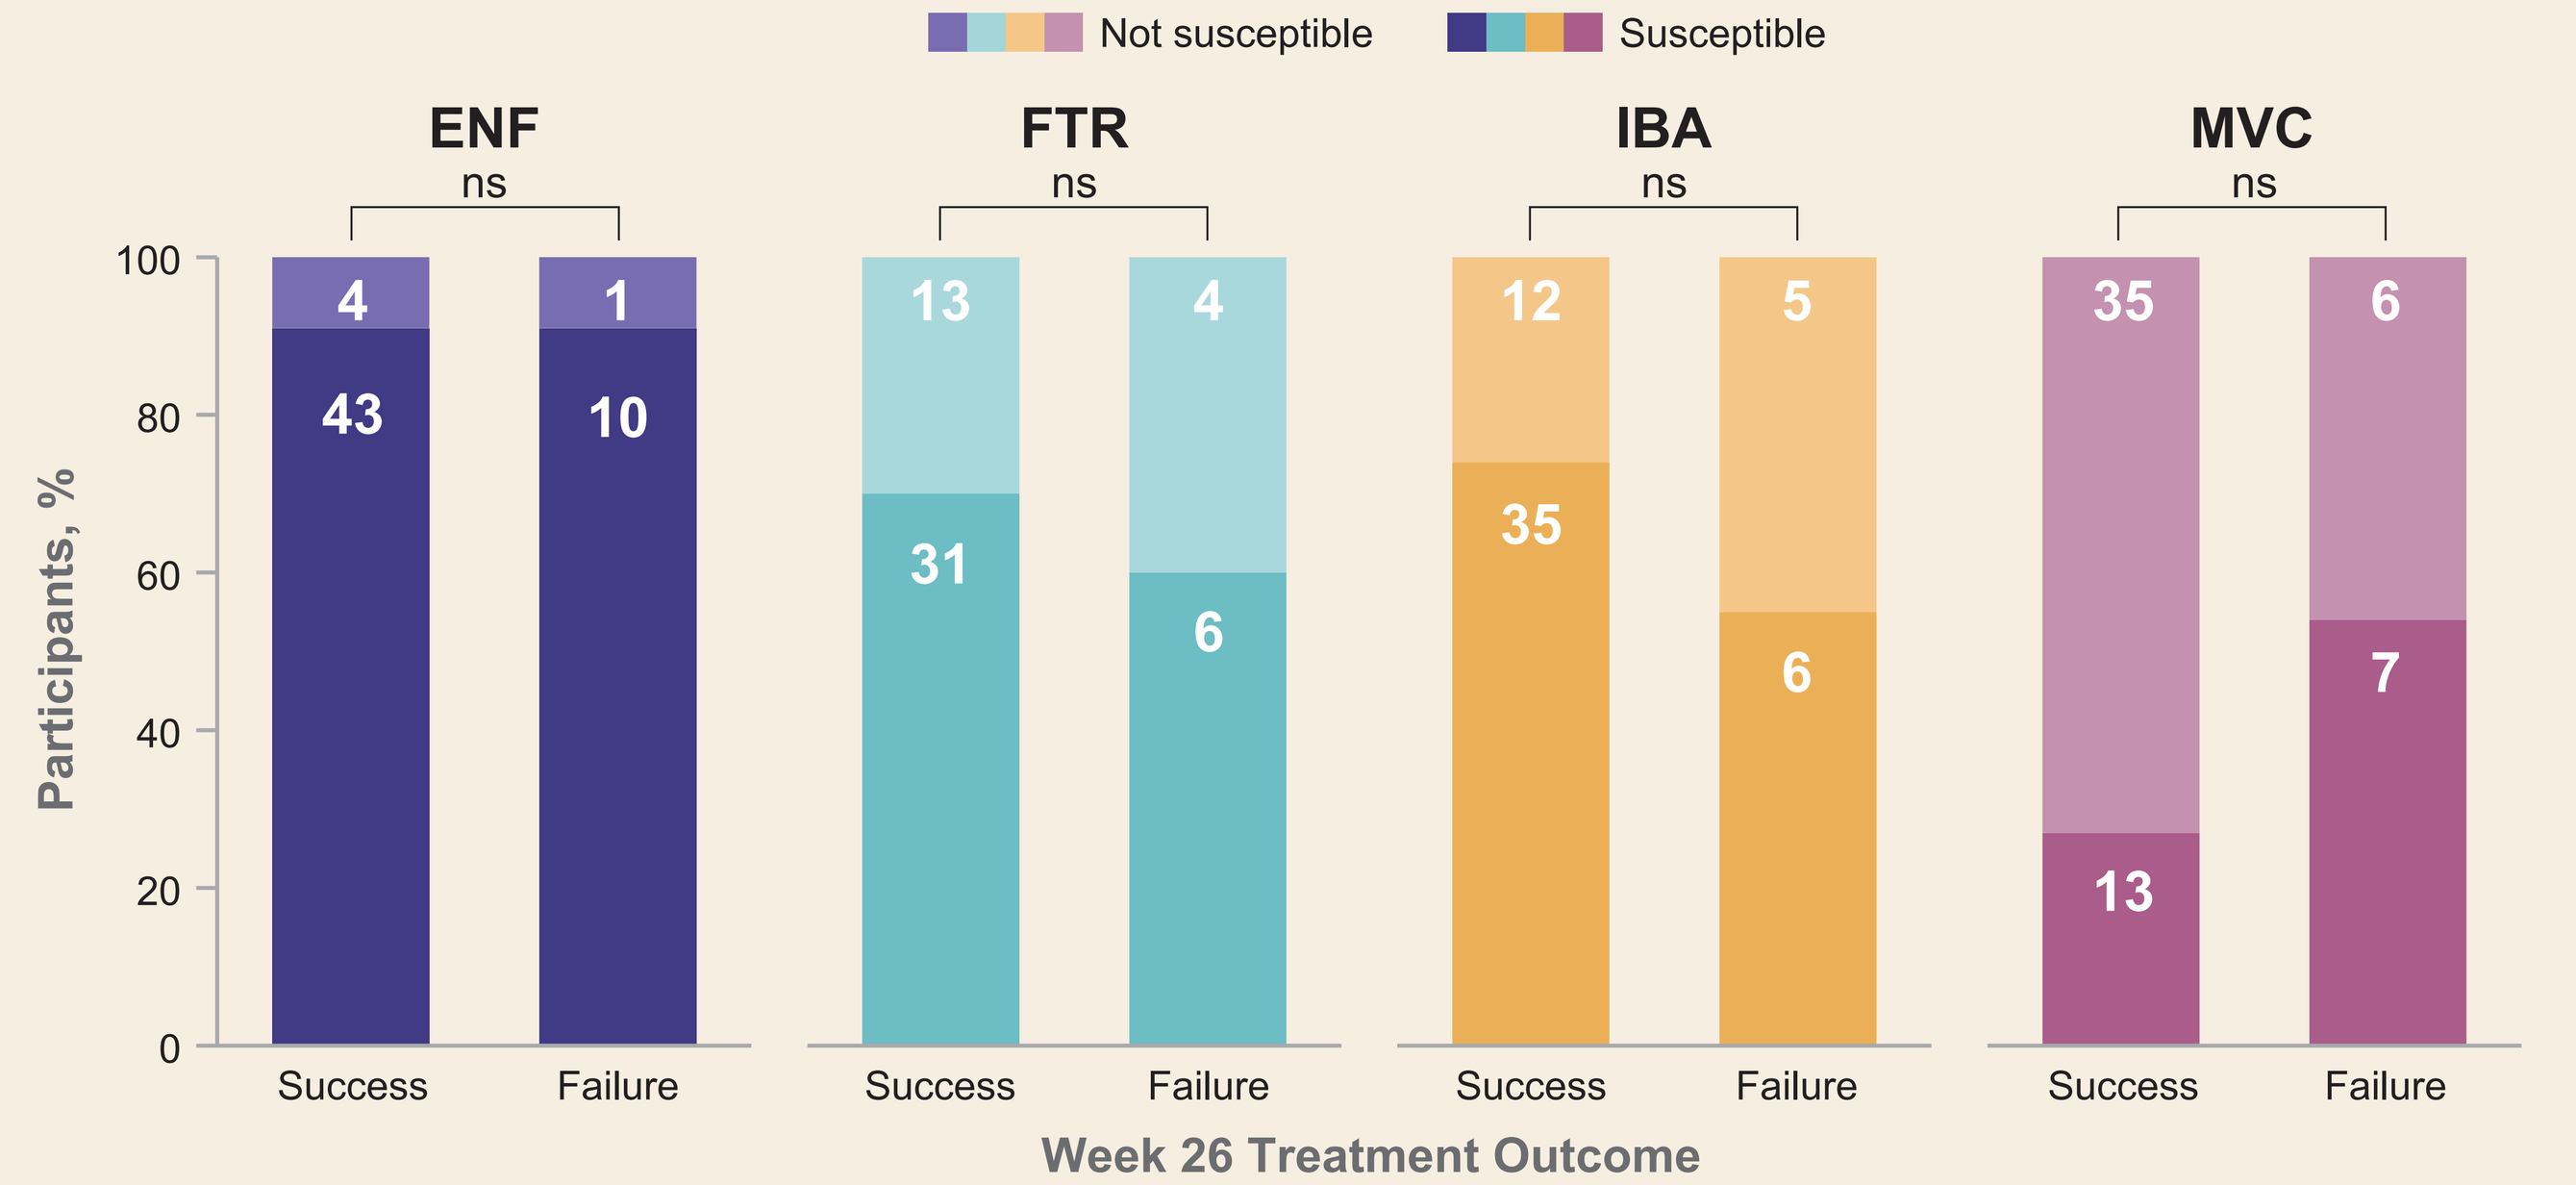
Not susceptible
Susceptible
ENF
ns
FTR
ns
IBA
ns
MVC
ns
Participants, %
100
80
60
40
20
0
4
43
1
10
13
31
4
6
12
35
5
6
35
13
6
7
Success
Failure
Week 26 Treatment Outcome

*FDA Snapshot algorithm; success: HIV-1 RNA <50 copies/mL at Week 26; failure: HIV-1 RNA ≥50 copies/mL or no virologic data at Week 26. ns, not significant.

- Baseline EI susceptibility in participants with treatment success was not different from that of participants with treatment failure at Week 26 (Fisher exact test: p>0.10)
 - Response to LEN + OBR was unaffected by EI susceptibility

Clinical Response to LEN[9-11]
Heavily Treatment-Experienced Participants With MDR

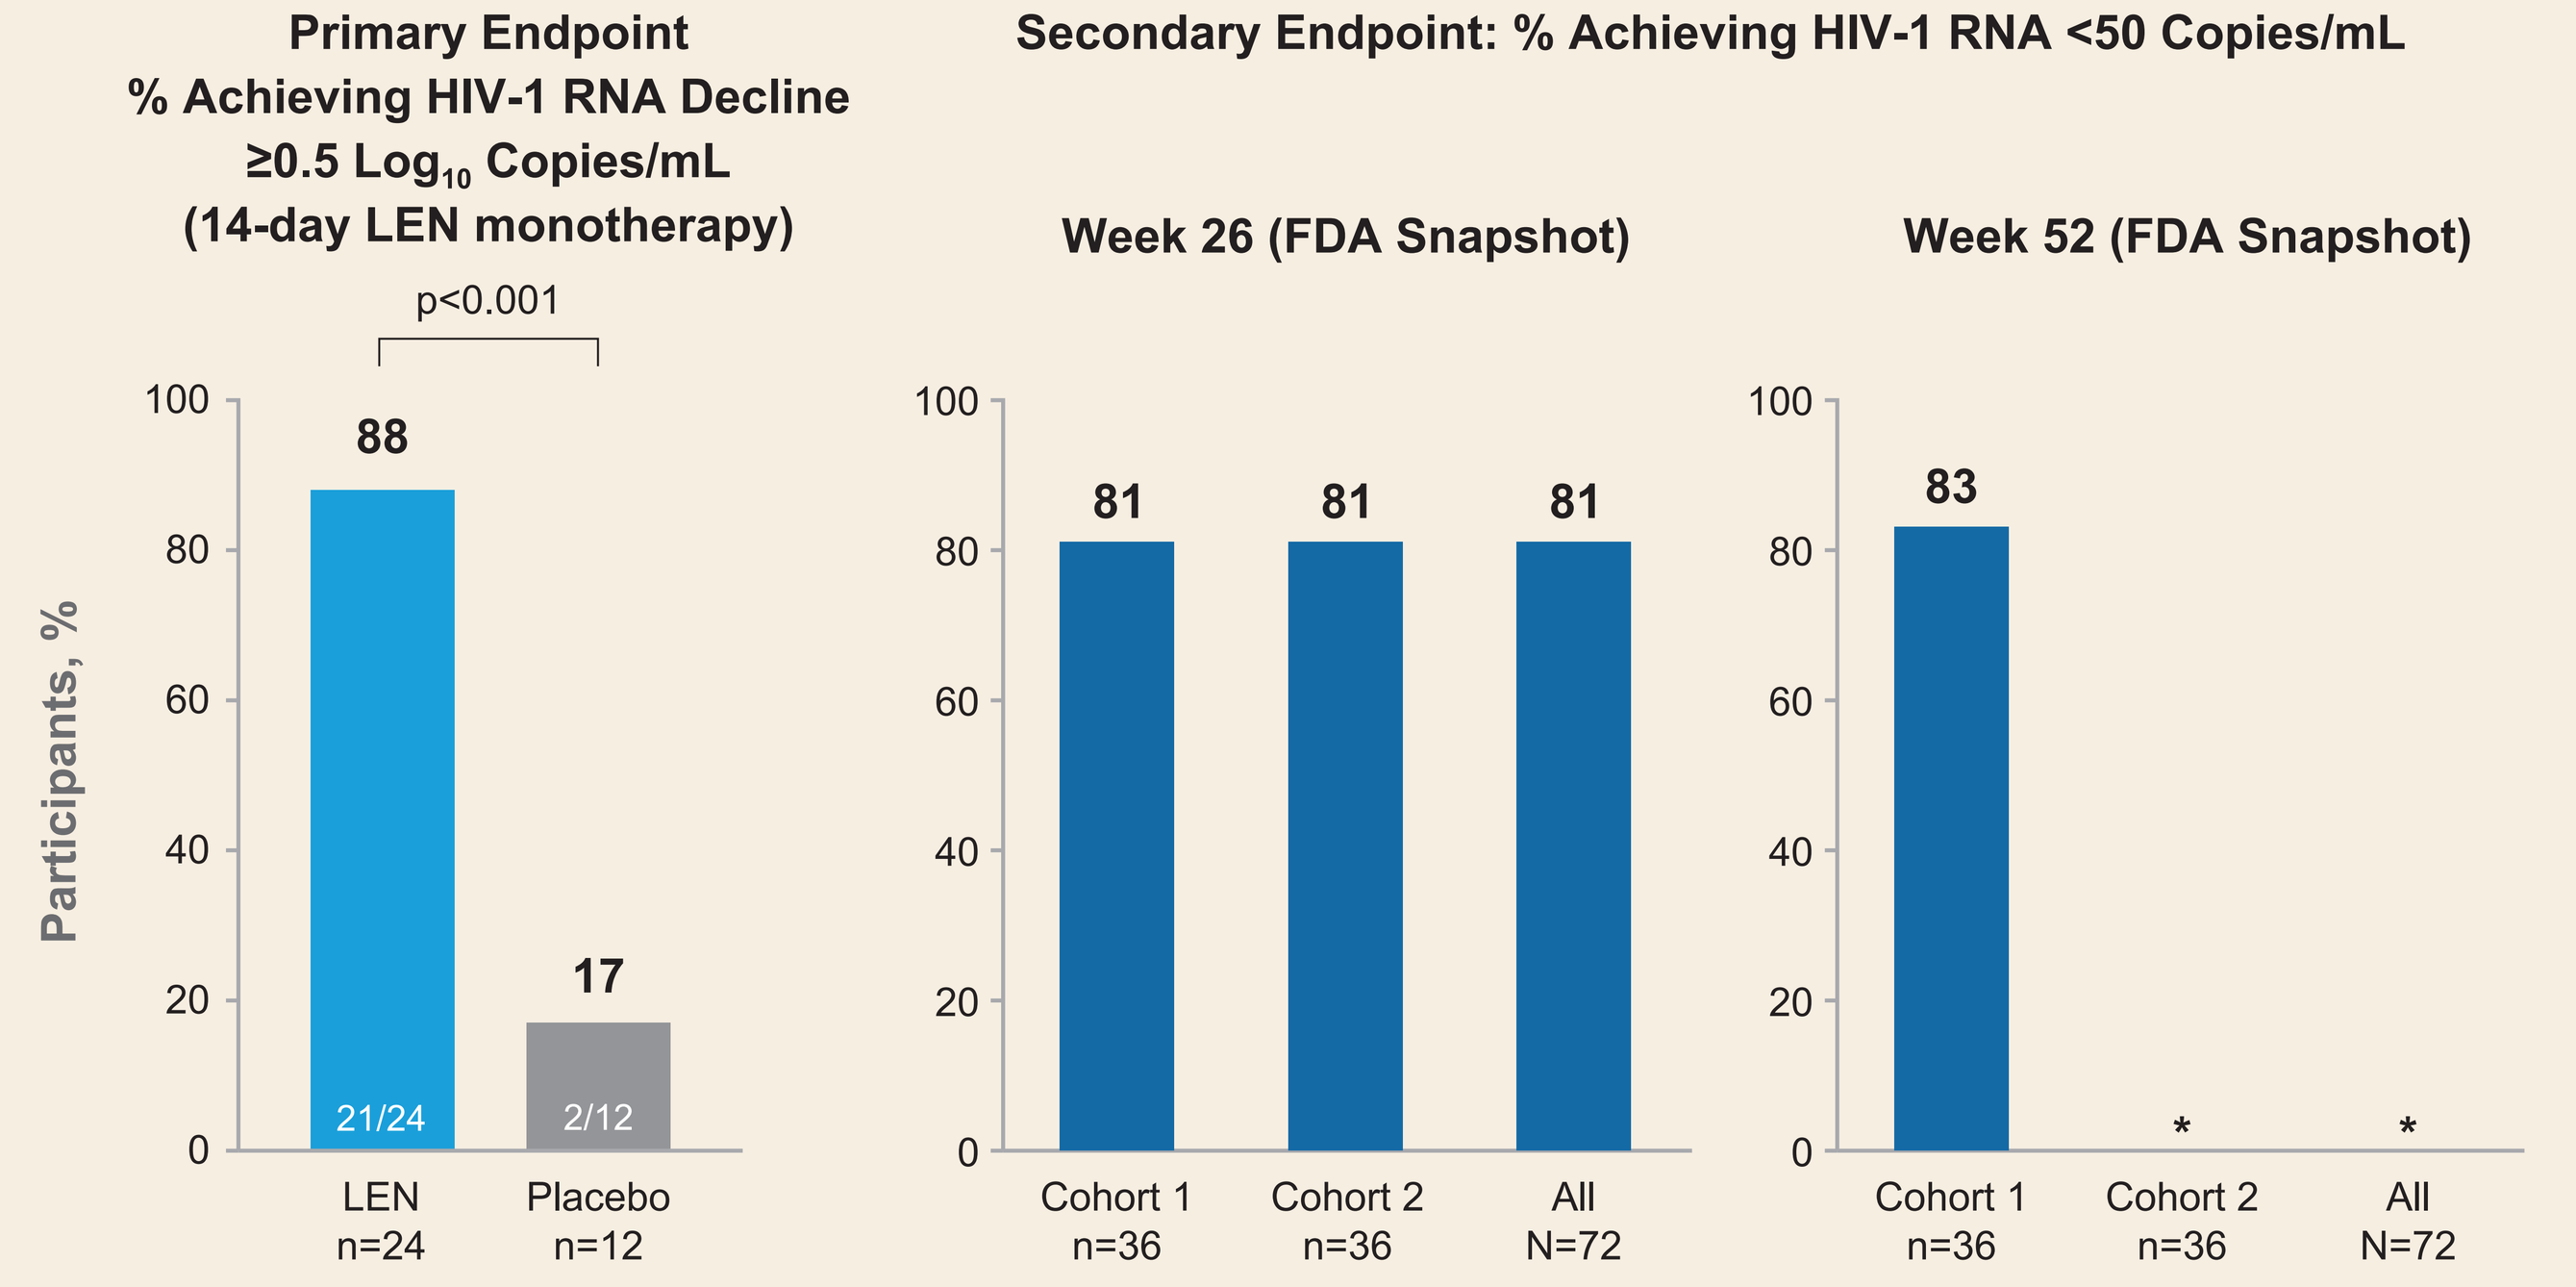
Primary Endpoint
% Achieving HIV-1 RNA Decline ≥0.5 $\log_{10}$ Copies/mL
(14-day LEN monotherapy)
Secondary Endpoint: % Achieving HIV-1 RNA <50 Copies/mL
Week 26 (FDA Snapshot)
Week 52 (FDA Snapshot)
p<0.001
Participants, %
100
80
60
40
20
0
88
21/24
17
2/12
LEN n=24
Placebo n=12
81
81
81
Cohort 1 n=36
Cohort 2 n=36
All N=72
83
*
*

*Data not available as most participants in Cohort 2 have not reached Week 52 time point.

Conclusions

- The gag sequence from EI-nonsusceptible isolates did not affect LEN susceptibility
 - There was no association between EI susceptibility and LEN antiviral activity
- These data complement prior data showing absence of cross-resistance between LEN and other ARV classes (NRTI, NNRTI, PI, and INSTI) and MIs
 - LEN resistance profile is orthogonal to that of all other ARV classes
- Treatment response to LEN + OBR was unaffected by EI susceptibility
- Lack of cross-resistance across ARV classes supports the use of LEN with OBR in PWH regardless of treatment history

References: **1.** Link JO, et al. Nature 2020;584:614-8; **2.** Zila V, et al. Cell 2021;184:1032-46.e18; **3.** Margot N, et al. Antimicrob Agents Chemother 2021;65:e02057-20; **4.** VanderVeen L, et al. CROI 2021, oral 128; **5.** Begley R, et al. AIDS 2020, abstr PEB0265; **6.** Daar E, et al. CROI 2020, poster 469; **7.** Gupta SK, et al. CROI 2022, abstr 1292; **8.** Gupta SK, et al. IAS 2021, oral OALB0302; **9.** Molina J-M, et al. IAS 2021, oral OALX01LB; **10.** Ogbuagu O, et al. CROI 2022, abstr 1047; **11.** Segal-Maurer S, et al. CROI 2021, oral 127; **12.** Jacobson JM, et al. Antimicrob Agents Chemother 2009;53:450-7; **13.** Lataillade M, et al. J Acquir Immune Defic Syndr 2018;77:299-307. **Acknowledgments:** We are grateful to all the individuals who participated in this trial, their partners and families, and the participating study investigators and their study teams: *Canada:* J Brunetto, B Trottier; *Dominican Republic:* E Koenig; *France:* J-M Molina, S Ronot-Bregigeon, Y Yazdanpanah; *Germany:* H-J Stellbrink; *Italy:* A Antinori, A Castagna, F Castelli; *Japan:* T Shirasaka, Y Yokomaku; *South Africa:* M Rassool; *Spain:* J Mallolas; *Taiwan:* C-C Hung; *Thailand:* A Avihingsanon, P Chetchotisakd, K Siripassorn, W Ratanasuwan; *USA:* DS Berger, M Berhe, C Brinson, CM Creticos, GE Crofoot, E DeJesus, D Hagins, T Hodge, K Lichtenstein, JP McGowan, O Ogbuagu, O Osiyemi, GJ Richmond, MN Ramgopal, PJ Ruane, W Sanchez, S Segal-Maurer, J Sims, GI Sinclair, DA Wheeler, A Wiznia, K Workowski, C Zurawski. We also thank Monogram Biosciences for the resistance analyses. This study was funded by Gilead Sciences, Inc. Editing and production assistance were provided by BioScience Communications, New York, NY, funded by Gilead.

Presented at *Virtual* CROI 2022, February 12–16, 2022

© 2022 Gilead Sciences, Inc. All rights reserved.