SUSCEPTIBILITY TO 3BNC117 AND 10-1074 IN ART SUPPRESSED CHRONICALLY INFECTED PERSONS

Luis J. Montaner[1], Kenneth Lynn[2], Livio Azzoni[1], Emmanouil Papasavvas[1], Matthew Fair[1], Giorgio Cocchella[2], Charles Kang[3], Jacqueline D. Reeves[3], Linden Lalley-Chareczko[4], Jay R. Kostman[5], William R. Short[2], Karam Mounzer[4], Christos J. Petropoulos[3], Pablo Tebas[2]

[1]The Wistar Institute, Philadelphia, PA, USA; [2]University of Pennsylvania, Philadelphia, PA, USA; [3]Labcorp-Monogram Biosciences, South San Francisco, CA; [4]Jonathan Lax Immune Disorders Treatment Center, Philadelphia Field Initiating Group for HIV-1 Trials, Philadelphia, PA, USA; [5]John Bell Health Center, Philadelphia Field Initiating Group for HIV-1 Trials, Philadelphia, PA, USA

THE WISTAR INSTITUTE BEAT-HIV DELANEY COLLABORATORY

Introduction

- Broadly neutralizing anti-HIV monoclonal antibodies (bNAbs) may have a role in the prevention and treatment of HIV.
- Preexisting resistance and the rapid development of resistance when used as monotherapy may limit their utility
- The PhenoSense mAb Assay is the only CLIA/CAP compliant screening test available to assess bNAb susceptibility in people with HIV infection.
- As part of the screening process for the BEAT2 trial we evaluated the sensitivity of the HIV reservoir in suppressed participants against two bNAbs: a CD4 binding antibody 3BNC117 and a V1-V2 loop binding antibody 10-1074

Subjects, Materials & Methods

Study participants

- 61 HIV^+ individuals were screened.
- All participants were chronically suppressed on ART at <20 HIV-1 copies/ml and had $CD4^+$ T cell count ≥450 $cells/mm^3$ and nadir $CD4^+$ T cell count >200 $cells/mm^3$.

Methods

- The PhenoSense Monoclonal Antibody (mAb) Assay (Labcorp-Monogram Biosciences) assess the susceptibility of pseudo-virions bearing plasma or PMBC derived HIV-1 envelope proteins to anti-envelope mAbs.
- Full-length envelope sequences, amplified from PBMC-associated HIV DNA were cloned into an envelope expression vector.
- Pseudo-virions were then tested for neutralization sensitivity to 3BNC117 and 10-1074 bNAbs. The concentration of bNAb required to inhibit virus infectivity by 50%, 80%, 90% and 95% (IC50, IC80, IC90 and IC95, respectively), as well as the maximum percent inhibition (MPI) was assessed.
- Clinical cut-off values for neutralization sensitivity have not been established for the PhenoSense mAb Assay.
- For the BEAT2 study, the following exploratory values were utilized to define mAb sensitivity: IC90 < 2 μg/mL for 3BNC117 and < 1.5 μg/mL for 10-1074 and a maximum percent inhibition (MPI) > 98%.

Figure 1. PhenoSense Monoclonal Antibody (mAb) Assay

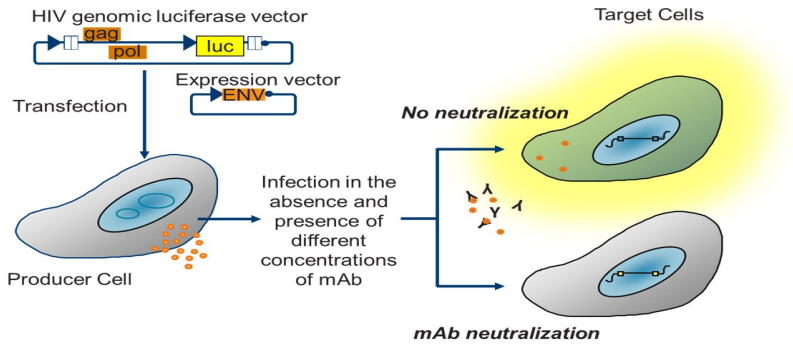
HIV genomic luciferase vector
gag
pol
luc
Expression vector
ENV
Transfection
Producer Cell
Infection in the absence and presence of different concentrations of mAb
Target Cells
No neutralization
mAb neutralization

Results

Figure 2 Screening subjects and amplification results

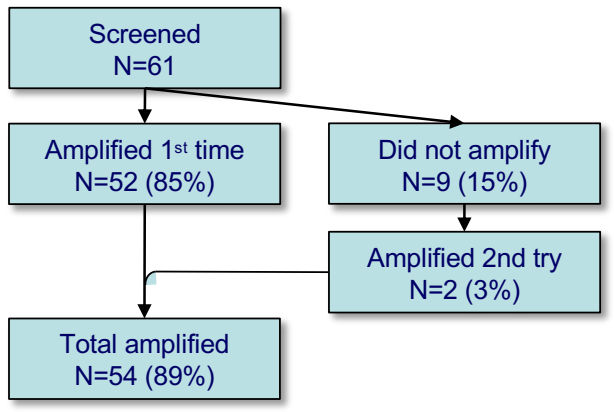
Screened
N=61
Amplified 1st time
N=52 (85%)
Did not amplify
N=9 (15%)
Amplified 2nd try
N=2 (3%)
Total amplified
N=54 (89%)

- 9 (15%) were not amplifiable on first pass, ere not amplifiable, of those 2 amplified on a 2nd attempt

Table 1. Demographic information (n=61)

Ethnicity	Hispanic	3 (5%)
Race	Caucasian	8 (13%)
	Black	53 (87%)
Gender	Male	52 (85%)
	Female	9 (15%)
Age (median, IQR)		50 (40-57)

- Demographic characteristics were not associated with sensitivity to bNAb.

Figure 3. Distribution of IC90, , cut offs and percentage of isolates with IC90 above the 10-1074 and the 3BNC117 cutoff, and spearman correlation between bNAbs susceptibility

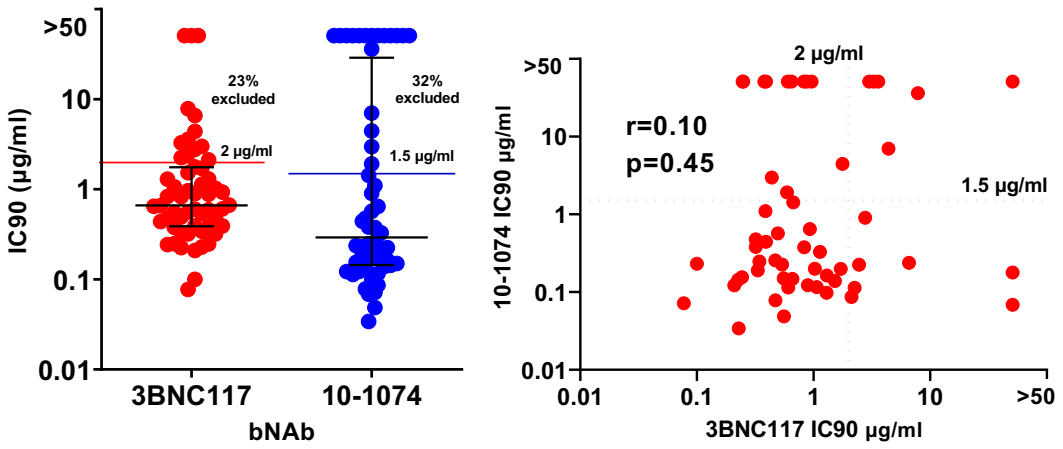
>50
10
1
0.1
0.01
IC90 (μg/ml)
23% excluded
2 μg/ml
32% excluded
1.5 μg/ml
3BNC117
10-1074
bNAb
10-1074 IC90 μg/ml
r=0.10
p=0.45
2 μg/ml
1.5 μg/ml
0.01
0.1
1
10
>50
3BNC117 IC90 μg/ml

Figure 4. Distribution of IC50, IC80, IC90, IC95 and MPI of all isolates. The MPI values identify 10-1074 resistant isolates.

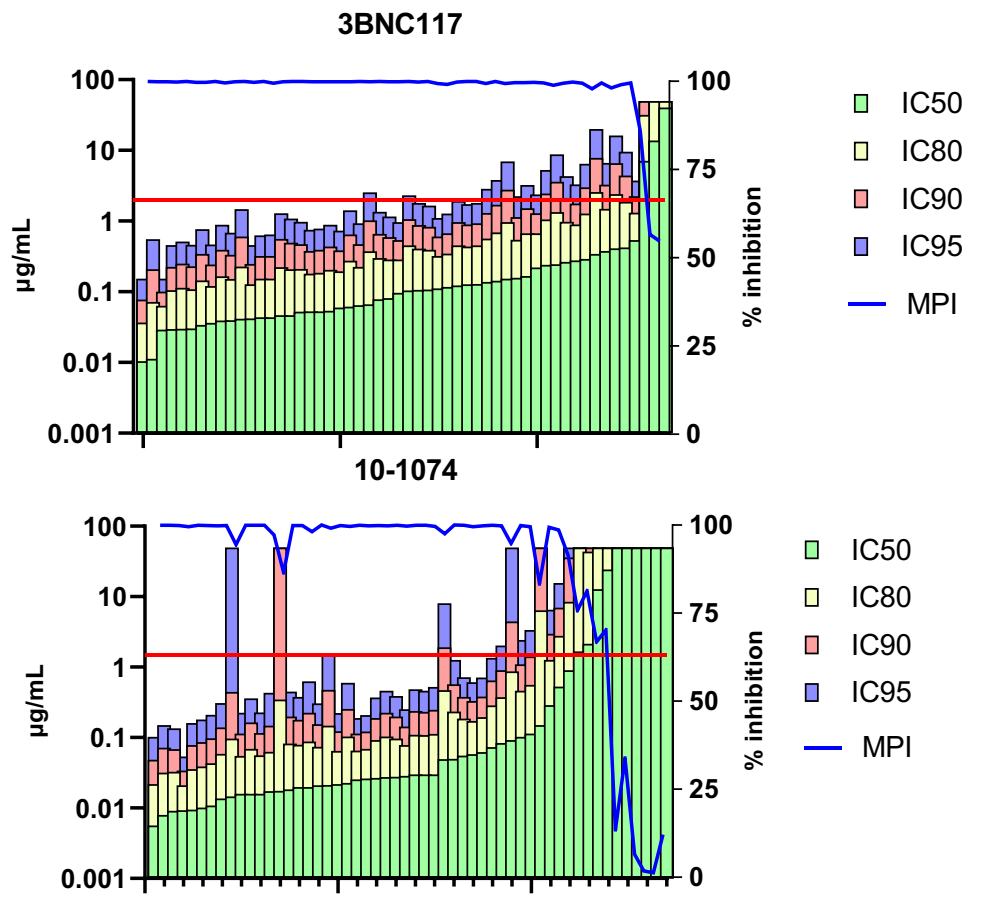
3BNC117
100
10
1
0.1
0.01
0.001
μg/mL
% inhibition
100
75
50
25
0
IC50
IC80
IC90
IC95
MPI
10-1074

Conclusions

- 15% can not be amplified initially, reduced to 11% on second try
- Preexisting resistance was common
- Only 50% of the chronically infected, virologically suppressed individuals harbored virus sensitive to both 3BNC117 and 10-1074.
- Sensitivity of the two antibodies were independent of one another
- MPI values identify virus resistant to 101-1074
- Demographic characteristics were no associated with sensitivity to bNAbs.

Interpretation

- Our data can be used in sample size calculations for the rate of screening failure when using combination bNAbs for maintenance and curative strategies.
- As the reservoir of chronically infected individuals is a representation of the circulating and transmissible virus our data suggest that the combination of 2 bNABs will not neutralize infection in approximately 50% of potentially transmissible virus.

Future Direction

Definition and validation of the clinical correlates of bNAb susceptibility thresholds for therapeutic maintenance protocols and curative treatment strategies

References

1. Petropoulos CJ, Parkin NT, Limoli KL, Lie YS, Wrin T, Huang W, Tian H, Smith D, Winslow GA, Capon DJ, Whitcomb JM. A novel phenotypic drug susceptibility assay for human immunodeficiency virus type 1.. Antimicrobial agents and chemotherapy. 2000 Apr 1;44(4):920-8.
2. Binley JM, Wrin T, Korber B, Zwick MB, Wang M, Chappey C, Stiegler G, Kunert R, Zolla-Pazner S, Katinger H, Petropoulos CJ, Burton DR. Comprehensive cross-clade neutralization analysis of a panel of anti-human immunodeficiency virus type 1 monoclonal antibodies. J Virol. 2004 Dec;78(23):13232-52. doi: 10.1128/JVI.78.23.13232-13252.2004. PMID: 15542675; PMCID: PMC524984……

Acknowledgements and contact

We would like to thank the study participants and their providers.

Support:

NIAID grant U01AI110434 to LJ Montaner, the BEAT-HIV Delaney Collaboratory UM1AI126620, co-funded by NIAID, NIMH, NINDS, and NIDA; Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA; Philadelphia FIGHT; Pennsylvania Department of Health; The The Philadelphia Foundation (Robert I. Jacobs Fund); the Penn Center for AIDS Research (P30 AI 045008); The Wistar Cancer Center Support Grant (P30 CA10815). Dr. Luis Montaner is supported by the Herbert Kean, M.D., Family Professorship

Luis J. Montaner, D.V.M., M.Sc., D.Phil.,
Director of HIV-1 Immunopathogenesis Laboratory,
The Wistar Institute, montaner@wistar.org