DURABLE HUMAN HIV-SPECIFIC bnAb PRODUCTION 3 YEARS AFTER AAV8 MEDIATED GENE TRANSFER

00498

JP Casazza[1], EM Cale[1], SR Narpala[1], L Novik[1], G Yamshchikov[1], BC Lin[1], JP Pandey[2], AB McDermott[1], MR Roederer[1], AB Balazs[3], D Baltimore[4], RA Koup[1], JE Ledgerwood[1], JR Mascola[1] and the VRC603 Team[1]

[1]Vaccine Research Center/NIAID/NIH, Bethesda, MD, USA, [2]Department of Microbiology and Immunology, Medical University of South Carolina, Charleston, SC, USA, [3]Ragon Institute of MGH, MIT and Harvard, Cambridge, MA, USA; [4]Division of Biology and Biological Engineering, California Institute of Technology, Pasadena, CA, USA

BACKGROUND .

- Although multiple sites of viral vulnerability are now identified, the unusual features of antibodies that recognize those sites make the design of immunogens capable of eliciting bnAbs difficult
- Repeated injections of bnAbs can maintain Abs at a protective concentration.
- Gene transfer protocols offer an alternative to repeated injections of bNAbs as a means of maintaining effective immunoprophylaxis. VRC07 is a bnAb targeting the CD4 binding site of the HIV-1 envelope glycoprotein.

METHODS

- An AAV8-VRC07 vector (Fig. 1) using a single open reading frame design packaged between two inverted terminal repeats (ITR) with a CASI promoter, a truncated woodchuck hepatitis post-translational regulatory element (WPRE) and a single SV40 poly A sequence (SV40pA) was used to carry the nucleotide sequence for VRC07 into HIV-infected trial participants with VL<50 copies/ ml. AAV transgenes persist in the nucleus as stable episomal DNA.
- 8 participants were given either $5x10^{10}$, $5x10^{11}$ or $2.5x10^{12}$ vector genomes/kg AAV8-VRC07 by IM injection with VRC07 concentrations measured between 1-3 years after IM injection of AAV8-VRC07.
- *In vitro* produced VRC07 was purified from ARVs and then pseudoviral neutralizing assays performed to assess biologic activity.

Vector Construction

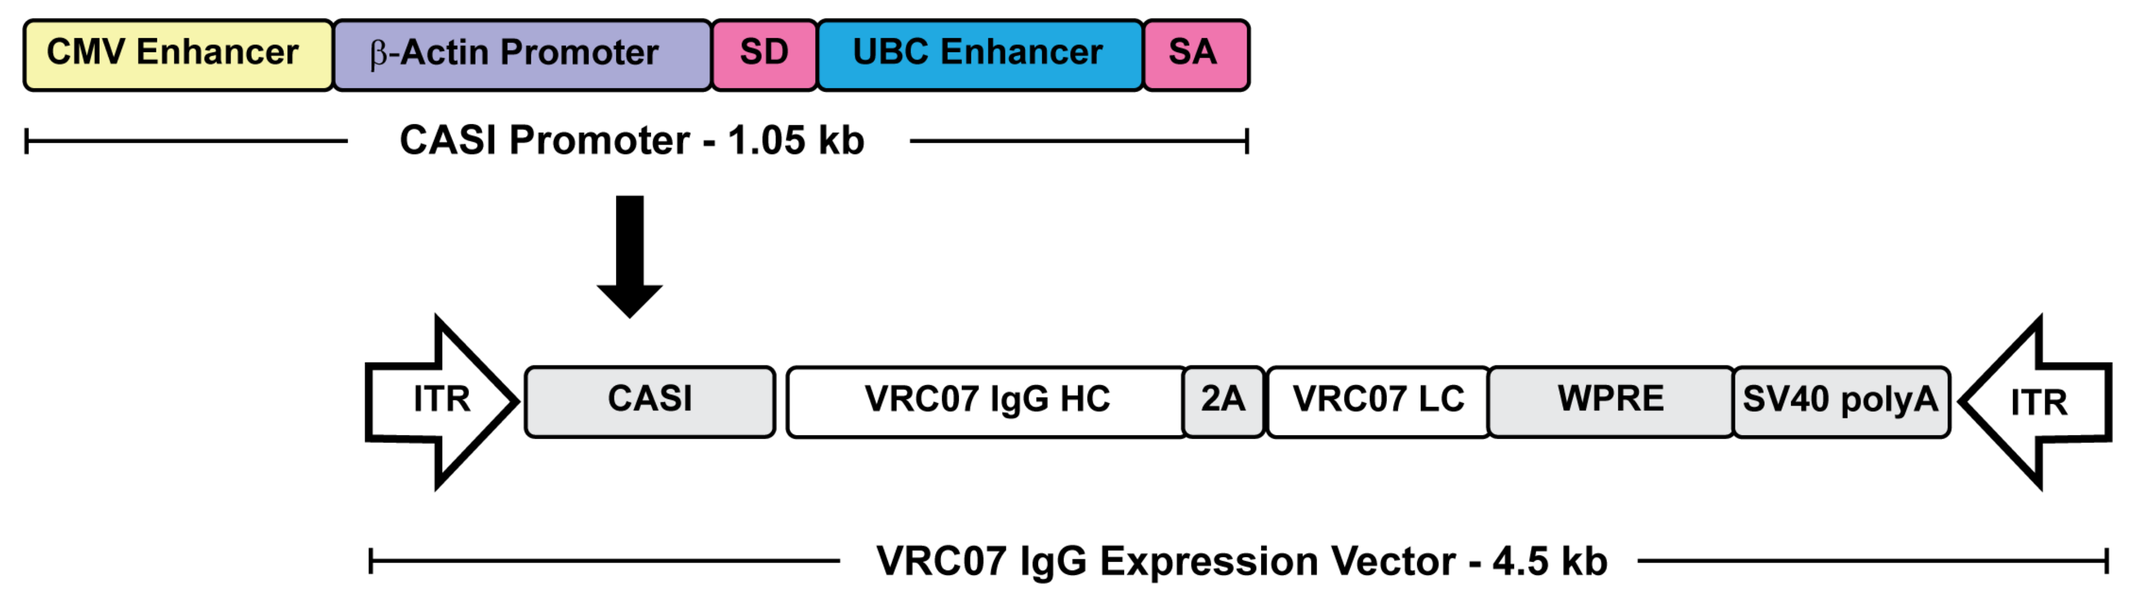
CMV Enhancer
β-Actin Promoter
SD
UBC Enhancer
SA
CASI Promoter - 1.05 kb
ITR
CASI
VRC07 IgG HC
2A
VRC07 LC
WPRE
SV40 polyA
ITR
VRC07 IgG Expression Vector - 4.5 kb

Fig 1. Schematic Representation of VRC07-AAV8

- AAV8 VRC07 is an adeno-associated gene transfer vector coding for the bNAb VRC07. It contains a bicistronic cassette that utilizes a self-processing F2A insert which allows the production of the light and heavy IgGs from a single transcribed mRNA (Fig 1).
- AAV8 VRC07 is safe and well tolerated in HIV-infected individuals.
- ADA responses were observed in 3 of 8 subjects. In individuals without dectable ADAs, VRC07 production was durable produced being measurable for up to 3 years after administration by IM injection. Three participants in this study had VRC07 concentrations >1µg/ml, one achieved a concentration of >3µg/ml (Fig 3).
- *In vivo* and *ex vivo* produced VRC07 had similar neutralization ID50s (Fig 4)

RESULTS

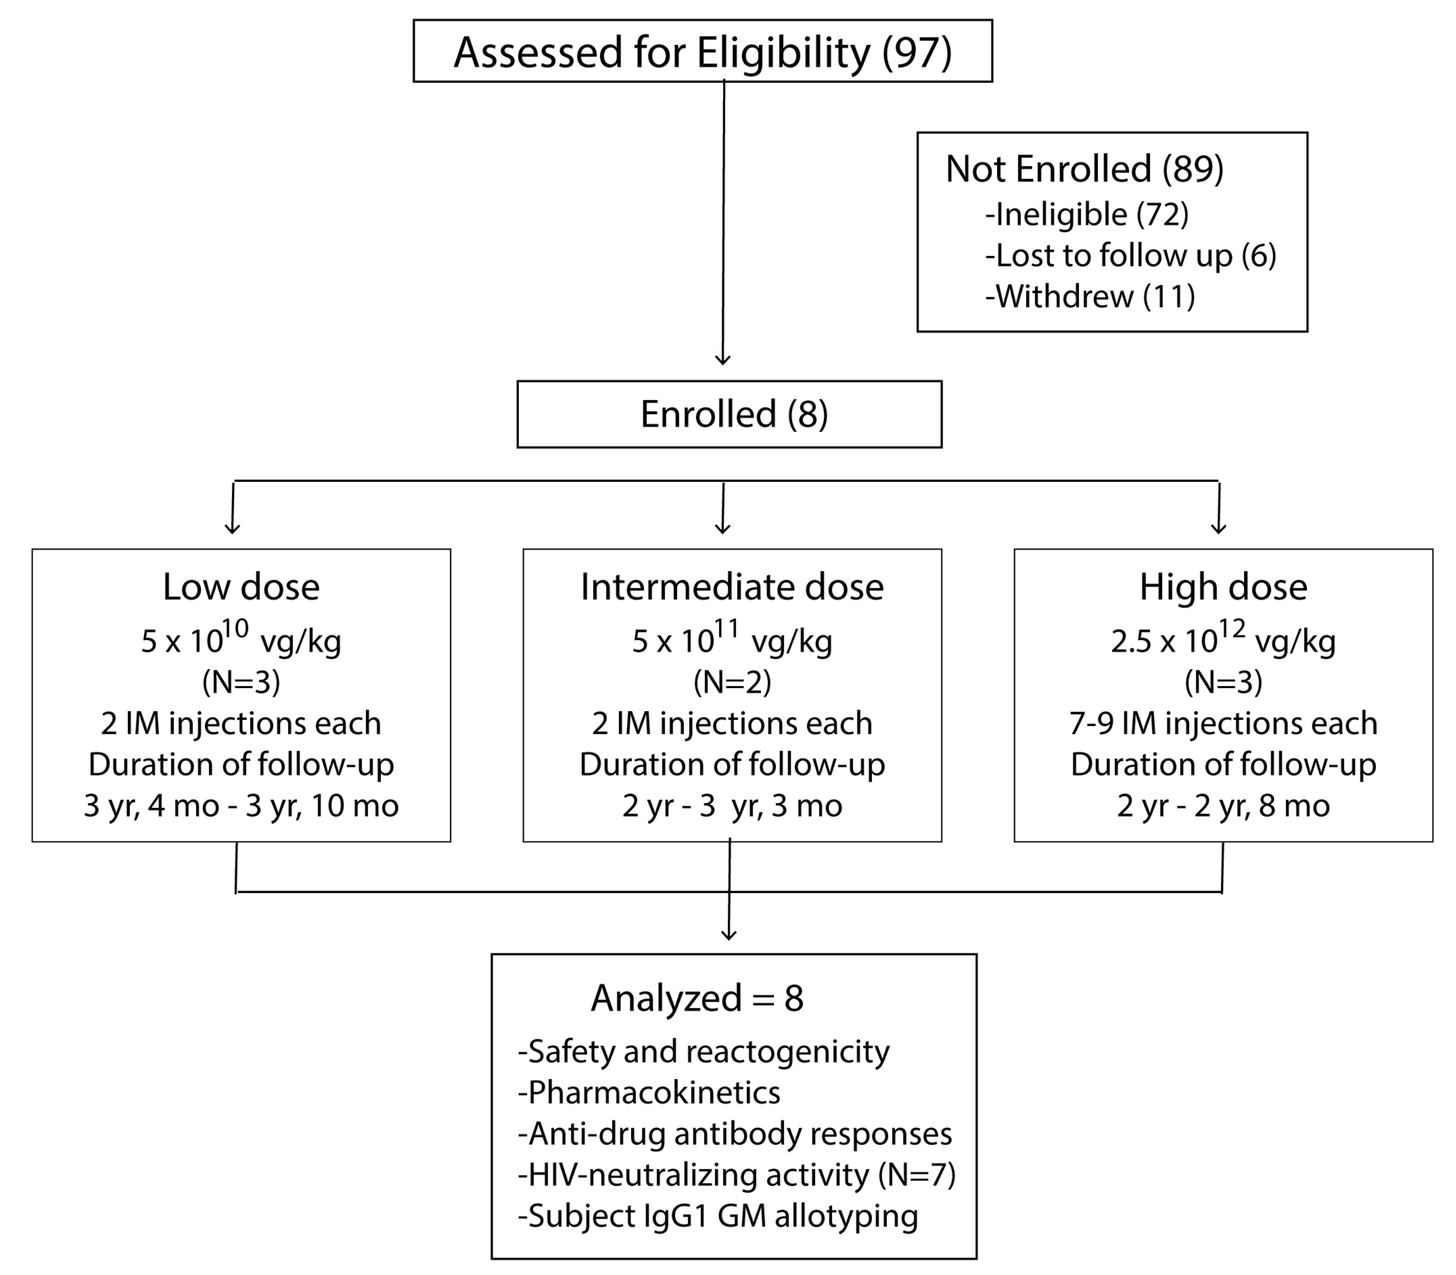
Assessed for Eligibility (97)
Not Enrolled (89)
-Ineligible (72)
-Lost to follow up (6)
-Withdrew (11)
Enrolled (8)
Low dose
5 x 10^10 vg/kg
(N=3)
2 IM injections each
Duration of follow-up
3 yr, 4 mo - 3 yr, 10 mo
Intermediate dose
5 x 10^11 vg/kg
(N=2)
2 IM injections each
Duration of follow-up
2 yr - 3 yr, 3 mo
High dose
2.5 x 10^12 vg/kg
(N=3)
7-9 IM injections each
Duration of follow-up
2 yr - 2 yr, 8 mo
Analyzed = 8
-Safety and reactogenicity
-Pharmacokinetics
-Anti-drug antibody responses
-HIV-neutralizing activity (N=7)
-Subject IgG1 GM allotyping

CONSORT DIAGRAM

Demographic characteristics of study participants

Category	Subcategory	Group 1 (N=3)	Group 2 (N=2)	Group 3 (N=3)	Overall (N=8)
		N (%)			
Gender	Male	3 (100)	2 (100)	1 (33)	6 (75)
	Female	0 (0)	0 (0)	2 (66)	2 (25)
Race	Asian	0 (0)	0 (0)	0 (0)	0 (0)
	Black/African	1 (33)	1 (50)	3 (100)	5 (62.5)
	White	2 (66.7)	1 (50)	0 (0)	3 (37.5)
Ethnicity	Non-Hispanic/Latino	3(100)	2(100)	3(100)	8(100)
	Hispanic/Latino	0(0)	0(0)	0(0)	0(0)
Median Age	Median Years (range)	56 (36-60)	41 (30-52)	52 (32-56)	52 (30-60)
Median Weight	Median Kg weight (range)	79 (66-82)	74 (73-75)	82 (71-91)	77 (66-91)

Maximum local and systemic reactogenicity up to day 7 post IM injection of AAV8-VRC07*

	$5 x 10^{10}$ (N=3)	$5 x 10^{11}$ (N=2)	$2.5 x 10^{12}$ (N-3)	Overall (N=8)
	N (%)			
Maximum local reactogenicity up to 7 days post IM injection of AAV8- VRC07*				
Pain/Tenderness				
None	3 (100)	2 (100)	0 (0)	5 (62.5)
Mild	0 (0)	0 (0)	3 (100)	3(37.5)
Swelling				
None	3 (100)	2 (100)	3 (100)	8 (100)
Mild	0 (0)	0 (0)	0 (0)	0 (0)
Redness				
None	3 (100)	2 (100)	3 (100)	8 (100)
Mild	0 (0)	0 (0)	0 (0)	0 (0)
Any local symptoms				
None	3 (100)	2 (100)	0 (0)	5 (62.5)
Mild	0 (0)	0 (0)	3 (100)	3(37.5)
Self-reported systemic reactogenicity up to 7 days post IM injection of AAV8- VRC07*				
Malaise				
None	3 (100)	2 (100)	3 (100)	8 (100)
Mild	0 (0)	0 (0)	0 (0)	0 (0)
Myalgia				
None	0 (0)	1 (50)	2 (66.7)	6 (75)
Mild	0 (0)	1 (50)	1 (33.3)	2 (25
Headache				
None	3 (100)	2 (100)	2 (66.7)	7 (87.5)
Mild	0 (0)	0 (0)	1 (33.3)	1 (12.5)
Chills				
None	3 (100)	2 (100)	3 (100)	8 (100)
Mild	0 (0)	0 (0)	0 (0)	0 (0)
Nausea				
None	3 (100)	2 (100)	3 (100)	8 (100)
Mild	0 (0)	0 (0)	0 (0)	0 (0)
Temperature Elevation				
None	3 (100)	2 (100)	3 (100)	8 (100)
Mild	0 (0)	0 (0)	0 (0)	0 (0)
Joint Pain				
None	3 (100)	2 (100)	3 (100)	8 (100)
Mild	0 (0)	0 (0)	0 (0)	0 (0)
Any systemic symptoms				
None	3 (100)	1 (50)	2 (66.7)	6 (75)
Mild	0 (0)	1 (50)	1 (33.3)	2 (25)

* There were no moderate or severe reactogenicity throughout the trial

AAV8 VRC08 DNA is rapidly cleared from the plasma

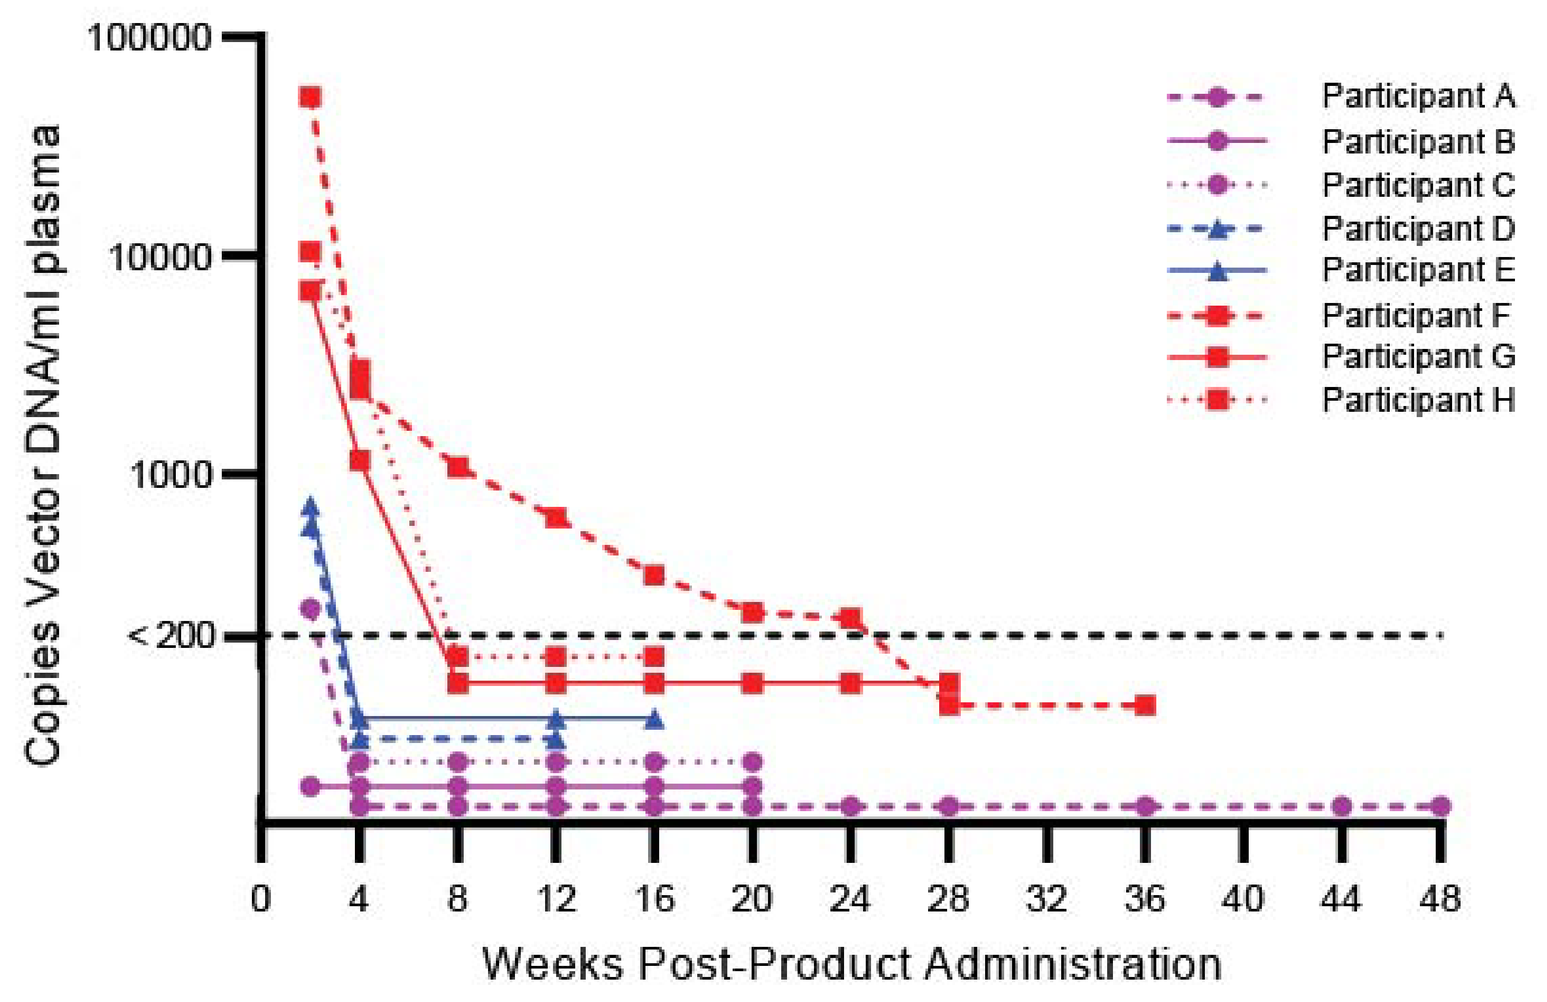
Copies Vector DNA/ml plasma
100000
10000
1000
< 200
Participant A
Participant B
Participant C
Participant D
Participant E
Participant F
Participant G
Participant H
0 4 8 12 16 20 24 28 32 36 40 44 48
Weeks Post-Product Administration

Fig 2. Longitudinal AAV8 VRC07 Plasma vector DNA. Plasma AAV8 VRC07 measured by PCR after extraction from sera. Concentration of product given is denoted by symbol shape and color: $5x10^{10}$ (●), 5 x 10^{11} (▲), and $2.5x10^{12}$ (■) vg/kg.

VRC07 persists in participants without ADA

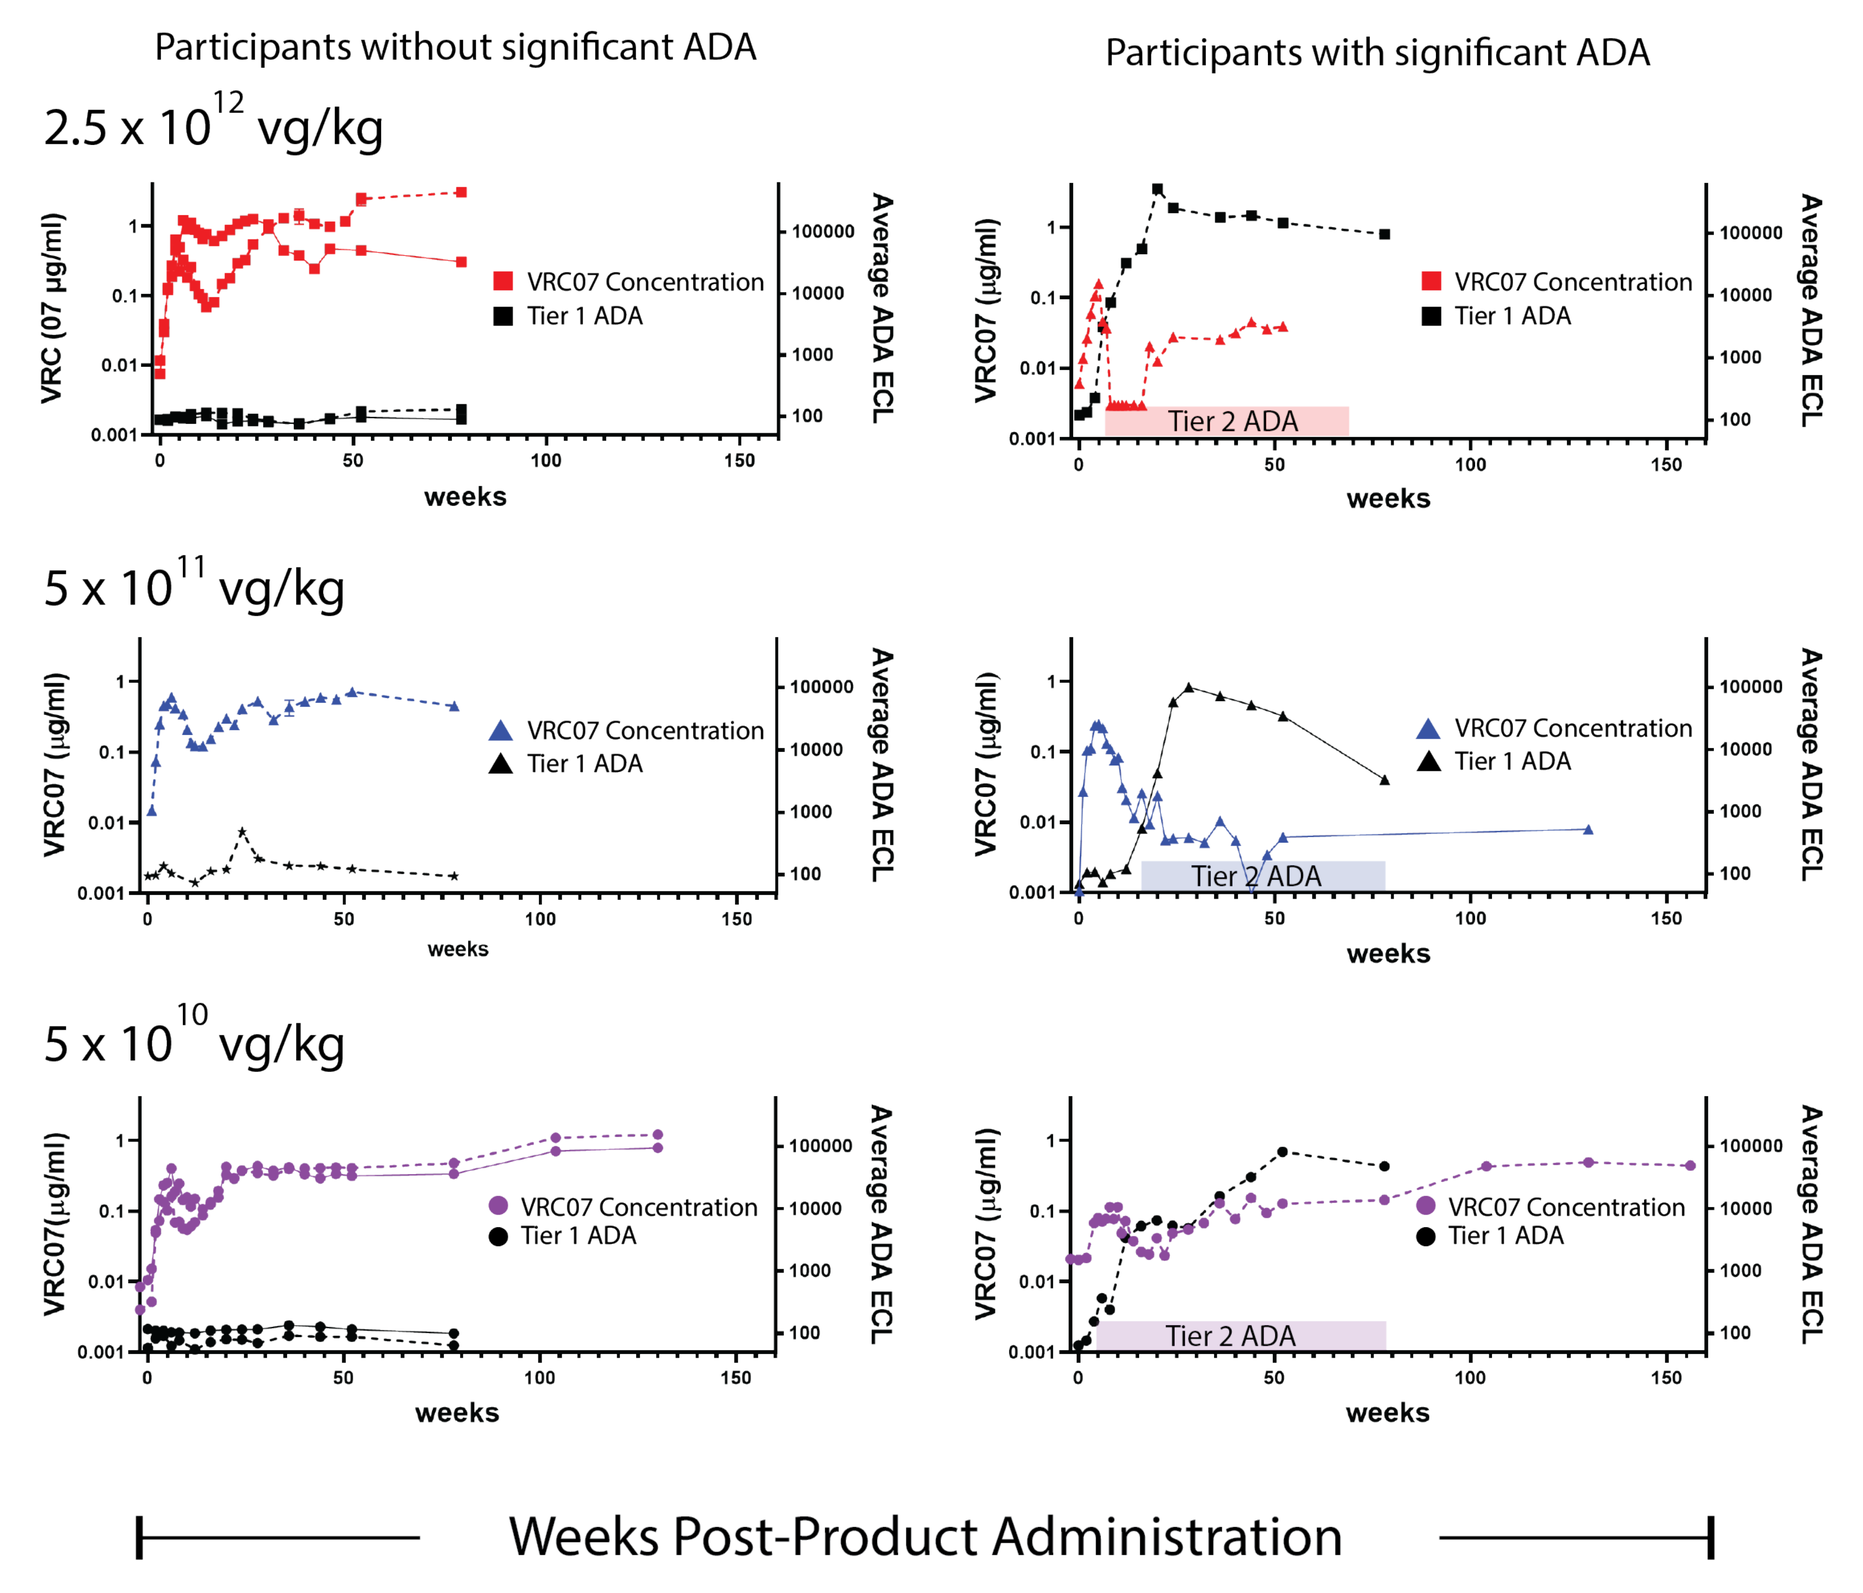
Participants without significant ADA
Participants with significant ADA
2.5 x 10^12 vg/kg
5 x 10^11 vg/kg
5 x 10^10 vg/kg
VRC07 Concentration
Tier 1 ADA
Tier 2 ADA
Average ADA ECL
weeks
Weeks Post-Product Administration

Fig 3. Longitudinal serum VRC07 concentration and tier 1 ADA (measured by binding of serum to *ex vivo* produced membrane bound VRC07) confirm by *ex vivo* VRC07 competition assays shown by solid-colored bars (tier 2 ADA). VRC07 and ADA were measured from immediately prior to IM injection of AAV8-VRC07 to between 80-156 weeks after IM injection of product. Concentration of product administer is denoted by VRC07 concentration marker color. Subjects within each dose group are denoted by line pattern for both VRC07 concentration and ADA. ADA is represented by black lines

Neutralization activity of in vitro produced VRC07 was similar to *ex vivo* produced VRC07

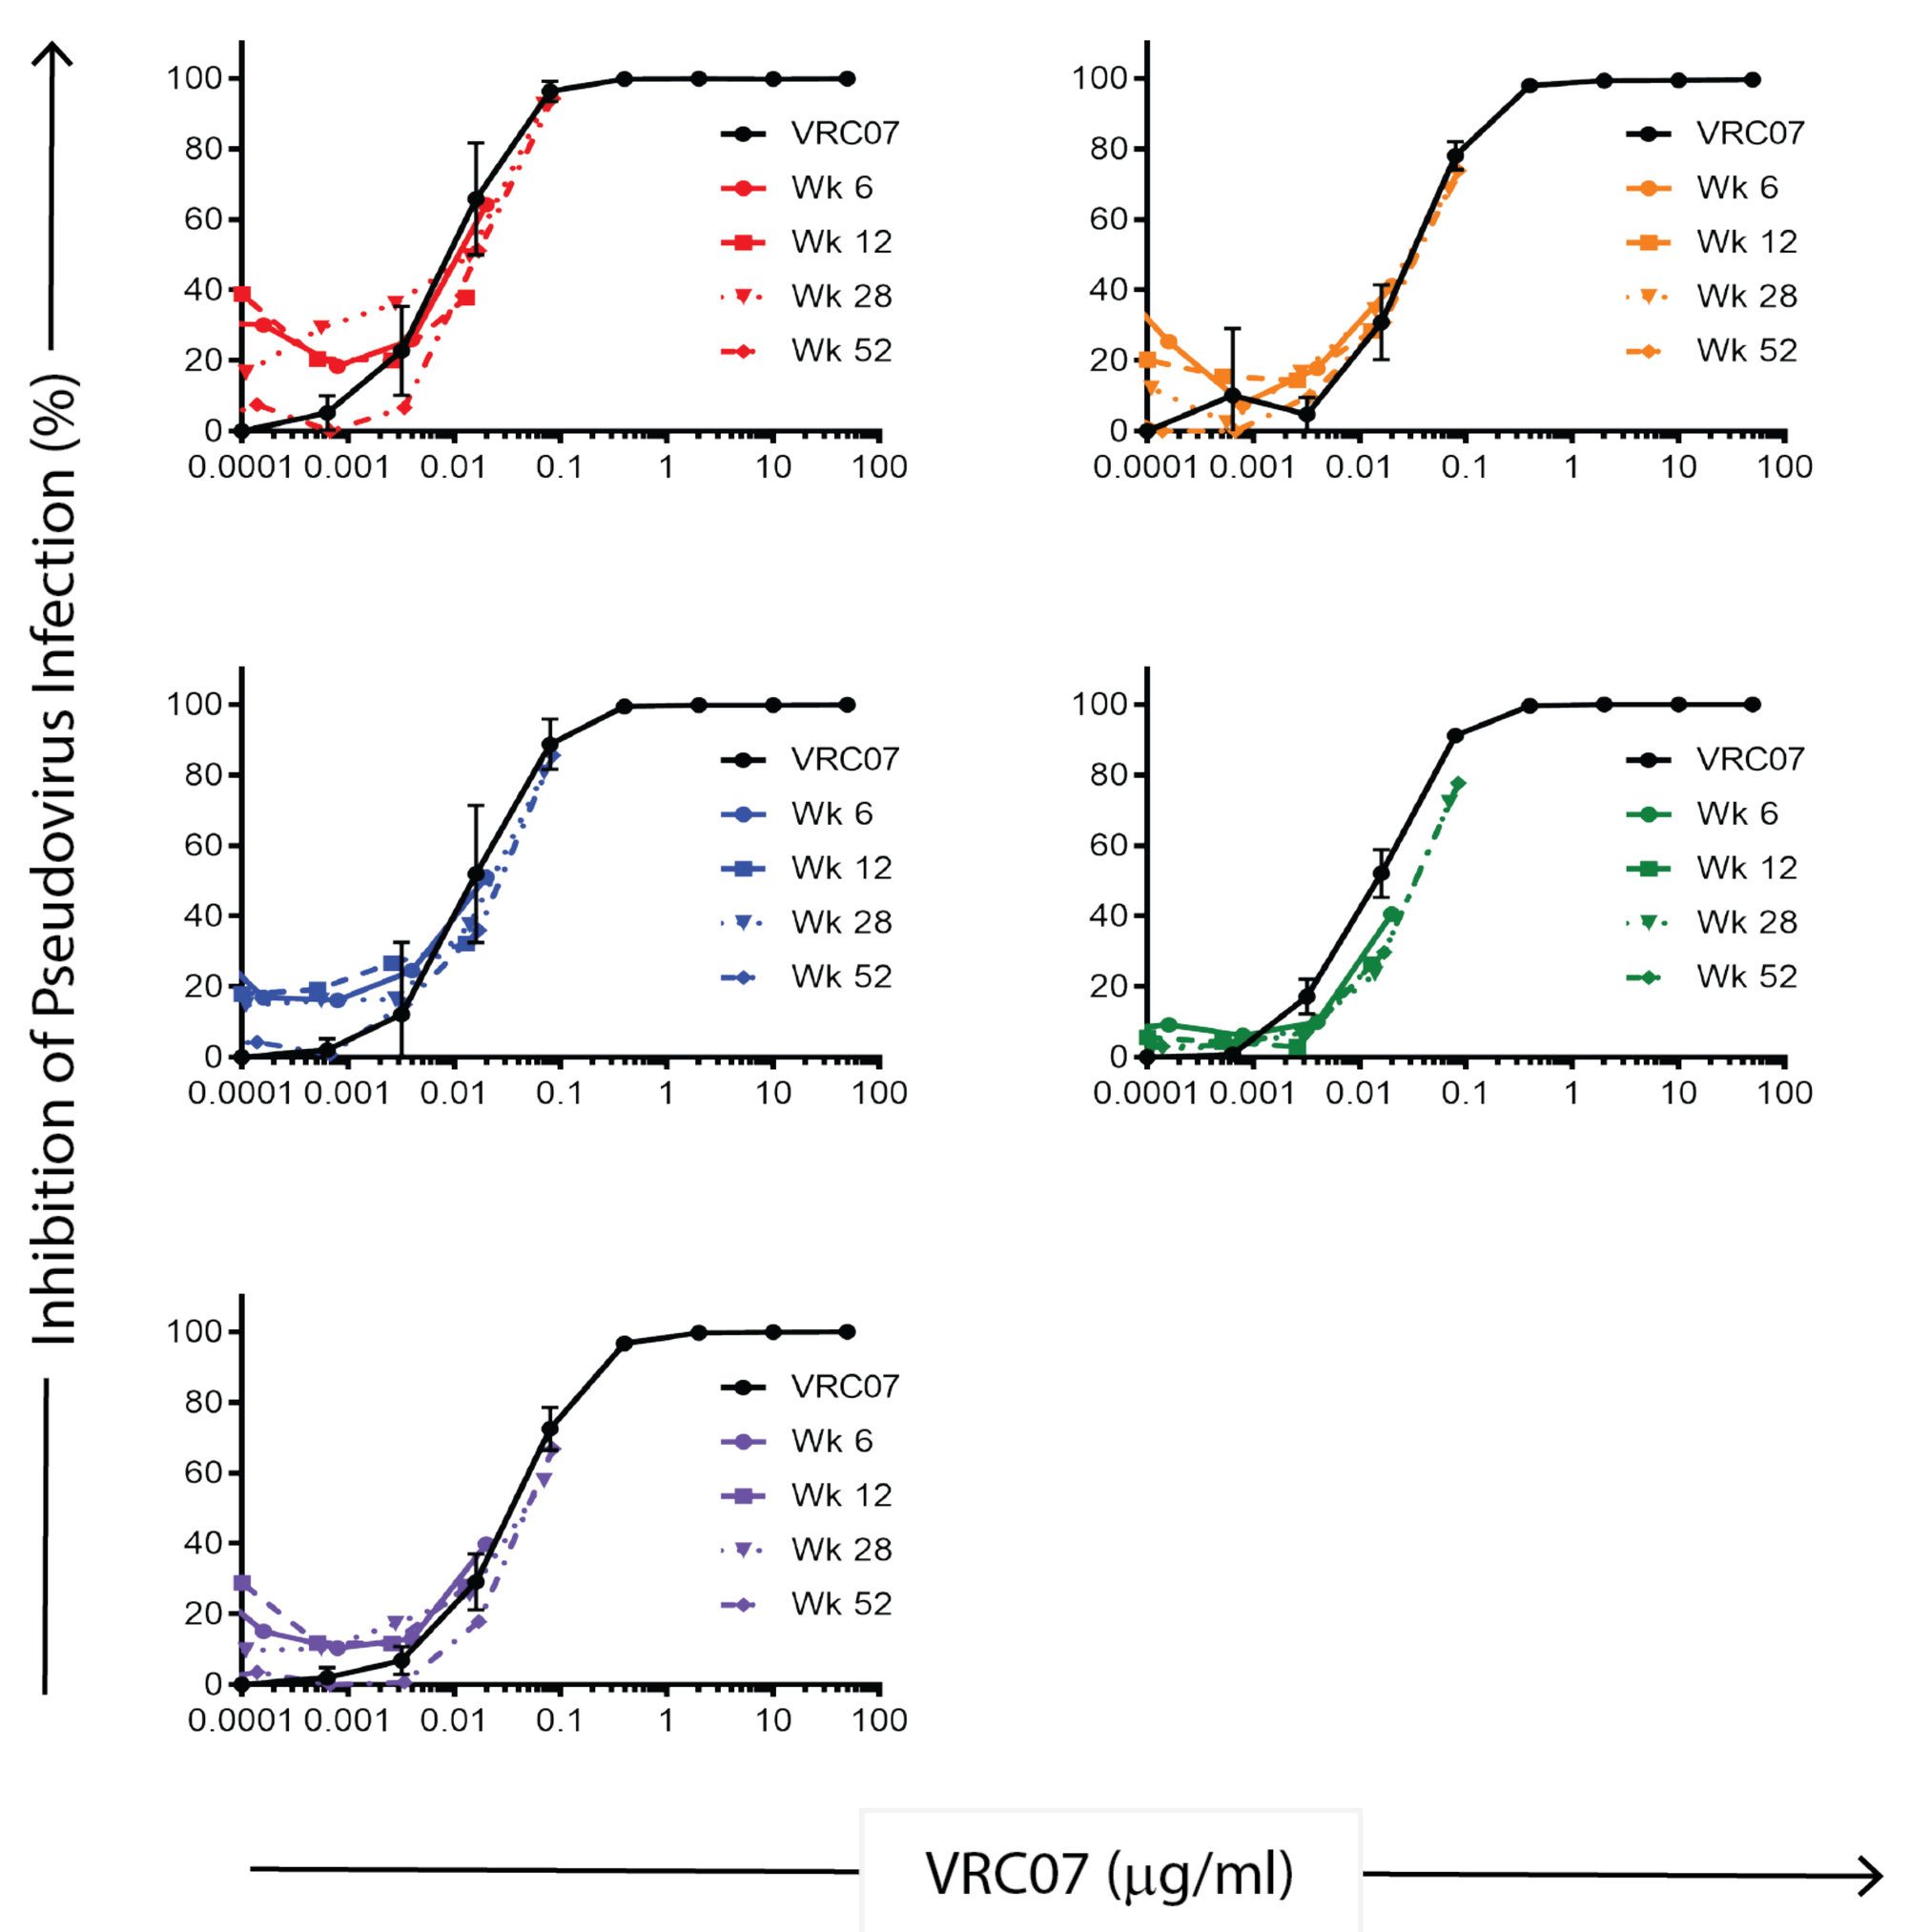
Inhibition of Pseudovirus Infection (%)
VRC07
Wk 6
Wk 12
Wk 28
Wk 52
0.0001 0.001 0.01 0.1 1 10 100
VRC07 (µg/ml)

Fig 4. Longitudinal comparison of *in vitro* and *ex vivo* produced VRC07 pseudoviral neutralization curves. Neutralization curves for strains 001428 (--), Q842 (--), T33-7 (--), JRFLIB (--), and TZBD (--) adjusted for the concentration of VRC07 in purified IgG serum samples are shown for a study participant at 0, 6, 12, 24 and 52 weeks after IM injection of AAV8-VRC07. Neutralization curves for purified *in vitro* produced VRC07 for each pseudoviral species are overlaid on each plot (-●-). Data points for neutralization assays of *ex vivo* produced VRC07 represent the average of 6 replicate assays, error bars are + SD. Data points for *in vitro* produced VRC07 represent a single assay, These data are representative of data from six additional participants.

Geometric means + 2 SD for *ex vivo* and *in vivo* produced VRC07

Pseudovirus	Geometric mean	-2 SD	+ 2 SD
ID50s for *ex vivo* produce VRC07 (µg/ml)			
001428	0.0093	0.0032	0.0271
JRFLJB	0.0147	0.0038	0.0570
Q842	0.0304	0.0147	0.0632
T33-7	0.0146	0.0086	0.0247
TZBD	0.0355	0.0194	0.0647
ID50s from purified IgGs from all (N=7) *in vivo* produced VRC07 (µg/ml)			
001428	0.0081	0.0018	0.0364
JRFLJB	0.0128	0.0026	0.0641
Q842	0.0193	0.0026	0.144
T33-7	0.0375	0.027	0.0521
TZBD	0.0357	0.0178	0.0711

CONCLUSIONS

- Administration of AAV8 VRC07 by IM injection is safe and well tolerated.
- In participants without measurable ADA, VRC07 production was durable with a concentration of >1µg/ml achieved in 3 participant and >3 µg/ml in one participant.
- Although challenges remain, these data provide proof-of-concept that an AAV vector can be used to induce the durable *in vivo* production of difficult-to-elicit antibodies that retain their neutralizing potency.

ACKNOWLEDGEMENT

- We wish to express our thanks to the volunteers who are participating in VRC 603. Without their continued participation this trial would not be possible.