PREDICTORS OF VIRAL SUPPRESSION FOLLOWING ENHANCED ADHERENCE COUNSELING: VISEND TRIAL

00490

Danielle C.M Engamba[1,2,6], C. William Wester[3,4], Melody Mbewe[1], Alepalli Praven Kumar [1], Bob Chirwa[1], Getrude Phiri [1], Edford Sinkala[1,2], Sombo Fwoloshi[1,2,5], Aggrey Mweemba [6], Davies Kampamba[1,5], Nyuma Mbewe[1,2,6], Mpanji Siwingwa[1,2], Andrew Hill [7], Lloyd Mulenga[1,2,3,4,5], Lameck Chirwa[1,2], The VISEND Study Team

[1]University Teaching Hospital, Adult Infectious Diseases Center, Lusaka, Zambia [2]University of Zambia, School of Medicine, Division of Infectious Diseases, Internal Medicine, Lusaka, Zambia, [3]Vanderbilt University Medical Center (VUMC), Department of Medicine, Division of Infectious Diseases, Nashville, TN, USA, [4]Vanderbilt Institute for Global Health (VIGH), Nashville, TN, USA, [5]Ministry of Health, Ndeke House, Lusaka, Zambia, [6] Levy Mwanawasa University Teaching Hospital, [7]Liverpool University, Department of Translational Medicine, Liverpool.

BACKGROUND

The WHO recommends enhanced adherence counselling (EAC) before regimen switch for all HIV-positive individuals on ART with non-suppressed viral loads (VL)[1].

However, there is a paucity of data, especially within a clinical trial setting, on the determinants of viral load suppression (VLS) following EAC among HIV positive adults failing ART. We thus evaluated predictors of VLS among Zambian HIV positive adults failing Integrase Strand Transfer Inhibitor (INSTI) or Protease Inhibitor (PI) based ART who had undergone EAC in the VISEND clinical trial.

METHODS

The VISEND clinical trial is a randomized 144-week open label non-inferiority study with HIV positive Zambian adults with VL < 1000 copies/mL (Arm A) or VL ≥ 1000 copies/mL (Arm B) on ART of tenofovir disoproxil fumarate (TDF), lamivudine (3TC) plus efavirenz (EFV) or nevirapine (NVP), switched to 1) TDF,3TC,DTG **[TLD]** 2) tenofovir alafenamide (TAF), emtricitabine (FTC) plus DTG [**TAFED]** 3) zidovudine (ZDV), 3TC, lopinavir/ritonavir (LPV/r) or 4) zidovudine (ZDV), 3TC, atazanavir/r (ATV/r). HIV treatment failure was defined as VL ≥ 1000 copies/mL after three sessions of EAC.

VLs and other biomarkers were collected at weeks 12, 24, 48, 72, 96 and 144. Adults with VL ≥ 1000 copies/mL at each of these time points underwent EAC involving 3 sessions over a period of 3 months according to existing guidelines. We calculated proportions of individuals who achieved VLS post EAC and analyzed factors (demographic and clinical) independently associated with VLS post EAC. Using multivariable log regression models, associations were analyzed as crude risk ratios (CRR) and adjusted risk ratios (ARR).

Figure 1: Enhanced Adherence Counselling (EAC) Procedure in the VISEND Clinical Trial

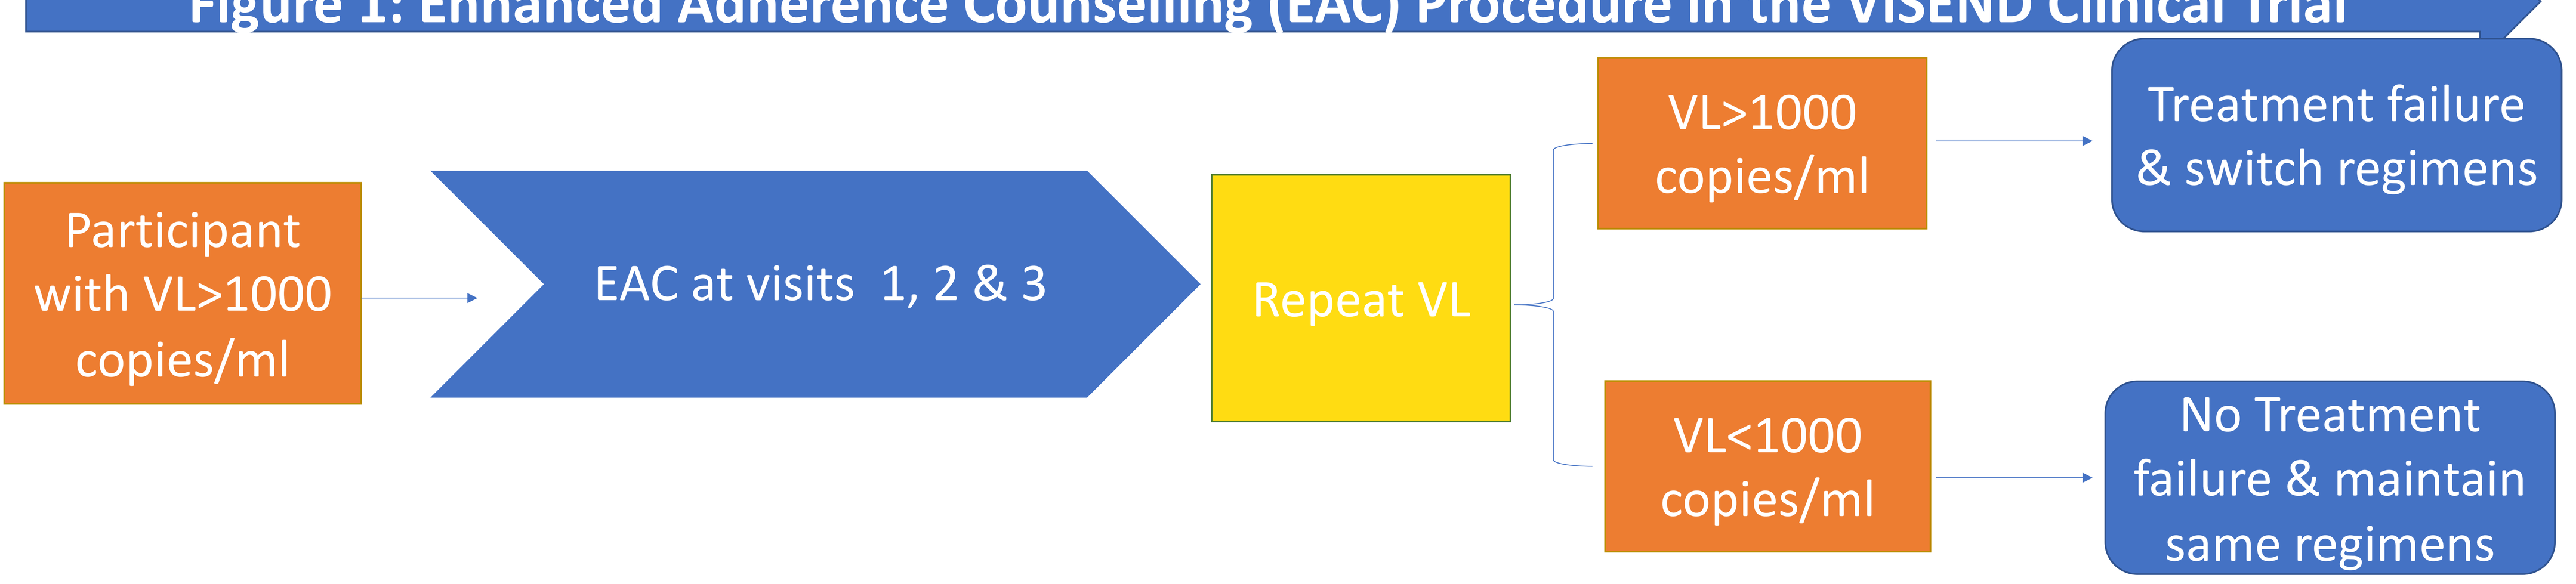
Participant with VL>1000 copies/ml
EAC at visits 1, 2 & 3
Repeat VL
VL>1000 copies/ml
Treatment failure & switch regimens
VL<1000 copies/ml
No Treatment failure & maintain same regimens

RESULTS

Figure 2: Participant Recruitment

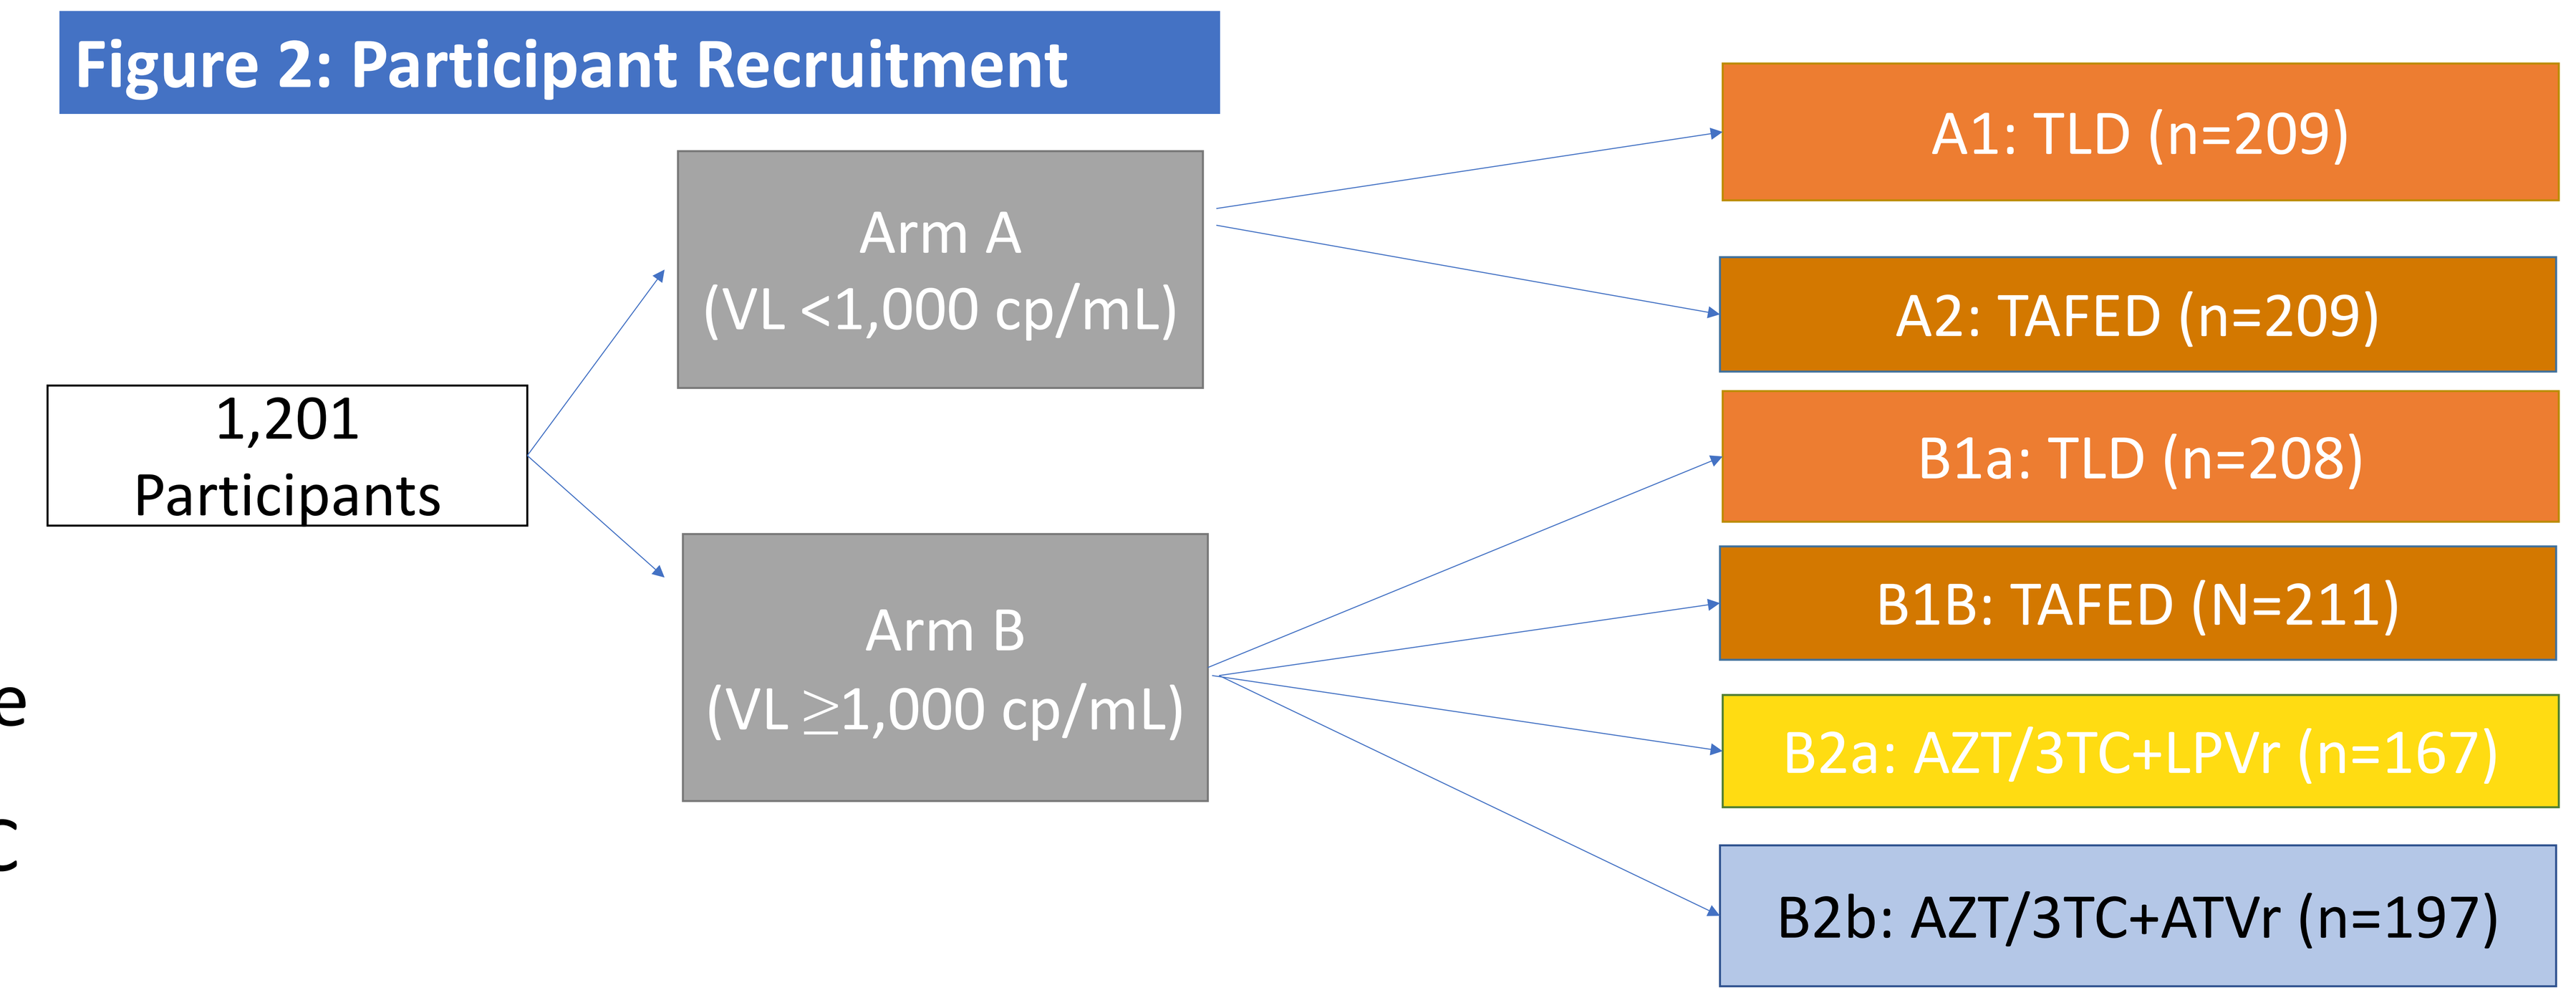
1,201 Participants
Arm A (VL <1,000 cp/mL)
Arm B (VL ≥1,000 cp/mL)
A1: TLD (n=209)
A2: TAFED (n=209)
B1a: TLD (n=208)
B1B: TAFED (N=211)
B2a: AZT/3TC+LPVr (n=167)
B2b: AZT/3TC+ATVr (n=197)

Figure 3: Distribution of participants with VL ≥1,000 cp/mL)

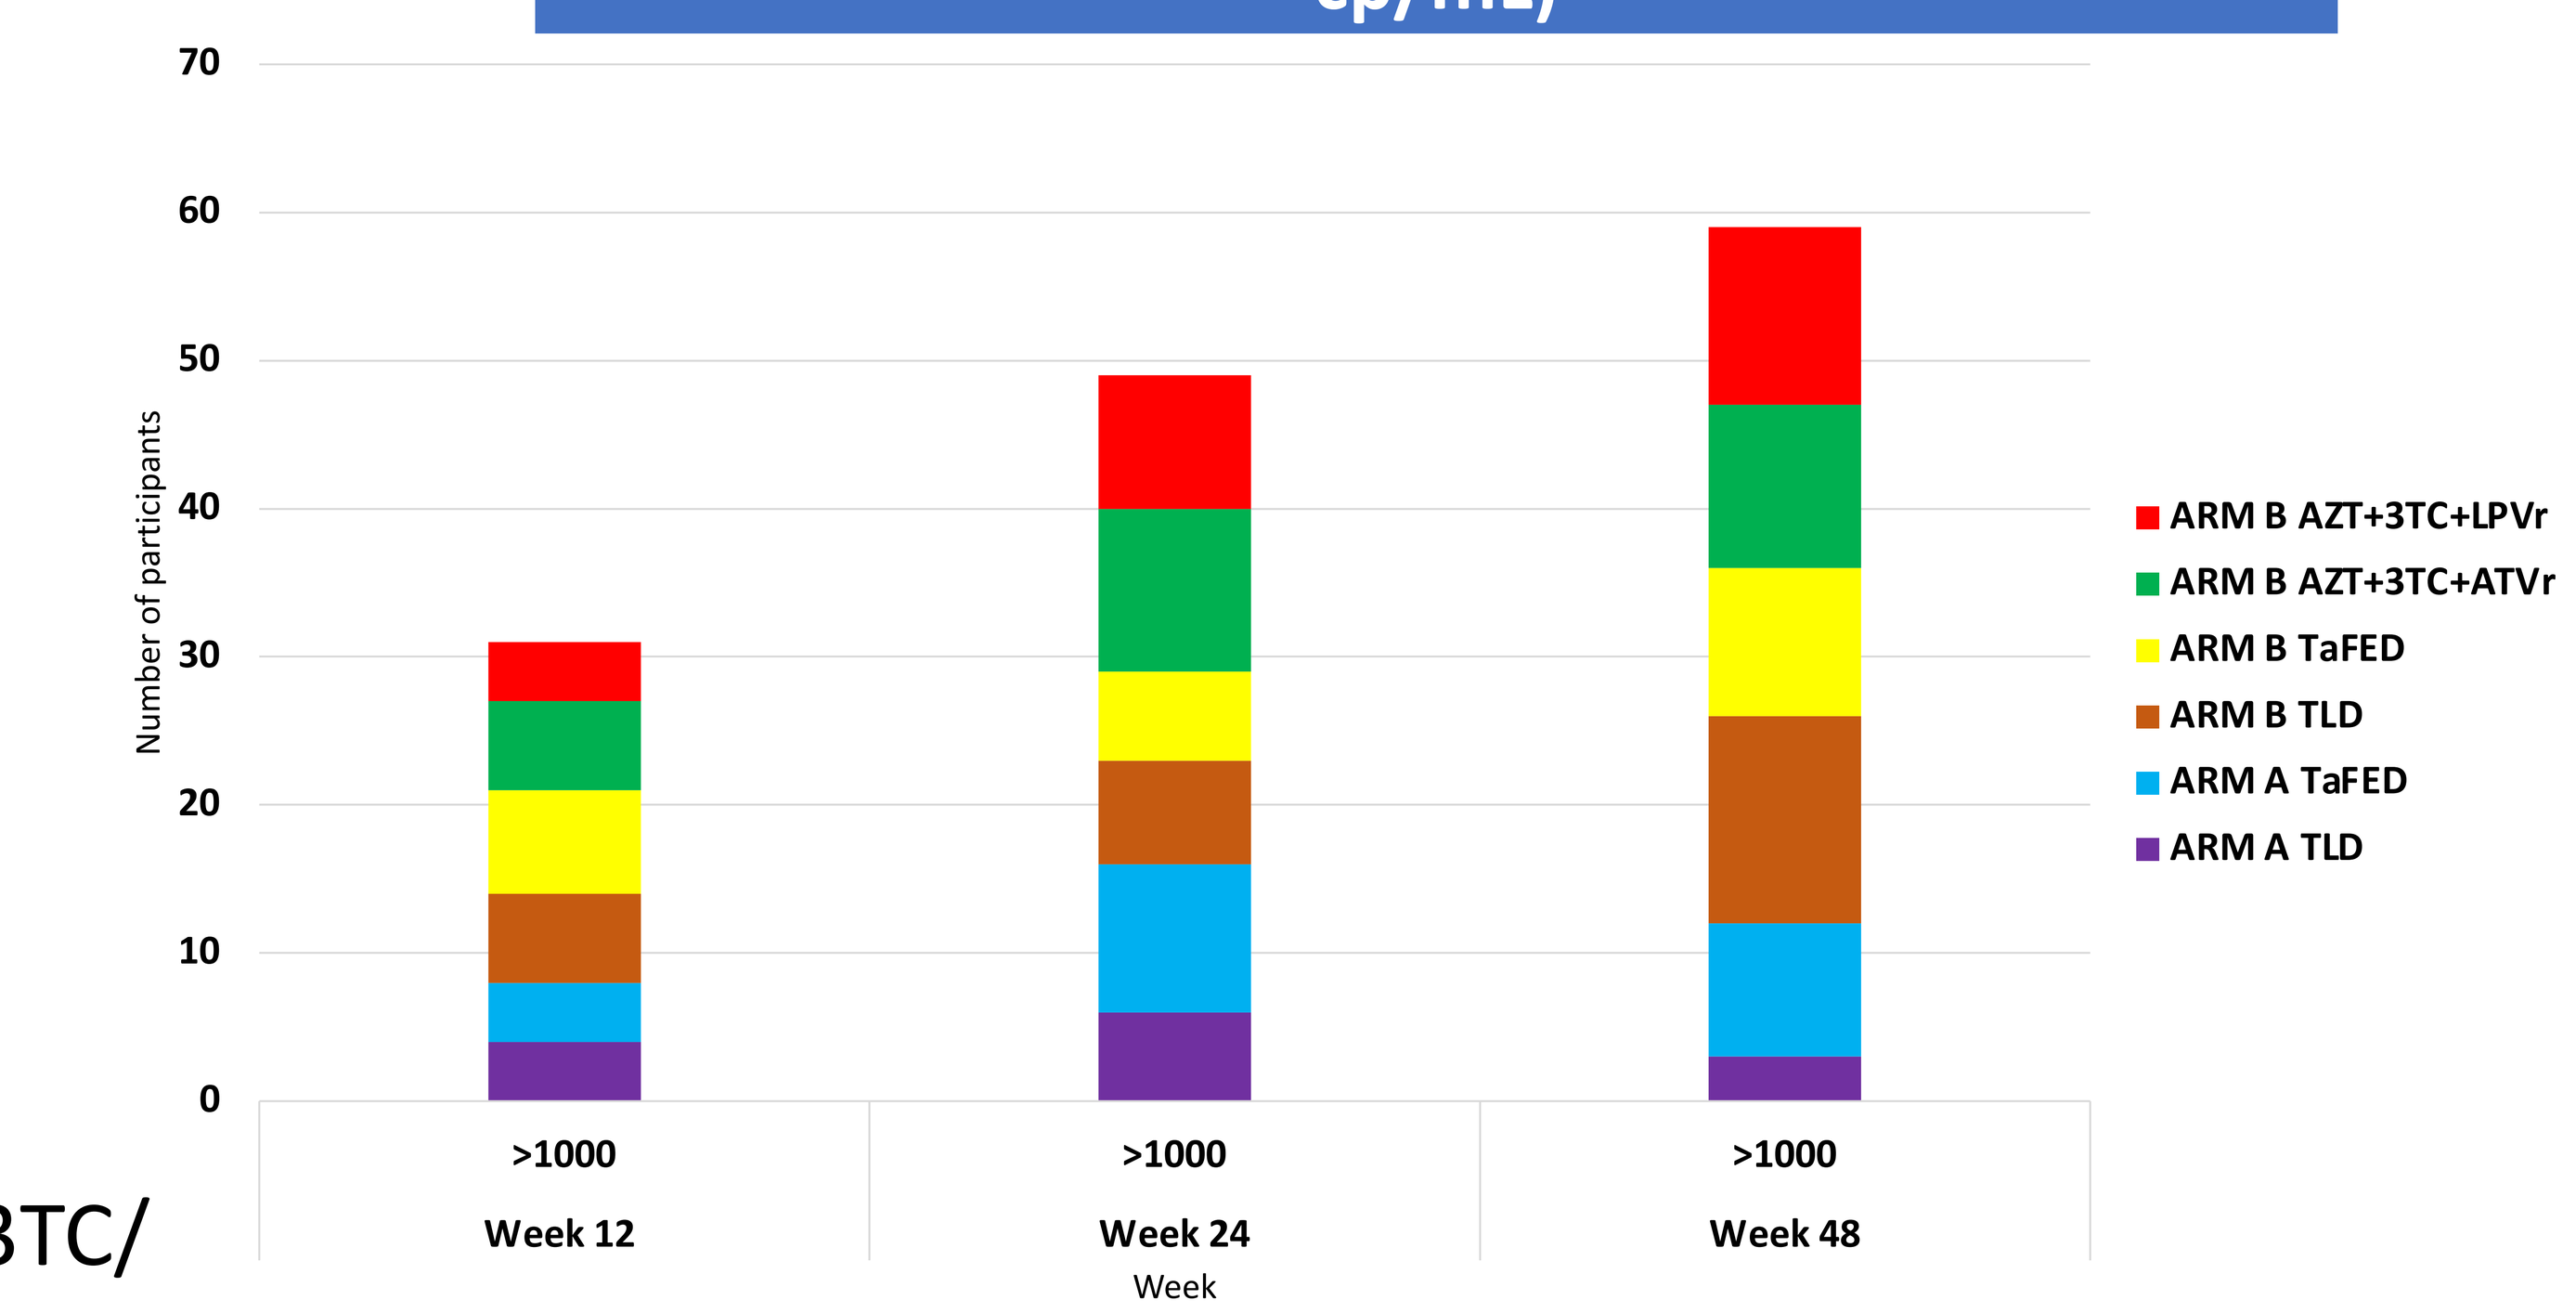
70
60
50
40
30
20
10
0
Number of participants
>1000
Week 12
>1000
Week 24
>1000
Week 48
Week
ARM B AZT+3TC+LPVr
ARM B AZT+3TC+ATVr
ARM B TaFED
ARM B TLD
ARM A TaFED
ARM A TLD

- The overall VLS post EAC was 66% distributed as 78% on TAF/ FTC/DTG , 71% on TDF/3TC/ DTG, 62% on ZDV/3TC/ATV/r and 53% on ZDV/3TC/LPV/r.

Table 1: Factors Associated With Viral Load Suppression Post EAC

Variable	CRR (95% CI)	ARR (95% CI)	p-value
Sex			
Male	1	1	
Female	1.48 [1.05 – 2.09] *	3.49 [2.17 – 5.62]	**<0.001**
Age			
18 – 24	1	-	
25 – 34	1.75 [0.73– 4.17]	-	
35 – 59	2.14 [0.95 – 4.85]	-	
≥ 60	2.0 [0.65 – 6.20]	-	
Educational level			
No formal education	1	1	
Primary	1.48 [0.55 – 4.04]	1.55 [1.32 – 1.81]	**<0.001**
Secondary	1.16 [0.42 – 3.18]	1.93 [1.65 – 2.27]	**<0.001**
Tertiary	1.27 [0.43 – 3.74]		
Duration on ART (years)			
< 2	1	1	
2 – 4	0.67 [0.53 – 0.86] *	-	
≥ 5	0.60 [0.51 – 0.72] *	0.75 [0.75 – 0.75]	<0.001
First viral load (copies/mL)			
1,000 – 5,000	1	1	
5,001 – 10,000	0.84 [0.42 – 1.69]	1.50 [1.09 – 2.05]	0.013
>10,000	0.61 [0.38 – 0.97] *	0.48 [0.48 – 0.48]	**<0.001**
Baseline CD4 cell count (cells/mm³)	1	1	
< 200	0.99 [0.65 – 1.52]	0.43 [0.37 – 0.50]	**<0.001**
200 – 499	0.78 [0.36 – 1.70]	0.11 [0.02 – 0.68]	0.017
≥ 500			
SARS-CoV-2 status (PCR results)			
Negative	1	1	
Positive	0.67 [0.32 – 1.42]	0.59 [0.22 – 1.58]	0.295
Concomitant Medications			
No	1	1	
Yes	0.74 [0.56 – 0.96]	0.67 [0.58 0.79]	**<0.001**
Comorbidities			
No	1	1	
Yes	1.01 [0.73 – 1.40]	0.77 [0.66 – 0.90]	**0.001**

CONCLUSION

In the VISEND trial, EAC led to VS rates near the WHO target of 70% with disparities in outcomes according to gender, education, and other factors.[1,2]

There is a need to routinely incorporate EAC into clinical trials and practice before regimen switch to maximize outcomes.

REFERENCES

1. World Health Organization. Consolidated guidelines on the use of antiretroviral drugs for treating and preventing HIV infection: recommendations for a public health approach. Geneva: World Health Organization; 2016

2. Nakalega, R., Mukiza, N., Debem, H. *et al.* Linkage to intensive adherence counselling among HIV-positive persons on ART with detectable viral load in Gomba district, rural Uganda. *AIDS Res Ther* 18, 15 (2021). https://doi.org/10.1186/s12981-021-00349-9

FUNDING

The study was sponsored by the :

1. Viatris (Mylan) Laboratories Ltd
2. Zambian Ministry of Health through the Global Fund to Fight AIDS, Tuberculosis and Malaria