0446

Long acting doravirine for treatment and prevention of vaginal HIV transmission

Martina Kovarova[1,2,3], Manse Kim[1,2,3], Sarah E. Wessel[1,2,3], J. Victor Garcia[1,2,3]

[1]International Center for the Advancement of Translational Science, University of North Carolina at Chapel Hill, Chapel Hill, NC, USA.

[2]Division of Infectious Diseases, Department of Medicine, University of North Carolina at Chapel Hill, Chapel Hill, NC, USA.

[3]Center for AIDS Research, University of North Carolina at Chapel Hill, Chapel Hill, NC, USA.

BACKGROUND

Non-adherence to HIV treatment is an important health care problem with subsequent development of drug resistance and disease progression. Doravirine (DOR) is a novel non-nucleoside reverse transcriptase inhibitor (NNRTI) with high activity against both wild-type virus (IC95 8.1 ng/ml) and the most prevalent NNRTI-resistant HIV variants. We developed injectable long-acting (LA) formulations of DOR that form an implant after subcutaneous administration, can deliver drug >5 months and can be removed to stop drug delivery. Importantly, DOR delivered in this way efficiently suppresses systemic HIV infection and prevents vaginal HIV transmission.

METHODS

- Injectable in situ forming implant formulations of DOR were developed and optimized to release DOR for extended time after subcutaneous administration with 28G needle.
- Extended release of DOR from optimized LA-DOR formulations was assessed in BALB/c mice (n=4 per formulation).
- Drug levels in selected tissues including lymph nodes, spleen, vagina, cervix, uterus, rectum, and ileum were evaluated 5 weeks post administration.
- Ability to suppressed established HIV infection in plasma and cervicovaginal secretions (CVS) was assessed in BLT humanized mice infected with transmitted/founder HIV_{RHPA} (n=3).
- Efficacy to prevent HIV vaginal transmission was evaluated using BLT humanized mice treated with LA-DOR (n=5) or placebo (n=5).
- BLT mice were vaginally challenge with HIV_{RHPA} 2-, 4-, and 6-weeks post LA-DOR administration. Peripheral blood was collected weekly and analyzed for HIV-RNA, CD4 T cells, and doravirine concentration.
- Ten weeks post administration (4 weeks after the last challenge), LA-DOR implants were removed to stop drug delivery and viremia was monitored for additional 4 weeks.

Injectable long-acting ISFI formulation of doravirine protects from multiple high-dose vaginal HIV exposures.

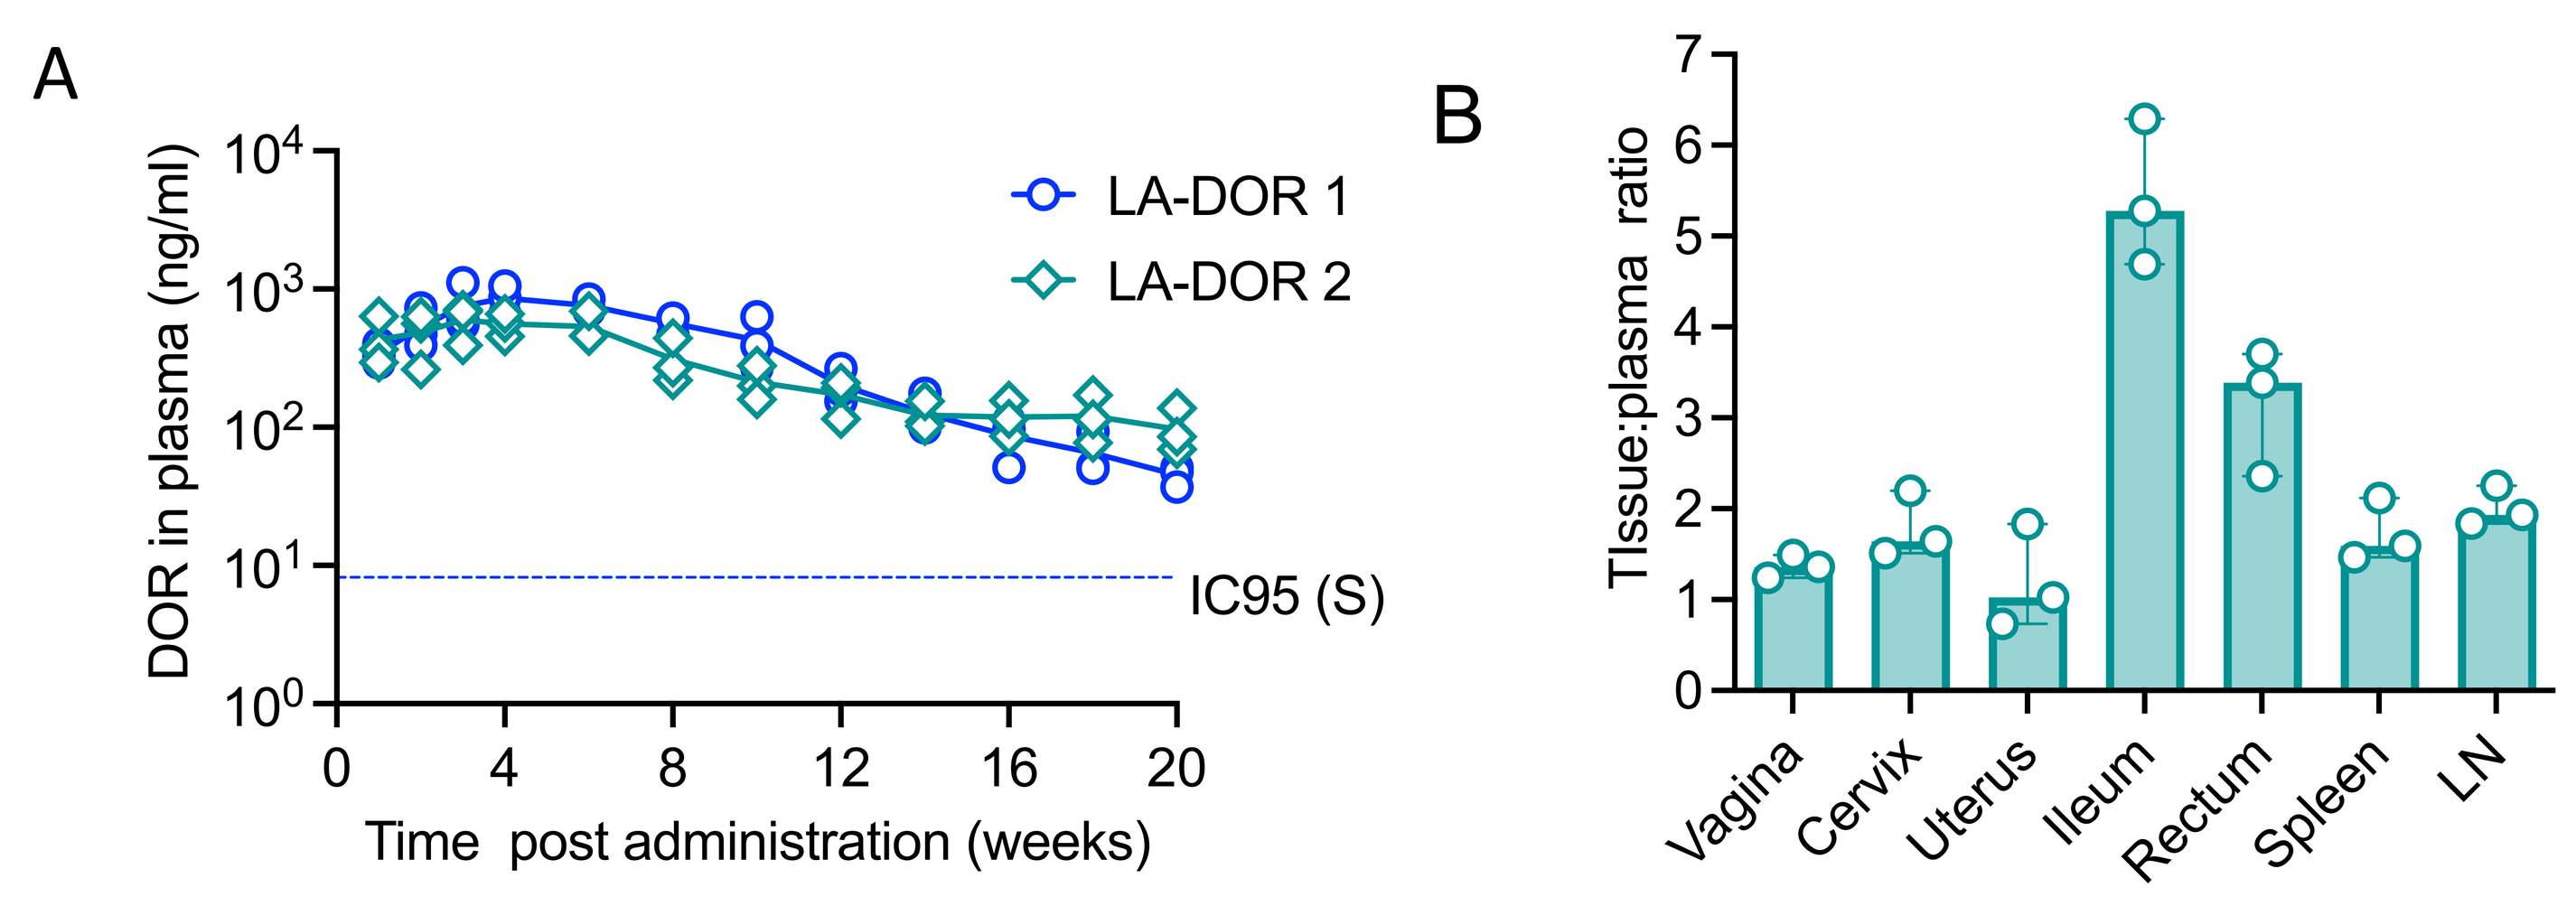

A
DOR in plasma (ng/ml)
LA-DOR 1
LA-DOR 2
IC95 (S)
Time post administration (weeks)
B
Tissue:plasma ratio
Vagina
Cervix
Uterus
Ileum
Rectum
Spleen
LN

Figure 1. Extended release of DOR from LA-DOR ISFI formulations. (A) BALB/c mice were administered with optimized LA-DOR formulation (6.8 mg DOR per mouse) and plasma concentration was measure for 20 weeks; n=4, serum adjusted IC95(S) is 8.2 ng/ml. (B) Tissue: plasma DOR concentration ratios in selected tissues 5 weeks after LA-DOR 2 administration, n=3, LN = lymph nodes.

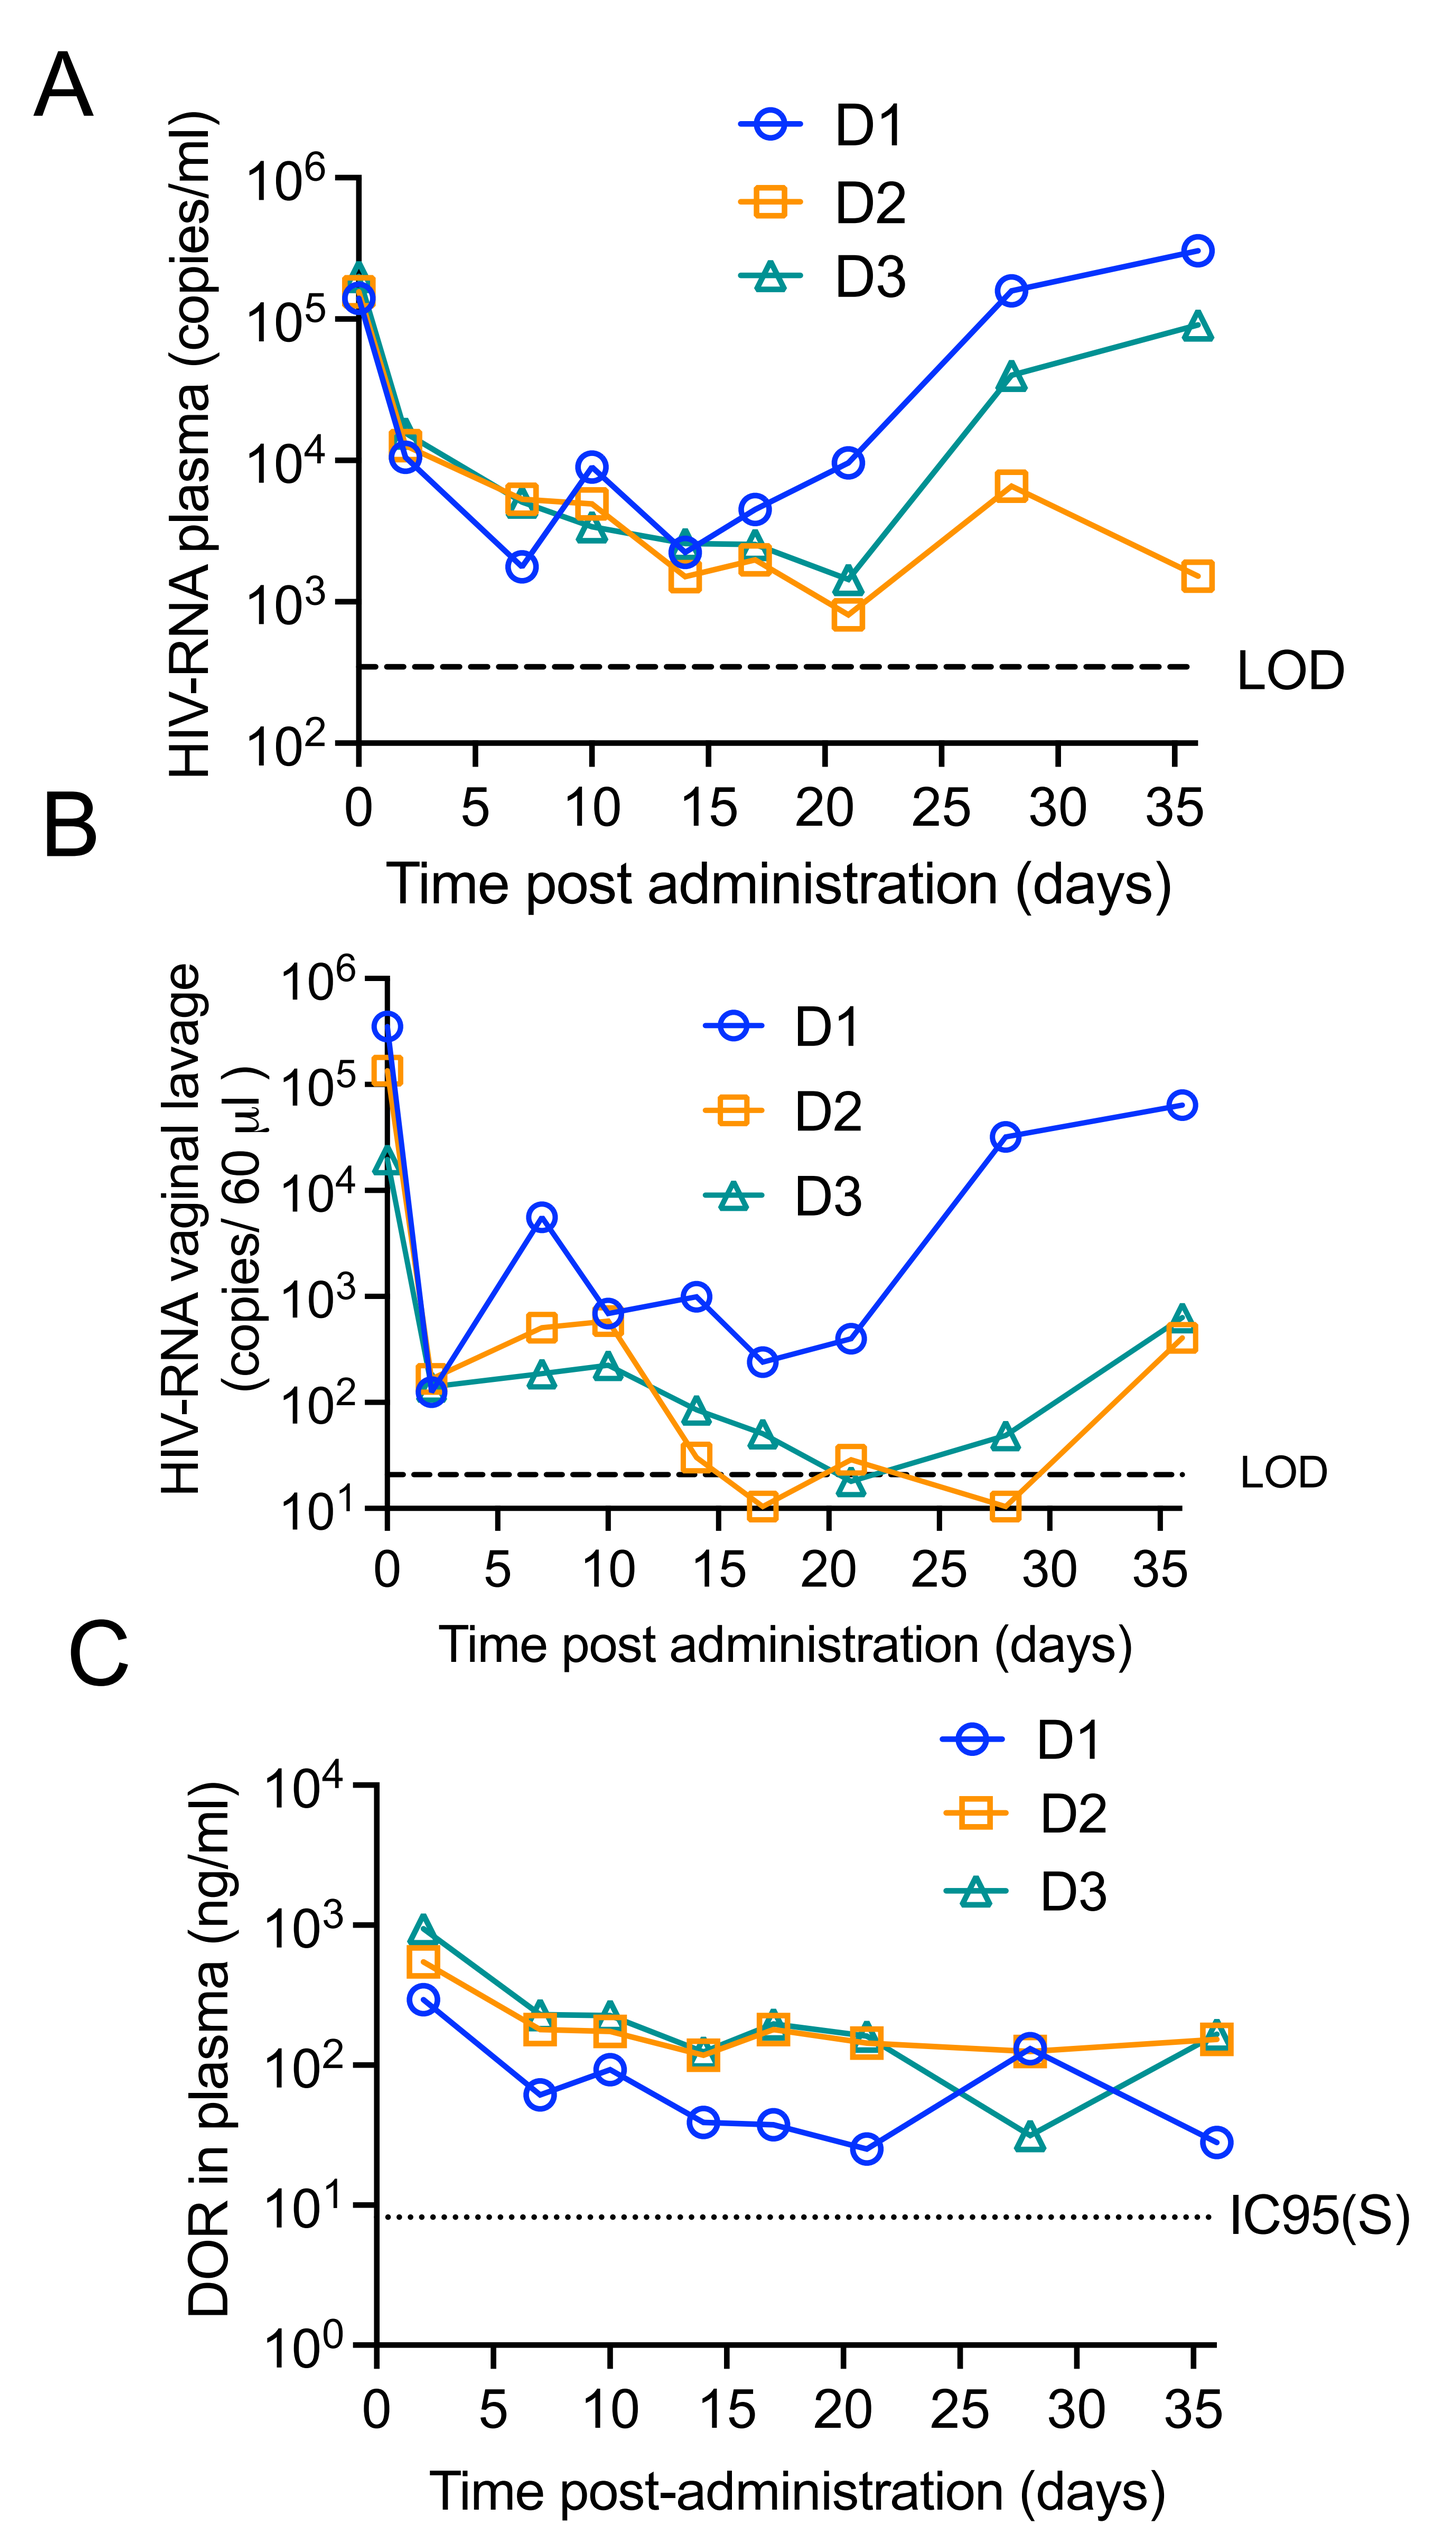

A
HIV-RNA plasma (copies/ml)
D1
D2
D3
LOD
Time post administration (days)
B
HIV-RNA vaginal lavage (copies/ 60 µl)
D1
D2
D3
LOD
Time post administration (days)
C
DOR in plasma (ng/ml)
D1
D2
D3
IC95(S)
Time post-administration (days)

Figure 2. LA-DOR reduces HIV in peripheral blood HIV_{RHPA} infected mice were treated with LA-DOR 1 (6.8 mg DOR per mouse) and transient suppression of HIV-RNA in peripheral blood and CVS was monitored until HIV rebound (A) HIV-RNA in plasma of individual mice.(B) HIV-RNA in CVS of individual mice. Dotted lines indicate limits of detection (346 copies/ml). (C) DOR concentration in plasma od individual mice. Dotted line indicates a human serum adjusted IC95 (8.1 ng/ml). DOR limit of detection 3ng/ml.

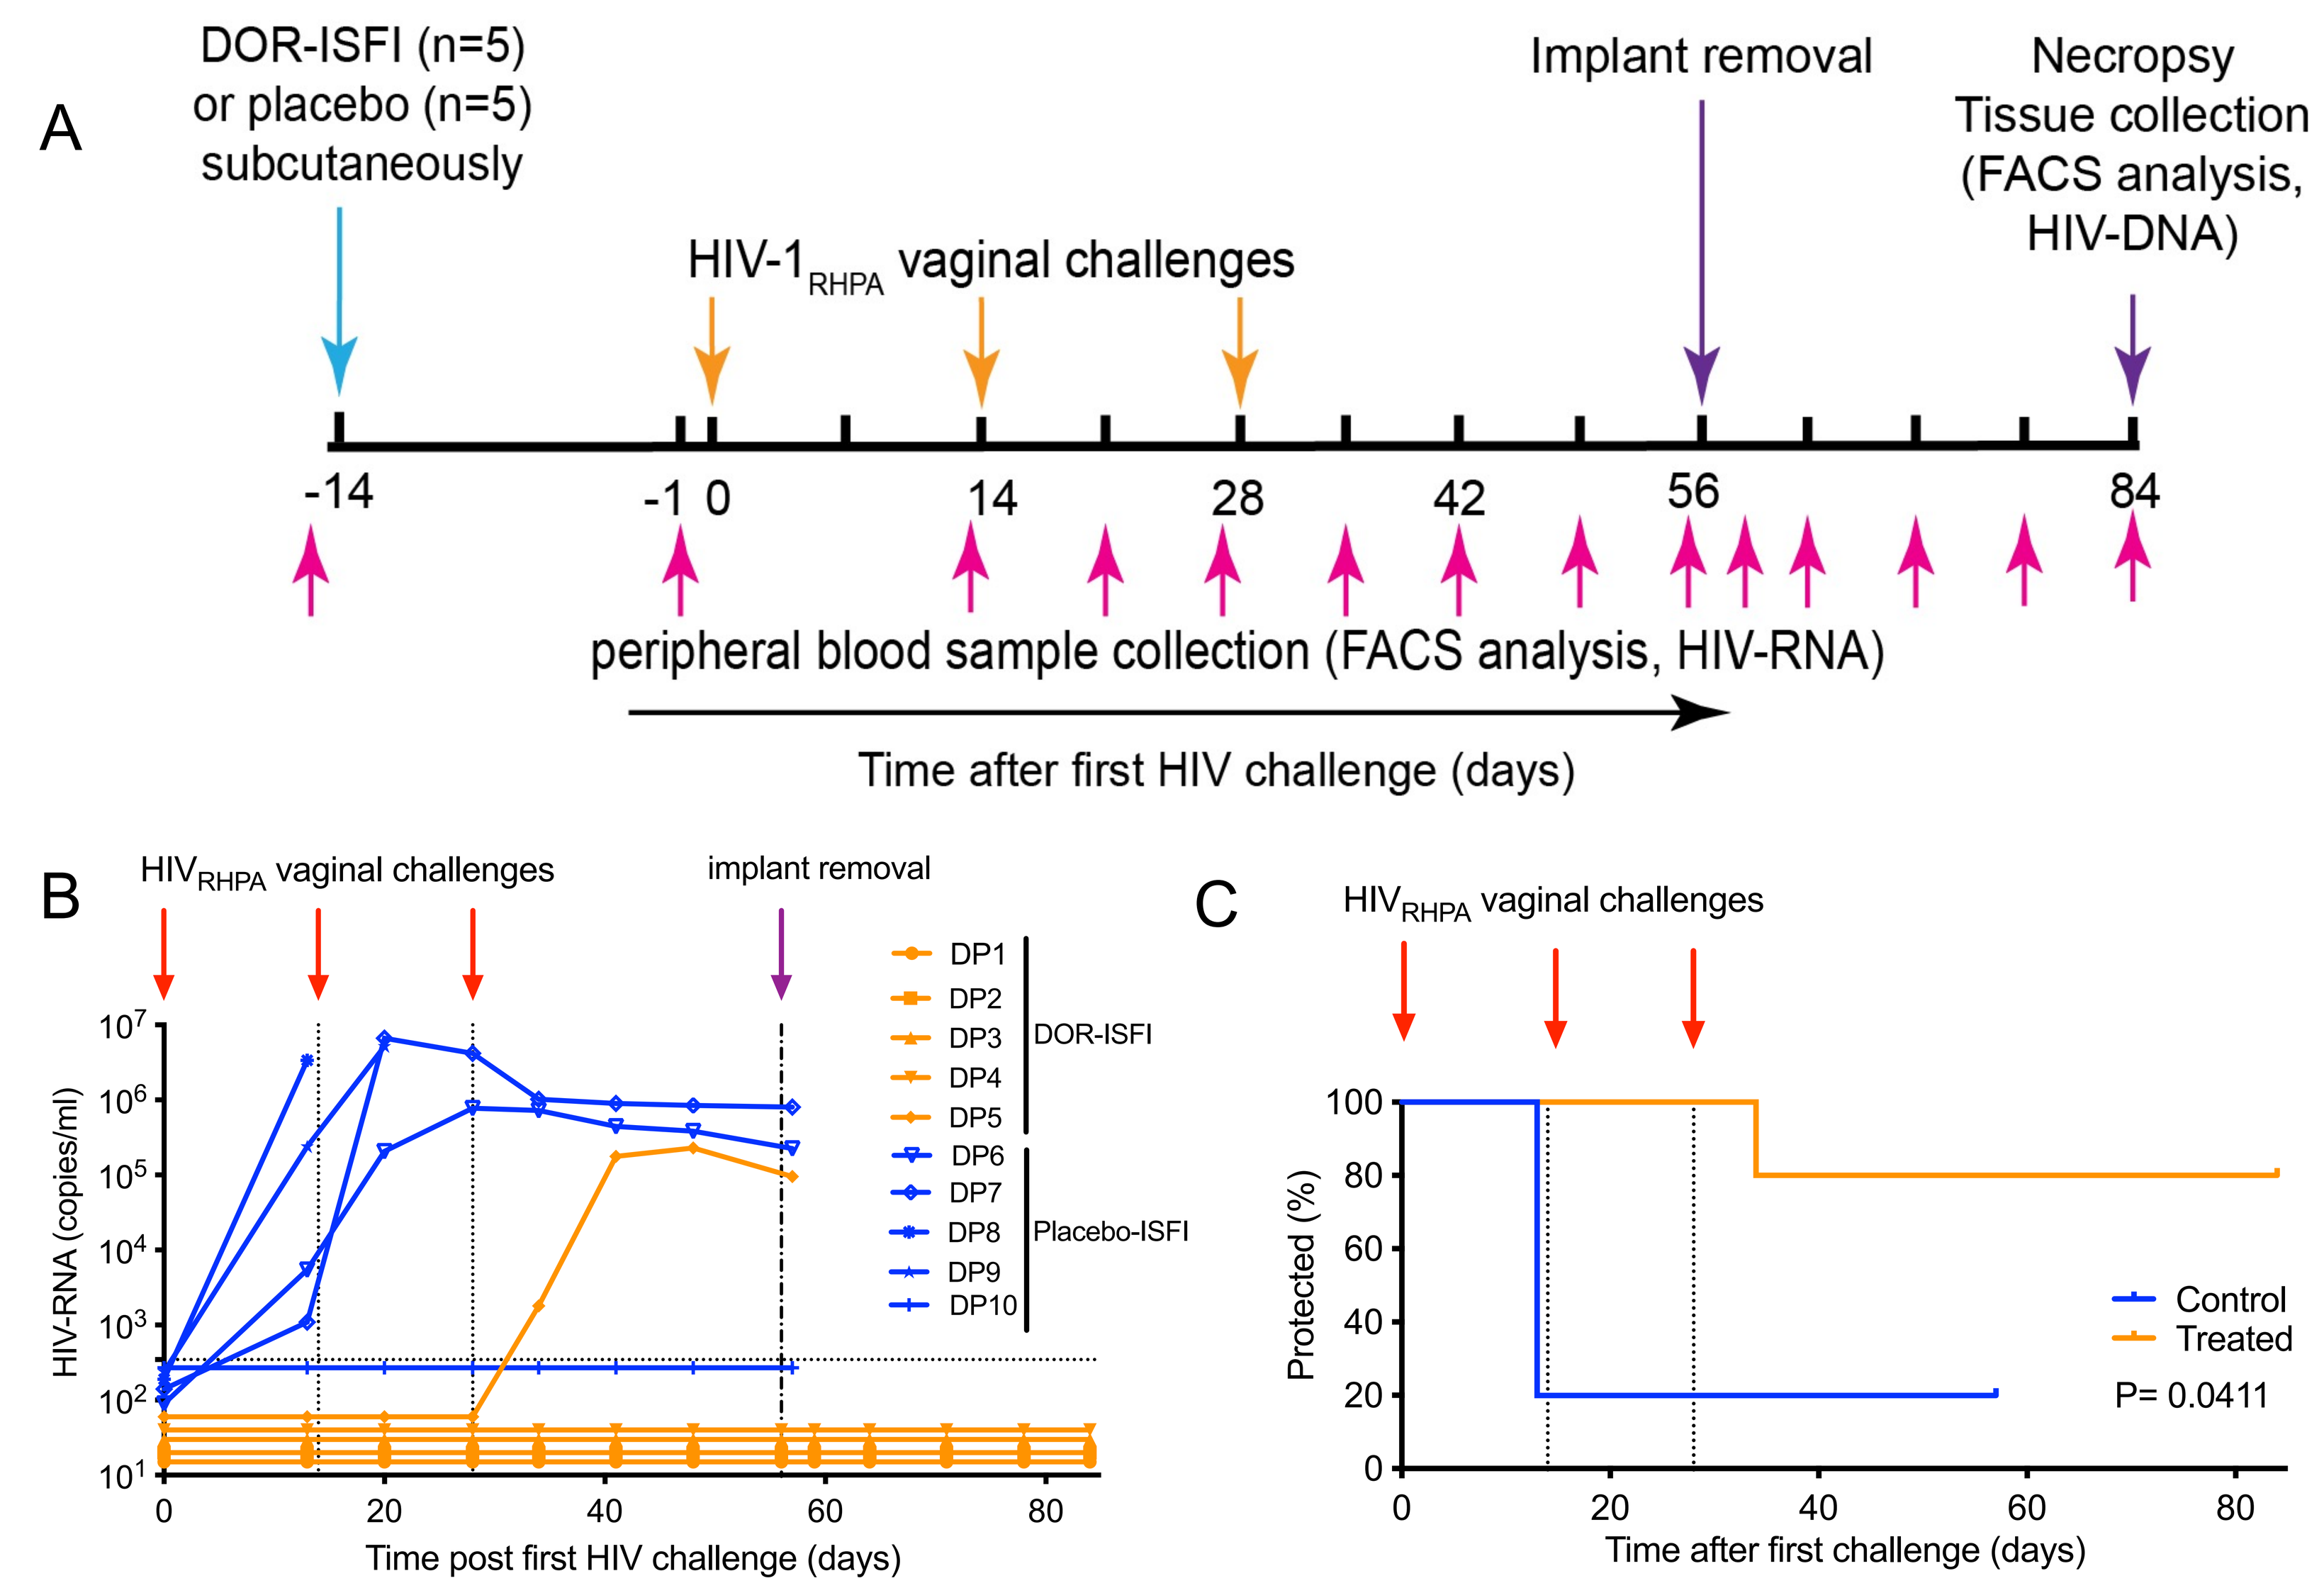

A
DOR-ISFI (n=5)
or placebo (n=5)
subcutaneously
HIV-1RHPA vaginal challenges
Implant removal
Necropsy
Tissue collection
(FACS analysis,
HIV-DNA)
-14
-1 0
14
28
42
56
84
peripheral blood sample collection (FACS analysis, HIV-RNA)
Time after first HIV challenge (days)
B
HIVRHPA vaginal challenges
implant removal
HIV-RNA (copies/ml)
DP1
DP2
DP3
DP4
DP5
DOR-ISFI
DP6
DP7
DP8
DP9
DP10
Placebo-ISFI
Time post first HIV challenge (days)
C
HIVRHPA vaginal challenges
Protected (%)
Control
Treated
P= 0.0411
Time after first challenge (days)

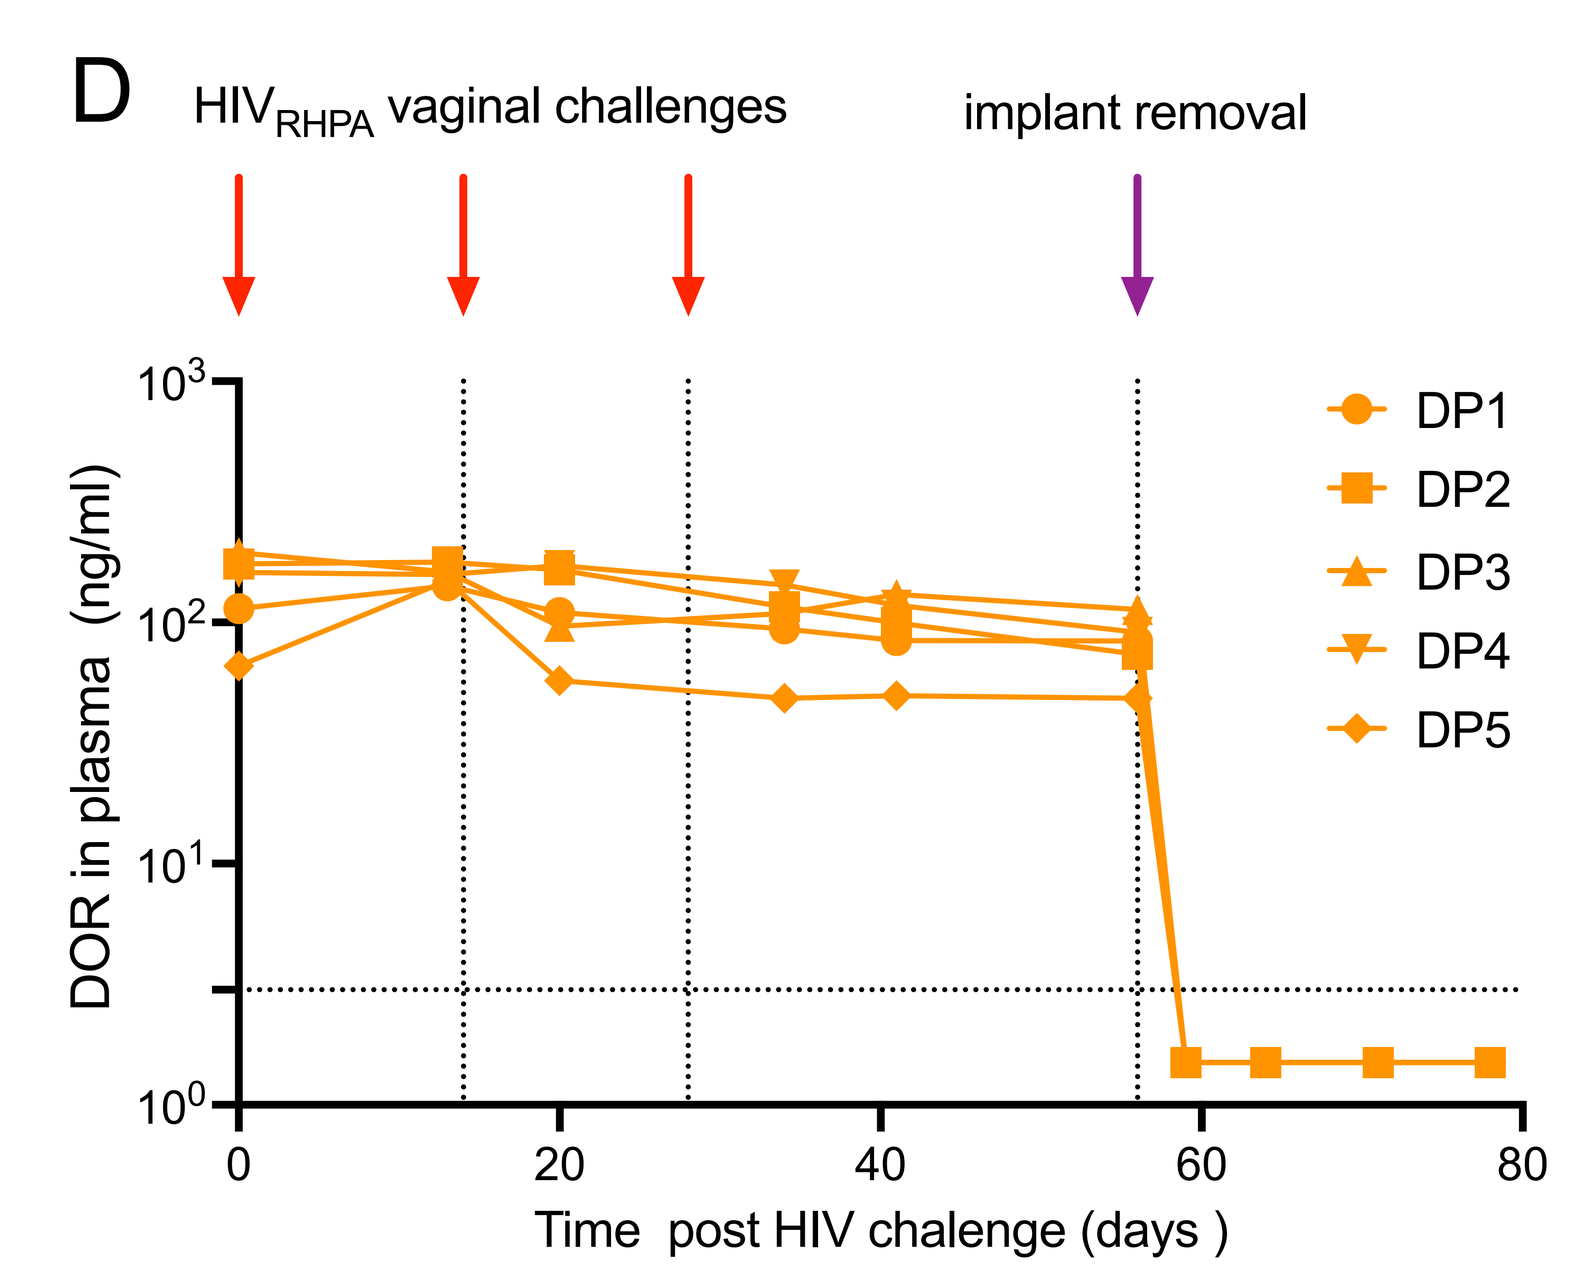

D
HIVRHPA vaginal challenges
implant removal
DOR in plasma (ng/ml)
DP1
DP2
DP3
DP4
DP5
Time post HIV chalenge (days)

Figure 3. LA-DOR protects from multiple vaginal HIV transmission. (A) Experimental design. BLT humanized mice were threated with a single subcutaneous injection of LA-DOR (6.8 mg DOR, n=5) or placebo (n=5). 2-, 4-, and 6- weeks later, mice were challenged with HIV_{RHPA} (3.5 x10^5 TCIU). Four week after the last challenge, LA-DOR implants were removed, and viremia monitored for additional 4 weeks. Cell-associated HIV-DNA was analyzed in multiple tissues in the end of the experiment to confirm protection (B) HIV-RNA in plasma of individual mice. The dotted line indicates the HIV-RNA limit of detection (346 copies/ml) (C) Kaplan-Mayer graph showing significant protection of LA-DOR treated BLT mice from vaginal HIV transmission. (D) Analysis of plasma DOR concentrations in individual mice during the experiment. The dotted line indicates DOR limit of detection (3 ng/ml).

RESULTS

- Two LA-DOR formulations with similar in vivo properties delivered DOR plasma concentrations above 10xIC95 (serum adjusted) for 16 weeks and above 1x IC95 for > 20 weeks (**Figure 1A**).
- DOR released from the LA-DOR showed high penetration to tissues, tissue:plasma ratios of DOR concentration was >1 in all tissue tested (**Figure 1B**).
- Median (range) tissue DOR concentrations five-weeks post injection were: vagina 206 ng/g (38.2-229), cervix 271.2 ng/g (42.5-336), uterus 122.3 ng/g (51.6-157), ileum 778.5 ng/g (177-808), rectum 391.4 ng/g (95.2-567), spleen 224.3 ng/g (44.7-351), and lymph nodes 281 ng/g (54.4-374).
- A single injection of LA-DOR (6.8 mg DOR) temporarily suppressed established HIV infection in plasma (2 logs) and CVS (3-5 logs) in all treated BLT humanized mice (**Figure 2**).
- When used for pre-exposure prophylaxis, 4 of 5 controls administered with placebo became infected compared to 1 of 8 LA-DOR treated animals. After implant removal, no additional viremia was identified in LA-DOR treated mice (**Figure 3**).

CONCLUSIONS

The LA-DOR formulation can deliver drug for more than five months after a single subcutaneous injection, penetrate to relevant tissues and efficiently prevent vaginal HIV transmission after multiple HIV challenges.

SUPPORT:

This work was supported by funding from National Institutes of Health grants R01AI111899 (JVG)