PHASE 2 CLINICAL TRIAL OF VEDOLIZUMAB AND ART IN SUBJECTS WITH NO PREVIOUS ART

María Reyes Jiménez-León, Carmen Gasca-Capote, Cristina Roca-Oporto, Nuria Espinosa, Salvador Sobrino, María Fontillon, Inmaculada Rivas-Jeremias, Karin Neukam, Jose German Sanchez-Hernandez, Raul Rigo-Bonnin, Antonio J. Cervera, Rosario Mesones, Pompeyo Viciana, Luis F. Lopez-Cortes, Ezequiel Ruiz-Mateos.

Clinical Unit of Infectious Diseases, Microbiology, and Preventive Medicine, Institute of Biomedicine of Seville, Virgen del Rocio University Hospital/CSIC/University of Seville[1], Institute of Biomedicine of Seville, Sevilla, Spain[2], Institute de Salud Carlos III, Majadahonda, Spain[3], Centro Sandoval, Madrid, Spain[4], Fundacion Jimenez Diaz, Madrid, Spain[5], Hospital Clinic of Barcelona, Barcelona, Spain[6]

CROI Conference on Retroviruses and Opportunistic Infections

00359

reyesjimenezleon@gmail.com

INTRODUCTION

Complete HIV remission off treatment has not been possible. The aim of this study was to evaluate the safety and efficacy of Vedolizumab (anti-α4β7 mAb) combined with ART to achieve permanent virological remission in ART naïve subjects after ART interruption.

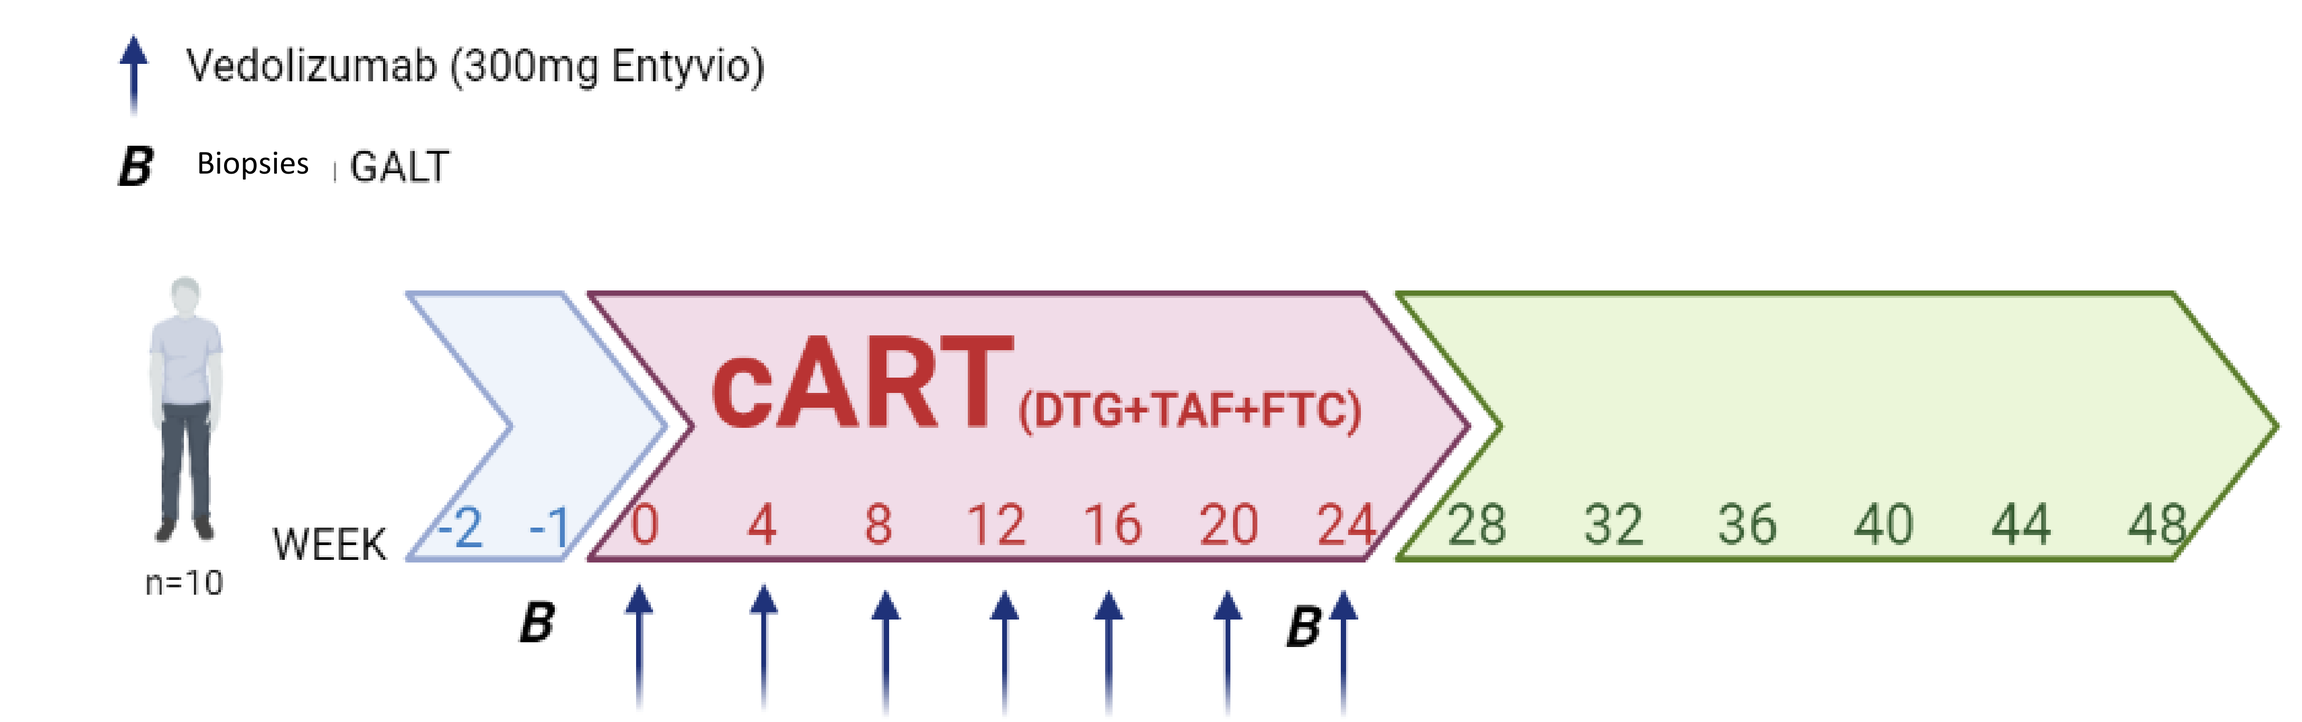
Vedolizumab (300mg Entyvio)
B Biopsies GALT
n=10
WEEK
-2 -1
cART (DTG+TAF+FTC)
0 4 8 12 16 20 24
28 32 36 40 44 48
B
B

MATERIALS AND METHODS

Open-label, single-arm phase 2 clinical trial. Ten patients were enrolled with CD4-T cells count of > 350 cells/µl and viral load >10.000 copies/ml. The time of infection was 75 [40-82] days. Patients started ART together with Vedolizumab infusions (300mg) at week 0, 4, 8, 12, 16, 20 and 24 weeks. At this time point ART and Vedolizumab treatment were interrupted. Biopsies were obtained from ileum and caecum at baseline (BL) and week 24. Subjects were monitored monthly by measuring CD4+T-cell counts, plasma viremia, Vedolizumab levels, HIV reservoir and flow cytometry to measure α4β7 levels and immune check point molecules among other markers. Criteria to restart ART were CD4 T-cells below 350 cell/µl or viral load >10^5 HIV-RNA copies/ml in two consecutive measurements.

RESULTS

1_ Four patients restarted ART due to an increase of viral load (>10^5 HIV-RNA copies/ml). The other six patients did not show criteria to restart ART and completed the follow up with of 1590, 6250, 10000, 36450 and 4300 HIV-RNA copies/ml.

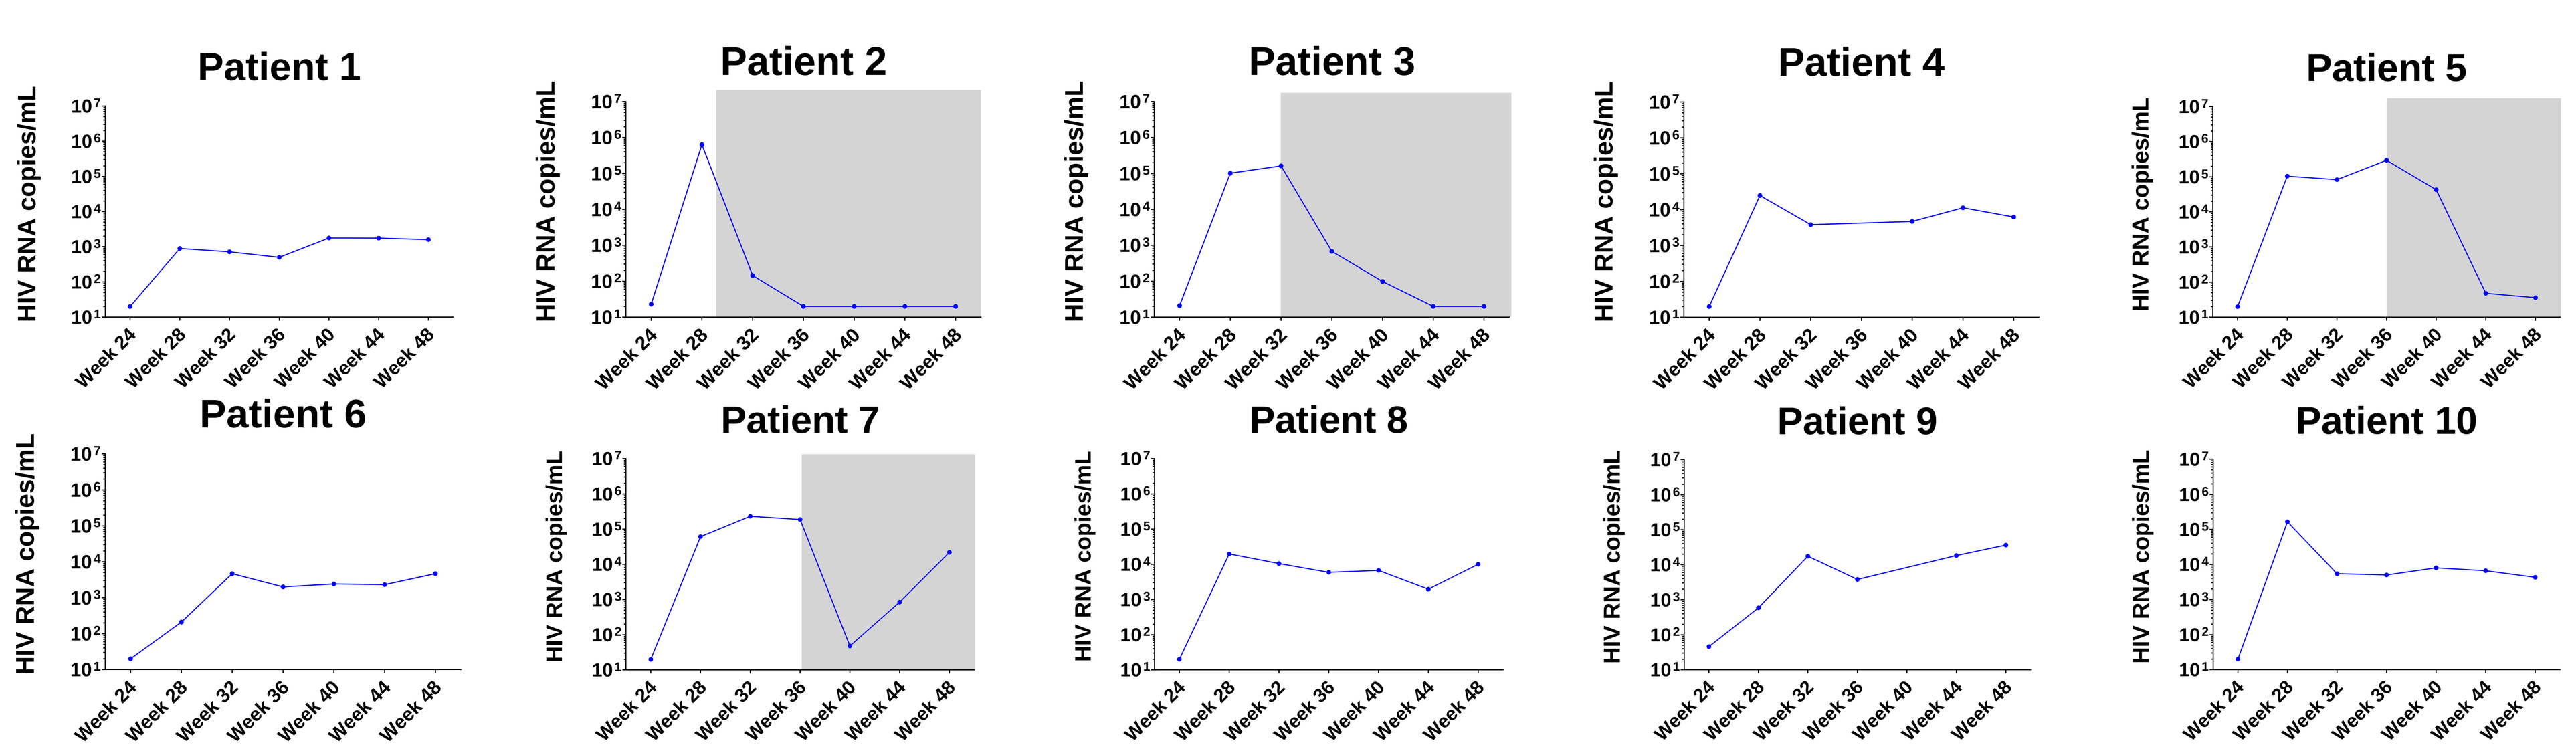
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5
Patient 6
Patient 7
Patient 8
Patient 9
Patient 10
HIV RNA copies/mL
Week 24 Week 28 Week 32 Week 36 Week 40 Week 44 Week 48

2_ No differences on either time to restart ART or time to first plasma viremia of >1000 HIV-RNA copies/ml when compared to historical controls of ART interruption (n=24) were observed (A). Nevertheless, Vedolizumab trial group present a viral load set point significant lower to the historical control group (B).

A

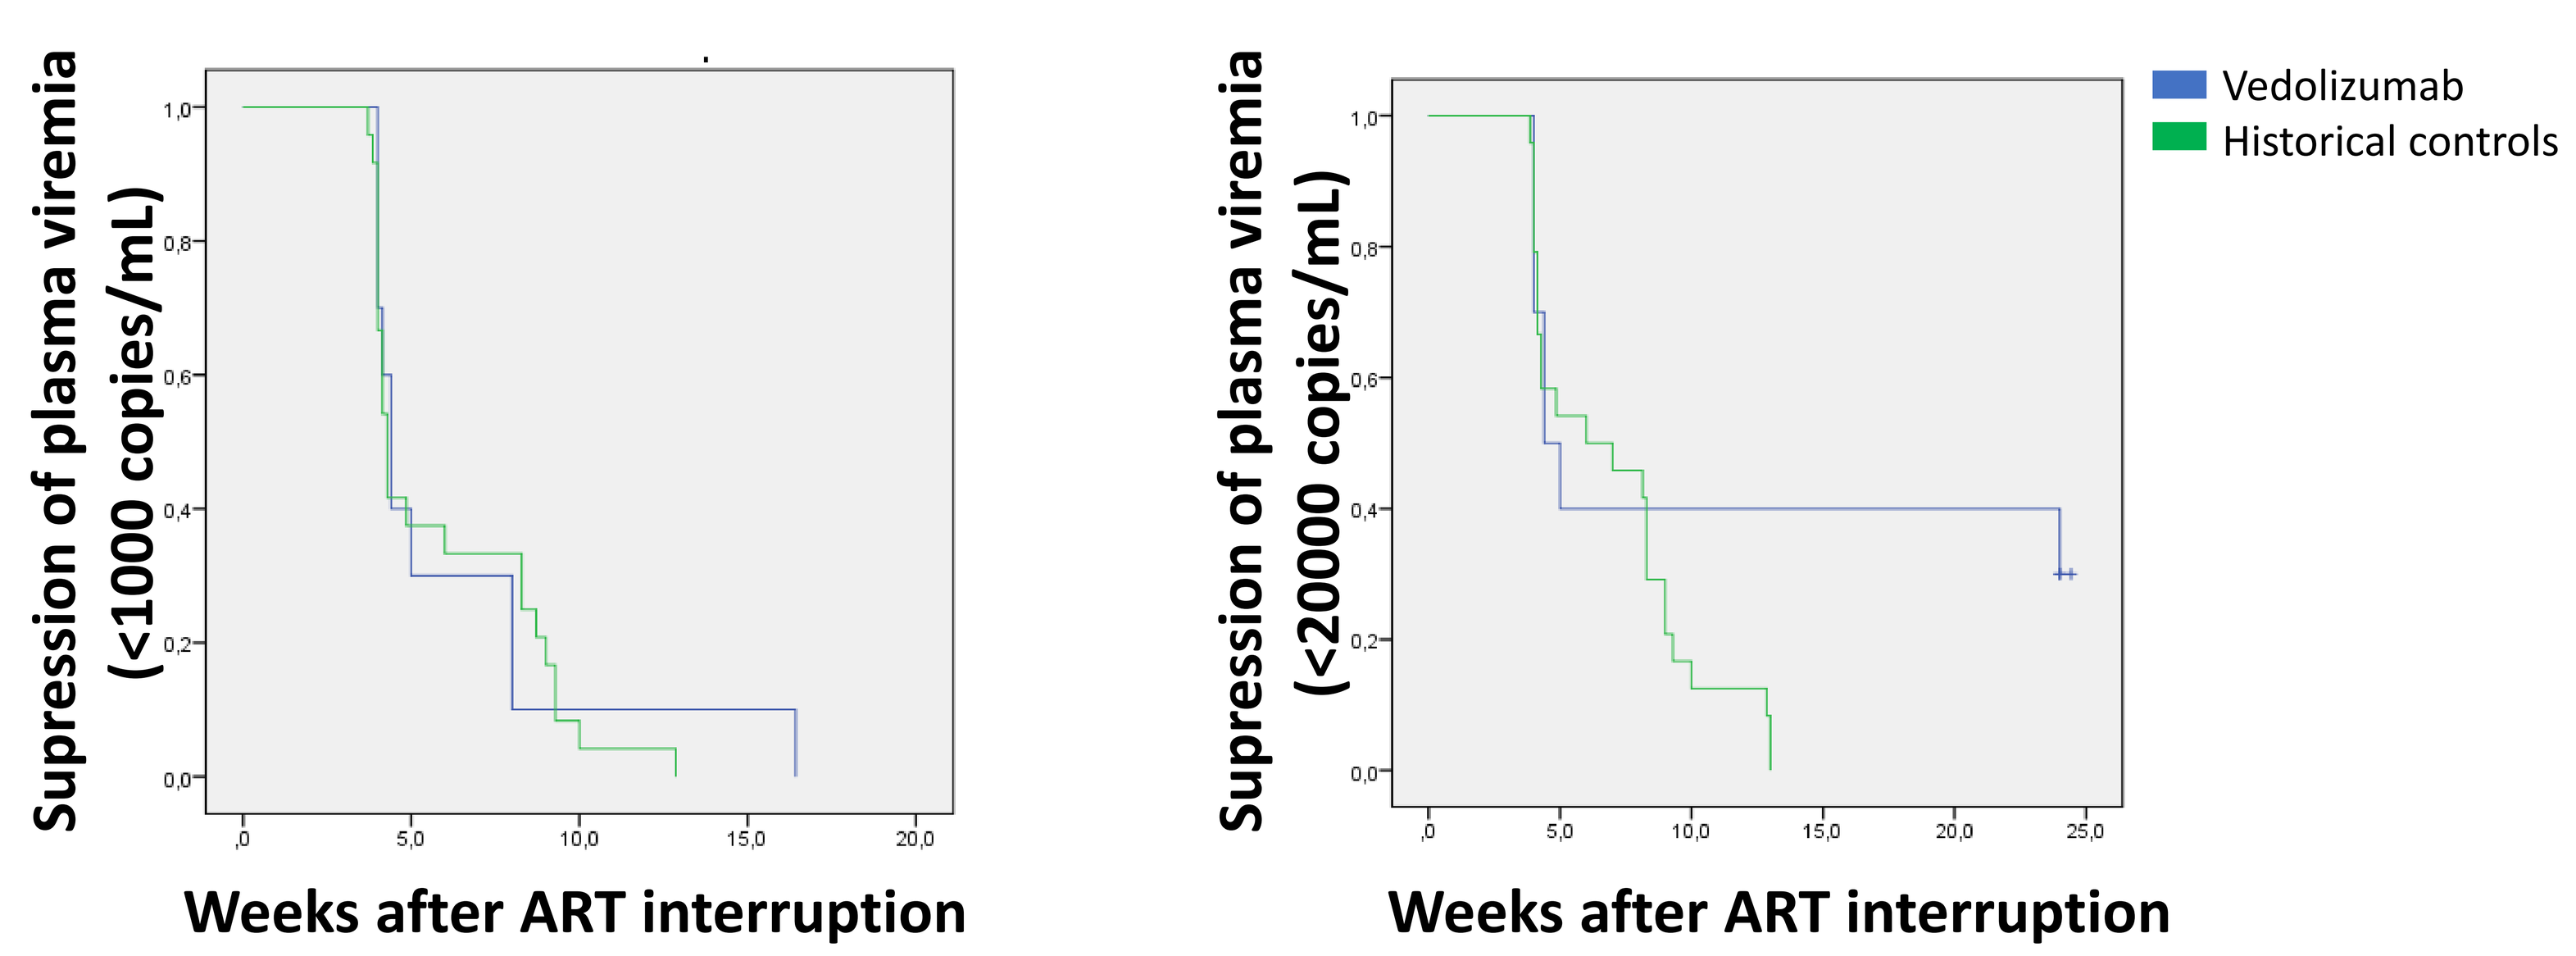
Supression of plasma viremia (<1000 copies/mL)
Weeks after ART interruption
Supression of plasma viremia (<20000 copies/mL)
Weeks after ART interruption
Vedolizumab
Historical controls

B

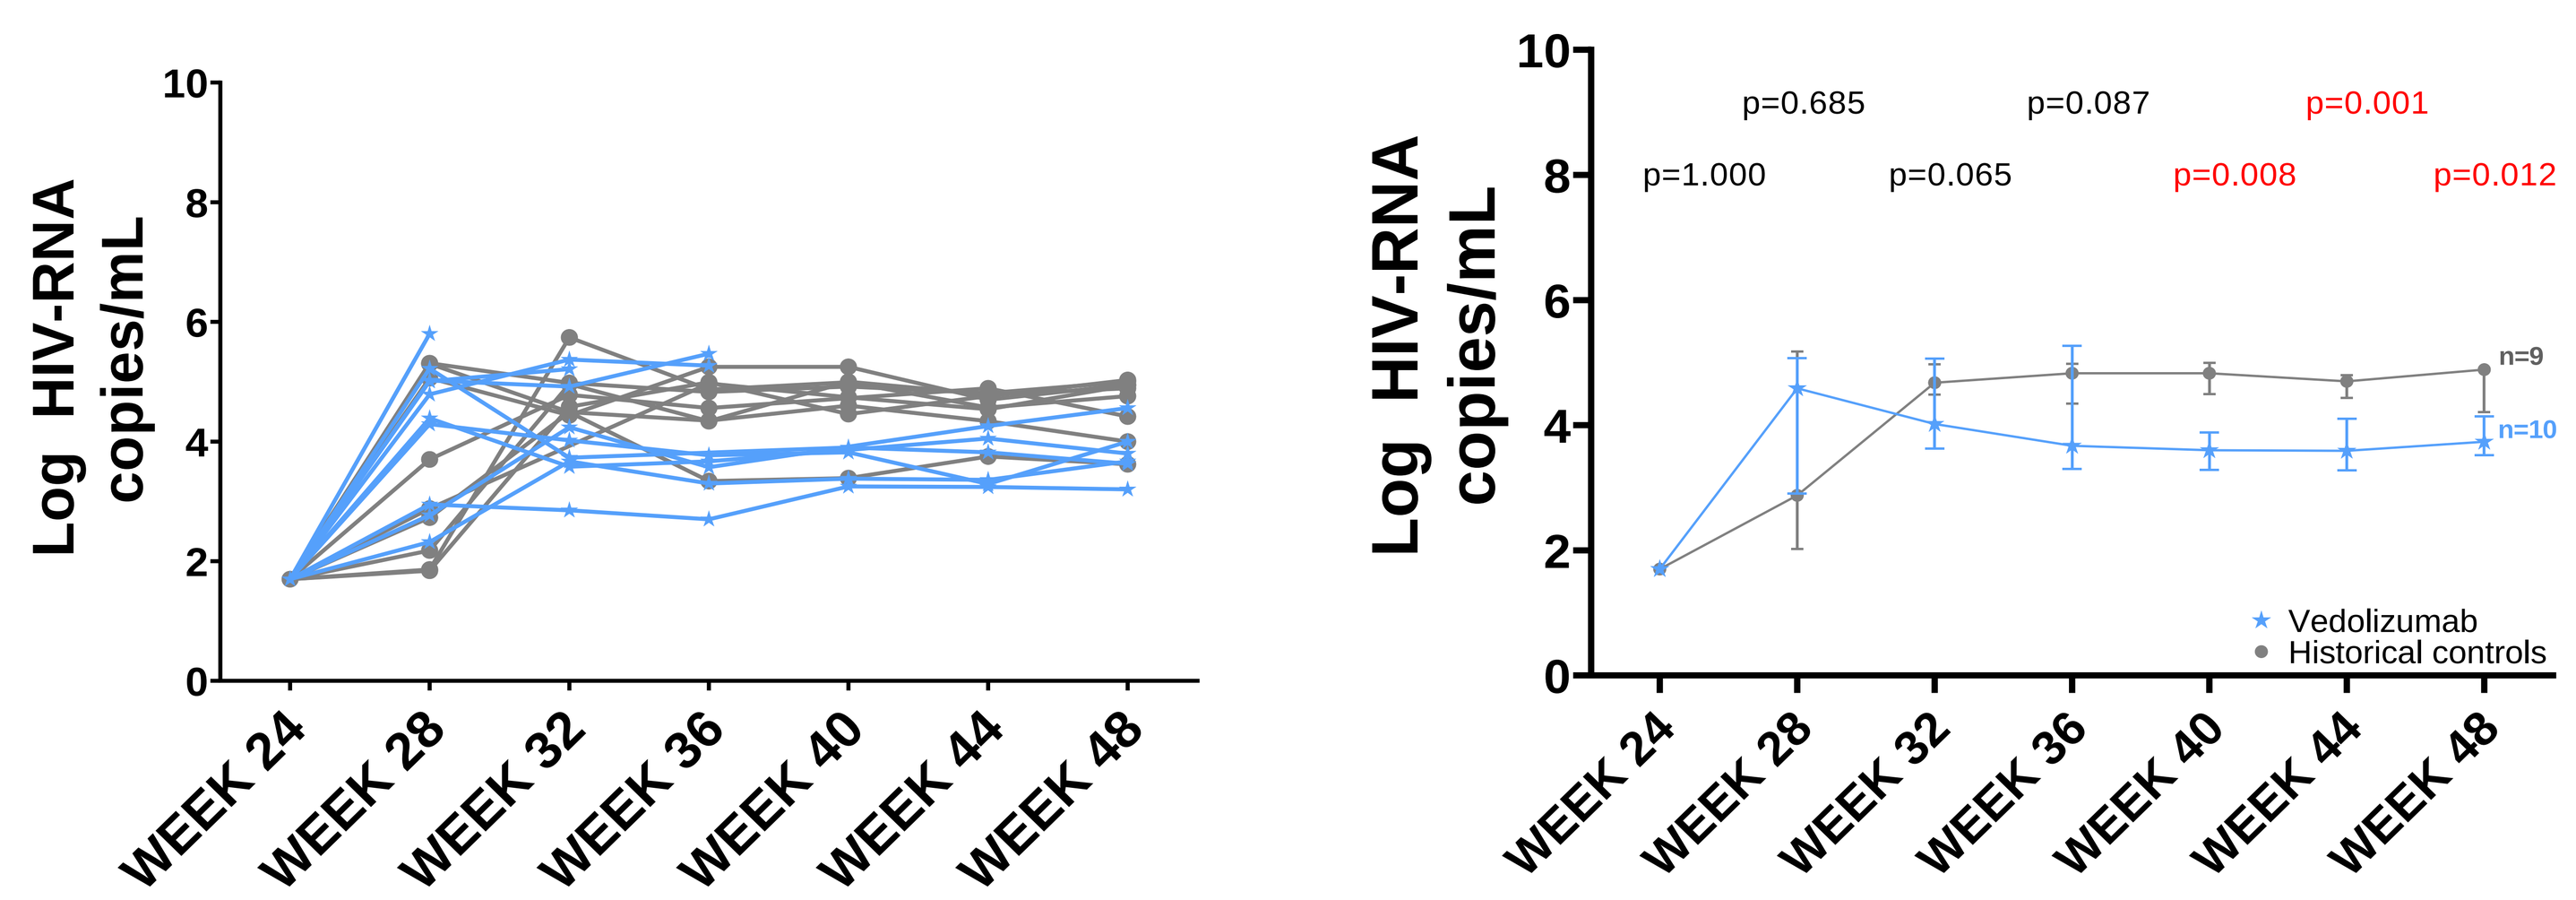
Log HIV-RNA copies/mL
WEEK 24 WEEK 28 WEEK 32 WEEK 36 WEEK 40 WEEK 44 WEEK 48
p=1.000
p=0.685
p=0.065
p=0.087
p=0.008
p=0.001
p=0.012
n=9
n=10
Vedolizumab
Historical controls

3_ We observed a decrease in HIV-DNA (p=0.027) and cell-associated HIV RNA on peripheral PBMCs (p=0.003), ileum and caecum cells over the follow up(A). The decrease on PBMCs was prominent in sorted CD4+CD45RO+ β7+ cells (B). Patients who restart ART presented higher levels of HIV reservoir on PBMCs (C).

A B

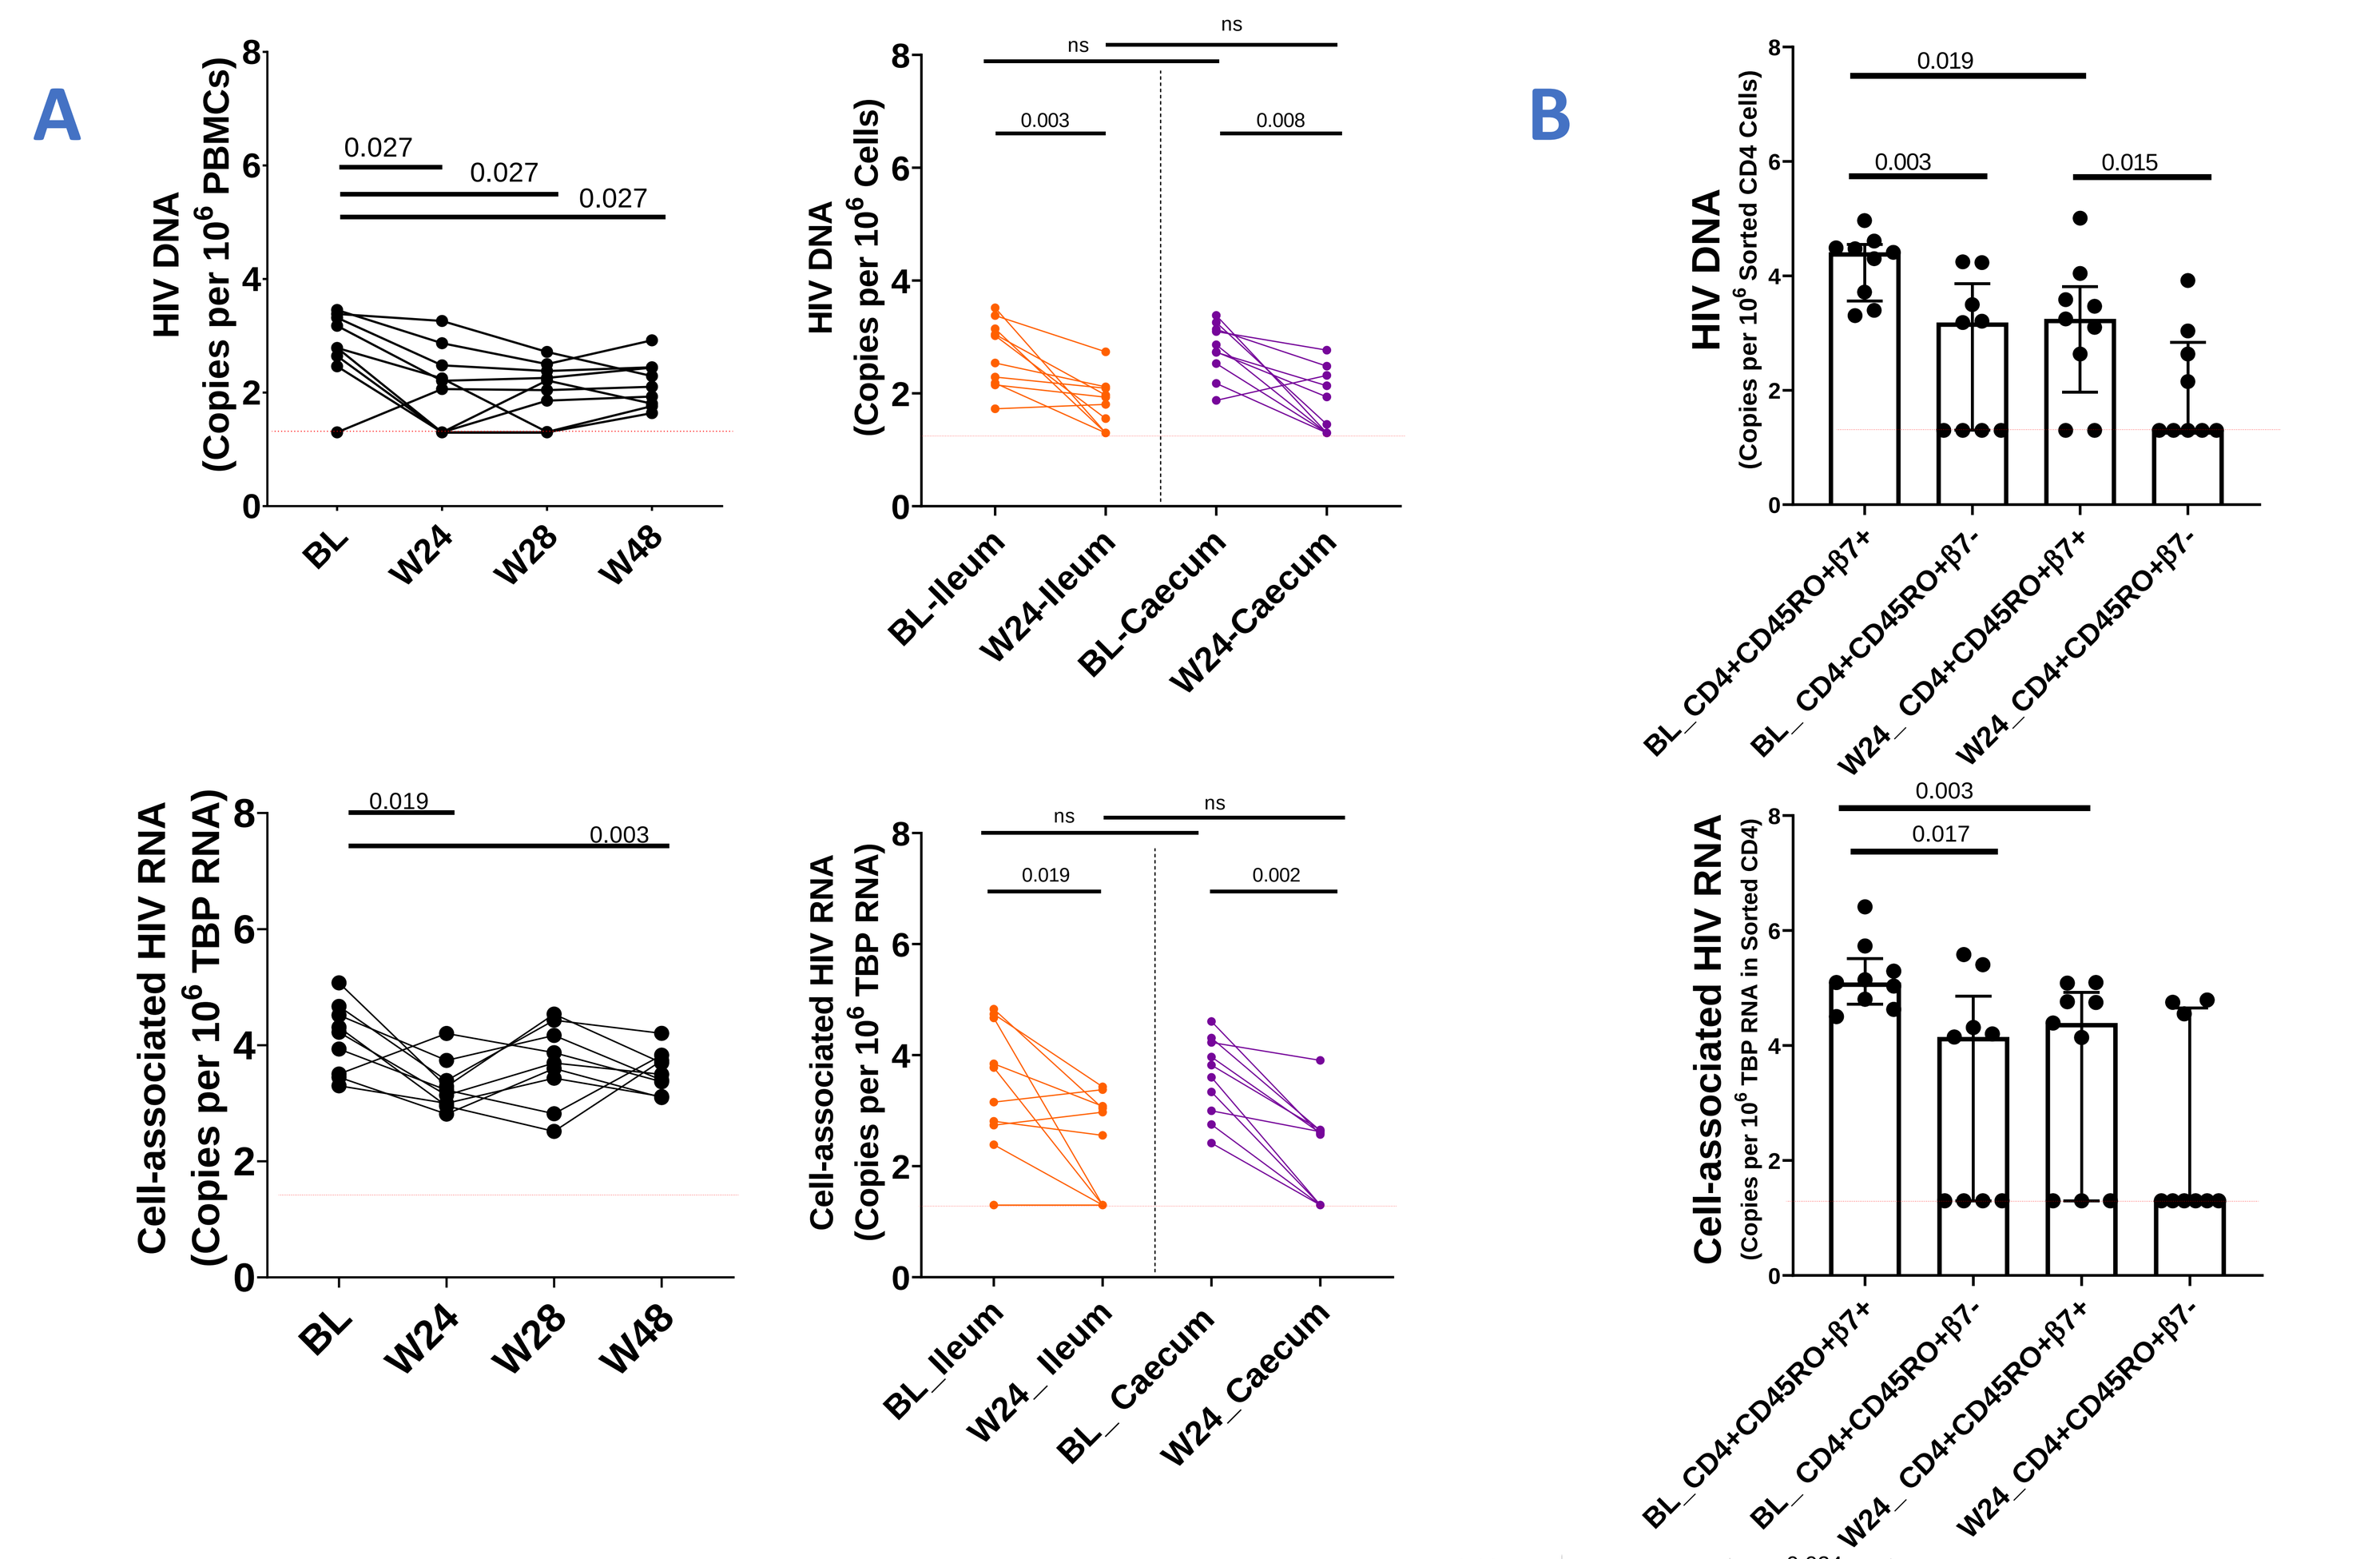
HIV DNA (Copies per 10^6 PBMCs)
0.027
0.027
0.027
BL W24 W28 W48
HIV DNA (Copies per 10^6 Cells)
ns
ns
0.003
0.008
BL-Ileum W24-Ileum BL-Caecum W24-Caecum
HIV DNA (Copies per 10^6 Sorted CD4 Cells)
0.019
0.003
0.015
BL_CD4+CD45RO+β7+ BL_CD4+CD45RO+β7- W24_CD4+CD45RO+β7+ W24_CD4+CD45RO+β7-
Cell-associated HIV RNA (Copies per 10^6 TBP RNA)
0.019
0.003
BL W24 W28 W48
Cell-associated HIV RNA (Copies per 10^6 TBP RNA)
ns
ns
0.019
0.002
BL_Ileum W24_Ileum BL_Caecum W24_Caecum
Cell-associated HIV RNA (Copies per 10^6 TBP RNA in Sorted CD4)
0.003
0.017

C

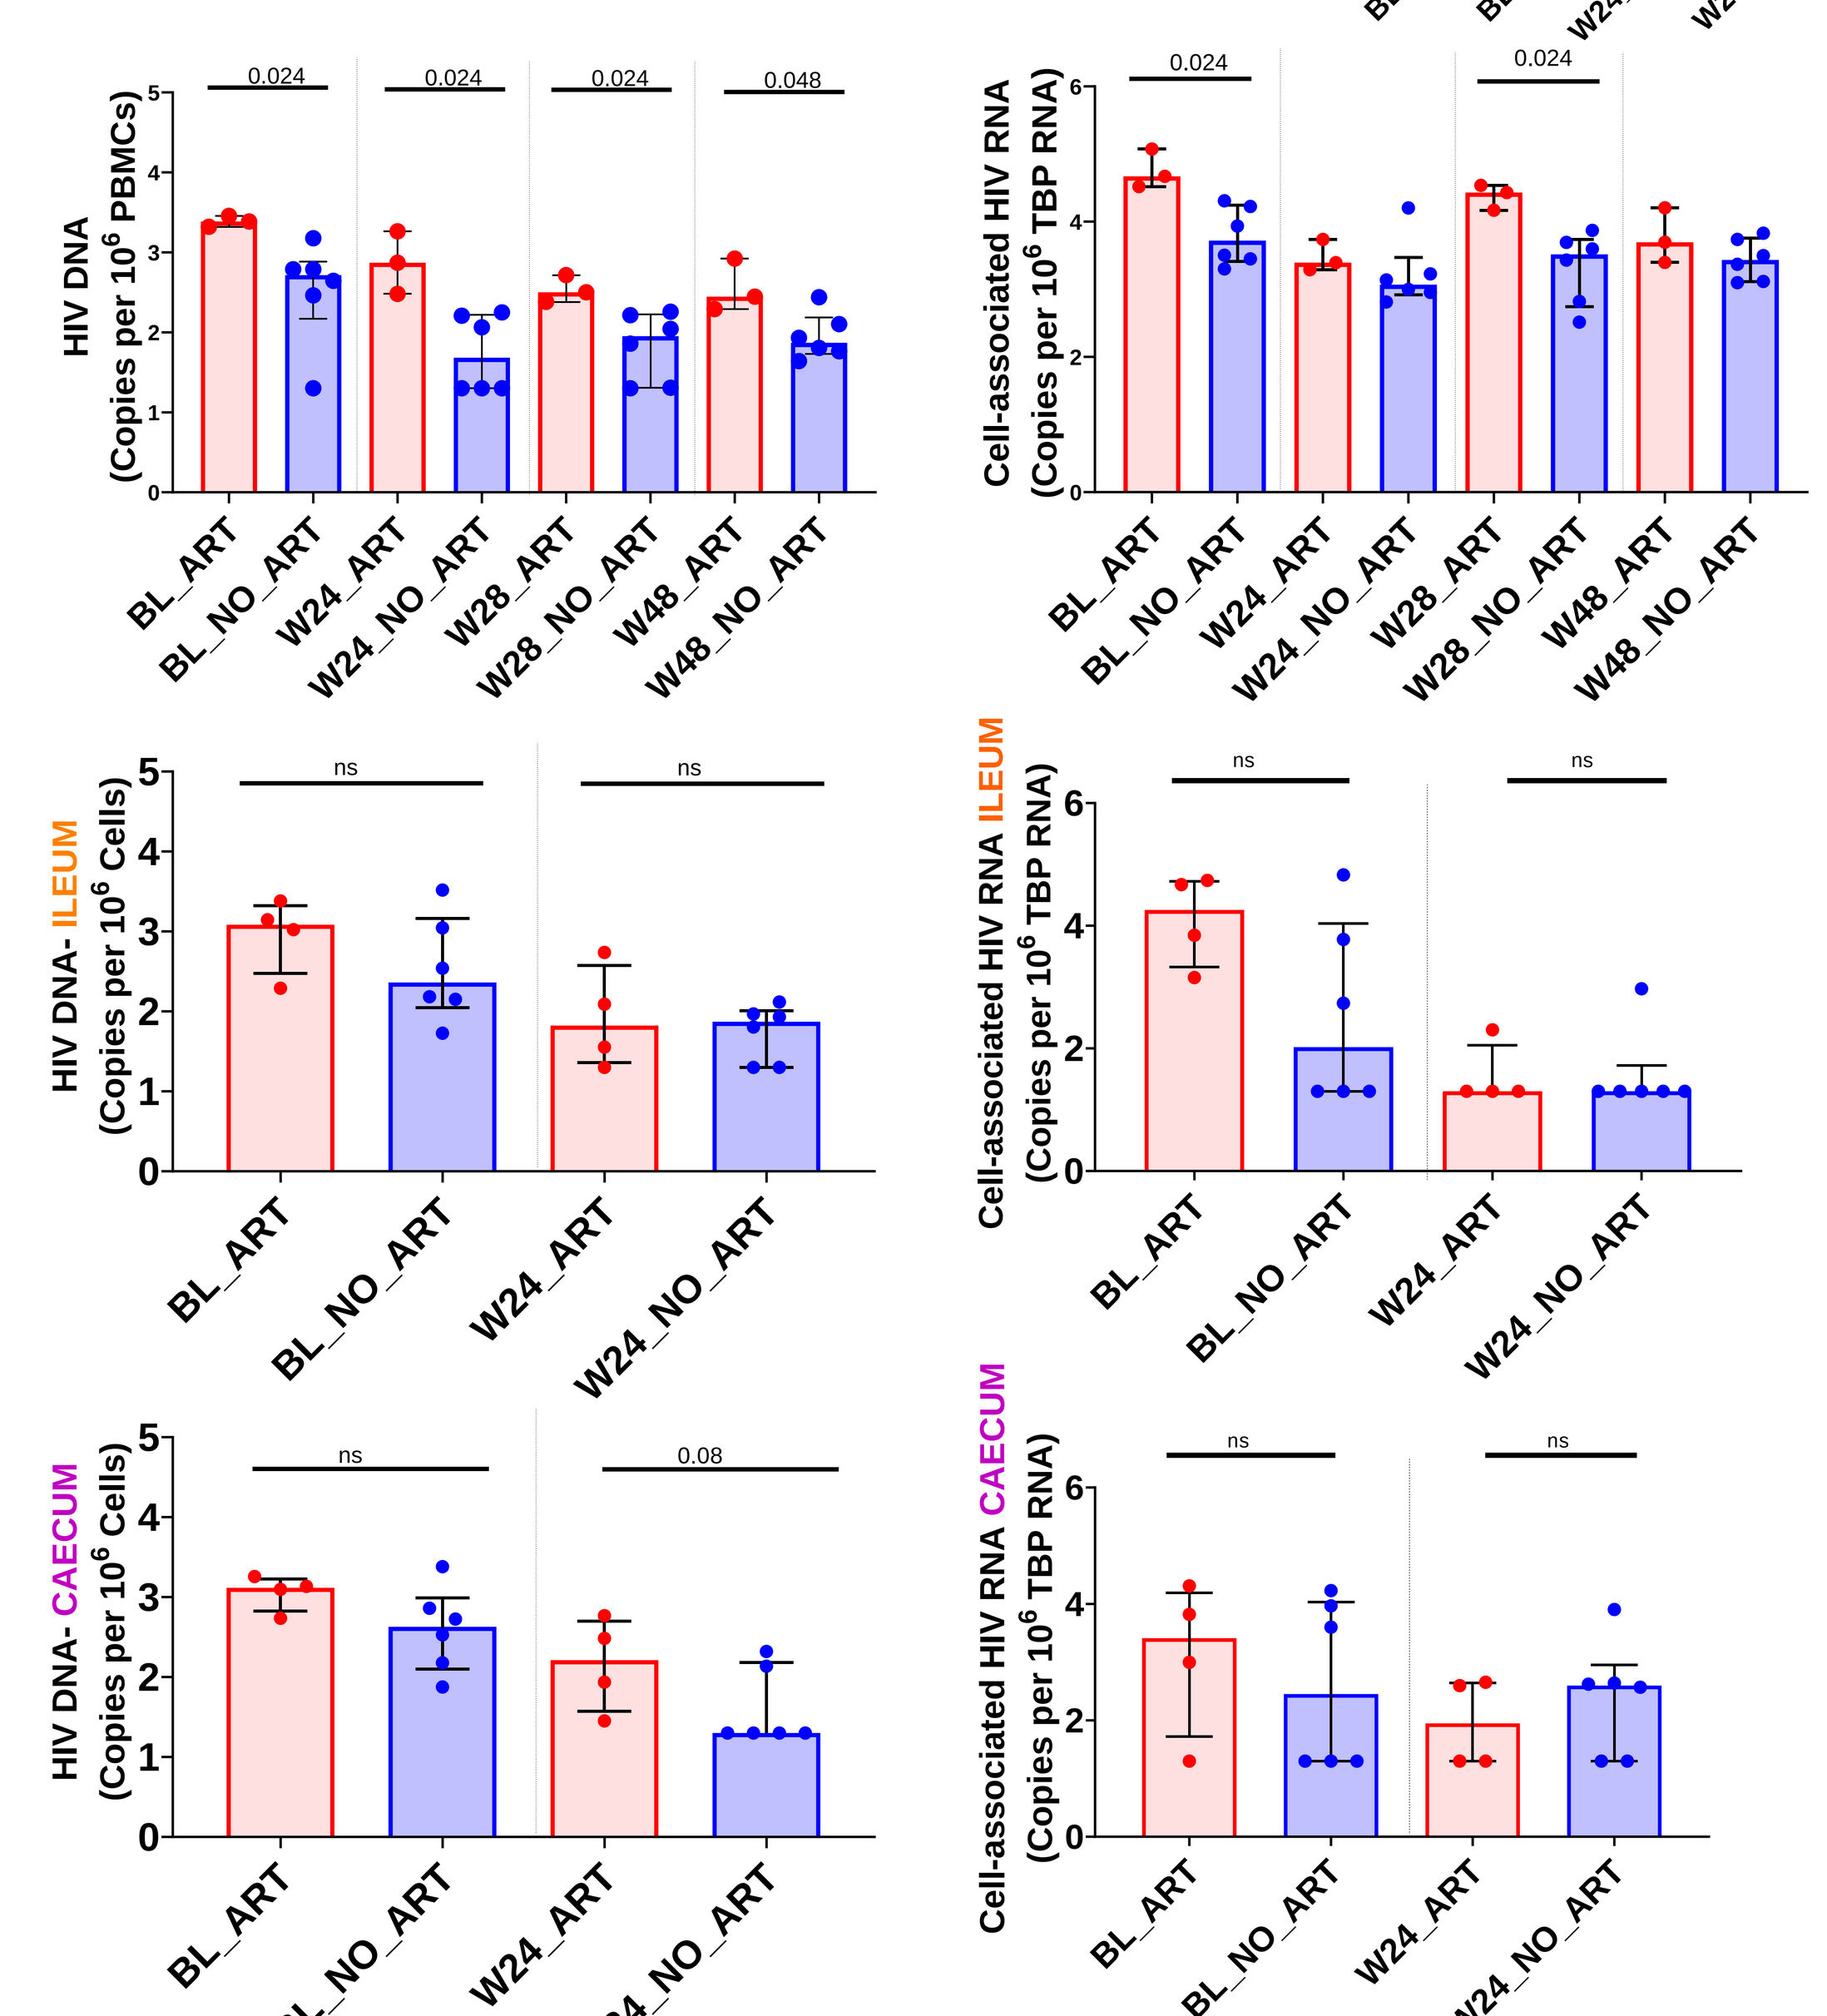
HIV DNA (Copies per 10^6 PBMCs)
0.024 0.024 0.024 0.048
BL_ART BL_NO_ART W24_ART W24_NO_ART W28_ART W28_NO_ART W48_ART W48_NO_ART
Cell-associated HIV RNA (Copies per 10^6 TBP RNA)
0.024 0.024
HIV DNA- ILEUM (Copies per 10^6 Cells)
ns ns
Cell-associated HIV RNA ILEUM (Copies per 10^6 TBP RNA)
ns ns
HIV DNA- CAECUM (Copies per 10^6 Cells)
ns 0.08
Cell-associated HIV RNA CAECUM (Copies per 10^6 TBP RNA)
ns ns
BL_ART BL_NO_ART W24_ART W24_NO_ART

4_ At W24, α4β7 was completely blocked by Vedolizumab on peripheral CD4+ T-Cells, unlike on gut (A). HIV reservoir was associated with expression levels of α4β7 integrin (B).

A

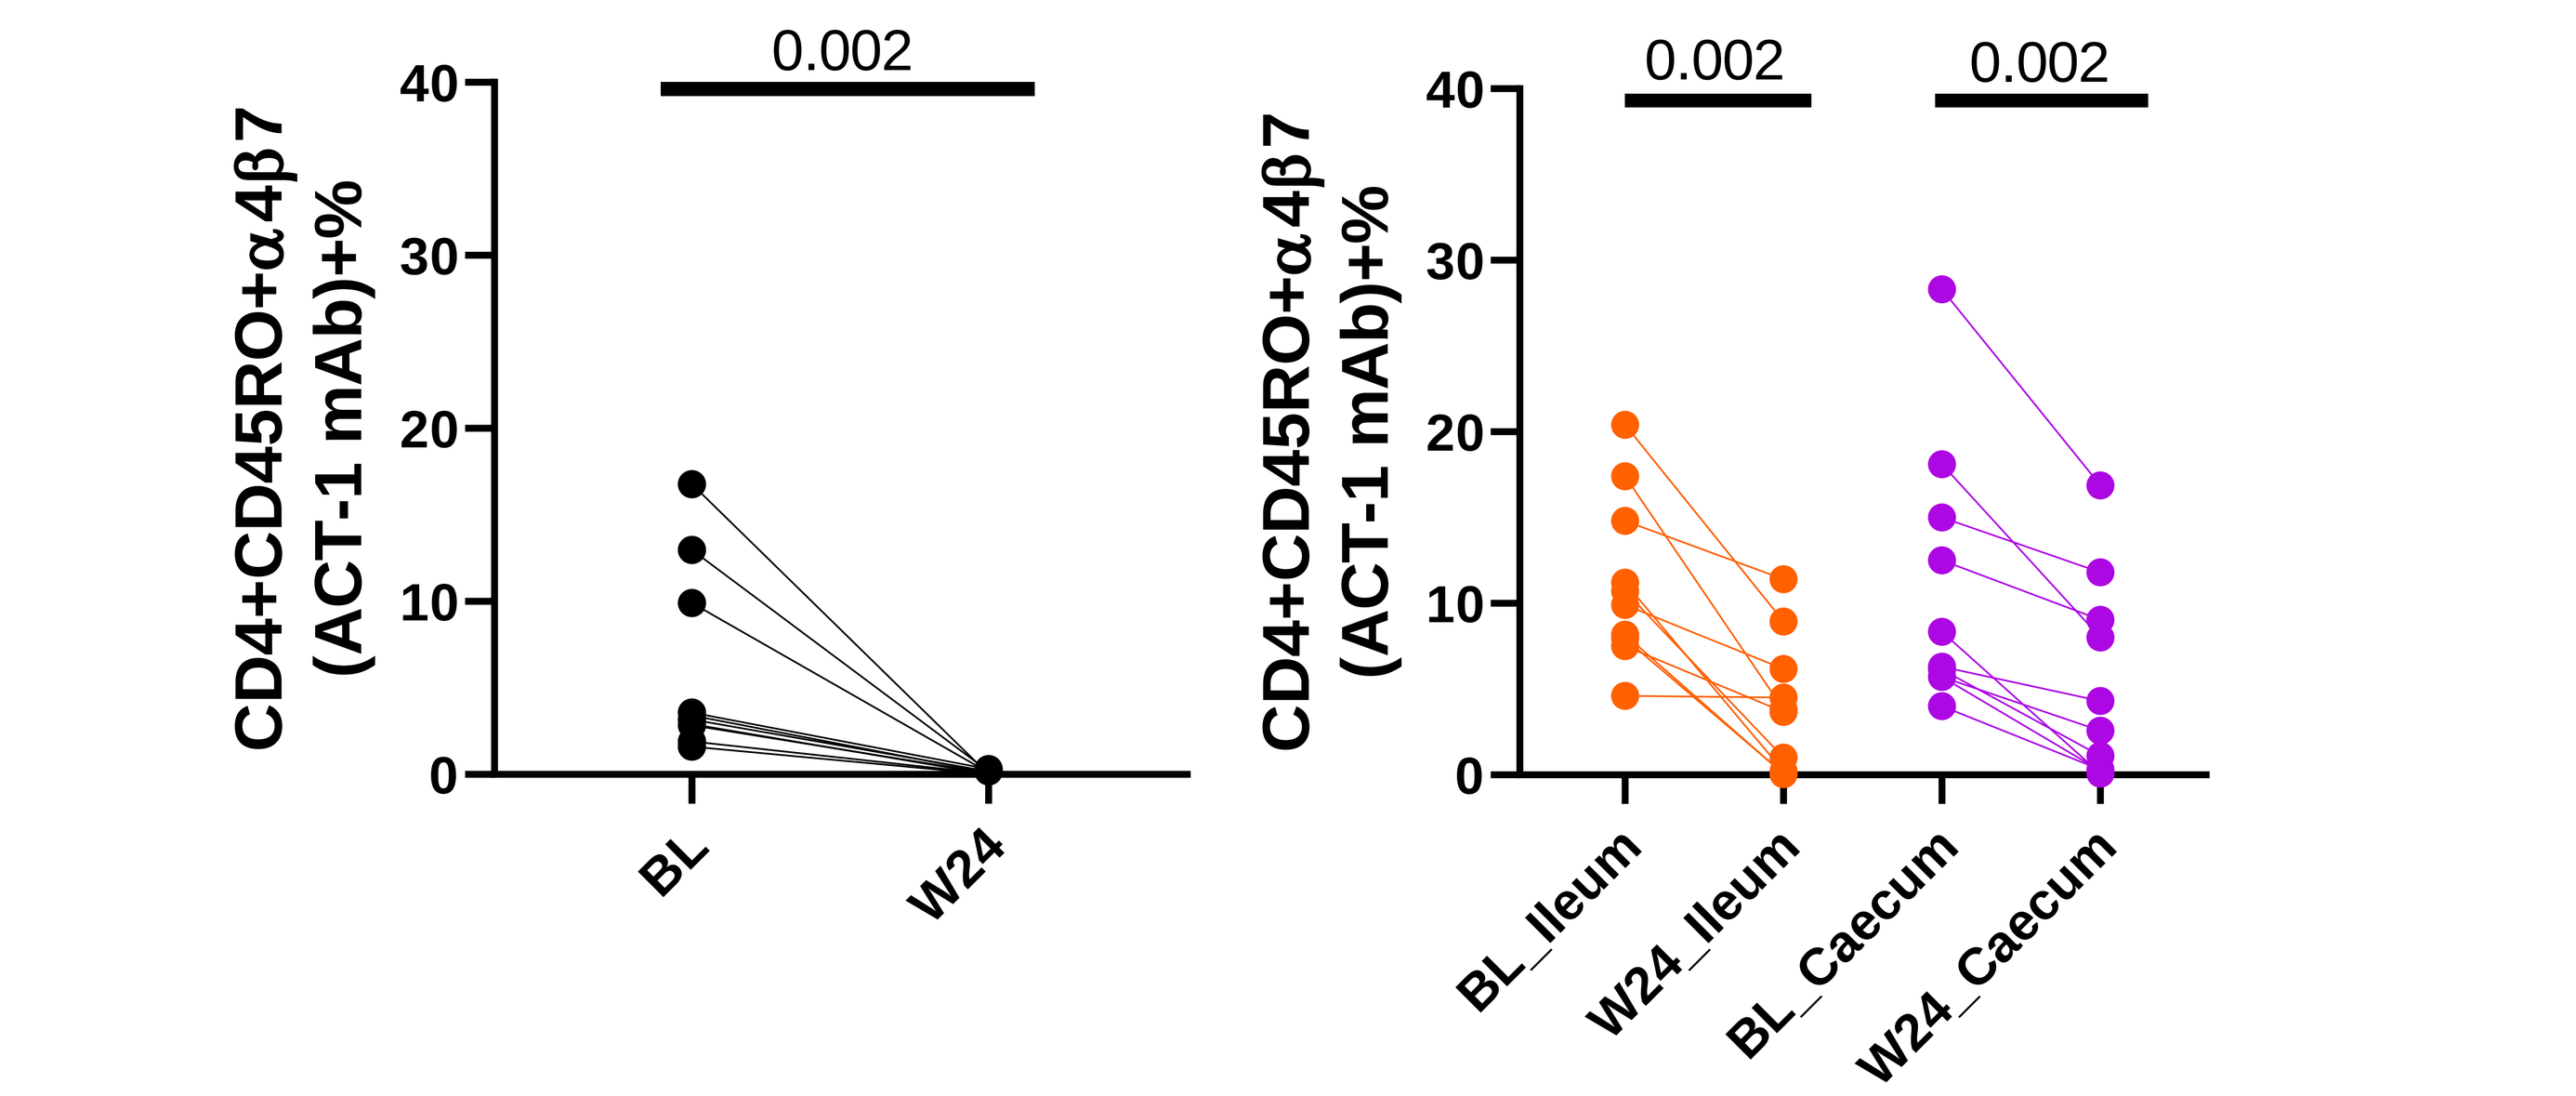
CD4+CD45RO+α4β7 (ACT-1 mAb)+%
0.002
BL W24
0.002 0.002
BL_Ileum W24_Ileum BL_Caecum W24_Caecum

B

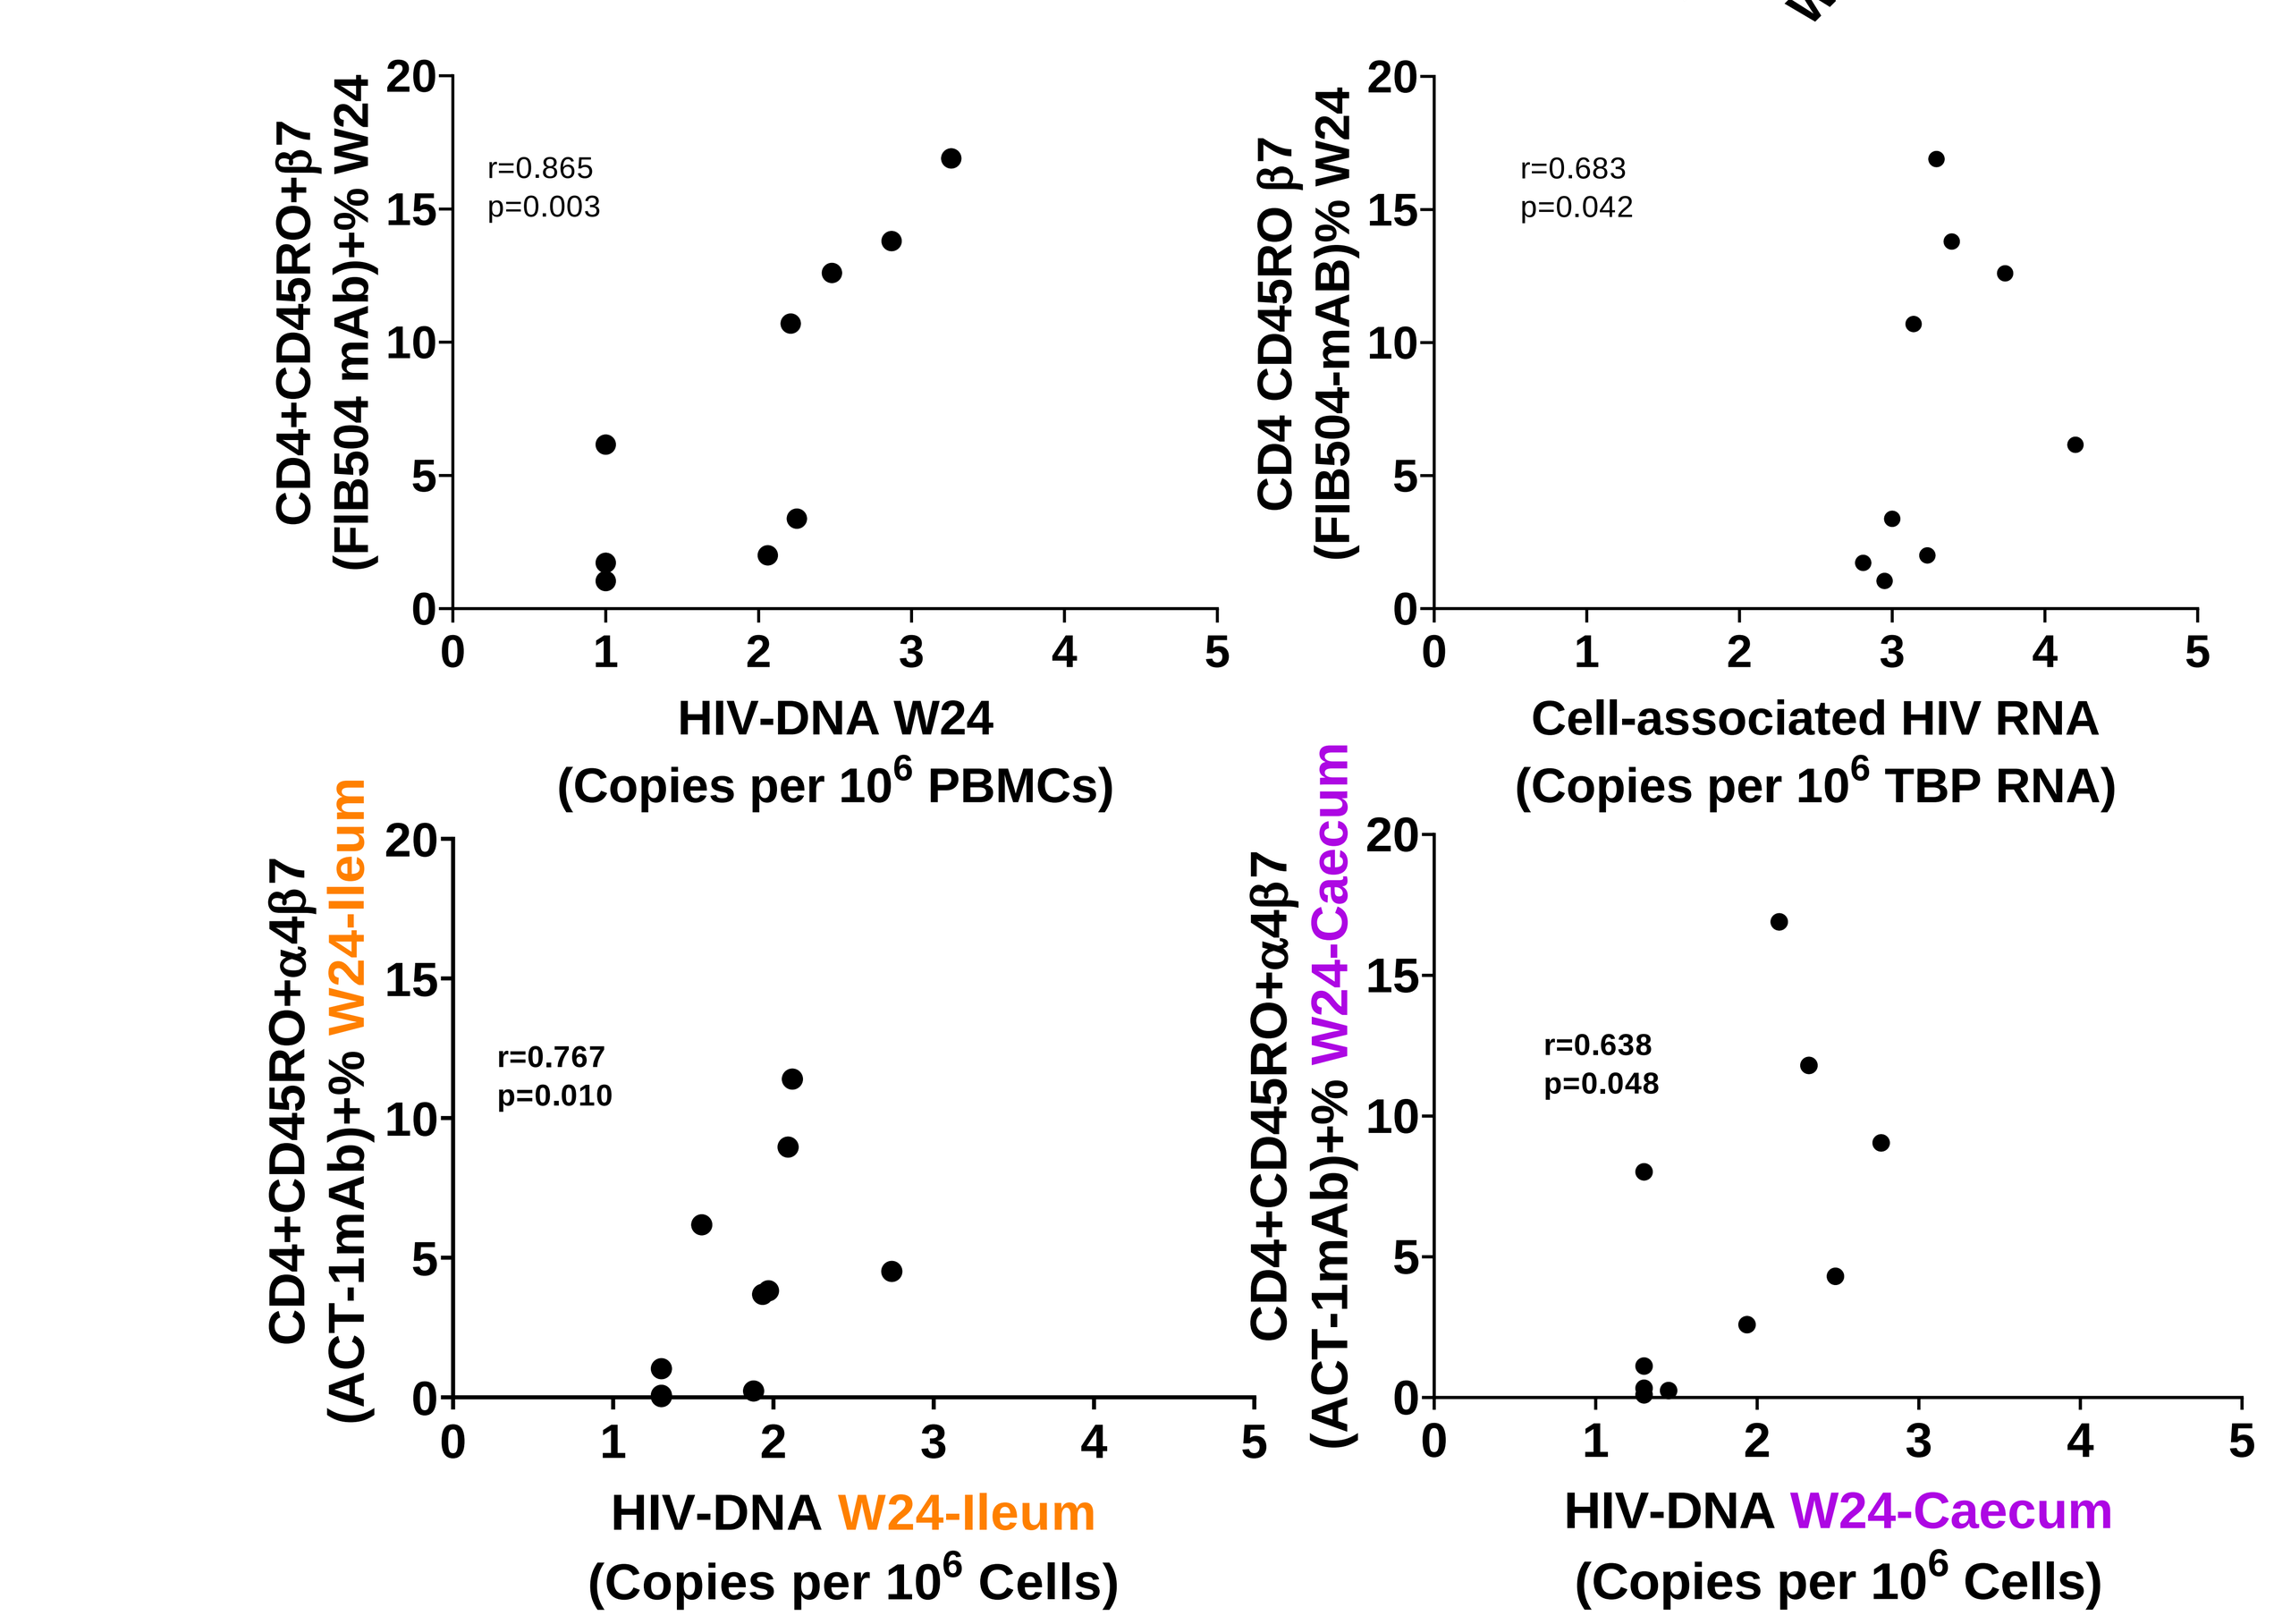
CD4+CD45RO+β7 (FIB504 mAb)+% W24
r=0.865
p=0.003
HIV-DNA W24 (Copies per 10^6 PBMCs)
CD4 CD45RO β7 (FIB504-mAB)% W24
r=0.683
p=0.042
Cell-associated HIV RNA (Copies per 10^6 TBP RNA)
CD4+CD45RO+α4β7 (ACT-1mAb)+% W24-Ileum
r=0.767
p=0.010
HIV-DNA W24-Ileum (Copies per 10^6 Cells)
CD4+CD45RO+α4β7 (ACT-1mAb)+% W24-Caecum
r=0.638
p=0.048
HIV-DNA W24-Caecum (Copies per 10^6 Cells)

5_ HIV reservoir on PBMCs was associated with expression of PD1 and TIGIT on CD4+ T-Cells at week 24.

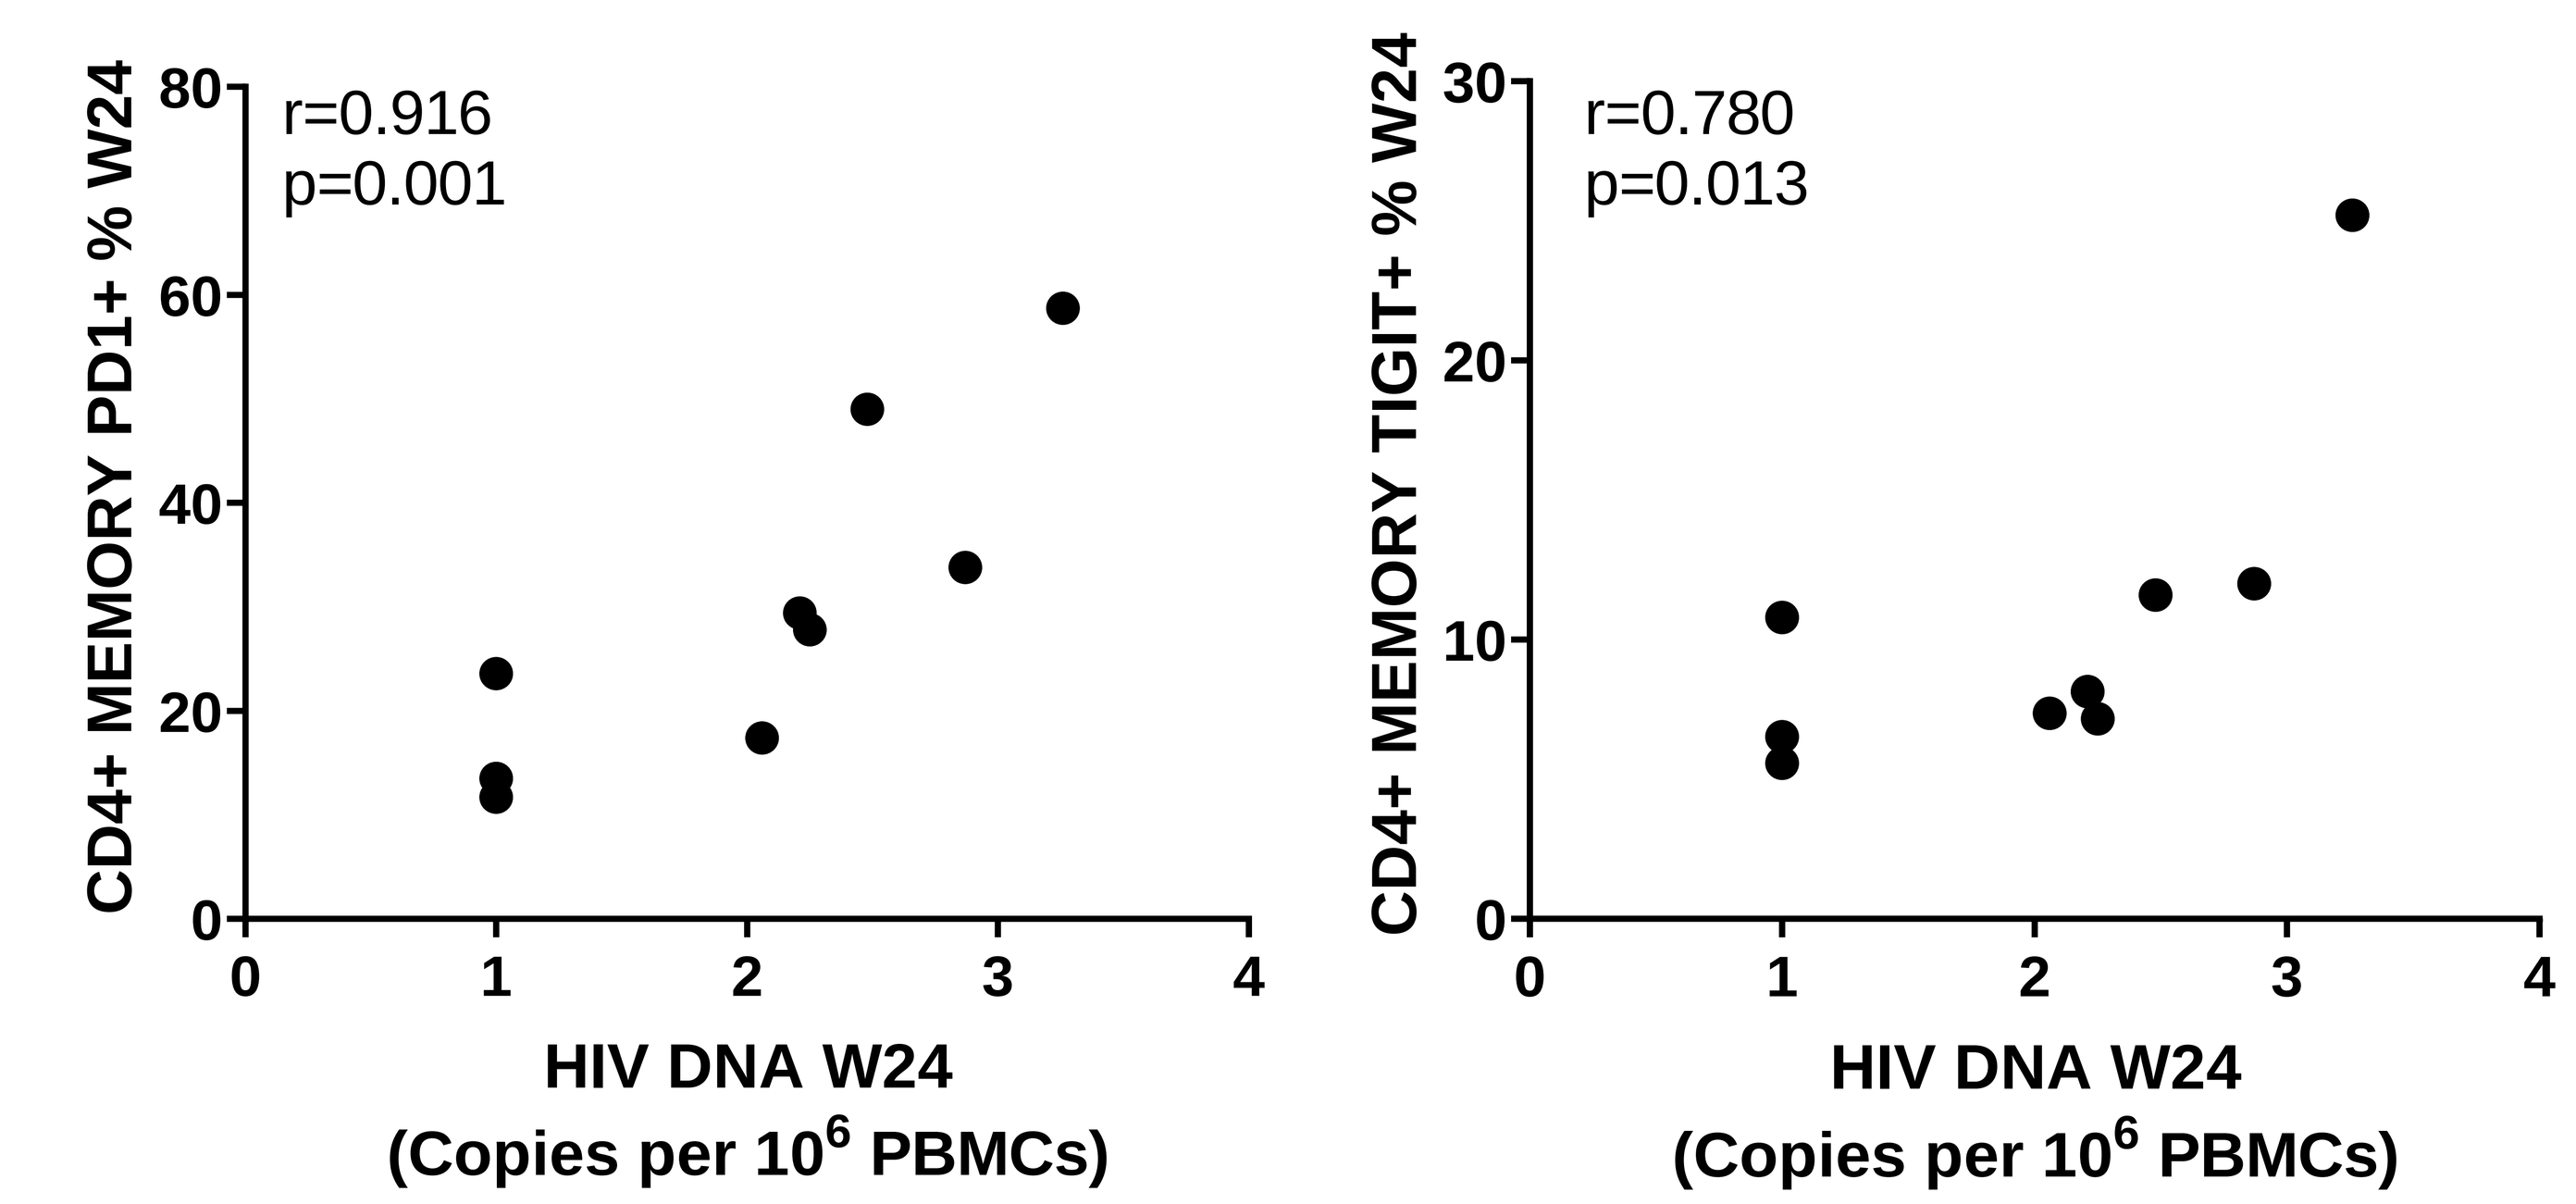
CD4+ MEMORY PD1+ % W24
r=0.916
p=0.001
HIV DNA W24 (Copies per 10^6 PBMCs)
CD4+ MEMORY TIGIT+ % W24
r=0.780
p=0.013
HIV DNA W24 (Copies per 10^6 PBMCs)

DISCUSSION

Vedolizumab was safe and well tolerated. No dramatic virological remission after ART interruption was found in naïve subjects. Reservoir establishment in PBMCs, IL and CC was associated with α4β7 and immune check point molecules expression.

Red de Investigación en Sida
Proyecto: RD16/0025/0020

IBIS INSTITUTO DE BIOMEDICINA DE SEVILLA

GOBIERNO DE ESPAÑA MINISTERIO DE CIENCIA, INNOVACIÓN Y UNIVERSIDADES Instituto de Salud Carlos III

UNIÓN EUROPEA Fondo Europeo de Desarrollo Regional "Una manera de hacer Europa"