HIV-1 viral reservoir disruption with panobinostat and IFN-α

00357

Marie Armani-Tourret[1], Ciputra A. Hartana[1], Yelizaveta Rassadkina[1], Ce Gao[1], Katrina Shea[2], Amy Sbrolla[2], Xu Yu[1], Rajesh T. Gandhi[2], Daniel R. Kuritzkes[3], Mathias Lichterfeld[1]

[1]Ragon Institute of MGH, MIT and Harvard, Cambridge, MA, United States, [2]Massachusetts General Hospital, Boston, MA, United States, [3]Brigham and Women's Hospital, Boston, MA, United States

BACKGROUND

Current antiretroviral therapies (ART) are unable to eradicate HIV-1, mainly because the virus persists in latently infected cells that can fuel viral rebound after treatment interruption. Pharmacological reactivation of viral transcription may sensitize viral reservoir cells to immune-mediated killing and reduce the long-term persistence of virus-infected CD4+ T cells.

The **ACTIVATE** study is a prospective, randomized clinical trial in which the histone deacetylase inhibitor **panobinostat** (PBT) is administered as a latency-reversing agent in combination with **pegylated IFN-α2a** (IFN-α2a) as an innate immune modulator to reduce the viral reservoir.

METHODS

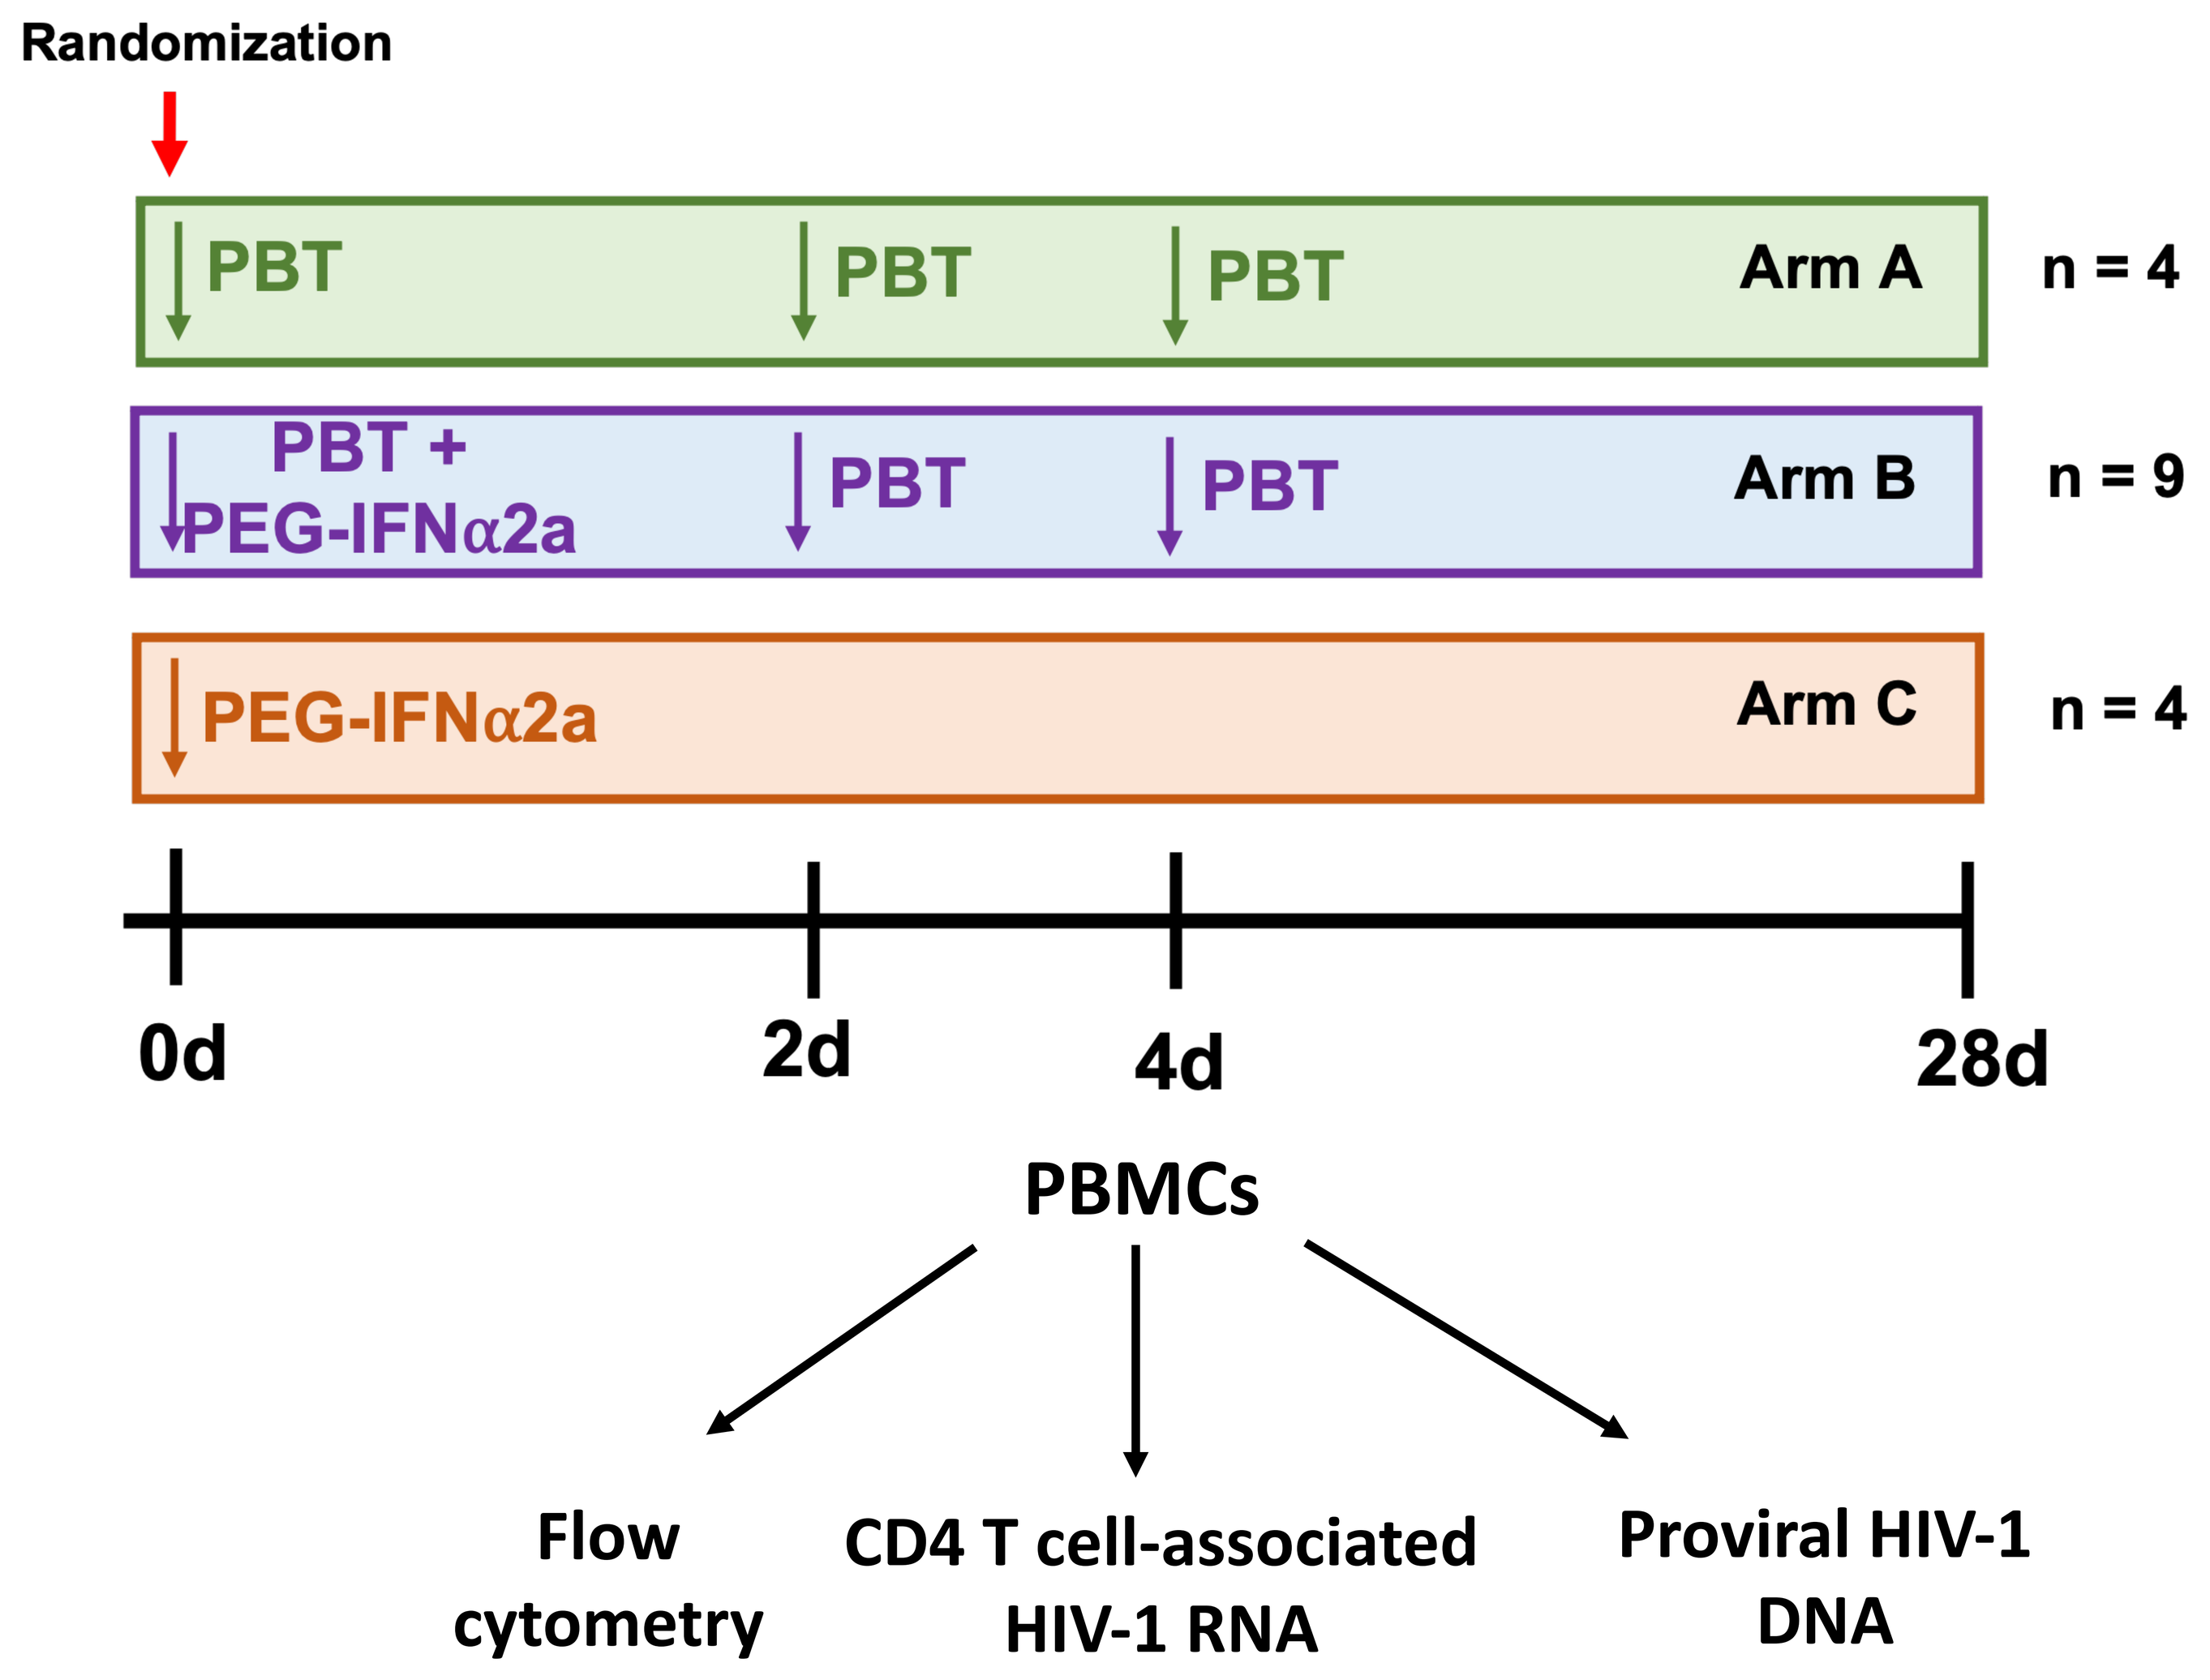
Randomization
PBT
PBT
PBT
Arm A
n = 4
PBT + PEG-IFNα2a
PBT
PBT
Arm B
n = 9
PEG-IFNα2a
Arm C
n = 4
0d
2d
4d
28d
PBMCs
Flow cytometry
CD4 T cell-associated HIV-1 RNA
Proviral HIV-1 DNA

Figure 1. Study design. ART-treated participants were randomized to receive PBT alone (n=4), the combination of PBT and IFN-a2a (n=9) or IFN-a2a alone (n=4). H3 acetylation and innate and adaptive cellular immune responses were analyzed by flow cytometry. CD4 T cell-associated HIV-1 RNA was quantified by RT-ddPCR assay. Proviral HIV-1 DNA was analyzed using the intact proviral DNA assay (IPDA).

RESULTS

• Panobinostat treatment increases histone acetylation and viral replication

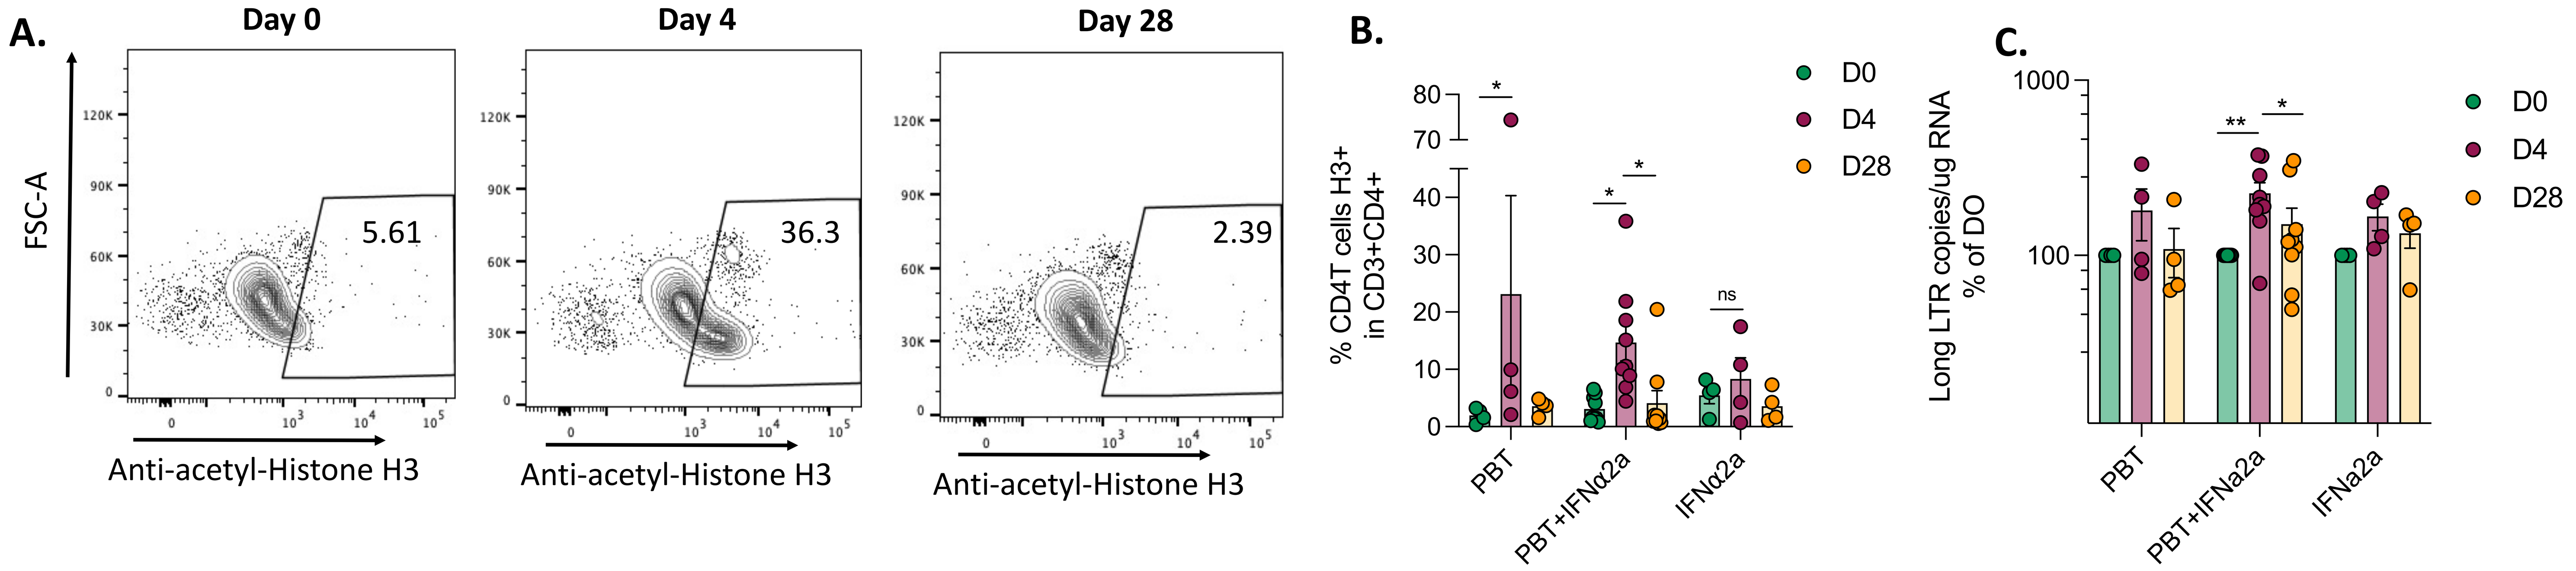
A.
Day 0
Day 4
Day 28
FSC-A
5.61
36.3
2.39
Anti-acetyl-Histone H3
B.
% CD4T cells H3+ in CD3+CD4+
C.
Long LTR copies/ug RNA % of DO
D0
D4
D28
PBT
PBT+IFNα2a
IFNα2a

Figure 2. A. Representative dot plots of histone acetylation following treatment with PBT at D0, D4 and D28. **B.** Bar charts reflecting proportions of acetylated histone H3-positive CD4 T cells in total CD4 T cells, in patients treated with PBT or pegIFNa alone or in combination (PBT+IFNa). *p<0.05; Friedman Test. **C.** Using RT-ddPCR *(Yukl et al.2018)*, Long-LT HIV transcripts were measured at D0, D4 and D28. The results represented are normalized with the value obtained at D0 (100%). *p<0.05, **p<0.01; Friedman Test.

• IFN- α treatment induces immune activation

*** A. Increased proportion of CD4 and CD8 T cells *(data not shown)* expressing GrzA, GrzB and Perforin**

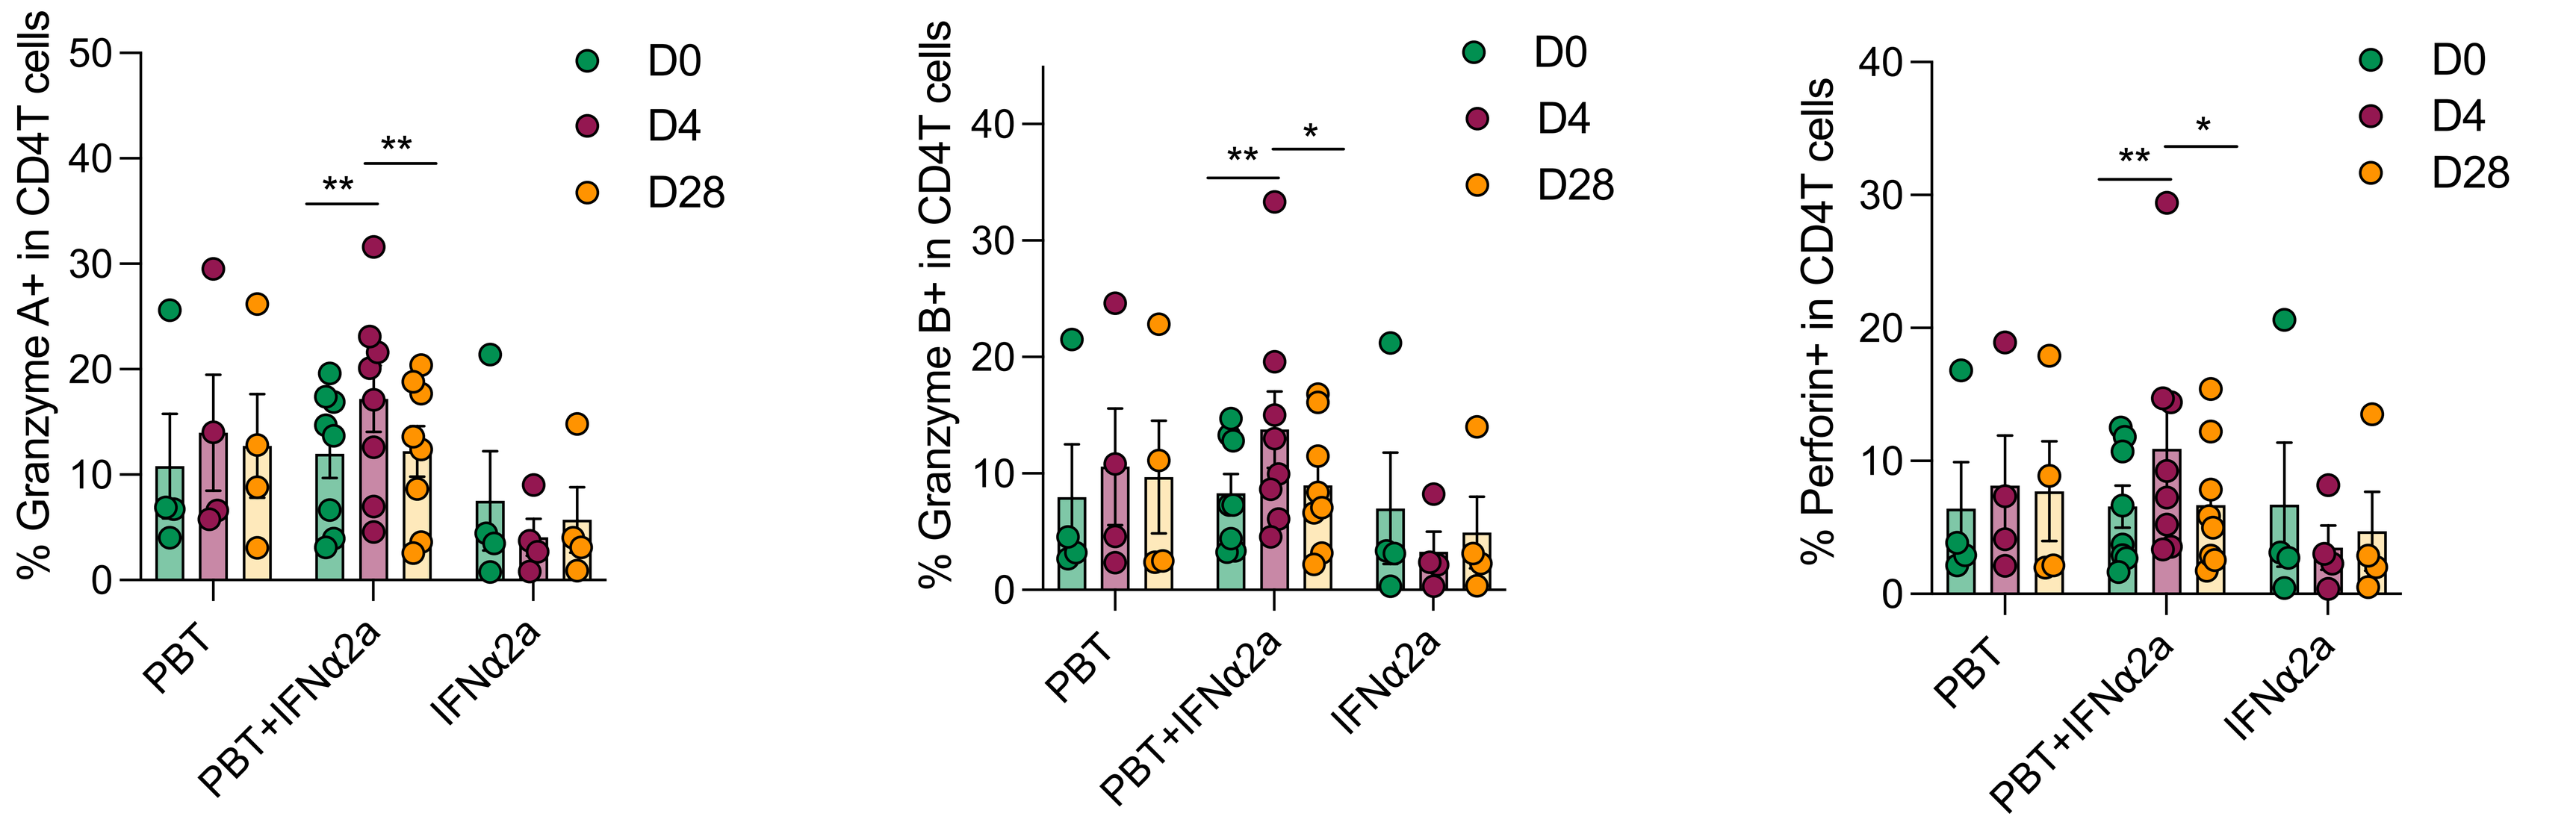
% Granzyme A+ in CD4T cells
% Granzyme B+ in CD4T cells
% Perforin+ in CD4T cells
D0
D4
D28
PBT
PBT+IFNα2a
IFNα2a

*** B. Increased proportion of mDC2-3 expressing co-stimulatory molecules**

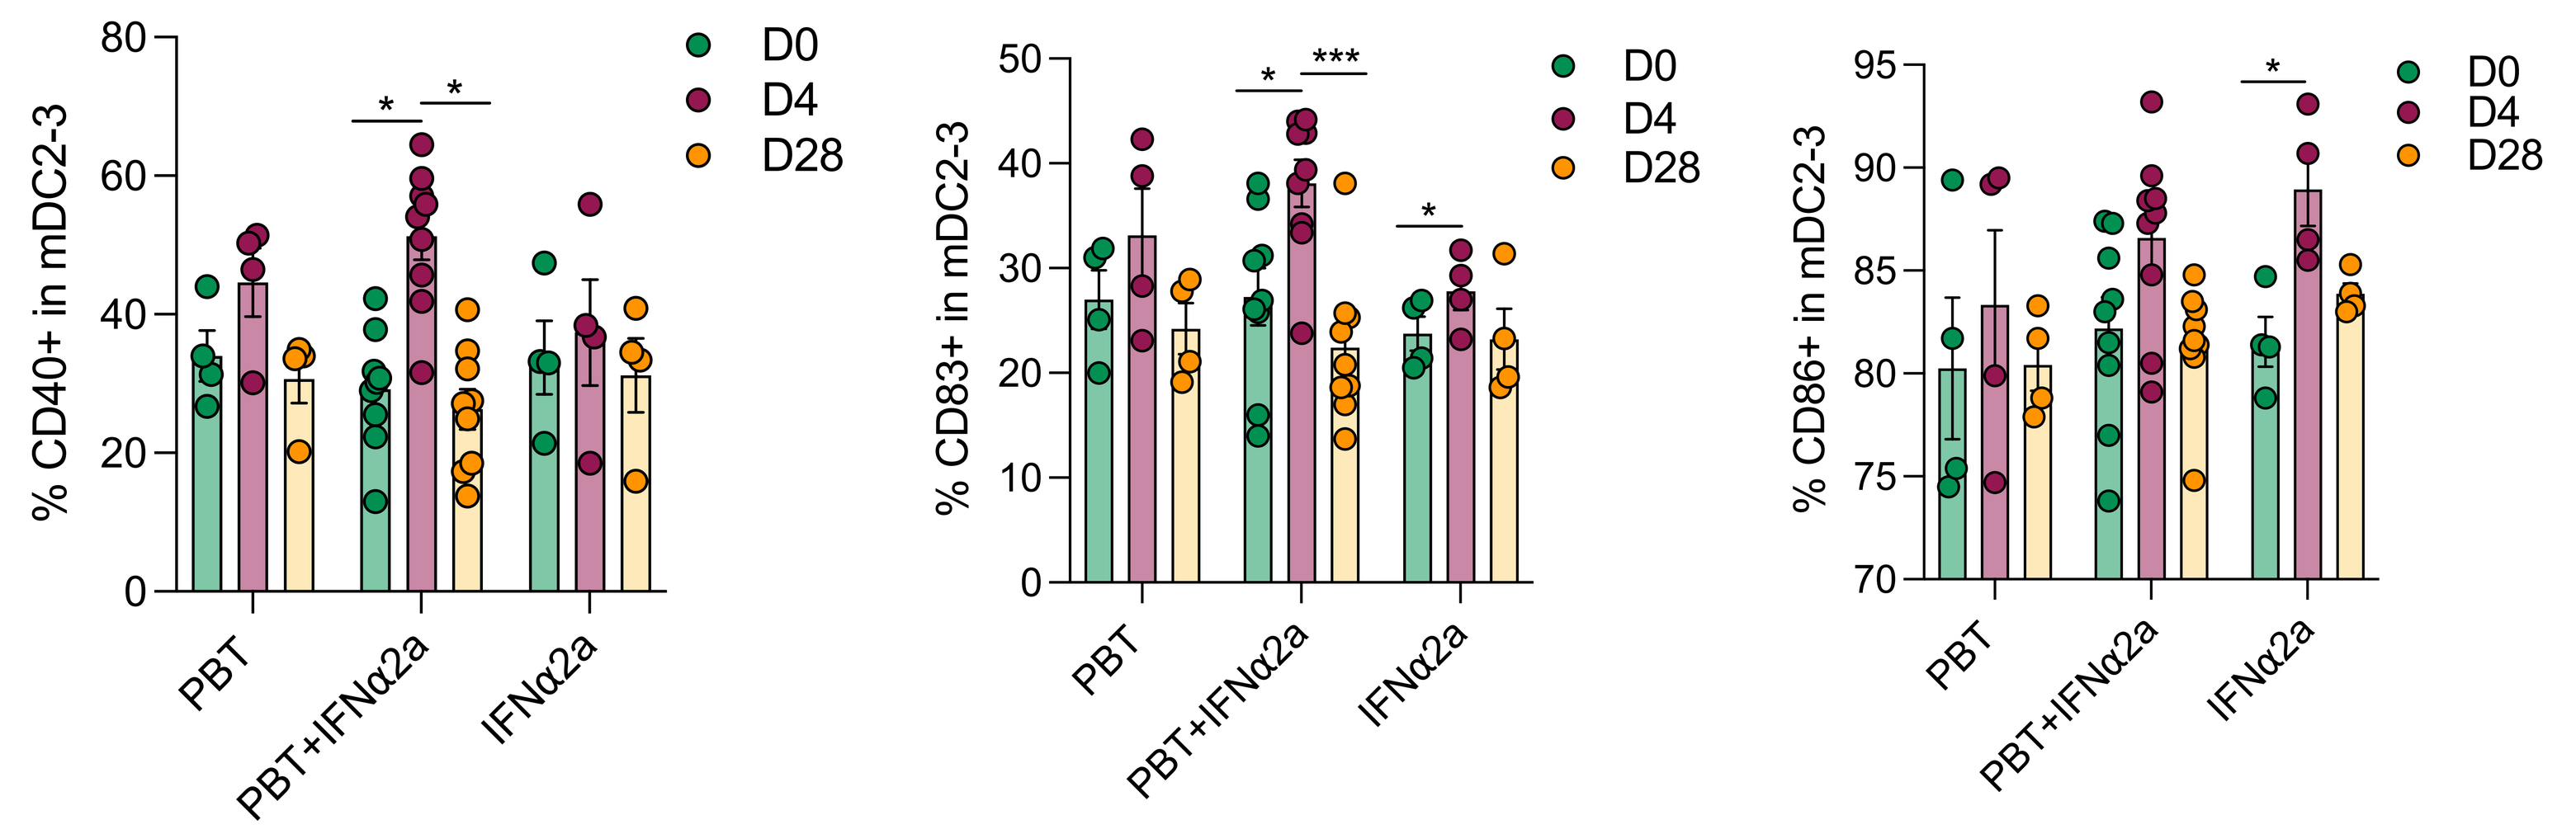
% CD40+ in mDC2-3
% CD83+ in mDC2-3
% CD86+ in mDC2-3
D0
D4
D28
PBT
PBT+IFNα2a
IFNα2a

*** C. Increased proportion of pDCs expressing co-stimulatory molecules and the migratory marker CCR7**

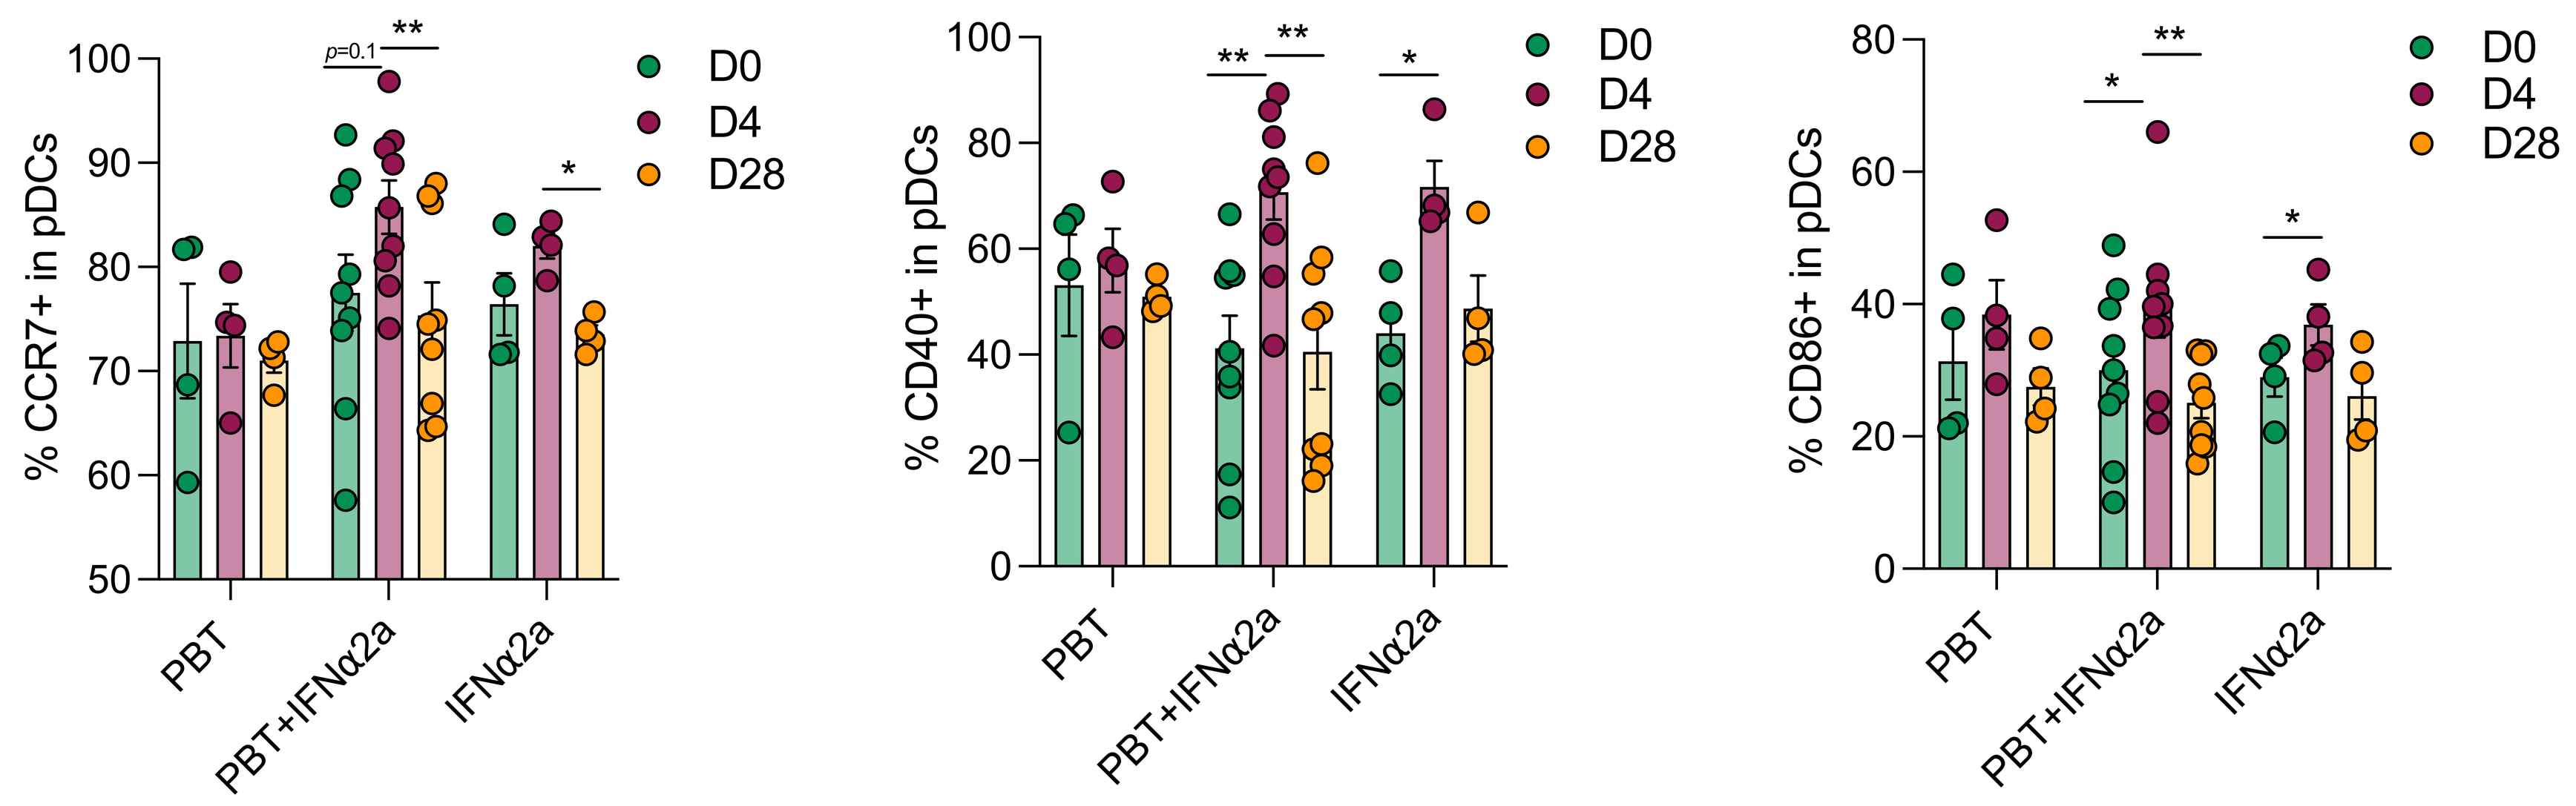
% CCR7+ in pDCs
% CD40+ in pDCs
% CD86+ in pDCs
D0
D4
D28
PBT
PBT+IFNα2a
IFNα2a

*** D. Increased frequency of activated CD38+ and NKp30+ cytotoxic NK cells (CD16+ CD56+)**

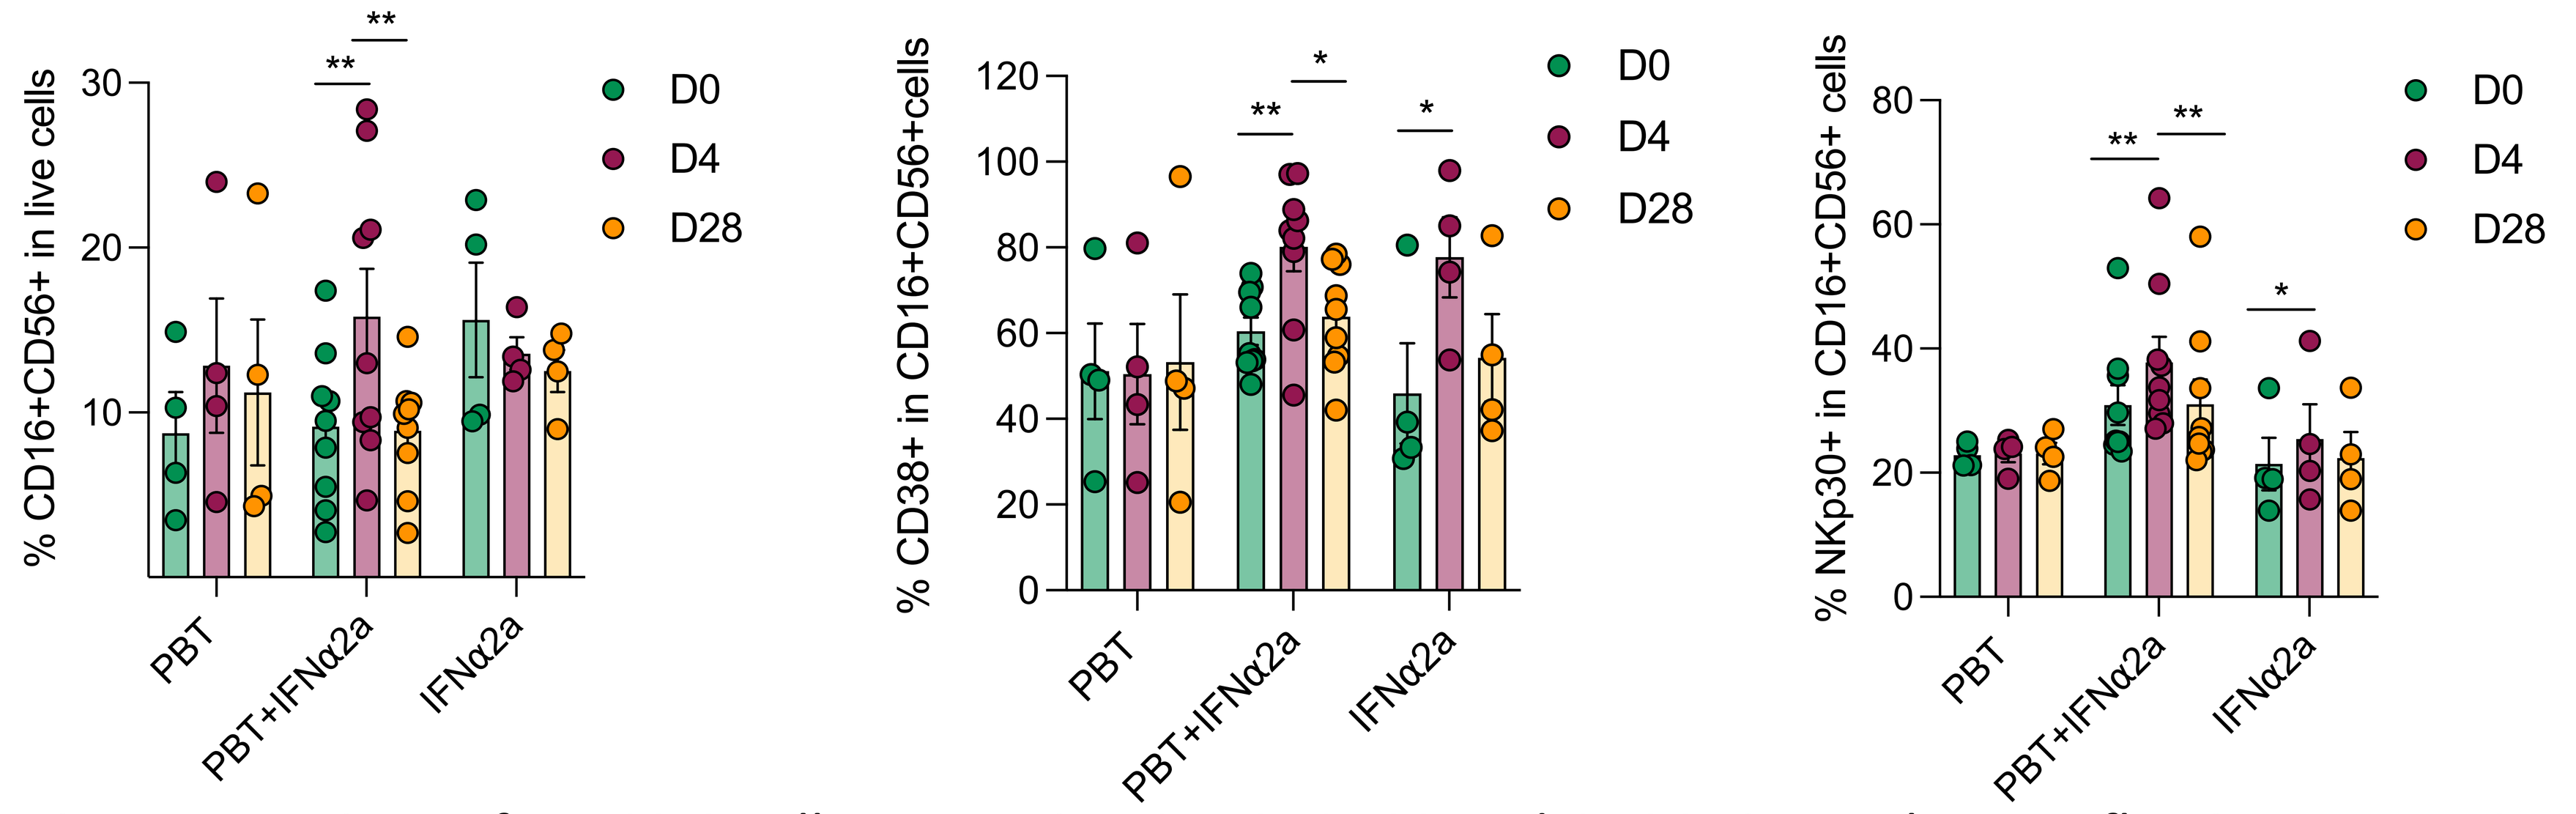
% CD16+CD56+ in live cells
% CD38+ in CD16+CD56+cells
% NKp30+ in CD16+CD56+ cells
D0
D4
D28
PBT
PBT+IFNα2a
IFNα2a

Figure 3. A-Bar charts reflecting proportions of CD4 T cells expressing Granzyme A, Granzyme B and Perforin. **B-**Bar charts reflecting proportions of mDC2-3 cells expressing CD40, CD83 and CD86. **C-**Bar charts reflecting proportions of pDCs cells expressing CCR7, CD40 and CD86. **D-** Bar charts reflecting proportions of NK cells expressing CD16+CD56+ then proportions of these CD16+CD56+ cells expressing CD38 and Nkp30.*p<0.05, **p<0.01; Friedman Test.

• Combined treatment do not induce changes in HIV-1 DNA level

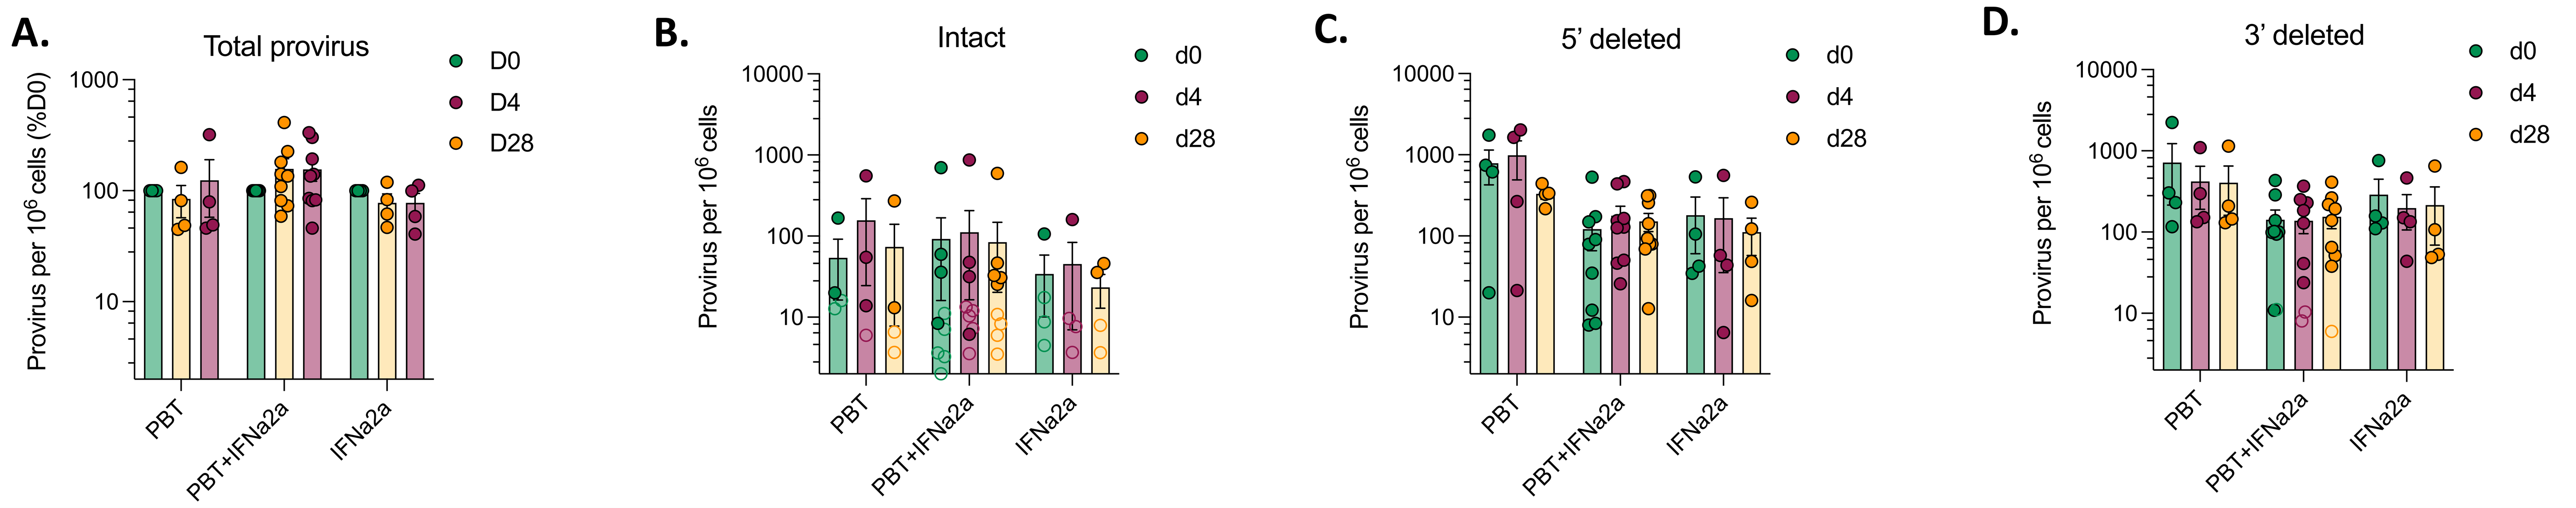
A.
Total provirus
Provirus per 10⁶ cells (%D0)
B.
Intact
C.
5' deleted
D.
3' deleted
Provirus per 10⁶ cells
D0
D4
D28
d0
d4
d28
PBT
PBT+IFNa2a
IFNa2a

Figure 4. A-D. Bar charts reflecting absolute frequencies of total (**A**), intact (**B**), 5'deleted (**C**) and 3' deleted (**D**) proviruses per 10^6 total CD4 T cells as measured by the Intact Proviral DNA Assay (IPDA). For **A**, the values have been normalized to D0 (100%).

CONCLUSIONS - Results from ACTIVATE study indicate that the study medication induces HIV-1 transcription and augments innate and adaptive immune effector cells, without appreciably affecting HIV-1 DNA levels in our current analysis.
Further studies will be conducted to evaluate possible changes in proviral positioning relative to activating epigenetic chromatin features.