995 - EVALUATION OF TARGETED HIV SELF-TEST KIT DISTRIBUTION VIA A DIGITAL VENDING MACHINE

A. Pollard[3], G. Dean[1], N. Pilarski[1], C. Peralta[2], L. Rodriguez[4], S. Soni[1], C. D. Llewellyn[3], J. Vera[1,3]

[1]Brighton and Sussex University Hospitals [2]University of Brighton [3]Brighton and Sussex Medical School [4]The Open University

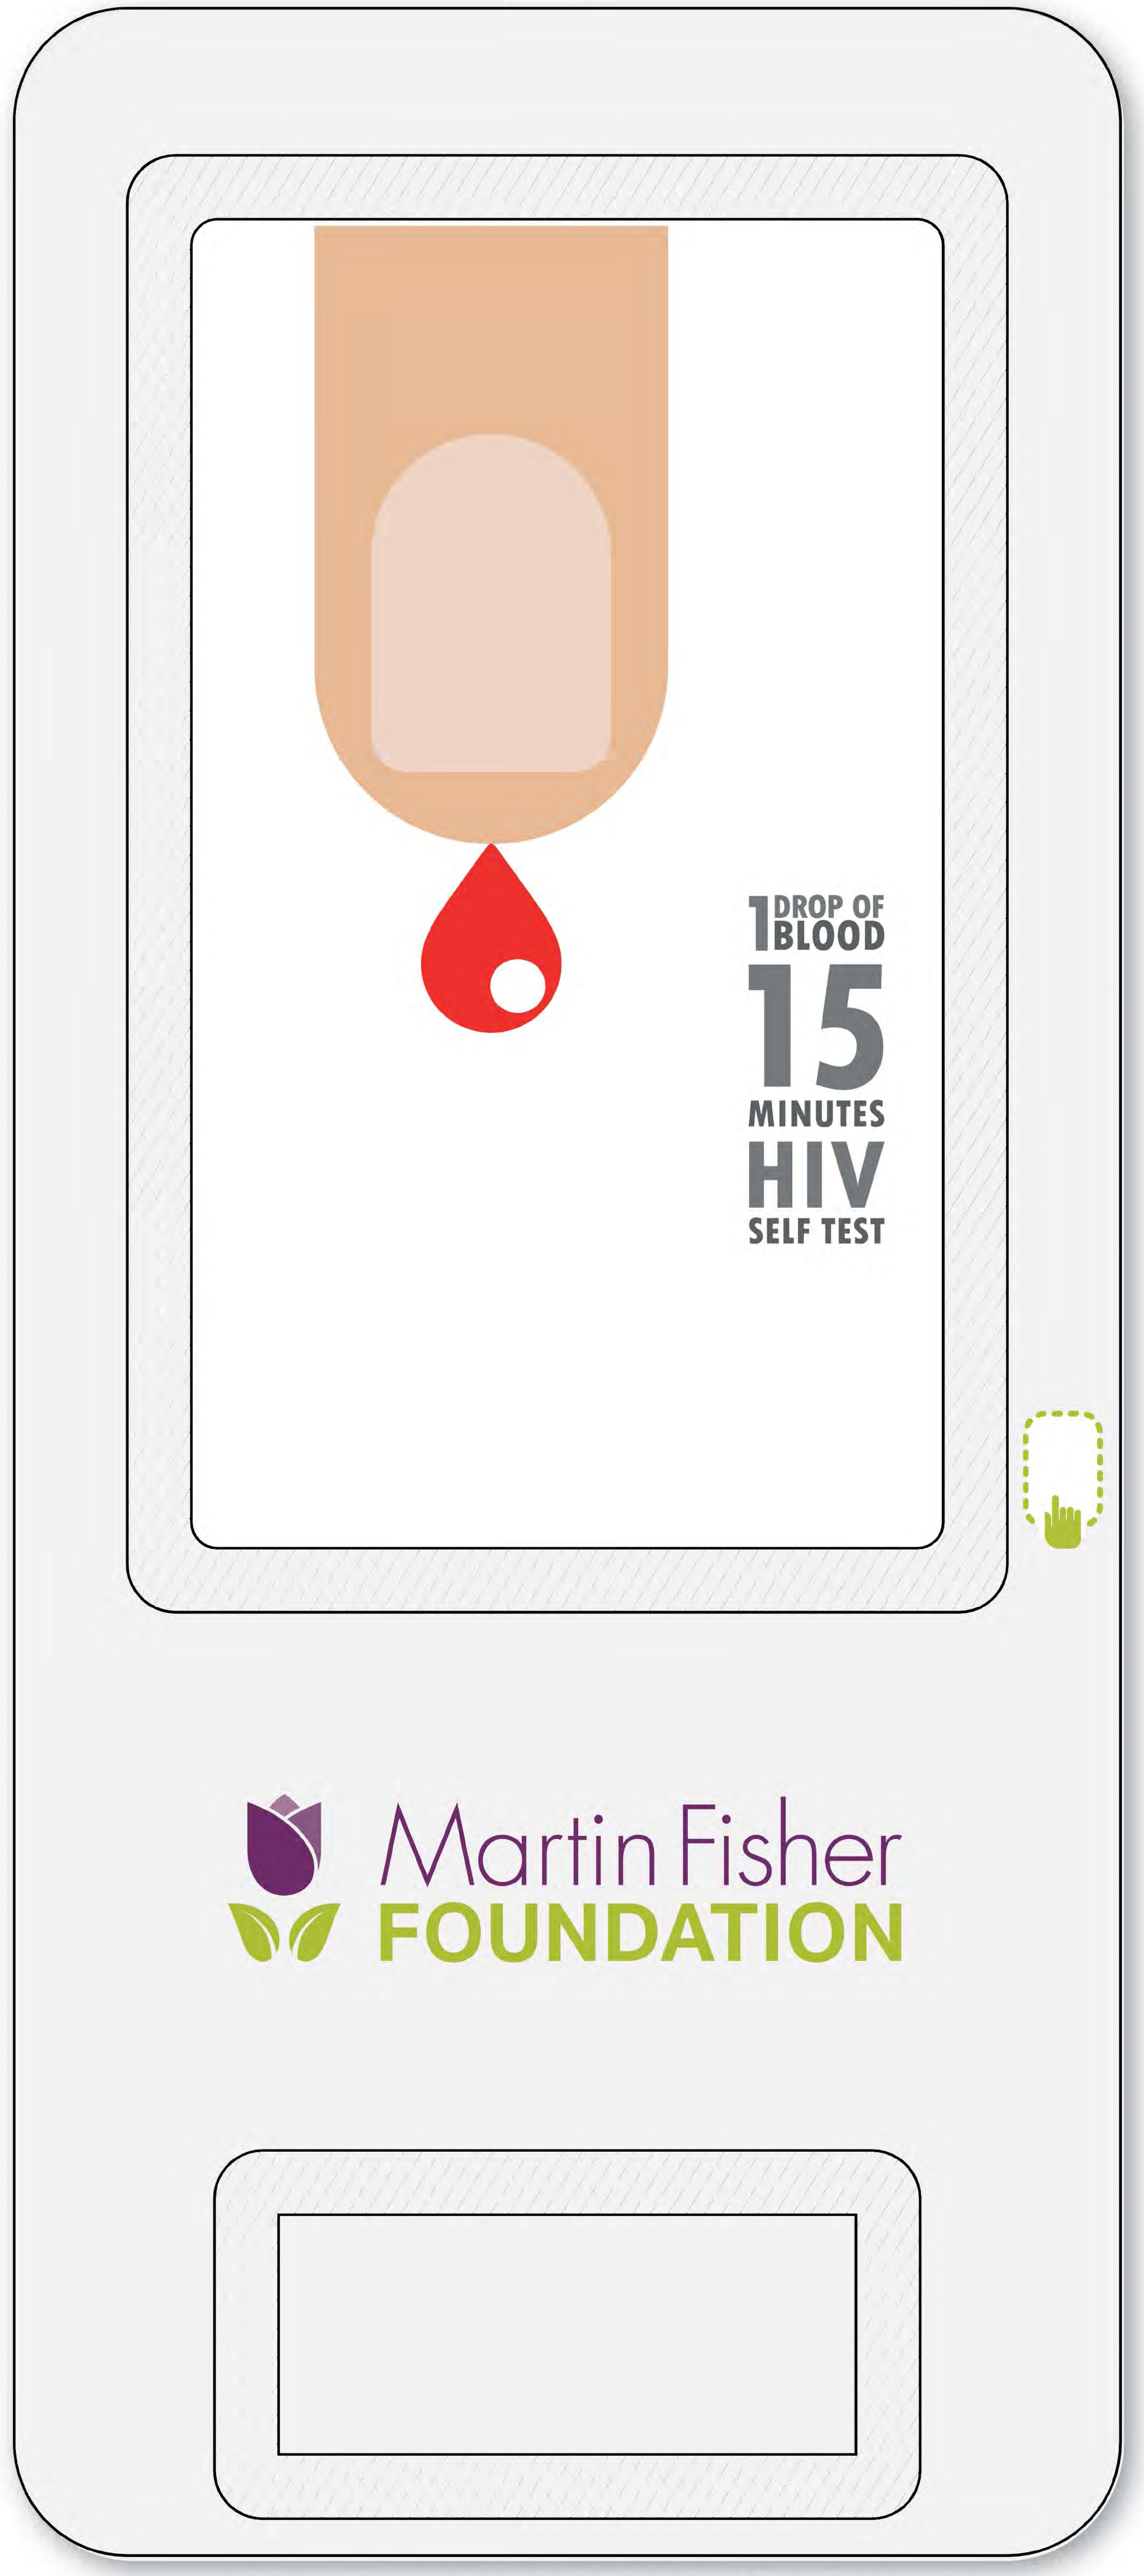
1 DROP OF BLOOD
15
MINUTES
HIV
SELF TEST
Martin Fisher
FOUNDATION

1:1 scale illustration of the HIV Self-test Vending Machine

We developed a pilot service promoting BioSURE® instant-result HIV self-testing kits (one drop of blood; 99.7% accurate[i]) to MSM via a bespoke digital vending machine in a gay sauna (Brighton, UK). This included co-design workshops to develop a campaign and digital interface to the vending machine, and extensive evaluation of user experience.

INTRODUCTION

HIV infection rates among men who have sex with men (MSM) are increasing and the uptake of STI and HIV testing in conventional clinical settings is suboptimal.
An estimated 14% of UK MSM with HIV are thought to be undiagnosed, and 30% of new diagnoses in MSM are late diagnoses[ii],[iii]. Innovative strategies are required to increase uptake of HIV testing.
One approach is to increase access to instant result HIV self-testing kits, enabling opportunities for providers to reach populations not actively engaged with clinical services.

METHODS

A mixed-methods evaluation included early co-design workshops, cross-sectional surveys and qualitative interviews to assess uptake and acceptability.

Co-Design workshops

Men using a sex-on-premises sauna were surveyed to assess acceptability and feasibility of a vending machine distributing HIV self-testing kits. Facilitated co-design workshops were held with volunteers and designers in LGBT community venues, discussing proposals for interventions.
The co-design workshops utilised service design tools such as personas (creating 'characters' to explore individual's thoughts and behaviours); the construction of 'user journeys'; and mock-ups of vending machine designs.
These workshops led to the development of a promotional campaign and the digital interface for the vending machine.

The Vending Machine

The bespoke digital vending machine was installed in a gay sauna. The interface included a touch-sensitive screen requiring users to enter demographics and a mobile phone number in order to receive a text (SMS) message with a four-digit code, which was entered into the machine to release the free self-test kit at no financial cost.
A leaflet with the kit directed users to onward care: reactive results to local on-line and telephone advice, and linkage to clinical services; negative results were encouraged to re-test to cover window periods.

Cross-sectional on-line survey

The text (SMS) message sent to user's phones also included a link to an on-line survey of 12 questions exploring demographics, HIV testing history, frequency of condomless sex/numbers of partners (https://brighton.onlinesurveys.ac.uk/testing).

Qualitative Interviews

Two phases of semi-structured interviews were conducted:

- 13 X face-to-face interviews with customers approached in the sauna lounge, about attitudes and acceptability of vending machines for HIV self-testing.
- 10 X 30-min telephone interviews with users of the vending machine and self-test, who had provided contact details via the on-line survey.

RESULTS

Key themes relating to visual campaign options were: a sense of community and support; clinical versus community settings; and giving clear information. There was excellent engagement in the process and many participants generated their own style, wording, or schema for the campaign.

Table 1 **Factors to encourage Self-testing:**	AWARENESS	"knowing more about self-test would persuade me to try"
	AVAILABILITY	"like more available at local establishments, uni, pubs, clubs"
	CONFIDENCE	"how easy it may be" QN8 "Confidence in result, ease of use"

Table 2 **Concerns about Self-testing:**	FAITH	"little faith in reliable result" "faith in results" to be sure it is done properly"
	FAMILIARITY	"did not know it existed"
	FEAR	"scared of blood" "may hurt myself taking sample" "fear of doing it wrong"
	SCREENING	"advice on sexual health, STI screening"
	SUPPORT	"if it was positive I'd want to talk to someone" "safe environment"

The participant-generated discussions developed a 'user journey' from options for HIV testing, to decision-making priorities, and routes to action. Participants explored benefits and disadvantages for the following populations identified as core targets for self-testing:

- Clinic attenders with high HIV risks
- MSM not engaged with clinical services
- Framing the campaign within the 'gay scene'.

The vending machine in the entrance of the gay sauna successfully dispensed 204 tests between 16th June 2017 and 31st December 2017 (average one/day).
The uptake of self-tests was higher via the vending machine compared to community outreach testing at the sauna over the same period (35 V 4.56 tests per month).

Demographics of vending machine users

		Previous HIV test		
Total self-tests vended	Mean Age	Never tested	More than 1 year ago	Within last year
204	31 (range: 18-70)	4%	17%	79%

48 survey respondents:

- Mean age: 42; range 17-70
- 34.7% last tested more than 12 months ago or Never
- 54.2% had condomless anal sex with a new or casual partner in the last 6 months; and 34% had 5 or more condomless partners in the last year
- 93.5% would recommend self-testing to others (6.5% [n=3] were unsure; none would not recommend).

29 of 48 survey respondents left free-text comments indicating high levels of acceptability with the vending machine and HIV self-testing:

A breakthrough in testing. Not just the test itself but the availability.
Felt safe and comfortable and well explained step by step.
The vending machine is an excellent idea!
Easy, quiet and private way of testing, but also very easy to get hold of. Would recommend expanding to other venues!
Firstly, thank you. Secondly, I tend to use the tests as preventative by testing when I ha new relationship and about to go bareback for the first time in that relationship. Ability to get two test kits or ability to get one test kit designed for two people would be ace.
Very quick and easy, would like to be able to order such a kit for free home delivery too.
A very easy test to use and convenient.
It's a great way to do it. The sexual health clinic is a difficult and intrusive experience with long waits.
Quick and easy no waiting for lab results!
It's good for those people who are shy or scared to go see someone.
Amazing service. Wish there were similar services for other STDs.
So much easier and accessible, less embarrassment. Great tool to use!!
This is absolutely amazing. Please continue this as it helps me to just grab a box and go home and do it in my own comfort without panicking and having to tell my entire sexual history.

Participants interviewed at the sauna (n=13) expressed high levels of acceptability for the convenience and discretion of both the vending machine and HIV self-testing.
Telephone interviewees, who had used the vending machine and self-test (n=10), all had previous experience of clinic-based testing, and most had also used community outreach testing services.

Self-tests were typically used within a few hours or days. Some participants additionally kept a test kit for use after anticipated future risks; but often with limited understanding of the (3-month) window period.

Age	Ever tested for HIV	Ever tested at clinics	Ever tested via community outreach	Ever tested via community outreach at a sauna
Mean age 40 (Range: 26-53)	Yes 10 No 0	Yes 10 No 0	Yes 7 No 3	Yes 7 No 3

DISCUSSION

Co-Design workshops
User co-design of this innovation has been central to the development of an acceptable and effective interface[iv].

The Vending Machine
Vending machines have not previously been used to deliver HIV self-testing kits in the UK. A single USA study using Orasure® rapid HIV tests is not comparable due to methodological differences[v].

Cross-sectional on-line survey
This study supports our previous research demonstrating high levels of acceptability for HIV self-testing as well as an unmet need[vi].
Vending machine distribution reached eligible users with high levels of HIV risk behaviour, many with low uptake of HIV testing.

Qualitative Interviews
Both phases of interviews reported high levels of acceptability and accessibility. However, concerns were identified by users in two key areas:

- The emotional and clinical risks of getting a reactive result in isolation, without immediate support, and with potential loss to care.
- The limitation to HIV testing alone, with varied levels of concern about other STIs.

Additional concerns were also identified by researchers:

- The convenience and discretion of HIV self-testing had displaced some participants' routine comprehensive STI screening at clinics.
- Several interviewees had tested with sexual partners as part of risk assessment prior to condomless sex – often without understanding the risks of high viral-load early in the window period.
- Knowledge and understanding of the window-period was frequently poor.

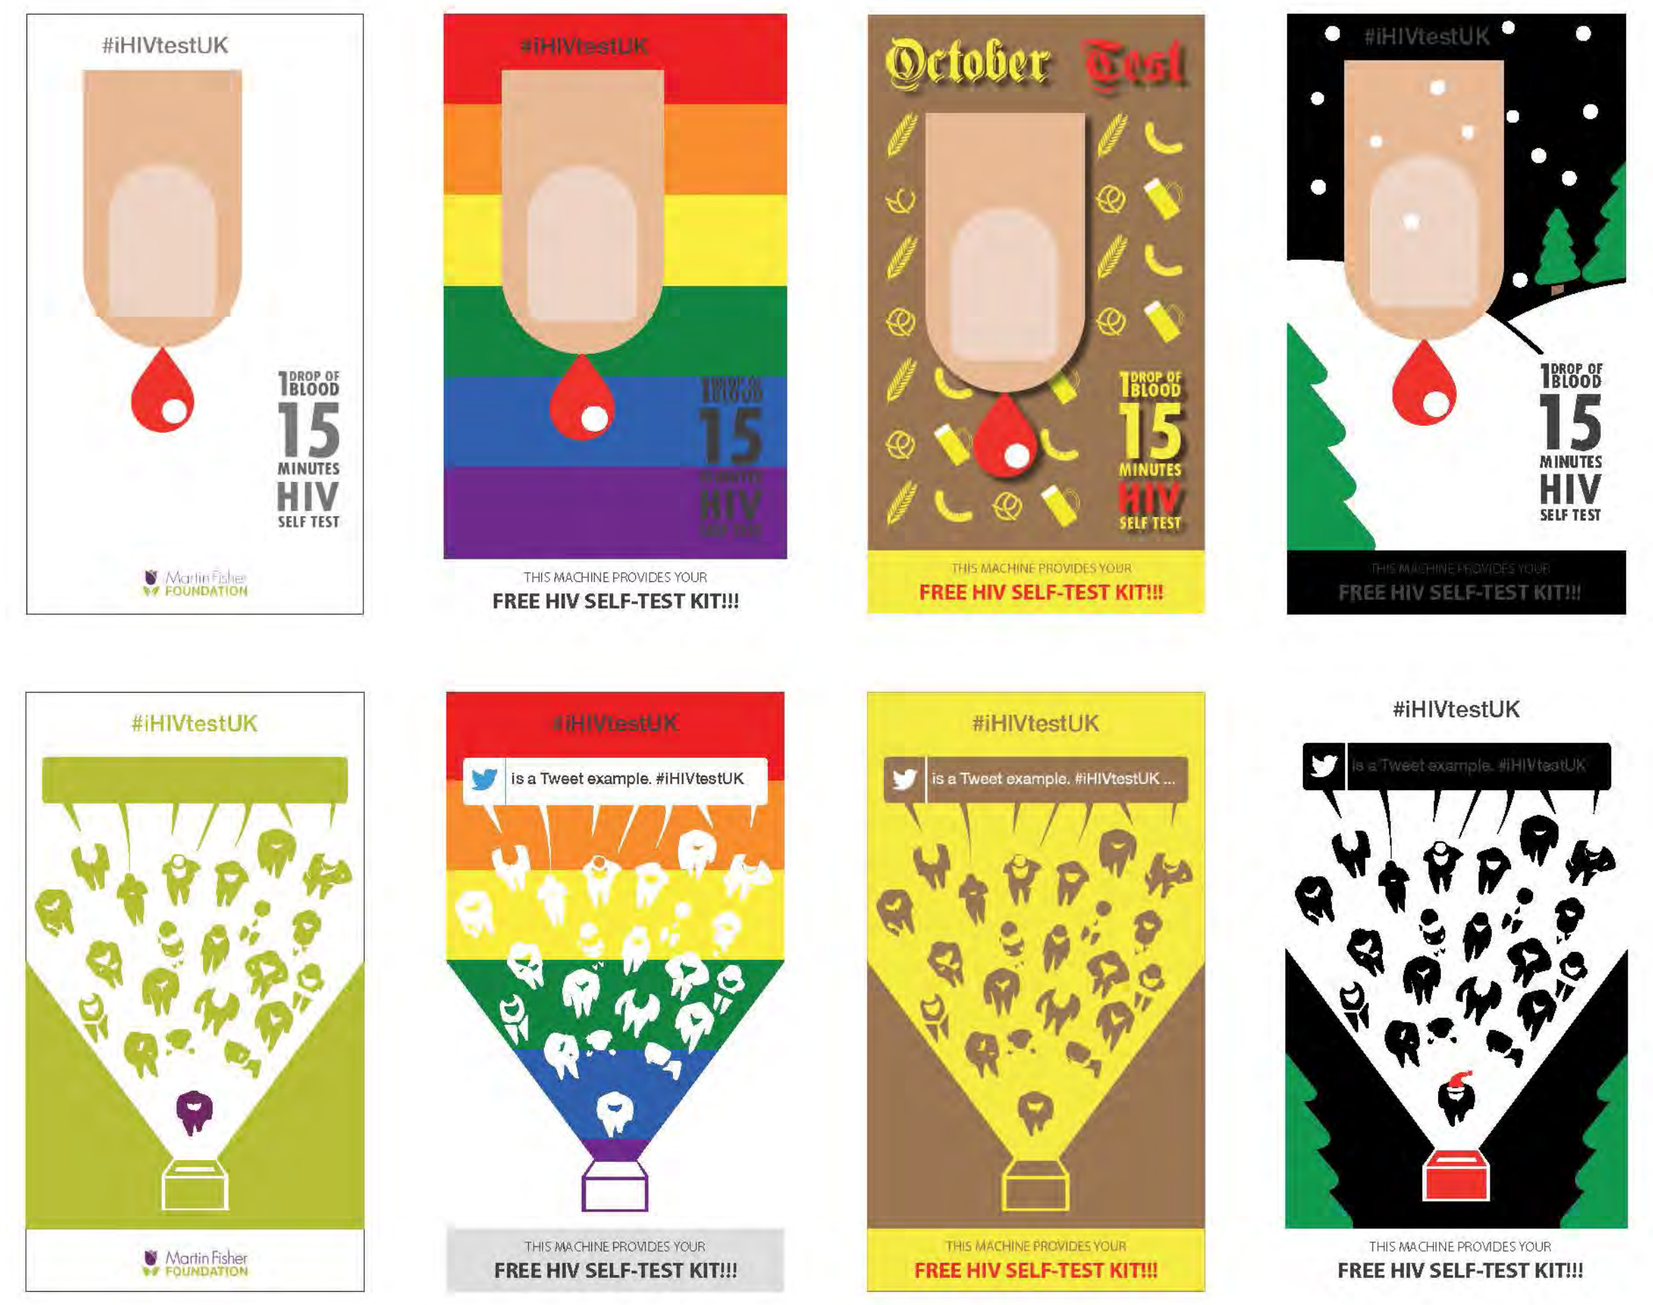
#iHIVtestUK
October Test
FREE HIV SELF-TEST KIT!!!

Seasonal vending machine interface posters to promote its use

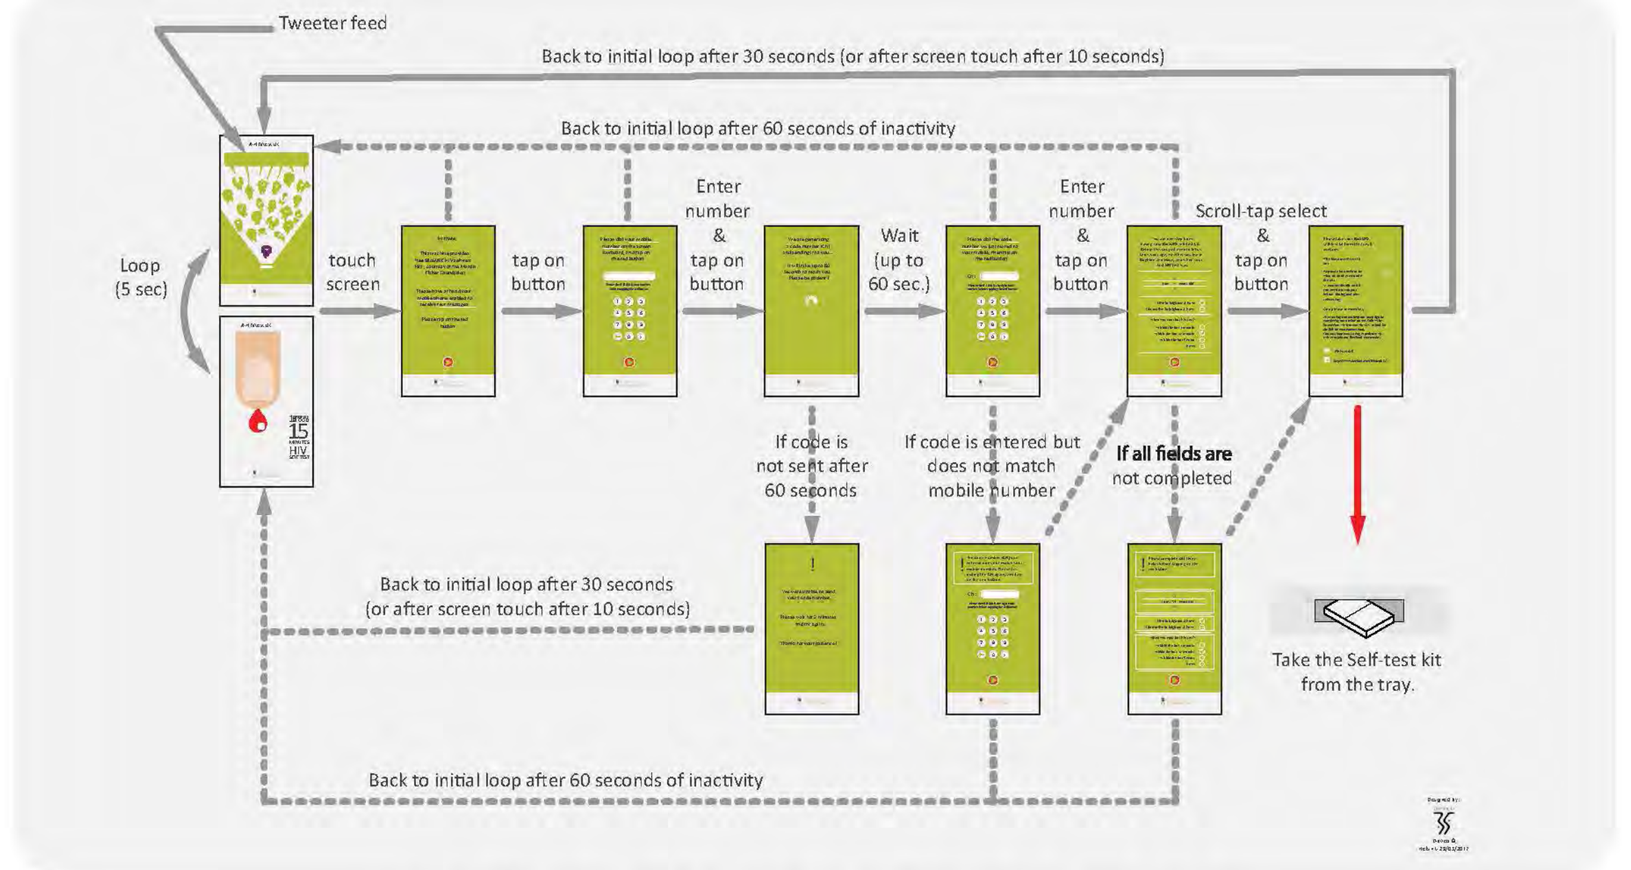
Tweeter feed
Back to initial loop after 30 seconds (or after screen touch after 10 seconds)
Back to initial loop after 60 seconds of inactivity
Loop (5 sec)
touch screen
tap on button
Enter number & tap on button
Wait (up to 60 sec.)
Scroll-tap select & tap on button
If code is not sent after 60 seconds
If code is entered but does not match mobile number
If all fields are not completed
Take the Self-test kit from the tray.

Vending machine interface sequence

CONCLUSIONS

Vending machine distribution of HIV self-testing kits is an accessible option to increase appropriately targeted testing among MSM. Frequent testing of men at high risk of HIV is thought to be crucial for elimination of HIV transmission (reference PHE report).

Collaboration with designers and target communities ensured a user-centered approach to this effective innovation in the distribution of HIV self-testing kits. The intervention was feasible and highly acceptable delivering low-cost HIV self-tests to men with low levels of prior testing.

Additional locations for vending machines such as gay bars/clubs, community pharmacies and GP practices, universities/colleges, sexual health clinics (as a 'short-cut' to HIV testing where comprehensive STI screening involved long appointments or delays) have been planned.

Further research is needed to evaluate the acceptability and impact of this intervention in other populations affected by HIV such as heterosexual men and women from areas of high-HIV prevalence.

NHS Brighton and Sussex University Hospitals NHS Trust
Martin Fisher FOUNDATION
University of Brighton
brighton and sussex medical school
Public Health England
DIPTICO

This study received ethical approval from the Brighton and Sussex Medical School Research Governance and Ethics Committee (BSMS RGEC) (UK). Contact: Dr Jaime Vera or Dr Gillian Dean 01273 664718.

REFERENCES: i) Public Health England. HIV diagnoses, late diagnoses and numbers accessing treatment and care. HIV official statistics overview: 2016. PHE publications gateway number: 2016349. ii) Skingsley A, Yin Z, Kirwan P, Croxford S, Chau C, Conti S, Presanis A, Nardone A, Were J, Ogaz D, Furegato M, Hibbert M, Aghaizu A, Murphy G, Tosswill J, Hughes G, Anderson J, Gill ON, Delpech VC and contributors. HIV in the UK – Situation Report 2015: data to end 2014. November 2015. Public Health England, London. iii) Young SD, Daniels J, Chiu CJ, Bolan RK, Flynn RP, Kwok J, et al. (2014) Acceptability of Using Electronic Vending Machines to Deliver Oral Rapid HIV Self-Testing Kits: A Qualitative Study. PLoS ONE 9(7): e103790. doi:10.1371/journal.pone.0103790 iv) Acceptability of HIV self-testing among men who have sex with men attending a sex on premises venue in Brighton: a cross sectional survey. Contact Dr Jaime Vera, BSMS for full details j.vera@bsms.ac.uk v) Donetto, S., Tsianakas, V. & Robert, G. (2014). Using Experience-based Co-design to improve the quality of healthcare: mapping where we are now and establishing future directions. London: King's College London.