A Phase 1 Study of ALT-803 (IL-15 Superagonist) to Clear Latent HIV Reservoirs

Poster #
356

Zachary Davis[1], Jodi Anderson[1], Ann Thorkelson[1], Hing C. Wong[2], Jonathan Karn[3], Curtis Dobrowlski[3], Jeffrey S. Miller[1], Sarah Cooley[1], Daniel C Douek[4], Timothy W Schacker[1]

[1]University of Minnesota, Minneapolis, MN, [2]Altor BioScience, a Nantworks company, Miramar, FL, [3]Case Western Reserve University, Cleveland, OH, [4]Vaccine Research Center, National Institutes of Health, Bethesda, MD

University of Minnesota
Driven to Discover

Background

Despite effective antiretroviral therapy (ART) yielding undetectable viral loads in infected patients, one important barrier to a cure for HIV is a significant latent reservoir in tissues. Efforts to clear these cells are hampered not only by defects in both adaptive and cellular immune responses, but in failure of the immune system to identify latently infected cells due to a lack of presentation of viral antigens or cell stress ligands. Therapies that would not only enhance immune cell function but reactivate latently infected cells could provide the means for a functional cure. Our focus is on IL-15, a cytokine that both stimulates NK and T cell function but also has potential to reverse NK and T-cell immunological defects to clear virus infected cells. The ability of IL-15 to enhance NK and CD8+ T-cell numbers has already been demonstrated and corresponds with a decrease in plasma viral load in treated patients (Ellis-Connell et al. J Virol 2017). This study will evaluate the effects of the IL-15 superagonist, ALT-80,3 on immune activation as well as reactivation of the latent viral reservoir.

Methods

We are conducting a Phase 1 dose escalation trial of ALT-803 in HIV+ adults. ALT-803 is an IL-15 superagonist/IL-15 receptor a complex (IL-15N72D/IL-15Ra-Fc) and is in clinical trials for hematologic and solid organ malignancies. Clinical trials in cancer patients have shown that subcutaneous (SQ) administration (compared to intravenous (IV)) is well tolerated and gives favorable pharmacokinetics at doses between 6-20 mcg/kg weekly. HIV+ people on ART with plasma viral load < 20 copies/ml and CD4 T cells > 500 cells/µl receive an IV or SQ injection of ALT-803 weekly for 3 weeks. Blood is obtained for assessments of cell activation and proliferation, and changes to the virus reservoir. Three people are planned to receive the drug at each dose level. The dose escalation scheme is 0.3 mcg/kg IV then 1, 3, and 6 mcg/kg SubQ.

IL-15 vs ALT-803

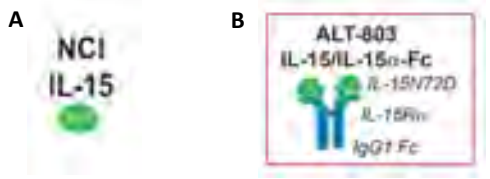
A
NCI
IL-15
B
ALT-803
IL-15/IL-15α-Fc
IL-15N72D
IgG1 Fc

Figure 1: Comparison of IL-15 drugs in clinical testing. (**A**) Monomeric IL-15 binds to IL-15Rα expressed on lymphocytes to initiate signaling. (**B**) ALT-803 consists of dimeric IL-15Rα bound to human IgG Fc via sushi domains. Each IL-15Rα is bound to an IL-15 molecule with an amino acid substitution at position 72 from an asparagine to aspartate which has been shown to increase affinity for the receptor and enhance signaling.

ALT-803 activates NK and T-cells *in vivo*

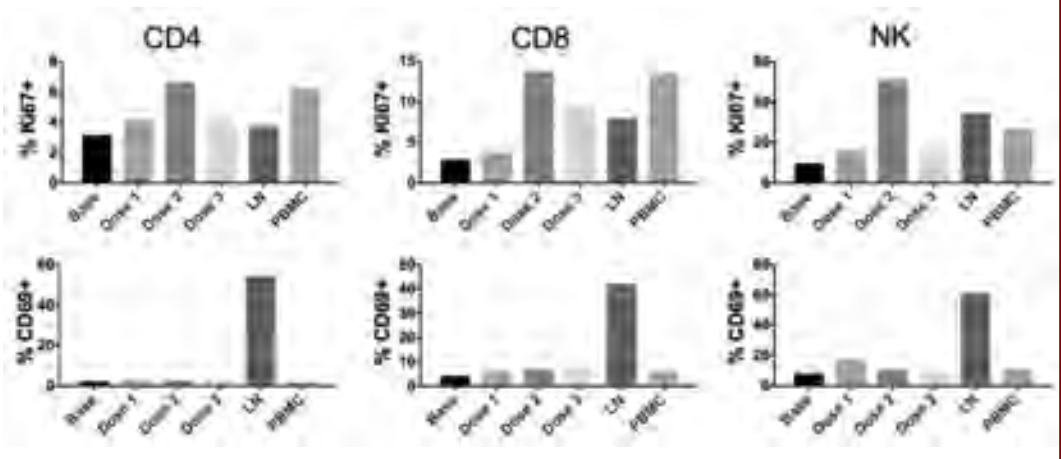
CD4
CD8
NK
% Ki67+
% CD69+
Base
Dose 1
Dose 2
Dose 3
LN
PBMC

Figure 2: Proliferation (Ki67) and activation (CD69) of CD4, CD8, and NK cells after each dose. Base = baseline/pre-dose 1. Dose 1 is 24 hours after the 1st dose. Dose 2 is 24 hours after the 2nd dose. Dose 3 is 24 hrs after the 3rd dose. LN and PBMC were obtained 48 hrs after the 3rd dose. Flow data was obtained from frozen samples at each of the time points indicated.

ALT-803 induces HIV transcription

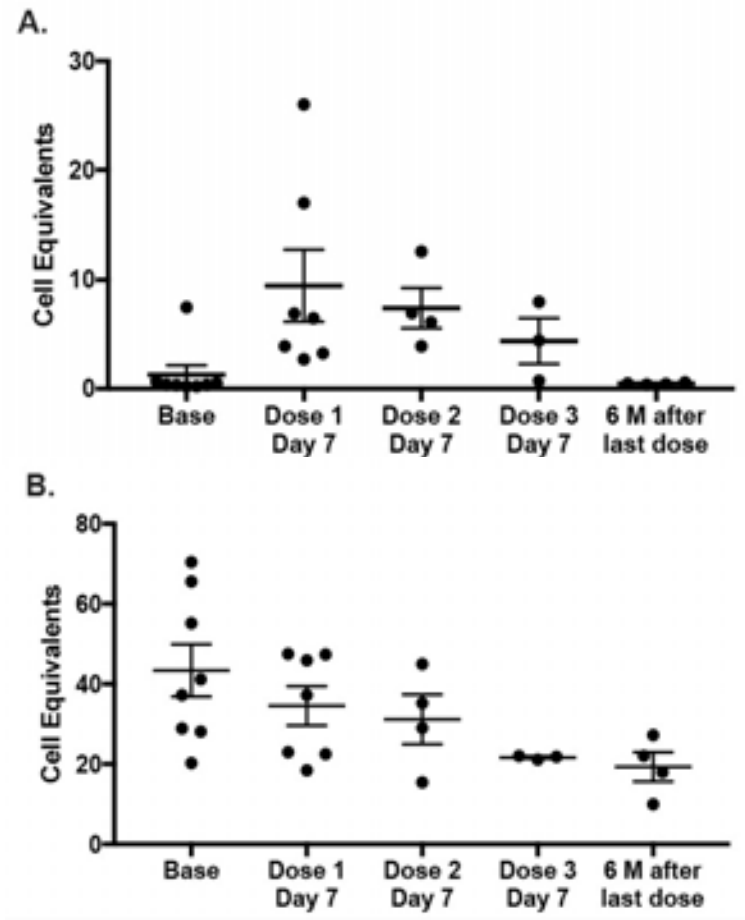
A.
B.
Cell Equivalents
30
20
10
0
80
60
40
20
0
Base
Dose 1
Day 7
Dose 2
Day 7
Dose 3
Day 7
6 M after
last dose

Figure 3: Mean frequency and SEM of HIV transcription events in 8 participants before and after receiving (**A**) ALT-803 or (**B**) ConcanamycinA stimulation. Dr. John Karn developed a novel Next Generation Sequencing (NGS)-based protocol, called EDITS (Envelope Detection by Induced Transcription-based Sequencing), to measure inducible cell-associated HIV RNA. Sequences of envelope are detected using a nested PCR on 1.25 x10^{6} resting memory cells from aviremic HIV-1-infected subjects before and after ConA stimulation.

ALT-803 reactivation of HIV by patient treated

		Dose 1			Dose 2			Dose 3		
	Base	8	16	24	8	16	24	8	16	24
2349	-	-	-	+	-	-	-	-	-	-
2354	+	-	-	-	-	-	+	-	-	+
2383	-	28	27	+						
2356	-	+	+	+	-	-	-	+	+	+
2413	-	-	-	-						
2470	+	-	-	+	-	-	+			
2403	-	-	-	-	-	-	-	-	-	-
2530	-	-	-	-	-	-	-	-	+	-
2543	-	-	35	+	+	+	+	-	23	+

Figure 4: Detection of HIV RNA in patient plasma at 8, 16, and 24 hours after ALT-803 doses (- not detected, + detected but <20 copies/ml. A numerical value is given if copies/ml exceed 20 copies/ml).

ALT-803 treatment results in accumulation of NK cells in LN

A
B

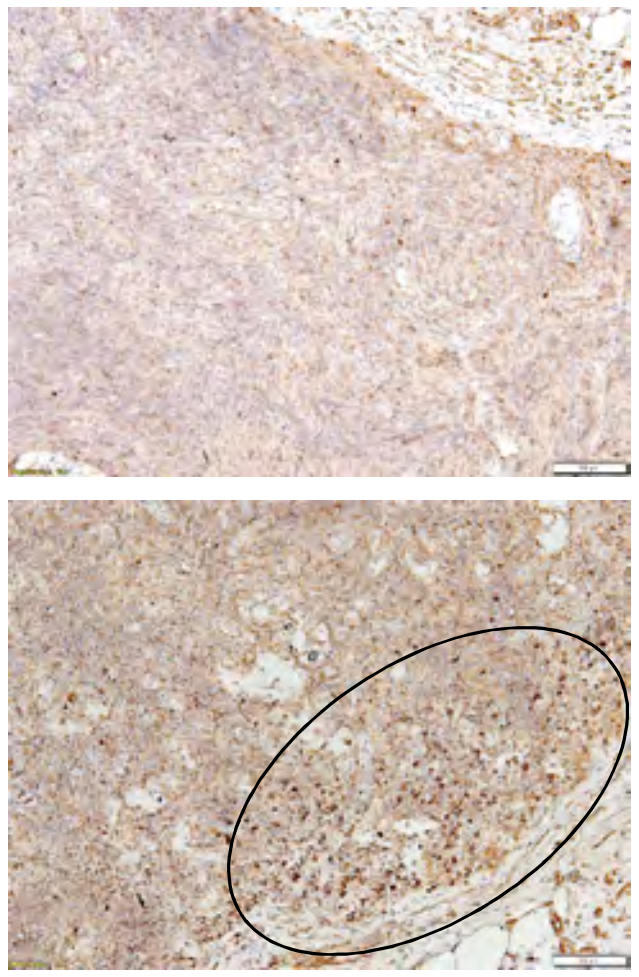

Figure 5: NK cells infiltrate secondary lymphoid tissues in response to ALT-803 treatment. CD56 Staining of LN before (**A**) and 1 week after (**B**) the 3rd dose of ALT-803 in participant 2543 (3.0 mcg/kg SQ). The arrow in **A** points to a CD56+ NK cell illustrating the paucity of these cells prior to ALT-803. The black oval in B shows an area of LN with a large number of CD56+ NK cells after ALT-803 therapy.

Results

A total of 7 individuals have been dosed to date as shown below. The mean age was 40 and mean CD4 T cell count was 755 cell/µl. The mean time from diagnosis was 8 years and the mean time on ART was 5 years. The first 2 participants received 0.3 mcg/kg IV and the remaining 5 received 1.0 mcg/kg SubQ.

PID	Age (yrs)	CD4	Yrs HIV+	Yrs on ART	ALT 803 Dose (mcg/kg)	# doses
2356	47	1323	12	10	1.0 (SQ)	3
2413	28	724	13	2	1.0 (SQ)	1
2470	51	611	12	1	1.0 (SQ)	2
2403	50	670	5	5	1.0 (SQ)	3
2530	35	828	9	5	1.0 (SQ)	3
2543	28	658	2	2	3.0 (SQ)	3

SubQ dosing was associated with an injection site rash and adenopathy. Eight of the first 13 doses were associated with transient low-level plasma viremia. We measured a 7.6-fold increase in NK cell activation, a 23-fold increase in CD4 activation and a 10-fold increase in CD8 T cell activation in LN 48 hours after dosing. We used the EDITS assay (Envelope Detection by Induced Transcription-based Sequencing, developed by Dr. Karn, CWRU) to measure inducible cell-associated HIV RNA. Prior to Con A stimulation we measured a significant increase in HIV transcription after the first 2 ALT-803 doses but after dose 3 there were fewer transcription events. With ConA stimulation there was a trend towards decreasing transcription events.

Conclusions

At these doses of ALT-803, the drug is safe and well-tolerated. The drug is biologically active and results in activation and proliferation of CD4 and CD8 T cells as well as NK cells. ALT-803 also induces transcription of HIV. Furthermore, treatment of ALT-803 results in NK cell infiltration of secondary lymphoid tissues where latently infected cells reside. These data suggest a potential role for ALT-803 in future cure studies.