A LONG-ACTING NANOFORMULATED CABOTEGRAVIR PRODRUG FOR IMPROVED ANTIRETROVIRAL THERAPY

Tian Zhou[1,2], Hang Su[2], Benson Edagwa[2], JoEllyn McMillan[2], Prasanta Dash[2] and Howard E. Gendelman[1,2]
Departments of Pharmaceutical Sciences[1] and Department of Pharmacology and Experimental Neuroscience[2]
University of Nebraska Medical Center, Omaha, NE68198

UNIVERSITY OF NEBRASKA MEDICAL CENTER℠

Poster # 439

Background: Long-acting parenteral (LAP) antiretroviral drugs (ARVs) have set a new bar for HIV/AIDS therapeutics. LAP ARVs can improve treatment adherence and positively affect drug resistance patterns and systemic toxicities. A single intramuscular (IM) injection of cabotegravir (CAB), a potent HIV integrase inhibitor, in phase III clinical trials (CAB-LAP), permits up to two month sustained plasma drug levels > 4x protein adjusted IC_{90} ($PA\text{-}IC_{90,}$ 660 ng/mL). We posit that CAB-LAP could be modified to reduce its injection volume and improve its pharmacokinetic (PK) and biodistribution profiles. To this end, a nanoformulated prodrug of CAB (we call NMCAB) was made to extend the drug's half-life and antiretroviral activities.

Methods: CAB was chemically modified with myristoyl chloride to affect its hydrophobic properties. MCAB was encased in poloxamer 407 by high-pressure homogenization. Uptake and retention in human monocyte-derived macrophages (MDM) were tested. Antiretroviral activity was evaluated by reverse transcriptase (RT) activity and HIV-1 p24 antigen levels. NMCAB test results were compared to parent drug formulations in Balb/c mice after a single IM injection of 15 or 45 mg/kg. The plasma drug levels were monitored for two months. Pharmacodynamics of NMCAB was evaluated in HIV-1 infected human PBL reconstituted NOD/scid-IL-$2R\gamma_c$ ^{null} (NSG) mice model.

Results: MDM efficiently took up the NMCAB nanoparticles with sustained slow release up to 30 days. Notably, the parent drug formulations were eliminated after the treatment day. Drug crystals were observed by transmission electron microscopy (TEM) in NMCAB treated MDM, but not in cells treated with parent drug formulations. NMCAB elicited sustained antiretroviral activity in MDM determined by RT activity and HIV-1 p24 staining for up to 15 days. In *in vivo* studies, NMCAB showed reduced burst release with reduced rate of elimination, resulting in plasma drug levels at 6-8 weeks being up to 300 times greater than the parent drug formulations with improved abilities to prevent HIV infection in a mouse model of disease.

Conclusion: NMCAB demonstrated improved antiretroviral activities and long-acting slow release properties as compared to CAB-LAP in the testing systems applied. Further refinements in drug delivery platforms will enable LAPs to be more easily administered.

MCAB synthesis and characterization

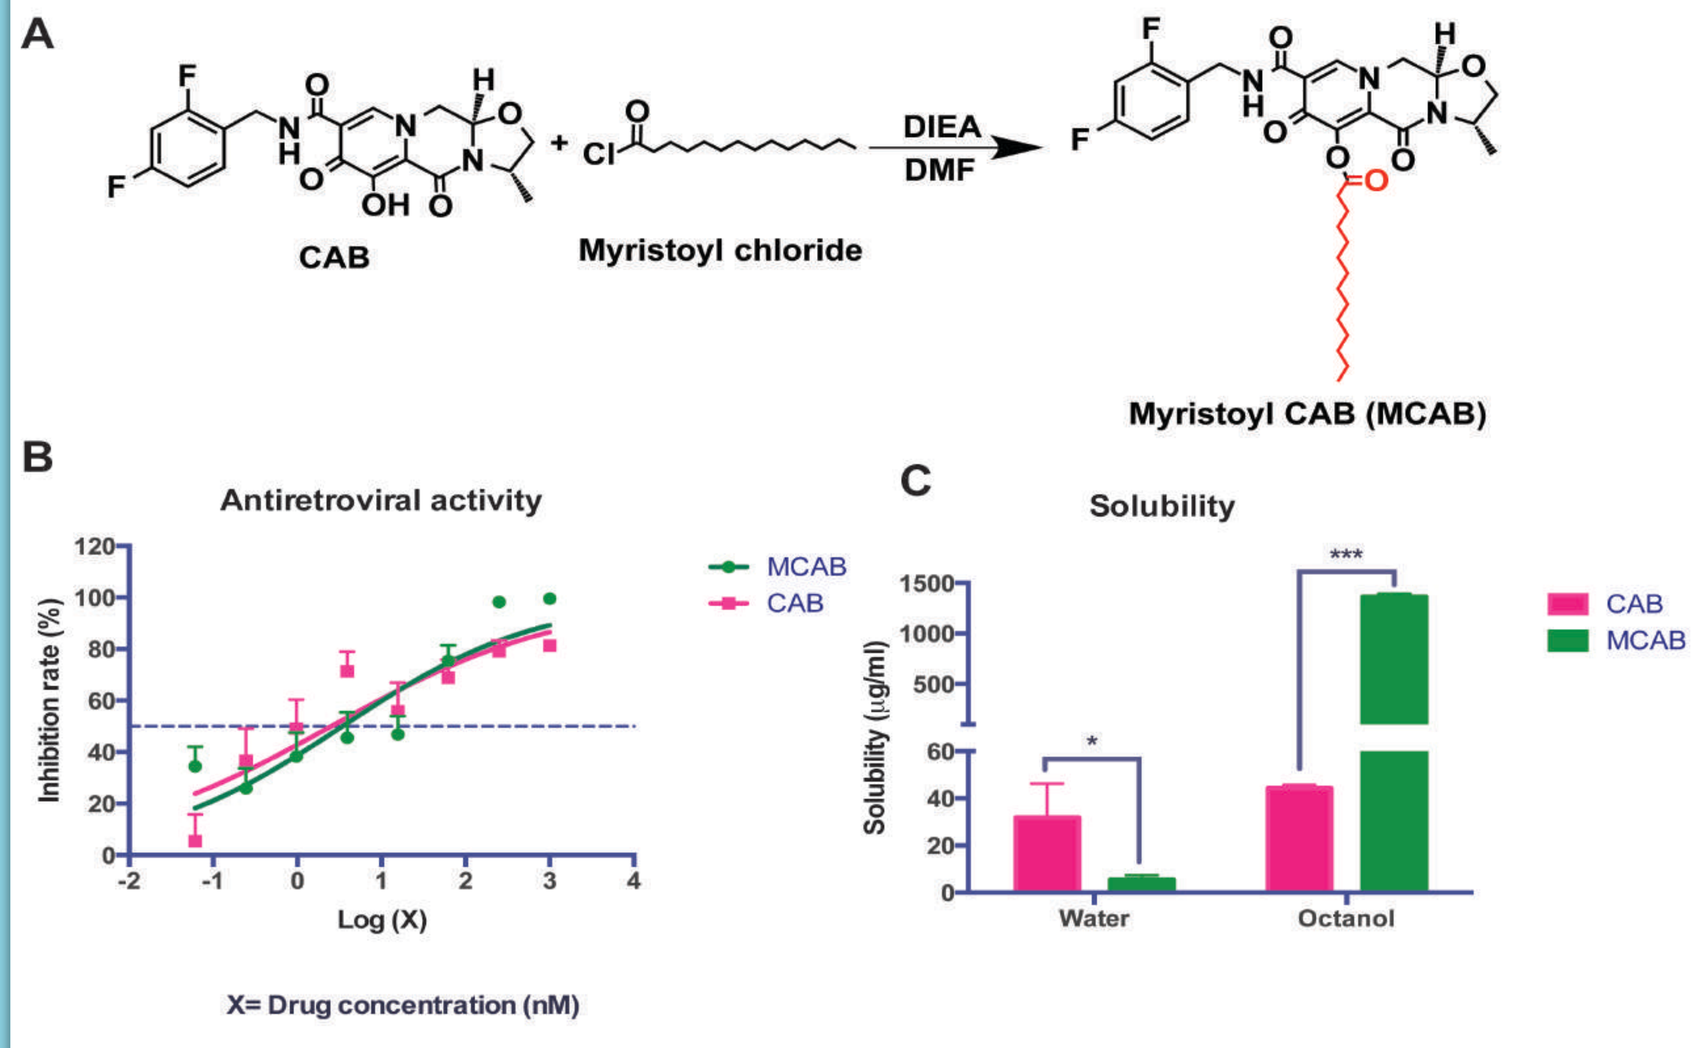
A
CAB
Myristoyl chloride
DIEA
DMF
Myristoyl CAB (MCAB)
B
Antiretroviral activity
MCAB
CAB
Inhibition rate (%)
Log (X)
X= Drug concentration (nM)
C
Solubility
CAB
MCAB
Solubility (µg/ml)
Water
Octanol

Figure 1. *MCAB synthesis and characterization. (A) Synthesis of myristoylated CAB (MCAB). (B) Antiretroviral activity of MCAB versus CAB in HIV-1_{ADA} infected MDM. (C) Solubility of CAB and NMCAB in water and 1-octanol measured at room temperature.*

Production of nanoformulated MCAB (NMCAB)

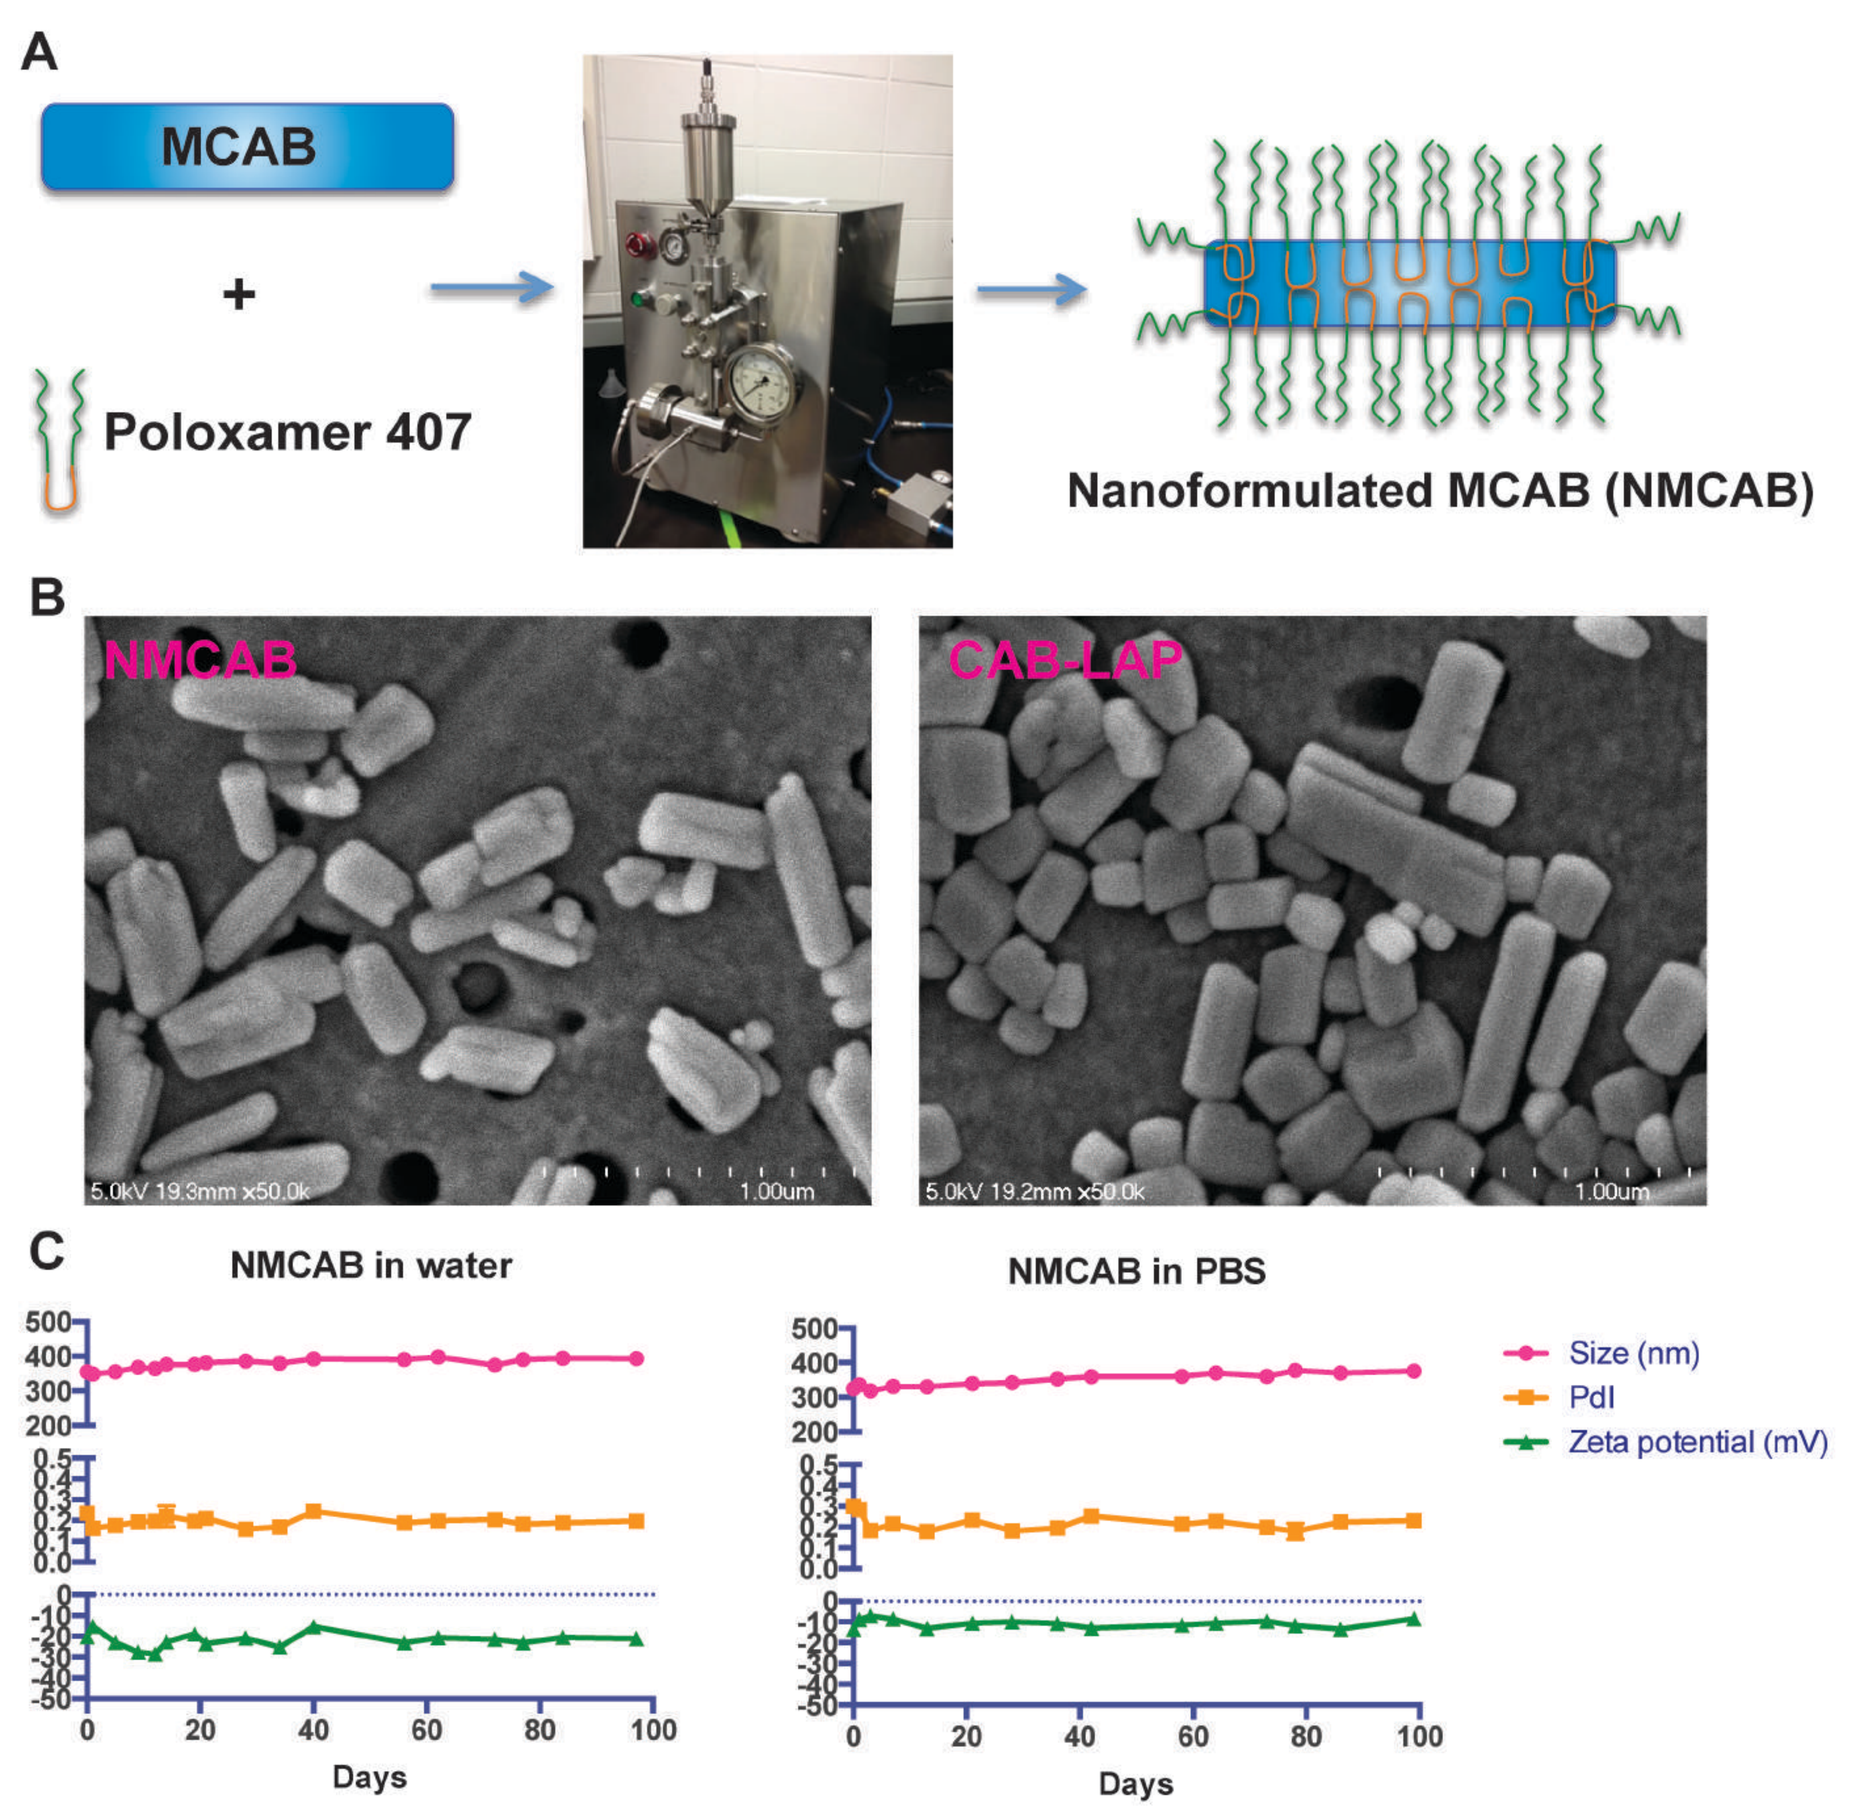
A
MCAB
+
Poloxamer 407
Nanoformulated MCAB (NMCAB)
B
NMCAB
CAB-LAP
C
NMCAB in water
NMCAB in PBS
Size (nm)
PdI
Zeta potential (mV)
Days

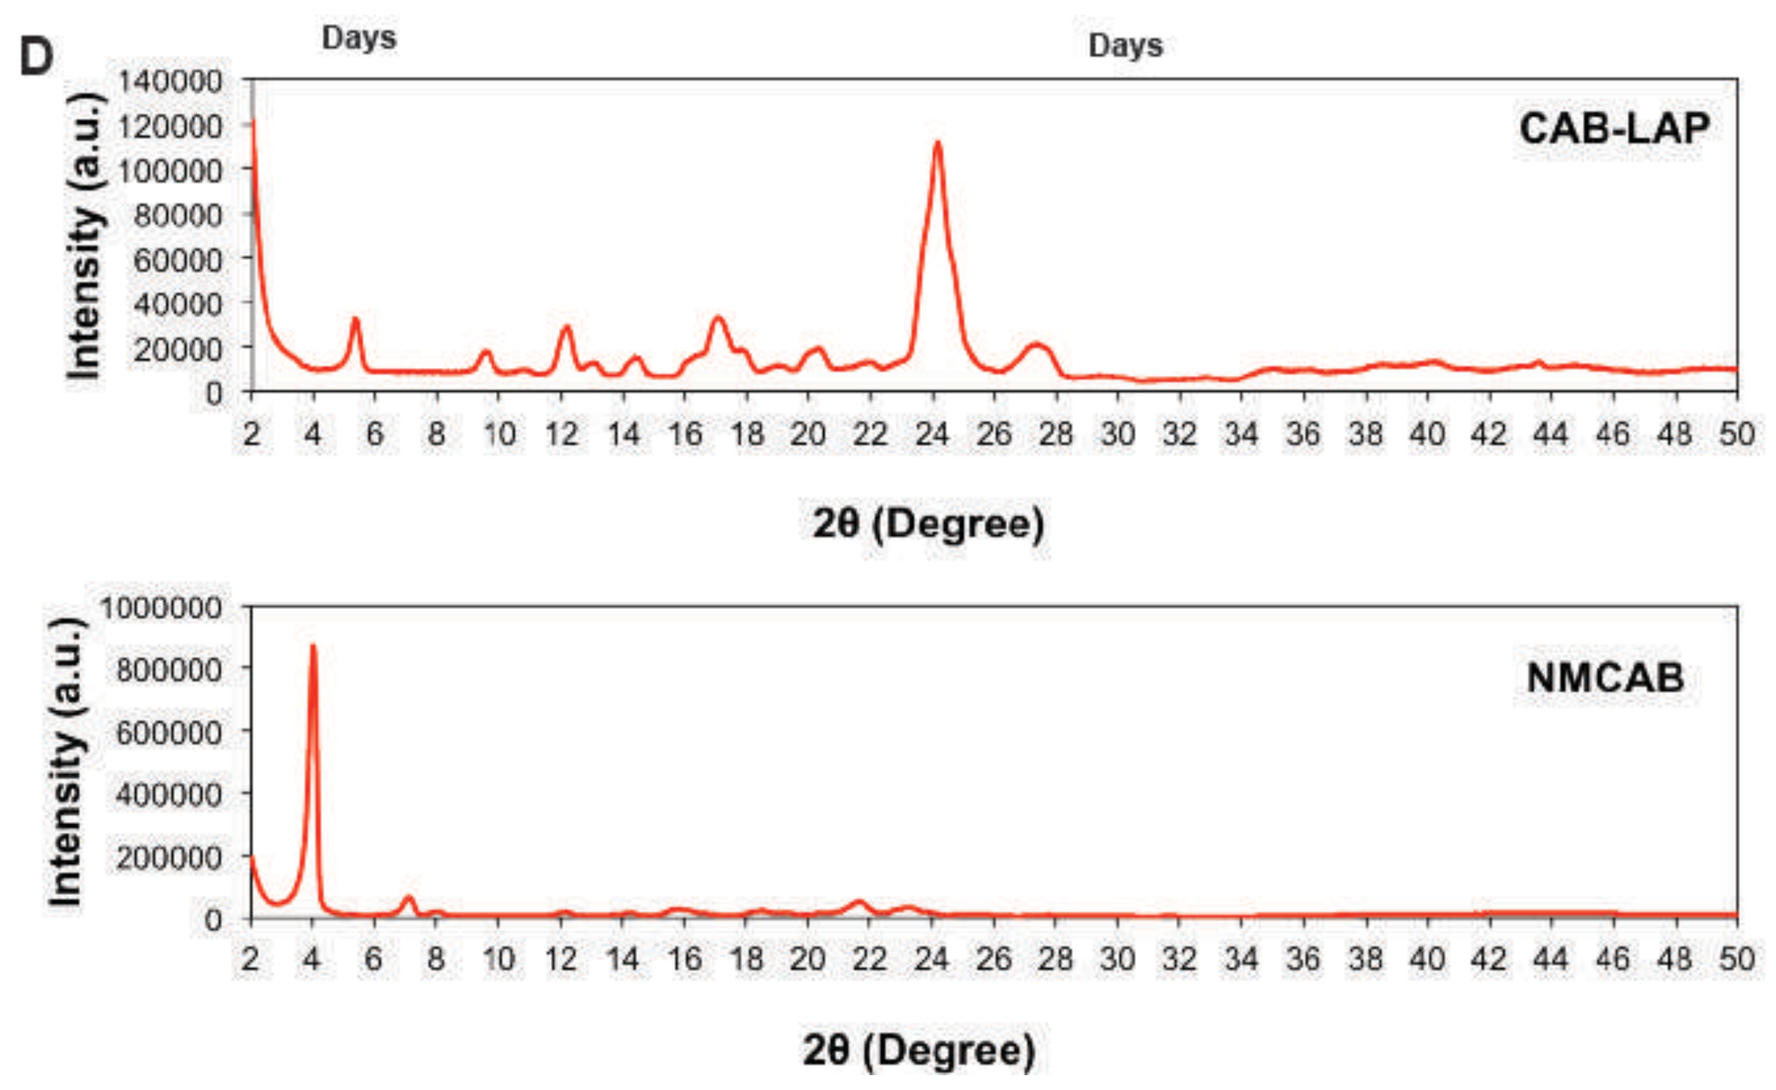
D
CAB-LAP
NMCAB
Intensity (a.u.)
2θ (Degree)

Figure 2. *NMCAB production and characterization. (A) P407-encased MCAB (NMCAB) was prepared by high pressure homogenization. (B) SEM of NMCAB and CAB-LAP shows rod-shaped particles. (C) Size, charge and PDI of NMCAB were evaluated in water and PBS over 3 months. (D) X-ray diffraction of CAB-LAP and NMCAB formulations illustrates the crystalline drug particles formed.*

NMCAB cellular reservoir formation

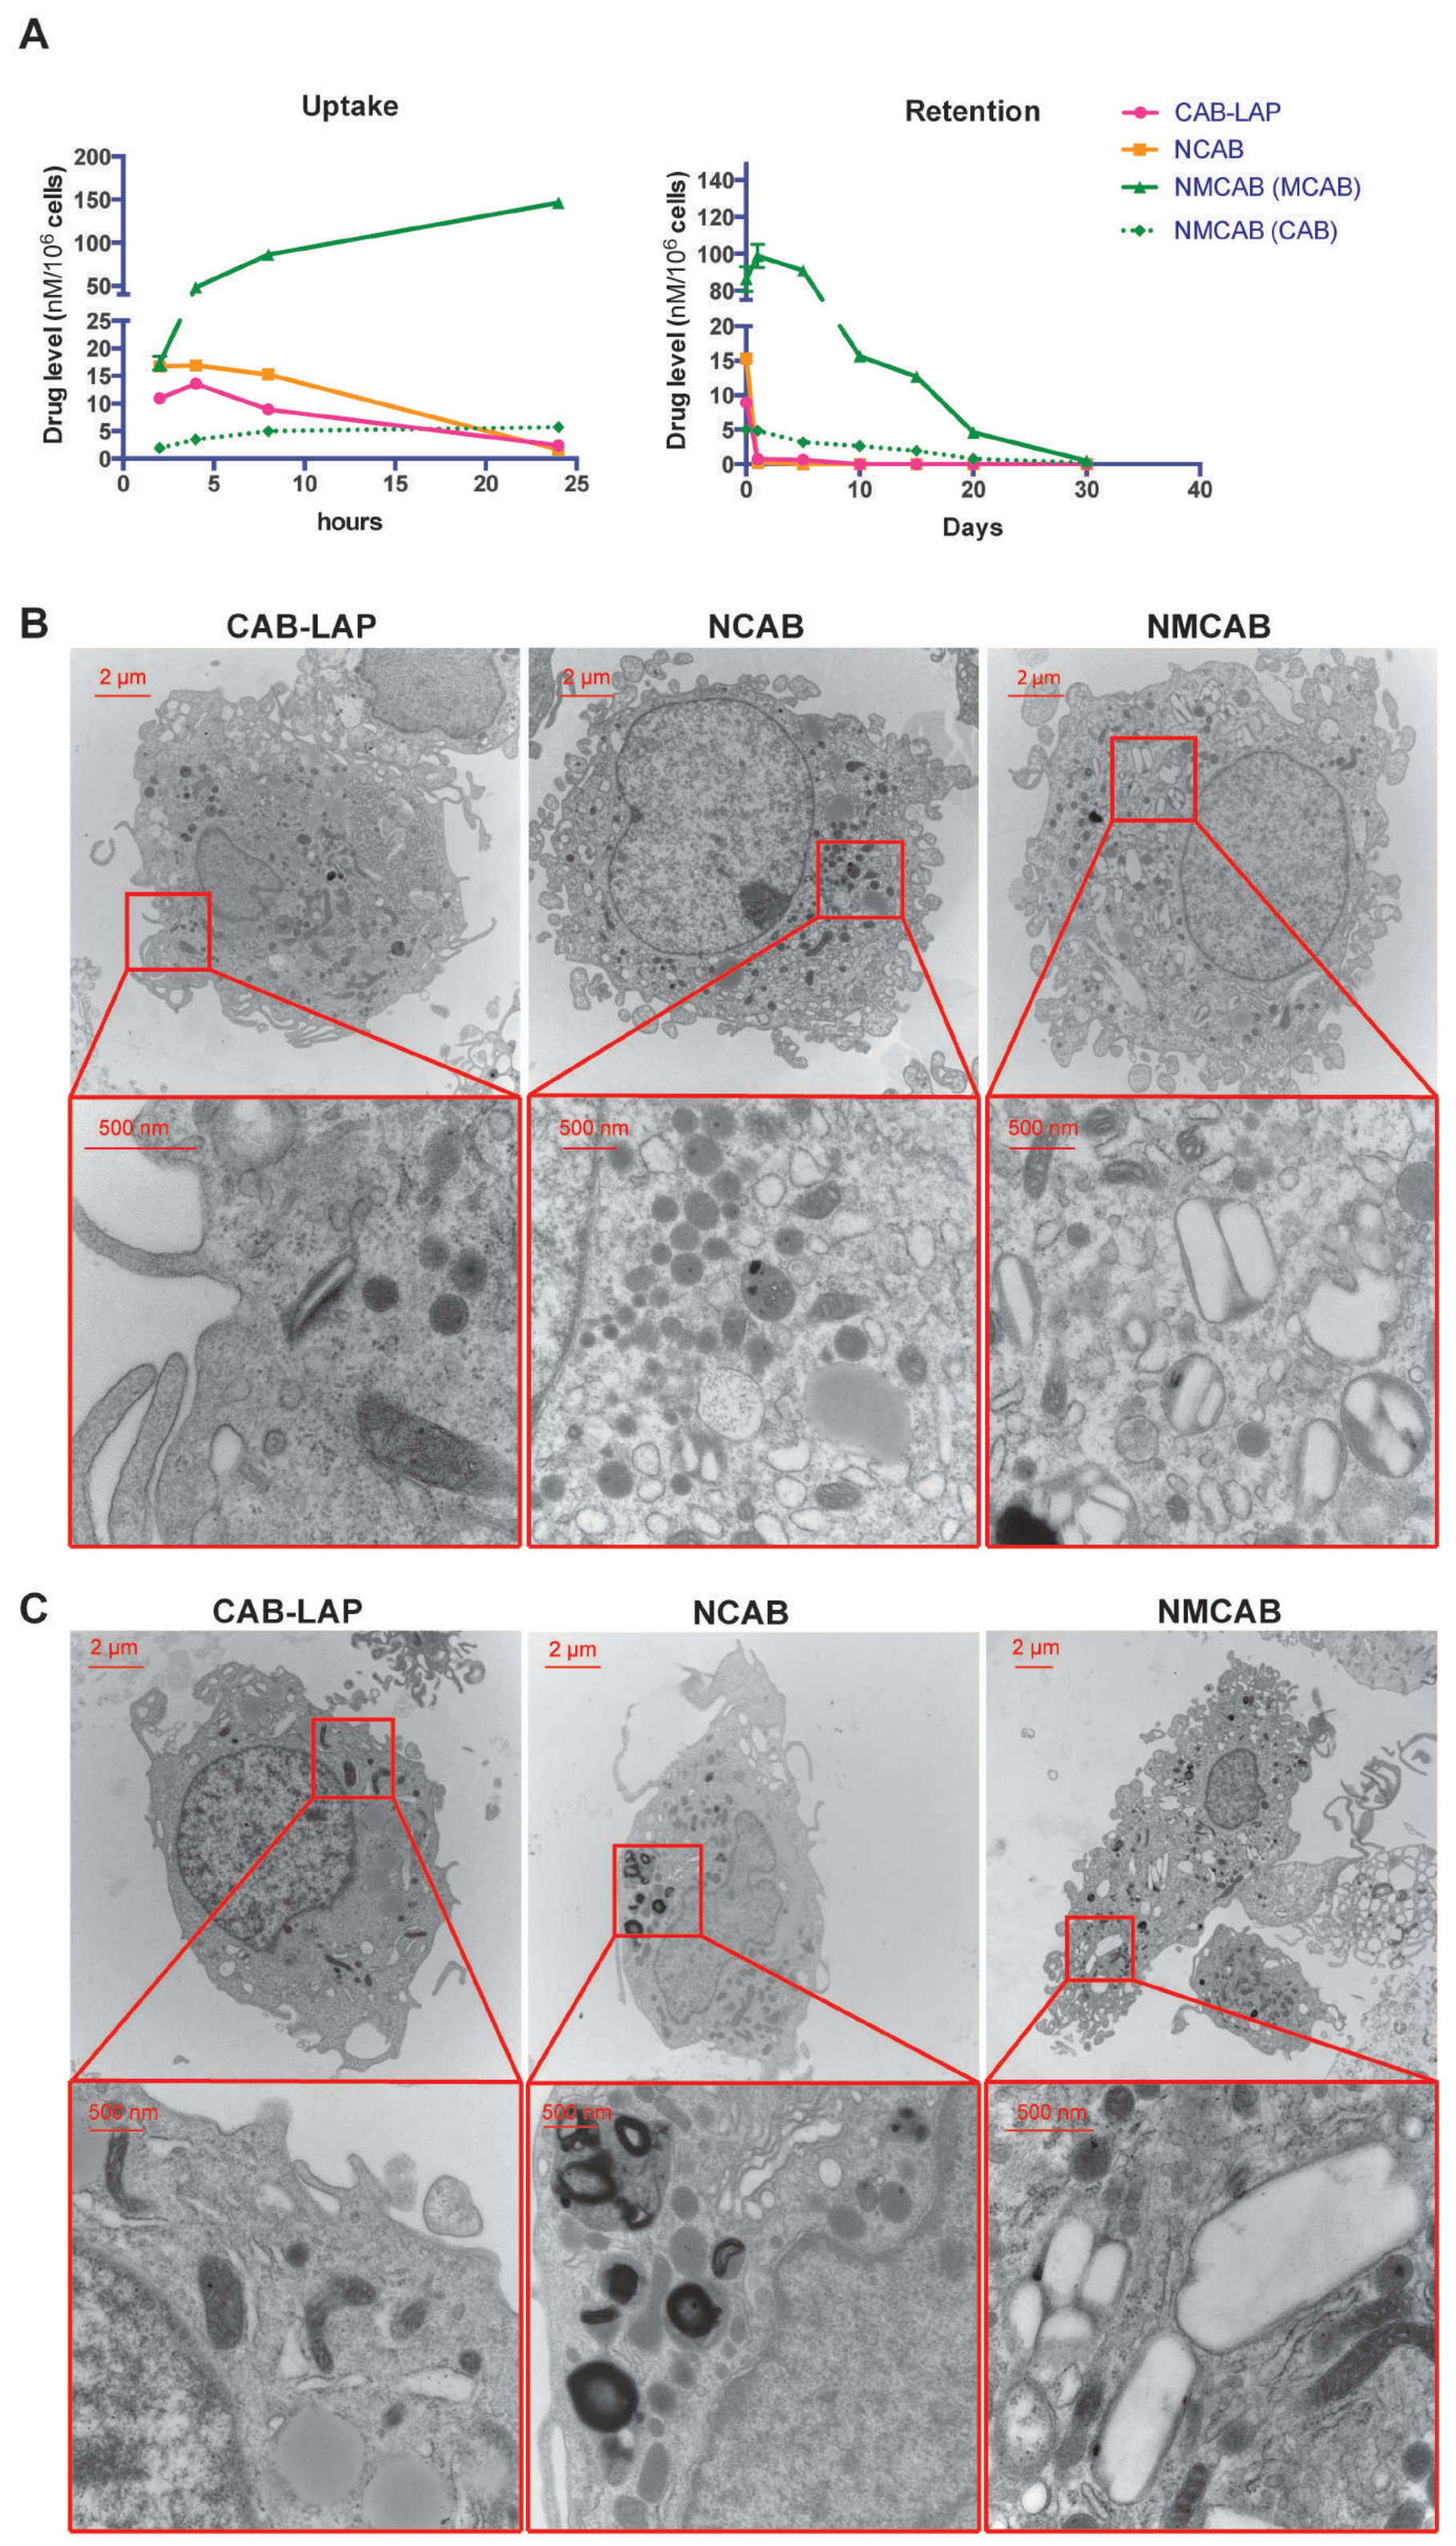
A
Uptake
Retention
CAB-LAP
NCAB
NMCAB (MCAB)
NMCAB (CAB)
Drug level (nM/10⁶ cells)
hours
Days
B
CAB-LAP
NCAB
NMCAB
C
CAB-LAP
NCAB
NMCAB

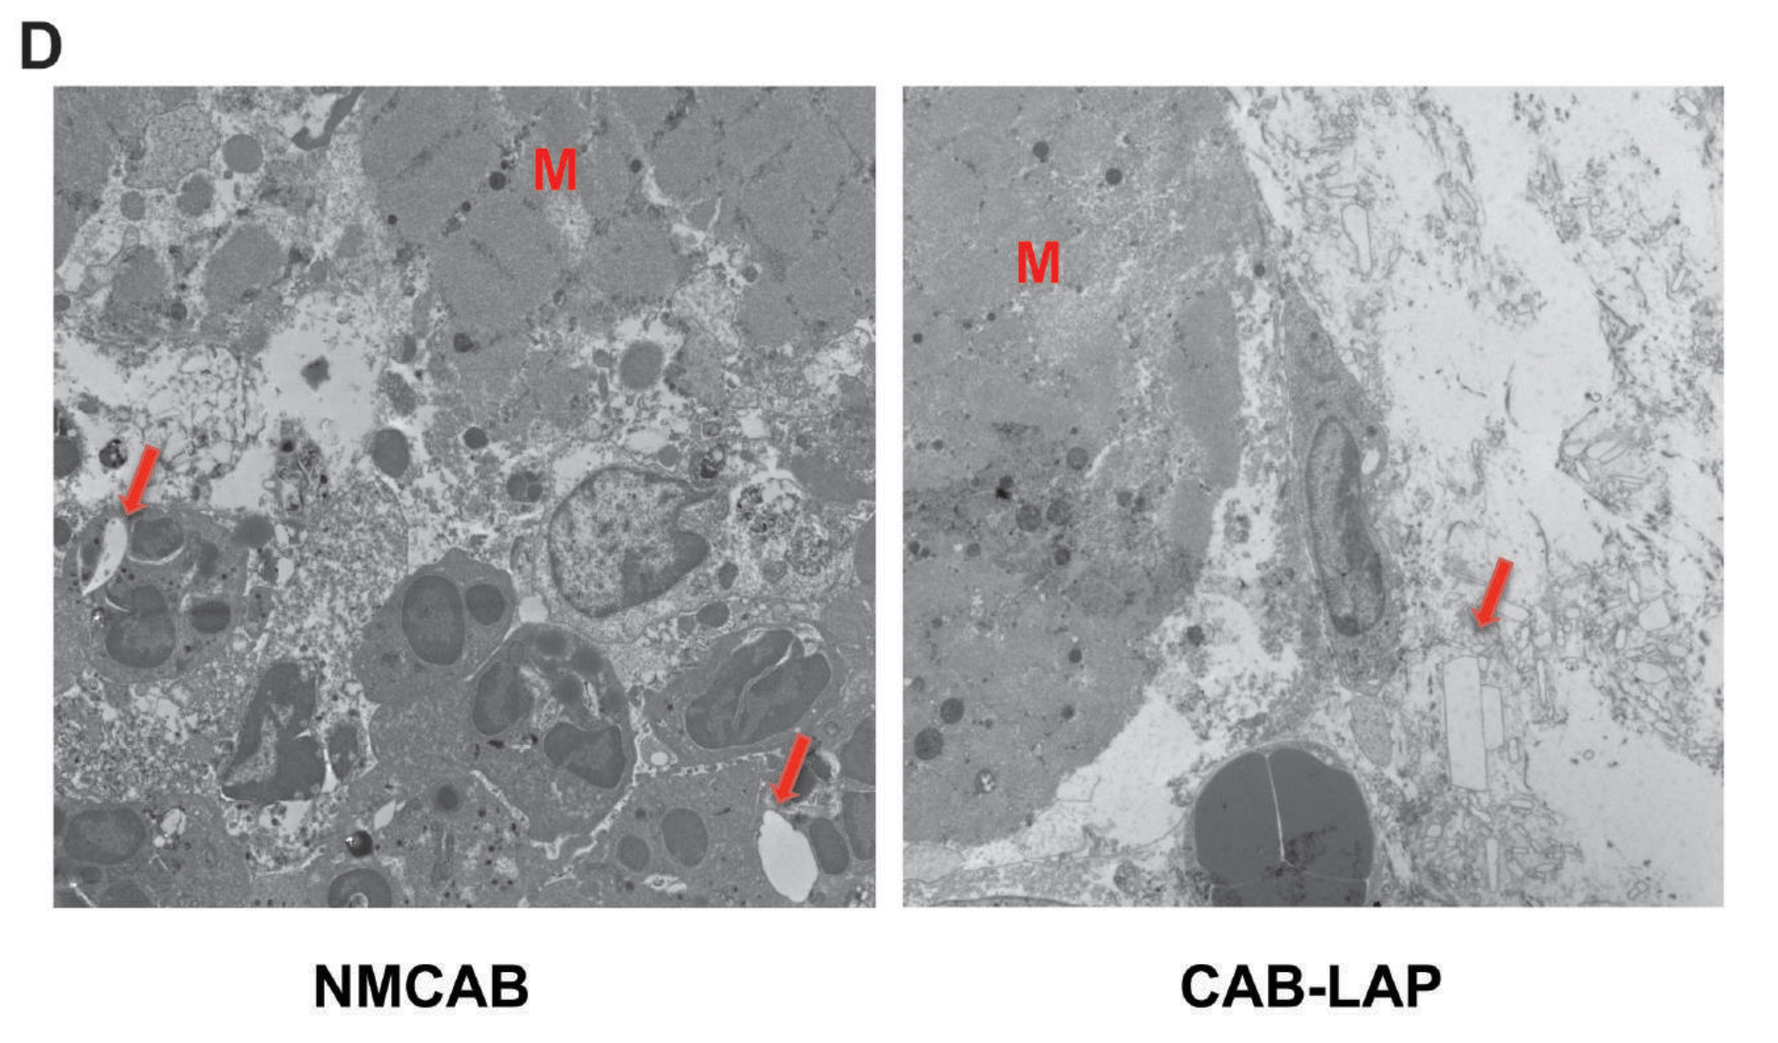
D
M
M
NMCAB
CAB-LAP

Figure 3. *NMCAB depot formation in MDM and at the site of injection (muscle). (A) NMCAB uptake (left) and retention (right) in human MDM. For uptake (left), drug concentration was determined in MDM treated with 100 µM NMCAB, NCAB or CAB-LAP for 2-24 hours. Cell drug retention (right) was determined after treatment of MDM for 8 hrs with 100 µM NMCAB, NCAB or CAB-LAP, followed by replacement with fresh medium without drug. Drug concentration in cells was determined from 1-30 days. Both MCAB (solid lines) and CAB (dashed lines) were measured in NMCAB treated cells. (B) TEM of MDM treated with CAB-LAP, NCAB, or NMCAB for 8 hrs or (C) treated with CAB-LAP, NCAB, or NMCAB for 8 hrs, then cultured in fresh medium for an additional 48 hrs. (D) TEM images of a cross-section of muscle following IM injection with NMCAB (left) or CAB-LAP (right). (M: muscle bundles; arrows: drug crystals)*

Antiretroviral Responses

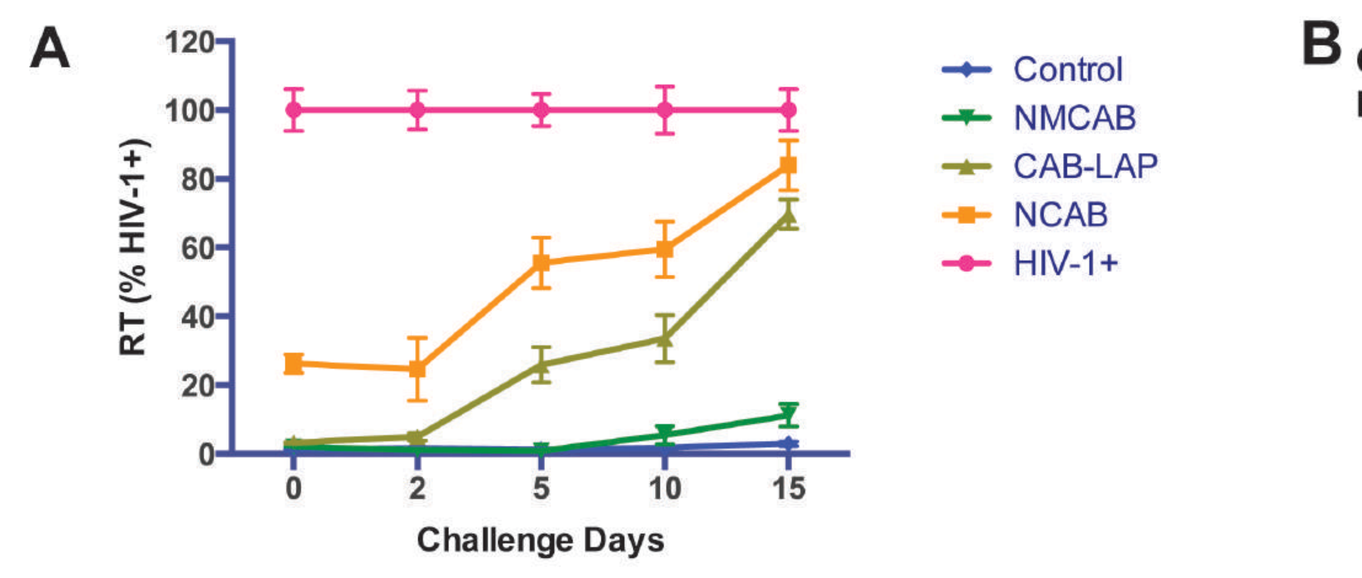
A
RT (% HIV-1+)
Challenge Days
Control
NMCAB
CAB-LAP
NCAB
HIV-1+

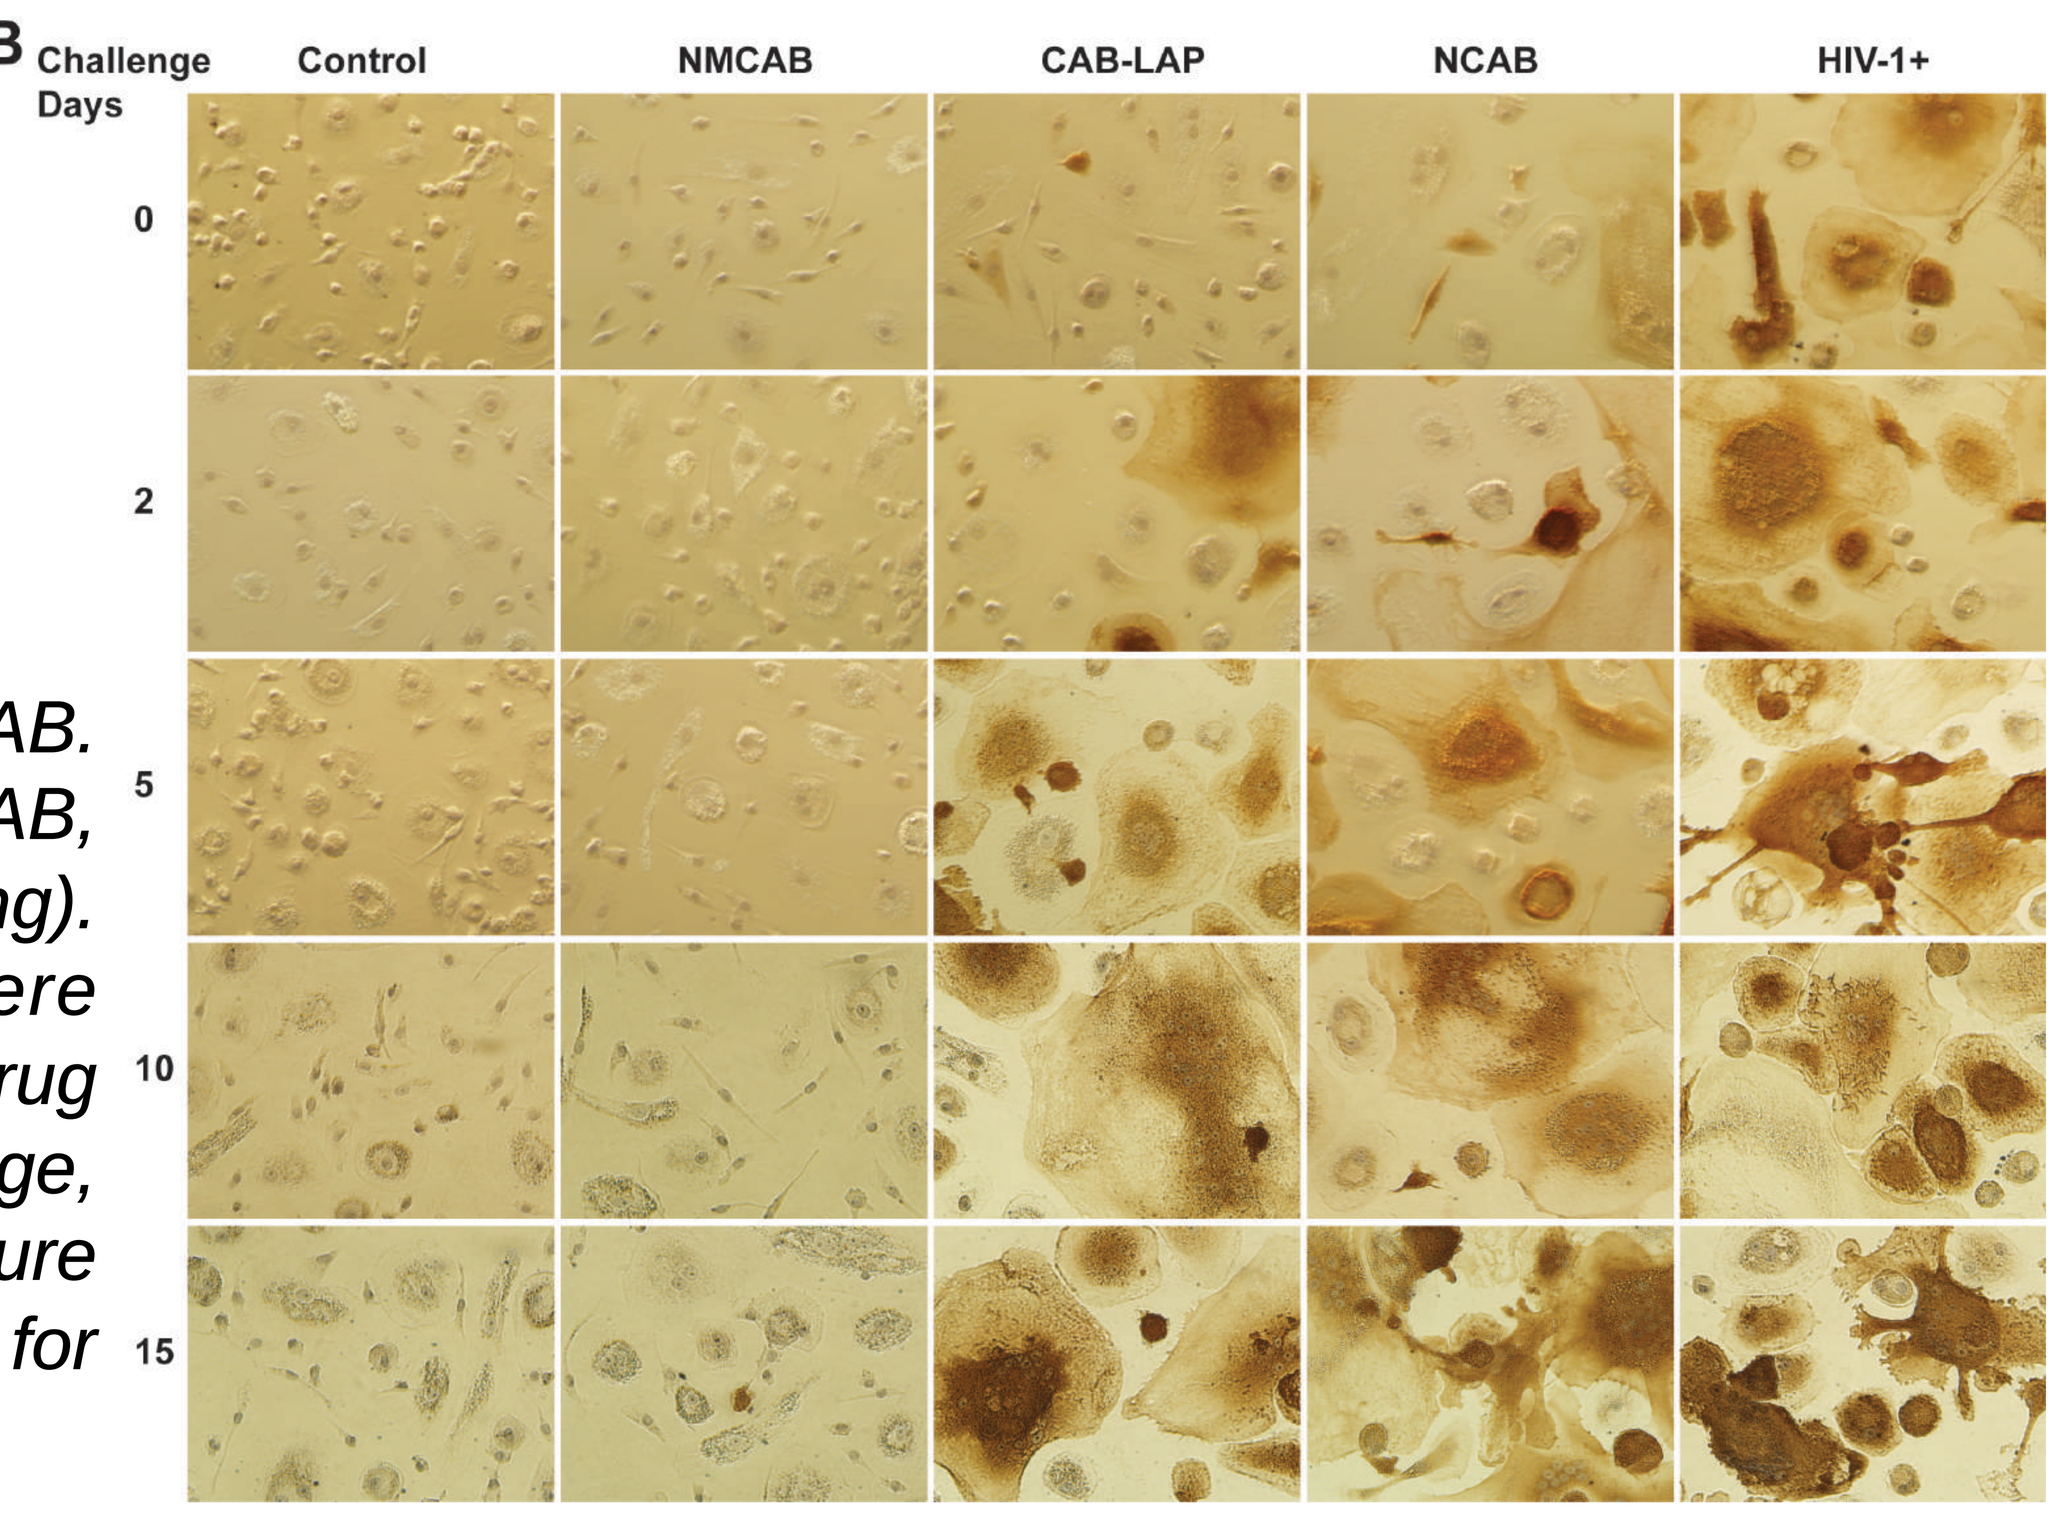
B
Challenge Days
Control
NMCAB
CAB-LAP
NCAB
HIV-1+
0
2
5
10
15

Figure 4. *Antiretroviral activity of NMCAB. MDM were treated with 100 µM NMCAB, CAB-LAP or NCAB for 8 hrs (drug loading). At the time points listed MDM were challenged with 0.1 MOI HIV-1_{ADA} after drug loading. Seven days after viral challenge, (A) RT activity was measured in culture medium and (B) cells were stained for HIV-1 p24 antigen.*

Pharmacokinetics

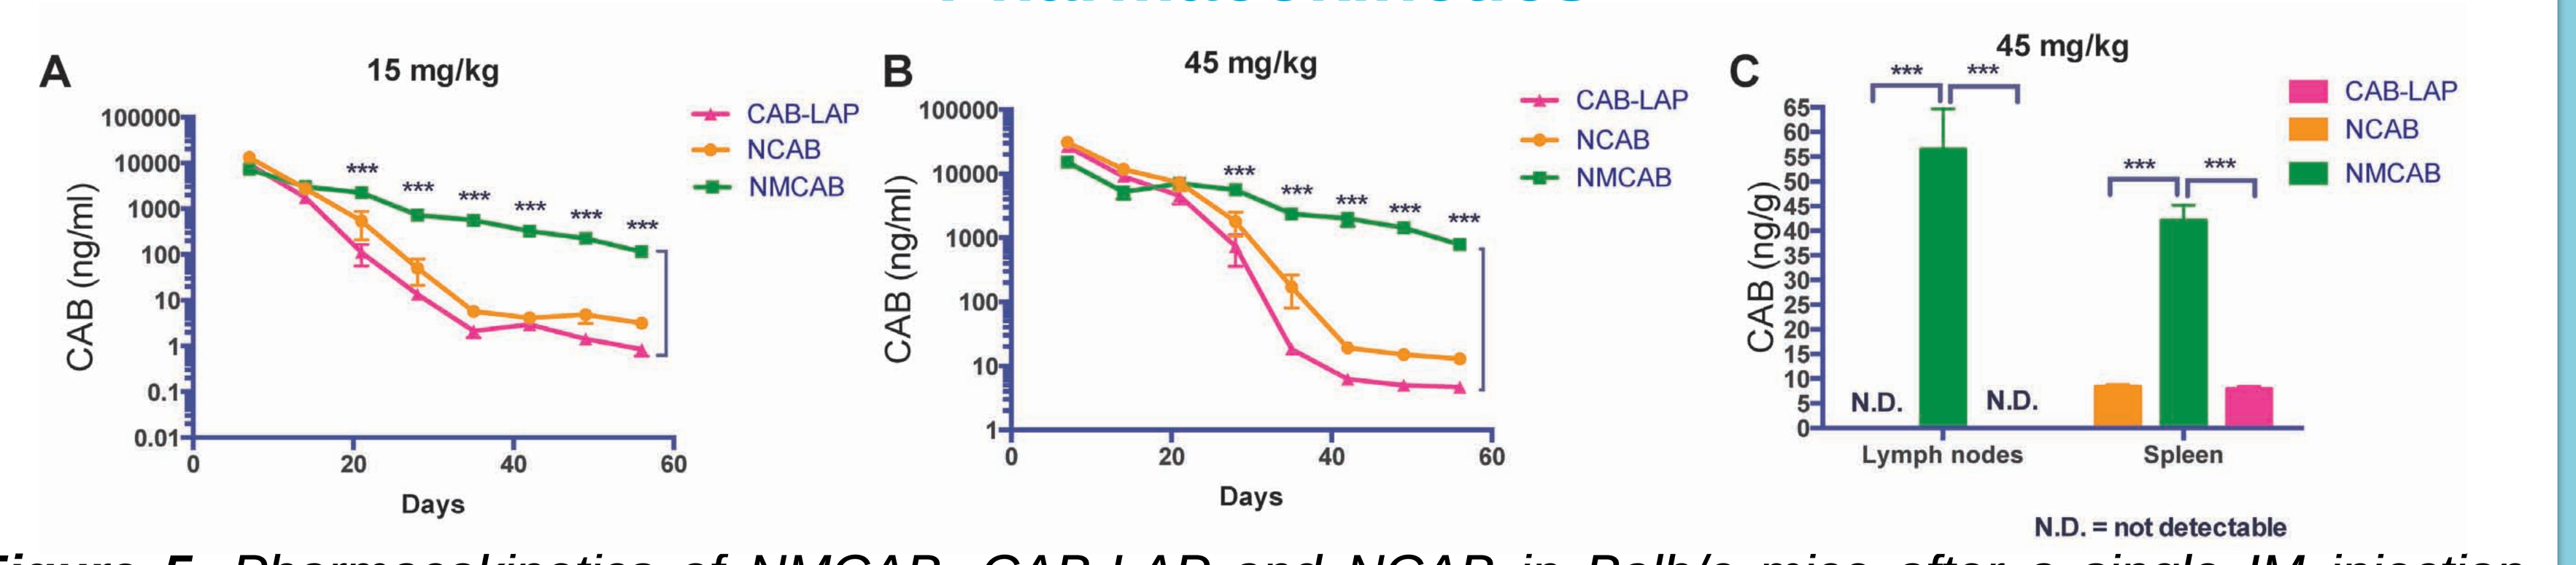
A
15 mg/kg
B
45 mg/kg
CAB-LAP
NCAB
NMCAB
CAB (ng/ml)
Days
C
45 mg/kg
CAB (ng/g)
N.D.
N.D.
Lymph nodes
Spleen
N.D. = not detectable

Figure 5. *Pharmacokinetics of NMCAB, CAB-LAP and NCAB in Balb/c mice after a single IM injection. Plasma CAB levels were determined over two months in mice treated with (A) 15 mg/kg and (B) 45 mg/kg CAB equivalents. (C) CAB levels in mouse lymph nodes and spleens 2 months after treatment with 45 mg/kg CAB equivalents.*

Pre-Exposure Prophylaxis

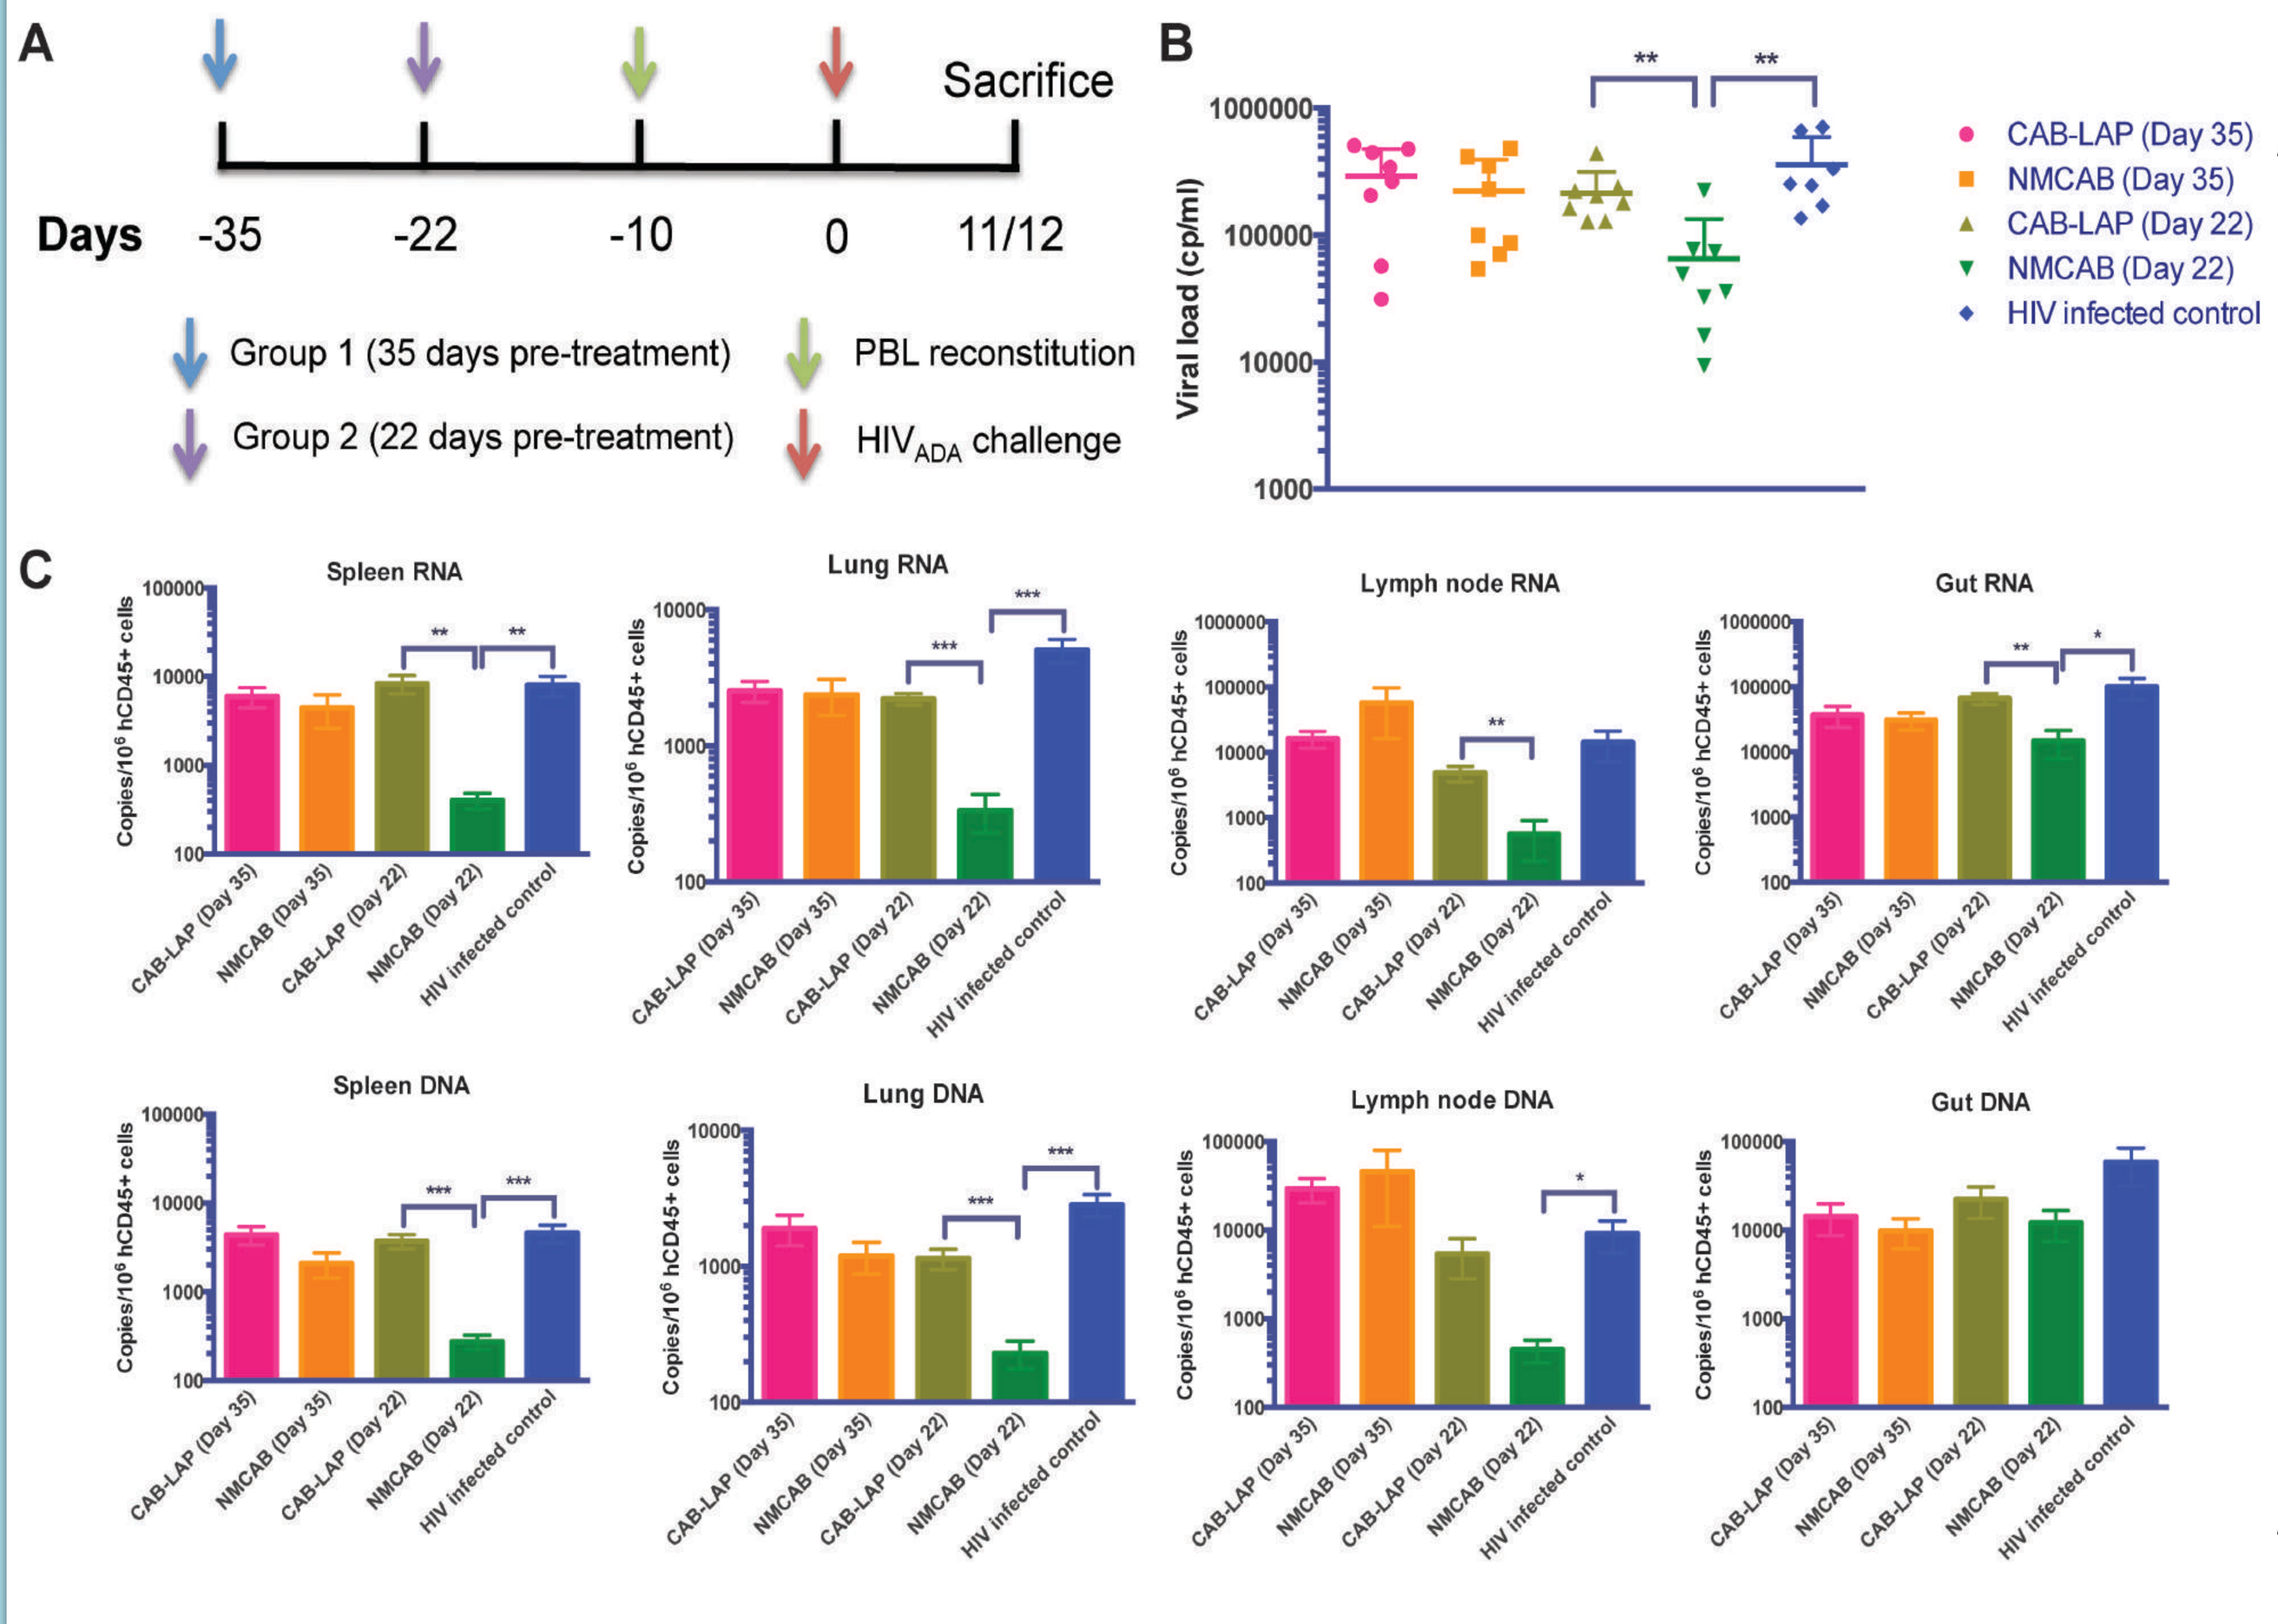
A
Sacrifice
Days -35 -22 -10 0 11/12
Group 1 (35 days pre-treatment)
Group 2 (22 days pre-treatment)
PBL reconstitution
HIV_ADA challenge
B
Viral load (cp/ml)
CAB-LAP (Day 35)
NMCAB (Day 35)
CAB-LAP (Day 22)
NMCAB (Day 22)
HIV infected control
C
Spleen RNA
Lung RNA
Lymph node RNA
Gut RNA
Spleen DNA
Lung DNA
Lymph node DNA
Gut DNA
Copies/10⁶ hCD45+ cells

Figure 6. *Pre-exposure prophylaxis of NMCAB versus CAB-LAP. NSG mice were treated intramuscularly with 15 mg/kg NMCAB or CAB-LAP at different times prior to reconstitution with human PBL and HIV-1_{ADA} challenge. Animals were sacrificed 11/12 days after HIV challenge. (A) Study timeline. (B) Plasma viral load. (C) Tissue HIV RNA and DNA. The data demonstrates a clear association from pharmacodynamics profiles of NMCAB with its improved pharmacokinetic test results.*

Acknowledgments: This work was supported by National Institutes of Health grants P01 DA028555, R01 NS36126, P01 NS31492, 2R01 NS034239, P01 MH64570, P01 NS43985, P30 MH062261, and AG043540 and by ViiV Healthcare.

University of Nebraska Medical Center