Background: Despite effective antiretroviral therapy (ART), people living with HIV (PLWH) still experience excess morbidity and mortality, with a growing awareness of the impact of non-AIDS defining events (NADE) and the factors potentially associated with the occurrence of these events including failure to normalise the CD4:CD8 ratio.

Method: Adult PLWH enrolled in the Mater ID Cohort Study who commenced ART after January 1st 2001 were included in an analysis determining prevalence of and associations with AIDS events and NADE. Demographic, laboratory (including HIV RNA, CD4+ and CD8+ T-cell counts, CD4:CD8 ratio) and clinical events (AIDS and NADE) were collated. Multivariable Cox proportional hazards regression models explored factors independently associated with the progression to NADE. Data are reported as median (IQR).

Results: Of 550 PLWH, 317 (58%) were male, 299 (54%) Caucasian, 220 (40%) African, 114 (21%) Men who have sex with Men and 131 (24%) Injecting drug users (IDU). 128 (23%) were co-infected with Hepatitis C. At ART initiation median age was 34 (29, 40) yrs, and nadir CD4+ count 187 (80, 284) cells/mm3. Of 135 NADE in 2557 person years of follow (crude incidence 5.3 per 100 PYFU), the commonest were pneumonia (n=39), liver disease (n=17), cardiovascular disease (CVD) (n=14) and non AIDS malignancies (n=12). Of 23 deaths, 5 were AIDS related and 11 were NADE (malignancy (n=7), liver disease (n=2), CVD (stroke) (n=1), abdominal sepsis (n=1)). In multivariable Cox models, older age at ART initiation, IDU risk and lower quintiles of pre-event CD4:CD8 ratio were independently associated with an increased risk of non-AIDS defining events, with male gender and non-Caucasian ethnicity independently associated with reduced risk of NADE. A sensitivity analysis in those virally suppressed revealed similar associations with age at ART initiation (HR=1.59, 95% CI 1.23-2.05), gender (HR=0.33, 95% CI 0.20-0.56) and lower pre-event CD4:CD8 ratio (CD4:CD8 ratio≤0.26; HR=3.11 (95% CI 1.44-6.71),CD4:CD8 ratio 0.27-0.43; HR=1.64 (0.74—3.66), CD4:CD8 ratio 0.44-0.59; HR= 1.67 (0.77-3.66), CD4:CD8 ratio 0.60-0.86; HR=1.26 (0.54-2.93)).

Conclusion: This is the first study to show an incremental association between pre-event CD4:CD8 ratio and NADE

Incremental Association between CD4:CD8 Ratio and Incidence of non-AIDS Events

Poster #710

P McGettrick [1,2], W Tinago[1], A Lacey[1] , A Macken [1], A Ni Fhlaitheartaigh [1], S Tennant[1], G Sheehan [1,2], J S Lambert[1,2] , PWG Mallon[1,2]

1. HIV Molecular Research Group, School of Medicine, University College Dublin, Dublin, Ireland
2. Department of Infectious Disease, Mater Misericordae University Hospital, Dublin

Corresponding Author: Padraig McGettrick
Address: HIV Molecular Research Group, School of Medicine, University College Dublin, Dublin Ireland
Email: padrigmcgettrick@gmail.com
Phone: +353 1 7164558

Introduction

- Despite effective antiretroviral therapy (ART), people living with HIV (PLWH) still experience excess morbidity and mortality.
- Non-AIDS defining events (NADE) have emerged as the leading cause of morbidity and mortality in this population[1,2].
- Reduced CD4:CD8 ratio in PLWH despite effective ART has been associated with an increased risk of NADE[3,4] and has been suggested as a marker to identify those at risk of developing these events.

Objective

- We aimed to determine the prevalence of AIDS events and NADE in our cohort and to investigate an association between NADE and the CD4:CD8 ratio

Methods

- Single center retrospective cohort analysis including all adult PLWH enrolled in the Mater ID Cohort Study who commenced ART after January 1st 2001.
- Demographic, laboratory and clinical events (AIDS and NADE) were collated.
- Multivariable Cox proportional hazards regression models, correcting for age, gender, ethinicity, and transmission risk, explored factors independently associated with the progression to NADE.
- Data are reported as median (IQR).

Results

- 550 PLWH were included in the analysis (See Table 1).
- There were 164 NADE of which 135 were first time events in 2557 person years of follow up (crude incidence 5.3 per 100 PYFU) (See Table 2).

Acknowledgements

We would like to thank the patients and staff of Mater Misericordae University Hospital, Dublin.The MMUH ID Cohort receives unrestricted funding from BMS, Jansen Cilag and MSD

Results continued

Table 1: Characteristics of subjects

	N=550
Gender: Male	317 (58%)
Age at ART initiation (median (IQR))	34 (29-40)
Ethnicity	
Caucasian	299 (54%)
African	220 (40%)
Other	31 (6%)
Mode of HIV infection	
Heterosexual contact	279 (51%)
Men having sex with men (MSM)	114 (21%)
Injecting drug use (IDU)	131 (24%)
Other	20 (4%)
Hepatitis C Ab: Positive	128 (23%)
Hepatitis B sAg: Positive	22 (4%)
Nadir CD4+ T-cell	187 (80-284)
Most recent CD4+ T-cell count	545 (389-717)
Most recent CD4+/CD8+ T-cell ratio	0.70 (0.39-0.92)
Most recent CD4+/CD8+ > 1.0 (n(%))	125 (23%)
Most recent HIV RNA<50 copies/ml	488 (89%)
Current ART regimes	
PI-based	235 (43%)
NNRTI-based	236 (43%)
INSTI-based	61 (11%)
Other	18 (3%)

Table 2: List of Non-AIDS Events

Non-AIDS clinical events	N=164
Cardiovascular events	
Ischemic Heart Disease	9
Cerebrovascular disease	3
Peripheral vascular disease	2
Renal Disease	6
Liver disease	17
Respiratory events	
Pneumonia	39
COPD	10
Non AIDS Malignancy	12
Other non-AIDS events	
Fractures	3
Pre-malignancies	47
Diabetes	10

- Of 23 deaths, 5 were AIDS related, 12 NADE and 6 from misadventure/trauma/unknown cause (See Table 3). The most common NADE related deaths were secondary to malignancy, lung carcinoma being the most frequent cause.

Table 3: Cause of Death

AIDS-related deaths	**N=5**
Lymphoma	3
Progressive multifocal leukoencephalopathy	1
Immunocompromised	1
Non-AIDS-related deaths	**N=12**
Respiratory malignancy	3
Oropharyngeal malignancy/other	2
Liver disease	2
Cardiovascular disease/Strioke	2
Pneumonia	2
Ecoli septicaemia	1
Unnatural/ Unknown	**6**

Figure 1: Factors associated with NADE

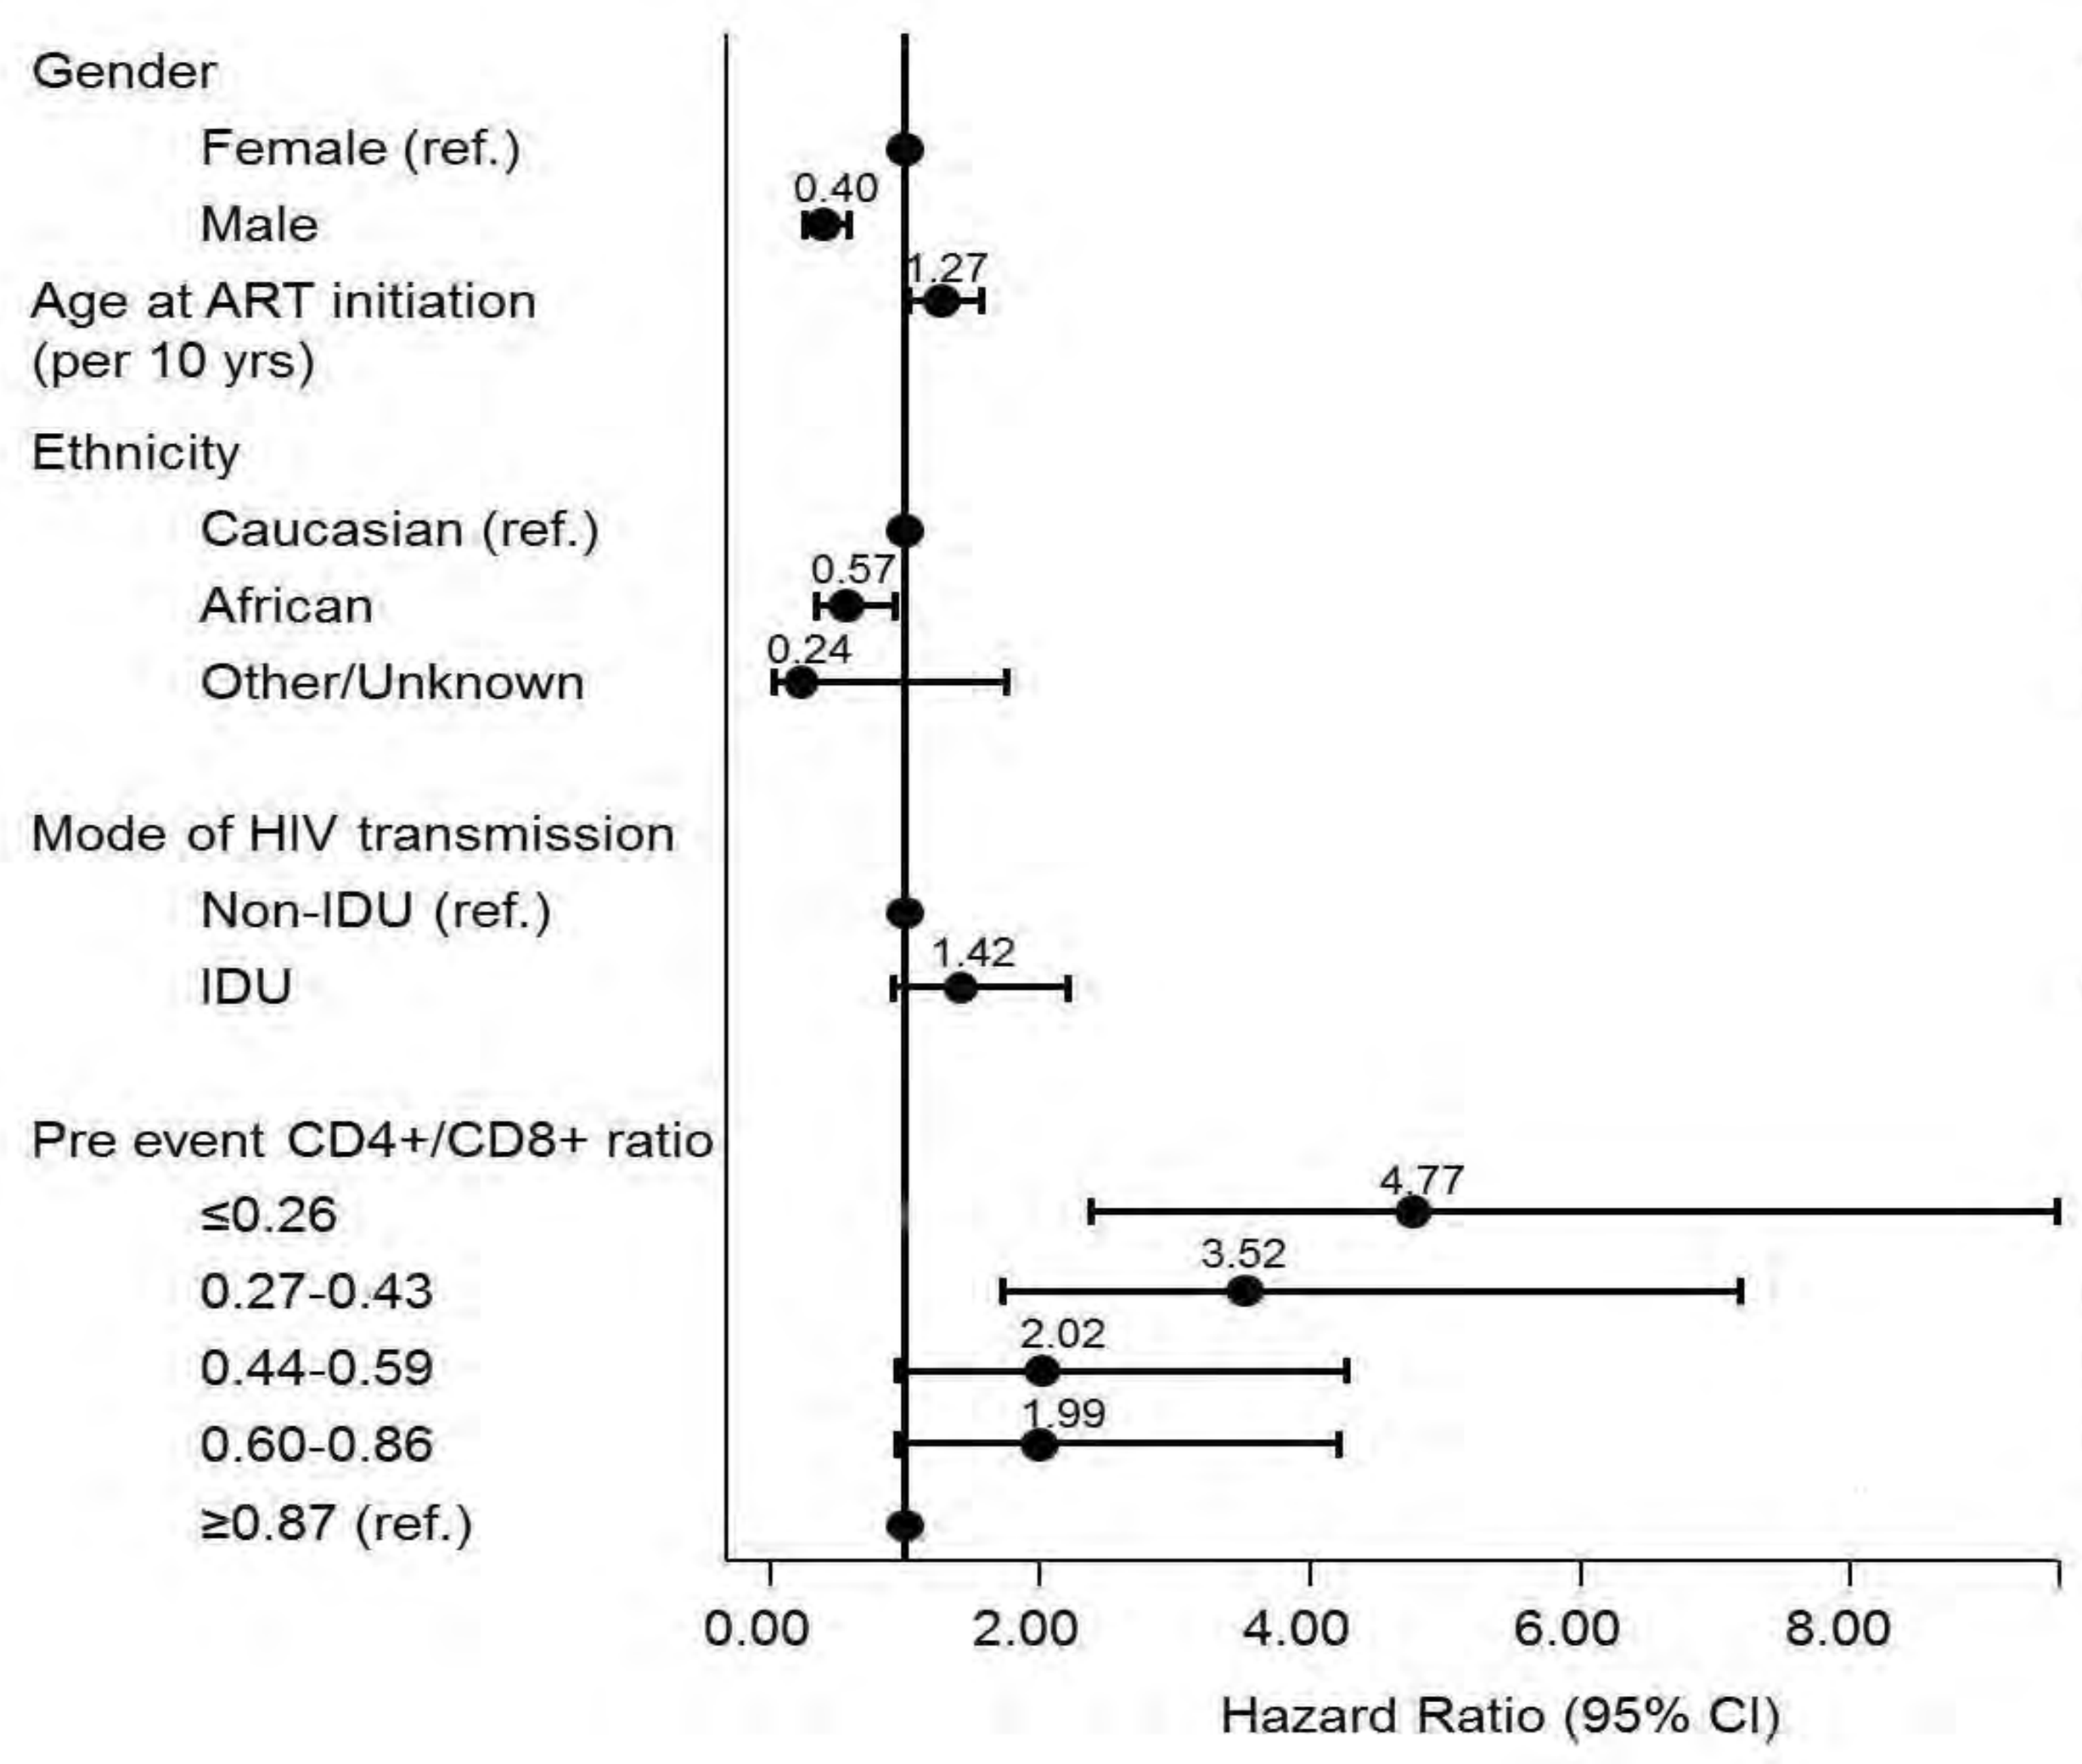
Gender
Female (ref.)
Male
0.40
<0.001
Age at ART initiation (per 10 yrs)
1.27
0.03
Ethnicity
Caucasian (ref.)
African
0.57
0.02
Other/Unknown
0.24
0.16
Mode of HIV transmission
Non-IDU (ref.)
IDU
1.42
0.12
Pre event CD4+/CD8+ ratio
≤0.26
4.77
<0.001
0.27-0.43
3.52
0.001
0.44-0.59
2.02
0.07
0.60-0.86
1.99
0.07
≥0.87 (ref.)
P-value
0.00
2.00
4.00
6.00
8.00
Hazard Ratio (95% CI)

- In multivariable Cox models, older age at ART initiation, IDU risk and lower quintiles of pre-event CD4:CD8 ratio were independently associated with an increased risk of NADE (See Figure 1).

Results continued

- Male gender and non-Caucasian ethnicity were independently associated with reduced risk of NADE
- A sensitivity analysis in those virally suppressed revealed similar associations with age at ART initiation (HR=1.59, 95% CI 1.23-2.05), gender (HR=0.33, 95% CI 0.20-0.56) and lower pre-event CD4:CD8 ratio (CD4:CD8 ratio≤0.26; HR=3.11 (95% CI 1.44-6.71), CD4:CD8 ratio 0.27-0.43; HR=1.64 (0.74—3.66), CD4:CD8 ratio 0.44-0.59; HR= 1.67 (0.77-3.66), CD4:CD8 ratio 0.60-0.86; HR=1.26 (0.54-2.93)).

Conclusions

- This is the first study to show an incremental association between pre-event CD4:CD8 ratio and NADE.
- This differs from previous studies which have demonstrated an increased risk of NADE below a threshold value[6,7,8] or per quantile decrease[3]
- This study is among the first to demonstrate an association between CD4:CD8 ratio and NADE including non inflammatory and non malignant conditions (i.e. Diabetes Mellitus, COPD, Serious Infection, Premalignancy) reflecting non-AIDS comorbidity
- It is yet to be determined what impact, if any, strategies to improve CD4:CD8 ratio will have on prevalence of NADE.

Limitations

- Single centre retrospective cohort analysis
- Due to low numbers of individual events, unable to assess association between specific types of event and CD4:CD8 ratio
- Unable to correct for traditional risk factors (e.g Smoking, family history) due to incomplete data

References

1.Smith CJ, Ryom L, Weber R, Morlat P, Pradier C, Reiss P, et al, Lancet (London, England). 2014;384(9939):241-8.
2.Mitsuyasu RT. Topics in antiviral medicine. 2014;22(3):660-5
3.Serrano-Villar S, Perez-Elias MJ, Dronda F, Casado JL, Moreno A, Royuela A, et al.,PloS one. 2014;9(1):e85798
4.Lu W, Mehraj V, Vyboh K, Cao W, Li T, Routy JP. Journal of the International AIDS Society. 2015;18:20052
5.Seng R, Goujard C, Krastinova E, Miailhes P, Orr S, Molina JM, et alA IDS (London, England). 2015;29(5):595-607
6.Mussini C, Lorenzini P, Cozzi-Lepri A, Lapadula G, Marchetti G, Nicastri E, et al.. The lancet HIV. 2015;2(3):e98-106
7.Bernal E, Serrano J, Perez A, Valero S, Garcia E, Marin I, et al. Journal of the International AIDS Society. 2014;17(4 Suppl 3):19723.
8.Cervero M, Torres R, Agud JL, Pastor S, Jusdado JJ.Medicina clinica. 2015.