P-432

Minimal Removal of Dolutegravir by Hemodialysis in HIV-Infected Patients

e-mail: jmolto@flsida.org

José Moltó[1,2], Fredzia Graterol[3], Cristina Miranda[1], Saye Khoo[4], Ioana Bancu[3], Alieu Amara[4], Anna Bonjoch[1], Bonaventura Clotet[1, 2, 5, 6].

[1]Lluita contra la Sida Foundation, Hospital Universitari Germans Trias i Pujol, Badalona, Spain. [2]Universitat Autónoma de Barcelona (UAB), Barcelona, Spain. [3]Nephrology Department, Hospital Universitari Germans Trias i Pujol, Badalona, Spain. [4]Department of Molecular and Clinical Pharmacology, University of Liverpool, UK. [5]Universitat de Vic (UVic), Vic, Spain. [6]IrsiCaixa Foundation, Hospital Universitari Germans Trias i Pujol, Badalona, Spain

SUMMARY

Data on dolutegravir removal by hemodialysis are lacking. To study this, we measured dolutegravir plasma concentrations in samples of blood entering and leaving the dialyzer, and resulting dialysate from 5 HIV-infected patients with end-stage renal disease. Median dolutegravir hemodialysis extraction ratio was 7%. Dolutegravir concentrations after the dialysis session remained far above the protein-binding-adjusted inhibitory concentration. Our results show minimal dolutegravir removal by hemodialysis with no specific dolutegravir dosage adjustment required in this setting.

BACKGROUND & OBJECTIVE

Chronic kidney disease may be considered epidemic among HIV-infected patients, and the number of patients with end-stage renal disease (ESRD) requiring hemodialysis as renal replacement therapy is increasing.

HIV drugs differ in the extent of their removal from blood by hemodialysis, and specific dosing recommendations may be required. Although no dolutegravir dosage adjustment is necessary for patients with creatinine clearance <30 ml/min, little is known about dolutegravir removal from plasma by hemodialysis.

Our objective was to evaluate the effect of hemodialysis on dolutegravir concentrations in HIV+ patients with ESRD.

METHODS

Single-centre, open-label, pilot study in HIV-infected patients with ESRD undergoing routine hemodialysis.

After enrolment (day 1), dolutegravir (Tivicay, Viiv Healthcare) 50 mg once daily was added to cART for five days. Patients were told to take dolutegravir in the morning, separated from other drugs that could interfere with dolutegravir absorption (e.g., antacids, multivitamins, chelating agents, etc.).

On day 5, dolutegravir concentrations in plasma were determined (LC-MS/MS) in blood samples collected from each patient at the beginning (C_{pre}) and at the end of a dialysis session (C_{post}), as well as in paired samples of blood entering (C_{in}) and leaving (C_{out}) the dialyzer, and in resulting dialysate.

The hemodialysis extraction ratio (ER) for dolutegravir was calculated as $ER(\%) = \frac{C_{in} - C_{out}}{C_{in}} \times 100$

where C_{in} is predialyzer dolutegravir concentration (i.e., blood entering the dialyzer), and C_{out} is postdialyzer dolutegravir concentration (i.e., blood leaving the dialyzer).

Due to the high protein binding of dolutegravir, postdialyzer concentrations (C_{out}) were corrected for hemoconcentration by a factor F based on total protein (TP) concentration pre- and postdialyzer: $F=TP_{in}/TP_{out}$

RESULTS

Five anuric HIV-infected patients (4 men, 1 woman), undergoing routine hemodialysis (n=3) or on-line hemodiafiltration (OL-HDF, n=2) were included in the study.

Median (range) age and body weight were 53.0 (41.3-69.5) years and 77.1 (57.5-91.9) kg, respectively.

The cART regimen included boosted protease inhibitors in three participants, and non-nucleoside reverse transcriptase inhibitors in two. Additionally, four of the patients were receiving raltegravir at enrolment.

Dolutegravir was well tolerated and all participants completed the study. At the start of the dialysis session on day 5, median (range) time after the last dolutegravir dose was 5.9 (5.6-6.4) hours.

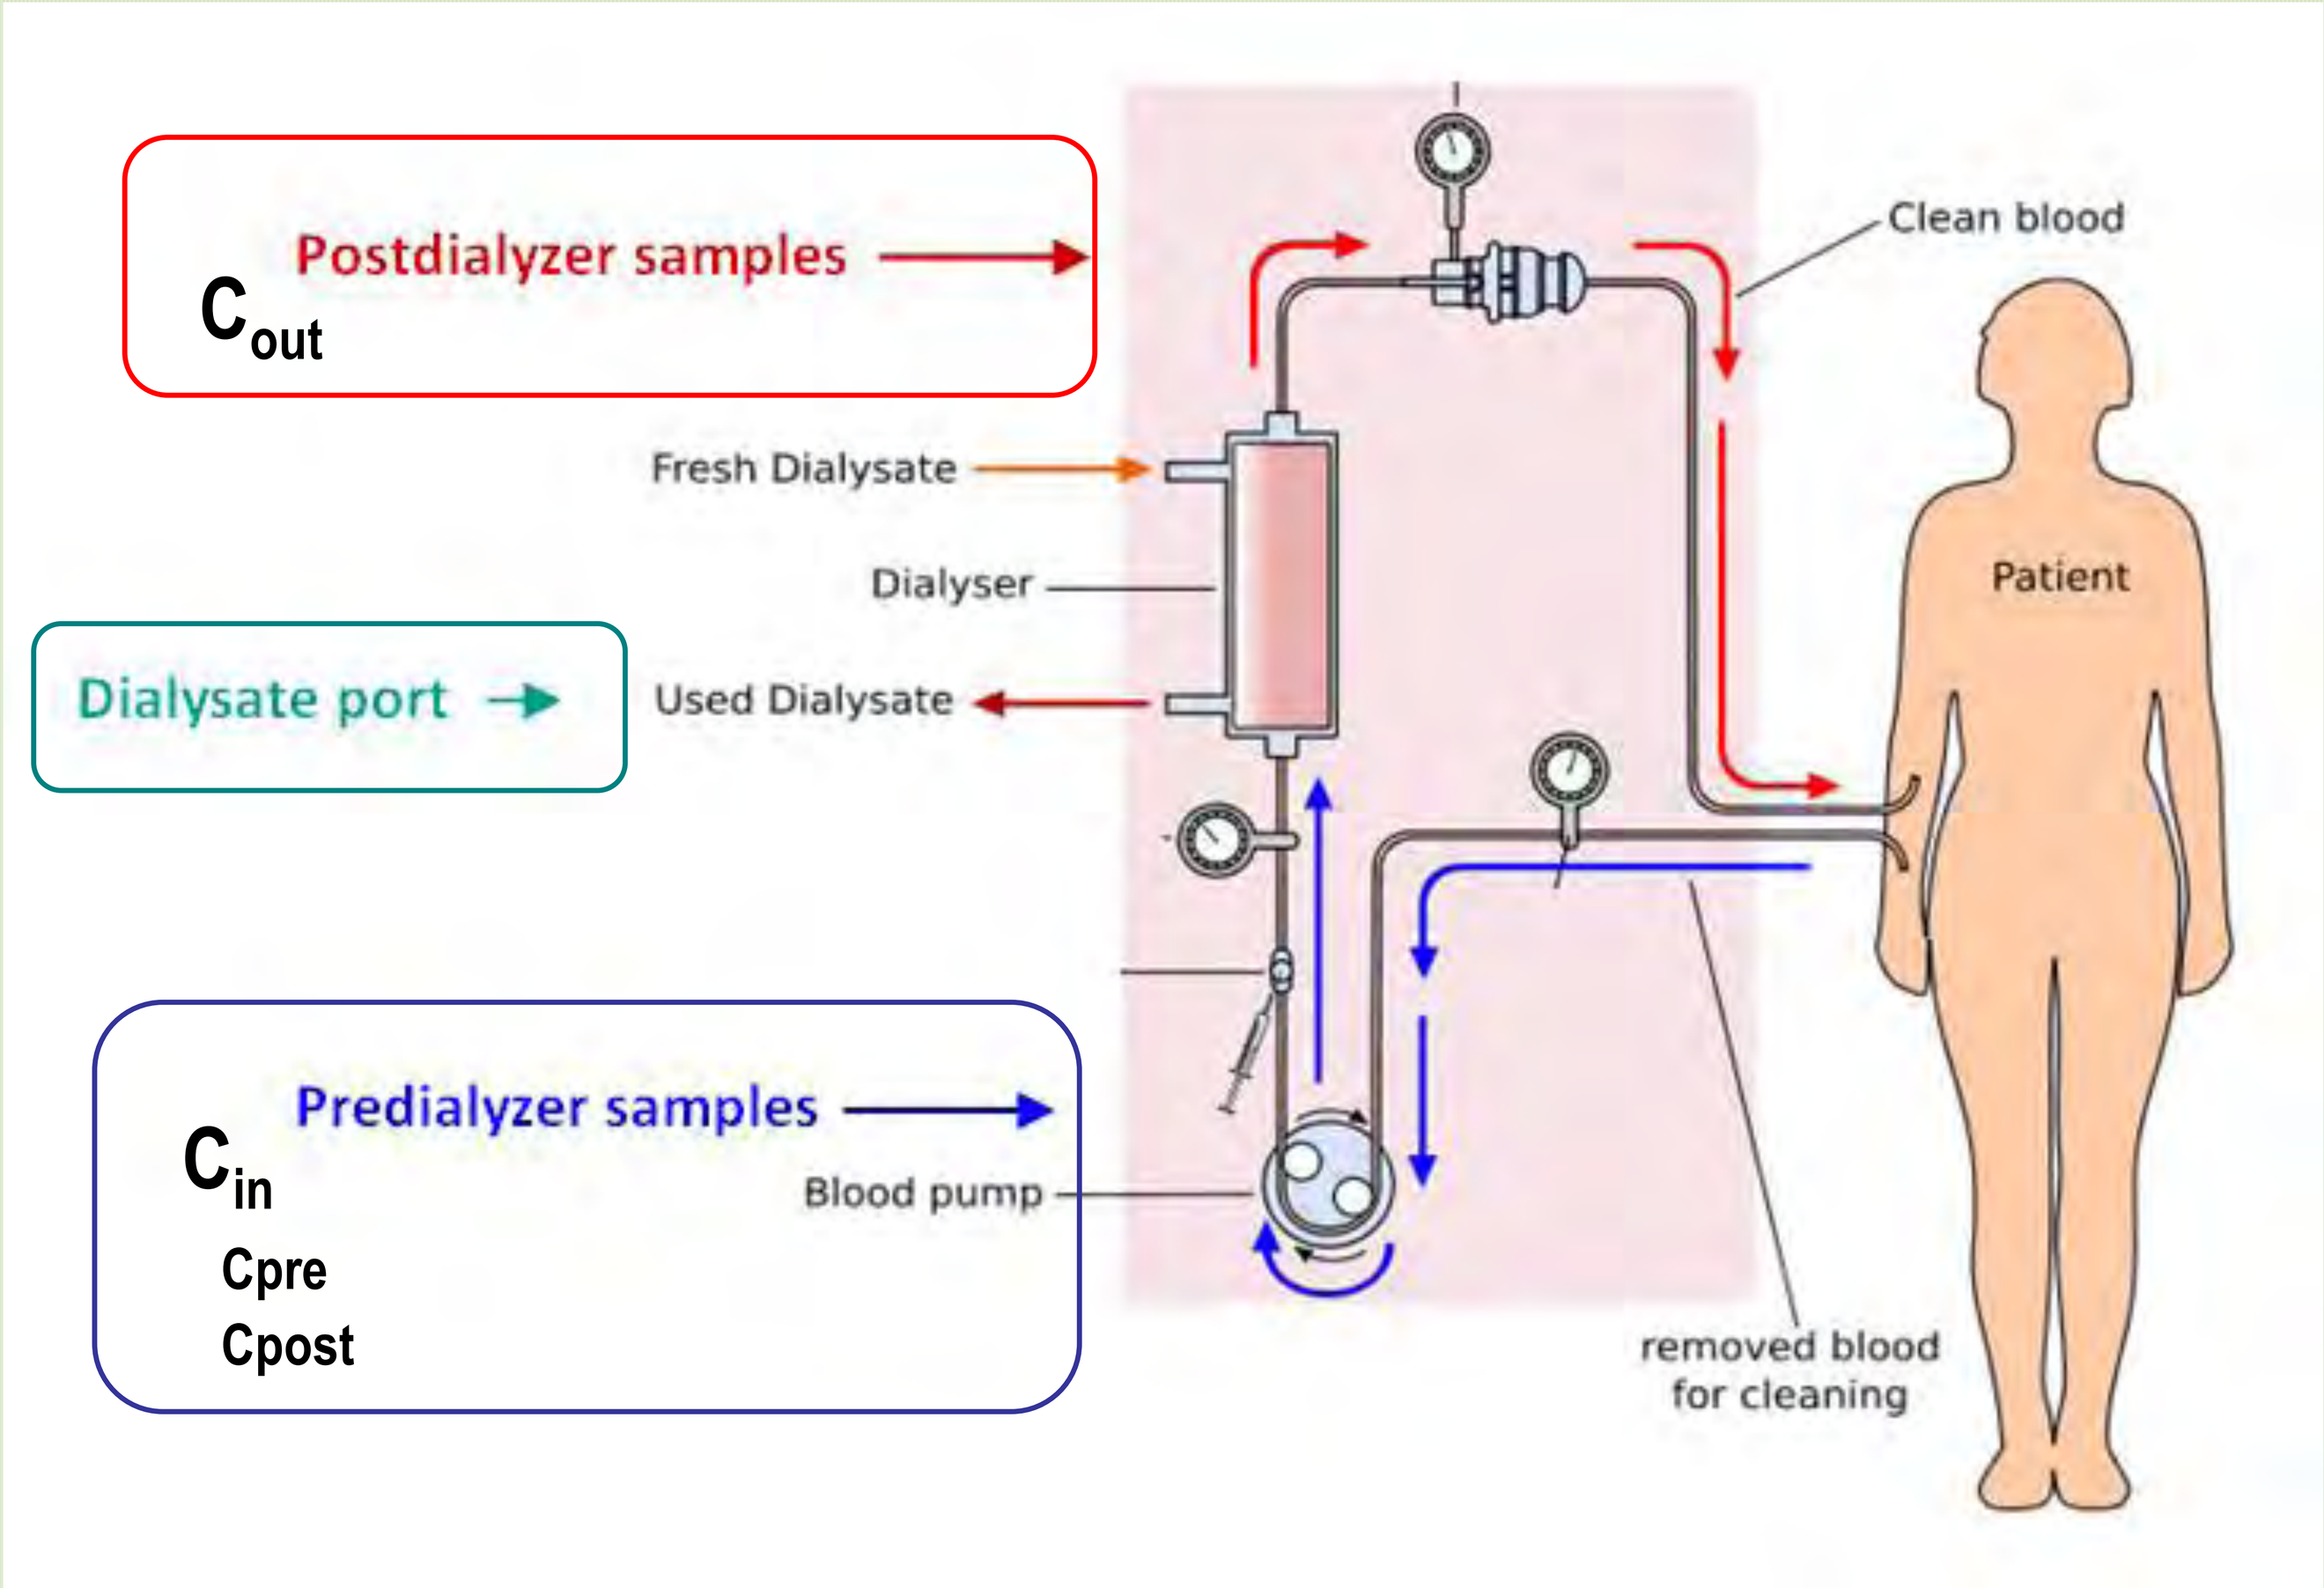
Postdialyzer samples
Cout
Fresh Dialysate
Dialyser
Dialysate port
Used Dialysate
Predialyzer samples
Cin
Cpre
Cpost
Blood pump
Clean blood
Patient
removed blood
for cleaning

Dialysis specifications

- Conventional hemodialysis: Revaclear®400, capillary dialyzer, membrane area 1.8 m2, bicarbonate cartridge Bicart® Gambro. Blood flow 300 ml/min. Dialysate flow 500 ml/min. Duration 4h.

-OL-HDF: Polyflux® 210H, capillary dialyzer, membrane area 2.1 m2, bicarbonate cartridge Bicart® Gambro. Blood flow 400 ml/min. Dialysate flow 500 ml/min.Duration 4h.

DOLUTEGRAVIR CONCENTRATIONS DURING THE DIALYSIS SESSION

Patient no.	Type of hemodialysis	C_{in} (mg/l)	C_{out} (mg/l)	Dialysate (mg/l)	ER (%)	C_{pre} (mg/l)	C_{post} (mg/l)
1	OL-HDF	0.47	0.98	0.02	1	0.48	0.86
2	Conventional	1.51	2.73	0.02	20	1.52	2.18
3	OL-HDF	1.90	3.90	0.03	7	1.62	2.77
4	Conventional	1.47	1.43	0.06	3	1.58	1.14
5	Conventional	2.13	2.59	0.04	25	2.90	2.52
Median		**1.51**	**2.59**	**0.03**	**7**	**1.52**	**2.18**

CONCLUSIONS

The minimal extraction ratio of dolutegravir by hemodialysis coupled with dolutegravir concentrations in plasma far above the protein-binding-adjusted IC_{90} (0.064 mg/l) in this study support no dolutegravir dose adjustment in HIV-infected patients with ESRD undergoing hemodialysis.

FUNDING

This work was funded by Viiv Healthcare and by the “Lluita contra la SIDA” Foundation. The funders had no role in study design, data collection and interpretation. We acknowledge infrastructural support from the Liverpool Biomedical Research Centre funded by Liverpool Health Partners.
The authors have no commercial or other association that might pose a conflict of interest.

Germans Trias i Pujol Hospital

FIGHT AIDS FOUNDATION

UNIVERSITY OF LIVERPOOL