Ishani Savant Landry, PhD
Associate Director
Clinical Pharmacology & Pharmacometrics
Virology
Bristol-Myers Squibb
PO Box 4000
Princeton NJ 08543 USA
Email: ishani.landry@bms.com
Phone: +1 (609) 252 7674

Poster 523

HIV-1 Attachment Inhibitor Prodrug BMS-663068: Interactions with DRV/r and/or ETR

I Savant Landry,[1] X Tao,[1] J Anderson,[1] M Hesney,[1] M Stonier,[1] S Lubin,[1] J Wang,[1] GJ Hanna,[1] DW Boulton[1]

[1]Bristol-Myers Squibb, Princeton, NJ, USA

BACKGROUND

- Novel antiretroviral agents are needed for treatment-experienced patients who may have limited treatment options due to drug resistance, drug intolerabilities and contraindications.[1,2]
- BMS-663068 is a prodrug metabolized to the active moiety BMS-626529, a first-in-class attachment inhibitor that binds to HIV-1 gp120, preventing initial viral attachment and entry into the host CD4+ T cell, and:[3,4]
 - has *in vitro* activity against HIV-1 viruses (except subtype AE and Group O)[5]
 - is active against CCR5-, CXCR4- and dual-tropic (R5X4) strains of HIV-1[5–8]
 - has a unique resistance profile with no *in vitro* cross-resistance to other classes of antiretrovirals.[5,8]
- In an ongoing Phase IIb study (AI438011) BMS-663068 has shown generally similar efficacy to ritonavir (RTV)-boosted atazanavir, each with tenofovir disoproxil fumarate and raltegravir, in HIV-1-infected, treatment-experienced patients.[9]
 - All BMS-663068 doses were generally well tolerated with no dose response safety signals reported.[9]

Study background and rationale

- AI438020 is a Phase I, open-label, single-sequence study in healthy subjects designed to evaluate the pharmacokinetic (PK) interaction of BMS-626529 (administered as the prodrug BMS-663068) with:
 - RTV-boosted darunavir (DRV/r) and/or etravirine (ETR).
- DRV/r and ETR are commonly used in HIV-positive, treatment-experienced patients. Thus, understanding potential drug-drug interactions between BMS-663068 and DRV/r and/or ETR is of clinical relevance.
- A previous Phase I study (n=44) of BMS-663068 1200 mg BID/single dose (SD), RTV 100 mg BID/SD and DRV 600 mg BID/SD in healthy subjects was terminated due to one instance of anaphylaxis (Grade 3 rash and Grade 1 angiodema) in a subject receiving BMS-663068 1200 mg + RTV 100 mg BID.[10]
 - Although RTV can be associated with angioedema and severe rash, a possible role of both RTV and BMS-663068 in the events could not be excluded.
 - Hence, the interaction study was conducted again to evaluate the safety profile of BMS-663068 when coadministered with DRV/r.
- BMS-626529 is a substrate of the P-glycoprotein transporter and predominately metabolized by an esterase-mediated hydrolysis pathway, with contributions from a cytochrome P450 (CYP)3A4-mediated oxidative pathway.
- DRV and RTV are inhibitors of CYP3A4, while ETR is an inducer of CYP3A4.
- Therefore, coadministration of DRV/r and/or ETR with BMS-663068 may affect BMS-626529 PK characteristics.
- Conversely, as BMS-626529 is not an inhibitor or inducer of DRV/r or ETR metabolism, no effect on the PK of these compounds is anticipated.

OBJECTIVES

Primary

- To assess the effects on the PK of BMS-626529 of coadministering BMS-663068 (600 mg BID) with:

DRV/r (600/100 mg BID)

ETR (200 mg BID)

ETR (200 mg BID) and DRV/r (600/100 mg BID)

Cohort 1 Cohort 2 Cohort 3

DRV/r, ritonavir-boosted darunavir; ETR, etravirine; PK, pharmacokinetics.

Secondary

- To assess the effect of coadministering BMS-663068 (600 mg BID) with:

DRV/r (600/100 mg BID) on the PK of DRV and RTV

ETR (200 mg BID) on the PK of ETR

ETR (200 mg BID) and DRV/r (600/100 mg BID) on the PK of ETR and DRV/r

Cohort 1 Cohort 2 Cohort 3

DRV/r, ritonavir-boosted darunavir; ETR, etravirine; PK, pharmacokinetics; RTV, ritonavir.

- To assess the safety and tolerability of BMS-663068 administered alone and in combination with DRV/r and/or ETR.

METHODS

Study population

- Healthy male and female subjects, 18–50 years of age and eligible to participate in the study.
 - Target of 42 subjects (14 per cohort).

Study design

- Eligible subjects were randomly assigned to Cohort 1, 2 or 3 before dosing on Day 1 (Figure 1).
- Dosing in each cohort was on Days 1–4, 7–16 and 17–26, with a 2-day washout on Days 5 and 6.
- Drug administration was under fed conditions.

Figure 1: AI438020 study design

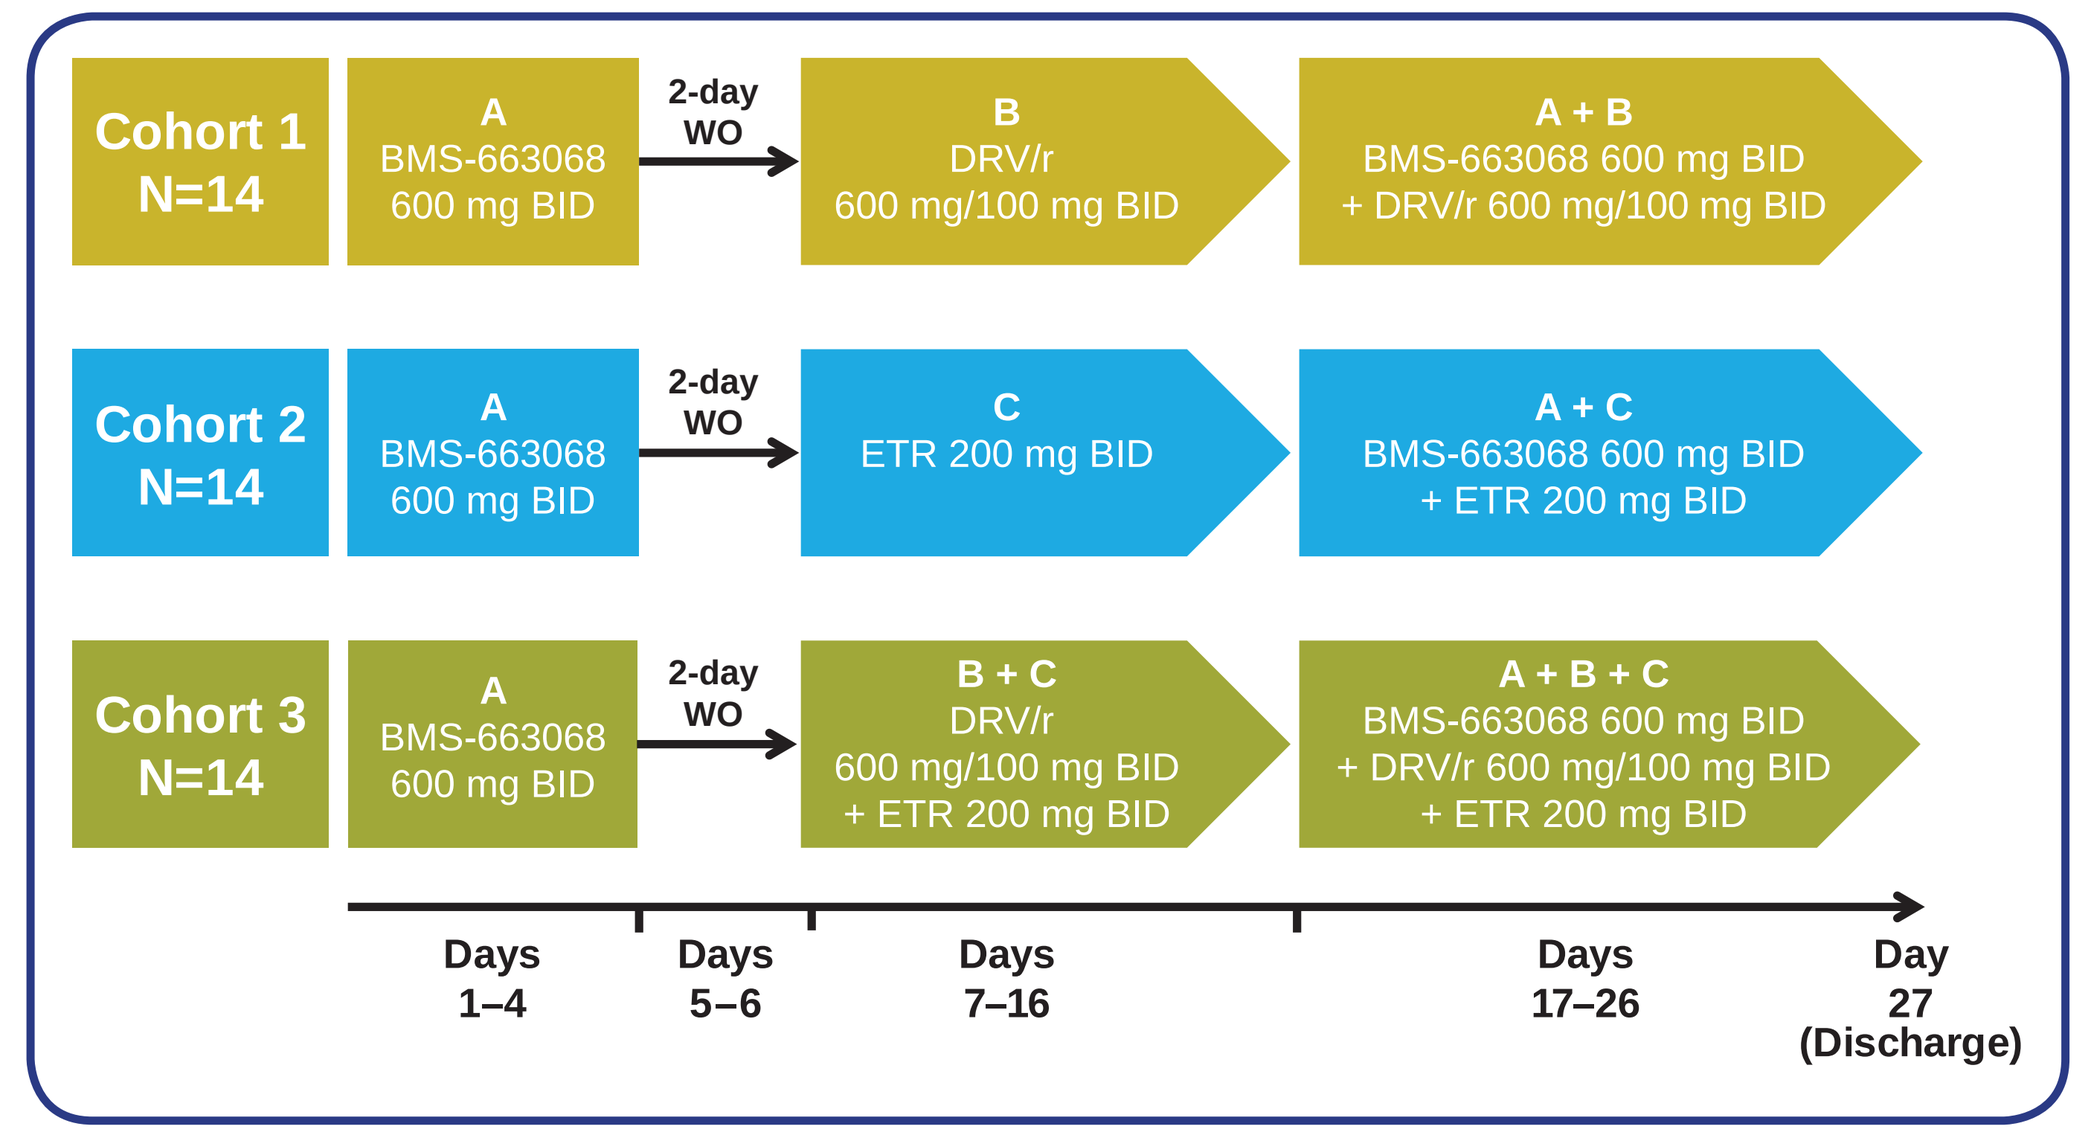
Cohort 1 N=14
A BMS-663068 600 mg BID
2-day WO
B DRV/r 600 mg/100 mg BID
A + B BMS-663068 600 mg BID + DRV/r 600 mg/100 mg BID
Cohort 2 N=14
A BMS-663068 600 mg BID
2-day WO
C ETR 200 mg BID
A + C BMS-663068 600 mg BID + ETR 200 mg BID
Cohort 3 N=14
A BMS-663068 600 mg BID
2-day WO
B + C DRV/r 600 mg/100 mg BID + ETR 200 mg BID
A + B + C BMS-663068 600 mg BID + DRV/r 600 mg/100 mg BID + ETR 200 mg BID
Days 1–4
Days 5–6
Days 7–16
Days 17–26
Day 27 (Discharge)

DRV/r, ritonavir-boosted darunavir; ETR, etravirine; WO, washout.

Assessments

- PK measures
 - Multiple-dose parameters (C_{max}, AUC_{tau}, T_{max}, C_{trough} [C_0 pre-dose] and C_{12}) were derived from plasma concentration versus time data for each of the analytes.
 - Samples were taken pre-dose, then at 30-minute to 12-hour intervals after dosing during each of the treatment periods.
- Safety measures
 - Reported adverse events (AEs), clinical laboratory tests, vital sign measurements, electrocardiogram measurements and physical examinations were recorded throughout the study.

Statistics

- Sample size determination was based on the precision of the estimate of the geometric mean ratios (GMRs) of C_{max} and AUC_{tau} of BMS-626529 with and without the other coadministered drugs.
 - No-effect 90% confidence interval (CI) limits of 0.75–1.70 were predefined for DRV and ETR.
- PK parameters were derived using non-compartmental methods.
- GMRs and 90% CIs were calculated from log-transformed C_{max}, AUC_{tau} and C_{12} using linear mixed-effect modeling on log-transformed data, with treatment as the fixed effect and within-subject assessments as the repeated measures.
- No formal statistical analysis of safety parameters was performed.

RESULTS

Subject disposition, demographics and disease characteristics

- Of 100 subjects enrolled, 46 were randomized.
 - Although the target randomization was 42 subjects, an additional four subjects were enrolled in Cohort 3 to replace subjects who discontinued due to AEs.
- Thirty-nine subjects completed the study.
 - Seven were discontinued because of AEs.
- Baseline characteristics and demographics were generally well balanced between cohorts (Table 1).

Table 1: Baseline characteristics and demographics

Characteristics	Cohort 1 n=14	Cohort 2 n=14	Cohort 3 n=18	Overall N=46
Age, years				
Mean (SD)	32.4 (7.61)	36.0 (6.19)	36.7 (8.10)	35.2 (7.48)
Range	24–47	28–46	23–49	23–49
Gender, n (%)				
Male	8 (57.1)	9 (64.3)	13 (72.2)	30 (65.2)
Female	6 (42.9)	5 (35.7)	5 (27.8)	16 (34.8)
Race, n (%)				
White	7 (50.0)	8 (57.1)	11 (61.1)	26 (56.5)
Black/African American	6 (42.9)	6 (42.9)	7 (38.9)	19 (41.3)
Asian	1 (7.1)	0	0	1 (2.2)
Ethnicity, n (%)				
Hispanic/Latino	4 (28.6)	6 (42.9)	6 (33.3)	16 (34.8)
Not Hispanic/Latino	10 (71.4)	8 (57.1)	12 (66.7)	30 (65.2)
Weight, kg				
Mean (SD)	73.11 (12.986)	77.44 (16.260)	78.48 (10.532)	76.53 (13.131)
Range	58.0–99.0	53.4–109.0	59.0–-94.3	53.4–109.0
Body mass index, kg/m^2				
Mean (SD)	26.27 (2.751)	26.85 (3.430)	26.56 (2.640)	26.56 (2.876)
Range	20.9–29.3	21.8–31.6	22.6–30.7	20.9–31.6

SD, standard deviation.

Effect of DRV/r on PK of BMS-626529

- Cohort 1
 - Compared with BMS-663068 alone, coadministration with DRV/r increased BMS-626529 C_{max}, AUC_{tau} and C_{12} by 52%, 63% and 88%, respectively (Table 2a & Figure 2a).

Table 2a: PK results and statistical analysis: BMS-626529
Figure 2a: Mean (+/-SD) plasma concentration–time profiles for the effect of DRV/r on BMS-626529

Cohort	Treatment and comparison	C_{max}, ng/mL (90% CI)	AUC_{tau}, ng.h/mL (90% CI)	C_{12}, ng/mL (90% CI)
		PK parameter, adjusted geometric means		
Cohort 1	BMS-663068 (n=14)	2033 (1619, 2553)	12632 (10142, 15735)	308 (213, 444)
Cohort 1	BMS-663068 + DRV/r (n=12)	3098 (2386, 4022)	20633 (16333, 26065)	577 (455, 732)
		GMR		
Cohort 1	BMS-663068 + DRV/r versus BMS-663068	1.524 (1.279, 1.815)	1.633 (1.423, 1.875)	1.875 (1.093, 3.216)

Cohort 1

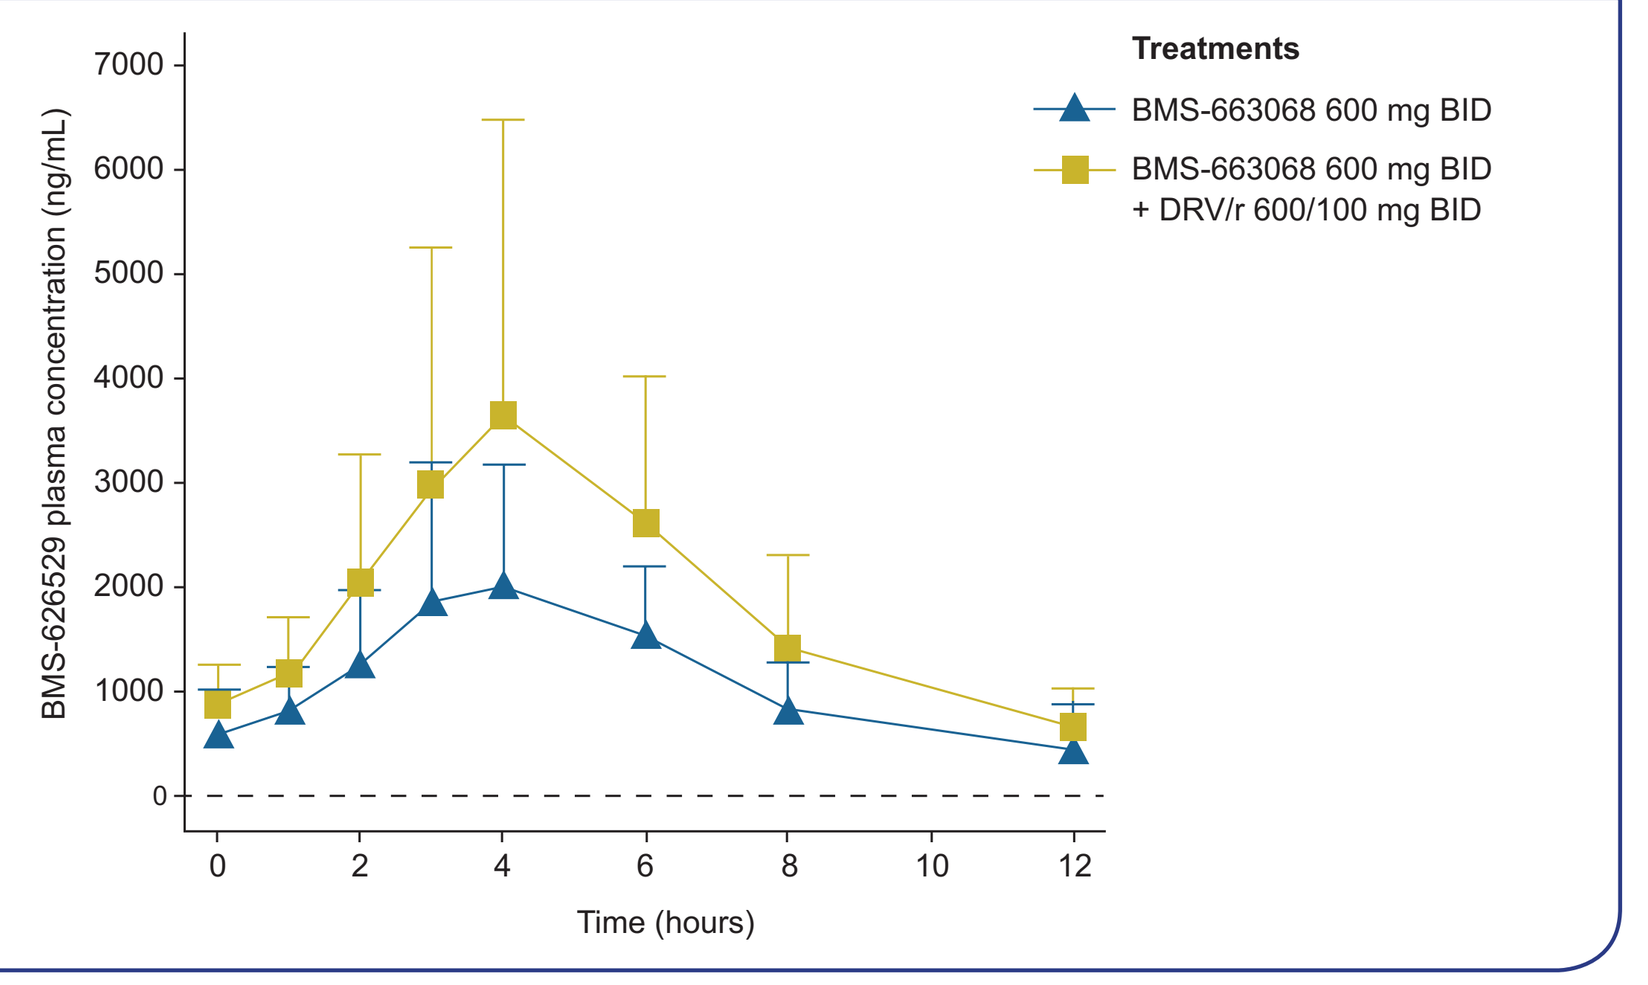
BMS-626529 plasma concentration (ng/mL)
Time (hours)
Treatments
BMS-663068 600 mg BID
BMS-663068 600 mg BID + DRV/r 600/100 mg BID

CI, confidence interval; DRV/r, ritonavir-boosted darunavir; GMR, geometric mean ratio; PK, pharmacokinetic; SD, standard deviation.

Effect of ETR on PK of BMS-626529

- Cohort 2
 - Compared with BMS-663068 alone, coadministration with ETR reduced BMS-626529 C_{max}, AUC_{tau} and C_{12} by 48%, 50% and 52%, respectively (Table 2b, Figure 2b).

Table 2b: PK results and statistical analysis: BMS-626529
Figure 2b: Mean (+/-SD) plasma concentration–time profiles for the effect of ETR on BMS-626529

Cohort	Treatment and comparison	C_{max}, ng/mL (90% CI)	AUC_{tau}, ng.h/mL (90% CI)	C_{12}, ng/mL (90% CI)
		PK parameter, adjusted geometric means		
Cohort 2	BMS-663068 (n=14)	1941 (1699, 2219)	13364 (11574, 15430)	479 (310, 740)
Cohort 2	BMS-663068 + ETR (n=14)	1003 (858, 1171)	6714 (5815, 7753)	231 (156, 344)
		GMR		
Cohort 2	BMS-663068 + ETR versus BMS-663068	0.516 (0.454, 0.587)	0.502 (0.442, 0.571)	0.483 (0.324, 0.720)

Cohort 2

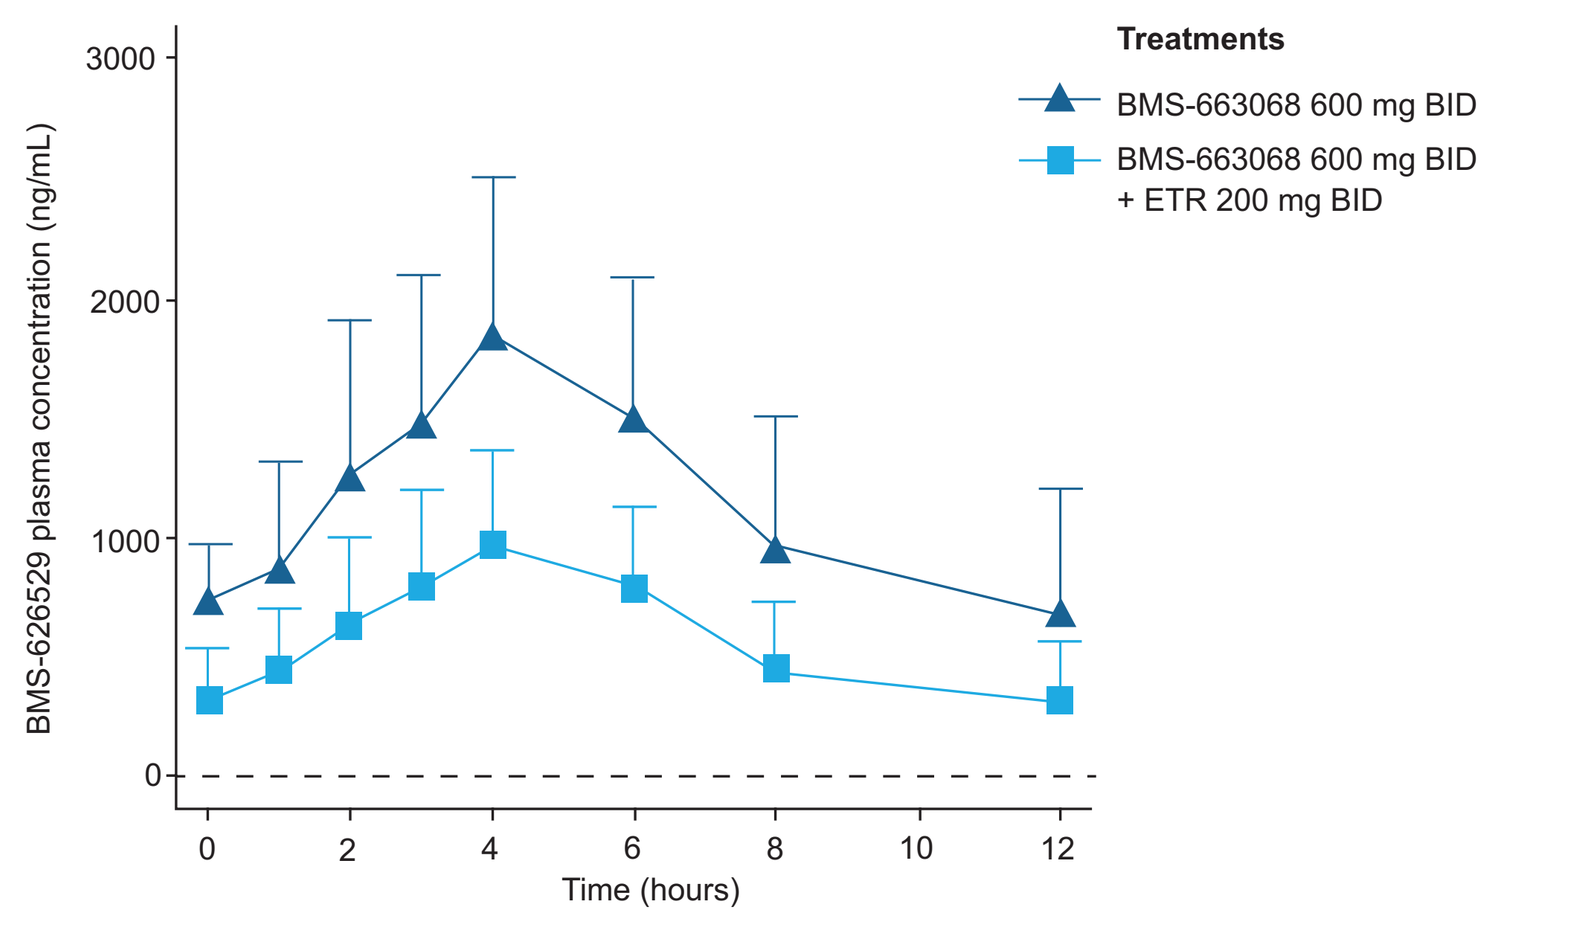
BMS-626529 plasma concentration (ng/mL)
Time (hours)
Treatments
BMS-663068 600 mg BID
BMS-663068 600 mg BID + ETR 200 mg BID

CI, confidence interval; DRV/r, ritonavir-boosted darunavir; ETR, etravirine; GMR, geometric mean ratio; PK, pharmacokinetic; SD, standard deviation.

Effect of DRV/r and ETR on PK of BMS-626529

- Cohort 3
 - Compared with BMS-663068 alone, coadministration with DRV/r and ETR increased BMS-626529 C_{max}, AUC_{tau}, and C_{12} by 53%, 34% and 33%, respectively (Table 2c, Figure 2c).

Table 2c: PK results and statistical analysis: BMS-626529
Figure 2c: Mean (+/-SD) plasma concentration–time profiles for the effect of DRV/r + ETR on BMS-626529

Cohort	Treatment and comparison	C_{max}, ng/mL (90% CI)	AUC_{tau}, ng.h/mL (90% CI)	C_{12}, ng/mL (90% CI)
		PK parameter, adjusted geometric means		
Cohort 3	BMS-663068 (n=18)	1568 (1362, 1805)	10339 (9136, 11700)	312 (221, 441)
Cohort 3	BMS-663068 + DRV/r + ETR (n=13)	2398 (2144, 2682)	13861 (11659, 16478)	416 (286, 603)
		GMR		
Cohort 3	BMS-663068 + DRV/r + ETR versus BMS-663068	1.529 (1.323, 1.768)	1.341 (1.172, 1.534)	1.332 (0.980, 1.809)

Cohort 3

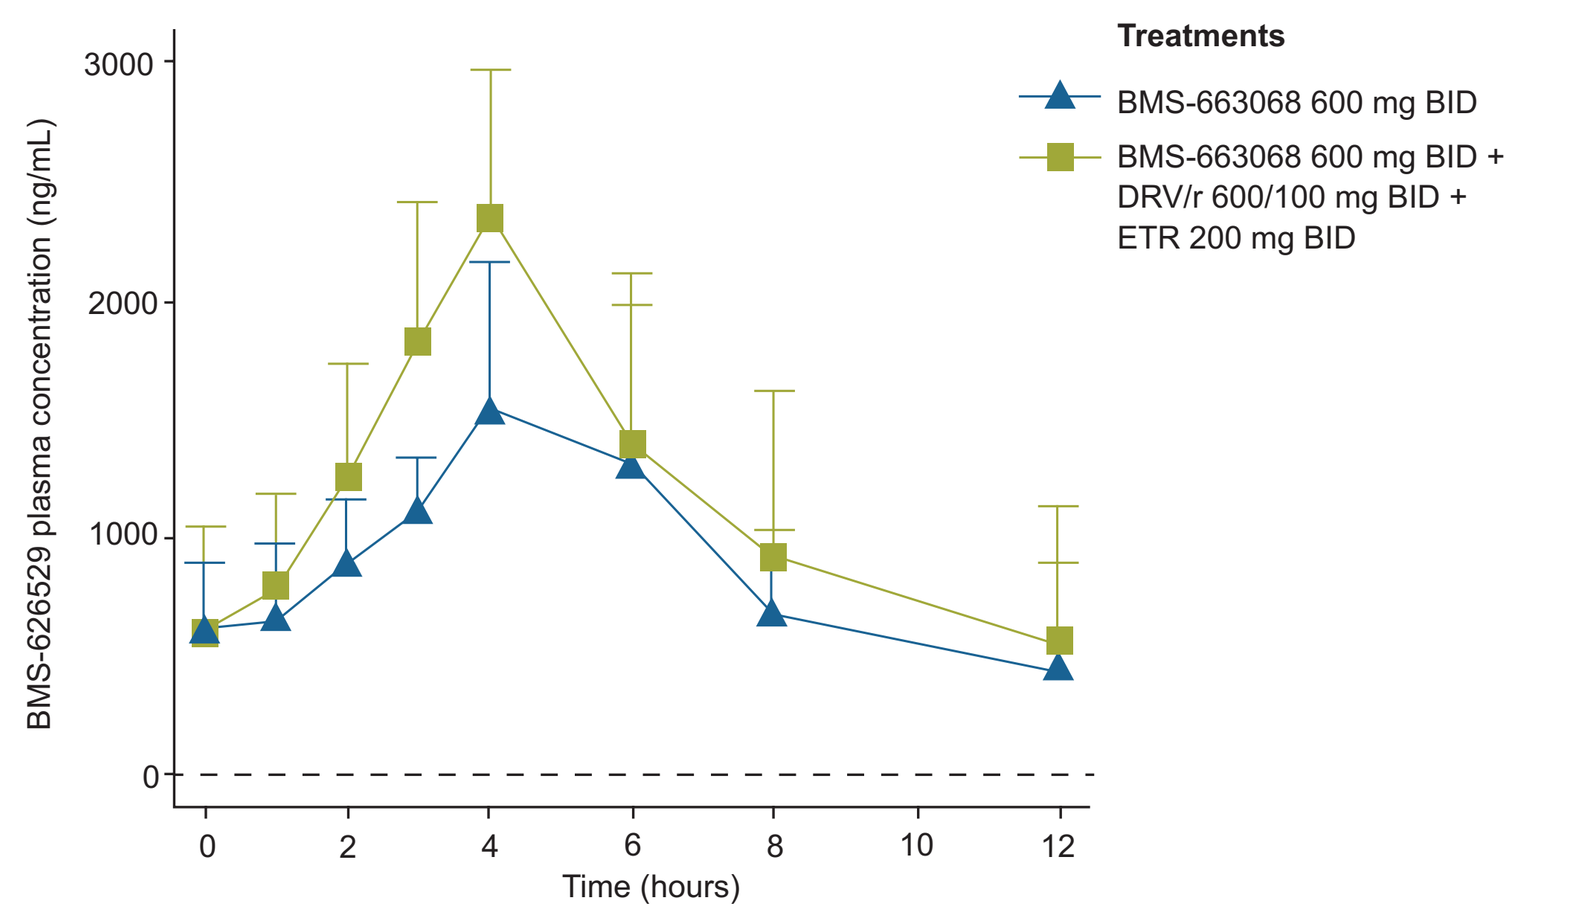
BMS-626529 plasma concentration (ng/mL)
Time (hours)
Treatments
BMS-663068 600 mg BID
BMS-663068 600 mg BID + DRV/r 600/100 mg BID + ETR 200 mg BID

CI, confidence interval; DRV/r, ritonavir-boosted darunavir; ETR, etravirine; GMR, geometric mean ratio; PK, pharmacokinetic; SD, standard deviation.

Effect of BMS-663068 coadministration on PK of DRV and ETR

- Cohort 1
 - Coadministration of BMS-663068 with DRV/r resulted in 2%, 6% and 5% decreases for C_{max}, AUC_{tau} and C_{12}, respectively for DRV (Table 3).
 - GMRs comparing BMS-663068 + DRV/r versus DRV were contained within the predefined 90% CI limits for DRV of 0.75–1.70.
- Cohort 3
 - Coadministration of BMS-663068 with DRV/r + ETR resulted in 5%, 6% and 12% decreases, respectively, for C_{max}, AUC_{tau} and C_{12} for DRV/r + ETR (Table 3).
 - GMRs comparing BMS-663068 + DRV/r + ETR versus DRV/r + ETR were contained within the predefined 90% CI limits for DRV and ETR of 0.75–1.70.

Table 3: PK results and statistical analysis: DRV

Cohort	Treatment and comparison	C_{max}, ng/mL (90% CI)	AUC_{tau}, ng.h/mL (90% CI)	C_{12}, ng/mL (90% CI)
		PK parameter, adjusted geometric means		
Cohort 1	DRV/r (n=13)	8785 (8181, 9434)	68380 (63785, 73306)	3633 (3255, 4056)
Cohort 1	BMS-663068 + DRV/r (n=12)	8636 (7983, 9342)	64543 (58485, 71228)	3445 (2956, 4016)
		GMR		
Cohort 1	BMS-663068 + DRV/r versus DRV/r	0.983 (0.931, 1.038)	0.944 (0.894, 0.996)	0.948 (0.865, 1.040)
		PK parameter, adjusted geometric means		
Cohort 3	DRV/r + ETR (n=13)	10105 (9524, 10721)	77739 (72510, 83344)	3672 (3171, 4252)
Cohort 3	BMS-663068 + DRV/r + ETR (n=13)	9636 (8920, 10409)	72907 (66302, 80171)	3234 (2628, 3979)
		GMR		
Cohort 3	DRV/r + ETR versus BMS-663068 + DRV/r + ETR	0.954 (0.903, 1.007)	0.938 (0.888, 0.991)	0.881 (0.769, 1.009)

CI, confidence interval; DRV/r, ritonavir-boosted darunavir, ETR, etravirine; GMR, geometric mean ratio; PK, pharmacokinetic.

Effect of BMS-663068 on PK of RTV

- Cohort 1
 - Coadministration of BMS-663068 with DRV/r resulted in no change for C_{max} and 15% and 19% increases for AUC_{tau} and C_{12}, respectively for RTV (Table 4).
- Cohort 3
 - Coadministration of BMS-663068 + DRV/r + ETR resulted in increases of 14%, 9% and 7%, for C_{max}, AUC_{tau} and C_{12}, respectively for RTV (Table 4).

Table 4: PK results and statistical analysis: RTV

Cohort	Treatment and comparison	C_{max}, ng/mL (90% CI)	AUC_{tau}, ng.h/mL (90% CI)	C_{12}, ng/mL (90% CI)
		PK parameter, adjusted geometric means		
Cohort 1	DRV/r (n=13)	1324 (1123, 1560)	7191 (6082, 8501)	229 (182, 288)
Cohort 1	BMS-663068 + DRV/r (n=12)	1317 (1050, 1651)	8256 (6457, 10557)	273 (204, 366)
		GMR		
Cohort 1	BMS-663068 + DRV/r versus DRV/r	0.995 (0.856, 1.156)	1.148 (0.992, 1.329)	1.192 (1.056, 1.347)
		PK parameter, adjusted geometric means		
Cohort 3	DRV/r + ETR (n=13)	862 (721, 1031)	4928 (4323, 5617)	157 (139, 178)
Cohort 3	BMS-663068 + DRV/r + ETR (n=13)	982 (828, 1165)	5380 (4912, 5892)	168 (148, 191)
		GMR		
Cohort 3	BMS-663068 + DRV/r + ETR versus DRV/r + ETR	1.139 (0.960, 1.352)	1.092 (0.979, 1.218)	1.067 (0.972, 1.171)

CI, confidence interval; DRV/r, ritonavir-boosted darunavir; ETR, etravirine; GMR, geometric mean ratio; PK, pharmacokinetic; RTV, ritonavir.

Effect of BMS-663068 coadministration on PK of ETR

- Cohort 2
 - Compared with ETR alone, coadministration with BMS-663068 resulted in a small increase in ETR C_{max}, AUC_{tau} and C_{12} by 11%, 11% and 14%, respectively.
 - GMRs comparing BMS-663068 + ETR versus ETR were contained within the predefined 90% CI limits of 0.75–1.70 (Table 5).
- Cohort 3
 - Compared with DRV/r + ETR, coadministration with BMS-663068 increased ETR C_{max}, AUC_{tau} and C_{12} by 18%, 28% and 28%, respectively.
 - GMRs comparing BMS-663068 + DRV/r + ETR versus DRV/r + ETR were contained within the predefined 90% CI limits of 0.75–1.70 (Table 5).

Table 5: PK results and statistical analysis: ETR

Cohort	Treatment and comparison	C_{max}, ng/mL (90% CI)	AUC_{tau}, ng.h/mL (90% CI)	C_{12}, ng/mL (90% CI)
		PK parameter, adjusted geometric means		
Cohort 2	ETR (n=14)	846 (739, 968)	7825 (6829, 8965)	457 (392, 534)
Cohort 2	BMS-663068 + ETR (n=14)	943 (811, 1096)	8670 (7542, 9968)	522 (448, 609)
		GMR		
Cohort 2	BMS-663068 + ETR versus ETR	1.114 (1.039, 1.194)	1.108 (1.053, 1.166)	1.142 (1.079, 1.208)
		PK parameter, adjusted geometric means		
Cohort 3	DRV/r + ETR (n=13)	838 (672, 1044)	7102 (5577, 9045)	417 (306, 570)
Cohort 3	BMS-663068 + DRV/r + ETR (n=13)	989 (767, 1274)	9071 (6790, 12118)	533 (373, 762)
		GMR		
Cohort 3	BMS-663068 + DRV/r + ETR versus DRV/r + ETR	1.180 (1.098, 1.267)	1.277 (1.198, 1.361)	1.278 (1.175, 1.389)

CI, confidence interval; DRV/r, ritonavir-boosted darunavir; ETR, etravirine; GMR, geometric mean ratio; PK, pharmacokinetic.

Safety

- A summary of AEs experienced by ≥2 subjects is shown in Table 6.
- Of the 46 subjects randomized to receive treatment, seven did not complete the study because of AEs:
 - two in Cohort 1
 - five in Cohort 3
 - in six of seven cases, etiology was most likely related to DRV/r and/or ETR based on timing of onset after BMS-663068 washout.
- The reasons for discontinuation were mild or moderate skin rash (n=5), increased alanine amino transaminase (3–4 times the upper limit of normal; n=1) and back pain (n=1).
- A total of 22 (48%) subjects reported AEs, of which skin rash was the most frequent (28%).
- There were no deaths, serious AEs or AEs categorized as severe.

Table 6: Summary of AEs experienced by ≥2 subjects

n (%)	A (N=46)	B (N=14)	A + B (N=13)	C (N=14)	A + C (N=14)	B + C (N=18)	A + B + C (N=13)	Total (N=46)
	Treatments received							
Subjects with AE	6 (13.0)	4 (28.6)	3 (23.1)	4 (28.6)	2 (14.3)	9 (50.%)	4 (30.8)	22 (47.8)
Drug eruption	0	3 (21.4)	0	2 (14.3)	0	7 (38.9)	1 (7.7)	13 (28.3)
Headache	1 (2.2)	1 (7.1)	1 (7.7)	0	0	3 (16.7)	1 (7.7)	6 (13.0)
Pruritus	1 (2.2)	1 (7.1)	1 (7.7)	0	0	0	3 (23.1)	6 (13.0)
Pruritus generalized	0	1 (7.1)	0	1 (7.1)	0	3 (16.7)	0	5 (10.9)
Back pain	0	1 (7.1)	0	0	0	2 (11.1)	0	3 (6.5)
Nausea	1 (2.2)	1 (7.1)	0	0	0	1 (5.6)	0	3 (6.5)
Abdominal distension	0	0	0	0	0	1 (5.6)	1 (7.7)	2 (4.3)
Aphthous stomatitis	0	0	0	0	0	1 (5.6)	1 (7.7)	2 (4.3)
Diarrhea	0	0	0	0	0	2 (11.1)	1 (7.7)	2 (4.3)
Dizziness	0	0	0	0	0	2 (11.1)	0	2 (4.3)
Dry mouth	1 (2.2)	0	0	0	0	1 (5.6)	0	2 (4.3)
Fatigue	0	1 (7.1)	0	0	0	1 (5.6)	0	2 (4.3)
Muscle spasms	0	0	0	0	0	2 (11.1)	0	2 (4.3)
Somnolence	0	0	0	0	0	2 (11.1)	0	2 (4.3)

Cohort 1 = A, B, A + B; Cohort 2 = A, C, A + C; Cohort 3 = A, B + C, A + B + C.
A = BMS-663068 600 mg BID; B = DRV/r 600/100 mg BID; C = ETR 200 mg BID.
AE, adverse event; DRV/r, ritonavir-boosted darunavir; ETR, etravirine.

CONCLUSIONS

- BMS-663068 and the active moiety BMS-626529 had no clinically relevant effect on the PK of DRV or ETR as 90% CIs were contained within the predefined limits of 0.75–1.70.
- Coadministration of DRV/r with BMS-663068 increased peak plasma concentration and systemic exposure of BMS-626529.
 - Increased exposures were not associated with evidence of an increased incidence of AEs or other safety signals that could be attributed to BMS-626529.
- Coadministration of BMS-663068 with ETR without a protease inhibitor decreased peak plasma concentration and systemic exposure of BMS-626529.
 - This decrease in exposure would not be expected to affect the efficacy based on the wide range of efficacy noted in AI438011 (with BMS-663068 doses of 400 and 800 mg BID, 600 mg and 1200 mg QD).
 - An increase in BMS-626529 exposure was noted when coadministered with DRV/r and ETR.
- In this study, coadministration of BMS-663068 with DRV/r + ETR was generally safe in healthy subjects; there were no deaths or serious AEs. Skin rash was consistent with ETR and DRV/r administration.
- This study shows that DRV/r alone, ETR alone or DRV/r + ETR may be coadministered with BMS-663068 without dose adjustment.

- Two accompanying posters are presented:
 - HIV-1 Attachment Inhibitor Prodrug BMS-663068: Pharmacokinetics, Pharmacodynamics and Model-Based Dose Selection (*Poster 509, Session P-H1, Tuesday, February 24th 2:30–4:00 pm PST*)
 - Attachment Inhibitor Prodrug BMS-663068 in Antiretroviral-Experienced Subjects: Week 48 Analysis (*Poster 545, Session P-J1, Tuesday, February 24th 2:30–4:00 pm PST*).

ACKNOWLEDGMENTS

- We would like to thank all of the AI438020 clinical trial participants and their families.
- Professional medical writing and editorial assistance was provided by Helen Brown at MediTech Media and was funded by Bristol-Myers Squibb.

REFERENCES

1. Imaz A *et al. AIDS Rev* 2011; 13: 180–193.
2. Tozzi V *et al. Antivir Ther* 2006; 11: 553–560.
3. Langley DR *et al. Proteins* 2015; 83: 331–350.
4. Brown J *et al. J Pharm Sci* 2013; 102: 1742–1751.
5. Nowicka-Sans B *et al. Antimicrob Agents Chemother* 2012; 56: 3498–3507.
6. Ray N *et al. JAIDS* 2013; 64: 7–15.
7. Zhou N *et al. JAC* 2014; 69: 573–581.
8. Li Z *et al. AAC* 2013; 57: 4172–4180.
9. Lalezari J *et al.* CROI 2014: Oral Abstract 86.
10. Data on File. AI438009 CSR. BMS, Princeton, NJ.

CROI 2015
23–26 February 2015, Seattle, WA, USA