A Phase I Clinical Trial of Autologous CD4+ T cells Modified with a Retroviral Vector Expressing the MazF Endoribonuclease in Patients with HIV-1

Jeffrey M. Jacobson, Hideto Chono, Meghan Metz, Gabriela Plesa, Julie Jadlowsky, Simon Lacey, Bruce Levine, Hirofumi Yoshioka, Tomohide Kidokoro, Daisuke Tomura, Junichi Mineno, Kazutoh Takesako, and Carl H. June

[1] Drexel University College of Medicine, Philadelphia, PA, USA, [2] University of Pennsylvania Translational Research Program, Philadelphia, PA, USA [3]Takara Bio Inc., Otsu, Shiga, Japan

#402

Abstract

Background: The *E.coli*-derived MazF endoribonuclease specifically cleaves single stranded RNAs at ACA sequences. HIV-1 contains over 240 ACA sequences, making it especially sensitive to MazF activity. In this study, autologous CD4⁺ T cells are modified using a retroviral vector containing the *mazF* gene expressed under the control of the HIV LTR. MazF expression is thus activated by Tat conditionally during HIV replication. This study is designed to evaluate the safety and durability of MazF-modified CD4⁺ T (MazF-T) cells, and assess related antiviral effects.

Methodology: This is an exploratory Phase I, Open Label, Dual Cohort study evaluating safety, tolerability and immunogenicity of autologous CD4⁺ T cells expressing the MazF endoribonuclease gene in HIV+ subjects. Both cohorts consist of subjects on combination antiretroviral therapy (cART) with undetectable HIV-1 RNA levels and with CD4 counts >350 cells/mm^3 in Cohort 1 and >450 cells/mm^3 in Cohort 2. Subjects in cohort 1 remain on cART throughout the duration of the study. Subjects in cohort 2 participate in a 16 week analytical treatment interruption (ATI) beginning 2 weeks post T cell infusion. All subjects are infused with a single dose of MazF-T cells and are evaluated for persistence of modified cells, impact on CD4 count and effects on HIV viral load.

Results: To date, all 6 subjects in Cohort 1 have each received a single infusion of 0.5-1x10^{10} cells. All 10 AE related to study drug have been grade 1 in severity. Based on available data, 3 of 6 subjects had an increase in CD4:CD8 ratio post-infusion before stabilizing near baseline. Absolute CD4 counts remained stable, or slightly increased compared to baseline. As all Cohort 1 subjects remained on cART, all viral loads remained undetectable. MazF DNA signal was detected by qPCR in the peripheral blood of all subjects evaluated to date at the most currently available timepoint.

Conclusions: These preliminary results suggest that autologous MazF-modified CD4⁺ T cells are safe and well-tolerated in aviremic HIV+ subjects and are able to persist out to 6 months post-infusion. Future results in subjects participating in an ATI (Cohort 2) will further elucidate the anti-HIV effects associated with MazF-T cells in the presence of viremia.

Study Design

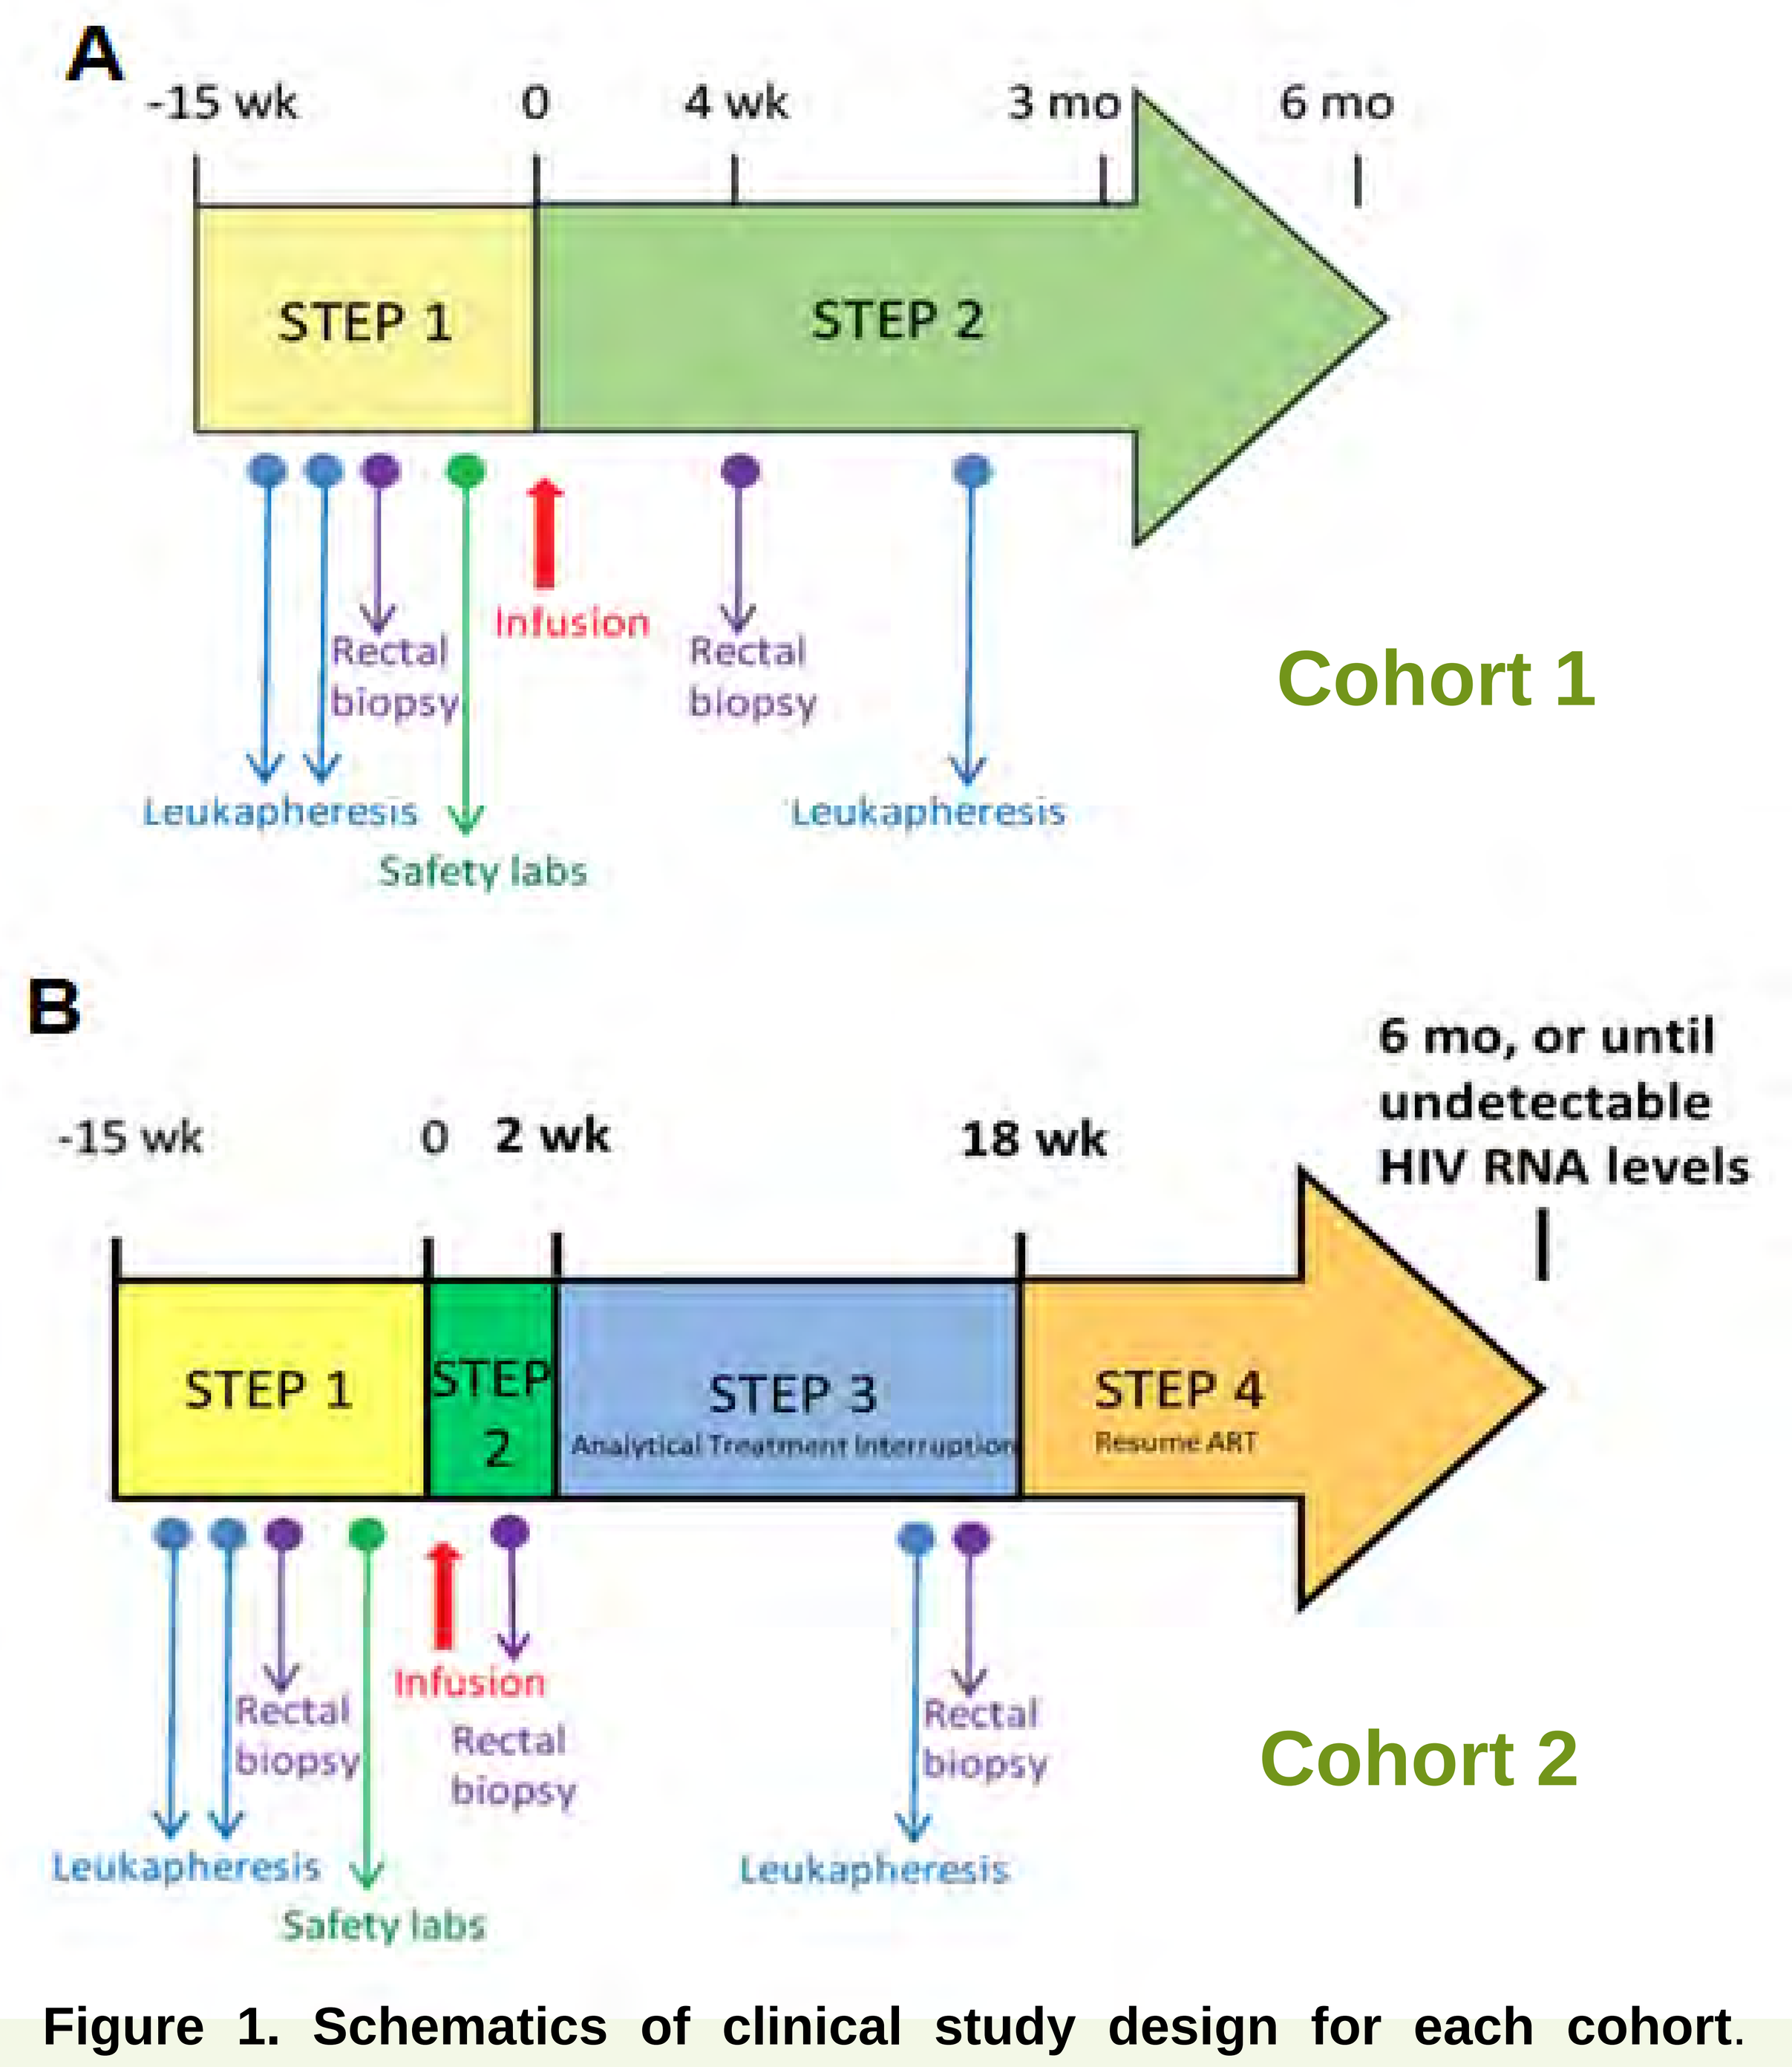

A
-15 wk
0
4 wk
3 mo
6 mo
STEP 1
STEP 2
Rectal biopsy
Infusion
Rectal biopsy
Leukapheresis
Safety labs
Leukapheresis
Cohort 1
B
-15 wk
0 2 wk
18 wk
6 mo, or until undetectable HIV RNA levels
STEP 1
STEP 2
STEP 3
Analytical Treatment Interruption
STEP 4
Resume ART
Rectal biopsy
Infusion
Rectal biopsy
Leukapheresis
Safety labs
Rectal biopsy
Leukapheresis
Cohort 2

Figure 1. Schematics of clinical study design for each cohort. Subjects in Cohort 1 remain on cART throughout the study. Two weeks after infusion of MazF-modified cells, subjects in Cohort 2 participate in an analytical treatment interruption lasting a maximum of 16 weeks.

Results

Safety- Cohort 1

- To date, all Cohort 1 subjects (N=6) have been infused. Each subject received a single infusion of 0.5-1x10^{10} MazF T cells.
- There has been 1 SAE, probably related to MazF-T. This event was a self-limited cytokine release syndrome which resolved within 48 hours.
- All 10 AEs related to study drug have been grade 1 in severity.
- Since subjects were on continuous cART, viral loads remained undetectable throughout the study, to date.

Results

Absolute CD4+ T cell counts- Cohort 1

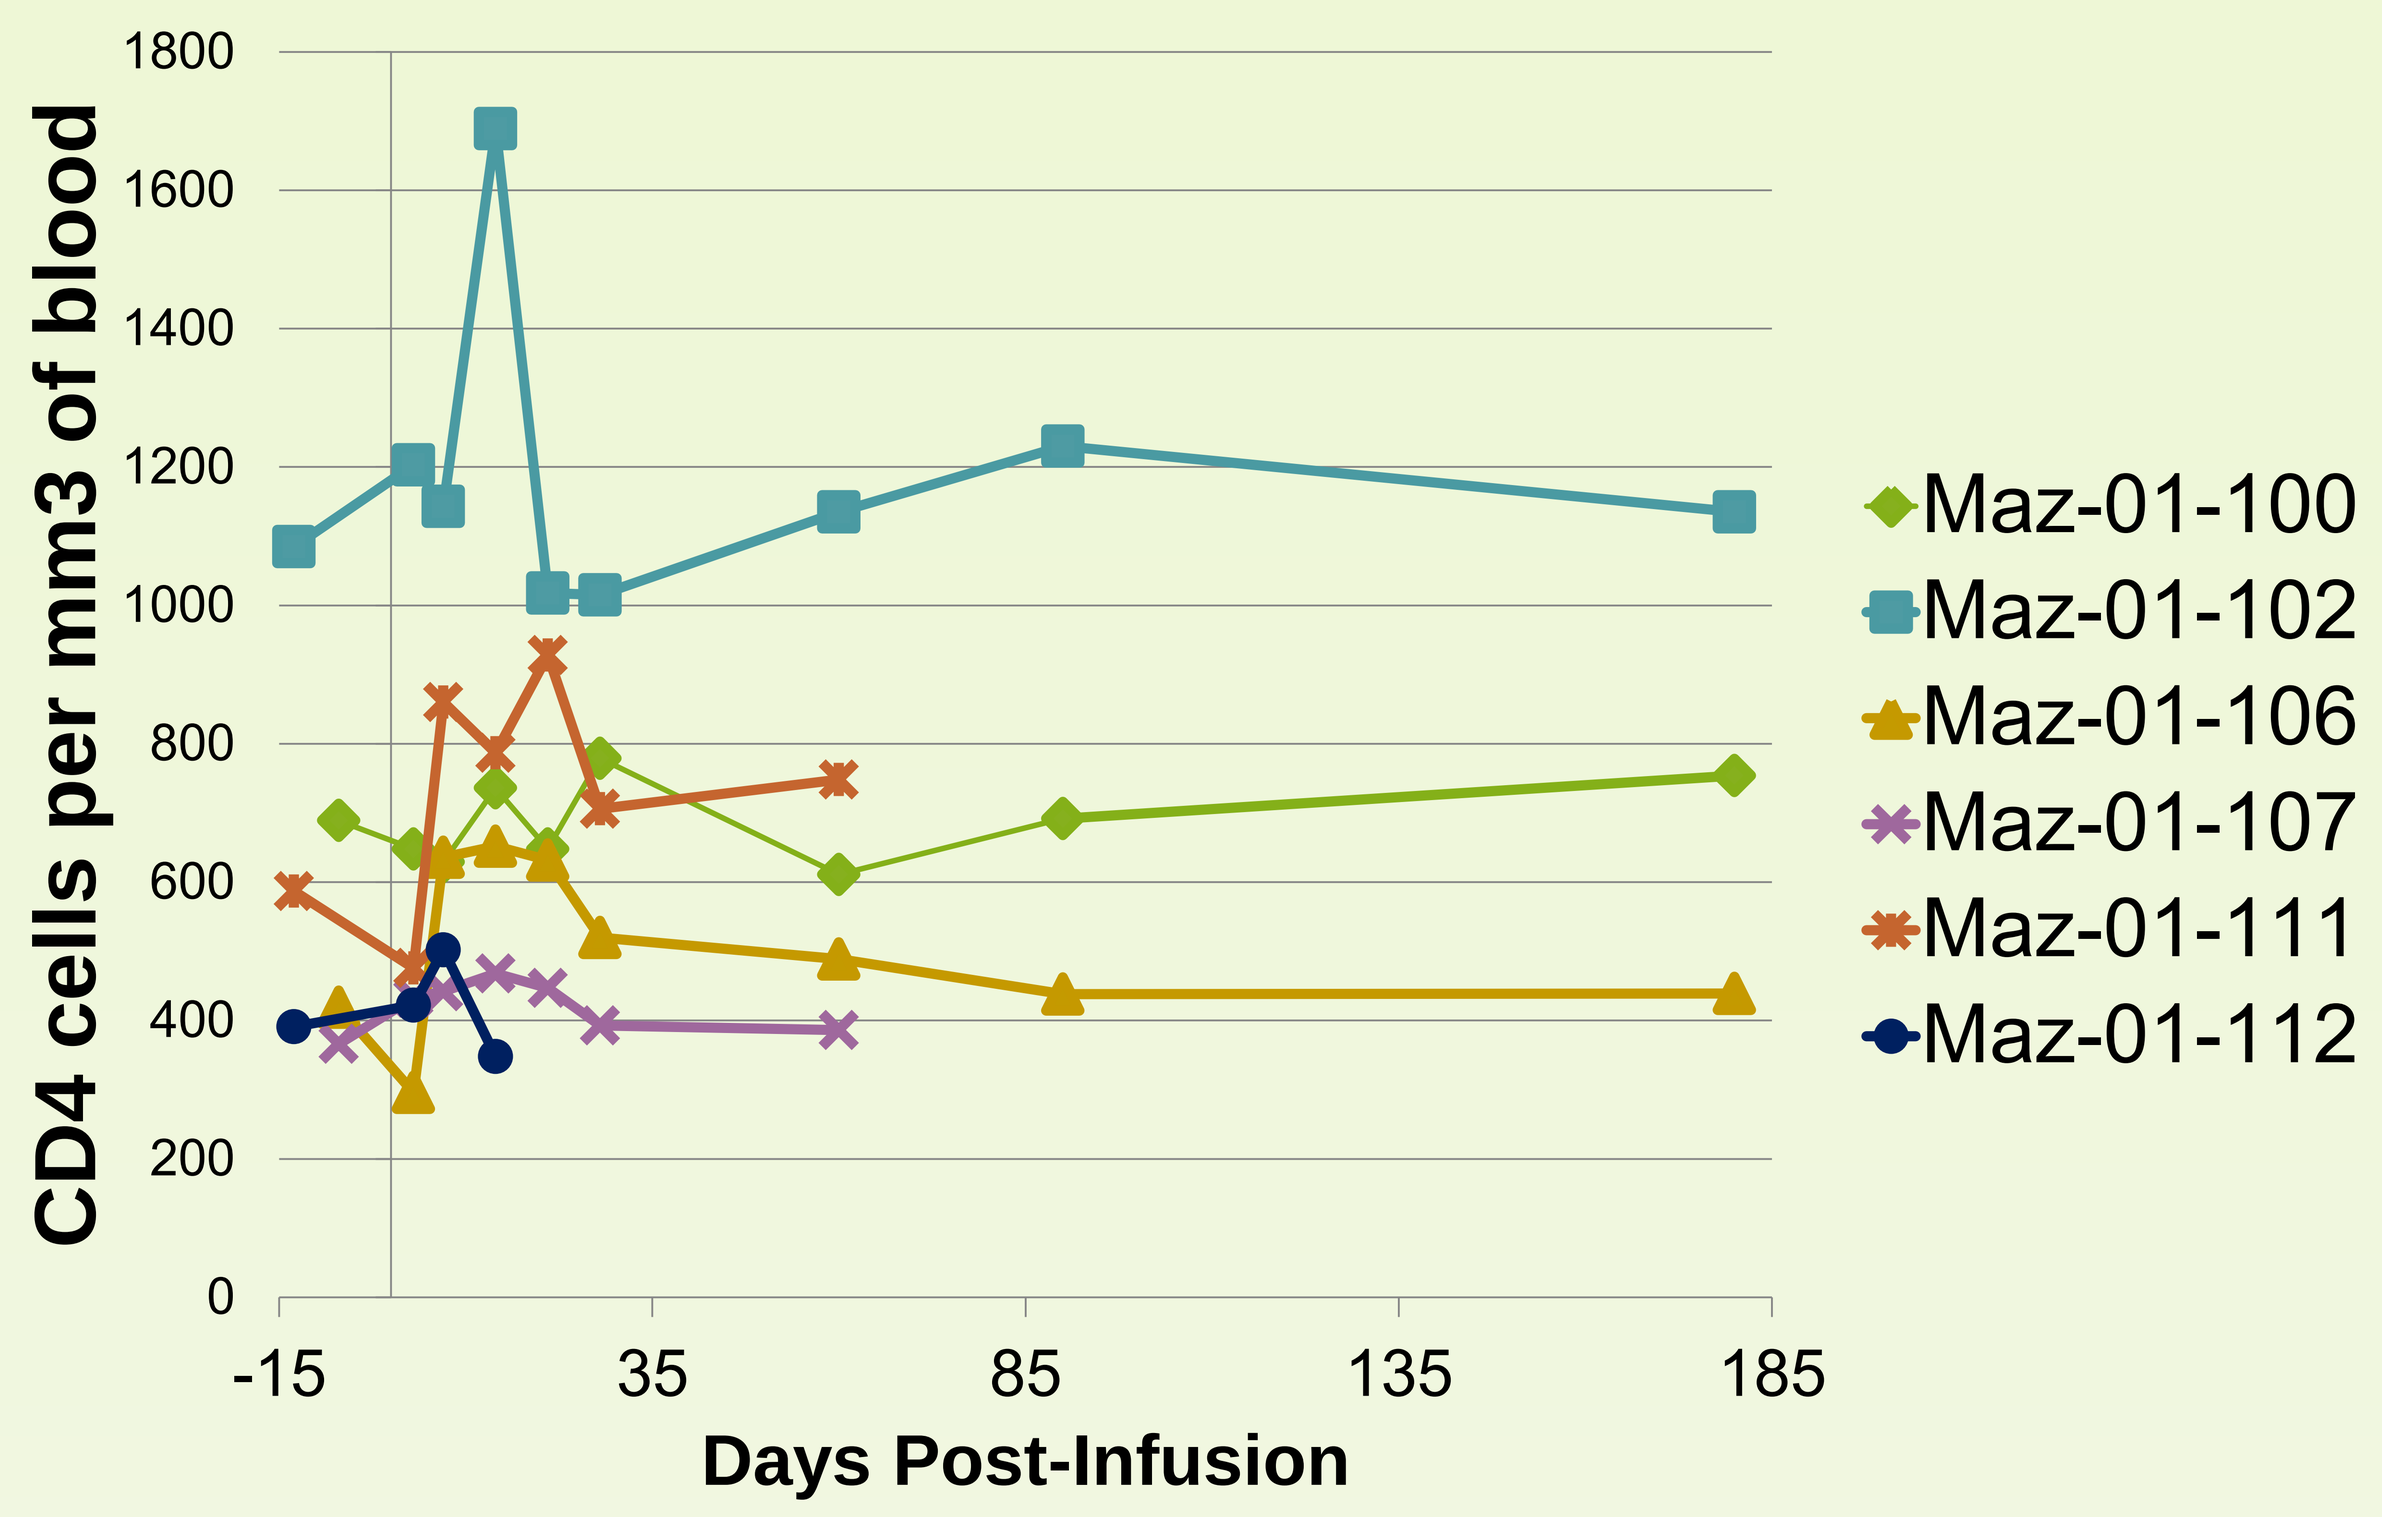

CD4 cells per mm3 of blood
1800
1600
1400
1200
1000
800
600
400
200
0
-15
35
85
135
185
Days Post-Infusion
Maz-01-100
Maz-01-102
Maz-01-106
Maz-01-107
Maz-01-111
Maz-01-112

Figure 2. Absolute CD4+ T cell count. The absolute CD4 count remained stable, or slightly increased to baseline CD4 counts in Cohort 1 subjects.

CD4:CD8 T cell Ratio-Cohort 1

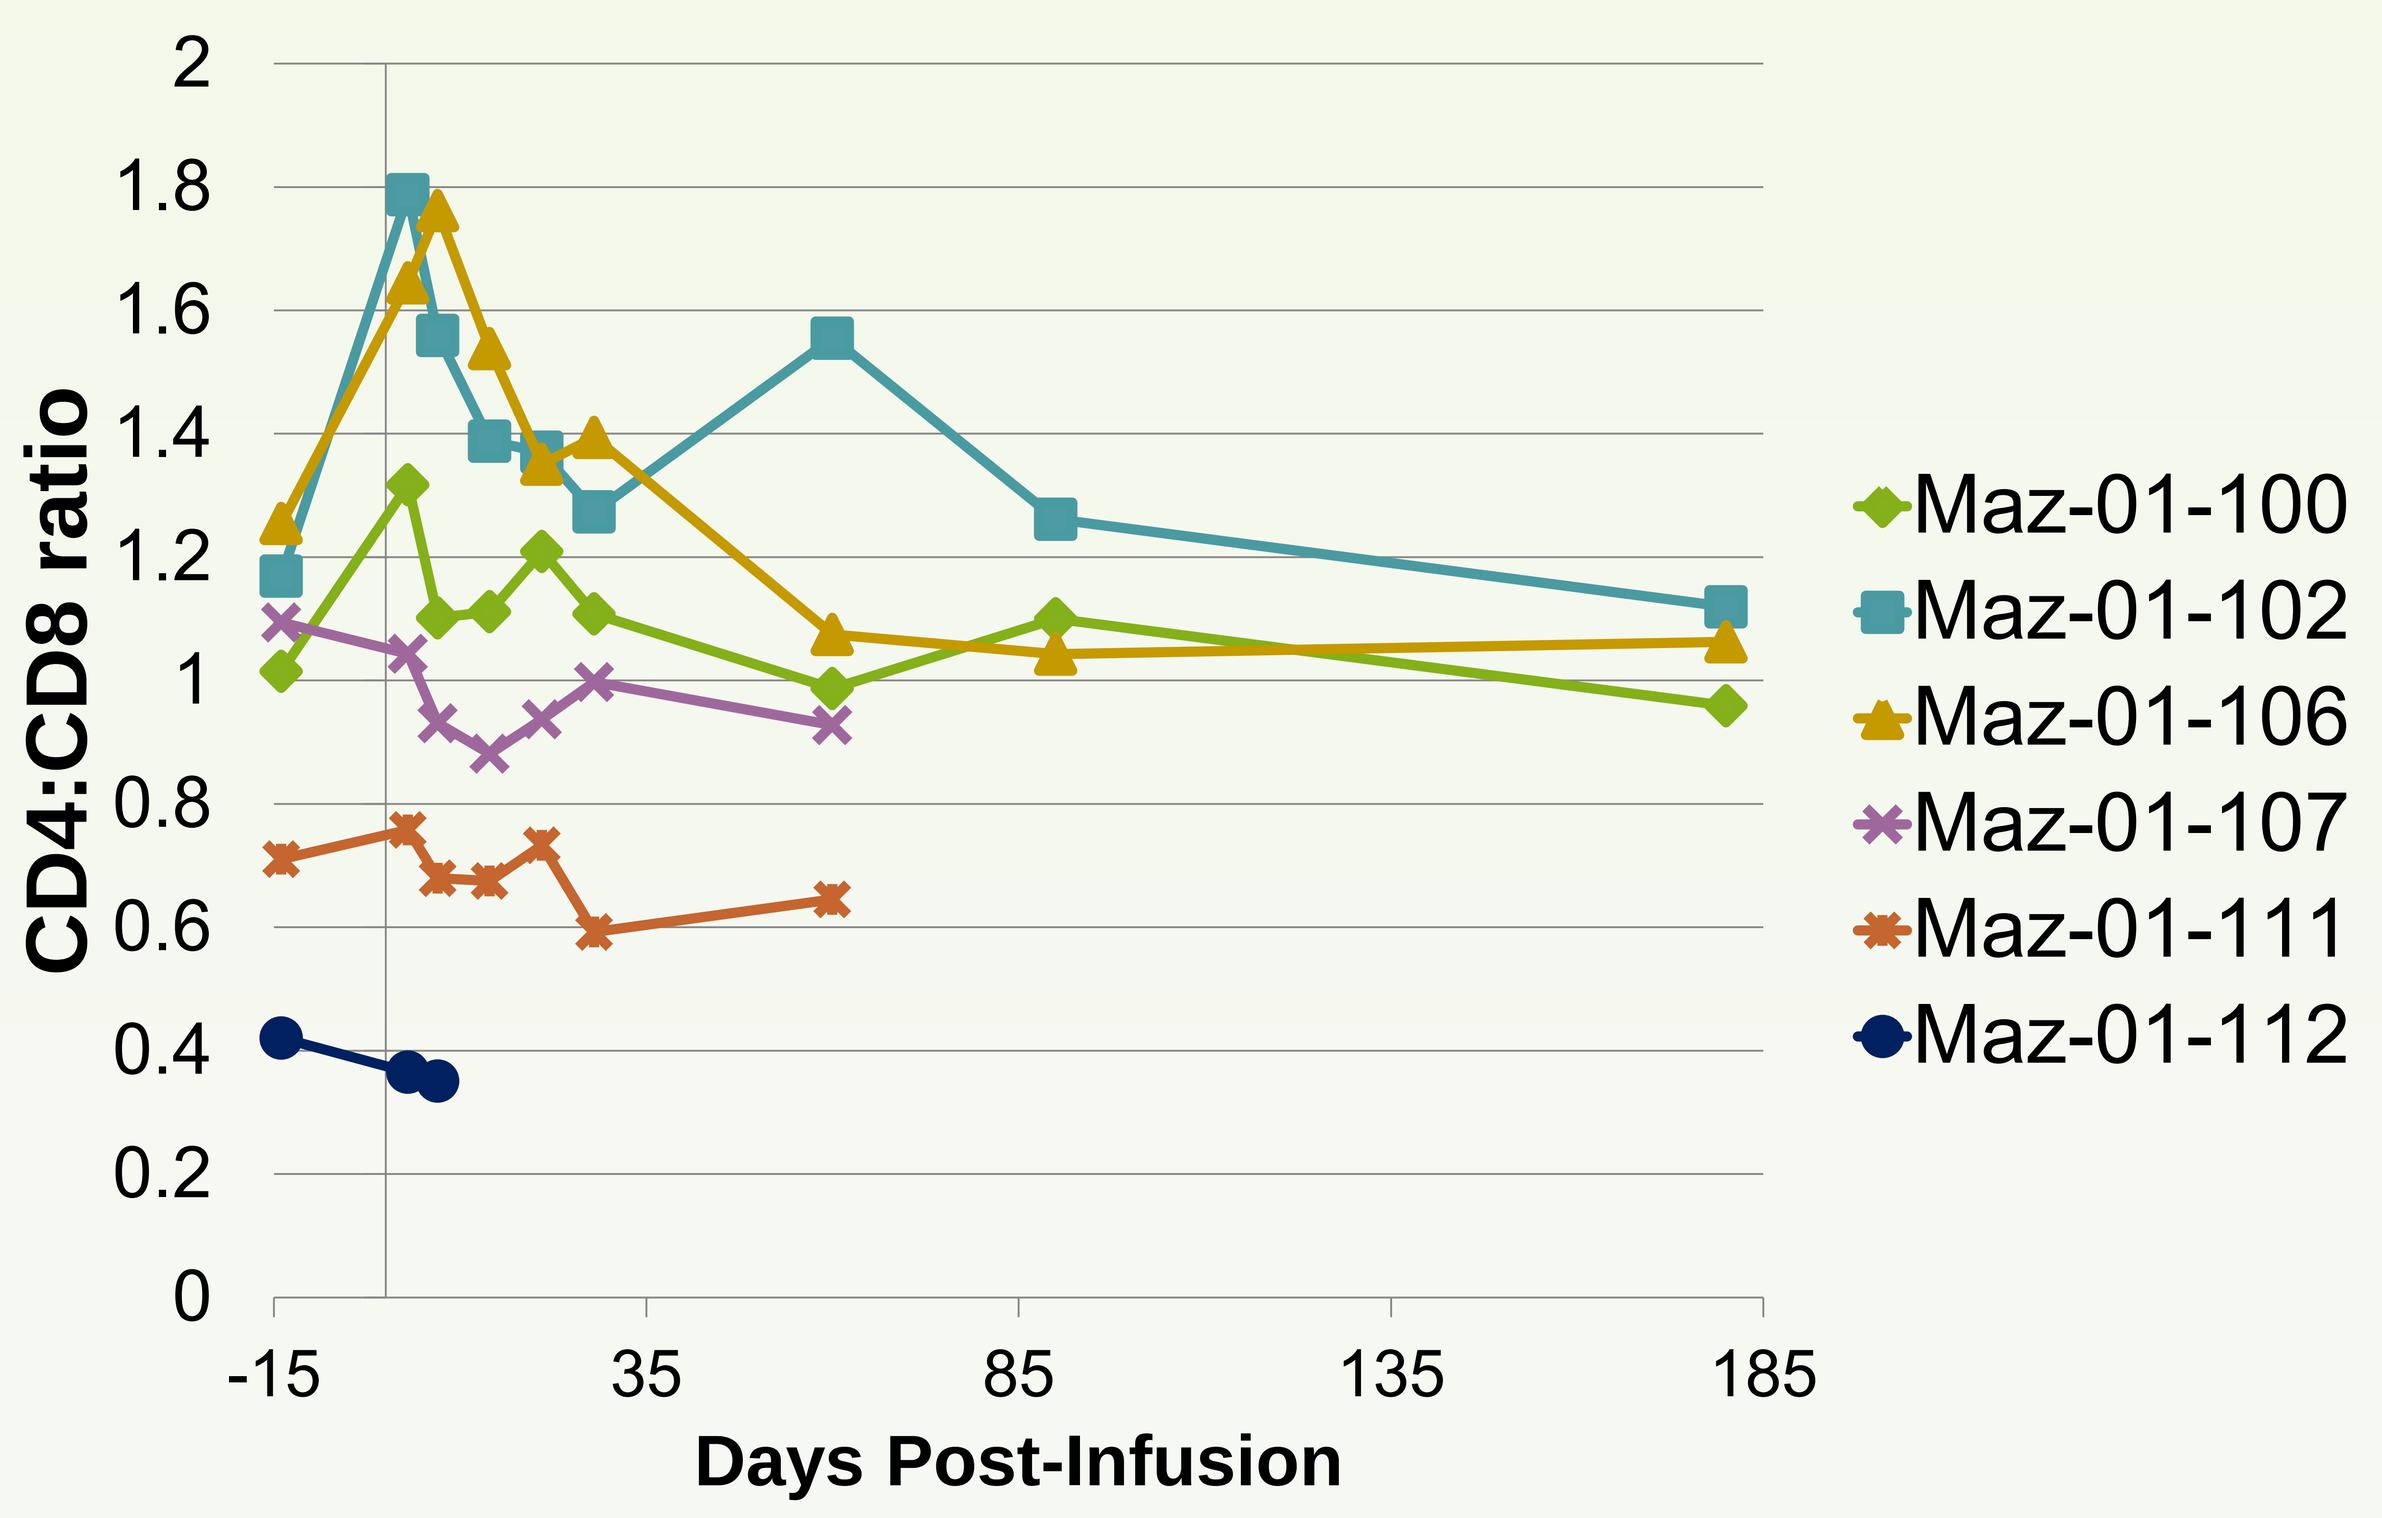

CD4:CD8 ratio
2
1.8
1.6
1.4
1.2
1
0.8
0.6
0.4
0.2
0
-15
35
85
135
185
Days Post-Infusion
Maz-01-100
Maz-01-102
Maz-01-106
Maz-01-107
Maz-01-111
Maz-01-112

Figure 3. CD4:CD8 T cell Ratio. The CD4:CD8 T cell ratio in Cohort 1 increased at least marginally in 4 of 6 subjects within 3 days of MazF T cell infusion.

Results

Persistence of MazF-modified cells in blood- Cohort 1

A

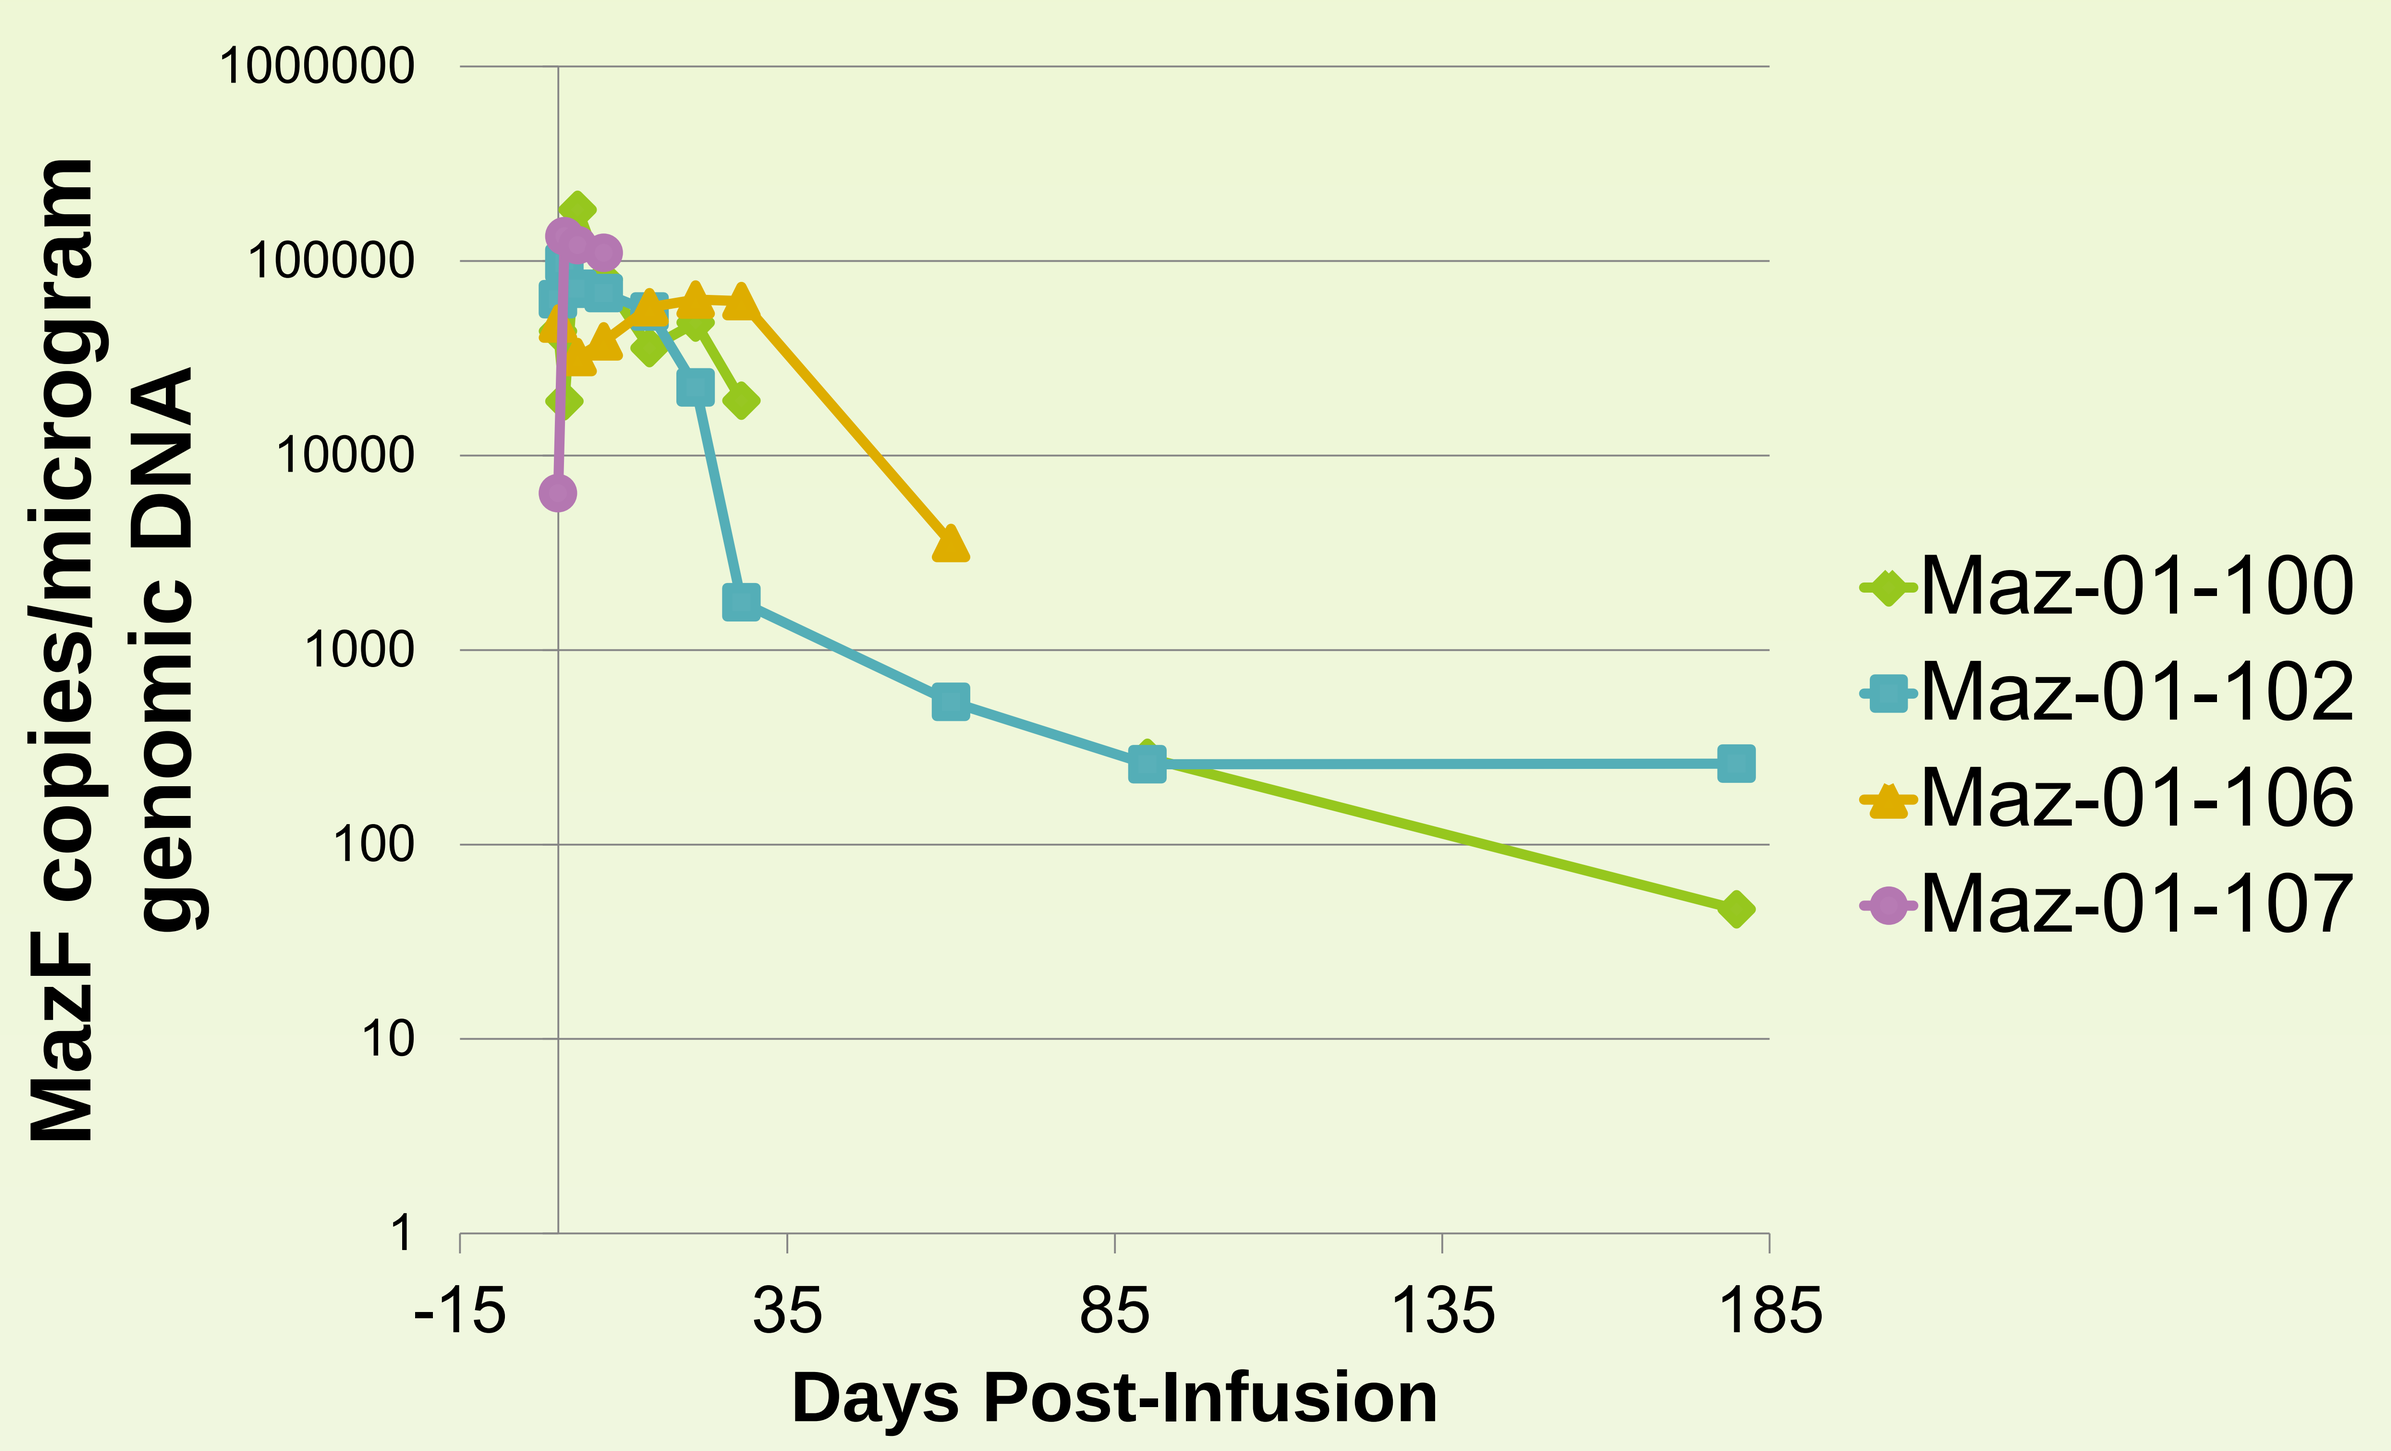

MazF copies/microgram genomic DNA
1000000
100000
10000
1000
100
10
1
-15
35
85
135
185
Days Post-Infusion
Maz-01-100
Maz-01-102
Maz-01-106
Maz-01-107

B

Study ID#	Safety Eval	Day 0 Post-infusion	Day 1	Day 3	Day 7	Day 14	Day 21	Day 28	Day 60	Day 90	Day 180
100	0	43547	18956	183260	80154	35667	48152	19105	0	279	47
102	0	63540	97080	71812	68205	54984	22375	1756	540	258	261
106	0	47594		32073	38579	57675	63200	62224	3542		
107	0	6384	134088	120394	109809						

Figure 4. MazF-modified CD4 T cell persistence in peripheral blood. MazF DNA signal was detected by qPCR in the peripheral blood of all subjects analyzed to date, including 2 subjects who have completed the final study visit at Day 180 post infusion. Data is shown graphically in panel (A). Data values expressed as MazF copies/microgram of genomic DNA are listed in the table (B).

Conclusion

Preliminary results suggest that autologous MazF-modified CD4⁺ T cells are safe and well-tolerated in aviremic HIV+ subjects and that MazF-T are able to persist at least 6 months post-infusion. Future results from Cohort 2, in which subjects participate in an analytical treatment interruption, are expected to further elucidate the anti-HIV effects associated with MazF-T cells in the presence of active HIV replication.

Funding Source:
Takara Bio Inc.
http://www.takara-bio.com