0338

Phase I Study of Gene-modified CD4+ Cells and CD34+ Cells With or Without Busulfan in HIV+ adults

Ronald T Mitsuyasu[1], Jay Lalezari[2], Bryan Burke[3], Mollie Barrett[3], Jeremy Casey[3], Alison Knop[3], Suparna Mishra[3], Jeffrey Ahlers[3], Louis Breton[3], Orit Wolstein[3], Jeffrey Bartlett[3], W. David Hardy[4], Louise Evans[3], Geoffrey Symonds[3]

[1]University of California Los Angeles, CA, USA, [2]Quest Clinical Research, San Francisco, CA, USA, [3]Calimmune, Inc., (now CSL Behring L.L.C) Pasadena, CA, USA, [4]John Hopkins University School of Medicine, Baltimore, MD, USA

BACKGROUND

It is estimated that >36 million individuals are currently living with HIV. HIV/AIDS is a disease that impairs immune function, primarily by decreasing CD4+ T lymphocytes.

HIV gene therapy potentially could, with a single treatment, reduce viral load (VL), preserve immunity and mitigate toxicities of antiretroviral therapy (ART). Calimmune, Inc. (now CSL Behring L.L.C), developed the CAL-1 lentiviral vector which encodes both a short hairpin RNA to silence the HIV co-receptor CCR5 (sh5) and the HIV fusion inhibitor peptide C46 (**Figure 1**).

In this cell-derived gene therapy approach, anti-HIV genes are introduced into hematopoietic stem cells (HSC). These HSC would then produce a population of CD4+ T lymphocytes that are protected from the effects of HIV.

Safety and efficacy have been demonstrated in pre-clinical *in vitro* and *in vivo* studies. Efficacy studies have shown effective marking of human target cells and effective expression of the therapeutic genes. Moreover, the HIV challenge assays demonstrate protection against a range of laboratory and primary viral isolates (Refs 1-3).

METHODS

The combined construct (CAL-1) has been tested in a phase I pilot clinical study in persons living with HIV who had previously been exposed to ART but were then not taking any antiretroviral agent (NCT02390297).

In the clinical study we assessed the safety and feasibility of introducing the anti-HIV-1 gene construct LVsh5/C46 (CAL-1) into autologous CD4+ T-cells (T^{tn}) and HSC (HSC^{tn}) along with non-myeloablative conditioning with busulfan to improve engraftment in HIV+ adults with a history of previous treatment with ART (**Figure 2**).

RESULTS

12 study participants (4 in each cohort) received both T^{tn} and HSC^{tn} cell products. Four participants completed the study without ART, while 8 resumed therapy prior to week 48. The reasons for restarting ART were mainly due to increasing or lack of suppression of plasma VL post transplant. Earliest time point to resume ART was 16 weeks post transplant.

Only 1 SAE was reported which was unrelated to study drug treatment.

Main Points:

Outpatient delivery of gene modified hematopoietic stem cell (HSC^{tn}) and CD4 cells (T^{tn}) was feasible and well tolerated

Use of **low- to moderate- dose busulfan** could be safely administered and tolerated in this population of HIV+ volunteers

The potential impact of HSC^{tn} and T^{tn} could not be fully assessed because of the *low level of gene marking* in peripheral blood

Busulfan dose correlated with higher level of marking

Potential reasons for the lack of long-term survival of CAL-1 transduced cells include persistent *HIV viremia* and associated *inflammation*

Further research should focus on continuing to *improve the engraftment* of gene modified cells in individuals living with HIV with *fully suppressed HIV* on antiretroviral therapy

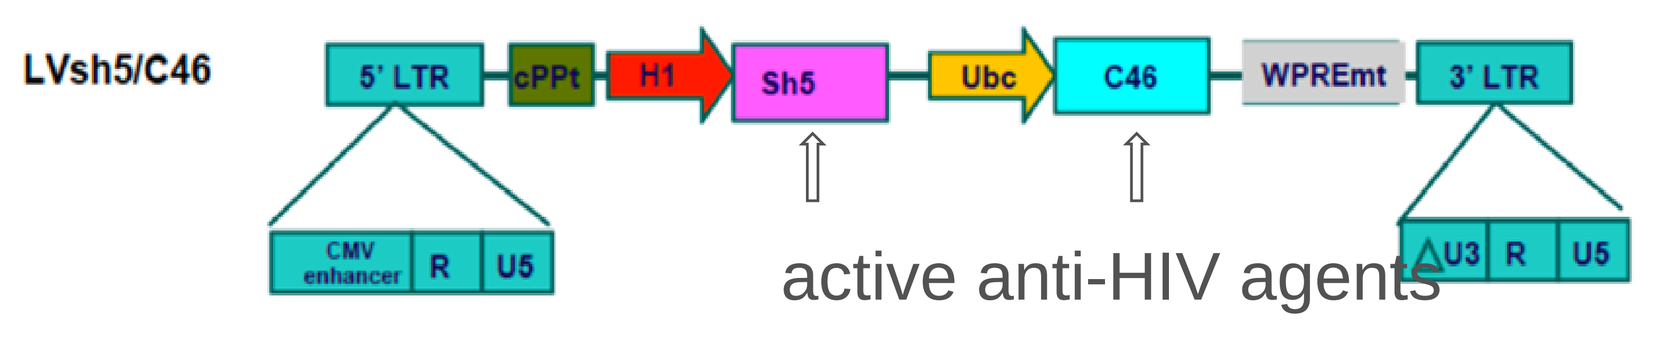
LVsh5/C46
5' LTR
cPPt
H1
Sh5
Ubc
C46
WPREmt
3' LTR
CMV enhancer
R
U5
ΔU3
active anti-HIV agents

Figure 1: CAL-1 Lentiviral vector

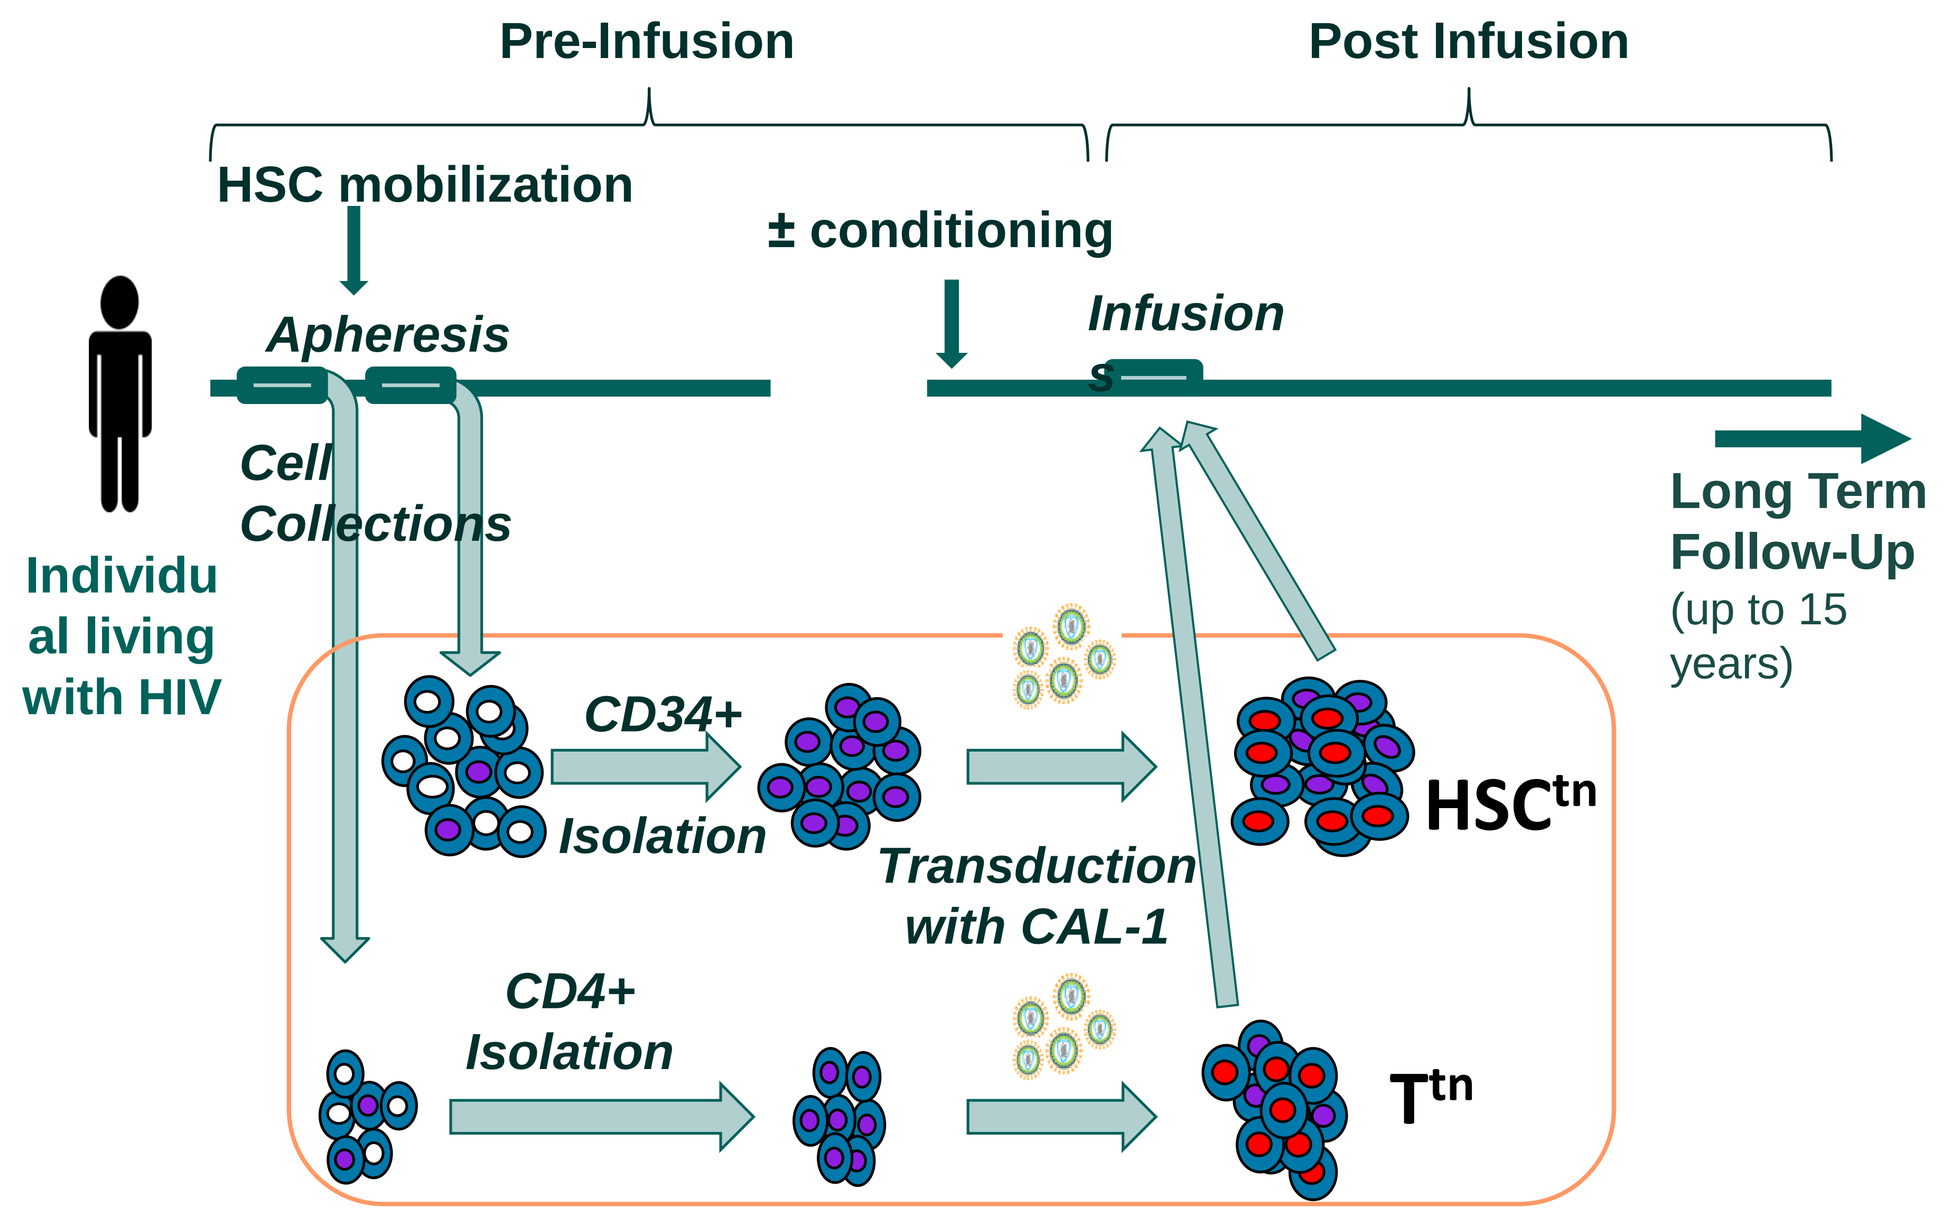
Pre-Infusion
Post Infusion
HSC mobilization
± conditioning
Apheresis
Infusion
Cell Collections
Individual living with HIV
CD34+ Isolation
Transduction with CAL-1
CD4+ Isolation
Long Term Follow-Up
(up to 15 years)

Figure 2: Clinical Study Design

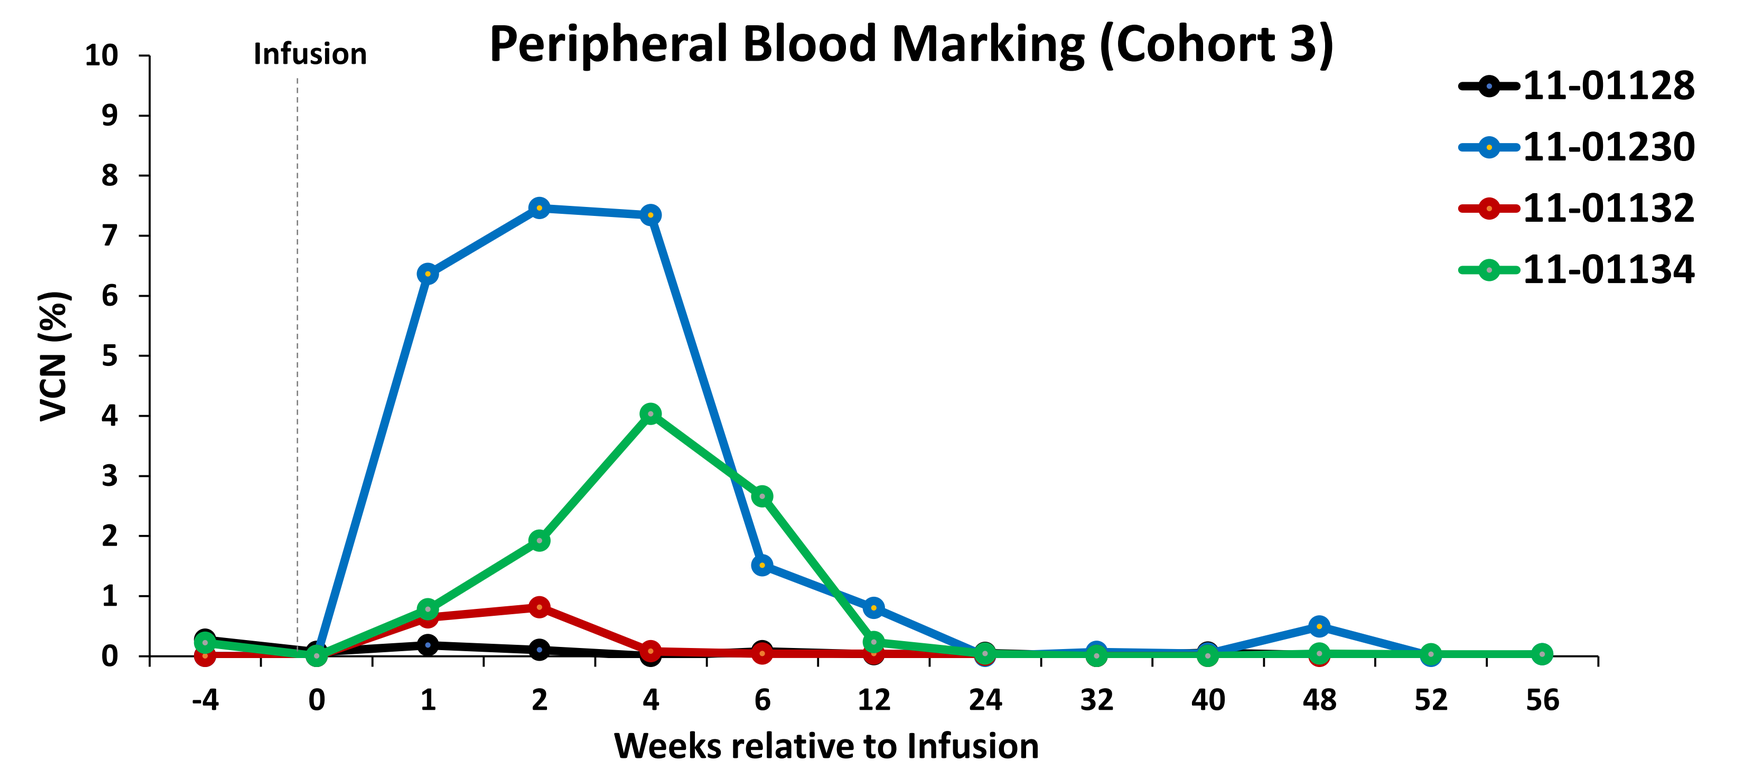
Peripheral Blood Marking (Cohort 3)
Infusion
VCN (%)
Weeks relative to Infusion
11-01128
11-01230
11-01132
11-01134

Figure 3: Cohort 3 marking showed the greatest extent of Cal-1 marking in terms of peak values and time period, but marking was not sustained

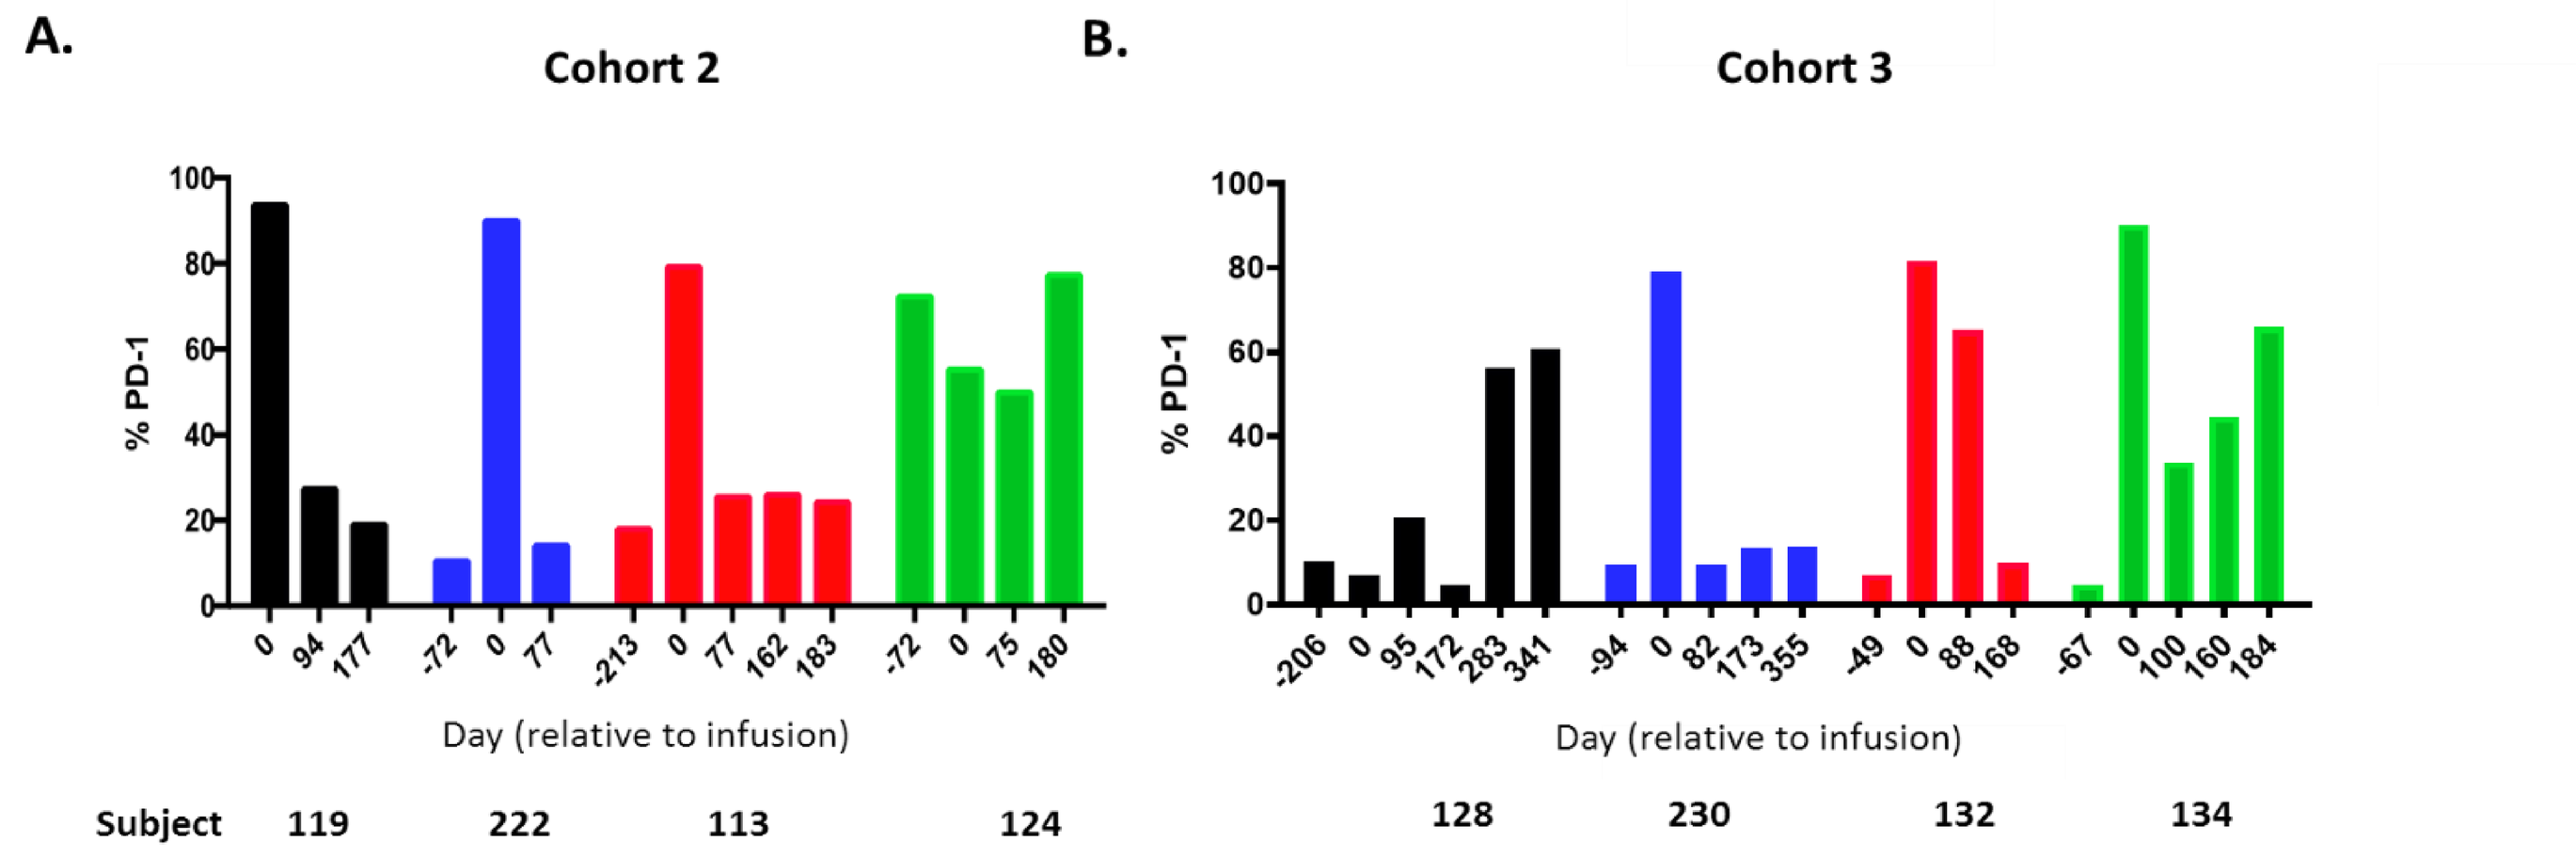
A.
Cohort 2
% PD-1
Day (relative to infusion)
Subject
119
222
113
124
B.
Cohort 3
128
230
132
134

Figure 4: Expression of exhaustion inflammatory marker PD-1 in cohorts 2 (A) & 3 (B) in the first 6 months post-infusion. Day 0 is Infusion day.

Study procedure-related AEs included neutropenia, thrombocytopenia, fatigue, nausea and back pain. All study participants who received busulfan developed transient, asymptomatic grade 4 neutropenia and or grade 4 thrombocytopenia. No consequent fever, infections or bleeding was seen in any participant.

Only 1 patient in cohort 1 had CAL-1 marking in peripheral blood (PB) >1% at Week 4 which was not sustained. All cohort 2 patients had >1% marking at early time points, and cohort 3 showed the highest levels of CAL-1 marking at peak and longest persistence (**Figure 3**), however marking in all cohorts was not sustained.

While no association was seen between CAL-1 marking and T^{tn} dose, there was a correlation between HSC^{tn} dose and peak CAL-1 marking in cohort 3. No significant marking was observed in the GALT samples for all cohorts. Very low levels of CAL-1 marking was seen in BM. Increased busulfan AUC was correlated with increased peak CAL-1 marking in PB. There was no effect of cell infusions on plasma HIV RNA. As anticipated, absolute CD4+ counts were reduced following apheresis but otherwise remained stable over time in Cohort 1 participants, who were not treated with busulfan and had lower apheresis volumes. There was a greater effect on CD4+ counts in Cohorts 2 and 3 subjects who received busulfan and had larger apheresis volumes. Conversely, percentages of CD4+ cells across all cohorts were relatively stable.

CONCLUSIONS

Delivery of HSC^{tn} and T^{tn} was feasible, well-tolerated, and the overall AE profile was generally consistent with the known AE profile of low- to moderate-dose busulfan coupled with large volume apheresis following mobilization in the context of ongoing viremia. No CAL-1-related SAE was observed.

This study demonstrated that low- to moderate-dose busulfan could be safely administered as an outpatient in this population of HIV+ patients. There was evidence that busulfan dose was correlated with higher levels of CAL-1 marking, however, the potential impact of HSC^{tn} and T^{tn} on the control of HIV replication could not be fully assessed because of the low level of marking of CAL-1 cells in PB. Potential reasons for the lack of long-term survival of CAL-1-transduced cells include persistent HIV viremia and associated inflammation. The relatively low transduction efficiency of the T^{tn} and HSC^{tn} products and the high frequency of cells expressing PD-1 on the day of infusion (**Figure 4**) also may have contributed to the low level of CAL-1 transduced cell survival.

Future research should focus on administering gene modified stem cells in virally suppressed individuals on ART at the time of cell infusion and techniques to enhance engraftment.

Acknowledgements

UCLA CFAR grant (P30 AI028697) and the UCLA CTRC grant (UL1 TR0001881). Both are USPHS, DHHS, NIH grants.

References:

1. Peterson CW et al. 2016. Mol Ther Methods Clin Dev.
2. Burke et al., 2015 Mol Ther Nucleic Acids; 4: e236.
3. Wolstein et al., 2014. Mol Ther Methods Clin Dev. 1:11